CAN DO Multiple Sclerosis™ | Lifestyle Empowerment Programs

THE **POWER** TO BE MORE THAN YOUR MS

Can Do Multiple Sclerosis | FOUR DAY

CAN DO® PROGRAM

Transforming Lives:

Evaluation of Immediate and 6-Month Outcomes from the CAN DO® PROGRAM

Authors: Lauri O'Brien and Sara Anne Tompkins, Ph.D.

ABSTRACT:

The CAN DO Program offers the most comprehensive view of MS, taking participants on an intensive four-day exploration of their unique conditions and empowering them to live fully with MS. Participants (PwMS and support partners) reported improved confidence to better manage MS symptoms, finding benefit in living with MS and improved MS specific communication at 6-months post CAN DO Program. The program is a personal and powerful MS program, effectively increasing confidence to transform challenges into possibilities.

BACKGROUND:

- The individualized course of MS calls for more empowering and person-specific interventions, as optimism can impact MS by promoting physical adjustment and healthy coping strategies, and it predicts positive physical health outcomes (de Ridder, 2000; Rasmussen, 2009).
- Using an interdisciplinary team, this intensive four-day experience goes beyond traditional programs by providing comprehensive assessments and education about MS – its effects, treatment options and lifestyle adaptation strategies.
- CAN DO Program goes beyond focusing on empowerment by incorporating a health-focused orientation of physical activity (PA) and complete wellness with research, showing PA can improve quality of life and MS symptoms (Motl & McAuley, 2009; Jensen et al., 2012).
- CAN DO Program brings multiple formats and professionals together to provide variety and options best suited to each and every participant. The overwhelmingly positive and hands-on orientation of Can Do MS and the CAN DO Program is unique with the mission of living more fully with MS.

METHOD:

- Pre, post and 6-month surveys collected from participants.
- Convenience sample collected from all participants of the CAN DO Program offered during 2012-2014 (three programs; PwMS and attending support partners (SP); Baseline sample contains 119 participants; 104 at post; 88 at 6 months).
- Outcomes: Multiple Sclerosis Self-Efficacy Scale (MSSE; Schwartz et al., 1996), Benefit Finding in Multiple Sclerosis Scale (BFiMSS; Pakenham & Cox, 2009), Couple Illness Communication Scale (CICS; Arden-Close et al., 2010), satisfaction, usage and qualitative feedback.

"I have successfully incorporated a balanced diet and exercise program into my life."

- CAN DO Program Participant

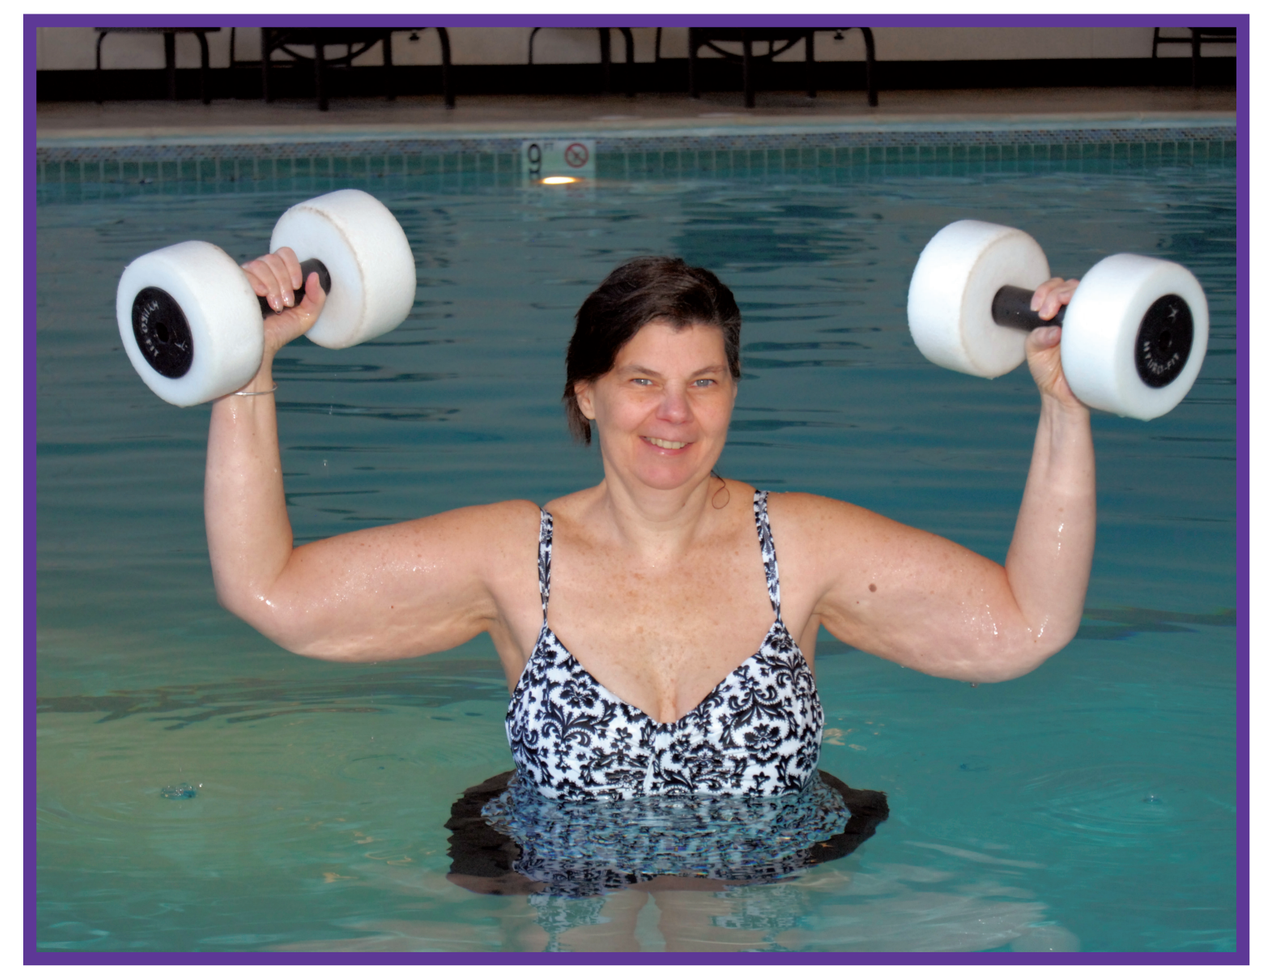

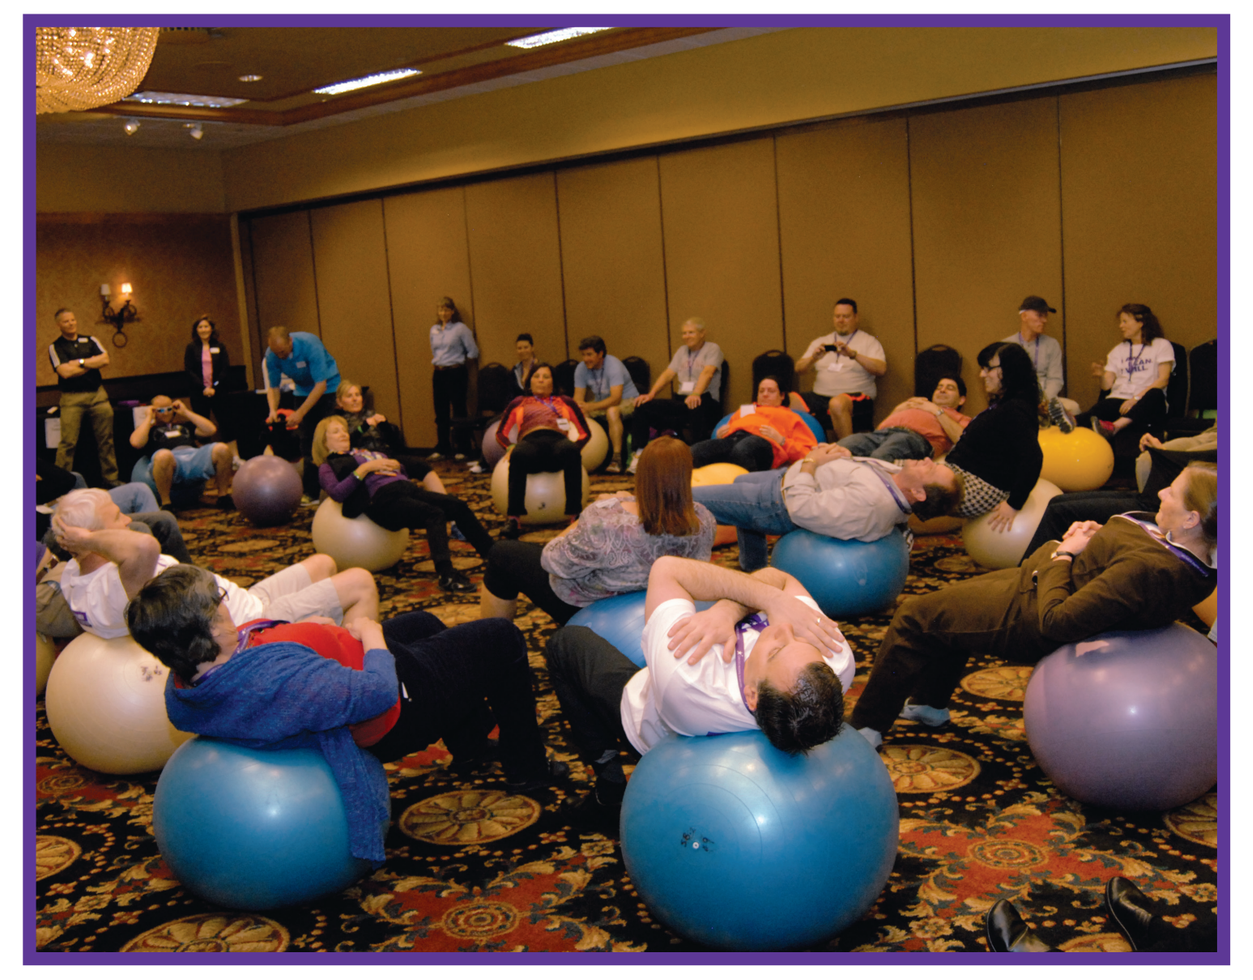

PROCEDURE:

- CAN DO Program is a world-renowned four-day program dedicated exclusively to helping individuals live their best lives with MS through a personal approach that addresses physical, emotional, interpersonal, intellectual and spiritual needs. Goals for participants include gaining knowledge, skills, support and confidence to transform challenges into possibilities.

CAN DO Program Participants

	Persons with MS	Support Partners
Age, mean (SD), y	50.56 (10.32)	53.42 (13.76)
Gender (female)	69 % (W)/ 31% (M)	36% (W)/ 64% (M)
Length of MS, mean (SD), y	10.84 (9.20)	N/A

RESULTS: Long-Term (6-Month) Outcomes

- CAN DO Program produced long-term positive results for participants (Fig. 1-3). Paired sample t-test (p<.05) indicate significant changes from before CAN DO Program to six months after:
 - A significant **increase in confidence in MS Symptom Management** for PwMS (Fig. 1; MS Self-Efficacy Scale (MSSE); Schwartz et al., 1996; t (38) = 6.27, p < .05).
 - A significant **increase in MS Benefit Finding (i.e., personal growth)** for PwMS (Fig. 2; BFiMSS; Pakenham & Cox, 2009; t (37) = 4.02, p < .05).
 - A significant **increase in MS Specific Communication** for PwMS (Fig. 3; Arden-Close et al., 2010; t (38) = 2.52, p < .05).
 - A significant **increase in MS Support Partner Benefit Finding (i.e., personal growth)** for support partners (BFiMSCare; Pakenham & Cox, 2008; t (36) = 2.07, p < .05).

"I have changed my thought process...I am building my team for physical and emotional support."

"As a support partner, my goals were to learn more about living with MS, the program took me above and beyond."

- CAN DO Program Participants

ADDITIONAL 6-MONTH OUTCOMES:

- Achievement of short-term goals set during CAN DO Program (91%).
- Usage of MS specific resources learned about during CAN DO Program (98%).
- Behavior and attitude changes: incorporated Can Do MS mission of empowerment in lives (98% PwMS; 96% SP); living more fully with MS (100% PwMS; 82% SP); made one or more positive behavior changes (e.g., exercise, nutrition; 100% PwMS; 93% SP).

Fig. 1: **Improved Confidence in MS Symptom Managment**

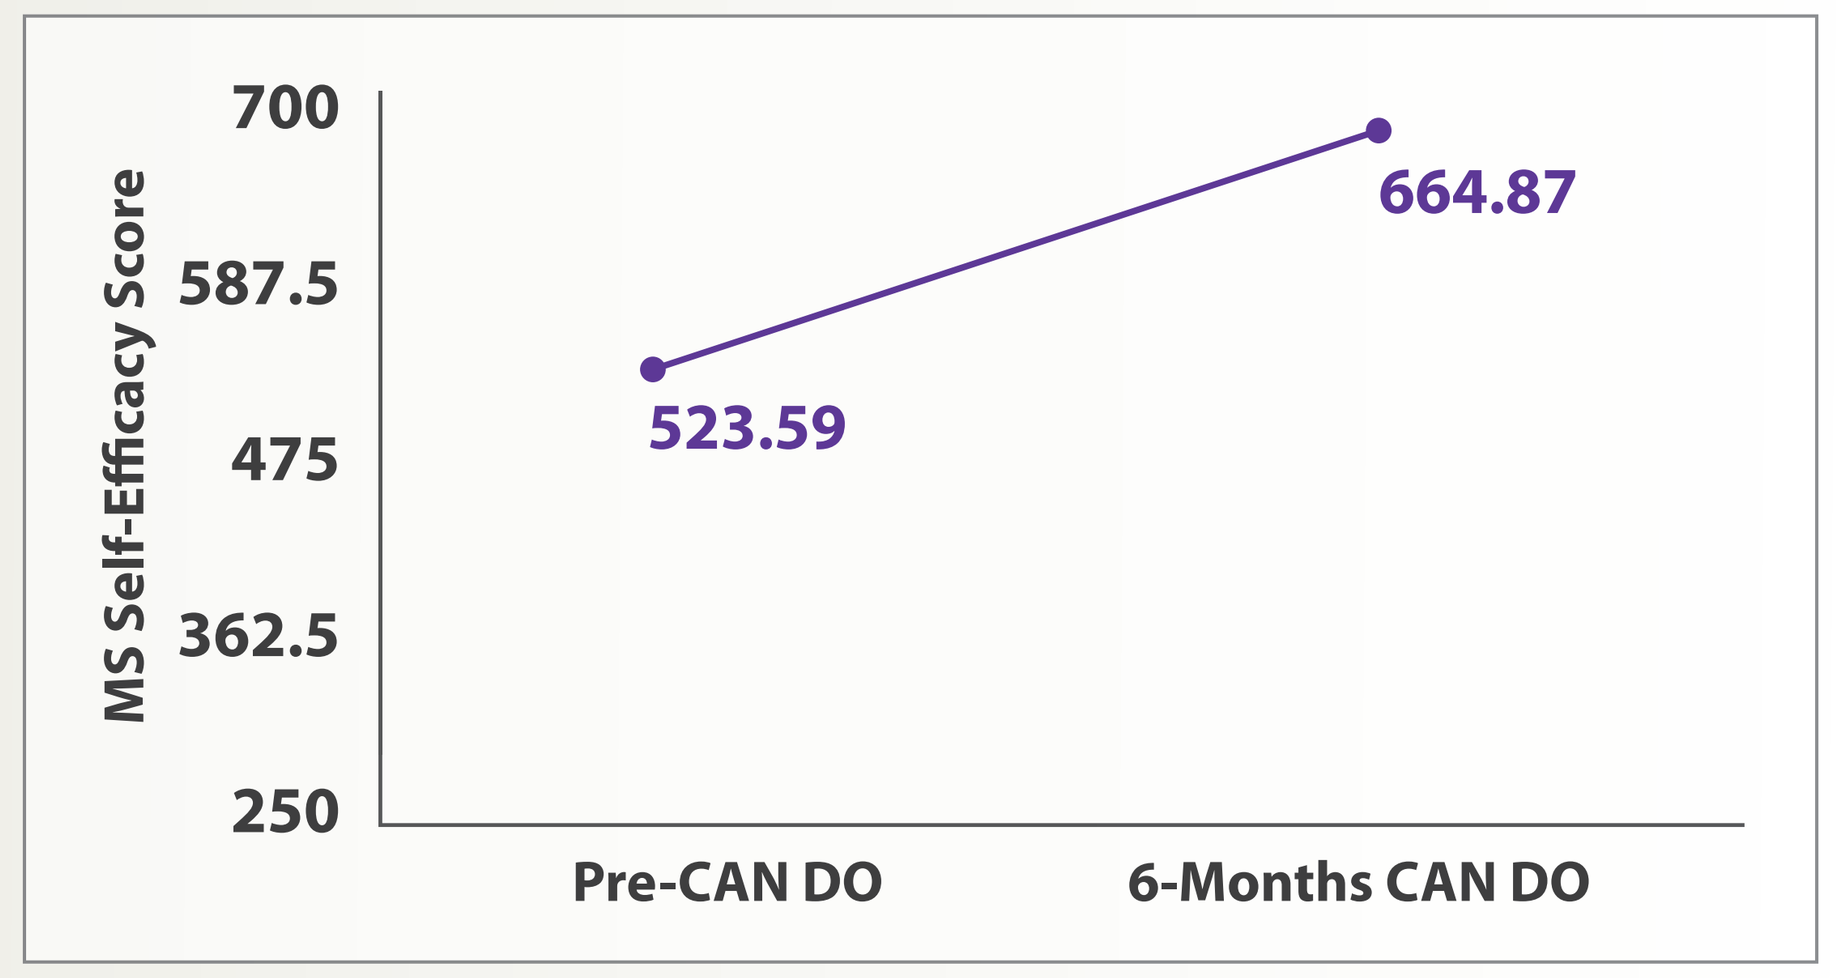

MS Self-Efficacy Score
700
587.5
475
362.5
250
523.59
664.87
Pre-CAN DO
6-Months CAN DO

Fig. 2: **Improved Personal Growth and Hope**

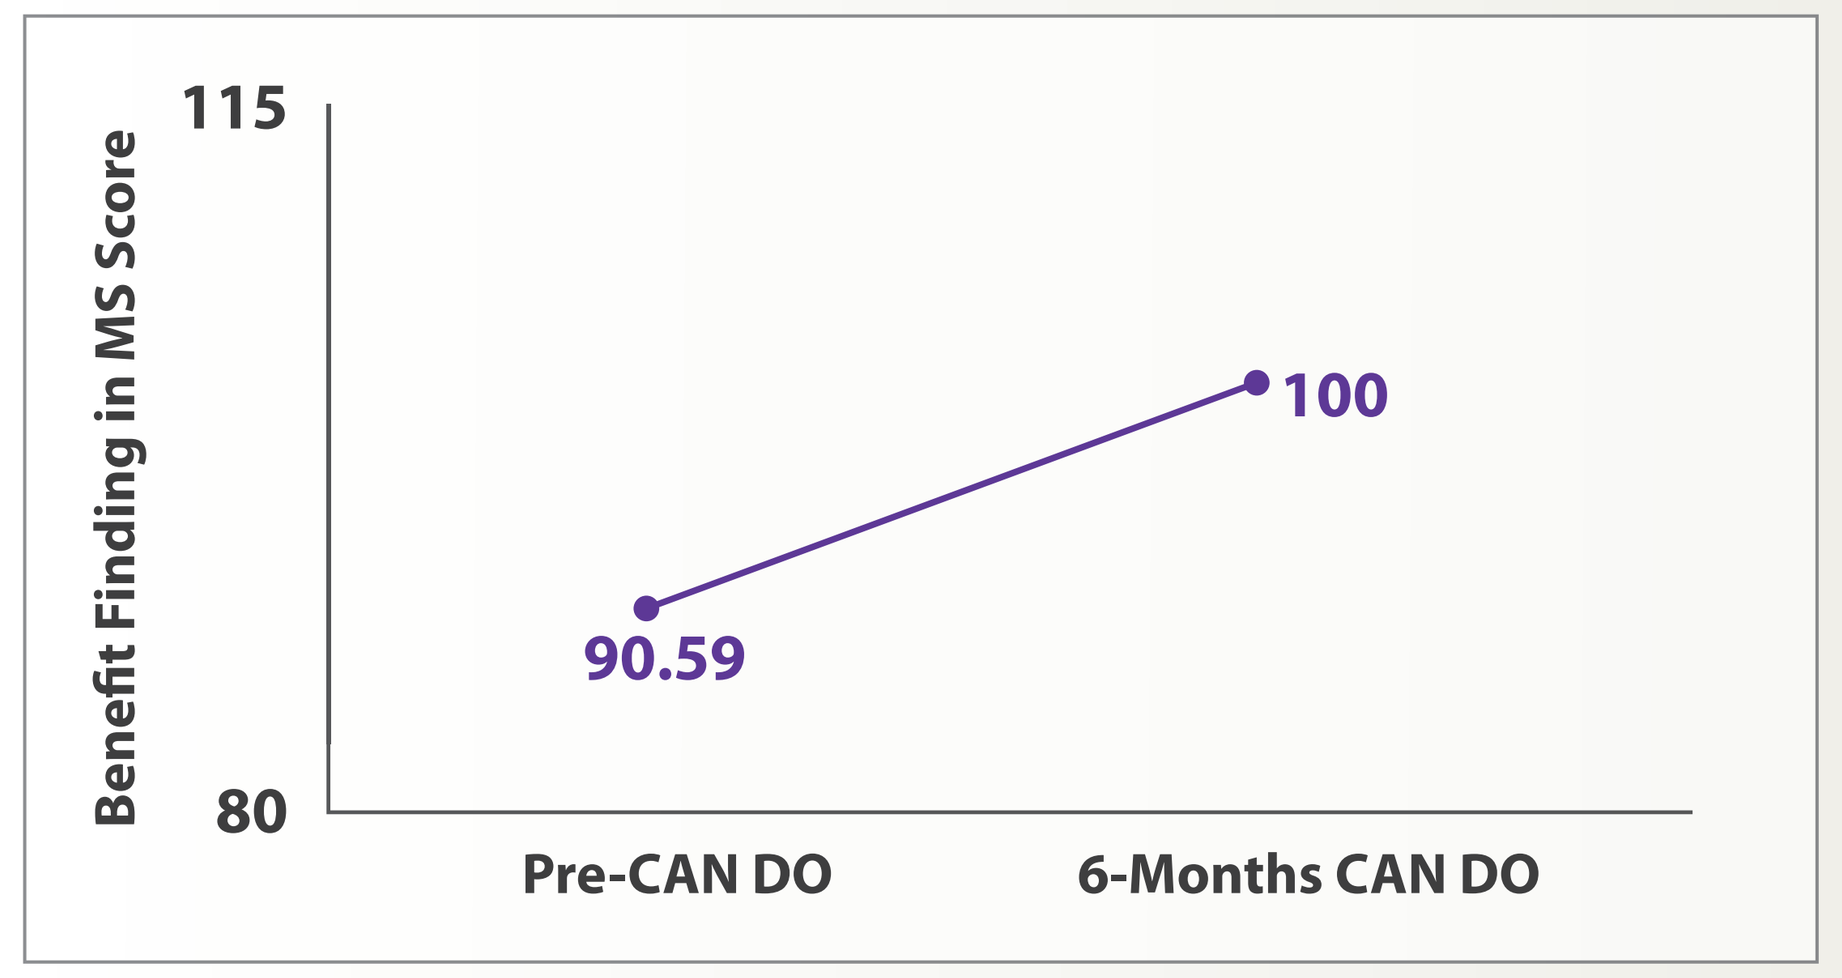

Benefit Finding in MS Score
115
80
90.59
100
Pre-CAN DO
6-Months CAN DO

Fig. 3: **Improved Communication about MS**

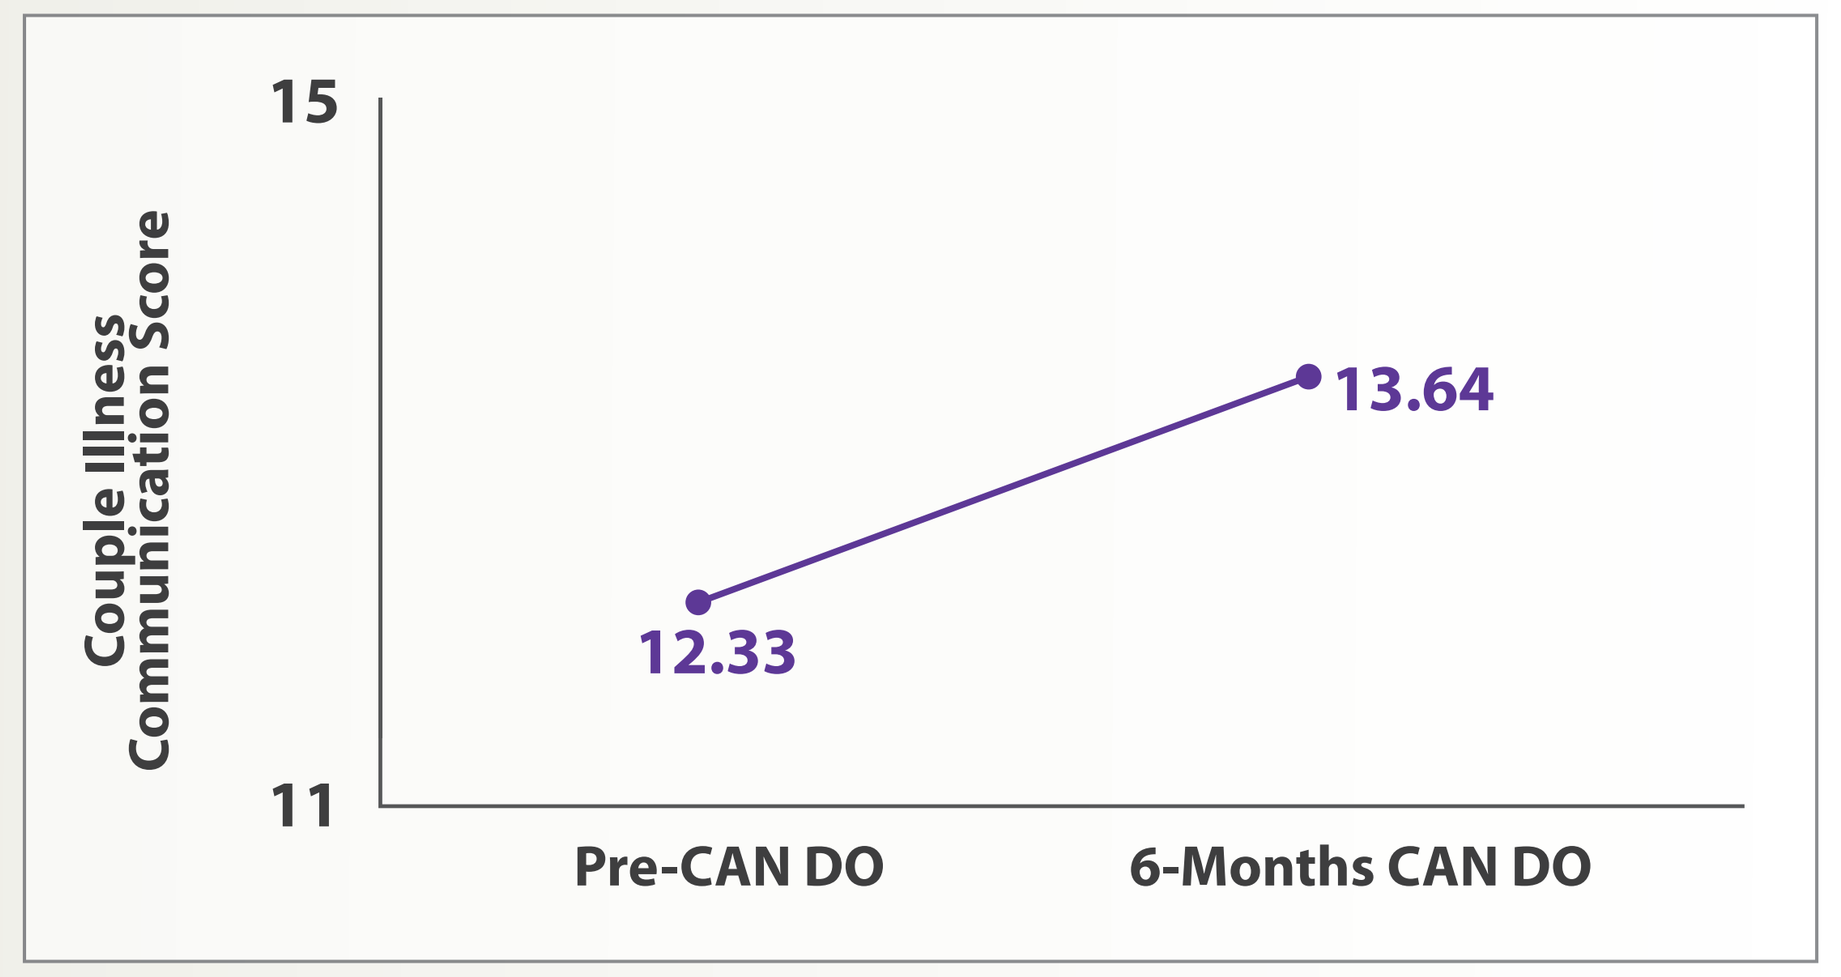

Couple Illness Communication Score
15
11
12.33
13.64
Pre-CAN DO
6-Months CAN DO

Fig. 4: **Behavior & Attitude Changes 6-months Post CAN DO**

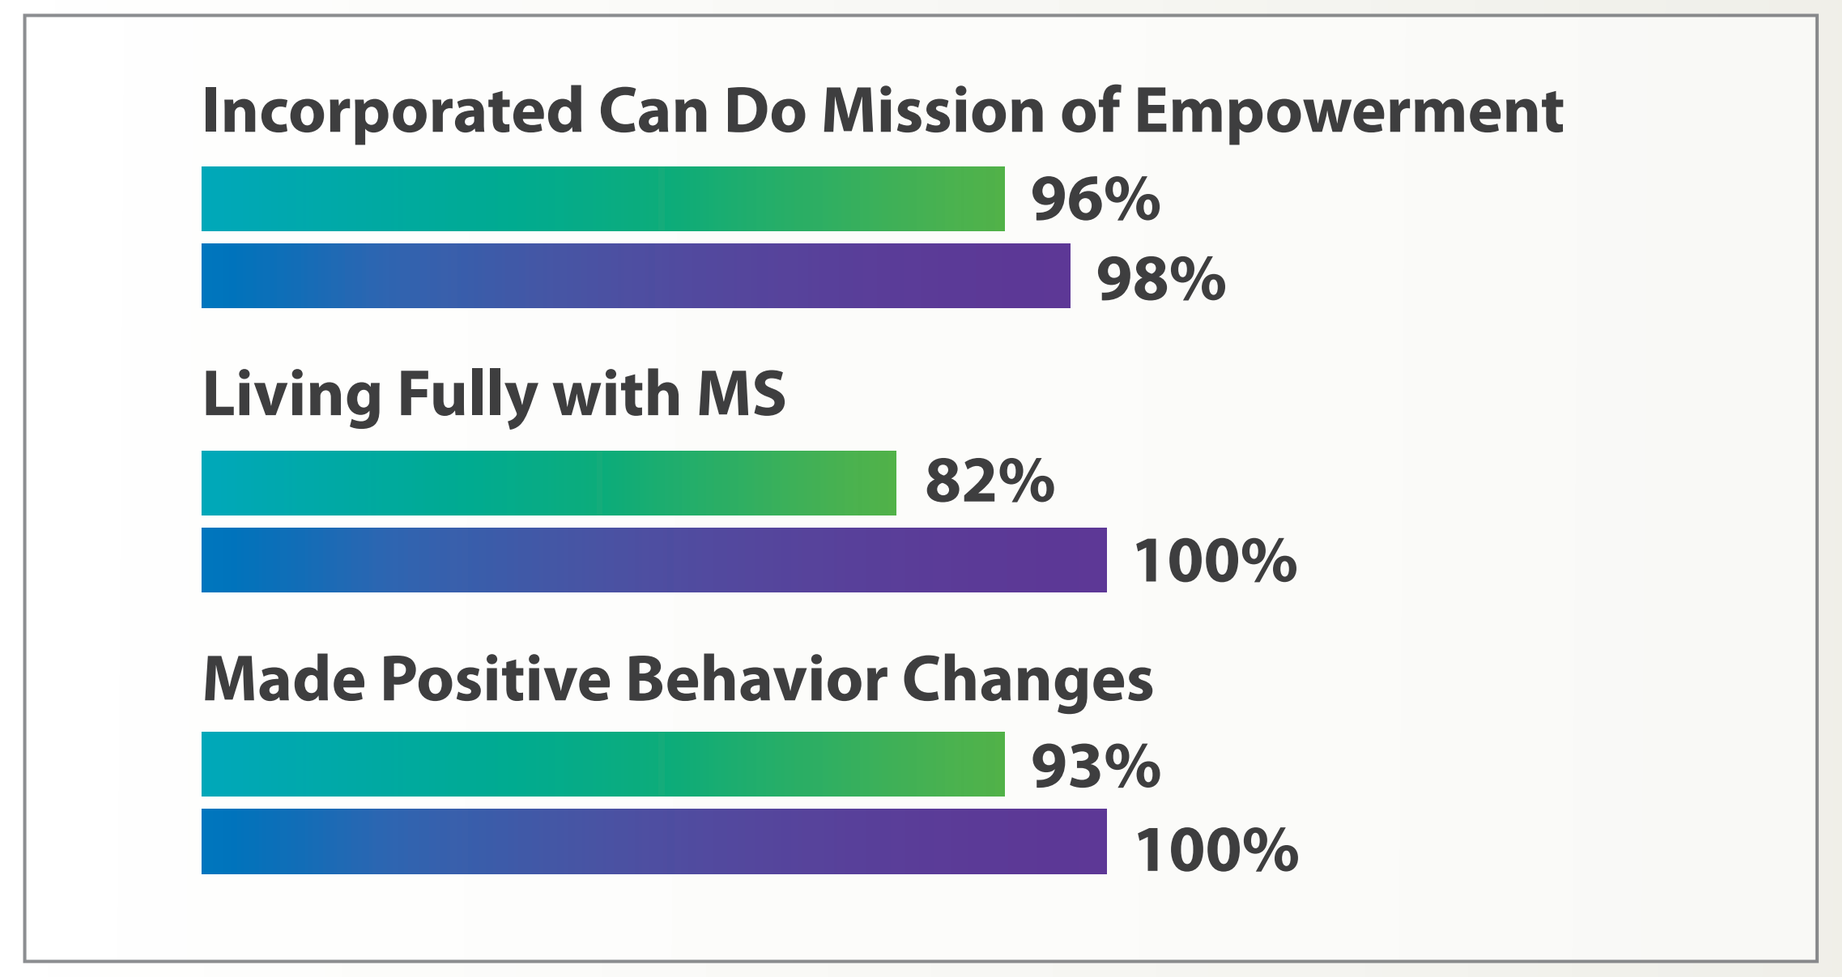

Incorporated Can Do Mission of Empowerment
96%
98%
Living Fully with MS
82%
100%
Made Positive Behavior Changes
93%
100%

CONCLUSION:

- The CAN DO Program is an effective way to create empowerment and assist in multiple aspects of an individual's MS journey. Specifically, CAN DO Program produced outcomes of benefit finding, confidence in MS management and illness specific communication at six months.
- Improved benefit finding is related to meaning-based coping strategies with direct effects on life satisfaction (Pakenham, 2006), demonstrating the Can Do MS mission of addressing multiple areas to help people live more fully with MS. Additionally, confidence in MS is linked to quality of life (Farrell et al., 2004) and may translate to improved management of MS.
- Implications of increasing positive constructs include better medication adherence, less morbidity and increased longevity (e.g., Cuffee et al., 2012; Moskowitz et al. 2008).
- As disease characteristics and clinical symptoms vary widely, interventions tailored to individuals which provide an empowering approach are needed. With a staff to participant ratio of 1.5:1, the CAN DO Program provides an unsurpassed level of personal attention in an intimate, unhurried environment.