DX57 Peginterferon Beta-1a and Management Strategies for Flu-Like Symptoms and Injection Site Reactions: Obtaining Recommendations Using the Delphi Technique

Newsome SD,[1] Centonze D,[2] Huang D,[3] Halper J,[4] Robertson C,[5] You X,[5] Sabatella G,[5] Evilevitch V,[5] Leahy L[5]

[1]Johns Hopkins School of Medicine, Baltimore, MD, USA; [2]Multiple Sclerosis Clinical Center Policlinico Universitario Tor Vergata, Rome, Italy; [3]Consortium of Multiple Sclerosis Centers, Hackensack, NJ, USA; [4]Neurology and Neuroscience Associates, Inc., Akron, OH, USA; [5]Biogen, Cambridge, MA, USA

2015 Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC)

May 27-30, 2015

Indianapolis, IN

INTRODUCTION

- Flu-like symptoms (FLS) and injection site reactions (ISR) have been reported by patients treated with interferon therapies for multiple sclerosis (MS).[1]
- FLS and ISR can impact patient adherence to therapy and thus affect treatment outcome in patients with MS.[2,3]
- FLS and ISR also have been reported for peginterferon beta-1a, which is approved for the treatment of relapsing MS.[4]
 - The Phase 3 ADVANCE study demonstrated that peginterferon beta-1a significantly reduced annualized relapse rate, magnetic resonance imaging lesion activity, and risk of relapse and disability progression vs. placebo.[4]
- Identifying management strategies for FLS and ISR may assist clinicians with improving patient adherence and could potentially impact patient outcomes.
- Here we report management strategies for FLS and ISR associated with peginterferon beta-1a treatment obtained in a consensus-generating method using the Delphi technique which utilizes iterative rounds of questionnaires to build consensus.[5]

METHODS

- ADVANCE was a 2-year, randomized, double-blind, placebo-controlled (Year 1 only) Phase 3 study to evaluate the efficacy and safety of peginterferon beta-1a 125 mcg subcutaneous administered every 2 or 4 weeks.[4]
- ADVANCE investigators with a predefined number of enrolled patients (≥ 2 enrolled patients in the United States and Western Europe or ≥ 10 patients in the rest of world) were offered the opportunity to participate in 2 sequential consensus-generating questionnaires using a modified Delphi methodology.[5]
- The development of the 2 questionnaires was overseen by an independent steering committee of expert clinicians (n=4).
- Those ADVANCE investigators who participated (i.e., Delphi responders) were provided access to 2 Web-based (SurveyMonkey, www.surveymonkey.com) questionnaires.
- Questionnaire 1 consisted of 150 questions designed to collect the responders' observations and experience with the frequency, duration, impact, and management of FLS and ISR in patients with MS treated with peginterferon beta-1a in ADVANCE.
 - Four question formats were used: Yes/no, multiple choice, ranking, and open-ended; both qualitative and quantitative techniques were used to analyze the results.
 - For relevant questions, responders were asked to provide their observations from 2 separate time periods of the study: 0–3 months of treatment (within the first 3 months of treatment) and > 3 months of treatment.
- After completion and review of questionnaire 1 responses, questionnaire 2 was generated (15 questions) to gain consensus on the characteristics of FLS and ISR and recommendations for management of these side effects.
 - All questions used a Likert scale and the response level for consensus was defined a priori by the steering committee as an average rating (AR) of ≥ 2.7 as based on the 4-point scale (1 = strongly disagree, 2 = disagree, 3 = agree, 4 = strongly agree).
- Reported here are results on the management strategies for FLS and ISR. Results on the characteristics and impact of these side effects are presented in poster DX35.[6]

RESULTS

- A total of 30 ADVANCE investigators (i.e., Delphi responders) completed questionnaire 1, and 29 also completed questionnaire 2.

Subcategories of FLS and ISR Observed by Responders

- Fever, myalgia, and chills were the most prevalent FLS reported by responders (Figure 1).
- More than 75% of responders reported that erythema was the most prevalent ISR during both time periods (Figure 1).

Figure 1. The most prevalent FLS and ISR reported by responders during each time period

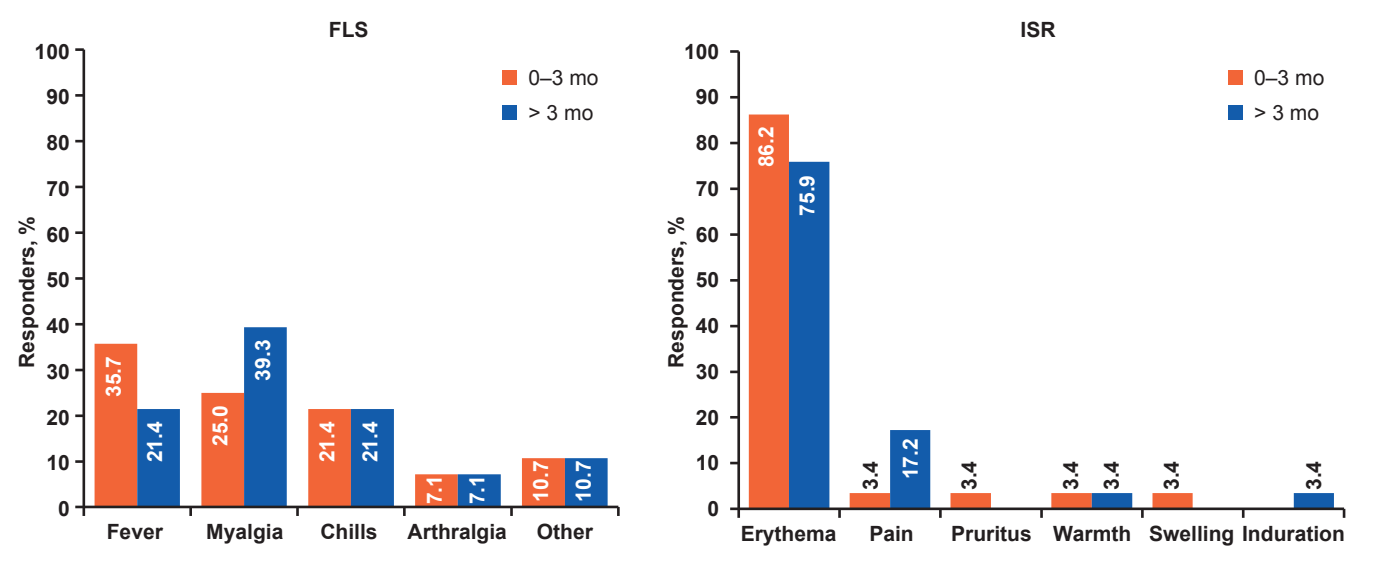
FLS
ISR
0–3 mo
> 3 mo
Responders, %
35.7
21.4
25.0
39.3
21.4
21.4
7.1
7.1
10.7
10.7
Fever
Myalgia
Chills
Arthralgia
Other
86.2
75.9
3.4
17.2
3.4
3.4
3.4
3.4
3.4
Erythema
Pain
Pruritus
Warmth
Swelling
Induration

Management Strategies Utilized in ADVANCE for FLS and ISR

- In questionnaire 1, all responders reported that in the ADVANCE study they recommended or encouraged the use of prophylactic therapy (acetaminophen/ibuprofen) to prevent or manage FLS and 97% of responders recommended the use of nonpharmacological interventions for managing ISR (Table).

Table. Management strategies utilized in ADVANCE for FLS and ISR

Strategy	FLS	ISR
Pharmacological therapy		
% who recommended therapy	100	10
Recommendation	Acetaminophen/ ibuprofen	Diphenhydramine, paracetamol, dimethindene
Additional pharmacological therapy		
% who recommended therapy	14	0
Recommendation	Paracetamol, pentoxifylline, naproxen sodium	NA
Nonpharmacological therapy		
% who recommended therapy	32	97
Recommendations[a,b]	• Altering injection timing • Rest • Cold water showers/ baths • Cooling of injection site • Caffeine • Administering medication at room temperature	• Rotation of injection site (93%) • Patient education (59%) • Warming of medication to room temperature (52%) • Applying ice to the site after injection (34%) • Injection before bed (34%)

NA=not applicable. [a]FLS: These recommendations were provided in an open-ended response. No percentages were obtained as not all responders provided details. [b]ISR: Other recommendations included avoidance of certain sites for injection (17%), warming medication to body temperature (7%), injection early in the day (7%), and apply heat (7%).

Responder Recommended Management of FLS

- In questionnaire 2, responders reached consensus on management strategies for FLS (Figure 2) and recommended that patients should use acetaminophen or a nonsteroidal anti-inflammatory drug (NSAID):
 - When beginning treatment (AR = 3.79)
 - On a scheduled basis (AR = 3.31)
 - As long as needed (AR = 3.52).
- In addition, responders agreed that changing the timing of peginterferon beta-1a injection was an additional management strategy for FLS (AR = 3.28).

Figure 2. Responder consensus on FLS management

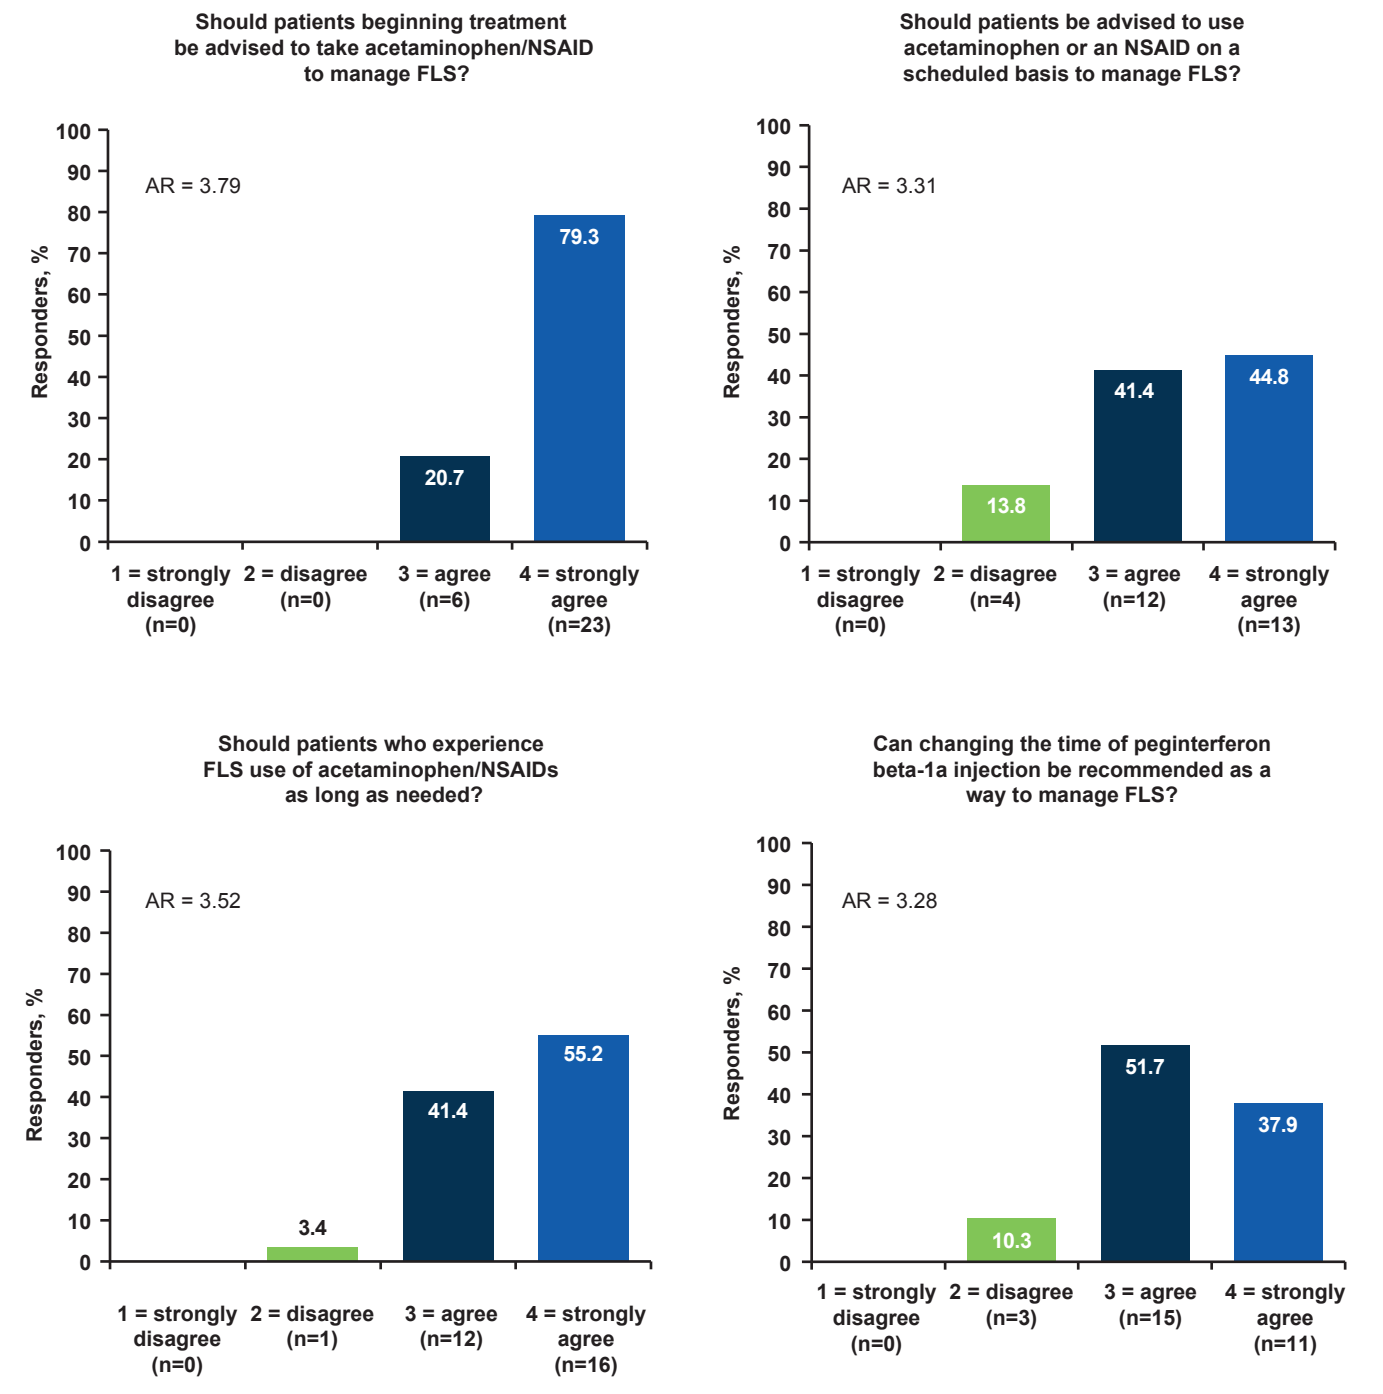
Should patients beginning treatment be advised to take acetaminophen/NSAID to manage FLS?
AR = 3.79
Responders, %
20.7
79.3
1 = strongly disagree (n=0)
2 = disagree (n=0)
3 = agree (n=6)
4 = strongly agree (n=23)
Should patients be advised to use acetaminophen or an NSAID on a scheduled basis to manage FLS?
AR = 3.31
13.8
41.4
44.8
1 = strongly disagree (n=0)
2 = disagree (n=4)
3 = agree (n=12)
4 = strongly agree (n=13)
Should patients who experience FLS use of acetaminophen/NSAIDs as long as needed?
AR = 3.52
3.4
41.4
55.2
1 = strongly disagree (n=0)
2 = disagree (n=1)
3 = agree (n=12)
4 = strongly agree (n=16)
Can changing the time of peginterferon beta-1a injection be recommended as a way to manage FLS?
AR = 3.28
10.3
51.7
37.9
1 = strongly disagree (n=0)
2 = disagree (n=3)
3 = agree (n=15)
4 = strongly agree (n=11)

Responder Consensus on ISR Management

- To prevent or manage ISR (Figure 3), responders recommended that patients should be advised to:
 - Rotate injection site (AR = 3.83)
 - Cool the injection site after injection (AR = 3.10)
 - Administer peginterferon beta-1a at room temperature (AR = 3.41).

Education on FLS and ISR

- In questionnaire 1, responders reported that information on FLS and ISR was provided to patients verbally before initiating treatment in ADVANCE (82% and 79%, respectively).
- In questionnaire 2, all responders agreed that patients beginning treatment should be educated on the characteristics and management of FLS (AR = 3.97) and ISR (AR = 3.83), including:
 - Frequency of FLS and ISR
 - Severity of FLS and ISR
 - Presentation of ISR (timing, size, skin changes)
 - Management of FLS and ISR.

Figure 3. Responder consensus on ISR management

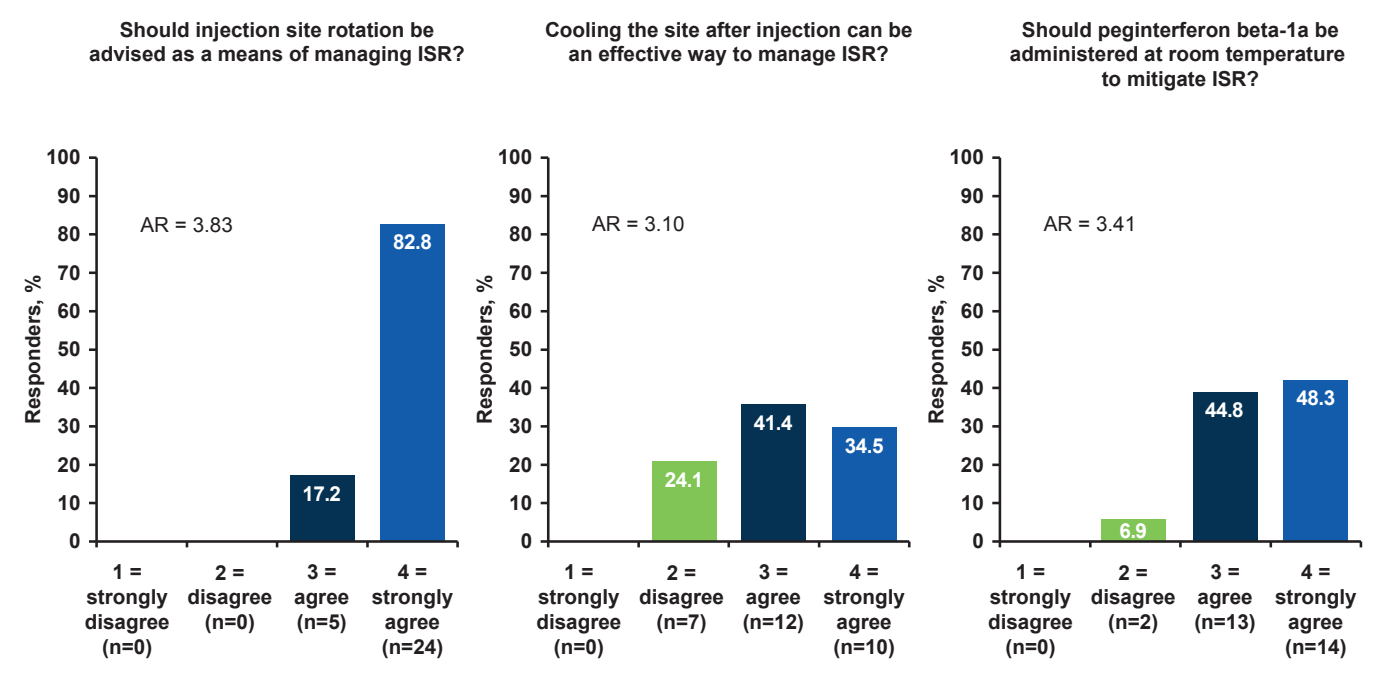
Should injection site rotation be advised as a means of managing ISR?
AR = 3.83
Responders, %
17.2
82.8
1 = strongly disagree (n=0)
2 = disagree (n=0)
3 = agree (n=5)
4 = strongly agree (n=24)
Cooling the site after injection can be an effective way to manage ISR?
AR = 3.10
24.1
41.4
34.5
1 = strongly disagree (n=0)
2 = disagree (n=7)
3 = agree (n=12)
4 = strongly agree (n=10)
Should peginterferon beta-1a be administered at room temperature to mitigate ISR?
AR = 3.41
6.9
44.8
48.3
1 = strongly disagree (n=0)
2 = disagree (n=2)
3 = agree (n=13)
4 = strongly agree (n=14)

CONCLUSIONS

- The use of a 2-part Delphi questionnaire resulted in consensus on the characteristics and management strategies for FLS and ISR.
- Delphi responders recommended prophylactic therapy (acetaminophen/NSAIDs) and change of injection timing to prevent or manage FLS, and injection site rotation/cooling and drug administration at room temperature to prevent or manage ISR.
- Delphi responders agreed that educating patients on the characteristics and management of FLS and ISR before starting treatment is critical to set treatment expectations and promote adherence.
- The Delphi responders were a small subset of investigators who participated in the study and their observations were based only on the number of patients enrolled at their site in a clinical study. Thus, these results should be confirmed after gaining more experience with peginterferon beta-1a in clinical practice.

References

1. Walther EU, Hohlfeld R. *Neurology*. 1999;53(8):1622-1627.
2. Steinberg SC, *et al. Clin Drug Investig*. 2010;30(2):89-100.
3. Costello K, *et al. Medscape J Med*. 2008;10(9):225.
4. Calabresi PA, *et al*; ADVANCE Study Investigators. *Lancet Neurol*. 2014;13(7):657-665.
5. Hsu C-C, Sandford BA. *Practical Assessment, Research & Evaluation*. 2007;12(10):1-8.
6. Huang D, *et al*. Evaluation of peginterferon beta-1a tolerability profile from the ADVANCE study: gaining consensus using the Delphi technique (poster DX35). Presented at: 2015 Annual Meeting of the Consortium of Multiple Sclerosis Centers; May 27–30, 2015; Indianapolis, IN.

Disclosures

This study was supported by Biogen (Cambridge, MA, USA). *SDN*: advisory board for Biogen, Genzyme, and Novartis; research support from Biogen and Novartis; *DC*: speaker/consulting fees and/or research support from Almirall, Bayer HealthCare, Biogen, Genzyme, GW Pharmaceuticals, Merck Serono, Novartis, Sanofi-Aventis, and Teva; *JH*: fees for non-CME services from Biogen; *DH*: consultant/advisory board for Biogen and Teva; *CR*, *XY*, *GS*, *VE*, and *LL*: employees of and stockholders in Biogen.

Acknowledgments

Writing and editorial support of this poster was provided by Maria Hovenden, PhD, (Excel Scientific Solutions, Southport, CT, USA); with funding provided by Biogen (Cambridge, MA, USA). Biogen reviewed and provided feedback on the poster. The authors had full editorial control of the poster and provided their final approval of all content.

For an electronic version of this poster, please scan code.