Relapse outcomes in fingolimod-treated patients previously exposed to natalizumab, interferon, or glatiramer acetate: results from the FIRST study

DX 52

Philipp von Rosenstiel,[1] Erik Burton[2]

[1]Novartis Pharma AG, Basel, Switzerland; [2]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA

CONCLUSIONS

- **During the first 4 months of fingolimod therapy, a beneficial effect on ARR was seen relative to historical pre-study relapse rates in patients recently treated with natalizumab, IFNβ, or GA.**
- **The peak in ARR during month 1 in patients who had discontinued natalizumab 3–6 months before the study coincides with the expected timing of disease reactivation.[13] The subsequent decrease in relapses by 4 months in this patient group suggests that fingolimod may reduce disease reactivation. However, the optimal gap between natalizumab discontinuation and fingolimod initiation should be further evaluated.**
- **Despite methodologic limitations, these post hoc analyses suggest that fingolimod may be a suitable therapy following the discontinuation of natalizumab, as well as in patients with MS who need to switch from IFNβs or GA to attain disease control.**

INTRODUCTION

- In multiple sclerosis (MS), it is common practice for patients to switch treatment from first-line disease-modifying therapies (DMTs)[1] because of suboptimal response to treatment, poor tolerability, or safety concerns.[1-3]
- Given that treatment-refractory, clinically active MS can quickly lead to irreversible disability,[4] there is a need for escalation to a treatment that provides rapid disease control[5]; however, treatment-escalation strategies in relapsing-remitting MS (RRMS) are limited.
- Fingolimod is a once-daily, oral sphingosine 1-phosphate receptor modulator approved in the United States and European Union for the treatment of relapsing MS.[6,7a]
- The phase 3 Trial Assessing Injectable Interferon vs Fingolimod Oral in Relapsing-Remitting Multiple Sclerosis (TRANSFORMS) extension showed that relapse rates were significantly lower 1 year after switching to once-daily oral fingolimod 0.5 mg (approved dose) than in the previous year when patients had been receiving intramuscular (IM) interferon (IFN) β-1a (0.22 vs 0.31, respectively).[8] Fingolimod is therefore a highly efficacious treatment option for patients who require a step-up in therapy from first-line DMTs.[9]
- In contrast, findings of clinical improvement after switching to fingolimod from natalizumab have been mixed.[10,11]
- In practice, many individuals initiating fingolimod therapy may have previously received other DMTs; it is therefore important to understand the efficacy profile of fingolimod in such patients.

OBJECTIVE

- To evaluate relapse outcomes associated with a change in therapy to fingolimod in patients previously treated with natalizumab, IFNβ, or glatiramer acetate (GA) during the Fingolimod Initiation and Cardiac Safety Trial (FIRST)

METHODS

Study Design and Patients

- These post hoc analyses included patients from FIRST.
- FIRST was a phase 3b, single-arm, open-label, 4-month, multicenter study of 2417 patients with RRMS conducted in 23 countries. The study assessed the safety and tolerability of fingolimod 0.5 mg in a broader population of patients than was previously included in the fingolimod phase 3 trials.
- Patients with a diagnosis of relapsing MS who were aged 18–65 years with an Expanded Disability Status Scale (EDSS) score of 0–6.5 were included in the study.
- Key exclusion criteria for FIRST were largely consistent with patient criteria in other phase 3 fingolimod studies; in contrast with previous studies, patients with diabetes or certain pre-existing cardiac conditions were eligible.[9,12]

Relapse Activity and Post Hoc Analyses

- All relapses were recorded, including those reported as serious adverse events. Subsequently, annualized relapse rates (ARR) and proportions of patients without relapses were calculated.
 - Relapses were assessed by the treating physician, not by an independent physician. Objective evidence of change in EDSS criteria or neurologic examination was not required.
- 2 separate post hoc analyses were conducted to evaluate ARR in the year before the study and during 4 months of fingolimod 0.5 mg treatment in
 - Patients who discontinued natalizumab >6 months (n=135) or 3–6 months (n=119) before the study or who had never received natalizumab (n=2163)
 - Patients who received IFNβ (n=1040) or GA (n=432) ≤6 months before the study or who had never received DMT (n=363)
- ARRs for each subgroup were calculated as the total number of relapses divided by the total duration (in days) of fingolimod exposure for all patients in that group multiplied by 365.25. ARRs for the 1 year before the study were calculated as the total number of relapses reported by patients divided by the number of patients in the group.

[a]*The approved indication may vary from country to country. In the United States, fingolimod is approved for the treatment of patients with relapsing forms of MS. In the European Union, fingolimod is approved for treatment of patients with highly active RRMS.*

RESULTS

- Baseline characteristics showed that patients were still experiencing relapses despite natalizumab therapy; longer disease duration and higher mean EDSS scores were observed in patients previously treated with natalizumab vs patients with no previous natalizumab treatment (**Table 1**).
- Similarly, patients with a recent history of IFNβ or GA treatment generally had longer disease duration and higher mean EDSS scores than treatment-naive patients (**Table 1**).

Table 1. Baseline demographics and disease characteristics

	Discontinued natalizumab >6 mo before study (n=135)	Discontinued natalizumab 3–6 mo before study (n=119)	No previous treatment with natalizumab (n=2163)	Received IFNβ ≤6 mo before study* (n=1040)	Received GA ≤6 mo before study* (n=432)	Treatment-naive (n=363)
Mean age, y	37.9	36.8	38.5	38.2	39.5	37.8
Women, n (%)	109 (80.7)	81 (68.1)	1583 (73.2)	761 (73.2)	330 (76.4)	247 (68.0)
Duration of MS since first symptoms, y						
Mean	12.3	10.6	9.1	9.0	9.9	6.8
Median (range)	11.7 (1.8–37.0)	9.3 (1.1–44.3)	7.5 (0.1–41.0)	7.2 (0.3–41.0)	8.3 (0.3–34.6)	4.6 (0.1–31.2)
EDSS score						
Mean	3.3	3.0	2.3	2.3	2.5	2.1
Median (range)	3.0 (0–6.5)	3.0 (0–6.5)	2.0 (0–6.5)	2.0 (0–6.5)	2.0 (0–6.5)	2.0 (0–6.5)
Relapses within previous 12 mo						
Mean/ARR	1.52	1.08	1.07	1.01	1.13	1.26
Total number of relapses requiring steroids, n (%)	183 (88.8)	105 (81.4)	1759 (75.9)	849 (80.6)	390 (79.8)	280 (61.1)

ARR=annualized relapse rate; EDSS=Expanded Disability Status Scale; GA=glatiramer acetate; IFN=interferon; MS=multiple sclerosis.
*The mean time since discontinuation of IFNβ or GA before starting fingolimod was approximately 1 month (32.9–34.3 d). The maximum duration since previous therapy exceeds 6 mo in some patients because of a gap of up to approximately 1 mo between the screening visit and the first dose of fingolimod.

ARRs 1 Year Before Study and During Fingolimod Therapy

- ARR in the year before study was highest in patients who had discontinued natalizumab >6 months before the study (1.52) and similar in patients who discontinued 3–6 months before the study (1.08) or had never received natalizumab (1.07; **Figure 1A**).
- ARR in the year before the study was lower in patients previously treated with IFNβ (1.01) or GA (1.13) compared with patients who were treatment-naive (1.26; **Figure 1B**).
- Treatment with fingolimod for 4 months reduced ARR compared with the year before the study in all patient groups (**Figures 1A, 1B**).
 - In patients who had discontinued natalizumab 3–6 months before the study, ARR during the study was higher than in the other patient groups.
 - This was driven by the peak in ARR during month 1, when more than 50% of the total number of relapses during the study occurred.

Figure 1. ARR in the year before and during FIRST in patients with or without (A) prior natalizumab or (B) prior IFNβ or GA treatment

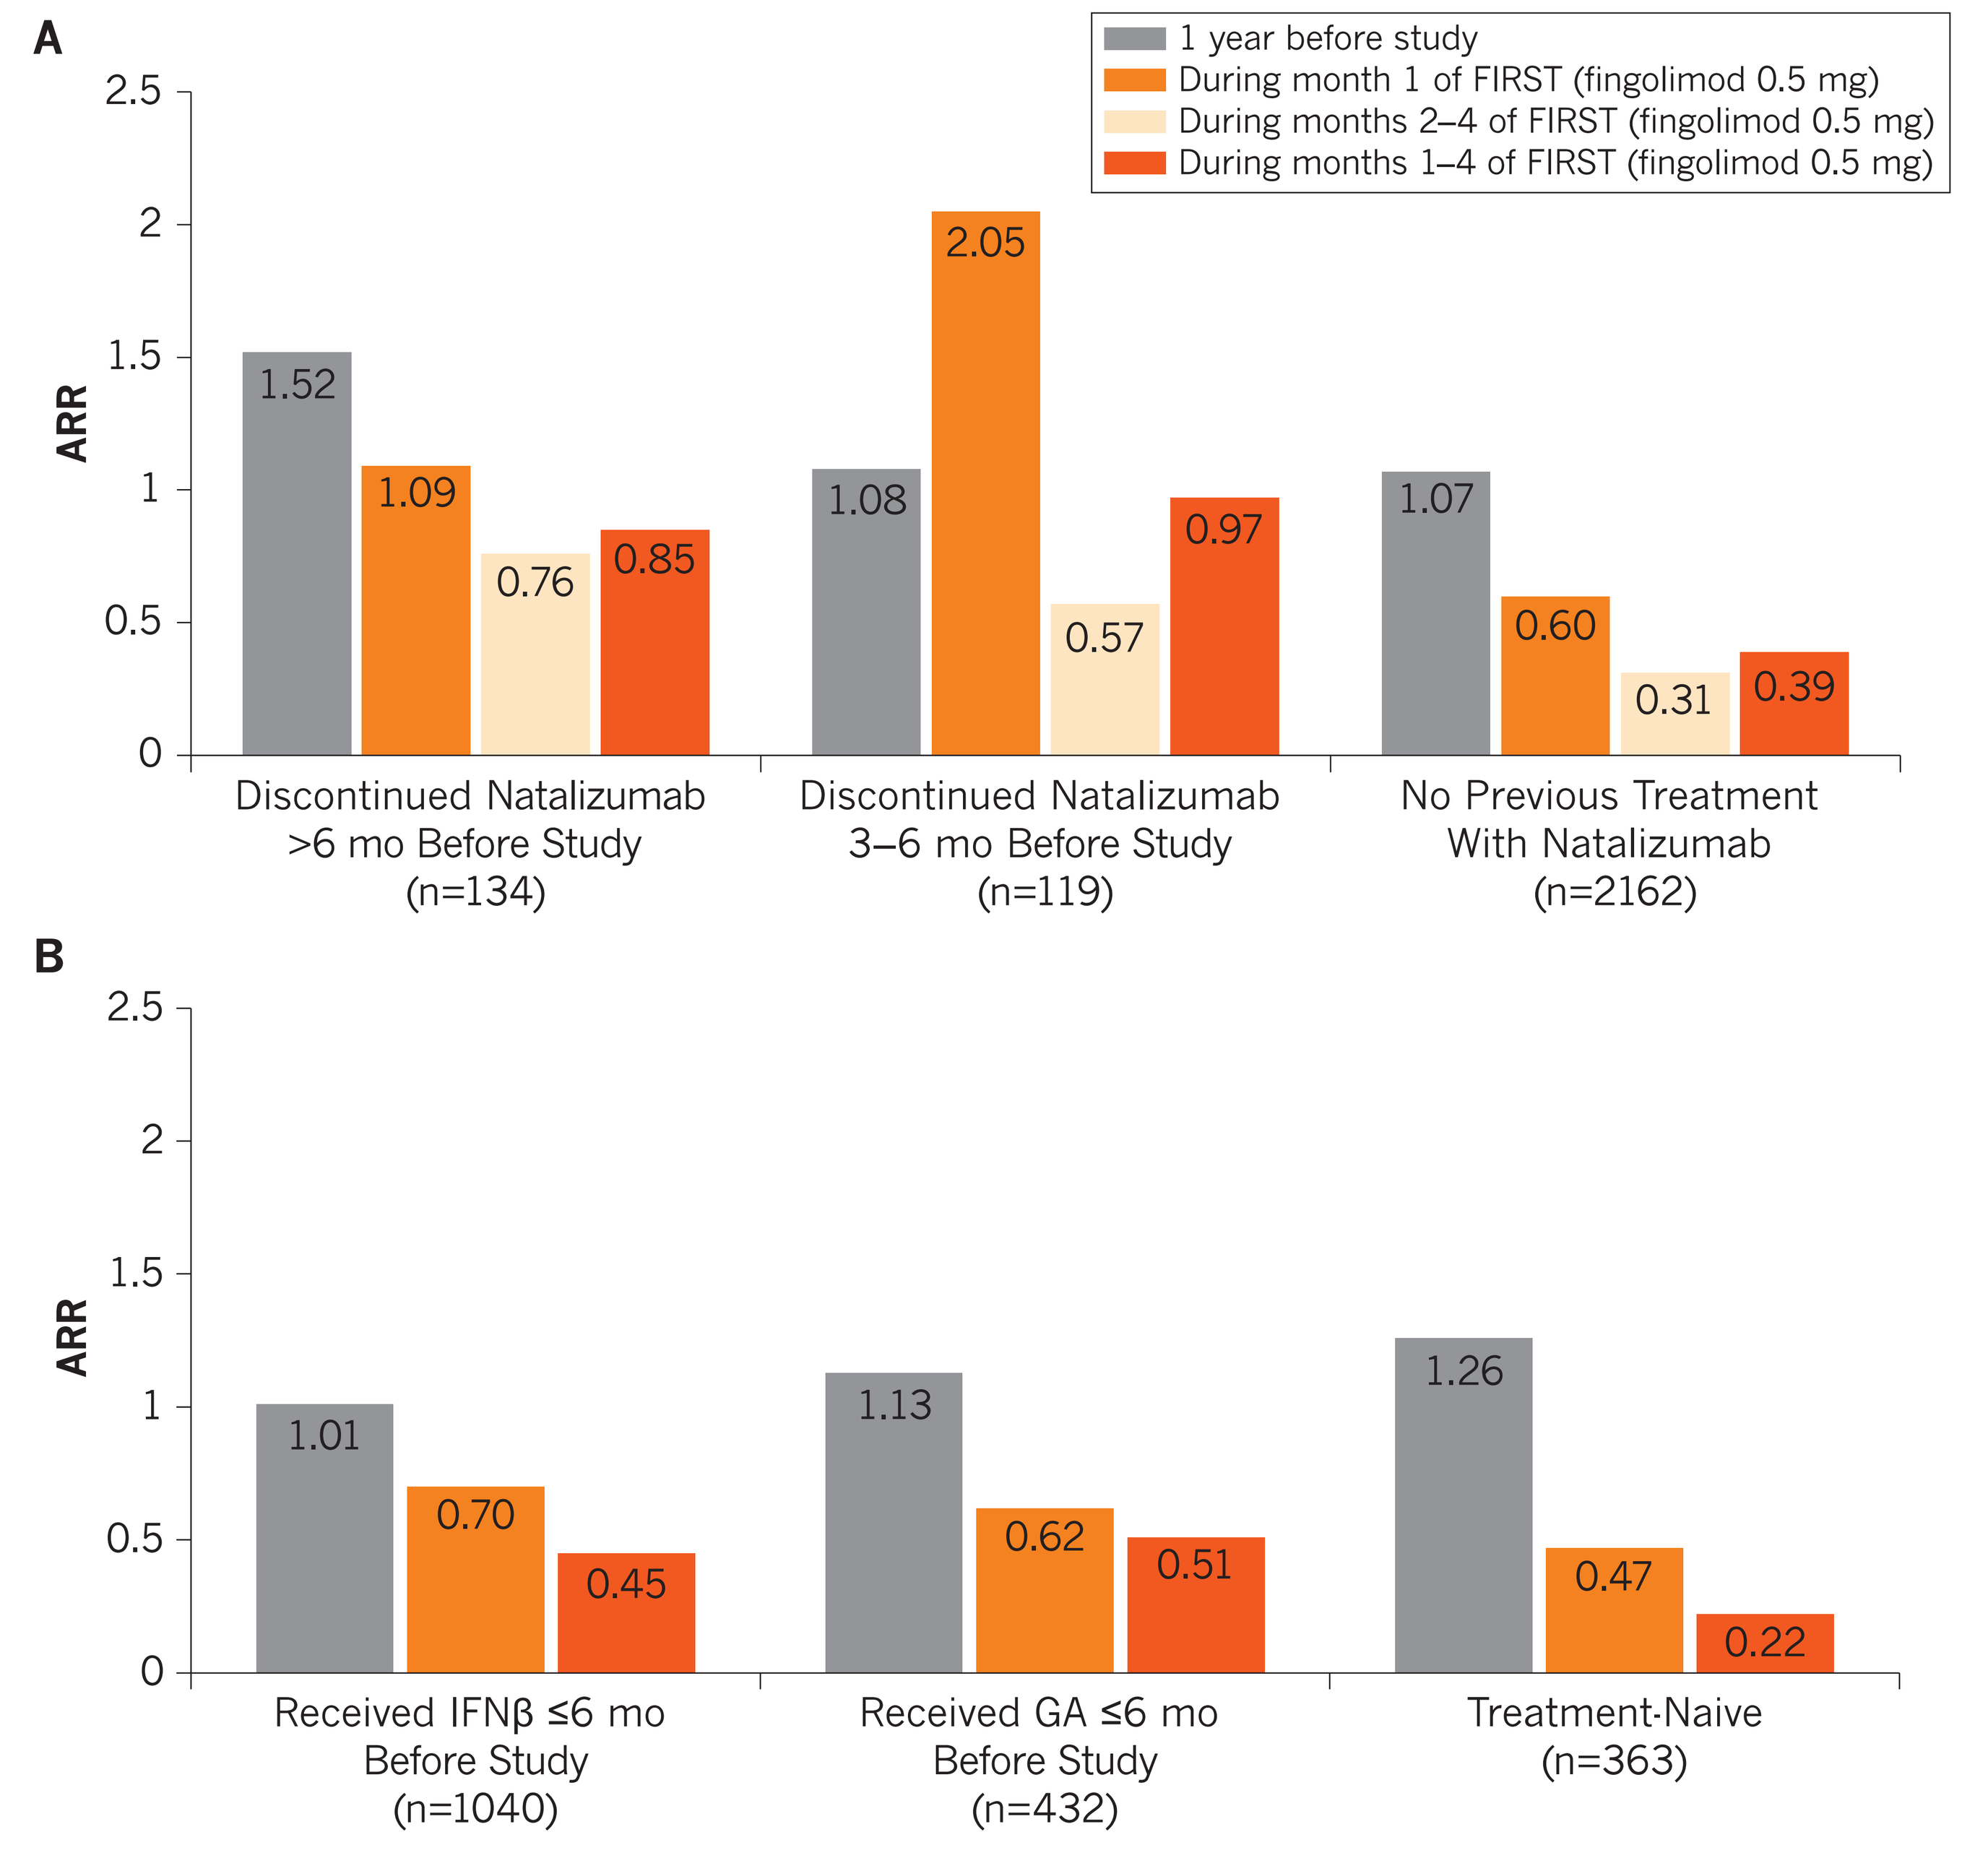
A
1 year before study
During month 1 of FIRST (fingolimod 0.5 mg)
During months 2–4 of FIRST (fingolimod 0.5 mg)
During months 1–4 of FIRST (fingolimod 0.5 mg)
ARR
2.5
2
1.5
1
0.5
0
1.52
1.09
0.76
0.85
1.08
2.05
0.57
0.97
1.07
0.60
0.31
0.39
Discontinued Natalizumab >6 mo Before Study (n=134)
Discontinued Natalizumab 3–6 mo Before Study (n=119)
No Previous Treatment With Natalizumab (n=2162)
B
ARR
2.5
2
1.5
1
0.5
0
1.01
0.70
0.45
1.13
0.62
0.51
1.26
0.47
0.22
Received IFNβ ≤6 mo Before Study (n=1040)
Received GA ≤6 mo Before Study (n=432)
Treatment-Naive (n=363)

ARR=annualized relapse rate; GA=glatiramer acetate; IFN=interferon.

Proportion of Patients Free From Relapse

- In the year before the study, the proportion of patients who were relapse-free was lowest in those who discontinued natalizumab >6 months before study (**Figure 2A**).
- In the IFNβ/GA analysis, the proportion of patients who were relapse-free in the year before the study was 37% in those who had received IFNβ, 32% in those who had received GA, and 14% in those who were treatment-naive (**Figure 2B**).
- Following 4 months of treatment with fingolimod, the majority of patients in all groups were relapse-free (**Figures 2A, 2B**).

Figure 2. Proportion of patients relapse-free in the year before and during FIRST in patients with or without (A) prior natalizumab or (B) prior IFNβ or GA treatment

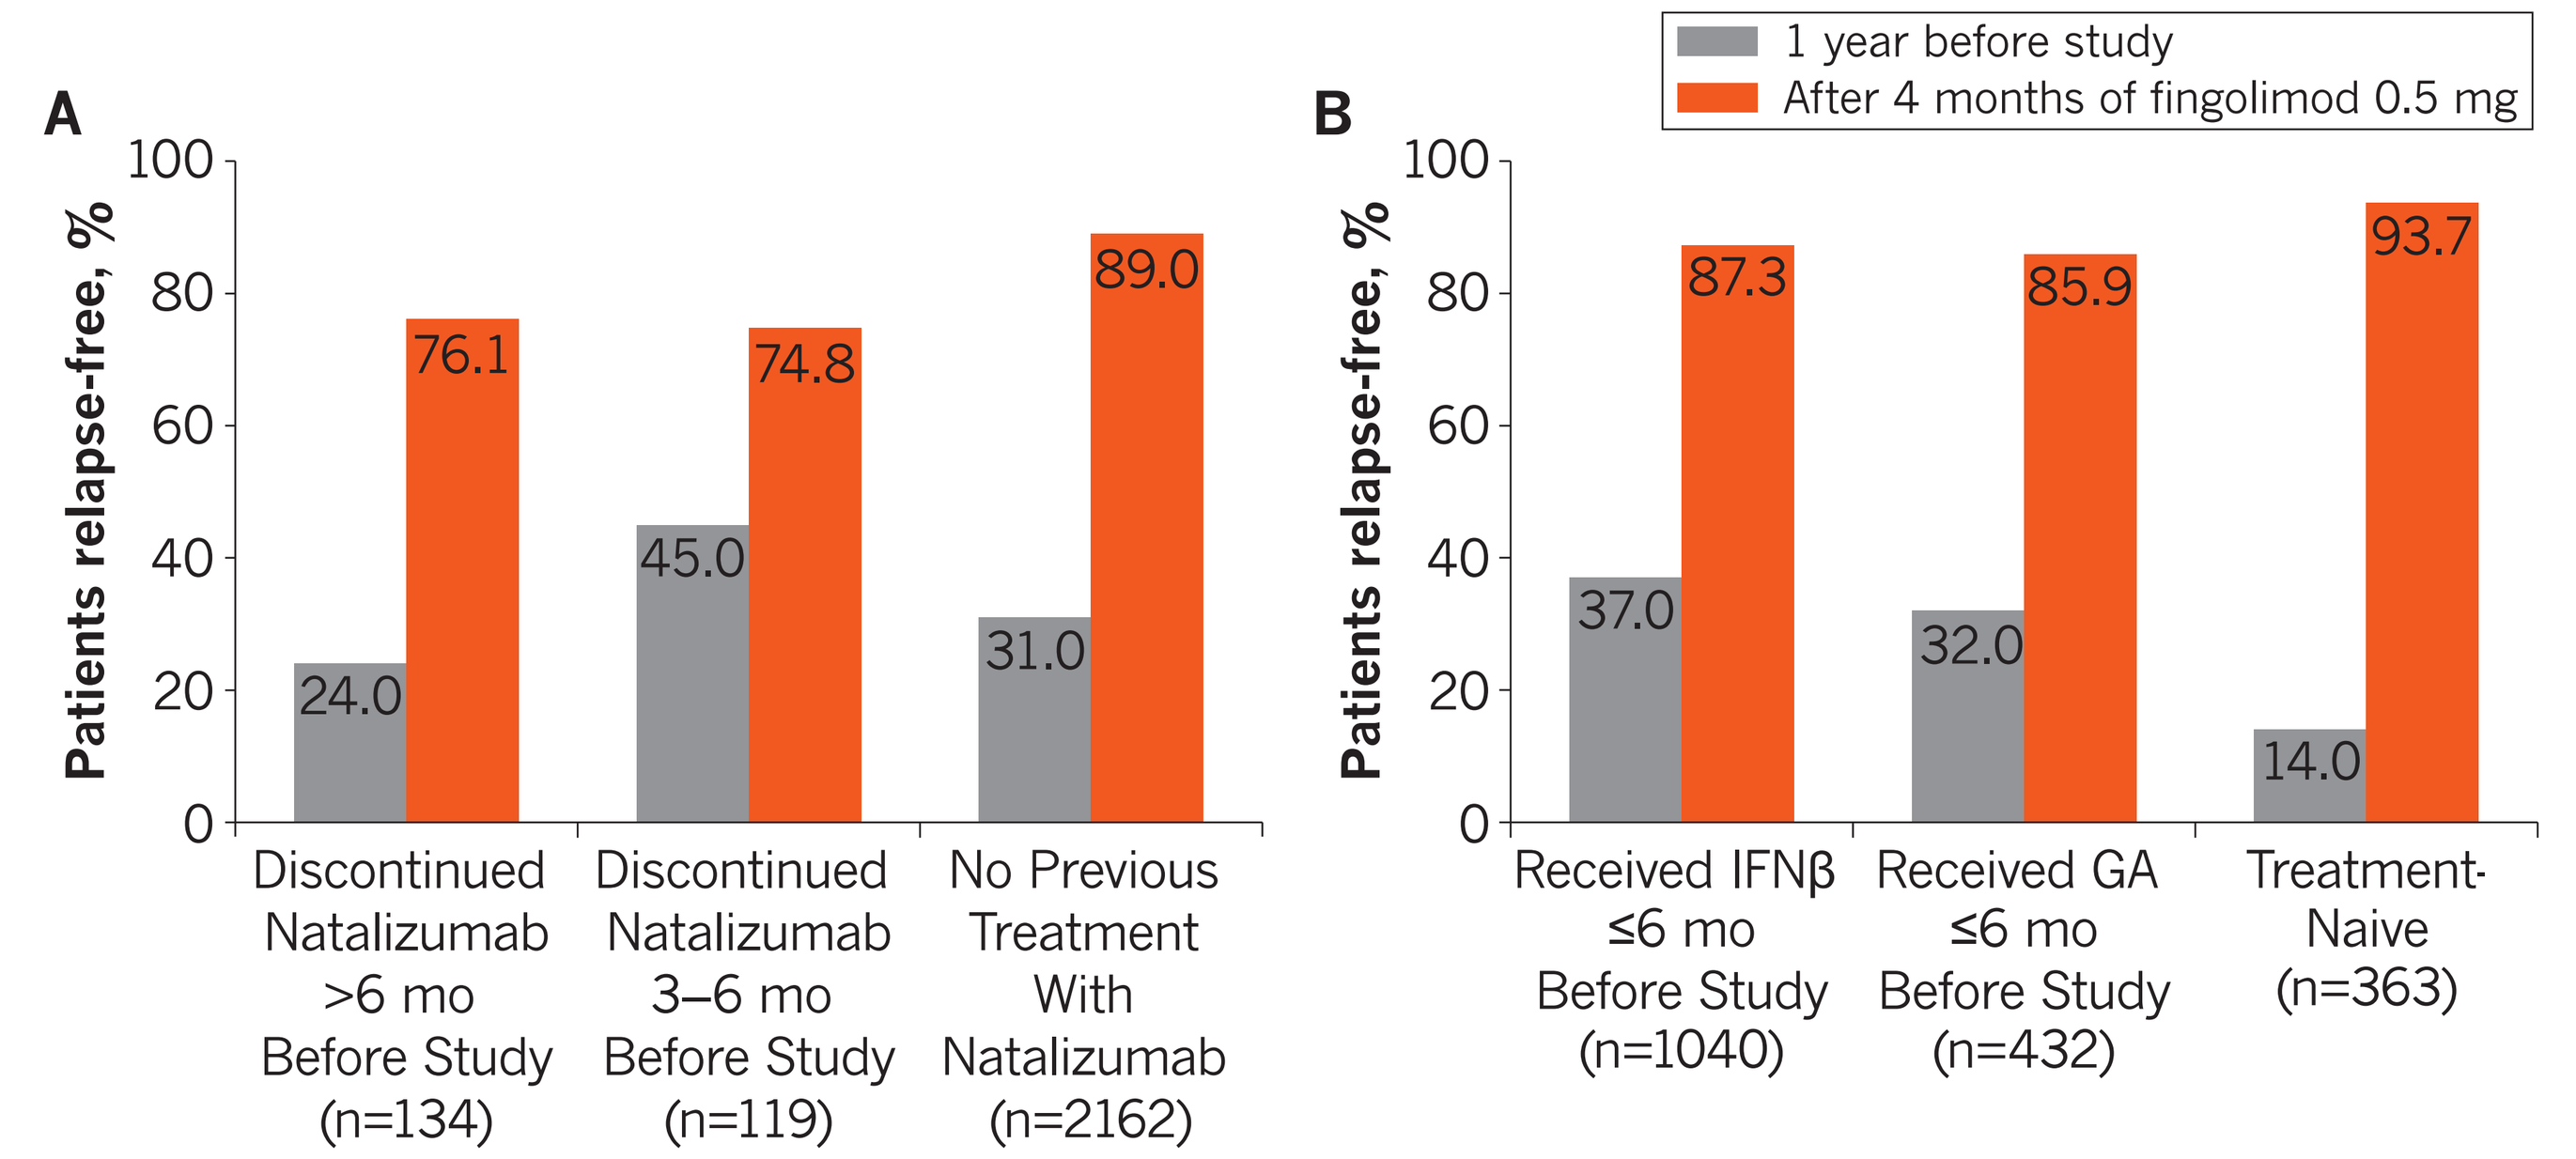
1 year before study
After 4 months of fingolimod 0.5 mg
A
Patients relapse-free, %
100
80
60
40
20
0
24.0
76.1
45.0
74.8
31.0
89.0
Discontinued Natalizumab >6 mo Before Study (n=134)
Discontinued Natalizumab 3–6 mo Before Study (n=119)
No Previous Treatment With Natalizumab (n=2162)
B
Patients relapse-free, %
100
80
60
40
20
0
37.0
87.3
32.0
85.9
14.0
93.7
Received IFNβ ≤6 mo Before Study (n=1040)
Received GA ≤6 mo Before Study (n=432)
Treatment-Naive (n=363)

GA=glatiramer acetate; IFN=interferon.

References

1. Gajofatto A, et al. *Mult Scler.* 2009;15:50-58.
2. Tysabri® (natalizumab). Full Prescribing Information, Biogen Idec, Inc., Cambridge, MA, 2013.
3. Sorensen PS, et al. *Lancet.* 2003;362:1184-1191.
4. Confavreux C and Vukusic S. *Clin Neurol Neurosurg.* 2006;108:327-332.
5. Rieckmann P, et al. *Ther Adv Neurol Disord.* 2008;1:181-192.
6. Gilenya® (fingolimod). Full Prescribing Information, Novartis Pharmaceuticals Corporation, East Hanover, NJ, 2014.
7. Gilenya® (fingolimod). Summary of Product Characteristics. Novartis Pharmaceuticals UK Ltd, Camberley, Surrey, UK. Available at: http://www.medicines.org.uk/emc/medicine/24443. Accessed May 13, 2014.
8. Khatri B, et al. *Lancet Neurol.* 2011;10:520-529.
9. Cohen JA, et al. *N Engl J Med.* 2010;362:402-415.
10. Havla J, et al. *J Neurol.* 2012;260:1382-1387.
11. Sempere AP, et al. *Acta Neurol Scand.* 2013;128:e6-e10.
12. Kappos L, et al. *N Engl J Med.* 2010;362:387-401.
13. O'Connor PW, et al. *Neurology.* 2011;76:1858-1865.

Disclosures

P. von Rosenstiel and E. Burton are employees of Novartis.

Acknowledgments

This study was supported by Novartis Pharmaceuticals Corporation. The authors thank Meg Church, MS, and Erica Wehner, RPh, CMPP, for medical writing support of this poster. Editorial support was provided by Complete Healthcare Communications, Inc. (Chadds Ford, PA). Medical writing and editorial support were funded by Novartis.

Scan to download a reprint of this poster

Downloading data may incur costs which can vary depending on your service provider and may be high if you are using your smartphones abroad. Please check your phone tariff or contact your service provider for more details.

This study was supported by Novartis Pharmaceuticals Corporation | Poster presented at the 2014 Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC) and the Sixth Cooperative Meeting with Americas Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS); May 28–31, 2014; Dallas, TX.