Gastrointestinal Symptoms in Early Dimethyl Fumarate Therapy

John Foley, Tamara Hoyt, Angelene Christensen, Ryan Metzger

Rocky Mountain MS Research Group, Salt Lake City, UT

BACKGROUND:

Dimethyl fumarate (DMF) is an oral therapy approved for treatment of relapsing forms of multiple sclerosis (MS). Though DMF is generally well tolerated, gastrointestinal (GI) side effects are not uncommon, particularly during initiation of treatment. A better understanding of the prevalence and intensity of specific GI symptoms occurring with DMF could prove useful in characterizing the GI response to DMF therapy, and help direct strategies to mitigate these symptoms.

OBJECTIVE:

The aim of this study was to further characterize the prevalence, intensity and frequency of specific GI symptoms during initiation of therapy.

METHODS:

Twenty-four patients with relapsing-remitting MS were enrolled into an IRB-approved study and assessed for GI symptoms over the first 2 weeks of DMF therapy, as part of a screening process to determine eligibility for a subsequent evaluation of mitigation strategies for GI symptoms. DMF was prescribed at 120 mg twice daily for the first 7 days, then increased to 240 mg twice daily on day 8. One patient terminated treatment due to flushing, and therefore was excluded. Patients recorded their daily GI symptoms using the Modified Acute GI Symptom Scale (MAGISS) - which evaluates 9 symptoms on a 10-point intensity scale: upper and lower abdominal pain, nausea, vomiting, bloating, flatulence, diarrhea, constipation, and indigestion. Scores of 4-10 were considered moderate-to-severe intensity. One patient did not report symptom intensity for 4 of the symptoms, therefore was excluded from some analyses. Data were analyzed using standard descriptive statistical methods using GraphPad Prism v6.05 (La Jolla, CA).

TABLE 1
PATIENT CHARACTERISTICS

	n = 23
Gender	83% female, 17% male
Body Weight (kg; median, range)	71, 55 - 118
Age (years; mean ± st. dev., range)	46 ± 13, 27-72
Disease Duration (years; median, range)	10, 0.2-38

RESULTS:

FIGURE 1
TREATMENT TERMINATION DUE TO GI SYMPTOMS WAS UNCOMMON

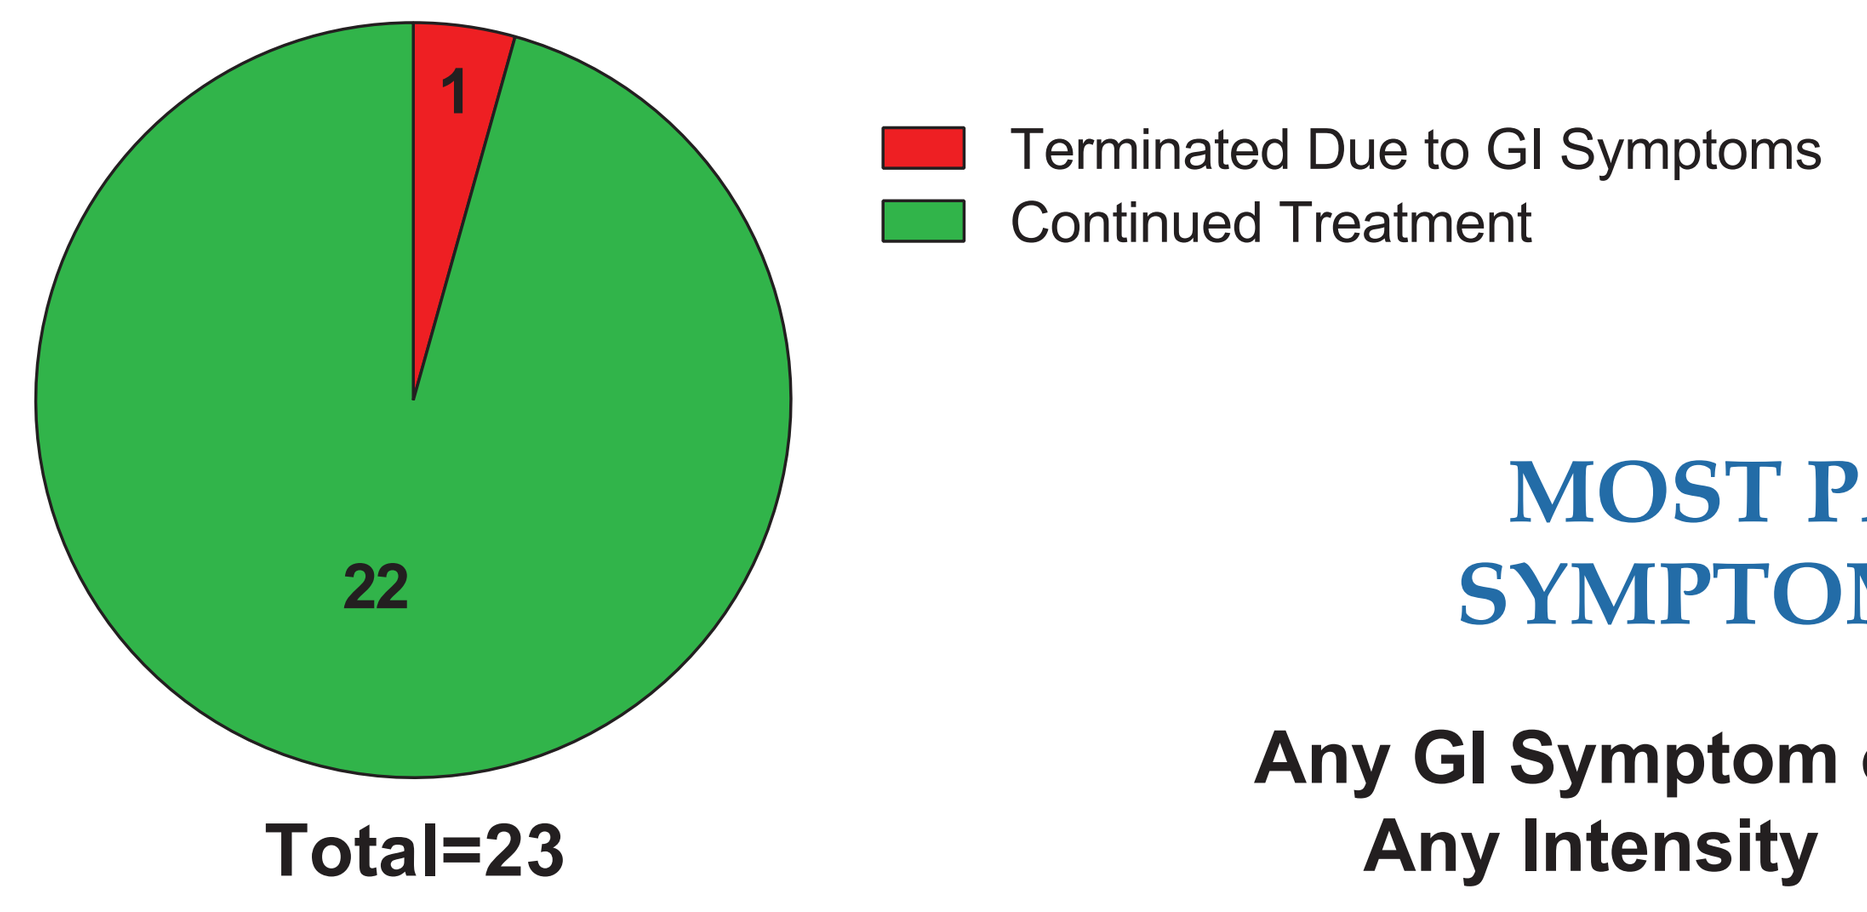
1
22
Terminated Due to GI Symptoms
Continued Treatment
Total=23

FIGURE 2
MOST PATIENTS EXPERIENCED GI SYMPTOMS FOR AT LEAST ONE DAY

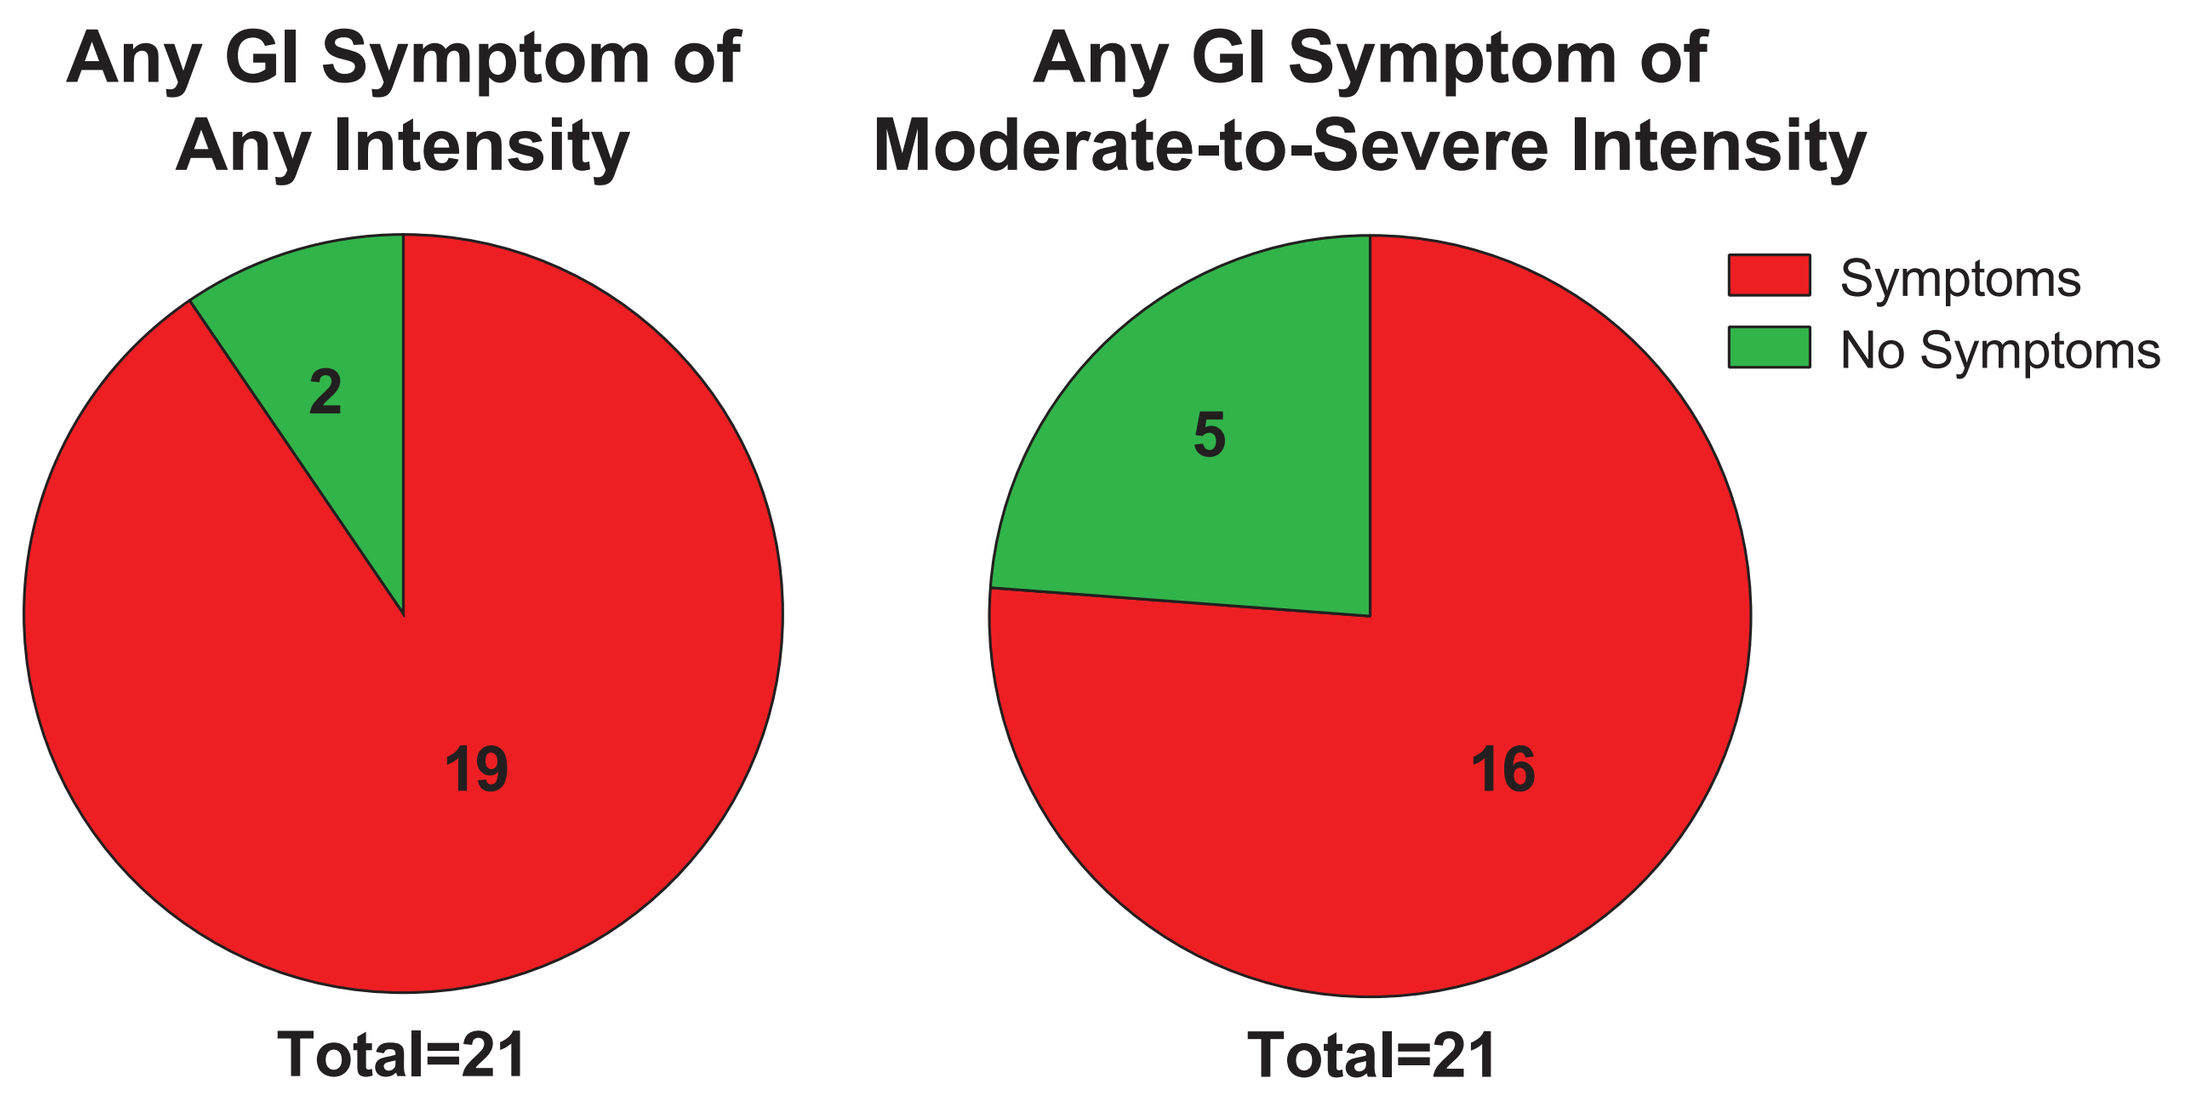
Any GI Symptom of Any Intensity
Any GI Symptom of Moderate-to-Severe Intensity
2
19
5
16
Symptoms
No Symptoms
Total=21
Total=21

*One patient did not rate several symptoms, and therefore was excluded here along with the patient who terminated treatment due to GI symptoms (Fig. 1)

FIGURE 3
PREVALANCE AND ACUITY OF EACH SYMPTOM

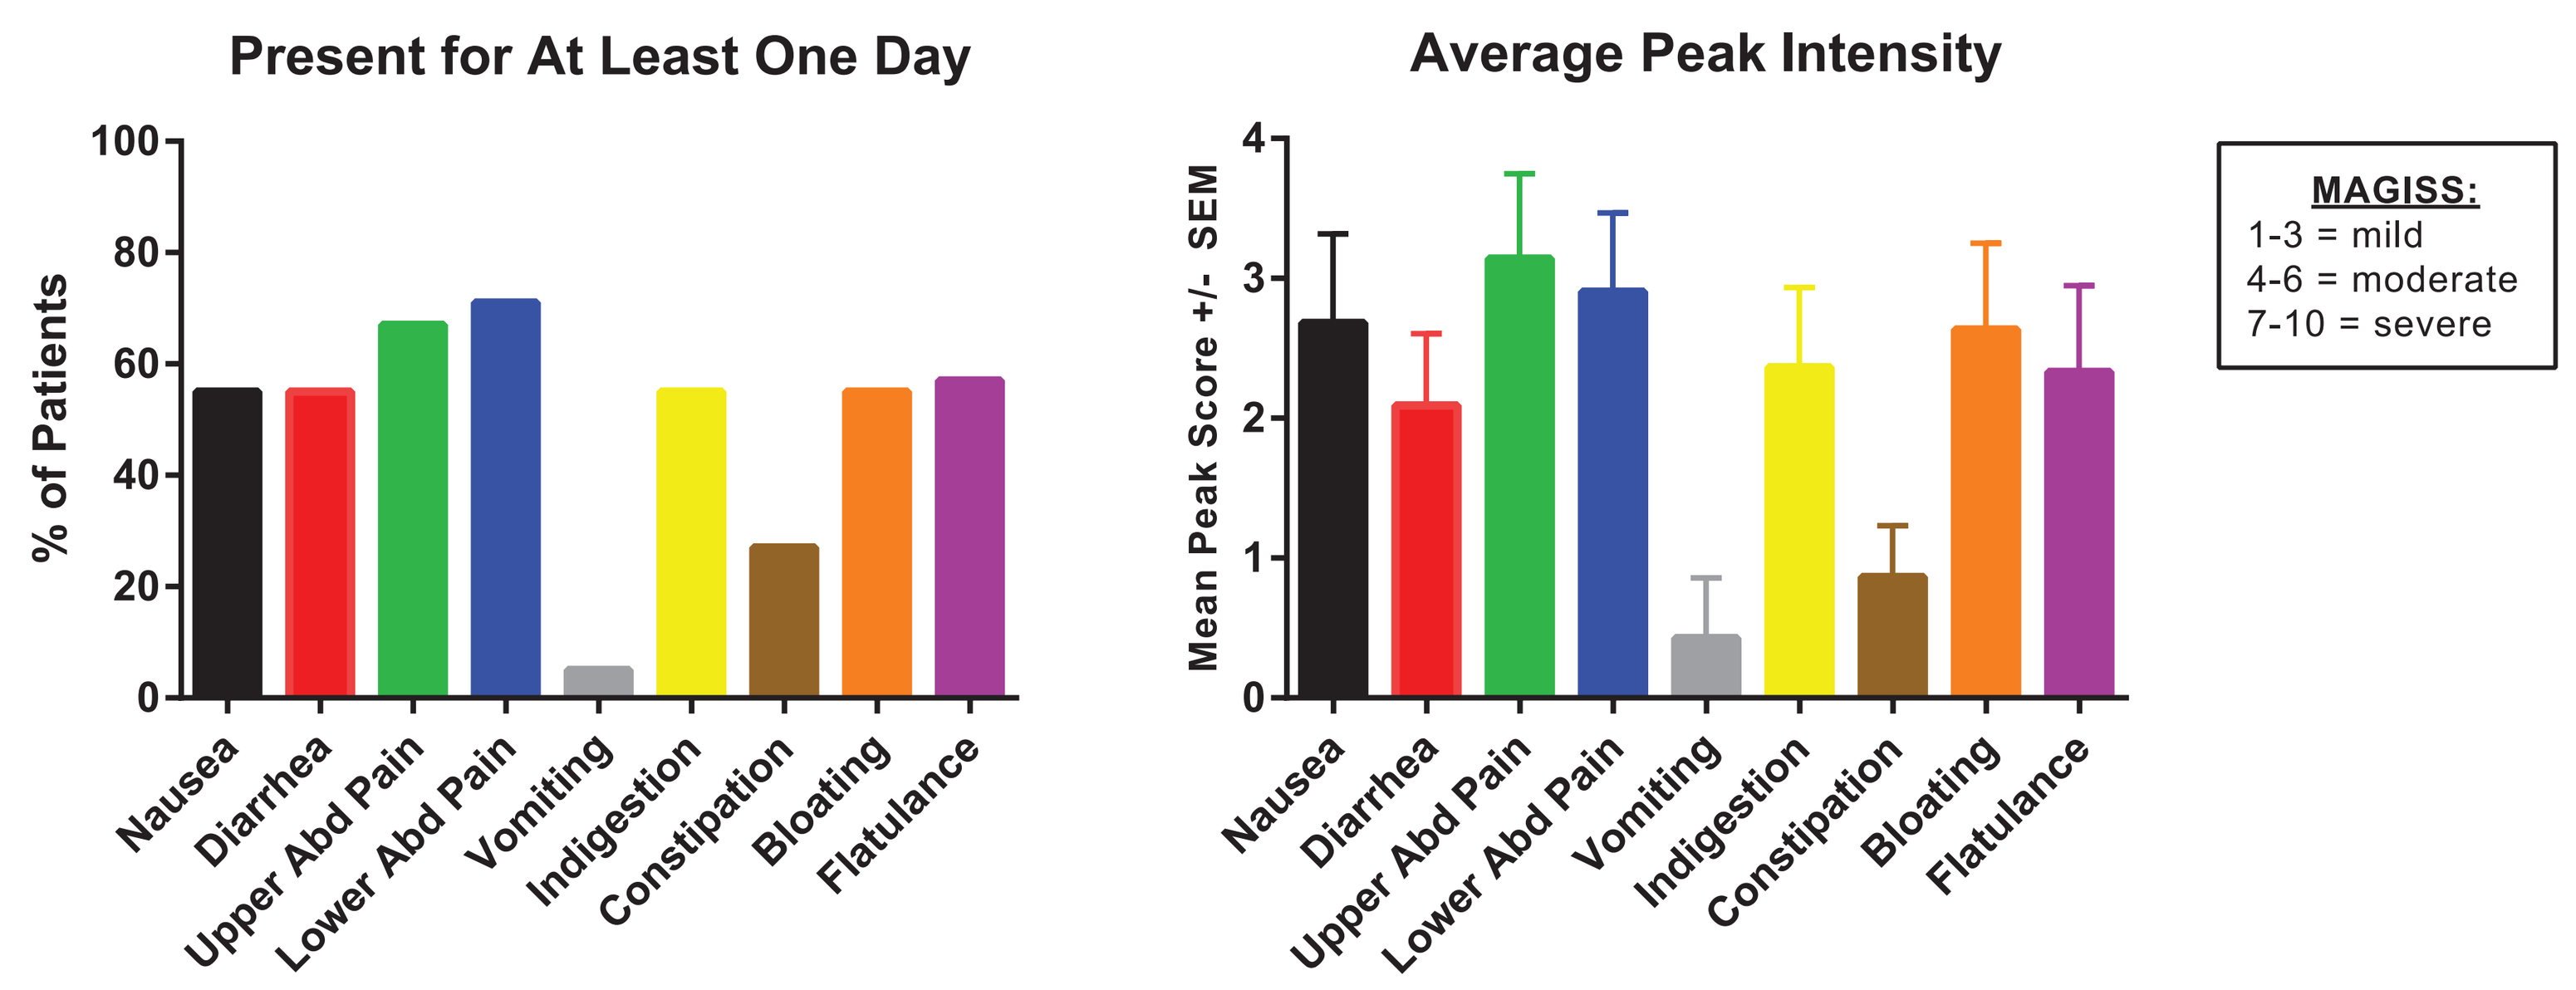
Present for At Least One Day
% of Patients
0
20
40
60
80
100
Average Peak Intensity
Mean Peak Score +/- SEM
0
1
2
3
4
Nausea
Diarrhea
Upper Abd Pain
Lower Abd Pain
Vomiting
Indigestion
Constipation
Bloating
Flatulance
MAGISS:
1-3 = mild
4-6 = moderate
7-10 = severe

FIGURE 4
MODERATE-TO-SEVERE SYMPTOMS ARE GENERALLY NOT SUSTAINED

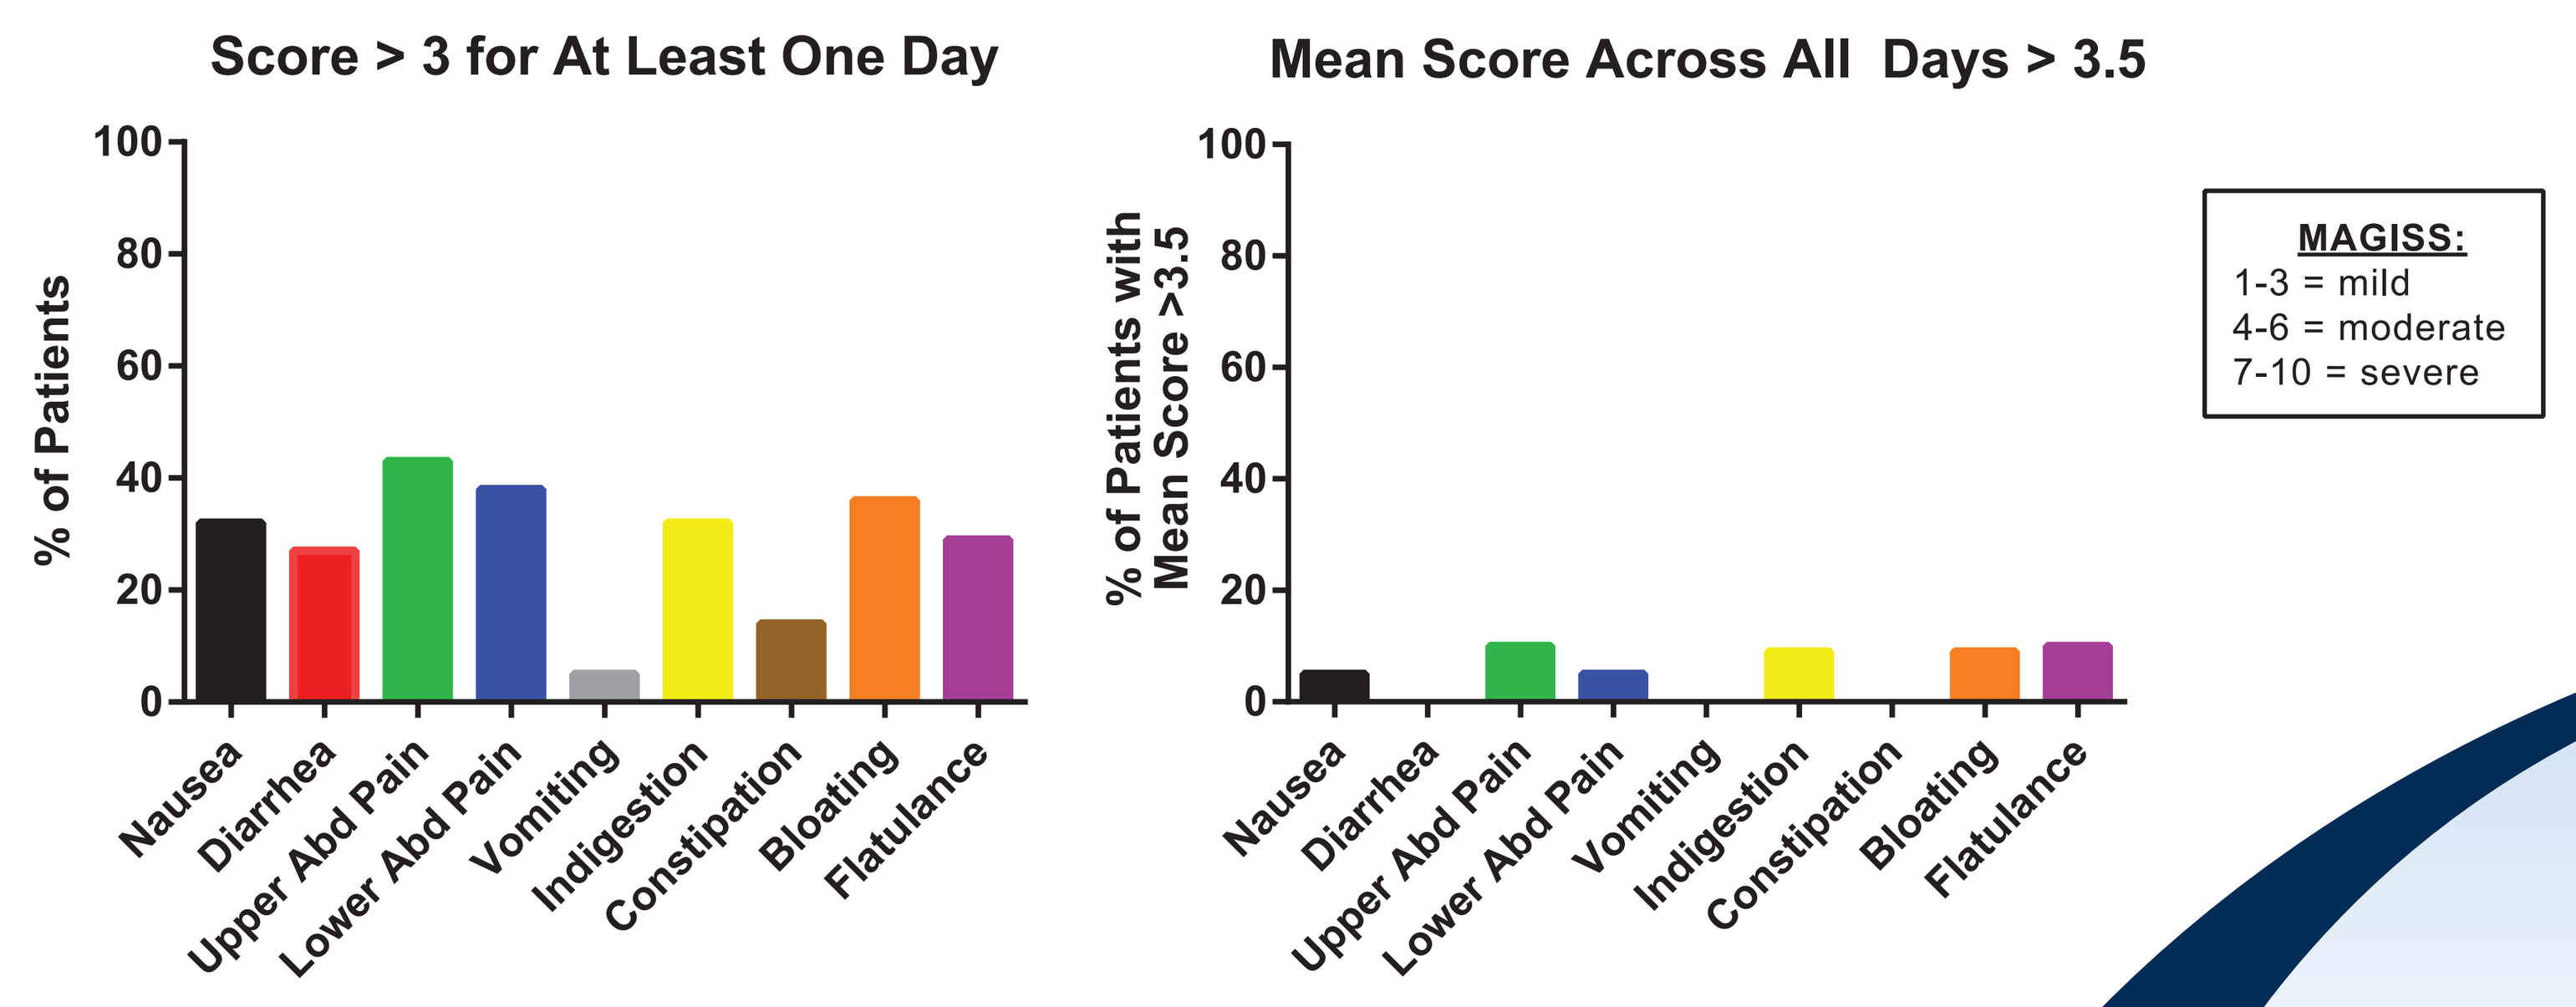
Score > 3 for At Least One Day
% of Patients
Mean Score Across All Days > 3.5
% of Patients with Mean Score >3.5
0
20
40
60
80
100
Nausea
Diarrhea
Upper Abd Pain
Lower Abd Pain
Vomiting
Indigestion
Constipation
Bloating
Flatulance
MAGISS:
1-3 = mild
4-6 = moderate
7-10 = severe

FIGURE 5
TIME-COURSE OF SYMPTOM SCORES OVER THE FIRST 14 DAYS OF DMF TREATMENT

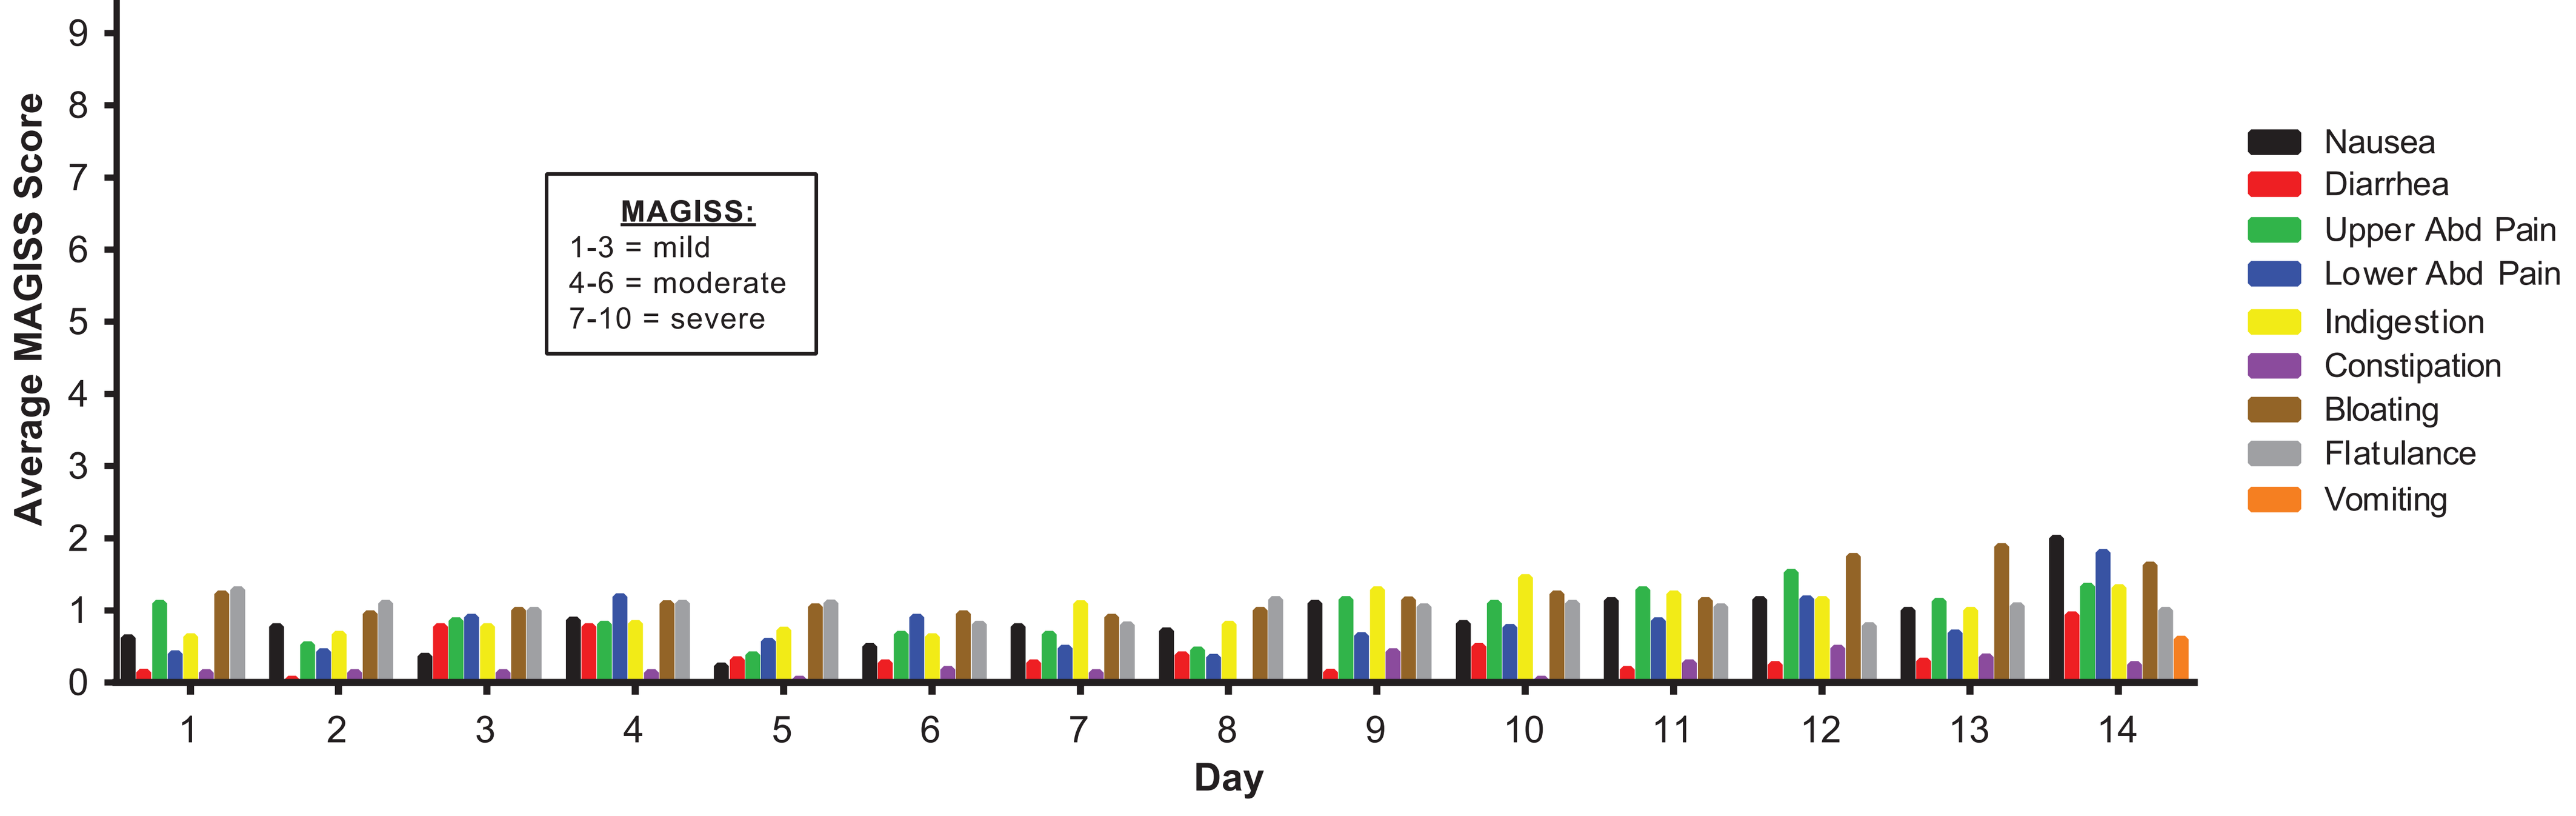
Average MAGISS Score
0
1
2
3
4
5
6
7
8
9
10
Day
1
2
3
4
5
6
7
8
9
10
11
12
13
14
MAGISS:
1-3 = mild
4-6 = moderate
7-10 = severe
Nausea
Diarrhea
Upper Abd Pain
Lower Abd Pain
Indigestion
Constipation
Bloating
Flatulance
Vomiting

FIGURE 6
ROCKY MOUNTAIN MS CLINIC PROTOCOL FOR MANAGEMENT OF GI SYMPTOMS FOR PATIENTS ON DMF THERAPY

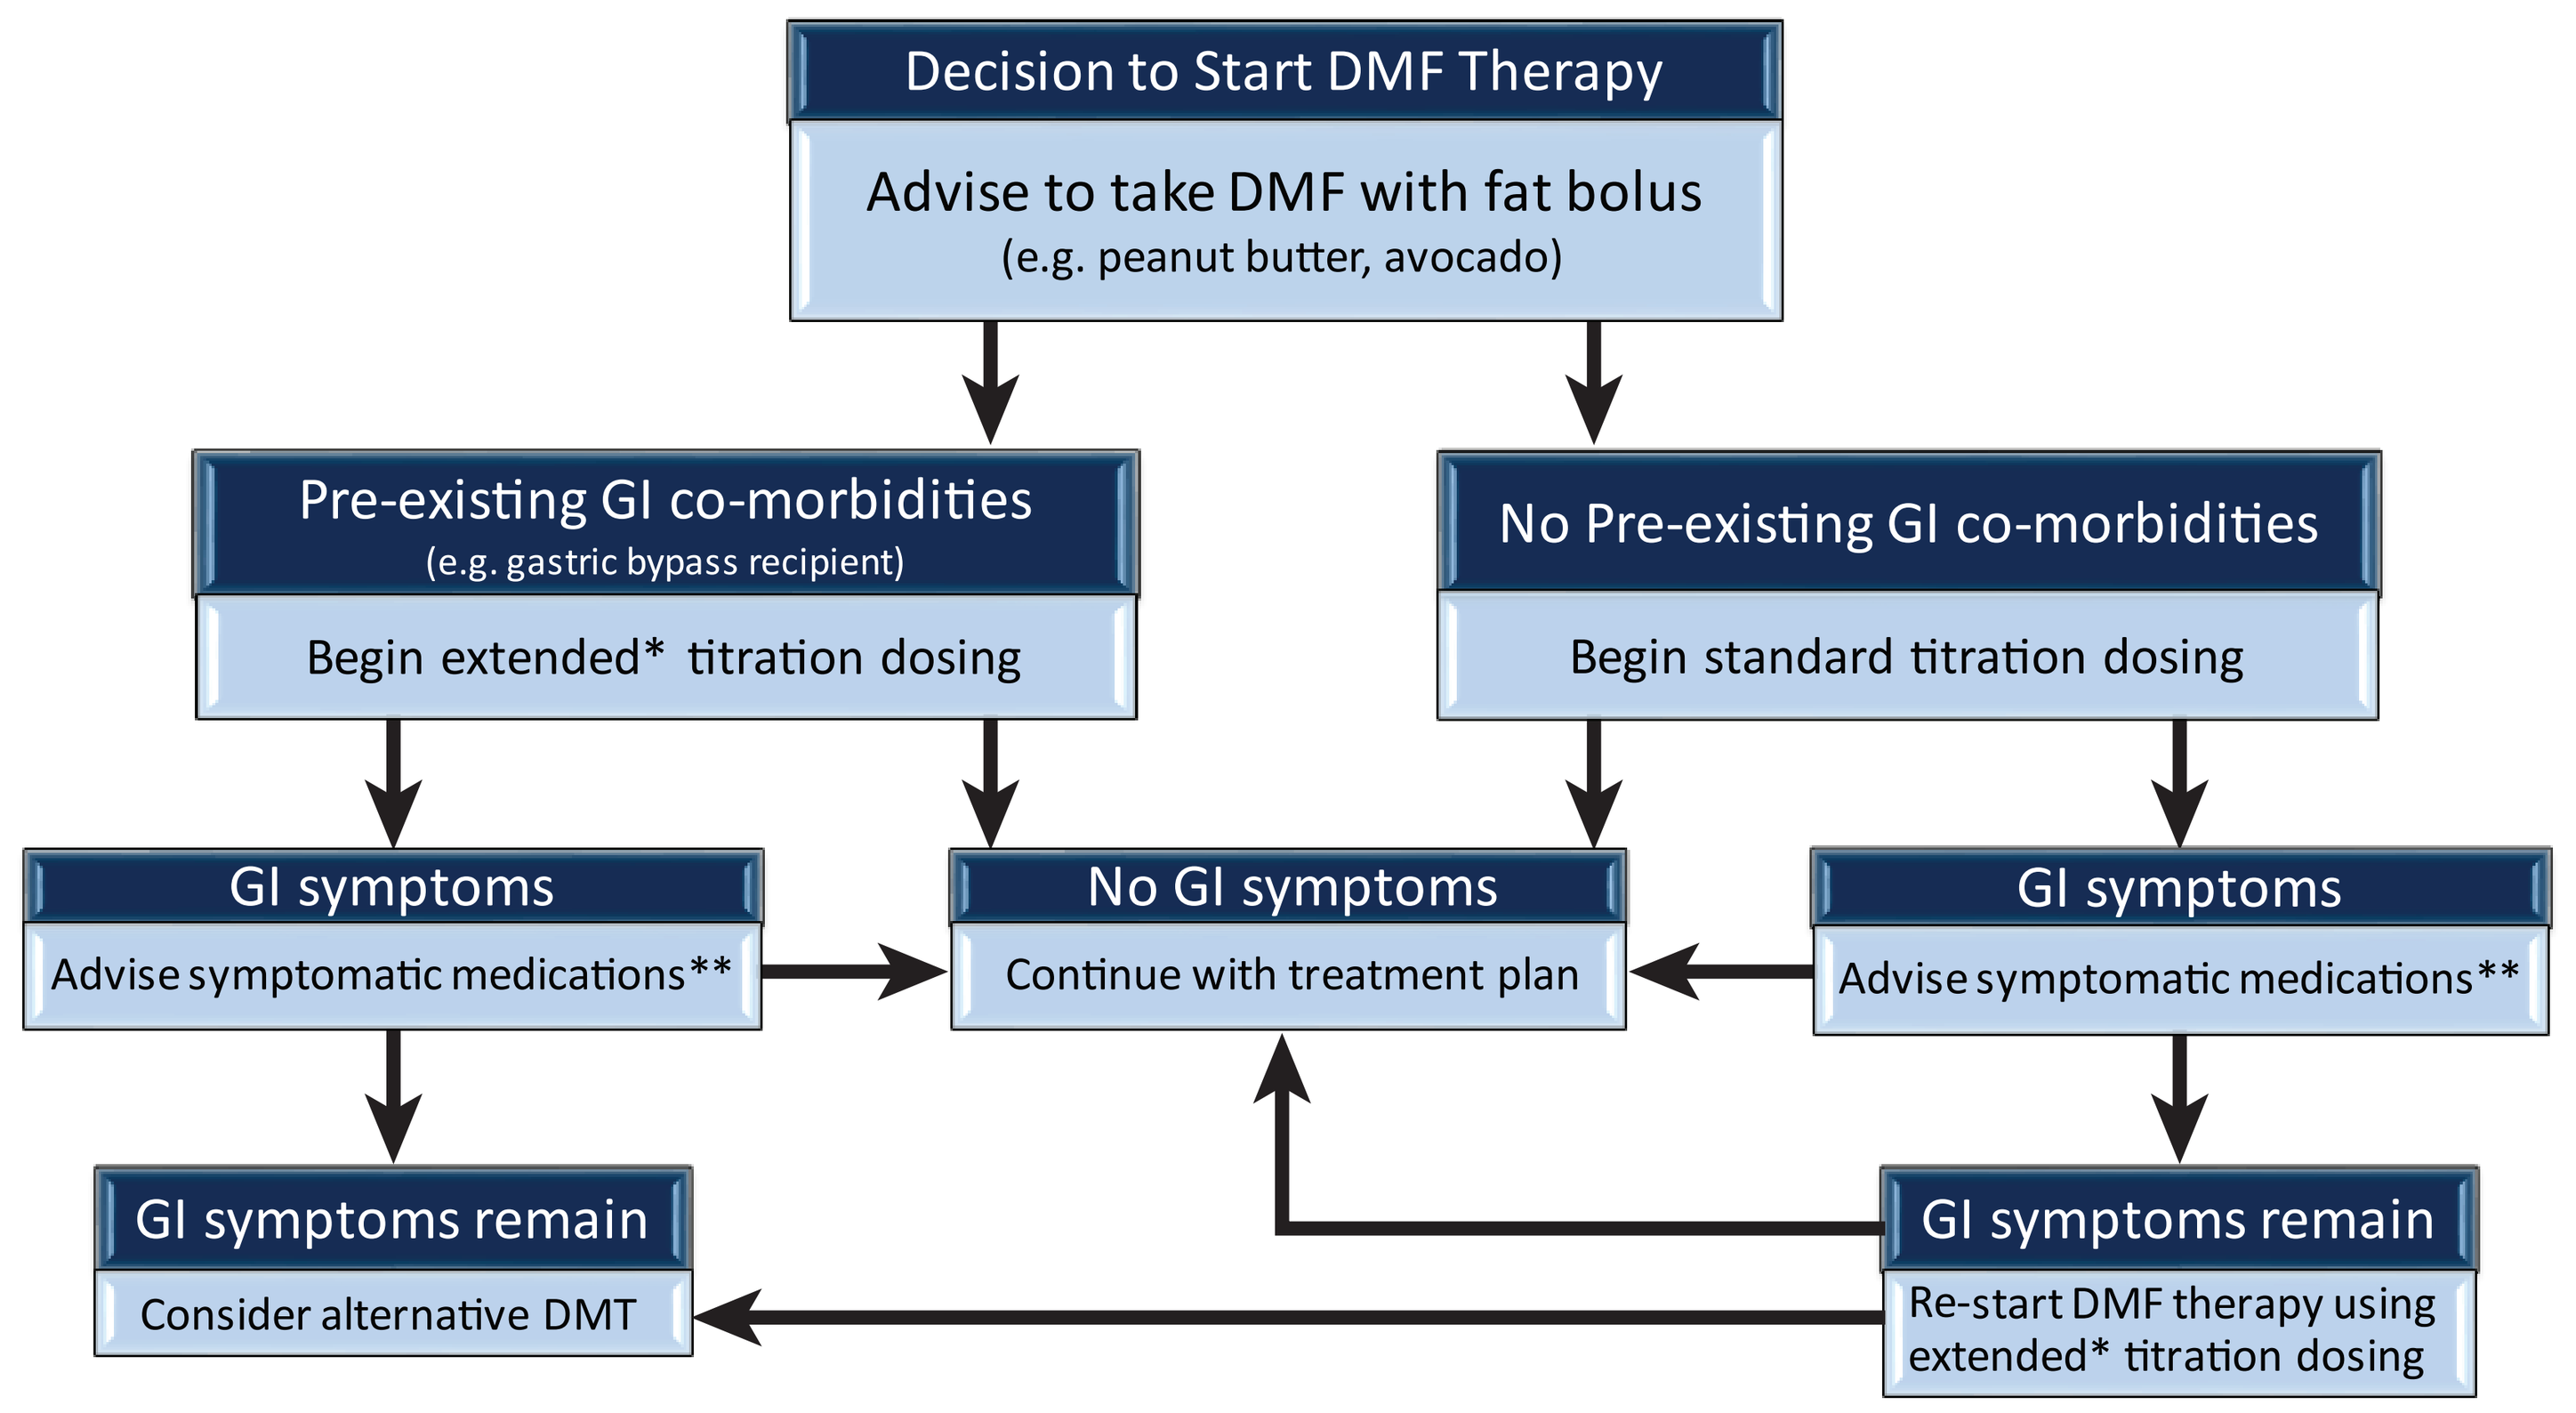
Decision to Start DMF Therapy
Advise to take DMF with fat bolus
(e.g. peanut butter, avocado)
Pre-existing GI co-morbidities
(e.g. gastric bypass recipient)
Begin extended* titration dosing
No Pre-existing GI co-morbidities
Begin standard titration dosing
GI symptoms
Advise symptomatic medications**
No GI symptoms
Continue with treatment plan
GI symptoms
Advise symptomatic medications**
GI symptoms remain
Consider alternative DMT
GI symptoms remain
Re-start DMF therapy using extended* titration dosing

**Symptomatic medications for GI symptoms

Symptom	Medication
Nausea	odansetron
Diarrhea	loperamide
Abdominal Pain	bismuth subsalicylate, simethicone
Indigestion	omeprazole, antacids
Bloating	simethicone
Flatulance	simethicone
Vomiting	odansetron

*Extended titration dosing = 120 mg once daily for 1 week, then 120 mg twice daily for 1 week, then 120 mg in morning + 240 mg in evening for 1 week, then 240 mg twice daily.

CONCLUSIONS:

- Over the first 2 weeks of DMF therapy, mild GI symptoms are common and wide-ranging.
- Abdominal pain, nausea, and bloating are the GI symptoms most likely to be of higher intensity during this time.
- Few patients experience moderate-to-severe symptoms for a sustained period.
- GI symptoms very rarely lead to treatment termination at this early stage of therapy.
- Strategies that may prove helpful in mitigating GI symptoms could include: 1) accompanying DMF administration with fat-containing food, 2) utilization of over-the-counter medications for GI symptoms, 3) extended titration dosing, or 4) dose reduction[1,2]. Indeed, experience at this center suggests improved tolerance of DMF utilizing these strategies.

DISCLOSURES:

- Study supported by an Investigator-Initiated Trial grant from Biogen.
- JF has received personal honoraria from Biogen, Genzyme and Teva Pharmaceuticals for consulting services.
- RM has a family member who is employed by Biogen.

REFERENCES:

1. Consensus Management of Gastrointestinal Events Associated with Delayed-Release Dimethyl Fumarate: A Delphi Study. Phillips TJ, Erwin AA, Agrella S et al. Neurol Ther. 2015 Dec;4(2):137-46.
2. Gastrointestinal Tolerability of Delayed-Release Dimethyl Fumarate in a Multicenter, Open-Label Study of Patients with Relapsing Forms of Multiple Sclerosis (MANAGE). Fox EJ, Vasquez A, Grainger W et al. MS Care. 2016 Jan-Feb;18(1):9-18