Four-year Expanded Disability Status Scale outcomes in patients treated with fingolimod in the phase 3 and extension trial program

DX17

Bruce Cree[1], Jeffrey Cohen[2], Peter Chin[3], Shannon Ritter[4], Daniela Piani Meier[5], Ludwig Kappos[6]

[1]Multiple Sclerosis Center, University of California, San Francisco, CA, USA; [2]Neurological Institute, Cleveland Clinic, Cleveland, OH, USA; [3]Formerly Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [4]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [5]Novartis Pharma AG, Basel, Switzerland; [6]Departments of Neurology and Biomedicine, University Hospital Basel, Basel, Switzerland

CONCLUSIONS

- **In the combined phase 3 and extension trials of patients with MS treated with fingolimod for up to 4 years, the vast majority remained ambulatory without the need for walking assistance**
 - **Approximately two-thirds of patients continuing on fingolimod treatment had stable EDSS scores after 2, 3 and 4 years of treatment: within this group 16–18% had improved scores**
- **Younger patients and those with more recent onset disease were more likely to demonstrate EDSS score improvement or stability while treated with fingolimod**
- **Patients with EDSS scores 3 or 4 and higher at the start of fingolimod treatment had better odds of improving or remaining stable, perhaps reflecting better treatment response or more reliable EDSS score determination in this range compared to EDSS scores between 0 and 1.5**
- **Absence of a control group and selective drop-outs may bias these results**
- **Additional analysis of factors associated with long-term EDSS status could help clinicians better understand and optimize long-term treatment with fingolimod**

INTRODUCTION

- Multiple sclerosis (MS) is a chronic neurological disease characterized by considerable variability in disability progression. Expanded Disability Status Scale (EDSS) score is the standard measure of disability in MS. The EDSS quantifies disability in eight functional systems and allows neurologists to assign scores. EDSS scores 1.0–4.5 refer to patients who are fully ambulatory, and EDSS scores 5.0–9.5 define impairment to ambulation. Assessment of long-term disability is important for characterizing the benefit–risk profile of disease-modifying MS therapies
- Once-daily oral fingolimod 0.5 mg (FTY720; Gilenya®, Novartis Pharma AG), a sphingosine 1-phosphate receptor modulator, is approved for the treatment of relapsing MS[a]
- Fingolimod 0.5 mg demonstrated efficacy on measures of MS disease activity including disability progression, relapses, MRI activity and brain volume loss in the FREEDOMS and TRANSFORMS extension studies[1,2]
- Here we explore longitudinal EDSS outcomes in the pooled cohort of patients treated with fingolimod in the phase 3 and extension trial program

[a]The approved indication may vary from country to country. In the European Union, fingolimod is approved for treatment of patients with highly active relapsing–remitting MS. In the United States, it is approved for the treatment of patients with relapsing forms of MS

OBJECTIVE

- To evaluate EDSS score over time in fingolimod-treated patients in the phase 3 FREEDOMS, FREEDOMS II and TRANSFORMS trials and their extensions

METHODS

- The analysis cohort consisted of patients initiating treatment with fingolimod in the 24-month FREEDOMS and FREEDOMS II studies, 12-month TRANSFORMS study or their respective extension studies (**Figure 1**)
- EDSS data for the intervals after the first dose of fingolimod were pooled for the *post hoc* analysis
- Kaplan–Meier estimates of proportions reaching EDSS scores ≥4 or ≥6 or ≥7 during fingolimod treatment were calculated from start of fingolimod treatment for the 0.5 mg and combined-dose (0.5 mg and 1.25 mg) cohorts
- Proportions with EDSS score less than or equal to the score at start of fingolimod (stable status) and proportions with decreased EDSS score compared with the start of fingolimod (improved status) were analyzed descriptively after 24, 36 and 48 months

Figure 1. Fingolimod studies pooled for the *post hoc* analysis of EDSS outcomes in patients treated with fingolimod in the phase 3 and extension trial program

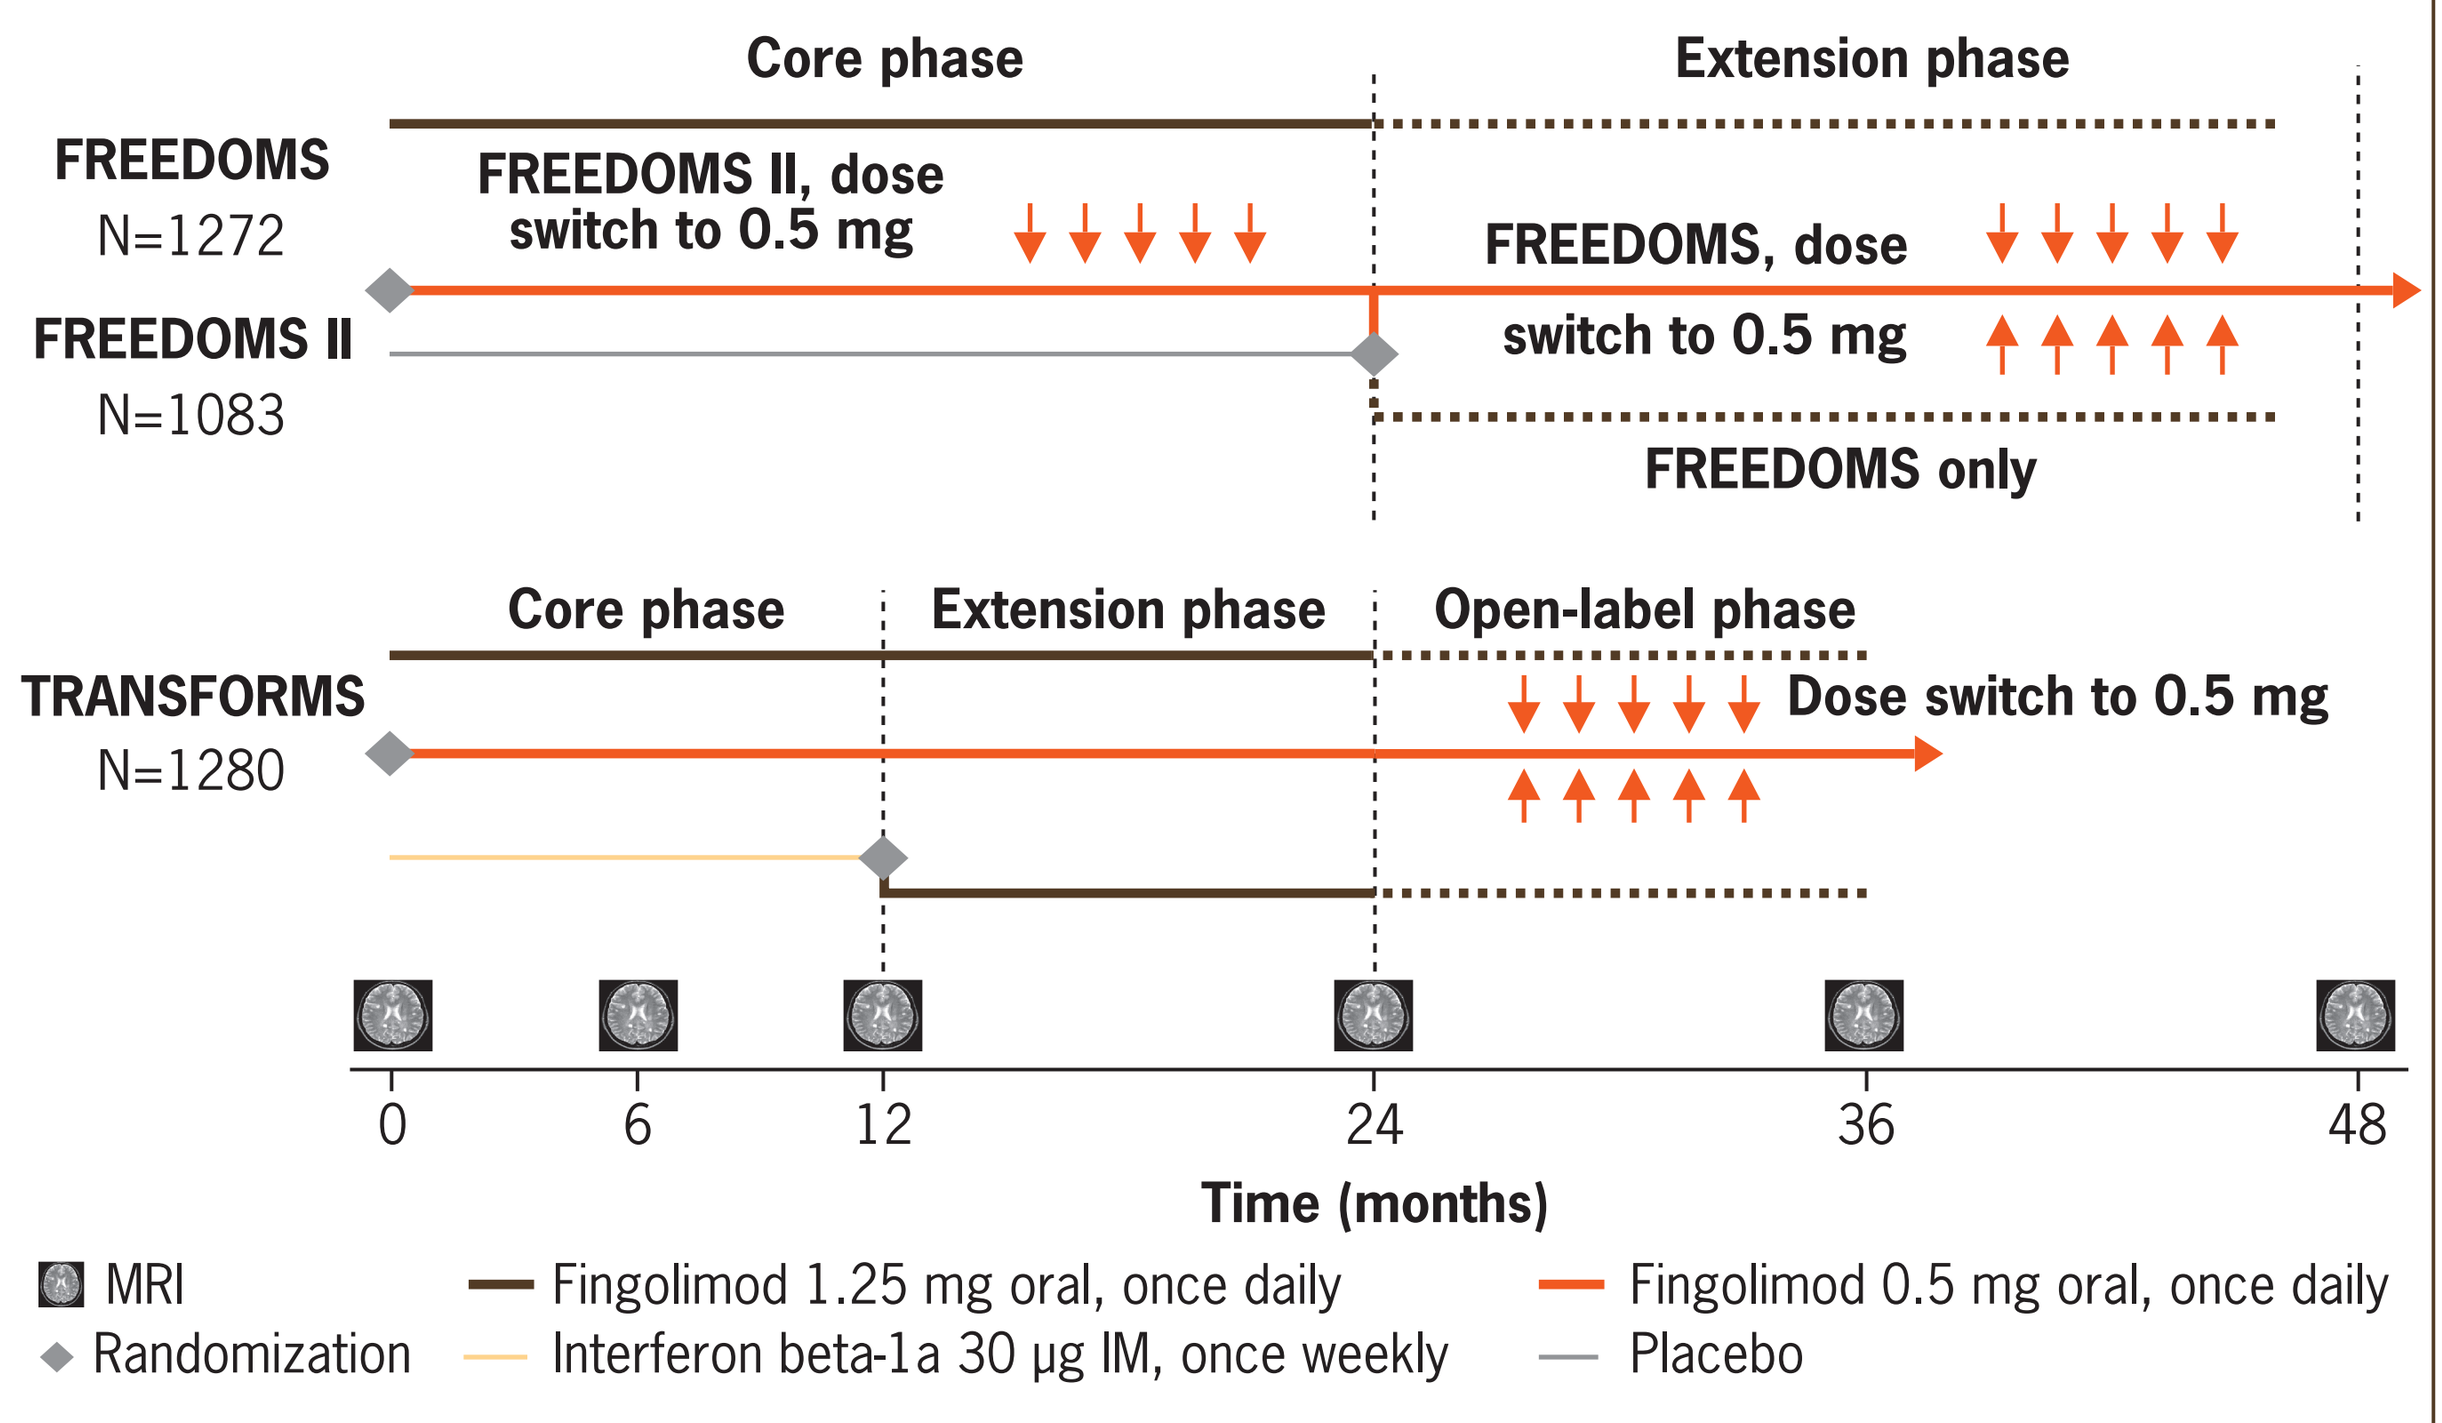
Core phase
Extension phase
FREEDOMS
N=1272
FREEDOMS II, dose switch to 0.5 mg
FREEDOMS, dose switch to 0.5 mg
FREEDOMS II
N=1083
FREEDOMS only
Core phase
Extension phase
Open-label phase
TRANSFORMS
N=1280
Dose switch to 0.5 mg
0
6
12
24
36
48
Time (months)
MRI
Randomization
Fingolimod 1.25 mg oral, once daily
Interferon beta-1a 30 µg IM, once weekly
Fingolimod 0.5 mg oral, once daily
Placebo

EDSS data from patients initiating fingolimod in the 24-month FREEDOMS and FREEDOMS II studies and 12-month TRANSFORMS study and their respective extension studies were pooled for *post hoc* analysis for the intervals after the first dose of fingolimod
EDSS, Expanded Disability Status Scale; IM, intramuscular; MRI, magnetic resonance imaging

- Logistic regression analyses were conducted in the larger combined-dose group to explore factors associated with stable EDSS score or improved EDSS score after 48 months as dependent variables. Fixed independent variables in the models included study, age and EDSS scores at the start of fingolimod treatment (categorized as 0–1.5, 2–2.5, 3–3.5 and ≥4)
- The following single variables were then evaluated, each in a separate logistic regression model that also included the fixed variables:
 - Sex; disease duration prior to core study entry; number of relapses in the 2 years prior to core study entry; prior disease-modifying treatment before start of core study; T2 lesion volume at start of fingolimod treatment; number of gadolinium (Gd)-enhancing T1 lesions at start of fingolimod treatment; and normalized brain volume at start of core study
 - Odds ratios (OR) with associated 95% confidence intervals together with P-values for each effect in the model are presented

RESULTS

- The pooled fingolimod 0.5 mg/all fingolimod dose (n=1641/3283) cohorts had mean and median (25th, 75th percentile) treatment exposures of 920/882 and 967/918 (556/482, 1343/1325) days. Numbers of patients by time point are shown in **Table 1**
- Kaplan–Meier estimates of the proportions of patients reaching EDSS scores ≥4, ≥6 or ≥7 indicate over 90% of fingolimod-treated patients remained ambulatory without assistance after 4 years (**Figure 2**)
- **Figure 3** shows the proportions of fingolimod-treated patients with stable or improved EDSS scores from the start of fingolimod treatment to months 24, 36 and 48

Baseline (at the start of the fingolimod treatment) factors associated with stable EDSS score at 48 months

- Fingolimod-treated patients with higher age (OR: 0.66, p<0.0001) and longer disease duration (OR: 0.76, p=0.01) at baseline were less likely to have a stable EDSS score after 4 years (**Figure 4**)
- Patients with an EDSS score ≥4 at the start of fingolimod had significantly higher odds of remaining stable compared to the reference group with an EDSS score 0–1.5 (**Figure 4**)
- An overall study effect was significant (p=0.04), with FREEDOMS patients exhibiting a greater trend toward stability; however, differences between individual studies were not significant

Baseline (at the start of the fingolimod treatment) factors associated with improved EDSS score at 48 months

- Older patients (OR: 0.56, p<0.0001) and those with longer disease duration (OR: 0.72, p=0.03) at the start of fingolimod treatment were less likely to have an improved EDSS score after 4 years (**Figure 4**)

Table 1. Duration of exposure to fingolimod by dose

	Fingolimod 0.5 mg (n=1641)	All fingolimod doses (n=3283)
Exposure (days)		
Mean (SD)	920 (469.5)	882 (480.1)
Median (range)	967 (2–1782)	918 (1–1782)
Duration of exposure (days), n (%)		
≥1	1641 (100.0)	3283 (100.0)
≥360 (1 year)	1361 (82.9)	2650 (80.7)
≥720 (2 years)	1087 (66.2)	2066 (62.9)
≥1080 (3 years)	724 (44.1)	1350 (41.1)
≥1440 (4 years)	193 (11.8)	360 (11.0)

All fingolimod dose group includes patients who took either fingolimod 1.25 mg or 0.5 mg

- Patients with EDSS scores ≥2 at the start of fingolimod treatment had higher odds of improving compared to the reference group with EDSS score 1–1.5 as follows: EDSS score 2–2.5 (OR: 1.8, p=0.01); EDSS score 3–3.5 (OR: 5.5, p<0.0001); EDSS score ≥4 (OR: 4.0, p<0.0001) (**Figure 4**)
- Normalized brain volume at randomization (OR: 1.34 per 100 cm^3, p=0.01) and number of Gd-enhancing lesions (OR: 1.04, p=0.03) at start of fingolimod were also associated with higher odds of improvement

Figure 2. Kaplan–Meier estimates for proportions of patients reaching EDSS scores ≥4, ≥6 and ≥7 at any time after first dose of fingolimod

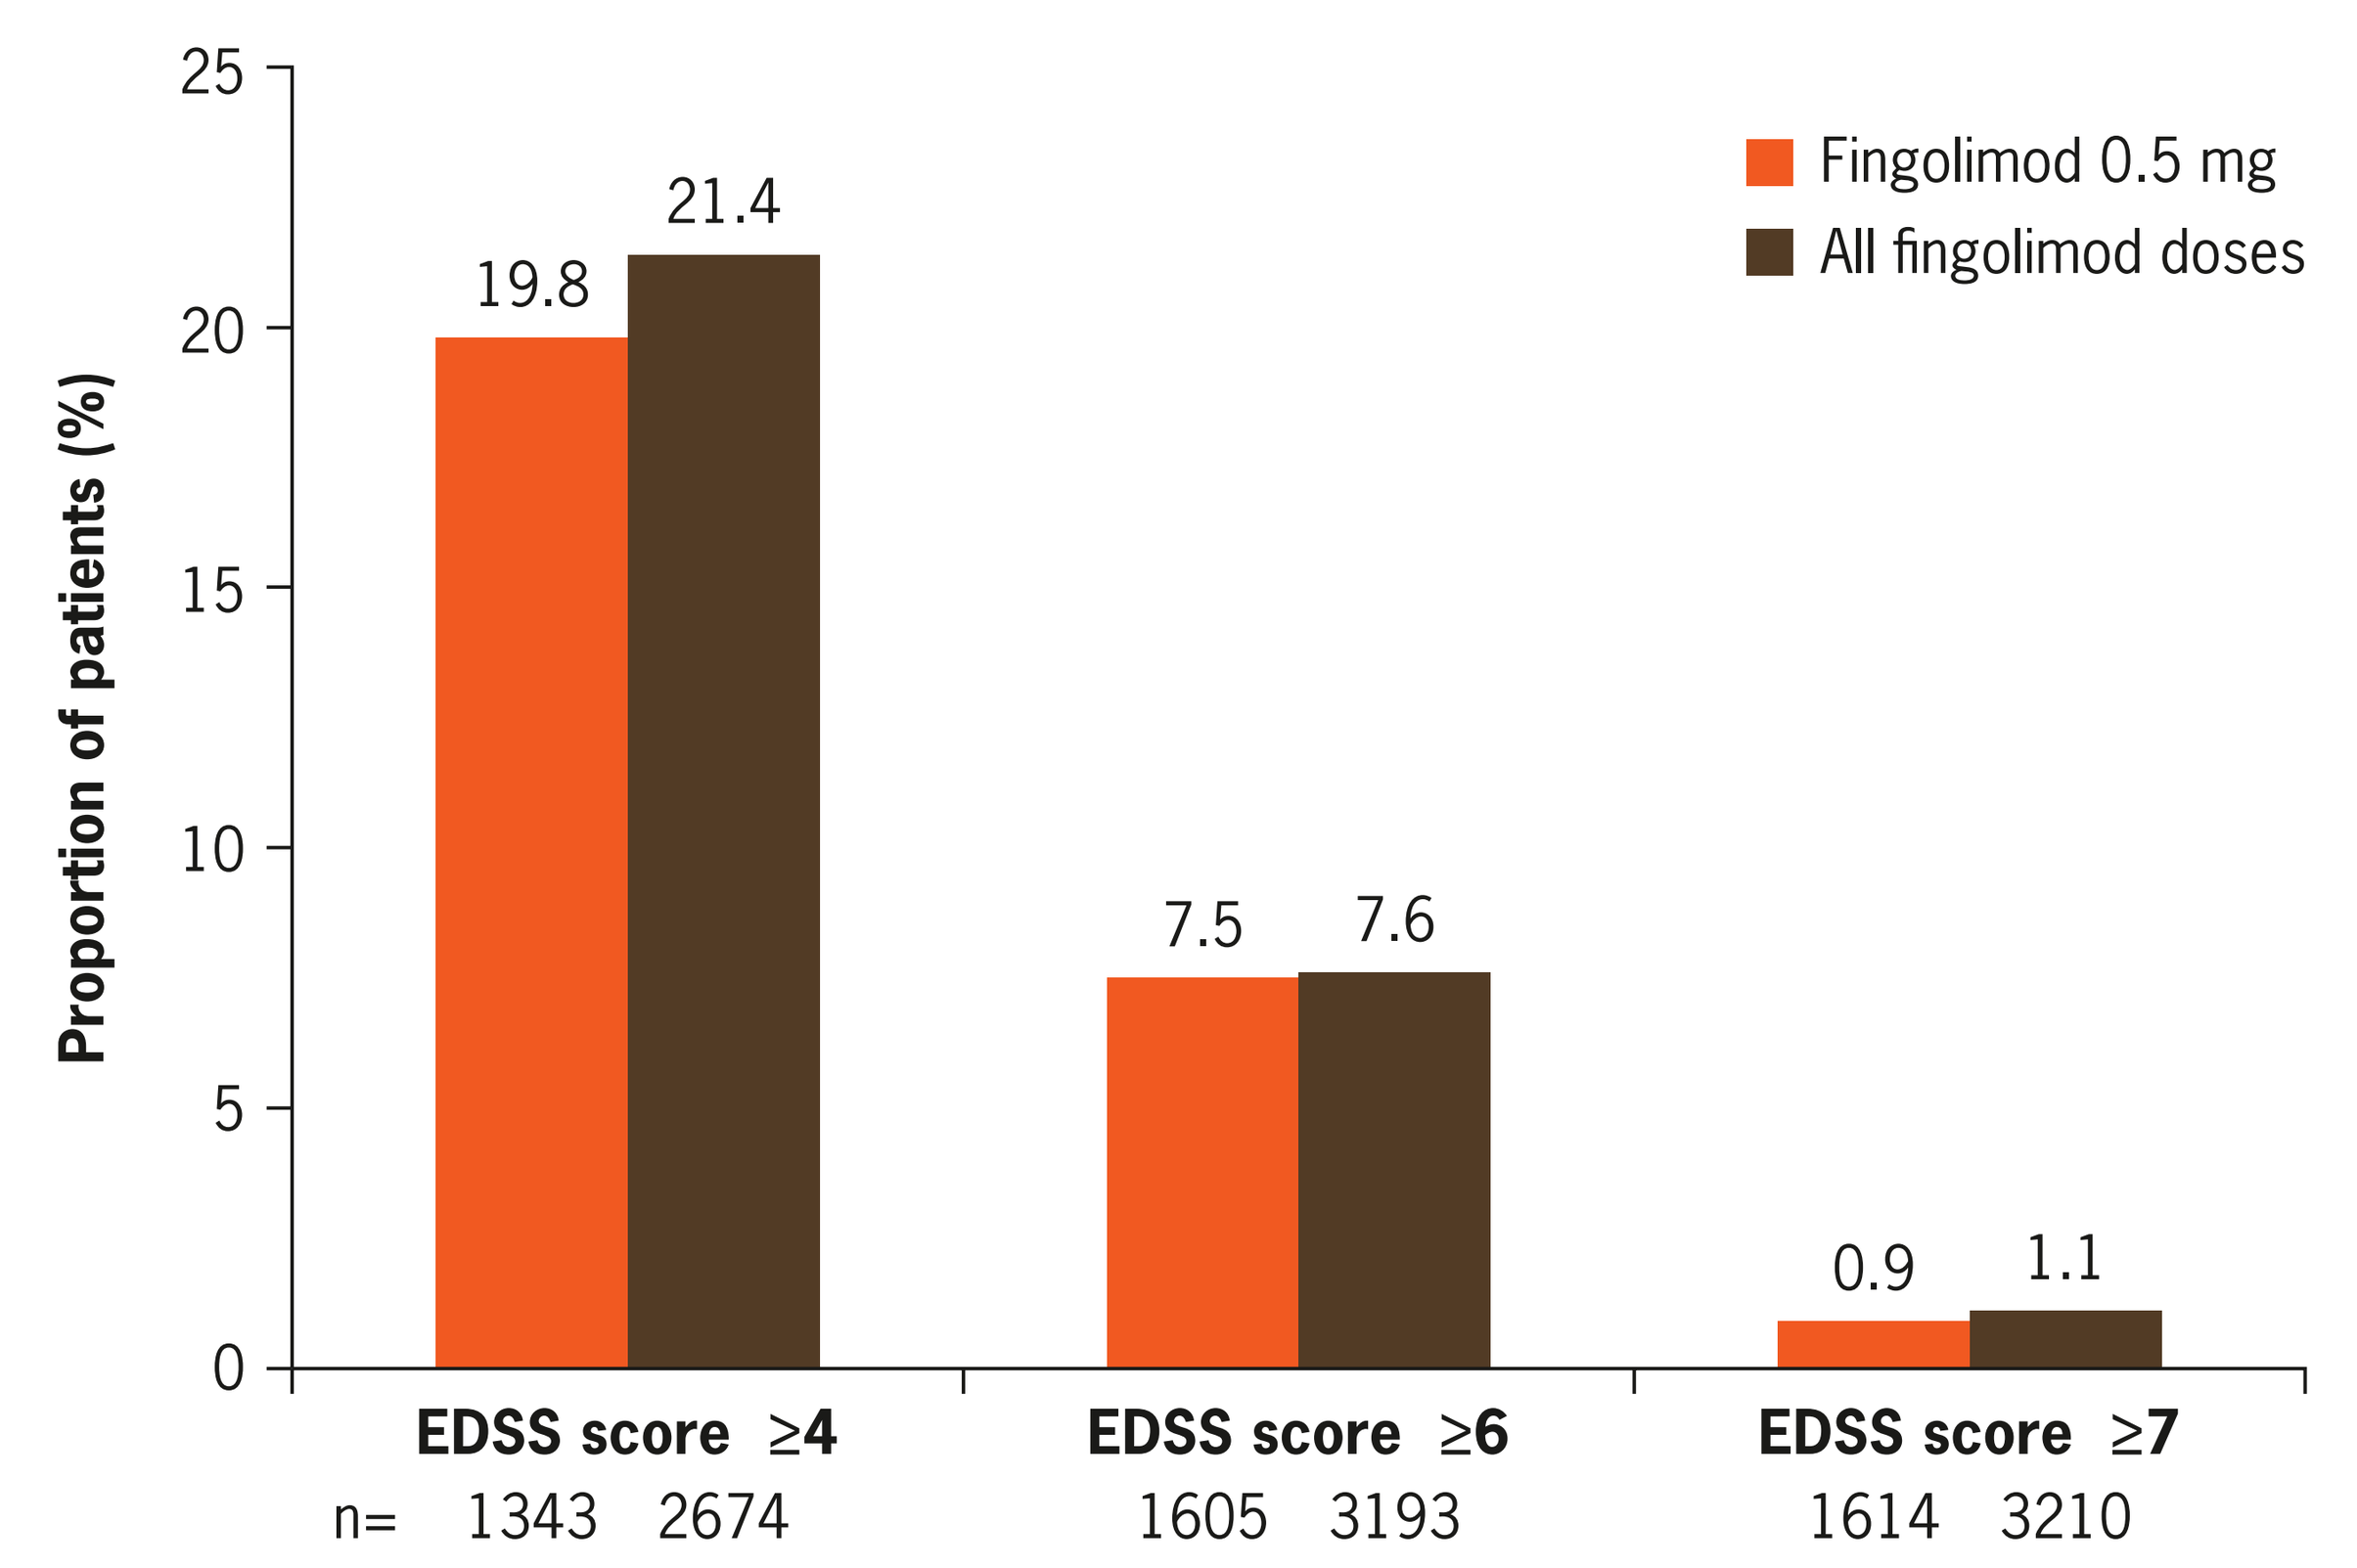
Fingolimod 0.5 mg
All fingolimod doses
Proportion of patients (%)
25
20
15
10
5
0
19.8
21.4
7.5
7.6
0.9
1.1
EDSS score ≥4
EDSS score ≥6
EDSS score ≥7
n= 1343 2674
1605 3193
1614 3210

n=number of patients assessed for EDSS scores
EDSS, Expanded Disability Status Scale

Figure 3. Proportions of fingolimod-treated patients with stable or improved EDSS scores from the start of fingolimod treatment to months 24, 36 and 48

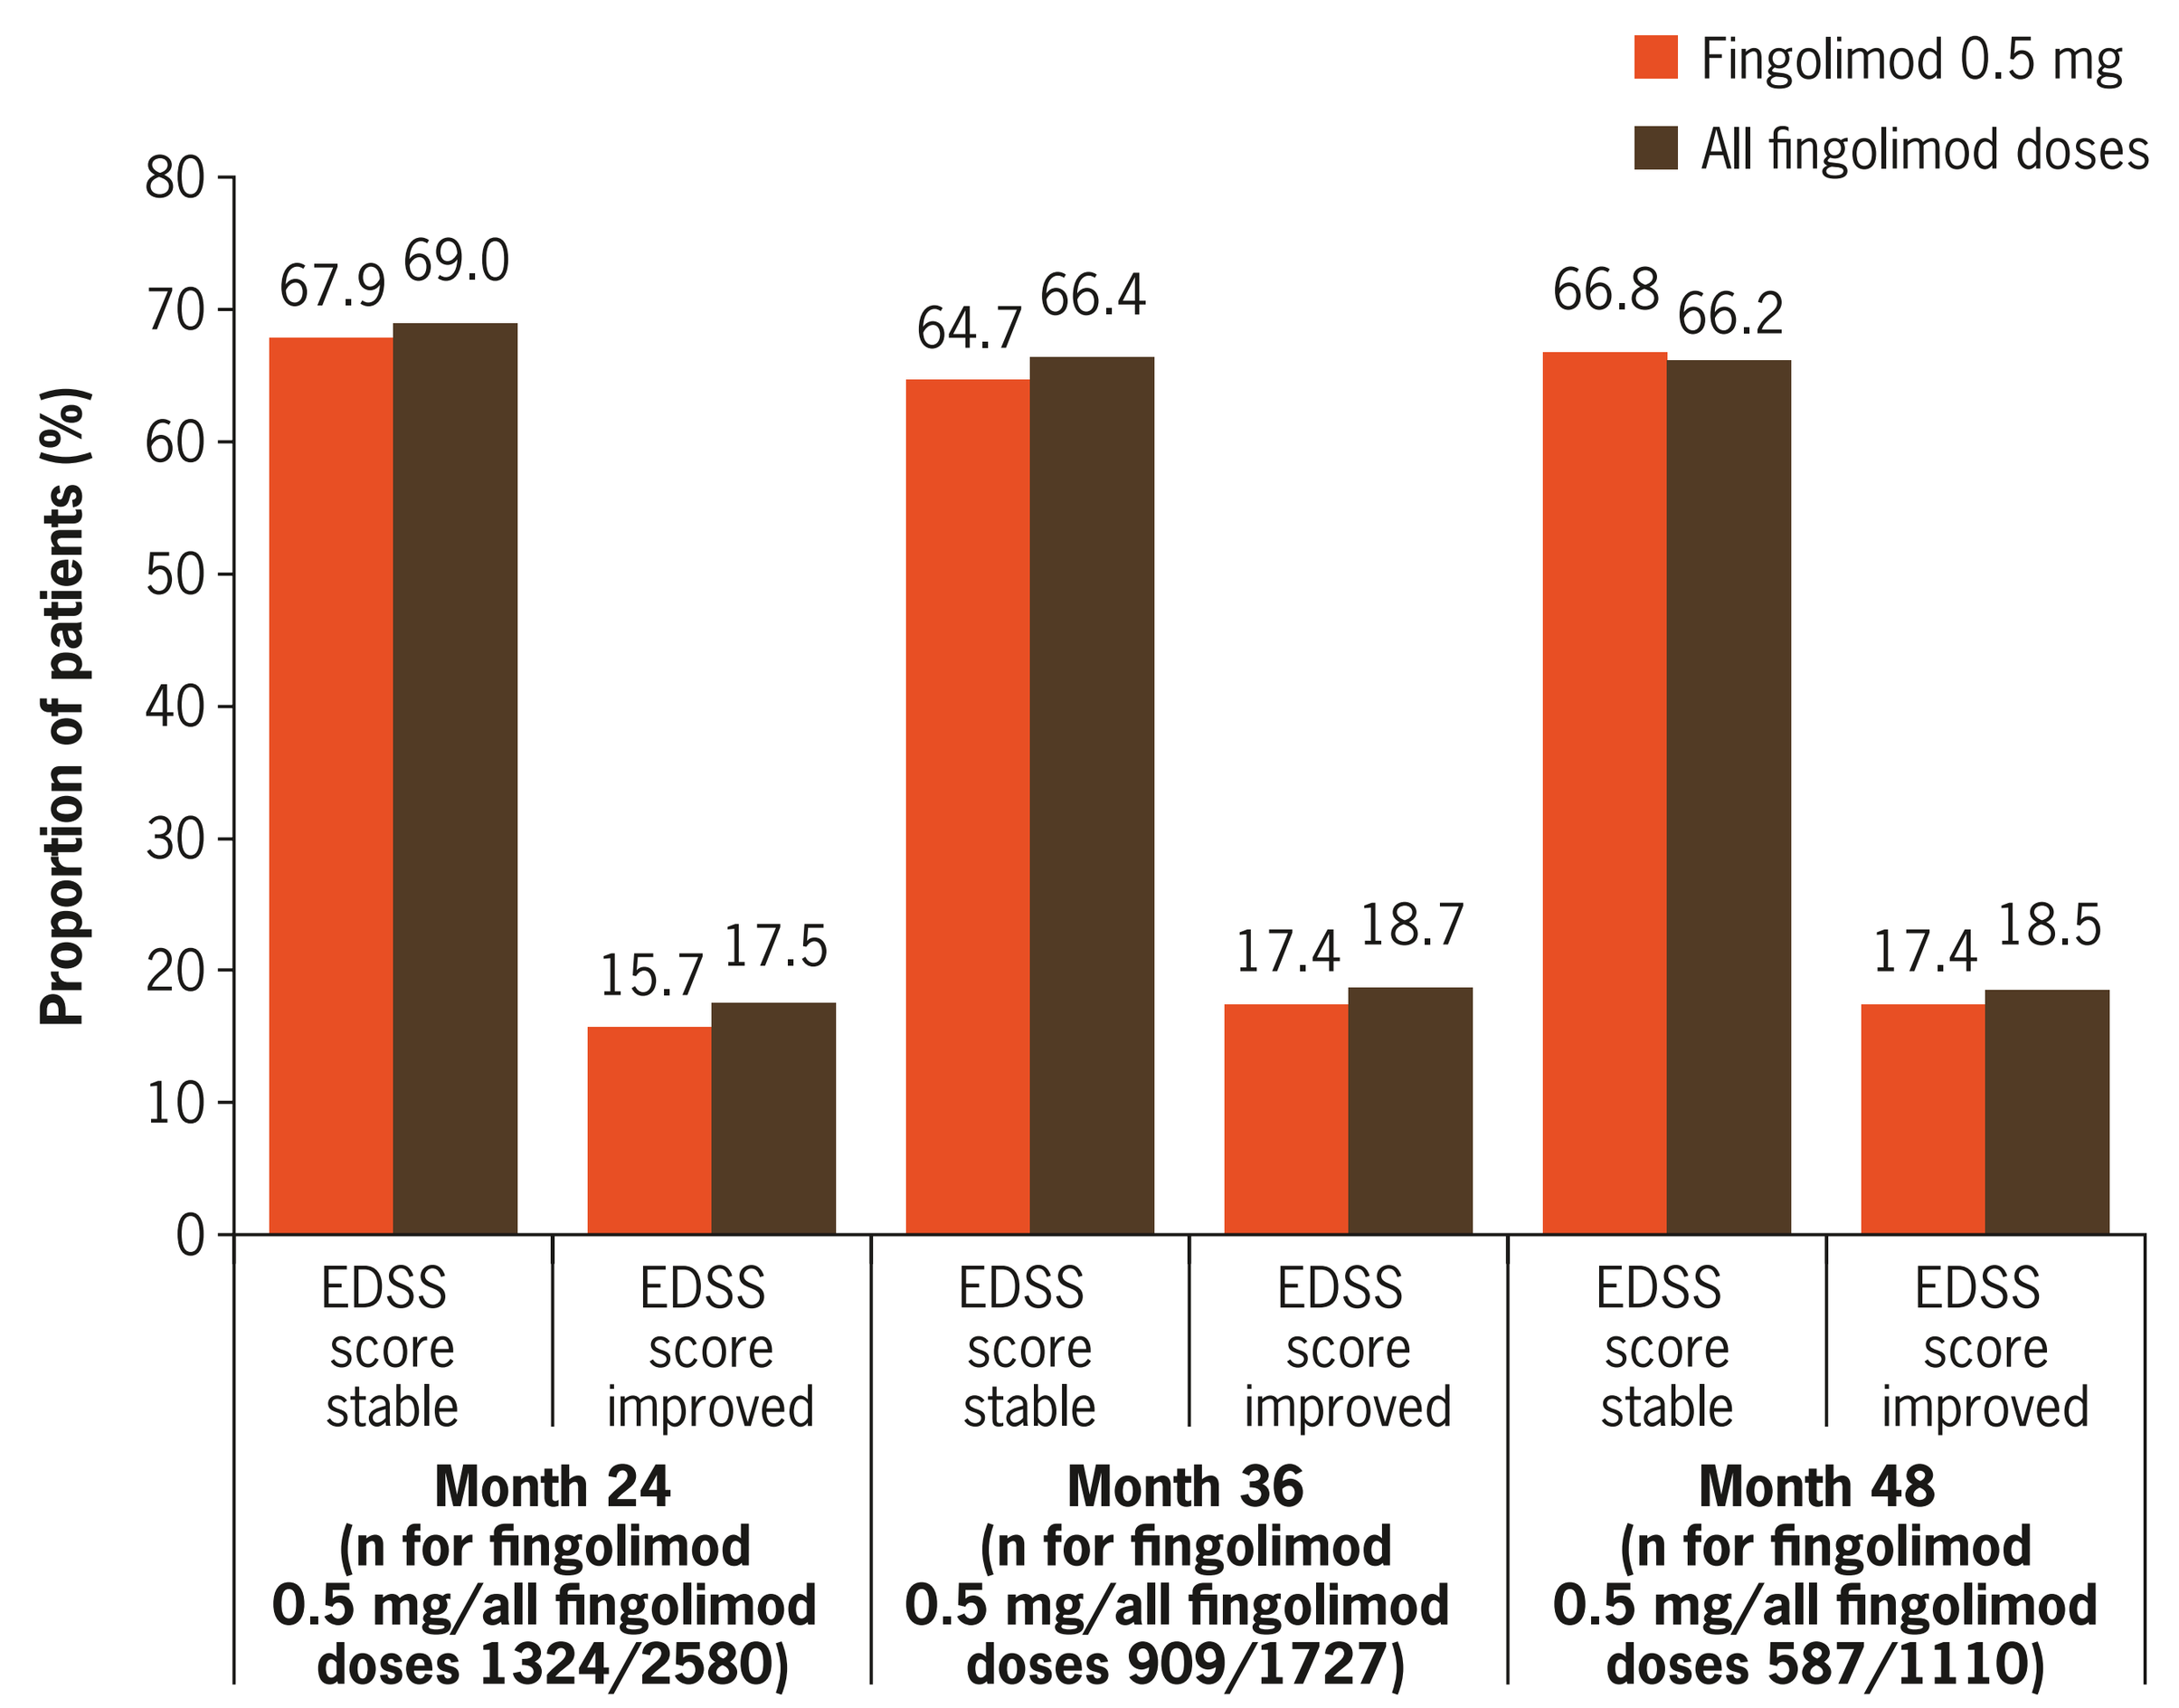
Fingolimod 0.5 mg
All fingolimod doses
Proportion of patients (%)
80
70
60
50
40
30
20
10
0
67.9
69.0
15.7
17.5
64.7
66.4
17.4
18.7
66.8
66.2
17.4
18.5
EDSS score stable
EDSS score improved
Month 24 (n for fingolimod 0.5 mg/all fingolimod doses 1324/2580)
Month 36 (n for fingolimod 0.5 mg/all fingolimod doses 909/1727)
Month 48 (n for fingolimod 0.5 mg/all fingolimod doses 587/1110)

n=number of patients with fingolimod baseline value and values at the specified visits
EDSS, Expanded Disability Status Scale

Figure 4. Logistic regression of stable and improved EDSS scores in fingolimod-treated patients from the start of fingolimod treatment to month 48

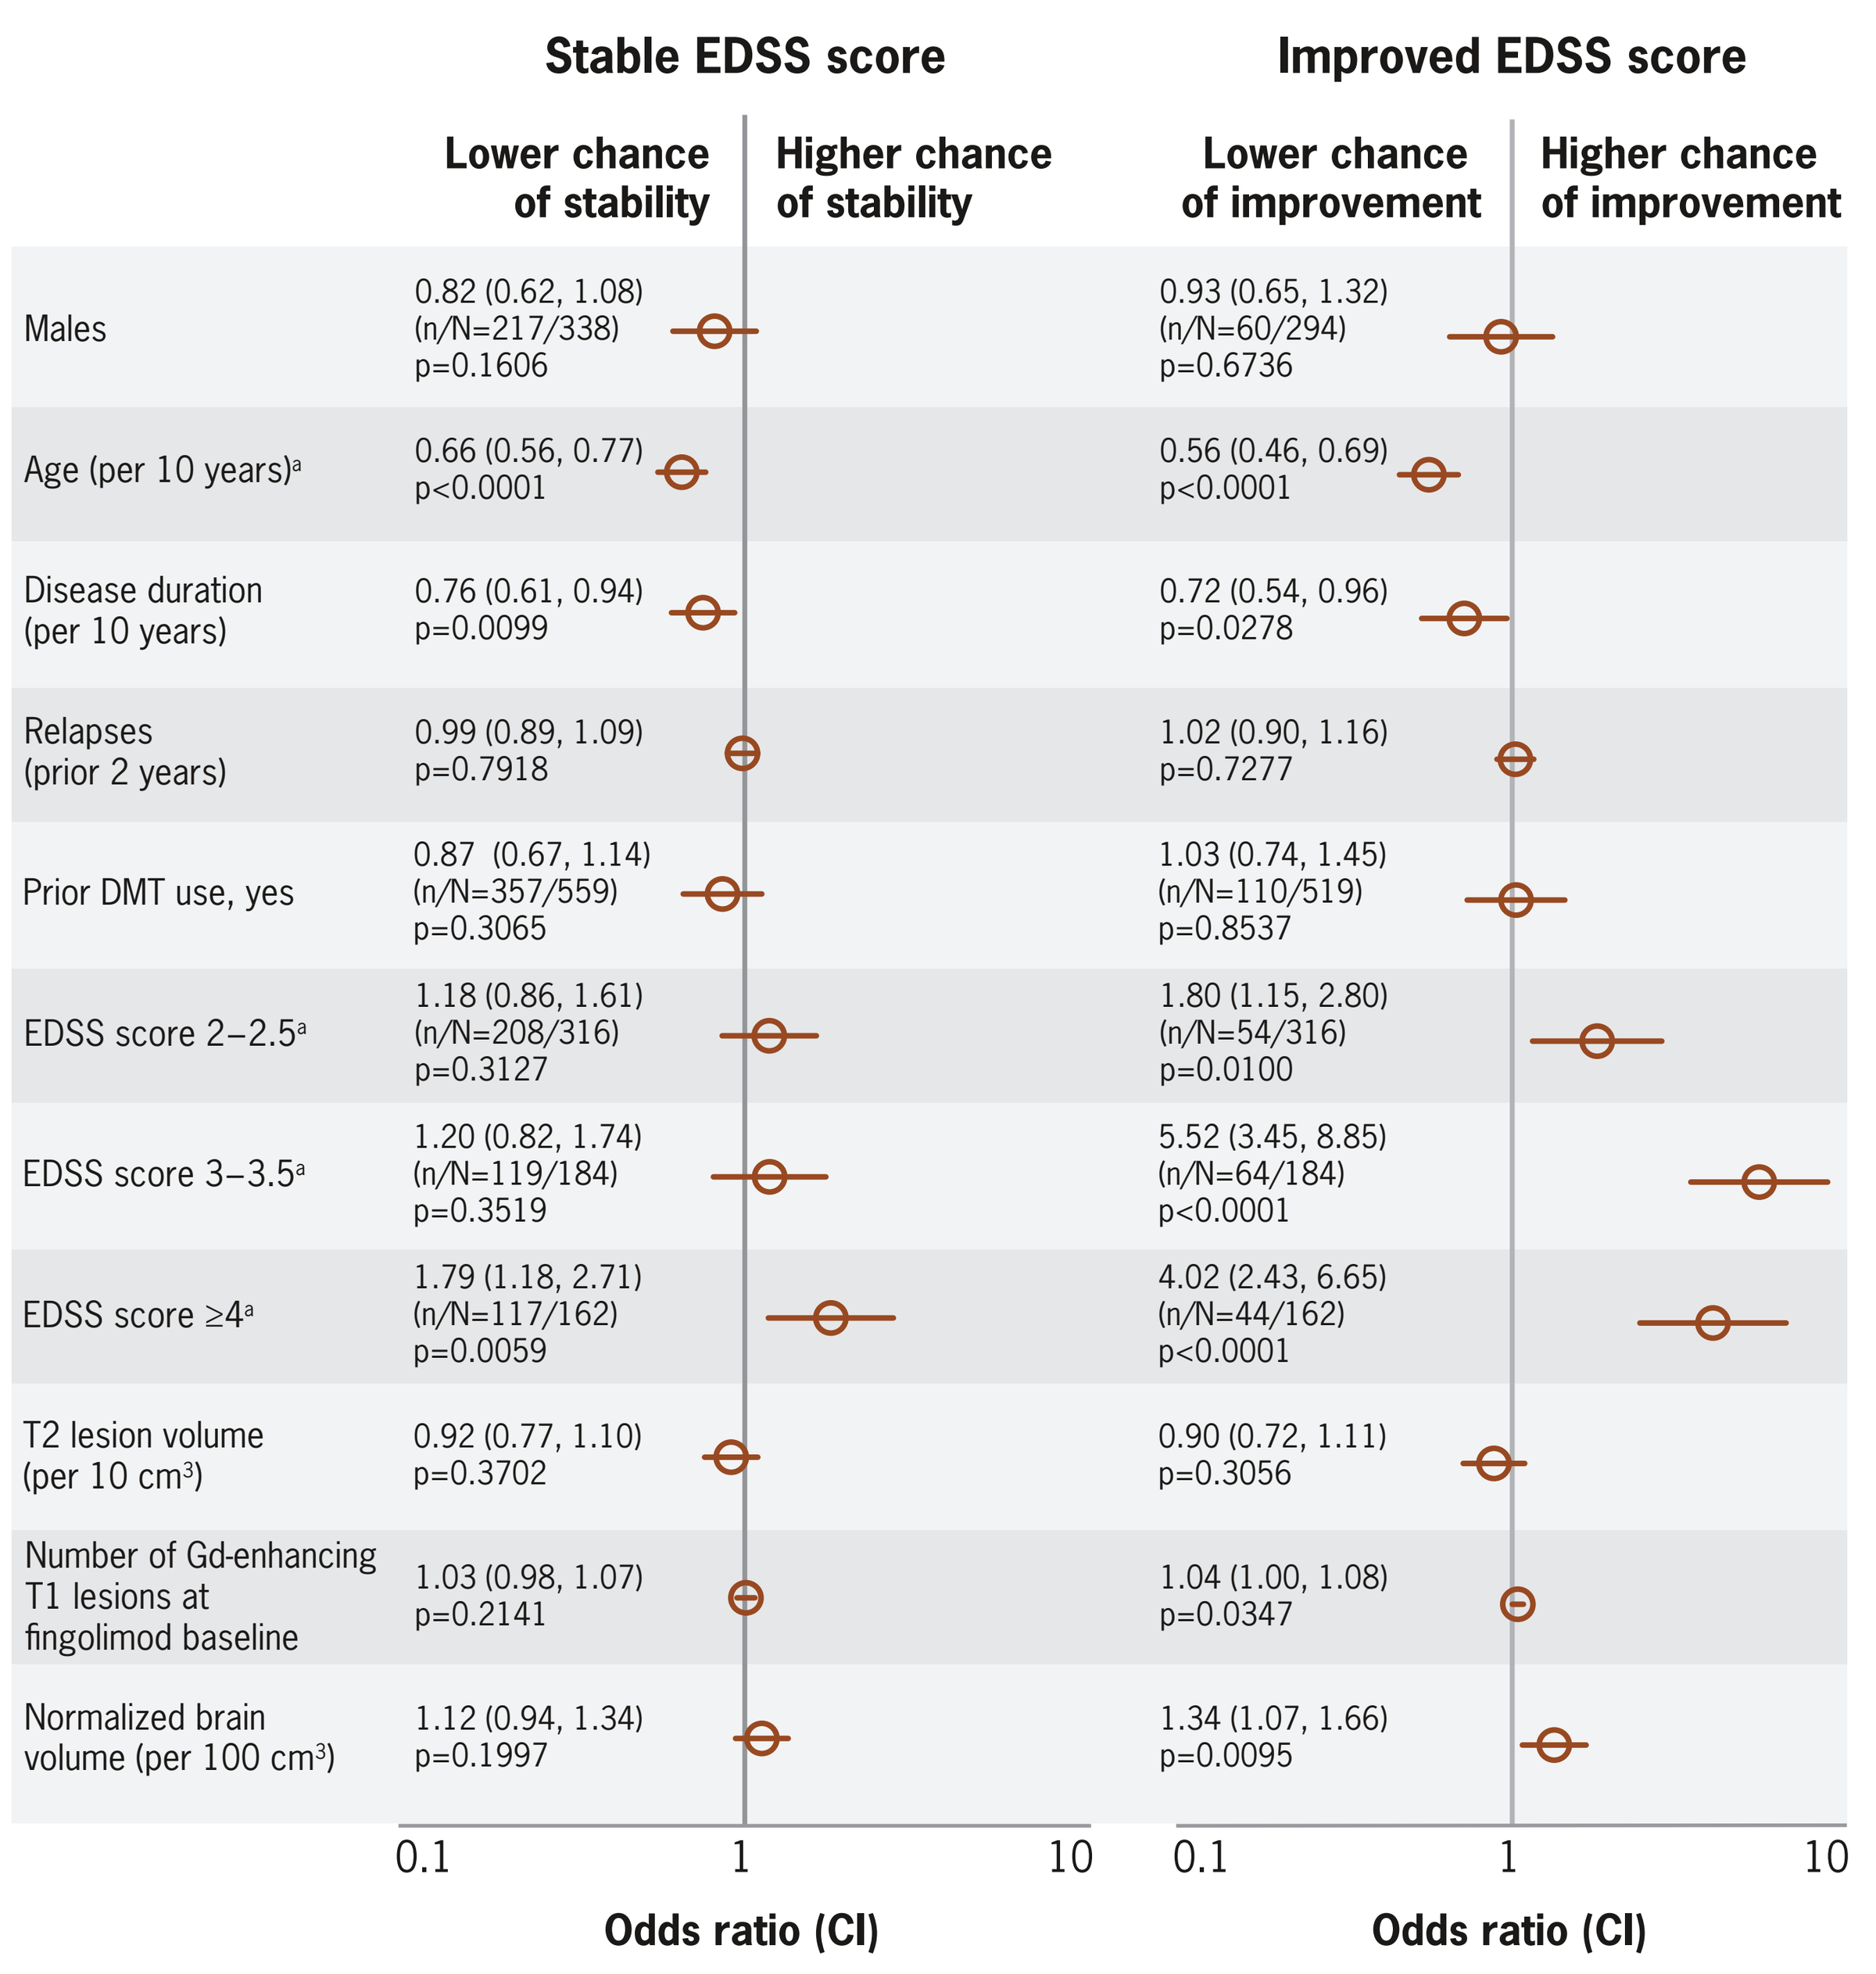
Stable EDSS score
Improved EDSS score
Lower chance of stability
Higher chance of stability
Lower chance of improvement
Higher chance of improvement
Males
0.82 (0.62, 1.08) (n/N=217/338) p=0.1606
0.93 (0.65, 1.32) (n/N=60/294) p=0.6736
Age (per 10 years)a
0.66 (0.56, 0.77) p<0.0001
0.56 (0.46, 0.69) p<0.0001
Disease duration (per 10 years)
0.76 (0.61, 0.94) p=0.0099
0.72 (0.54, 0.96) p=0.0278
Relapses (prior 2 years)
0.99 (0.89, 1.09) p=0.7918
1.02 (0.90, 1.16) p=0.7277
Prior DMT use, yes
0.87 (0.67, 1.14) (n/N=357/559) p=0.3065
1.03 (0.74, 1.45) (n/N=110/519) p=0.8537
EDSS score 2–2.5a
1.18 (0.86, 1.61) (n/N=208/316) p=0.3127
1.80 (1.15, 2.80) (n/N=54/316) p=0.0100
EDSS score 3–3.5a
1.20 (0.82, 1.74) (n/N=119/184) p=0.3519
5.52 (3.45, 8.85) (n/N=64/184) p<0.0001
EDSS score ≥4a
1.79 (1.18, 2.71) (n/N=117/162) p=0.0059
4.02 (2.43, 6.65) (n/N=44/162) p<0.0001
T2 lesion volume (per 10 cm3)
0.92 (0.77, 1.10) p=0.3702
0.90 (0.72, 1.11) p=0.3056
Number of Gd-enhancing T1 lesions at fingolimod baseline
1.03 (0.98, 1.07) p=0.2141
1.04 (1.00, 1.08) p=0.0347
Normalized brain volume (per 100 cm3)
1.12 (0.94, 1.34) p=0.1997
1.34 (1.07, 1.66) p=0.0095
0.1
1
10
Odds ratio (CI)

[a]Age and EDSS scores (categorized as 2–2.5, 3–3.5 and ≥4) are fixed independent variables in the models
Other variables were evaluated each in a separate logistic regression model that also included age and EDSS scores as fixed variables
n=number of patients with EDSS score improvement in the category
N=total number of patients in the category with assessments at the start of fingolimod treatment and a value at month 48
CI, confidence interval; DMT, disease-modifying therapy; EDSS, Expanded Disability Status Scale; Gd, gadolinium

References

1. Kappos L et al. Poster presented at ECTRIMS 2012, 10–13 October, 2012, Lyon, France, poster no. 979.
2. Montalban X et al. Poster presented at ECTRIMS 2012, 10–13 October, 2012, Lyon, France, poster no. 517.

Disclosures

Bruce Cree has served as a consultant for Biogen Idec, BioMS Medical, Elan Corporation, EMD Serono, Genentech, Genzyme, Sanofi-Aventis, Teva Pharmaceuticals, US National Institutes of Health and US National Multiple Sclerosis Society. **Jeffrey Cohen** has received personal compensation for serving as a consultant or speaker from Biogen Idec, Eli Lilly, Novartis and Vaccinex; and has received research support from EMD Serono, Genzyme, Genentech, Innate Immunotherapeutics, Novartis Pharmaceuticals Corporation, Biogen Idec, Consortium of MS Centers, US Department of Defense, Genzyme, Receptos, Synthon, Teva Pharmaceuticals. **Peter Chin** was an employee and stock holder of Novartis Pharmaceuticals Corporation at the time the study was conducted. **Shannon Ritter** and **Daniela Piani Meier** are employees and stock holders of Novartis Pharmaceuticals Corporation. **Ludwig Kappos** has received research support from Actelion Pharmaceuticals, Advancell, Allozyne, BaroFold, Bayer HealthCare Pharmaceuticals, Bayer Schering Pharma, Bayhill, Biogen Idec, BioMarin, CSL Behring, Elan Corporation, Genmab, GeNeuro SA, GenMark, Genzyme, GlaxoSmithKline, Eli Lilly, Merck Serono, MediciNova, Novartis Pharmaceuticals, Novo Nordisk, Peptimmune, Sanofi-Aventis, Santhera, Roche, Teva Pharmaceuticals, the Gianni Rubatto Research Foundation, the Novartis Pharmaceuticals Research Foundation, the Roche Research Foundation, the Swiss MS Society, the Swiss National Research Foundation, UCB and Wyeth.

Acknowledgments

The authors acknowledge Oxford PharmaGenesis, Oxford, UK, for editorial support, which was funded by Novartis Pharmaceuticals Corporation. The final responsibility for the content lies with the authors.

Scan to download a reprint of this poster

This study was funded by Novartis Pharmaceuticals Corporation | Poster presented at the 2015 Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC) | May 27–30, 2015 | Indianapolis, Indiana, USA