Real-world fingolimod first-dose effects in patients with hypertension, cardiac conditions and/or receiving selective-serotonin reuptake inhibitors

DX25

Daniel Wynn[1], Chris LaGanke[2], Lesley Schofield[3], Xiangyi Meng[3], Nadia Tenenbaum[3]

[1]Consultants in Neurology Multiple Sclerosis Center, Northbrook, IL, United States; [2]North Central Neurology Associates, Cullman, AL, Unites States; [3]Novartis Pharmaceuticals Corporation, East Hanover, NJ, United States

CONCLUSIONS

- **Fingolimod first-dose effects on heart rate, AV conduction and QT interval in patients with pre-existing hypertension with or without pre-existing cardiac conditions, and in patients receiving SSRIs, were similar to those effects observed among patients in the overall PREFER*MS* population, highlighting no additional safety implications in these subgroups**

BACKGROUND

- Approximately 134 000 patients have been treated with fingolimod in both the clinical trial and post-marketing settings; total patient exposure now exceeds 289 000 patient-years[1]
- First-dose findings from phase 3 studies show that fingolimod is generally well tolerated; a transient decrease in heart rate and asymptomatic atrioventricular (AV) conduction delays in a small number of patients are well-characterized pharmacological effects of fingolimod treatment initiation.[2–4] Symptomatic bradycardia and AV block are uncommon and typically require no intervention[2–4]
- These phase 3 findings are supported by the real-world 12-month prospective, randomized, active-controlled, open-label study to evaluate patient retention of fingolimod versus approved first-line disease-modifying therapies in adults with relapsing–remitting multiple sclerosis (PREFER*MS*)[5]
- However, it is important to understand the first-dose effects of fingolimod in specific patient subgroups, such as those with pre-existing hypertension with or without pre-existing cardiac conditions, and those receiving selective serotonin reuptake inhibitors (SSRIs)

OBJECTIVE

- To examine the first-dose effects of fingolimod in a real-world group of patients with relapsing–remitting MS (RRMS) that included individuals with pre-existing hypertension with or without pre-existing cardiac conditions, and those who were receiving a concomitant SSRI at first-dose administration

METHODS

- PREFER*MS* enrolled male and female patients (aged 18–65 years) with RRMS (Expanded Disability Status Scale score ≤6)
 - Patients were either treatment-naïve or had previously been treated with no more than one class of disease-modifying therapy (DMT), such as interferon beta, glatiramer acetate or dimethyl fumarate, for less than 2 months
 - Patients were randomized to receive once-daily fingolimod 0.5 mg or an injectable DMT for 12 months
 - Patients treated with fingolimod who underwent first-dose observation after initial randomization or switched to fingolimod during the study were included in the analysis
 - Patients with a resting heart rate lower than 45 beats per minute (bpm) were excluded from the trial
- Patient subgroups were defined *post hoc* according to whether the patients had:
 - pre-existing hypertension
 - pre-existing hypertension plus pre-existing cardiac conditions of special interest (first-degree AV block, bradycardia/sinus bradycardia, unstable angina, coronary artery disease and hypertensive heart disease)
 - received an SSRI at the time of fingolimod first-dose administration
- Electrocardiogram (ECG) monitoring was performed at baseline and 6 hours after the first dose of fingolimod. Vital signs (sitting heart rate and blood pressure) were recorded hourly
- Individuals were discharged after a 6-hour observation period unless they had a heart rate lower than 45 bpm, corrected QT (QTc) interval of at least 500 ms, new-onset second-degree or higher AV block, their lowest post-dose heart rate at the end of the observation period or the investigator determined that the patient had symptomatic bradycardia that required treatment. Patients meeting these criteria were required to undergo extended observation
- Analyses were based on change from baseline in sitting heart rate and incidence of newly occurring ECG abnormalities during first-dose observation
- The differences between the subgroups and the overall PREFER*MS* population are descriptive only and no statistical comparisons are presented

RESULTS

- In total, 687 patients were randomized to receive once-daily fingolimod 0.5 mg and are included in the analysis
 - 119 patients had pre-existing hypertension
 - 127 individuals had pre-existing hypertension with or without pre-existing cardiac conditions
 - In this subgroup, patients with pre-existing hypertension and cardiac conditions were combined with individuals with pre-existing hypertension without cardiac conditions because there were only eight patients in the former category. Therefore, patients (and associated events) could belong to more than one subgroup
 - 138 patients received a SSRI on the day of fingolimod first-dose administration
- The baseline demographics were generally similar; however, patients with pre-existing hypertension with or without cardiac conditions were older, weighed more and had a higher body mass index than the overall PREFER*MS* population (**Table 1**)

Table 1. Baseline demographics

	Overall PREFER*MS* population (n=687)	Pre-existing hypertension subgroup (n=119)	Pre-existing hypertension with or without cardiac conditions subgroup (n=127)	SSRI subgroup (n=138)
Age, years, mean (SD)	41.5 (10.7)	46.9 (10.2)	46.8 (10.5)	42.8 (10.4)
Age interval, years, n (%)				
18–30	127 (18.5)	11 (9.2)	14 (11.0)	22 (15.9)
31–40	201 (29.3)	20 (16.8)	20 (15.7)	40 (29.0)
41–55	281 (40.9)	60 (50.4)	62 (48.8)	55 (39.9)
>55	78 (11.4)	28 (23.5)	31 (24.4)	21 (15.2)
Sex, n (%)				
Male	184 (26.8)	28 (23.5)	30 (23.6)	29 (21.0)
Female	503 (73.2)	91 (76.5)	97 (76.4)	109 (79.0)
Weight, kg, mean (SD)	82.3 (20.4)	93.5 (24.1)	92.7 (23.7)	80.8 (19.9)
Body mass index, kg/m², mean (SD)	29.2 (7.0)	33.5 (8.4)	33.2 (8.3)	28.8 (7.0)
Race, n (%)				
Caucasian	563 (82.0)	92 (77.3)	97 (76.4)	126 (91.3)
Black	108 (15.7)	26 (21.8)	29 (22.8)	9 (6.5)
Asian	1 (0.1)	0 (0.0)	0 (0.0)	0 (0.0)
Other	15 (2.2)	1 (0.8)	1 (0.8)	3 (2.2)
Duration of MS since first symptom, years, mean (SD)	7.3 (7.8)	8.5 (9.3)	8.7 (9.5)	8.7 (9.0)
Number of relapses in the past year, mean (SD)	0.6 (1.0)	0.6 (0.9)	0.6 (0.9)	0.6 (0.9)
Number of relapses in the past 2 years, mean (SD)	0.9 (1.4)	0.9 (1.6)	0.9 (1.6)	0.9 (1.5)

Clinical experience

- In the overall PREFER*MS* population and the pre-existing hypertension, pre-existing hypertension with or without pre-existing cardiac conditions and SSRI subgroups, 88.1%, 85.7%, 85.8% and 89.9% of patients, respectively, were discharged at 6 hours post-dose (**Table 2**)
 - Extended monitoring was required in 11.5%, 14.3%, 14.2% and 9.4% of patients, respectively
- In the overall PREFER*MS* population, two patients (0.3%) required hospitalization, one patient (0.1%) had symptomatic bradycardia that did not require treatment and two patients (0.3%) reported a serious adverse event (SAE)

Table 2. Clinical experience during initiation of fingolimod treatment

Clinical experience, n (%)	Overall PREFER*MS* population (n=687)	Pre-existing hypertension subgroup (n=119)	Pre-existing hypertension with or without cardiac conditions subgroup (n=127)	SSRI subgroup (n=138)
Discharged at 6 hours	605 (88.1)	102 (85.7)	109 (85.8)	124 (89.9)
Required extended monitoring after 6 hours as per protocol	79 (11.5)	17 (14.3)	18 (14.2)	13 (9.4)
Hospitalized	2 (0.3)	0 (0.0)	0 (0.0)	0 (0.0)
Symptomatic bradycardia	1 (0.1)	0 (0.0)	0 (0.0)	0 (0.0)
SAE reported	2 (0.3)	0 (0.0)	0 (0.0)	0 (0.0)
First-degree AV block post-dose (6 hours)	41 (6.1)	8 (6.9)	12 (9.7)	7 (5.1)
Second-degree Mobitz I AV block post-dose (6 hours)	4 (0.6)	0 (0.0)	1 (0.8)	1 (0.7)
2:1 AV block post-dose (6 hours)	1 (0.1)	0 (0.0)	0 (0.0)	0 (0.0)

- Of the two patients who experienced SAEs:
 - one patient had second-degree Mobitz I AV block secondary to bradycardia. This patient was admitted to hospital for further observation as per study protocol and remained asymptomatic; study medication was discontinued
 - one patient had second-degree 2:1 AV block secondary to bradycardia and was admitted to hospital for further observation as per study protocol. The patient remained on fingolimod treatment (**Table 2**)
- No patient required hospitalization, had symptomatic bradycardia or reported SAEs in any of the pre-existing hypertension, pre-existing hypertension with or without pre-existing cardiac conditions or SSRI subgroups

Heart rate effects at 6 hours post-dose

- The lowest mean sitting heart rate was reached at 5 hours in the overall PREFER*MS* population and the pre-existing hypertension and pre-existing hypertension with or without cardiac conditions subgroups (–7.3, –6.4 and –6.5 bpm, respectively), and at 4 hours in the SSRI subgroup (–6.2 bpm). The heart rate began to recover by 6 hours (**Figure 1**)

Figure 1. Mean sitting heart rate in the 6 hours following the first dose of fingolimod

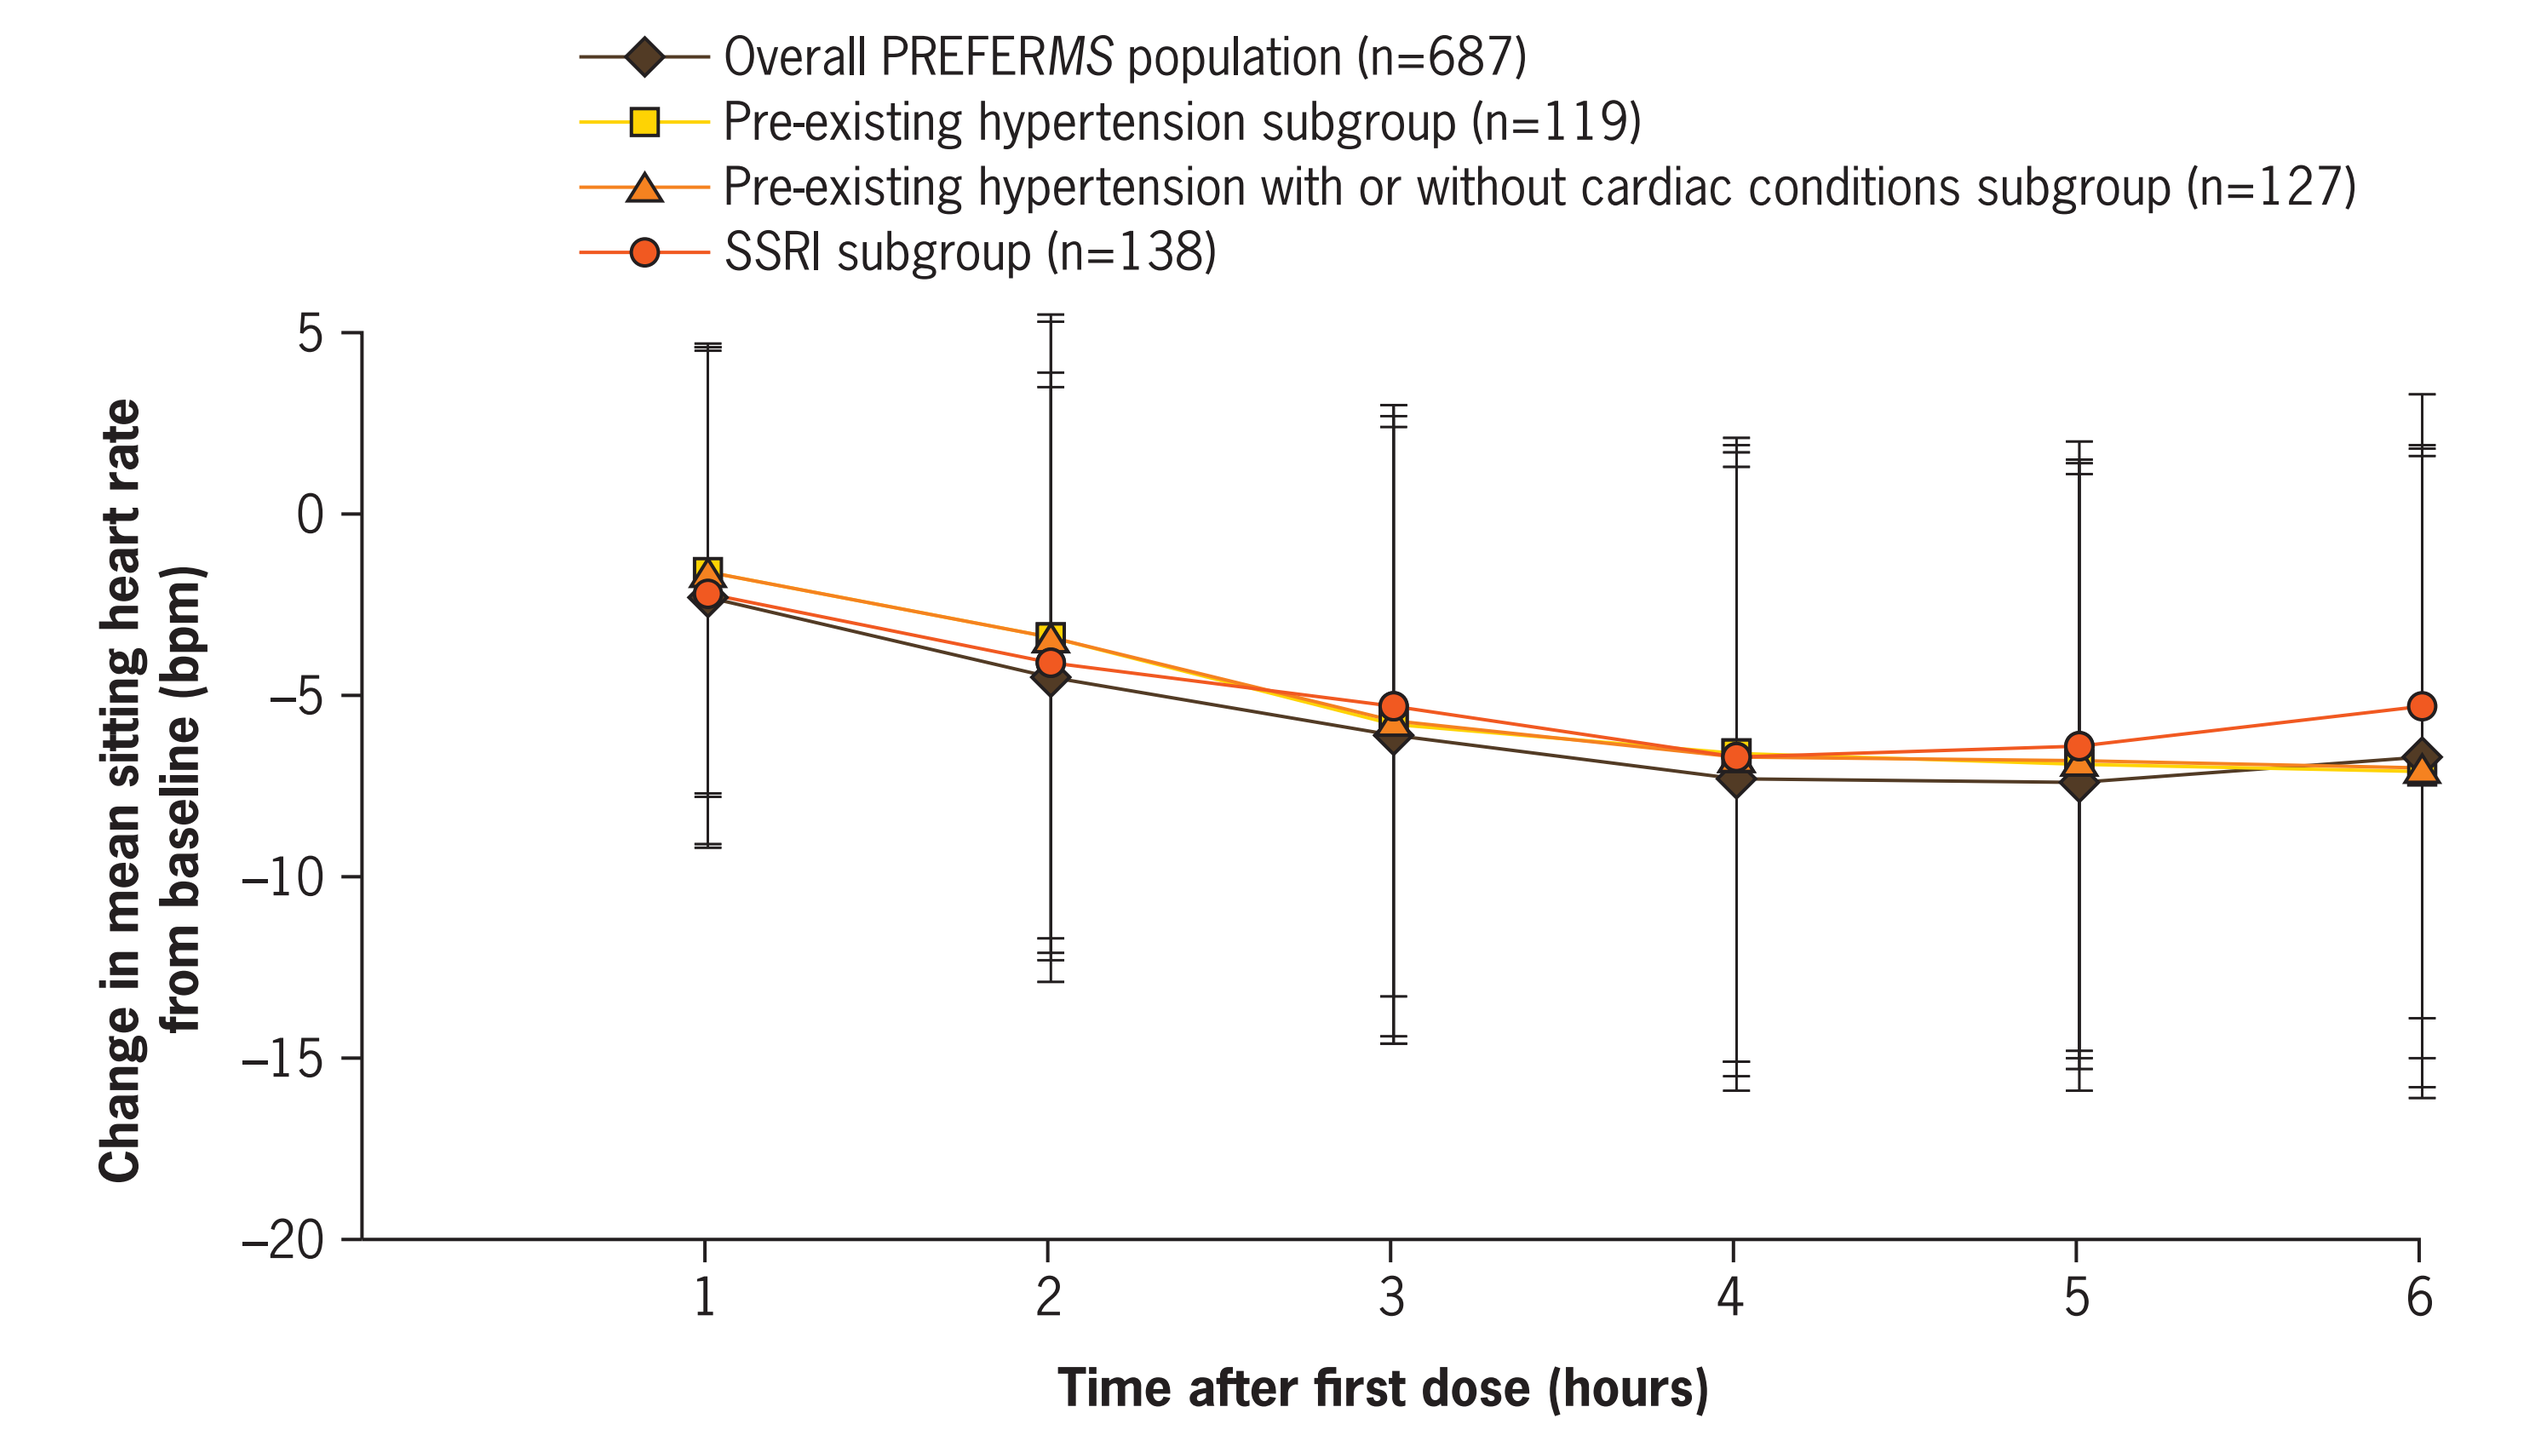
Overall PREFERMS population (n=687)
Pre-existing hypertension subgroup (n=119)
Pre-existing hypertension with or without cardiac conditions subgroup (n=127)
SSRI subgroup (n=138)
Change in mean sitting heart rate from baseline (bpm)
5
0
–5
–10
–15
–20
1
2
3
4
5
6
Time after first dose (hours)

Data points represent change from baseline in sitting heart rate (bpm), mean and standard deviation

- At 6 hours, mean heart rate change from baseline was –6.1, –5.9 and –4.5 bpm in the pre-existing hypertension, pre-existing hypertension with or without cardiac conditions and SSRI subgroups, respectively; these changes were similar to those in the overall PREFER*MS* population (–6.5 bpm)
- No patient in any subgroup had a heart rate of less than 45 bpm during first-dose fingolimod administration, and most patients maintained a heart rate of at least 55 bpm
 - Three patients in the overall PREFER*MS* population had a heart rate of less than 45 bpm at 2 hours post-dose

ECG findings at 6 hours post-dose

- One patient in each of the subgroups had a QTc interval longer than 450 ms (men) or 470 ms (women) according to the Fridericia correction method, and none had values greater than 500 ms (**Table 3**)

Table 3. QTc interval changes 6 hours after initiation of fingolimod treatment

	Overall PREFER*MS* population (n=687)	Pre-existing hypertension subgroup (n=119)	Pre-existing hypertension with or without cardiac conditions subgroup (n=127)	SSRI subgroup (n=138)
Patients with ECG data, n (%)	673	116	124	136
Fridericia correction Maximum increase from pre-dose, n (%)				
<30 ms	445 (66.1)	79 (68.1)	85 (68.5)	92 (67.6)
30–60 ms	35 (5.2)	5 (4.3)	5 (4.0)	4 (2.9)
>60 ms	1 (0.1)	1(0.9)	1 (0.8)	0 (0.0)
QTc value >450 ms (men) or >470 ms (women), n (%)	4 (0.6)	1 (0.9)	1 (0.8)	1 (0.7)
QTc values >500 ms (men) or >520 ms (women), n (%)	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)

- The maximum increase in QTc interval was less than 30 ms compared with the pre-dose value in 68.1%, 68.5% and 67.6% of patients in the pre-existing hypertension, pre-existing hypertension with or without cardiac conditions and SSRI subgroups, respectively, compared with 66.1% of patients in the overall PREFER*MS* population
- In the overall PREFER*MS* population, four patients (0.6%) had second-degree Mobitz I AV block at 6 hours post-dose. No 2:1 AV blocks were reported in any of the subgroups (**Table 2**)
- A total of 6.9%, 9.7% and 5.1% of patients in the pre-existing hypertension, pre-existing hypertension with or without cardiac conditions and SSRI subgroups, respectively, had first-degree AV block at 6 hours post-dose, compared with 6.1% in the overall PREFER*MS* population (**Table 2**)

References

1. Novartis. Novartis patient exposure. Data as of January, 2016; Q1 Novartis Pharmaceuticals Annual Report, January, 2016.
2. Kappos L, et al. N Engl J Med. 2010;362:387–401.
3. Cohen JA, et al. N Engl J Med. 2010;362:402–415.
4. Calabresi PA, et al. Lancet Neurol. 2014;13:545–556.
5. Tenenbaum N, et al. Poster DX74. Presented at the Annual Meeting of the Consortium of Multiple Sclerosis Centers, 7–30 May 2015, Indianapolis, IN, United States.

Disclosures

Daniel Wynn has received research support and/or consulting fees from Acorda, Actelion, Allergan, Avanir, Biogen, EMD Serono, GlaxoSmithKline, F. Hoffmann-La Roche, Novartis, Ono Pharmaceutical Co., Opexa Therapeutics, Osmotica, Pfizer, Questcor, SanBio, Sanofi, Sunovion, Teva and XenoPort. **Chris LaGanke** has received research support and has been a consultant and/or received honoraria from Acorda, Avanir, Bayer, Biogen Idec, EMD Serono, Genzyme, GlaxoSmithKline, Merz, Novartis, Opexa, Pfizer, Questcor, Sanofi, Teva Neuroscience and Vaccinex. **Xiangyi Meng**, **Lesley Schofield** and **Nadia Tenenbaum** are employees of Novartis Pharmaceuticals Corporation.

Acknowledgments

Editorial support was provided by Oxford PharmaGenesis, Oxford, UK, which was funded by Novartis Pharmaceuticals Corporation. The final responsibility for the content lies with the authors.

Your document will be available for download at the following URL:
http://novartis.medicalcongressposters.com/Default.aspx?doc=e56ca

And via Text Message (SMS)
Text: **Qe56ca**
To: **8NOVA (86682)** *US Only*
+18324604729 *North, Central and South Americas; Caribbean; China*

Note: downloading data may incur costs which can vary depending on your service provider and may be high if you are using your smartphone abroad. Please check your phone tariff or contact your service provider for more details.

Scan to download this poster

This study was funded by Novartis Pharmaceuticals Corporation, USA | Poster presented at the 2016 Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC) | June 1–4, 2016 | National Harbor, Maryland, United States