Evaluation of Didox as a Potential Oral Therapy for MS

[1]H.L. Elford, [2]C. Campbell, [2]R. Farrer, [2]C. Papadopoulos, [3]D. Feinstein, [5]J. Dupree, [4]J. Litz, [4]P.J. Yannie, [4]M. Joslyn, [4]A. Marcu, [4]G.H. DeVries

Molecules for Health, Inc., Hines VA Hospital, Hines, IL, Deparment of Anesthesiology, University of Illinois, Chicago, IL, McGuire VA Medical Center, Richmond, V A, Virginia Commonwealth University, Richmond, V A

Introduction

Multiple sclerosis (MS) is a chronic inflammatory autoimmune disease occurring in the central nervous system (CNS). It is a heterogeneous disease clinically and pathologically. The pathological hallmarks are neuroinflammation, myelin and axonal degeneration, and neurodegeneration. The etiology of the disease is unclear but is thought to involve an autoimmune pathogenesis .

Potential MS therapeutic – Didox, a Molecules for Health polyphenolic compound of low molecular weight, has biochemical attributes that have the potential to mitigate several of the pathological processes that are thought to contribute to initiation, promotion and maintenance of MS.

1.Potent free radical scavenger

2.Strong iron chelator

3.Inhibits T- and B-cell proliferation by virtue of being a potent inhibitor of DNA synthesis via its ability to inhibit ribonucleotide reductase

4.Inhibitor of T- and B-cell activation and cytokine production

5.Inhibits NF-kB activation

6.Inhibits migration of immune cells

To determine the potential of didox for MS therapy it was tested in the chronic experimental allergic encephalomyelitis (EAE) mouse model.

Materials and Methods

Chronic EAE was induced in C578Bl/6 mice and splenocytes were prepared as described by Feinstein et al 2002. Nitrite was measured by the standard Griess assay. Cytokines were quantitated via ELISA. Morphological studies were carried out as described by Dupree et.al (1999).

Results

Oral Didox Works!

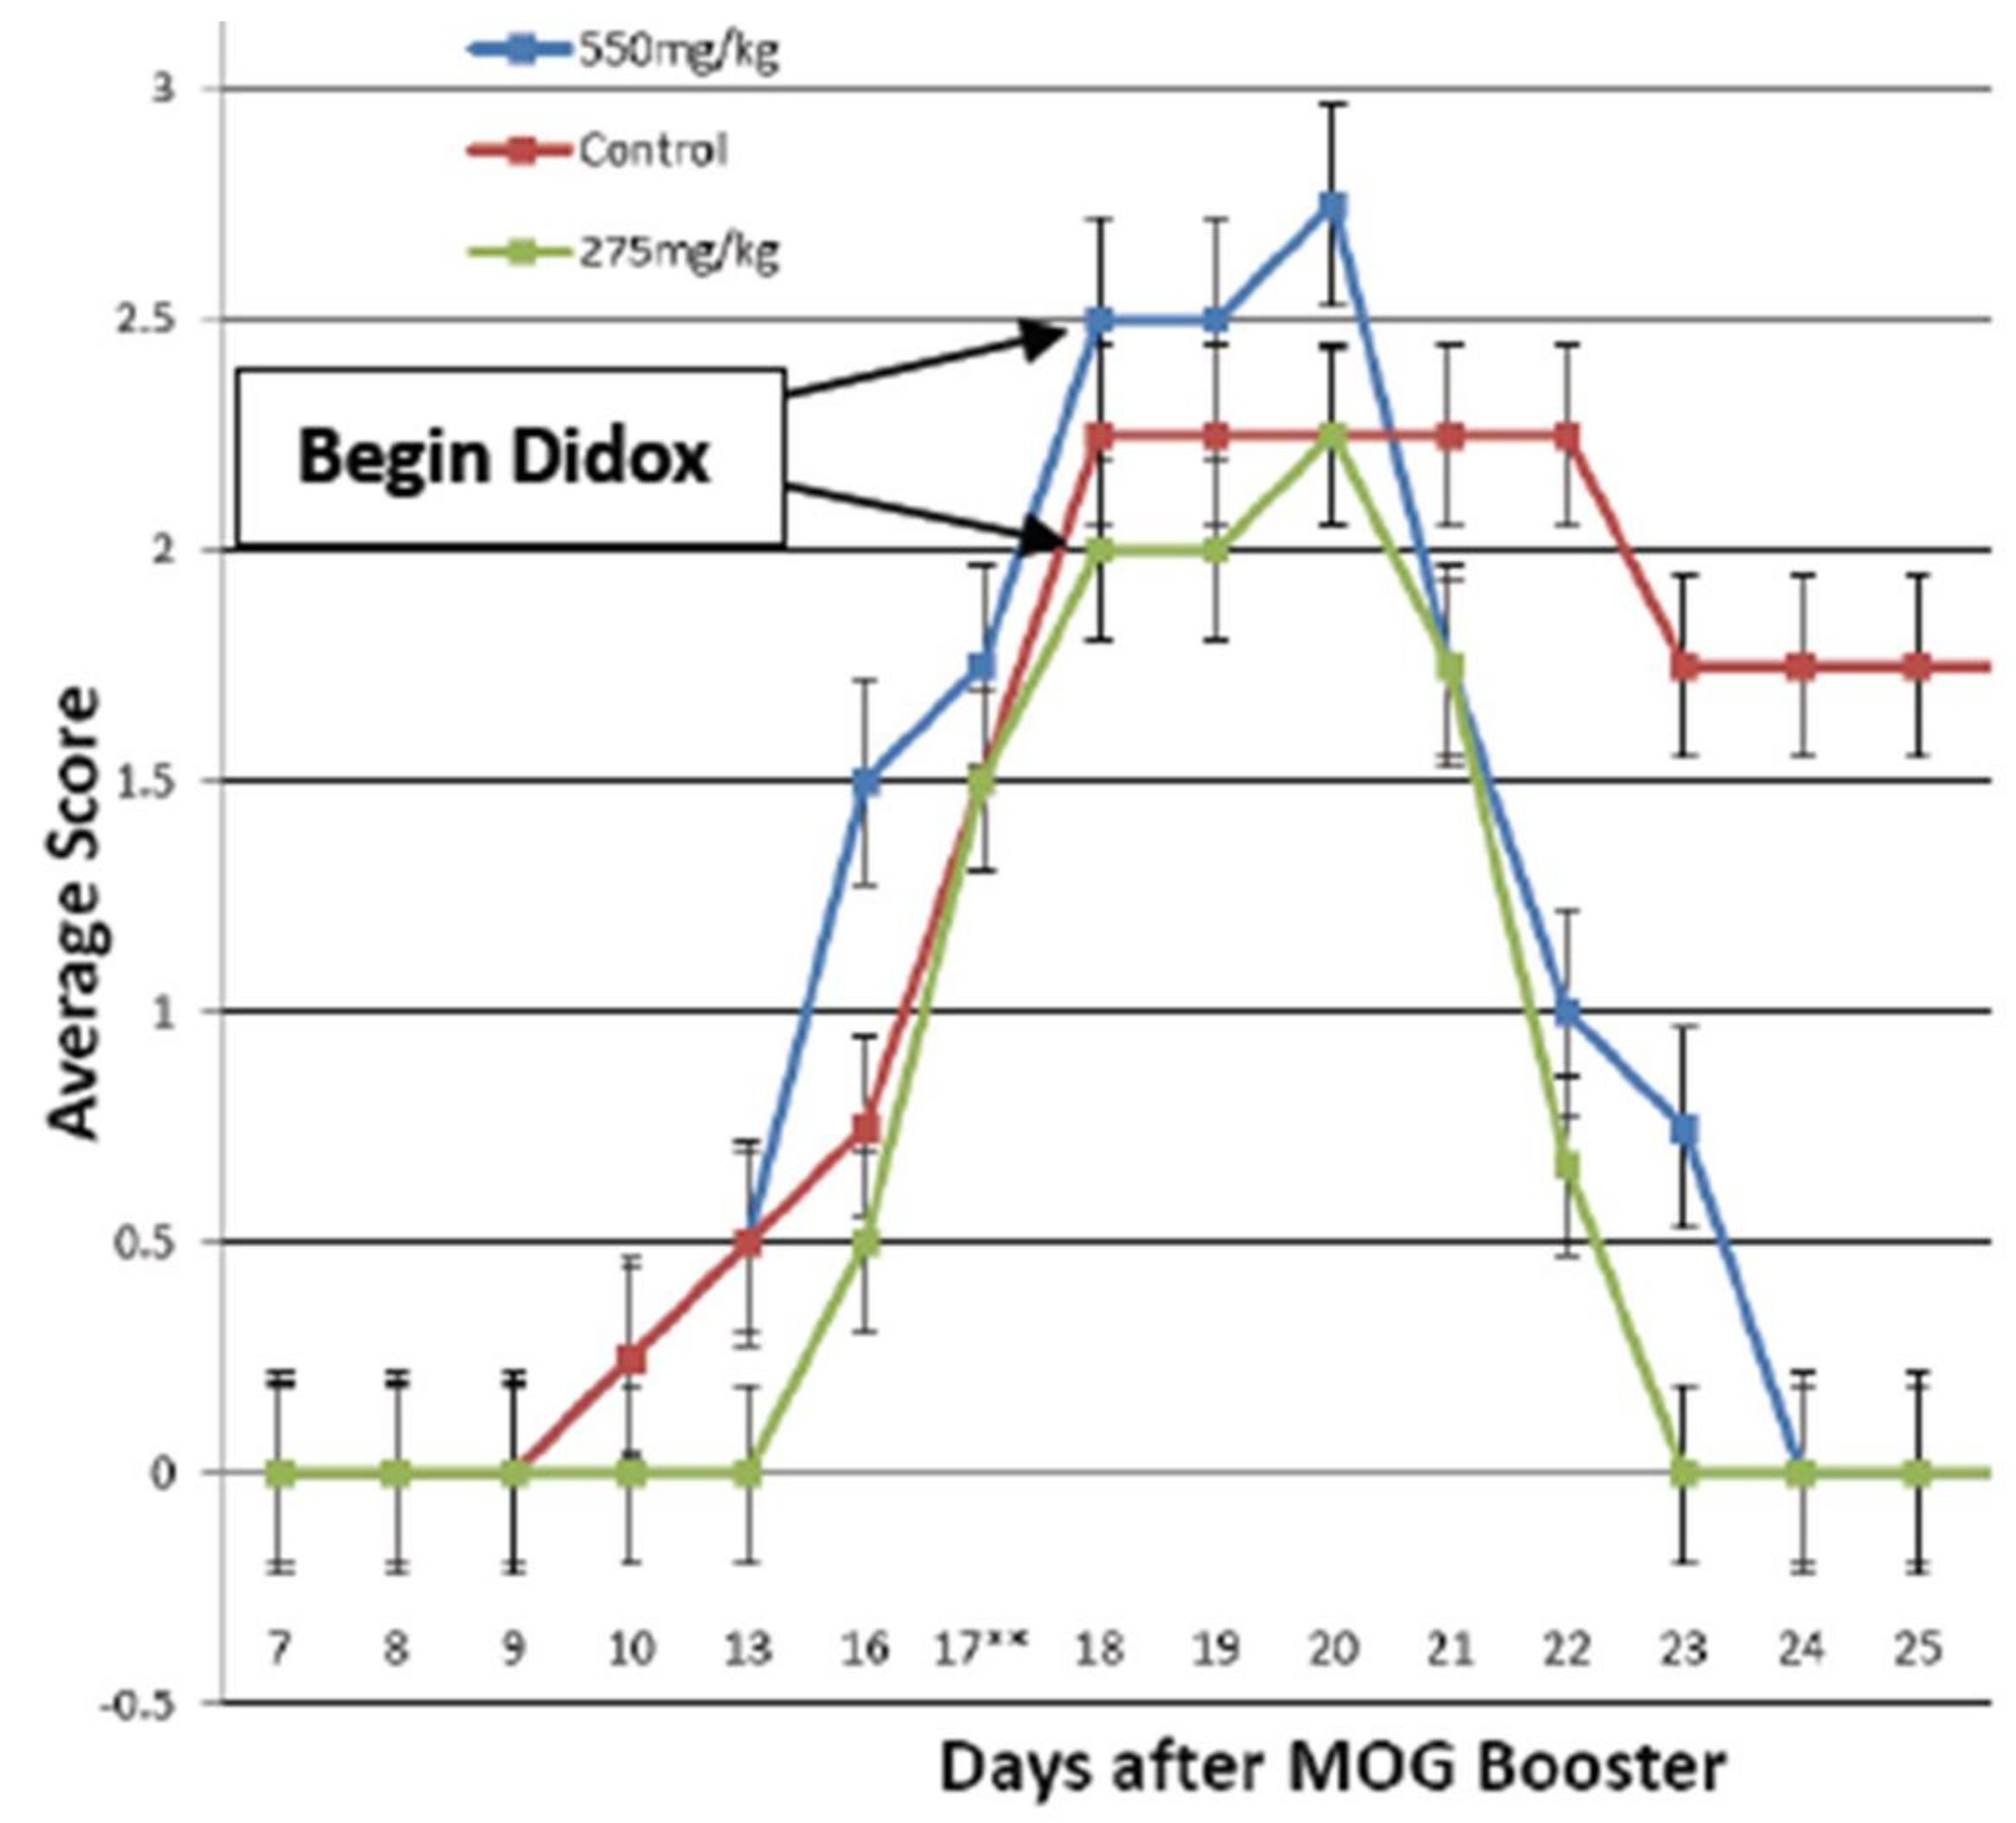
550mg/kg
Control
275mg/kg
Begin Didox
Average Score
Days after MOG Booster

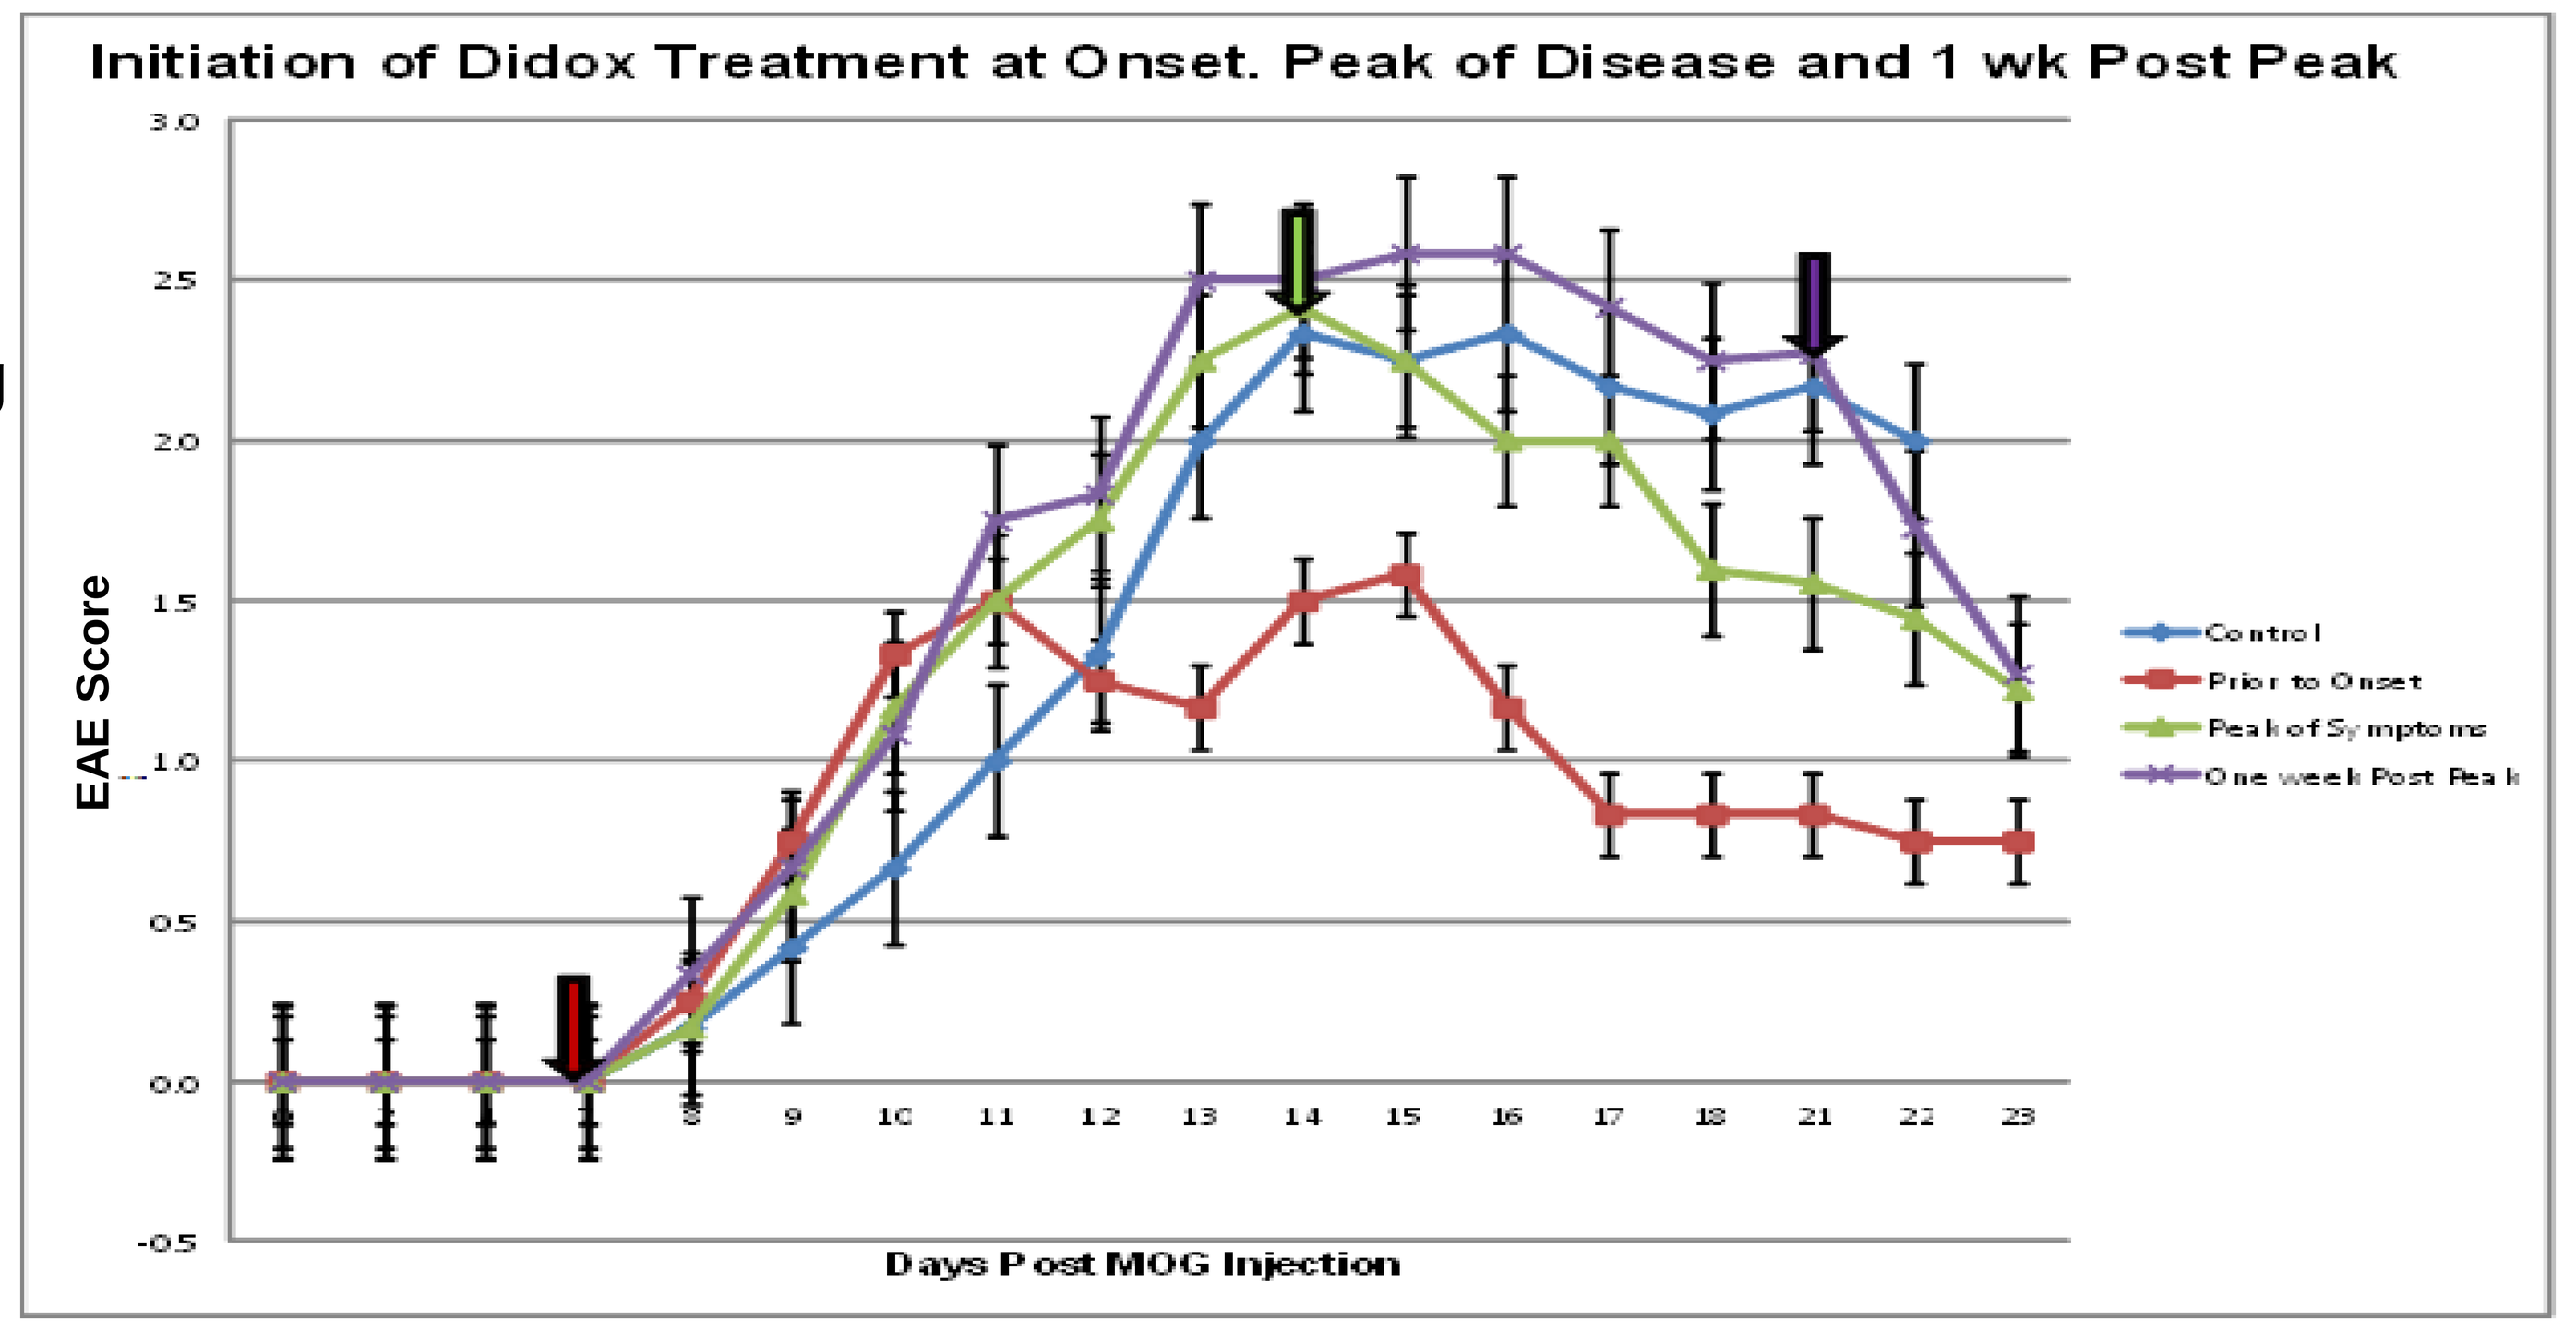
Initiation of Didox Treatment at Onset. Peak of Disease and 1 wk Post Peak
EAE Score
Days Post MOG Injection
Control
Prior to Onset
Peak of Symptoms
One week Post Peak

Didox VS. Mitox (Novanthrone) and Fumarate (BG-12)

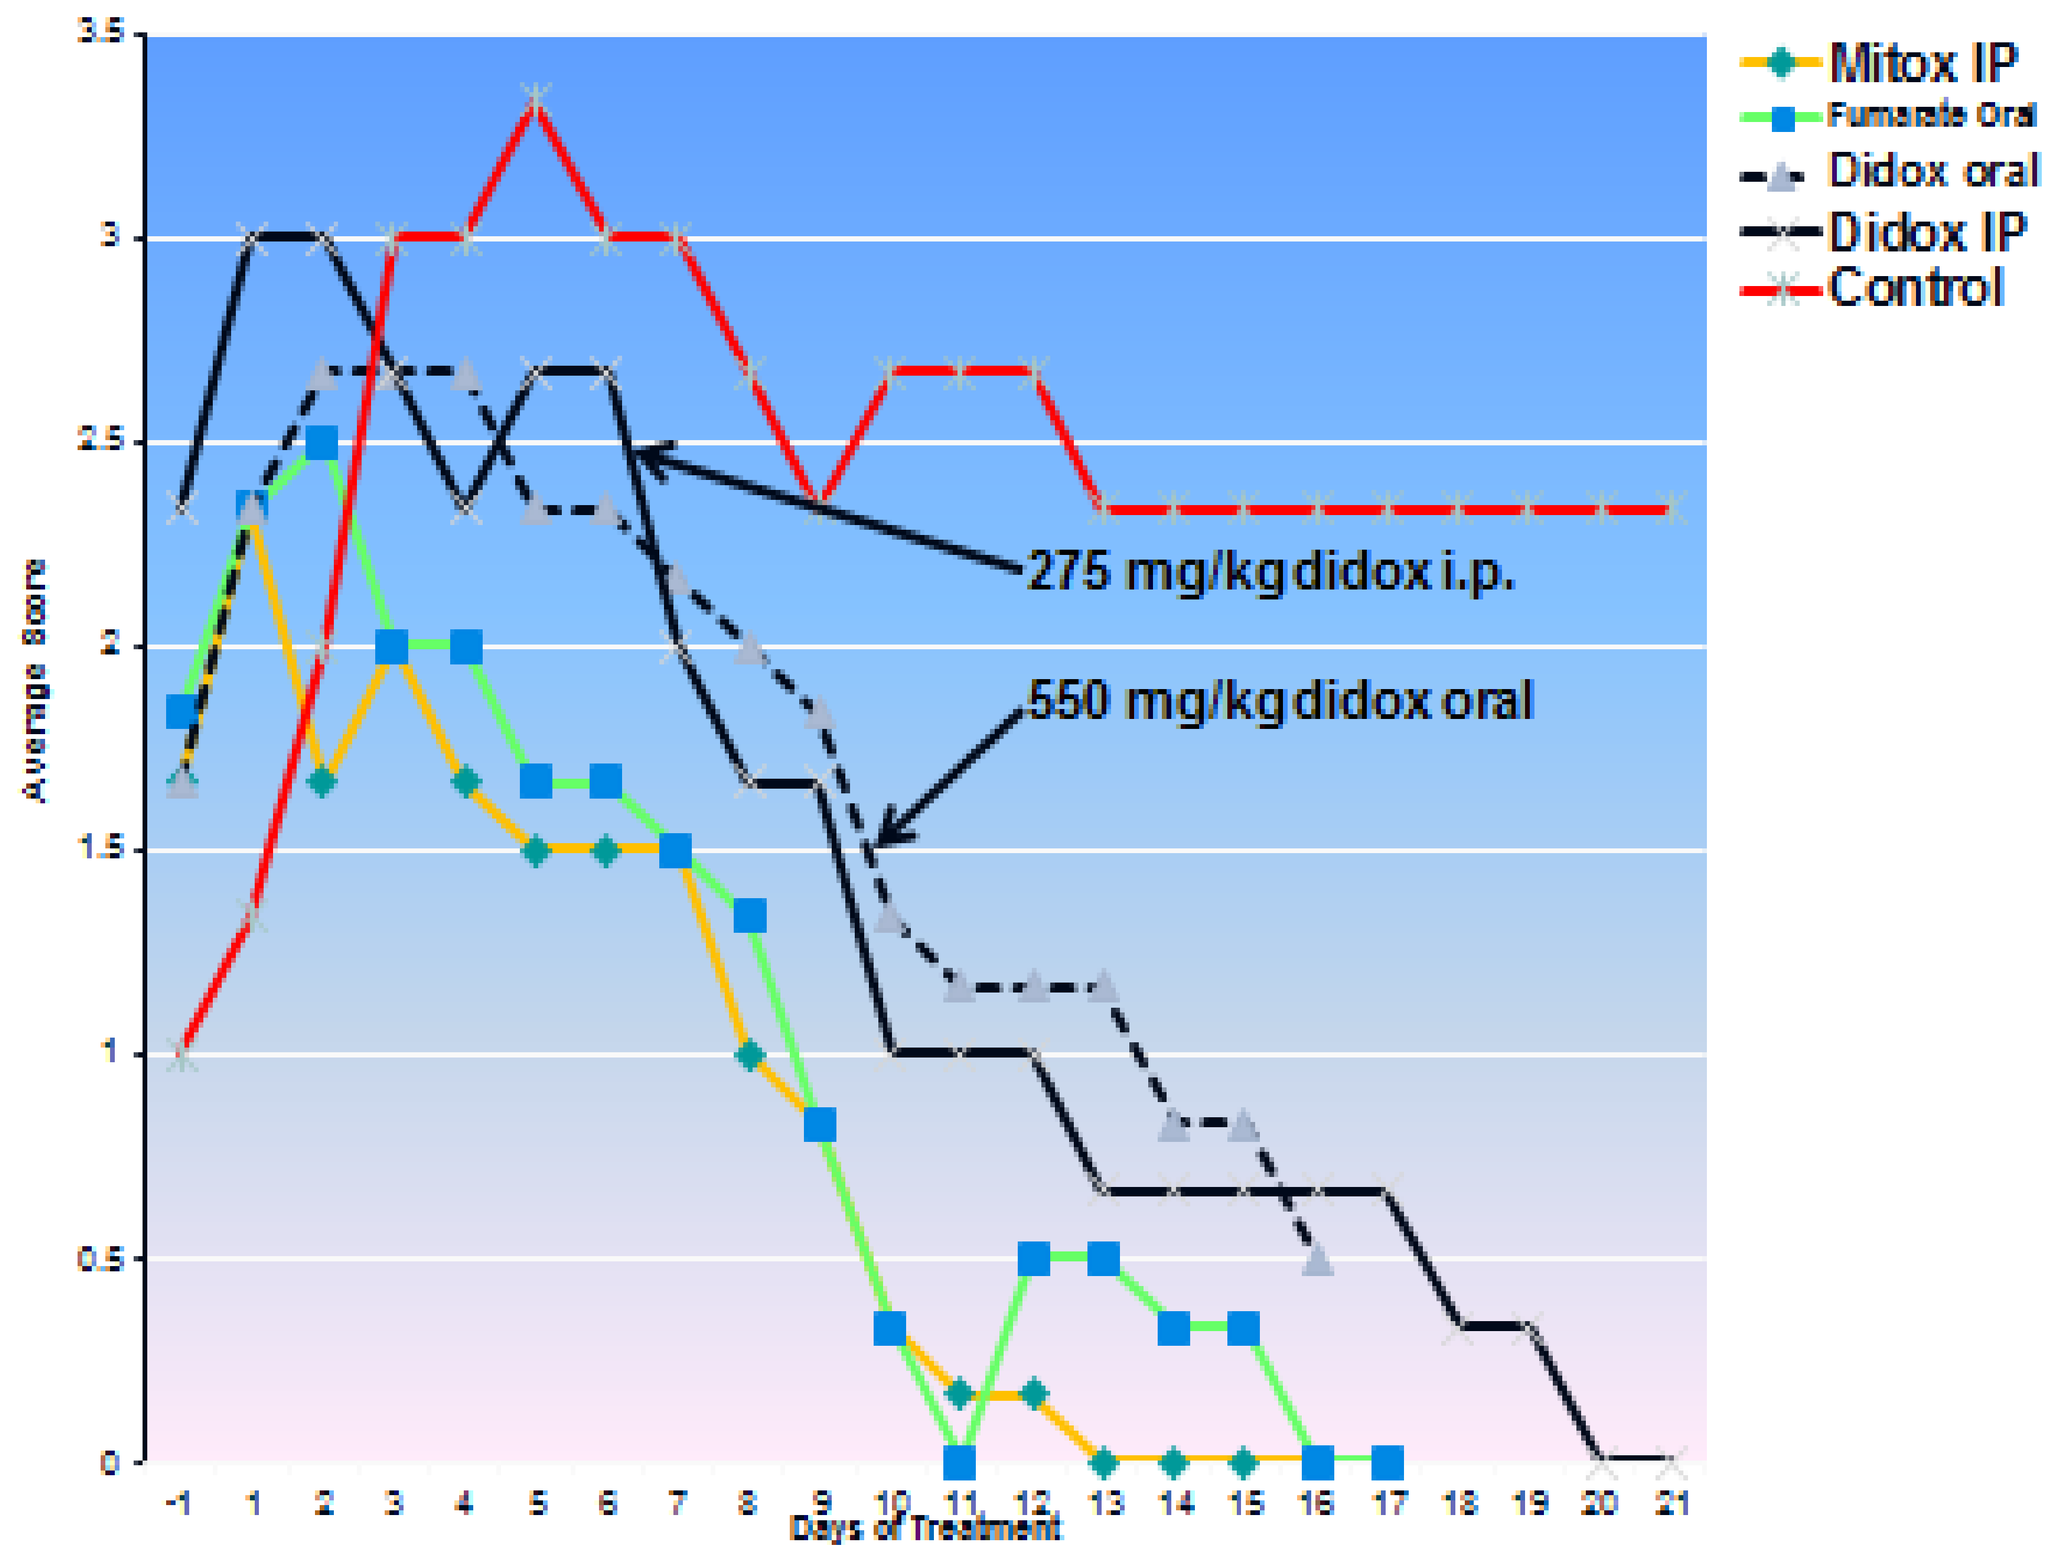
Mitox IP
Fumarate Oral
Didox oral
Didox IP
Control
275 mg/kg didox i.p.
550 mg/kg didox oral
Average Score
Days of Treatment

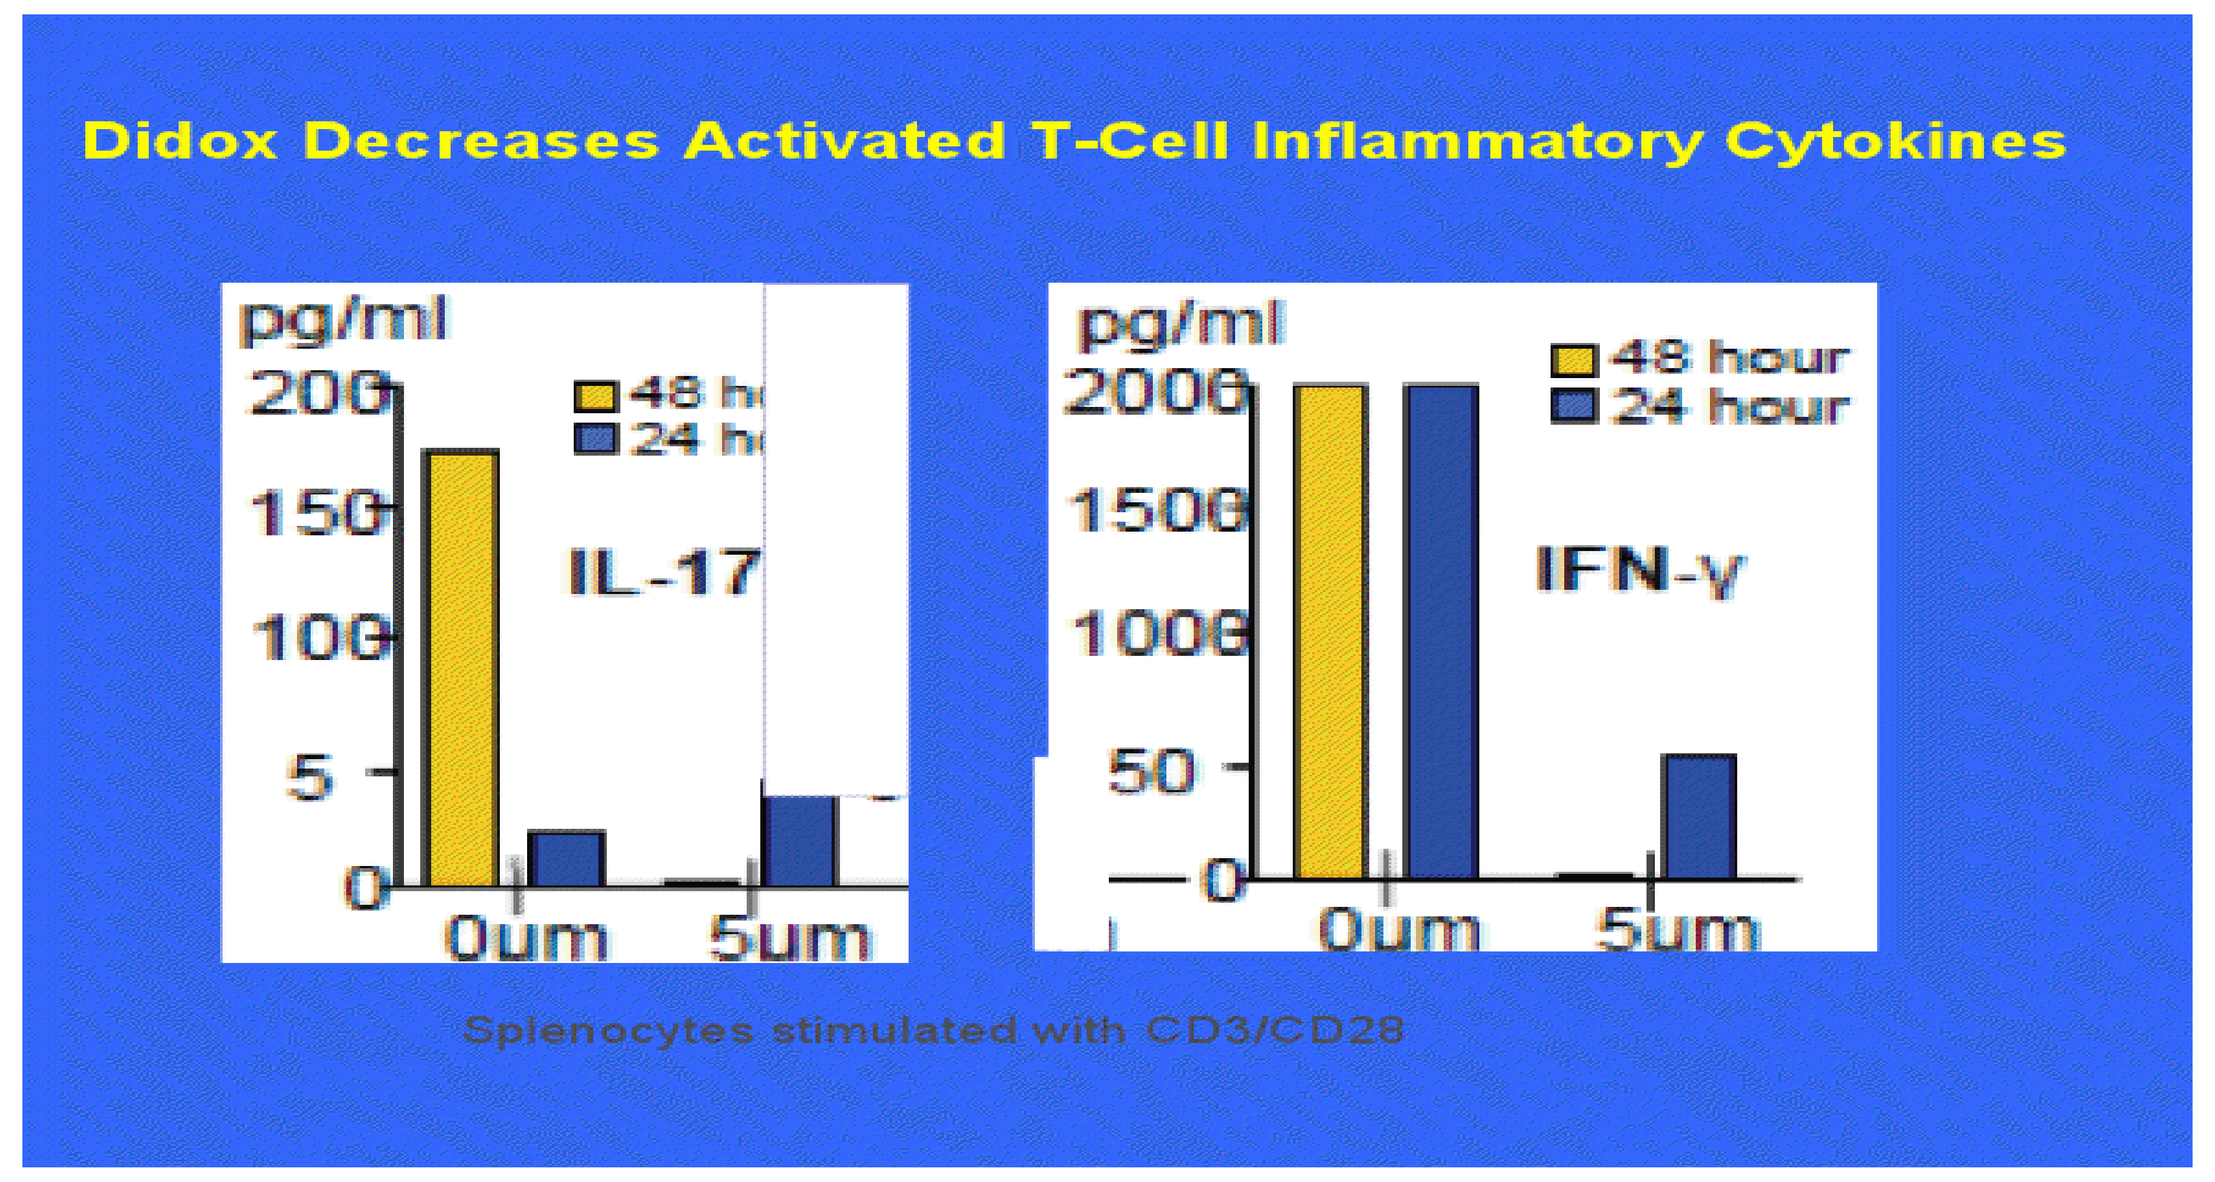
Didox Decreases Activated T-Cell Inflammatory Cytokines
pg/ml
IL-17
IFN-γ
48 hour
24 hour
0um
5um
Splenocytes stimulated with CD3/CD28

T-cells in a splenocyte preparation were stimulated for 72 hours with antibodies to CD3/CD28 followed by analysis of supernatant for cytokines using ELISA. Results shown are from 3-4 splenocyte preps.

Figure 5- Didox Inhibits Microglial Nitrite Production

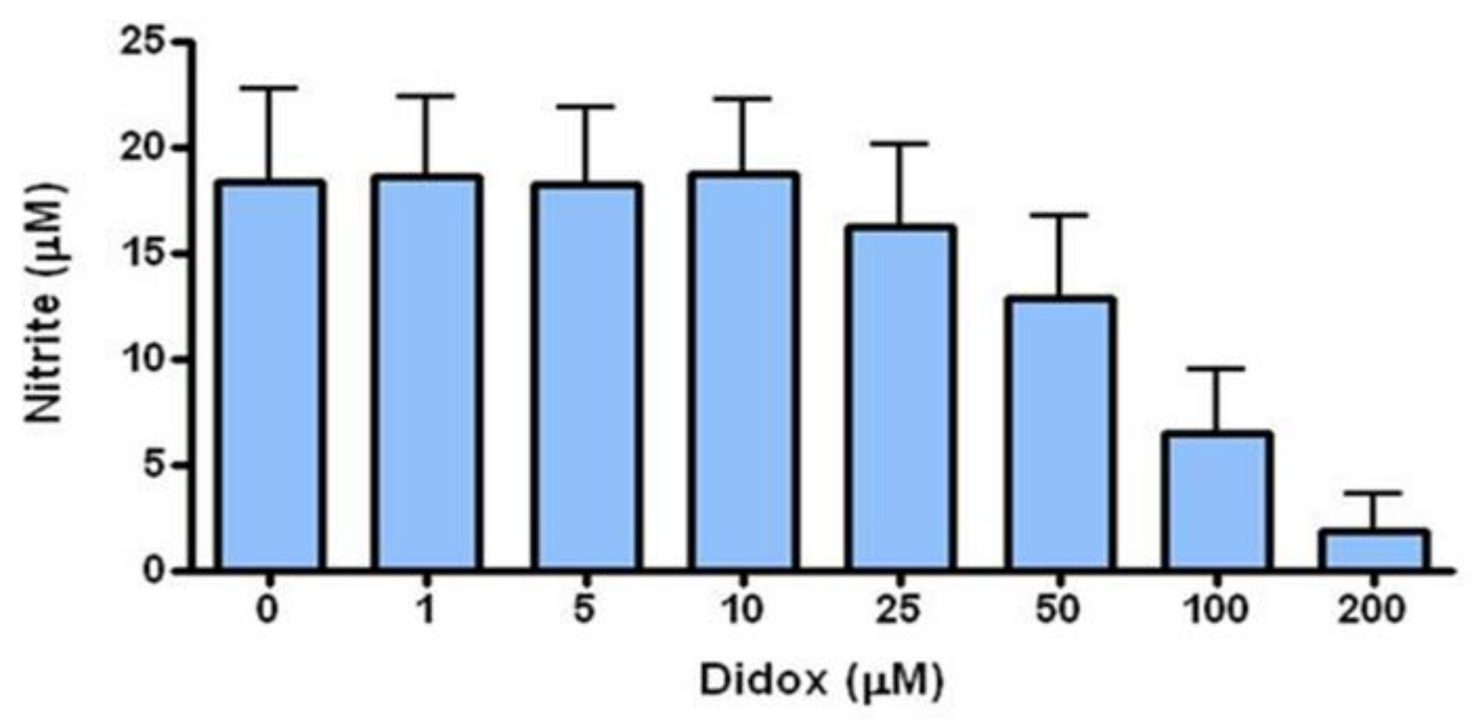
Nitrite (µM)
Didox (µM)

Figure 5. Microglia were isolated by shaking from a mixed glial cell bed, followed by stimulation with Lipopolysaccharide (LPS) and analysis of nitrite in the supernatant via the Griess Assay. Results shown are average of 3 preparations +/- S.D. of the mean.

Morphology Study

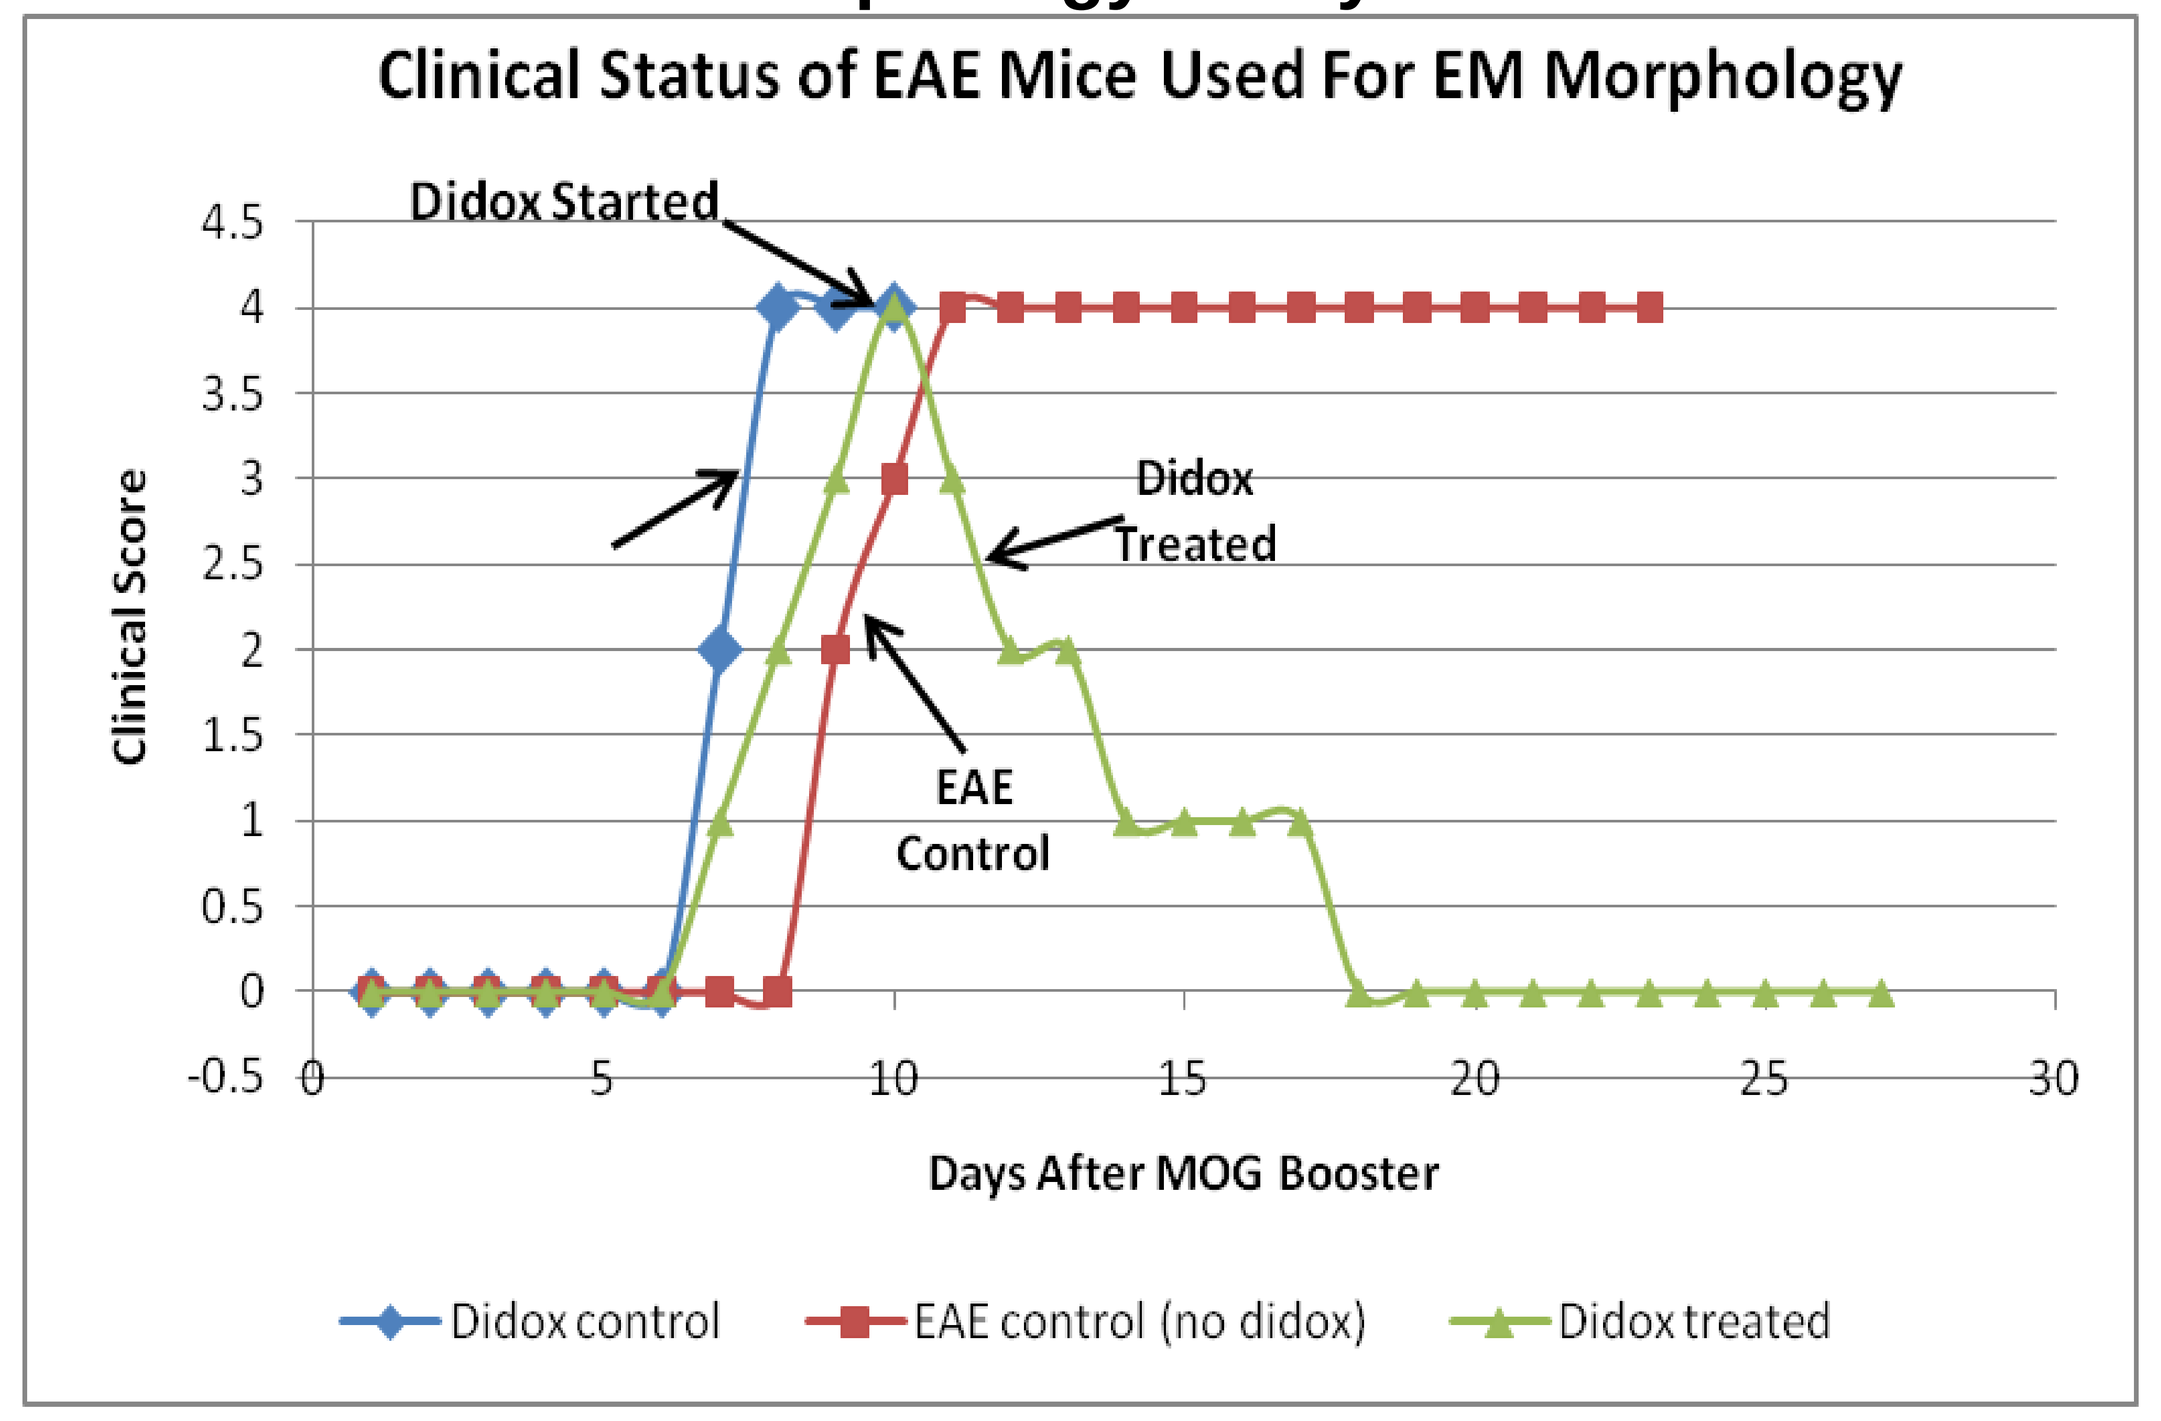
Clinical Status of EAE Mice Used For EM Morphology
Didox Started
Didox Treated
EAE Control
Clinical Score
Days After MOG Booster
Didox control
EAE control (no didox)
Didox treated

Figure 6. Chronic EAE was induced in C57Bl/6 mice. One control EAE mouse (blue line) was sacrificed and perfused at the time that Didox therapy (250mg/kg daily) was initiated (10days after MOG booster) in another mouse.. These three mice were perfused and processed for EM studies.

Figure 7- Didox Preserves Morphology Present at Initiation of Treatment

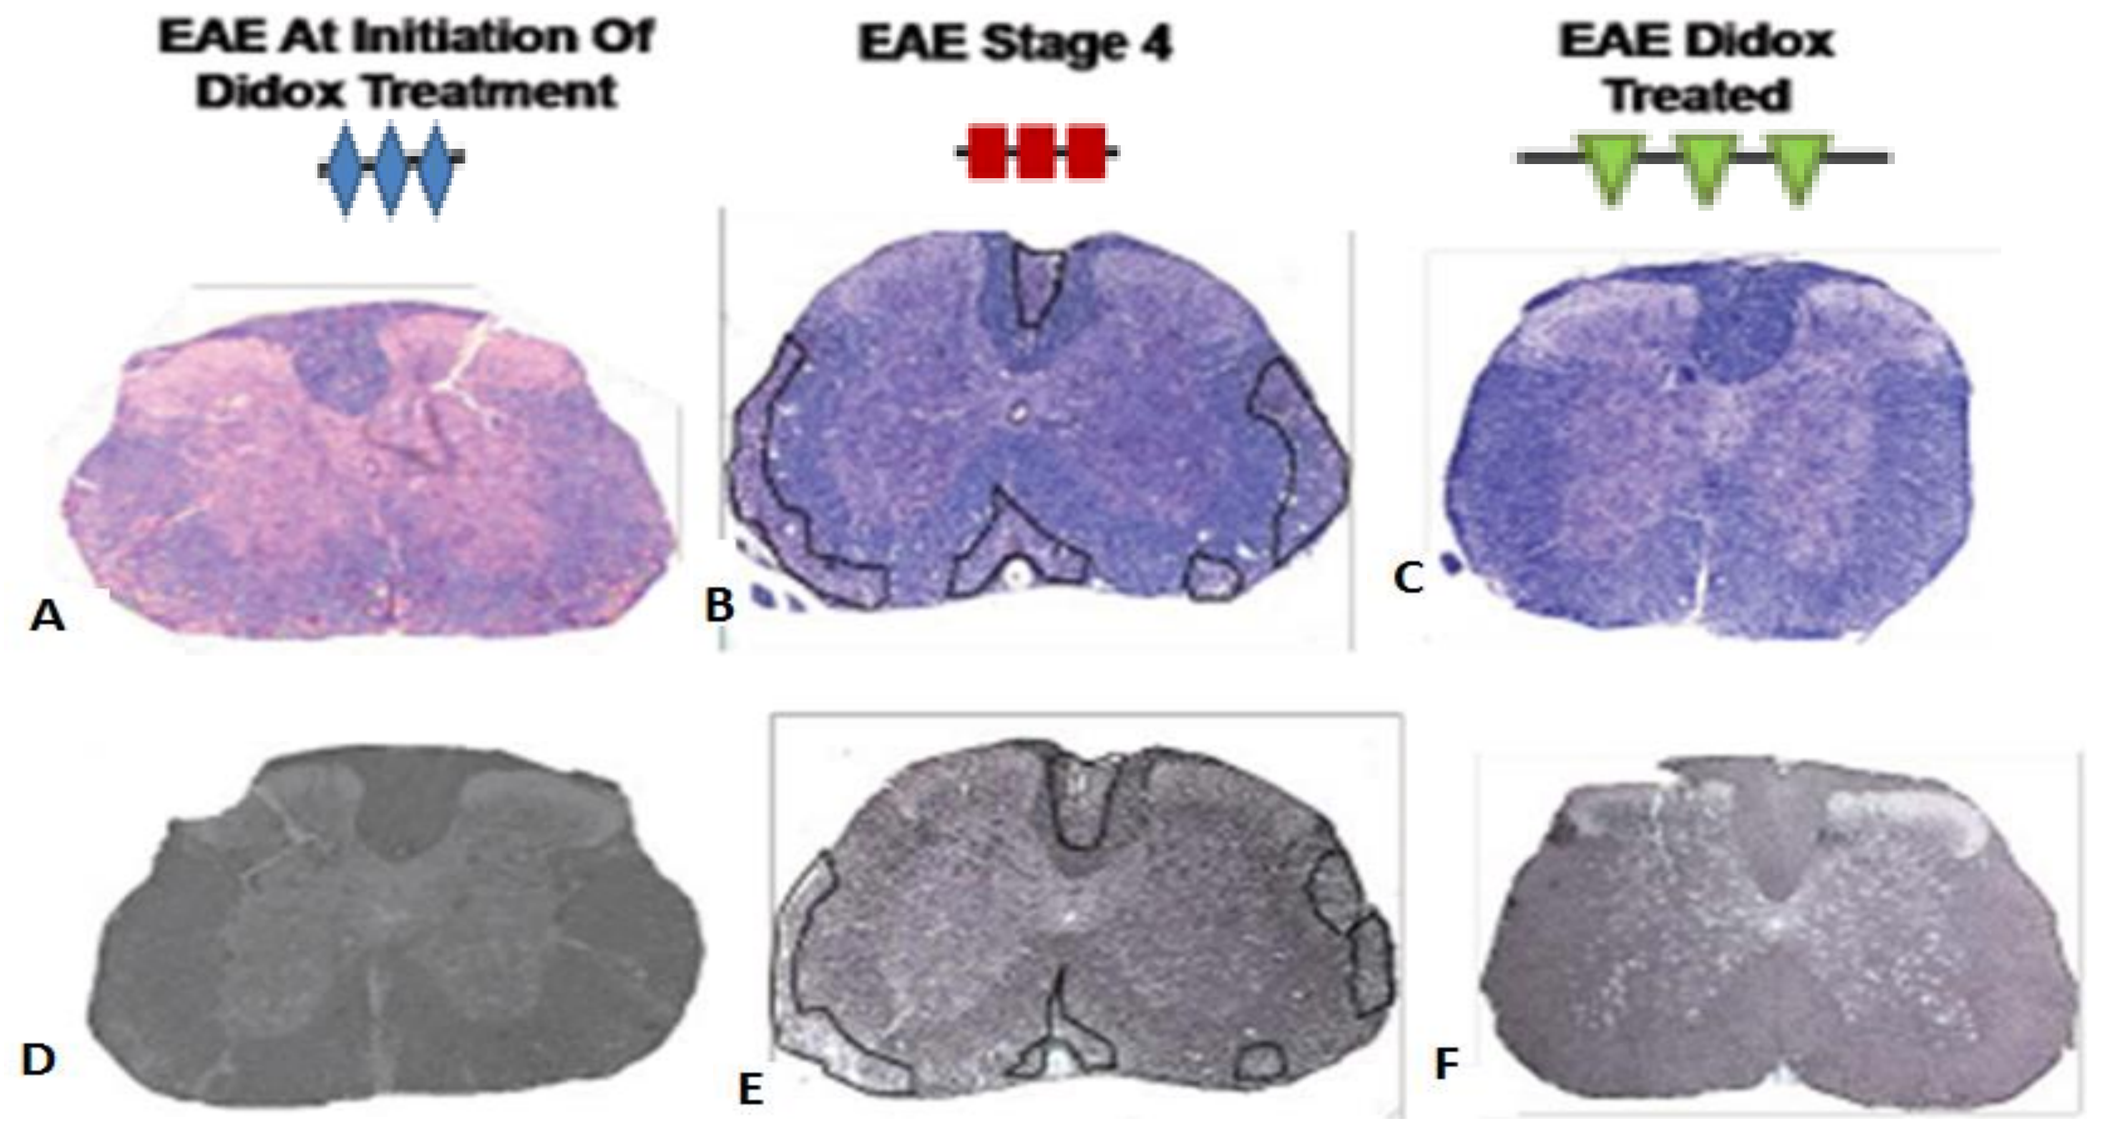
EAE At Initiation Of Didox Treatment
EAE Stage 4
EAE Didox Treated
A
B
C
D
E
F

Figure 7. Extensive demyelination is present in the untreated EAE spinal Cord (B). In contrast myelination in the didox treated cord is intact (C) while some limited demyelination is present in the spinal cord of Stage 4 EAE spinal cord (A) at the time of initiation of didox treatment. These changes are reflected in the Bielschowsky Axonal Stain where mild axonal degeneration is present in the untreated spinal cord (E), while the didox treated spinal cord (F) and the EAE spinal cord at the time of the initiation of didox treatment (D) look relatively normal.

Figure 8

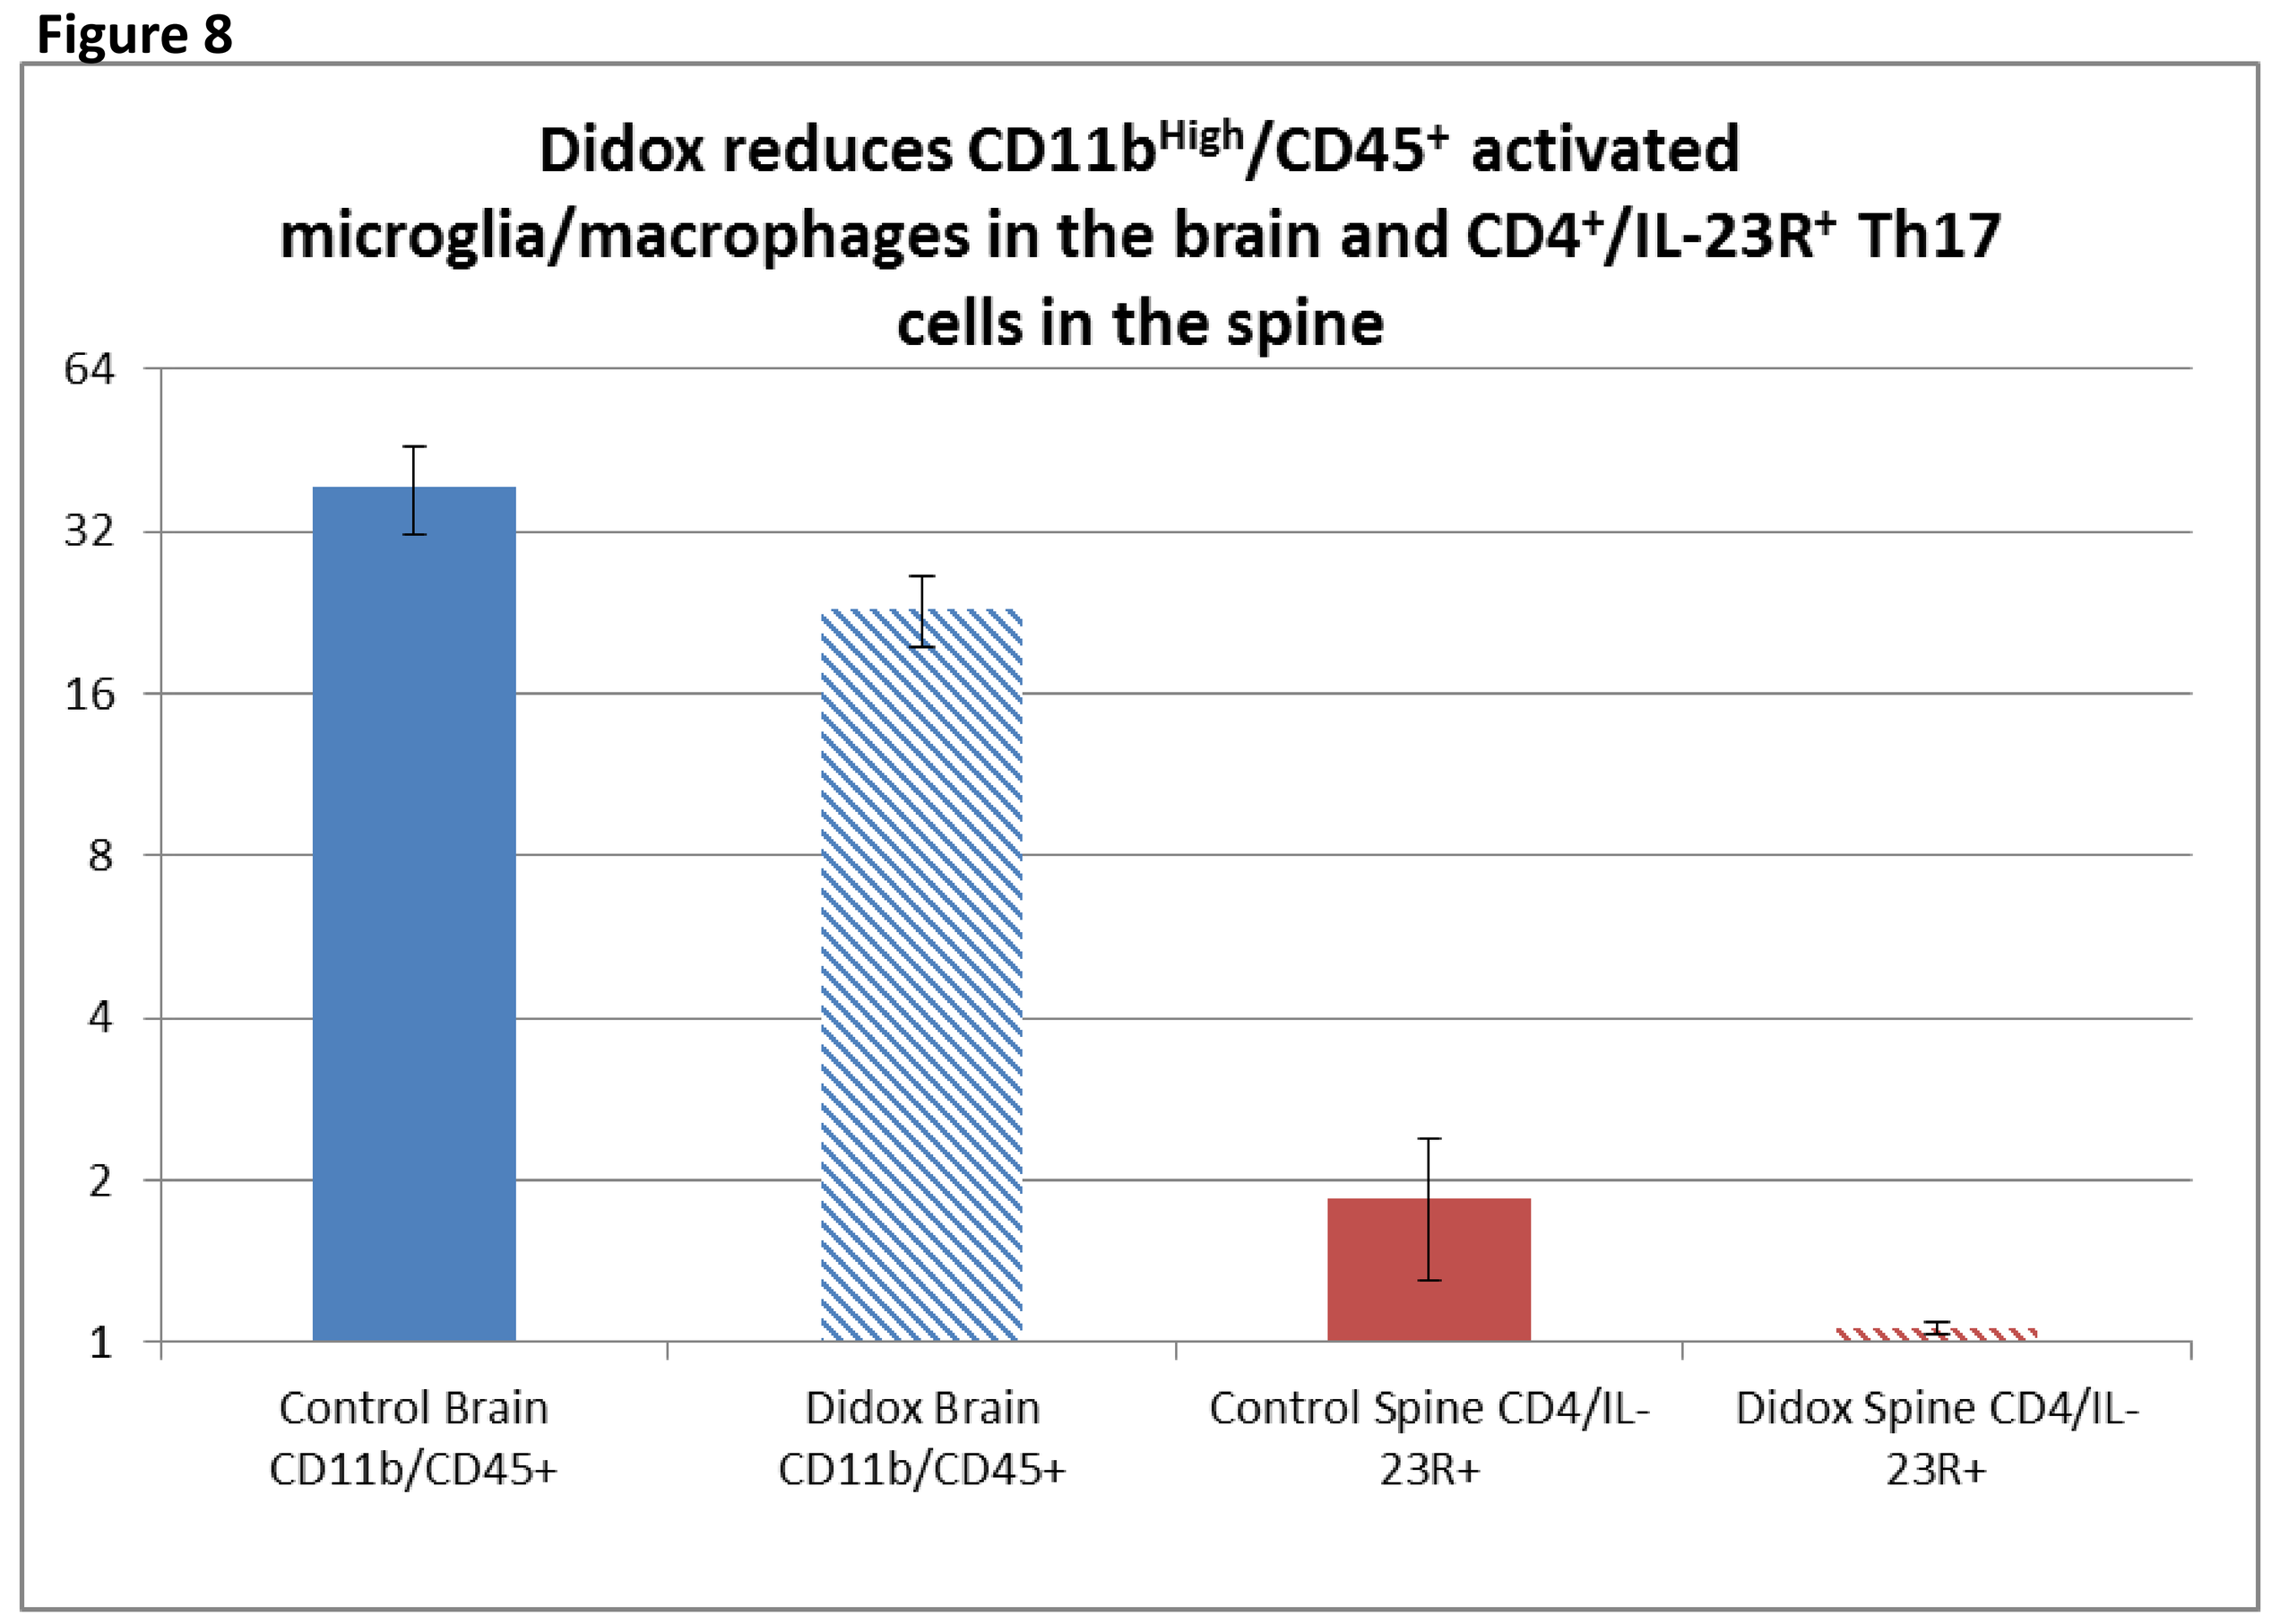
Didox reduces CD11b^{High}/CD45^{+} activated microglia/macrophages in the brain and CD4^{+}/IL-23R^{+} Th17 cells in the spine
Control Brain CD11b/CD45+
Didox Brain CD11b/CD45+
Control Spine CD4/IL-23R+
Didox Spine CD4/IL-23R+

Figure 9

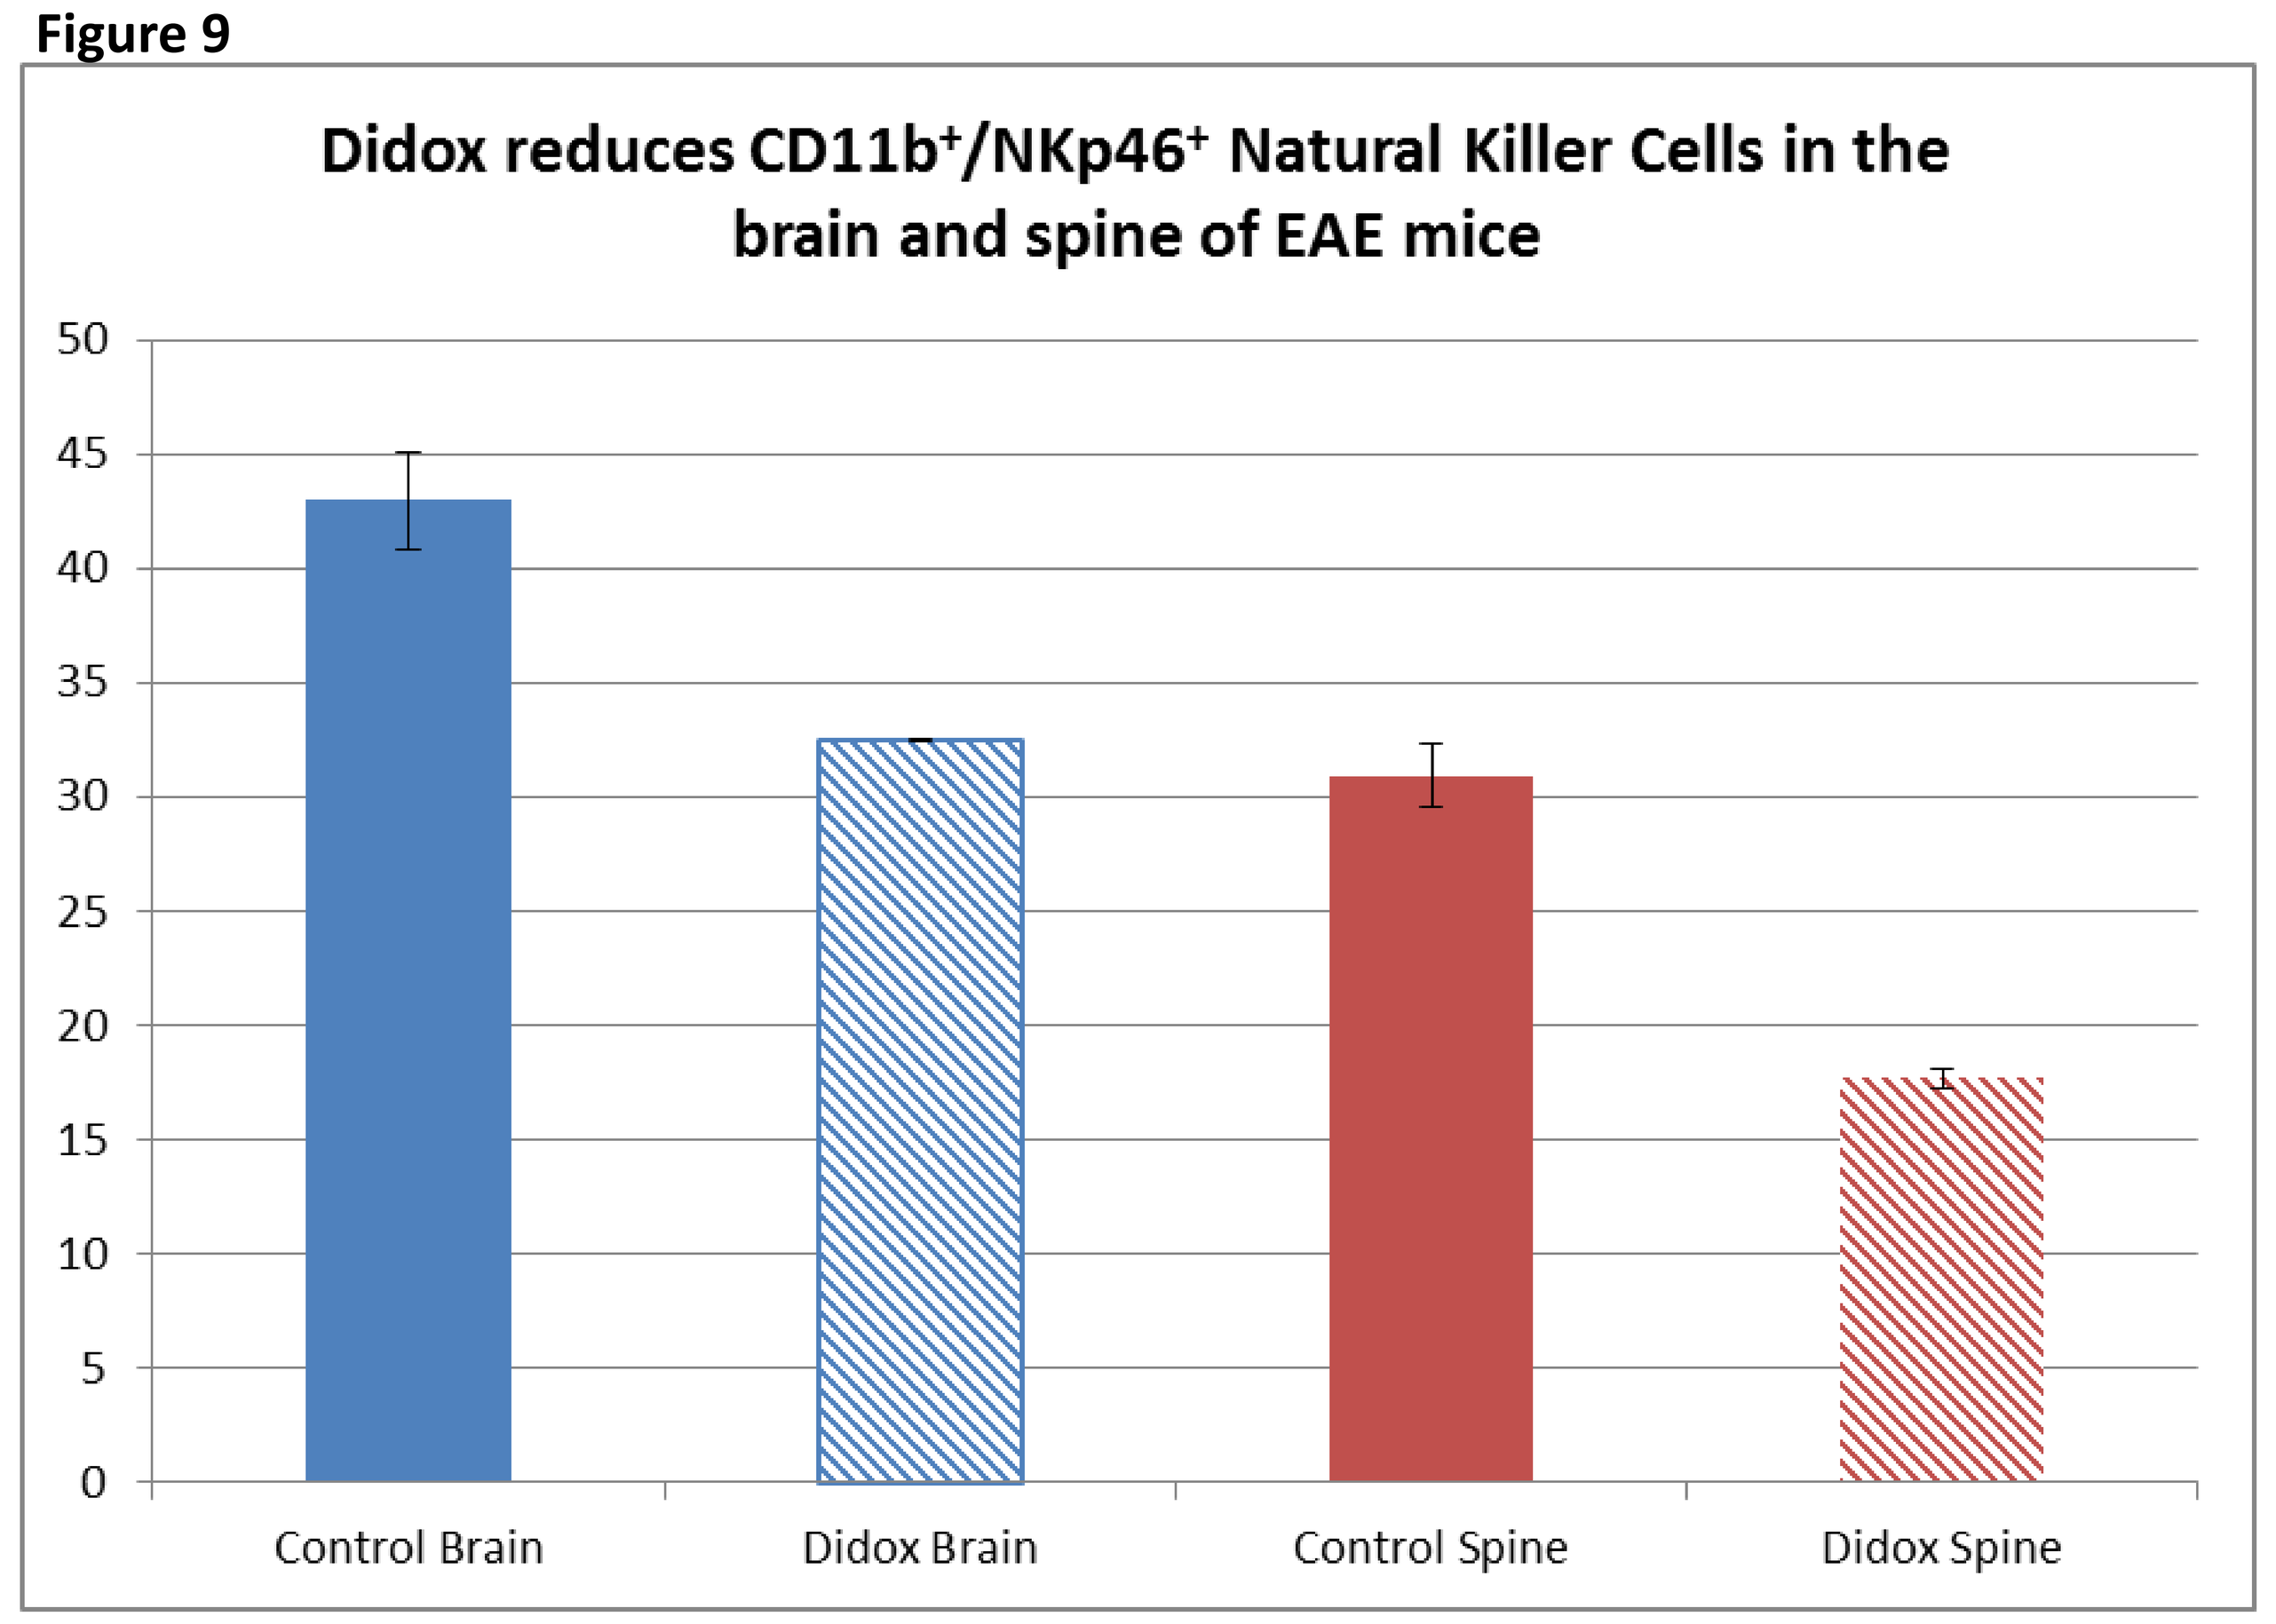
Didox reduces CD11b^{+}/NKp46^{+} Natural Killer Cells in the brain and spine of EAE mice
Control Brain
Didox Brain
Control Spine
Didox Spine

Figure 8 & 9. Flow Cytometry Analysis of Immune Cells In the Brain and Spinal Cord of EAE Mice:

Animals were anaesthetised with Avertin and perfursed through the left ventricle with PBS. The spinal cord and brain were removed and both tissues were minced and placed in Miltenyi Neural Tissue Dissociation solution followed by digestion according to manufacturers instructions. After a single cell suspension was obtained both brain and spine were clarified of myelin by centrifugation through PBS in 0.9M sucrose.

Cells were first incubated with a blocking antibody to CD16/32 and then incubated with antibodies attached to the desired flourophores, fixed after staining and analyzed with a Becton Dickinson FACSCalibur. Date analysis was carried out with FlowJo Software.

Conclusions

Oral Didox can prevent or reverse the clinical symptoms of EAE when given relatively early in the disease process.

Oral Didox was able to reverse established disease in animals that were at the highest severity, clinical score 4, when treatment was initiated.

Didox inhibits T-cell proliferation and secretion of IFN- γ .

Didox inhibits nitrite production by microglia.

EM analysis of didox-treated animals reveals preservation of morphology relative to untreated EAE animals at state 4.

EAE mice which were at stage 4 when didox treatment was initiated have only modest morphological changes.

Oral Didox deters the immune cell population in the brain and spinal cord of EAE mice to favor recovery and remyelination.

Didox is effective when administered orally.

These Didox effects have the potential to provide an environment conducive to recovery and myelin repair in MS.

References

Chofflon N (2005) BioDrugs 19(5);299-308.

Dupree JL, et al. (1999) J Neurocytol. 27(9):649-59.

Feinstein DL et al. (2002) Ann Neurol 51:694-702.

Papadopoulos CM et al. (2002) Ann Neurol 51(4):433-41.

Call4Posters