DX39

Pregnancy Outcomes With Teriflunomide: Female Patients and Partners of Male Patients

Lily Jung Henson,[1] Olaf Stuve,[2] Myriam Benamor,[3] Sandrine Turpault,[4] Françoise Menguy-Vacheron[3]

[1]Swedish Medical Center, Issaquah, WA, USA; [2]Department of Neurology, University of Texas Southwestern Medical Center at Dallas, TX, USA; [3]Sanofi R&D, Chilly-Mazarin, France; [4]Sanofi R&D US, Bridgewater, NJ, USA

BACKGROUND

- Teriflunomide is a once-daily, oral disease-modifying therapy approved for the treatment of patients with relapsing forms of multiple sclerosis (MS).[1]
 - Exposure across more than 10 years of the teriflunomide clinical program exceeds 3700 patient-years.
- Preclinical evidence regarding embryofetal toxicity demonstrates that teriflunomide:[1]
 - is non-mutagenic in vitro
 - does not affect chromosomes (mice, rats, hamsters) or fertility (rats)
 - has teratogenic effects in rats and rabbits.
- Teriflunomide is the principal active metabolite of leflunomide, used in the treatment of rheumatoid arthritis (RA) since 1998.[2]
 - Embryo-lethality and teratogenicity were observed in leflunomide animal studies.
 - In the prospective Organization of Teratology Information Services (OTIS) study, patients with RA were exposed to leflunomide in the first trimester of pregnancy and underwent accelerated elimination.[3]
 - No significant differences were observed in the overall rate of major structural defects compared with disease-matched and healthy controls, there was no evidence of teratogenicity, and the rate of spontaneous abortion was similar to that in the general population.[3,4]
- Based on animal studies, women of childbearing potential must use reliable contraception when receiving teriflunomide.[1]
- Teriflunomide has been detected in human semen; animal studies to evaluate risk of male-mediated fetal toxicity have not been conducted. In humans, the estimated female exposure via semen of a male treated patient is expected to be 100 times lower than the steady-state plasma exposure with 14 mg oral dosing. However, to minimize any possible risk, patients receiving teriflunomide should use reliable contraception.[1]
- If pregnancy is desired, patients—female or male—must discontinue treatment and undergo an elimination procedure with cholestyramine or activated charcoal until teriflunomide plasma concentrations are <0.02 µg/mL (**Figure**).

Figure. Plasma Elimination of Teriflunomide Using Accelerated Elimination Procedure

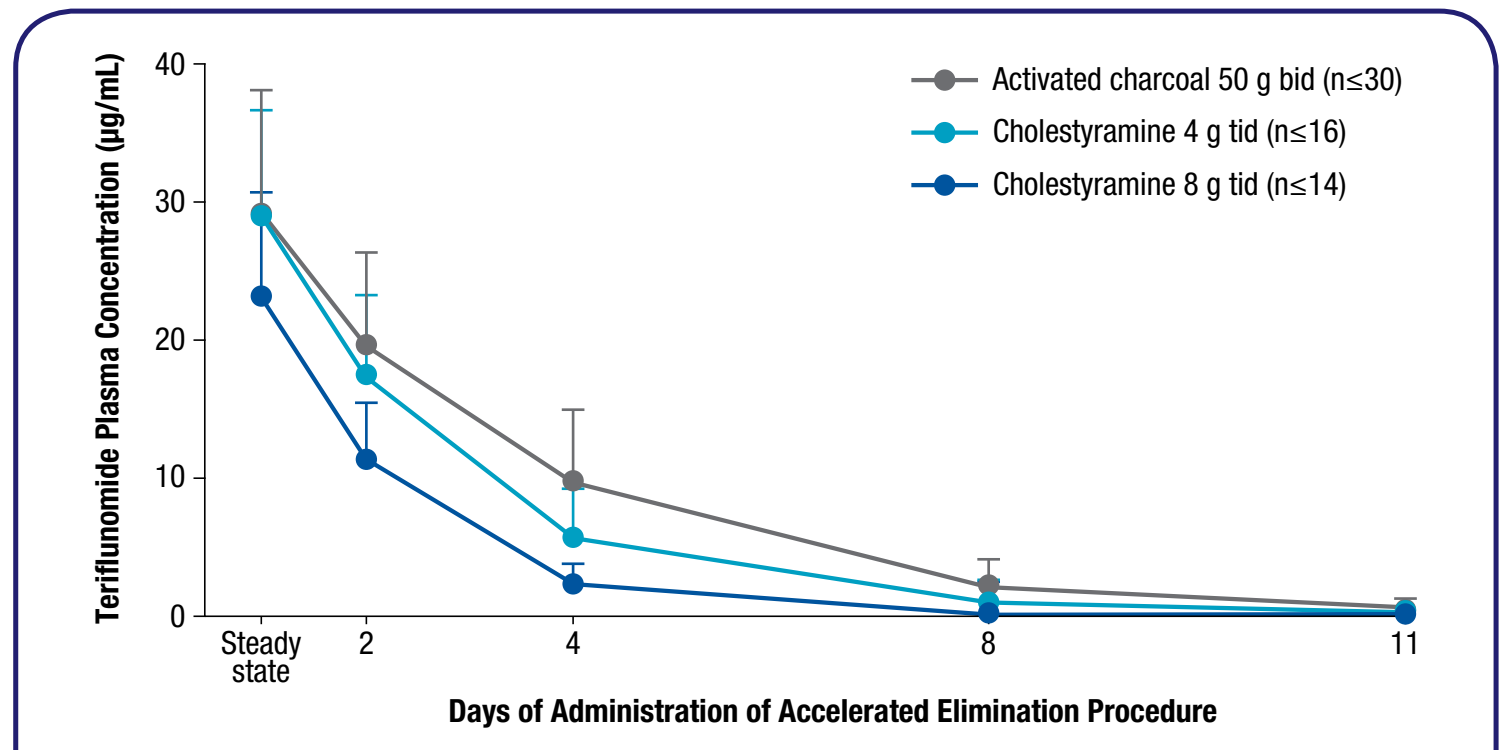
Teriflunomide Plasma Concentration (µg/mL)
40
30
20
10
0
Activated charcoal 50 g bid (n≤30)
Cholestyramine 4 g tid (n≤16)
Cholestyramine 8 g tid (n≤14)
Steady state
2
4
8
11
Days of Administration of Accelerated Elimination Procedure

Teriflunomide elimination can be accelerated by either of the following procedures:
- Administration of cholestyramine 8 g three times daily (tid) for 11 days. If cholestyramine 8 g tid is not well tolerated, cholestyramine 4 g tid can be used.
- Administration of 50 g oral activated charcoal powder twice daily (bid) for 11 days.

OBJECTIVE

- Summarize pregnancy outcomes in female patients and partners of males exposed to teriflunomide across 9 phase 2/3 clinical studies in the MS clinical development program.

METHODS

- Patients received teriflunomide (7 or 14 mg), interferon-β, placebo, or a combination of treatments in 9 different clinical studies (**Table 1**).
- Although reliable contraception was required during the studies for women of childbearing potential and men with partners of childbearing potential, a number of pregnancies were reported.

Table 1. Teriflunomide Clinical Development Program

Study / Phase	Treatment Arms	Treatment Duration	Randomized†
Monotherapy Studies			
Phase 2* / 2	Placebo, 7 mg, 14 mg teriflunomide	36 weeks	179 (Females: Placebo 41, 7 mg teriflunomide 46, 14 mg teriflunomide 45)
TERIVA / 2	IFN β, 7 mg, 14 mg teriflunomide	28 days, vaccination on Day 1	128 (Females: IFN β 32, 7 mg teriflunomide 28, 14 mg teriflunomide 29)
TEMSO* / 3	Placebo, 7 mg, 14 mg teriflunomide	108 weeks	1088 (Females: Placebo 275, 7 mg teriflunomide 255, 14 mg teriflunomide 255)
TOWER* / 3	Placebo, 7 mg, 14 mg teriflunomide	Range 48–152 weeks (mean 78 weeks)	1169 (Females: Placebo 273, 7 mg teriflunomide 300, 14 mg teriflunomide 258)
TENERE* / 3	IFN β-1a, 7 mg, 14 mg teriflunomide	Range 48–118 weeks (median 64 weeks)	324 (Females: IFN β 71, 7 mg teriflunomide 70, 14 mg teriflunomide 78)
TOPIC* / 3	Placebo, 7 mg, 14 mg teriflunomide	108 weeks	618 (Females: Placebo 135, 7 mg teriflunomide 130, 14 mg teriflunomide 154)
Adjunctive Therapy Studies			
Teri + GA* / 2	GA + Placebo or 7 mg, 14 mg teriflunomide	24 weeks	123 (Females: GA 32, 7 mg teriflunomide 33, 14 mg teriflunomide 32)
Teri + IFN β* / 2	IFN β + Placebo or 7 mg, 14 mg teriflunomide	24 weeks	118 (Females: IFN β + placebo 31, IFN β + 7 mg teriflunomide 25, IFN β + 14 mg teriflunomide 25)
TERACLES / 3	IFN β + Placebo or 7 mg, 14 mg teriflunomide	Variable	534

GA, glatiramer acetate; IFN β, interferon beta. *Study designs included optional extension. †Not all females were of childbearing potential.

- Information (where available) was collected using a Drug Exposure Via Parent form including treatment allocation, pregnancy outcome, patient and fetal exposure, and accelerated elimination procedure.
- The data cut-off was April 9, 2013.

RESULTS

Pregnancies in Female Patients

- Eighty-one pregnancies were reported in female patients across the studies.
 - 63 pregnancies occurred in patients exposed to teriflunomide.
 - 18 pregnancies occurred in patients treated with placebo or interferon-β, or who remained blinded with respect to treatment group; 1 pregnancy was identified at screening.
- Outcomes of the pregnancies are reported in **Table 2**. Of the 25 live births, 20 were born to patients exposed to teriflunomide.

Table 2. Pregnancy Outcomes in Female Patients

	Pregnancy Outcome, n			
Treatment	Live Birth	Induced Abortion	Spontaneous Abortion	Ongoing Pregnancy
Teriflunomide	20	26	12	5
Placebo	2	6	1	0
Interferon-β	2	0	0	0
Blinded therapy	1	3	1	2

- All 20 teriflunomide-treated women giving birth to live infants had already discontinued teriflunomide or discontinued a few days to 11 weeks after becoming pregnant.
 - All patients underwent an accelerated elimination procedure after discontinuing treatment except 2 patients who refused.
 - 7 became pregnant after completing the accelerated elimination procedure.
- All 20 newborns were healthy, without structural or functional problems.
 - Mean known birth weight was 3415 g (range 2780–4150 g) for 12 newborns.
 - Mean gestational age was 38 weeks (range 37–41 weeks).
 - No malformations were reported.
- The rate of spontaneous abortion in the teriflunomide group (19.0%) was within the range reported for the non-MS population.[5]

Pregnancies in Partners of Male Patients

- Twenty pregnancies were reported in partners of 17 men in teriflunomide clinical trials.
 - In 16 pregnancies, the father had been exposed to teriflunomide; in 1 case, the patient remained blinded with respect to treatment group.
- Pregnancy outcomes are reported in **Table 3**.

Table 3. Pregnancy Outcomes in Partners of Male Patients

	Pregnancy Outcome, n			
Treatment	Live Birth	Induced Abortion	Spontaneous Abortion	Ongoing Pregnancy
Teriflunomide	12	1	1	2
Placebo	3	0	0	0
Blinded therapy	0	1	0	0

- All newborns were healthy and free from structural and functional abnormalities.
- There was 1 spontaneous abortion reported in a partner of a man receiving teriflunomide.

CONCLUSIONS

- Teriflunomide clinical data to date have not shown a teratogenic signal, consistent with the findings of the leflunomide OTIS registry.
 - Pregnancy outcomes among women who received teriflunomide, including rates of spontaneous abortion, and gestational age and weight at birth, are consistent with those for the non-MS population.
 - Babies born to mothers or fathers who received teriflunomide had no structural or functional abnormalities at birth.
- Teriflunomide is a therapeutic option for women of childbearing potential, and for male patients with female partners of childbearing potential, when using reliable contraception.
 - If a woman receiving teriflunomide becomes or wishes to become pregnant, she should undergo an accelerated elimination procedure and verify teriflunomide plasma concentrations to be <0.02 µg/mL.
 - If a man receiving teriflunomide has a pregnant partner, he should discontinue treatment and undergo accelerated elimination.
- Planned teriflunomide pregnancy registries will provide additional information.

References

1. Aubagio® (teriflunomide) prescribing information. Genzyme Corporation. 2012.
2. Arava® (leflunomide) prescribing information. sanofi-aventis US LLC. 2011.
3. Chambers CD, et al. *Arthr Rheum.* 2010;62:1494-1503.
4. Cassina M, et al. *Arthr Rheum.* 2012;64:2085-2094.
5. Garcia-Enguidanosa A, et al. *Eur J Obstet Gynecol Reprod Biol.* 2002;102:111-119.

Disclosures

LJH has received research support from Biogen Idec, Genzyme, NIH, Novartis, Sanofi, and Opexa, and has consulted and performed other services for Questcor, Biogen Idec, Genzyme, Novartis, Pfizer, Sanofi, and Teva. OS has received research support from Teva Pharmaceuticals and has consulted and performed other services for Biogen Idec, Genzyme, Novartis, Roche, Sanofi, and Teva Neuroscience. MB, ST, and FMV are employees of Sanofi.

Acknowledgments

This study was supported by Genzyme, a Sanofi company.
Editorial assistance was provided by Melanie Watson, Fishawack Communications, also funded by Genzyme, a Sanofi company.

Fifth Cooperative Meeting of CMSC and ACTRIMS, May 29–June 1, 2013, Orlando, FL, USA