DX58

Long-term effect of fingolimod on disability: a categorical trend analysis over 8 years

Shannon Ritter[1], Anthony T Reder[2], Daniela Piani Meier[3], Davorka Tomic[3], Bruce A.C. Cree[4]

[1]Novartis Pharmaceuticals Corporation, East Hanover, NJ, United States; [2]Department of Neurology, University of Chicago, Chicago, IL, United States; [3]Novartis Pharma AG, Basel, Switzerland; [4]University of California San Francisco, San Francisco, CA, United States

CONCLUSIONS

- **Over 2 years, disability changed minimally or fluctuated in most patients with RRMS enrolled in the FREEDOMS trials**
- **Hence, longer follow-up periods are required to detect meaningful changes in disability evolution**
- **After 8 years, disability was stable or had improved in the majority of patients who received fingolimod continuously**

BACKGROUND

- Disability progression is a major clinical outcome in patients with multiple sclerosis (MS), and even moderate levels of disability can be highly disruptive to normal living.[1] It is important to identify disease-modifying therapies that can slow or halt accrual of disability in the long term
- Disability progression is most commonly measured in clinical trials by analyzing changes in the Expanded Disability Status Scale (EDSS) score, confirmed after 3 or 6 months, typically over periods of 2–3 years[2–4]
- In the 2-year, phase 3 FREEDOMS and FREEDOMS II trials, fingolimod reduced confirmed disability progression compared with placebo in patients with relapsing–remitting MS (RRMS)[5,6]
- However, MS is a lifelong disease, so evaluating disability evolution over longer periods is important to determine the real impact of treatment and to minimize bias from short-term fluctuations in EDSS score that are not sustained in the longer term
- Changes in disability that are sustained in the longer term are likely to be a more meaningful measure of outcome than those observed in the short term. Categorizing trends in EDSS score changes may be a good means of assessing long-term disability progression,[7] because these may capture the true impact of treatment
- In this analysis of the pooled FREEDOMS RRMS population, we examined trends in changes in EDSS scores over a maximum of 8 years to investigate the impact of early continuous treatment with fingolimod on long-term disability progression

OBJECTIVES

- To investigate over 96 months how patterns of disability evolved and the impact of fingolimod treatment on long-term disability, in the pooled FREEDOMS RRMS population, based on categorical analysis of change in EDSS score

METHODS

Analysis population

- *Post hoc* analyses were conducted using data collected for up to 96 months from baseline from patients randomized to fingolimod 0.5 mg (n=783) or to placebo (n=773) in the two 2-year FREEDOMS trials,[5,6] those who continued or switched to fingolimod 0.5 mg in the trial extensions,[8] and those who continued to receive fingolimod 0.5 mg in the observational LONGTERMS trial[9]
- Analyses were conducted in the full analysis set (FAS; individuals with values at baseline and at month 24, 48 or 96) and in the completer subgroup (CS; individuals with complete values at baseline and at months 24, 48 and 96)

Analyses

- Building on previously published work,[7] trends in disability progression observed at intervals up to 96 months were categorized as:
 - minimal (an increase or decrease of 0.5 points from baseline EDSS score if the baseline score was ≤5.5, or no change in score if the baseline score was >5.5)
 - improving (a decrease of ≥1.0 point from baseline EDSS score, either confirmed at 6 months only or confirmed at 6 months and sustained until month 24, 48 or 96)
 - worsening (an increase of ≥1.0 point from baseline EDSS score, either confirmed at 6 months only or confirmed at 6 months and sustained until month 24, 48 or 96)
 - stable/improving (minimal and improving categories combined)
 - fluctuating (changes in EDSS score that differed from those defined in other categories)
- Cross-sectional and longitudinal comparisons were made using the Cochran–Mantel–Haenszel test

Figure 1. Categorical disability at months 24, 48 and 96 (FAS)

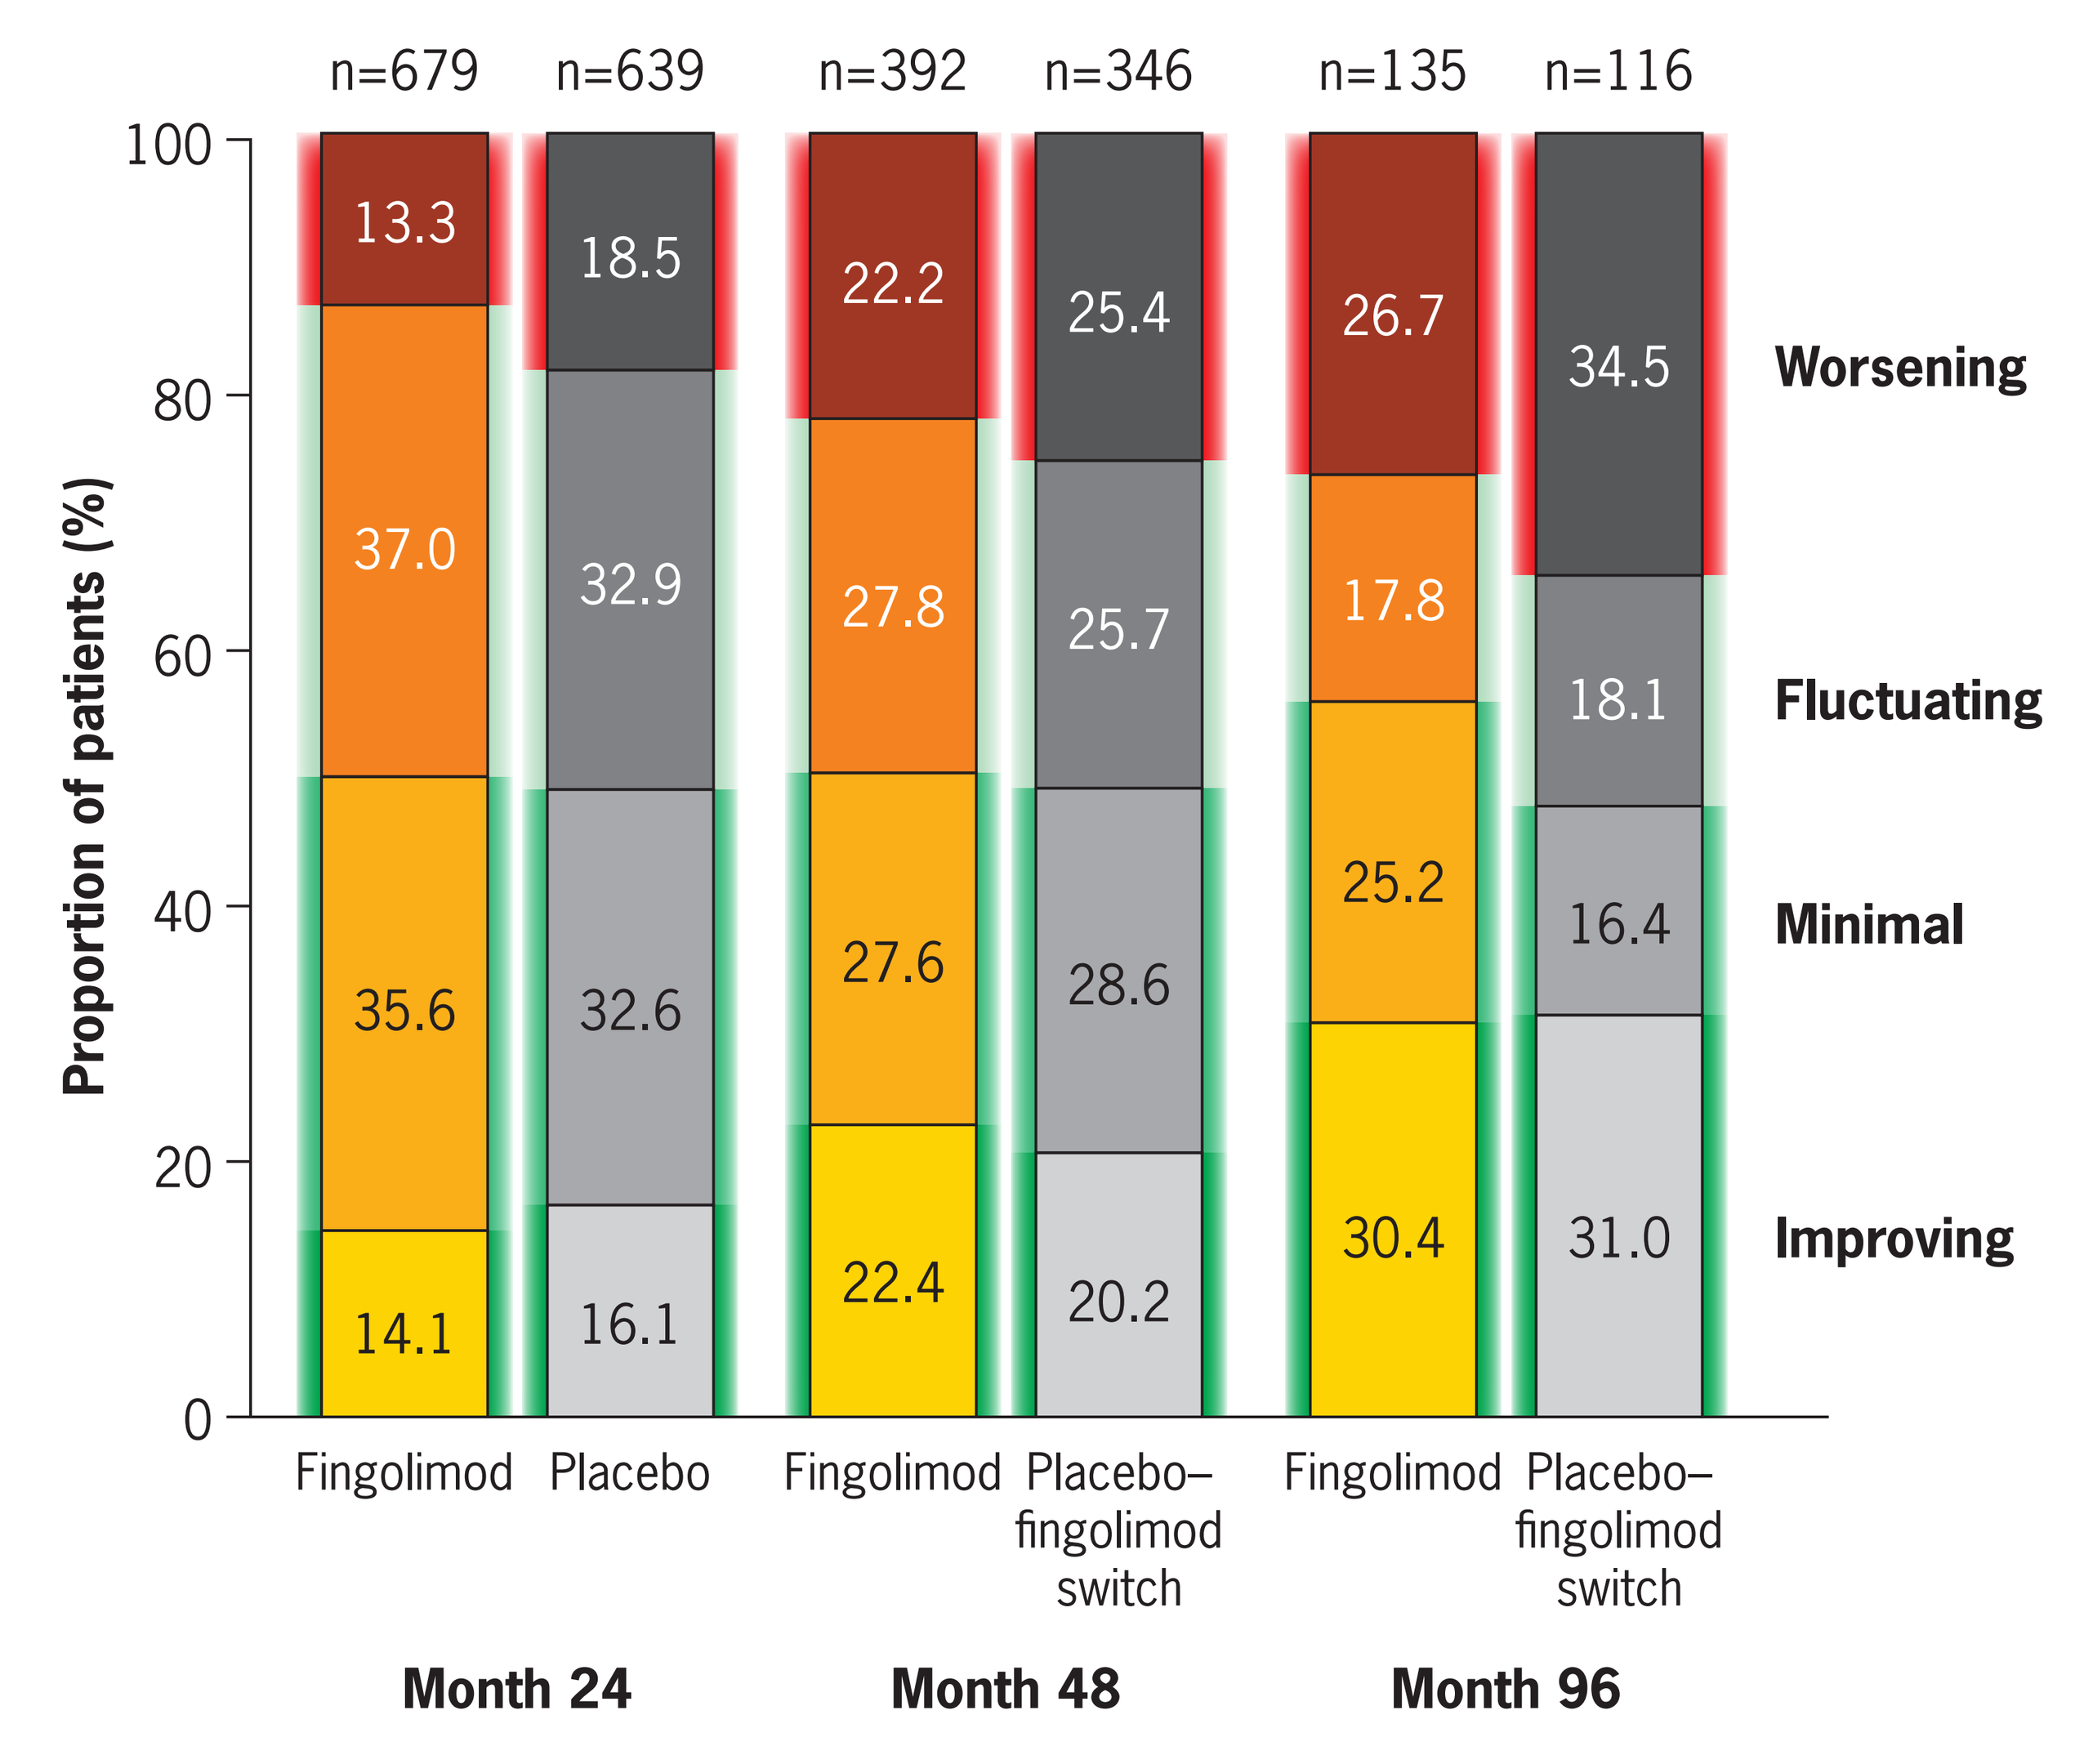
n=679
n=639
n=392
n=346
n=135
n=116
Proportion of patients (%)
0
20
40
60
80
100
13.3
37.0
35.6
14.1
18.5
32.9
32.6
16.1
22.2
27.8
27.6
22.4
25.4
25.7
28.6
20.2
26.7
17.8
25.2
30.4
34.5
18.1
16.4
31.0
Worsening
Fluctuating
Minimal
Improving
Fingolimod
Placebo
Fingolimod
Placebo–fingolimod switch
Fingolimod
Placebo–fingolimod switch
Month 24
Month 48
Month 96

Fingolimod dose was 0.5 mg

Figure 2. Categorical disability at months 24, 48 and 96 (CS)

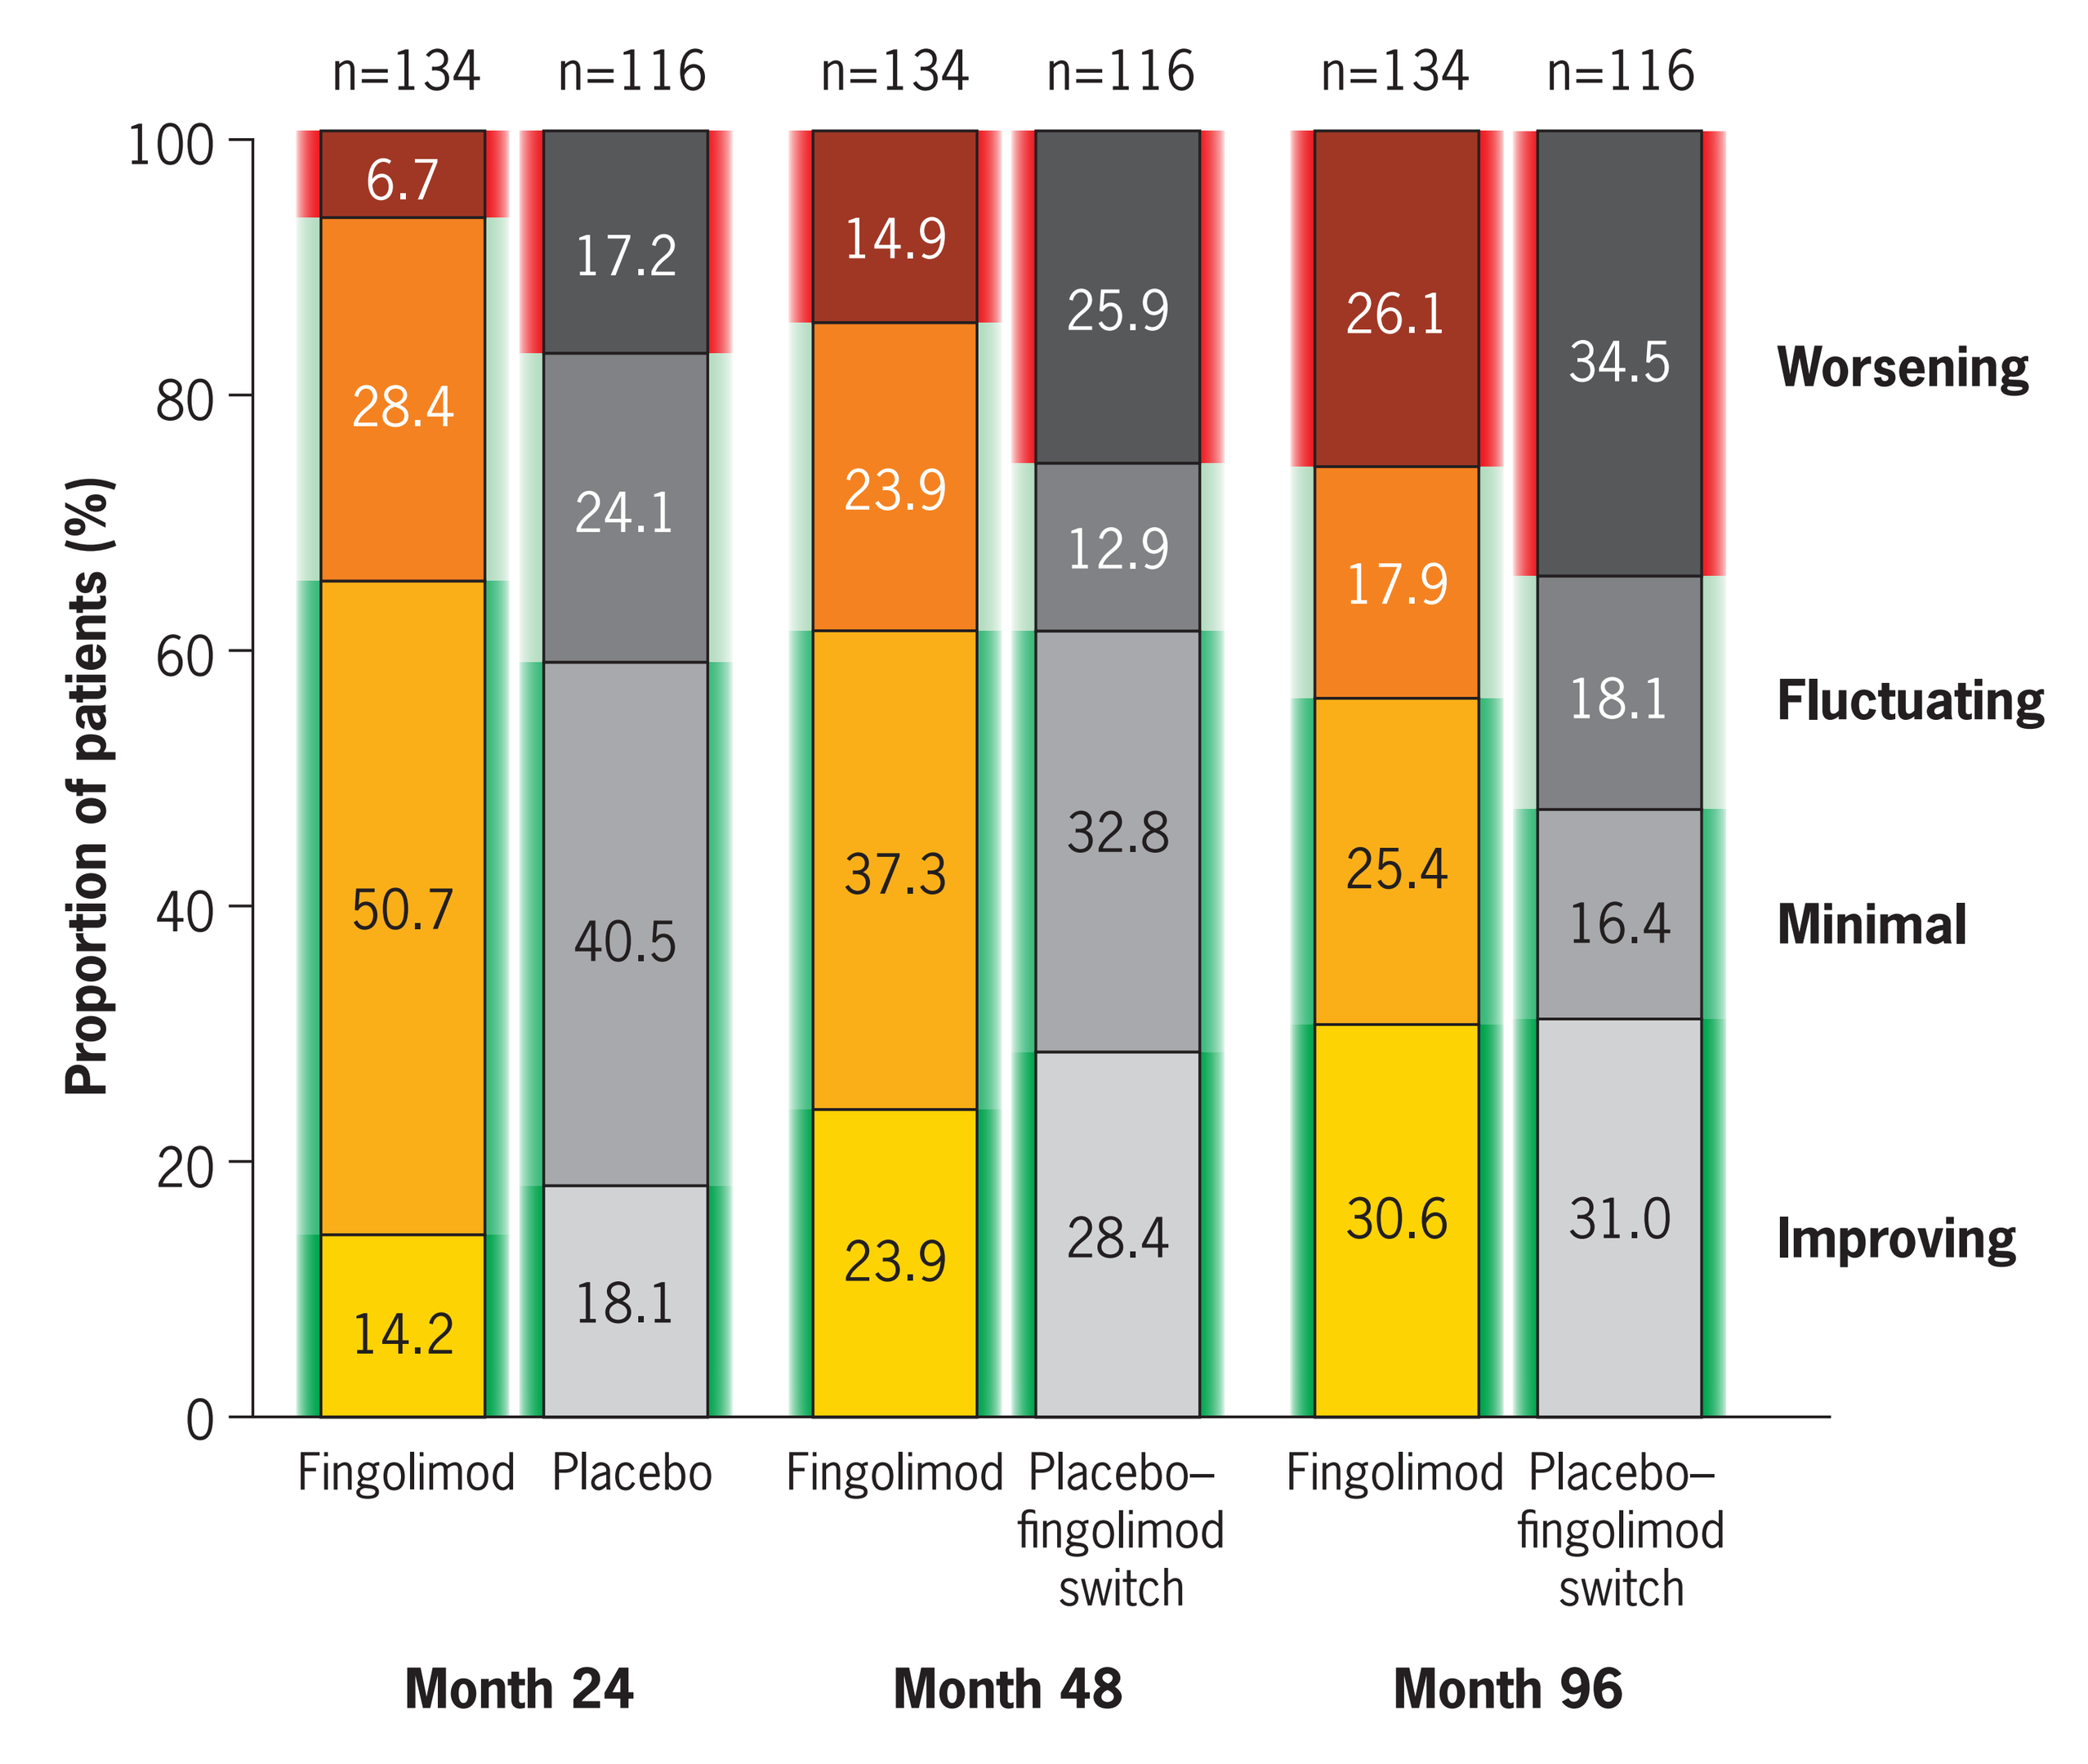
n=134
n=116
n=134
n=116
n=134
n=116
Proportion of patients (%)
0
20
40
60
80
100
6.7
28.4
50.7
14.2
17.2
24.1
40.5
18.1
14.9
23.9
37.3
23.9
25.9
12.9
32.8
28.4
26.1
17.9
25.4
30.6
34.5
18.1
16.4
31.0
Worsening
Fluctuating
Minimal
Improving
Fingolimod
Placebo
Fingolimod
Placebo–fingolimod switch
Fingolimod
Placebo–fingolimod switch
Month 24
Month 48
Month 96

Fingolimod dose was 0.5 mg

Figure 3. Short- and long-term effects of continuous fingolimod treatment: patients with stable/improving disability versus those with fluctuating or worsening disability at months 24, 48 and 96 in the a) FAS and b) CS

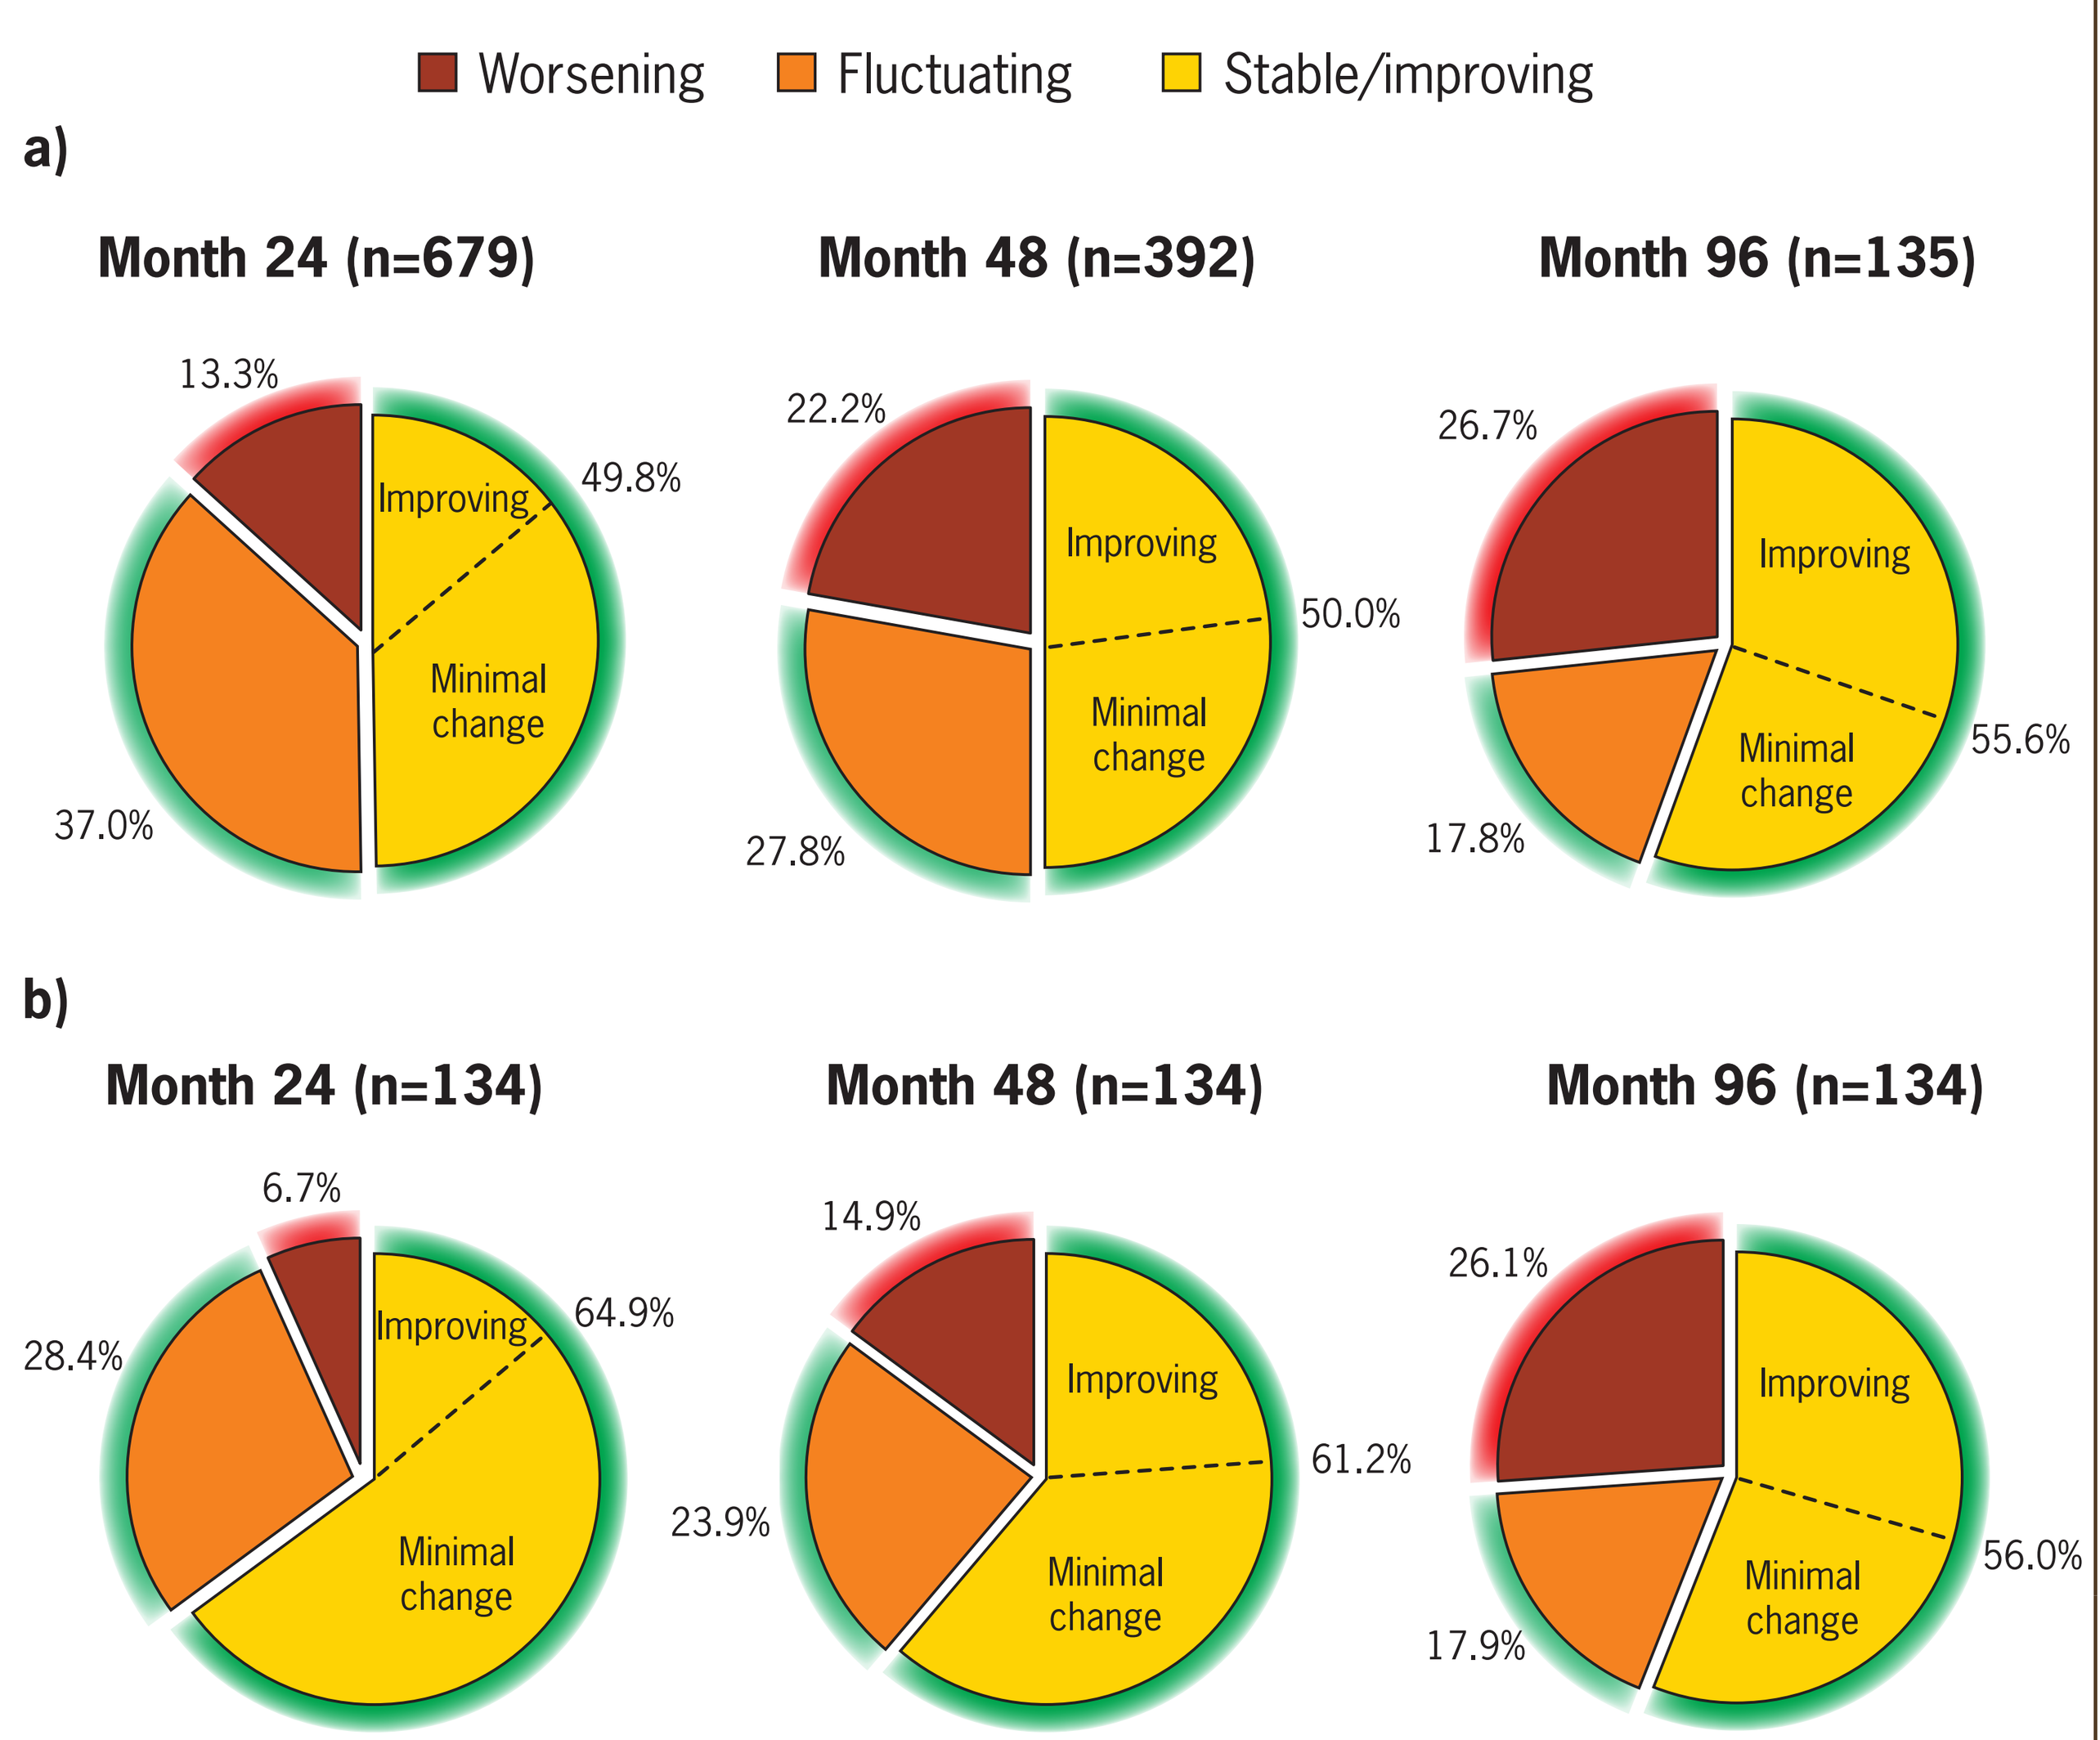
Worsening
Fluctuating
Stable/improving
a)
Month 24 (n=679)
13.3%
49.8%
37.0%
Month 48 (n=392)
22.2%
50.0%
27.8%
Month 96 (n=135)
26.7%
55.6%
17.8%
b)
Month 24 (n=134)
6.7%
64.9%
28.4%
Month 48 (n=134)
14.9%
61.2%
23.9%
Month 96 (n=134)
26.1%
56.0%
17.9%
Improving
Minimal change

RESULTS

Evolution of disability patterns over 8 years

- Categorical trend analysis of disability at 24 months and at 96 months supported the notion that meaningful changes are more likely to be observed over longer time periods
- In both the FAS (**Figure 1**) and CS (**Figure 2**), disability had fluctuated or changed minimally in most patients at 24 months (categories combined, 64.7–79.1%), but the proportions of patients in these categories had decreased by almost half at 96 months (categories combined, 34.5–43.3%)
- Among patients in the CS whose disability had fluctuated or changed minimally at 24 months:
 - in the continuous fingolimod group (n=106), 22.6% were improving, 22.6% were worsening and 54.7% had changed minimally or were still fluctuating at 96 months
 - in the switch group (n=75), 20.0% were improving, 28.0% were worsening and 52.0% had changed minimally or were still fluctuating at 96 months
- Overall, the proportions of patients in either the improving or worsening categories were greater at 96 months (26.1–34.5%) than at 24 months (6.7–18.5%)

Impact of delaying fingolimod treatment on long-term disability

- Between-group trends in the proportions of patients with worsening disability over 96 months supported the benefit of early fingolimod treatment (**Figures 1** and **2**)
 - Proportionately fewer patients had worsening disability in the continuous fingolimod group than in the switch group at 24 months (FAS, 13.3% vs 18.5%, p<0.01; CS, 6.7% vs 17.2%, p<0.01)
 - At 96 months, there were still proportionately fewer patients with worsening disability in the continuous fingolimod group than in the switch group, although differences were not significant (FAS, 26.7% vs 34.5%, p=0.18; CS, 26.1% vs 34.5%, p=0.15).

Long-term effect of fingolimod treatment on disability status

- Longitudinal comparison in the continuous fingolimod group suggested that the majority of patients had either stabilized or had improving disability over 96 months
 - Proportionately more patients had improving disability at 96 months than at 24 months (FAS, 30.4% vs 14.1%, p<0.01; CS, 30.6% vs 14.2%, p<0.01; **Figures 1** and **2**)
 - At least half of those receiving continuous fingolimod were categorized as stable/improving at all time points (**Figure 3**)
 - Patients whose disability was classified as fluctuating at 96 months might be regarded as stable because their disability had neither improved nor worsened overall. On this basis, nearly three-quarters (73.3–73.9%) of patients receiving continuous fingolimod could be considered as having stable/improving disability at 96 months (**Figure 3**)

References

1. Kobelt G, et al. J Neurol Neurosurg Psychiatry. 2006;77:918–926.
2. Kurtzke JF. Neurology. 1955;5:580–583.
3. Kurtzke JF. Neurology. 1983;33:1444–1452.
4. Wiendl H, Meuth S. Drugs. 2015;75:947–977.
5. Kappos L, et al. N Engl J Med. 2010;362:387–401.
6. Calabresi PA, et al. Lancet Neurol. 2014;13:545–556.
7. Liu C, Blumhardt LD. J Neurol Neurosurg Psychiatry. 1999;67:451–456.
8. Kappos L, et al. Neurology. 2015;84:1582–1591.
9. Cohen J, et al. Mult Scler. 2015;23(11 Suppl 1):P591.

Disclosures

Anthony T. Reder has received honoraria or research support from Abbott Laboratories, Acorda Therapeutics, Astra/Merck, Athena Neurosciences, Bayer, BioMS Medical Corp, Biogen Idec, Blue Cross Blue Shield, Boehringer Ingelheim, Caremark, Cephalon, Connective Therapeutics, Elan, Eli Lilly, F. Hoffmann-La Roche, Genentech, Genzyme, GlaxoSmithKline, Immunex, Neurocrine Biosciences, Novartis, Parke-Davis, Pfizer, Pharmacia & Upjohn, Questcor, Quintiles, Serono, Takeda Pharmaceuticals and Teva Marion Partners. **Jeffrey Cohen** has received research support and/or consulting fees from Biogen Idec, Consortium of Multiple Sclerosis Centers, Department of Defense (US), Genentech, Genzyme, National Institutes of Health, National MS Society (US), Novartis, Receptos, Synthon, Teva and Vaccinex, and has been a journal editor for *Multiple Sclerosis Journal – Experimental, Translational and Clinical*. **Daniela Piani Meier** and **Davorka Tomic** are employees of Novartis Pharma AG. **Shannon Ritter** is an employee of Novartis Pharmaceuticals Corporation. **Bruce A.C. Cree** has received personal compensation for consulting and/or contracted research support (including clinical trials) from AbbVie, Acorda, Biogen Idec, EMD Serono, F. Hoffmann-La Roche, Genzyme/Sanofi-Aventis, MedImmune, Novartis and Teva.

Acknowledgments

Editorial support was provided by Oxford PharmaGenesis, Oxford, UK, which was funded by Novartis Pharmaceuticals Corporation. The final responsibility for the content lies with the authors.

Your document will be available for download at the following URL:
http://novartis.medicalcongressposters.com/Default.aspx?doc=ceea7
And via text message (SMS)
Text: **Qceea7** *To:* **8NOVA (86682)** *US only*
+18324604729 *North, Central & South Americas; Caribbean; China*
Note: downloading data may incur costs which can vary depending on your service provider and may be high if you are using your smartphone abroad. Please check your phone tariff or contact your service provider for more details.

Scan to download this poster

This study was funded by Novartis Pharmaceuticals Corporation, USA | Poster presented at the 2016 Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC) | June 1–4, 2016 | National Harbor, Maryland, United States