Patient initiation of fingolimod treatment at home: cardiac monitoring and patient satisfaction from the Gilenya@Home program

John Osborne,[1] Jamie L Weiss,[2] Xiangyi Meng,[2] Brandon Brown[2]

[1]State of the Heart Cardiology, Grapevine, TX, United States;
[2]Novartis Pharmaceuticals Corporation, East Hanover, NJ, United States

MC04

Introduction

- Fingolimod (Gilenya®) 0.5 mg/day is approved for relapsing forms of multiple sclerosis (MS);[1] more than 204 000 patients with MS have been treated with fingolimod.
- Asymptomatic, transient decreases in heart rate (HR) can occur at first dose of fingolimod, a pharmacodynamic effect arising from its interaction with sphingosine 1-phosphate receptors on atrial myocytes.[2-4]
- Thus, fingolimod prescribing information requires first-dose observation (FDO) of patients for at least 6 hours, with monitoring of HR and blood pressure (BP).[1]
- Originally, fingolimod FDO was conducted only in medical facilities, but in October 2014 the Gilenya@Home program offered patients the convenience of starting treatment at home.
- Gilenya@Home is conducted by clinicians trained in fingolimod pharmacology, FDO procedure and cardiovascular life support.

Objective

- To report cardiac safety data and to assess patient-reported satisfaction among patients who have completed their FDO as part of the Gilenya@Home program.

Methods

Gilenya@Home program overview

- Fingolimod initiation in the Gilenya@Home program is summarized in **Figure 1**.
- Before scheduling a Gilenya@Home visit, each patient undergoes baseline assessment, including electrocardiogram (EKG) recording, clinical laboratory measurements, and medical history review for any contraindications (**Table 1**).
- Before fingolimod is administered during the Gilenya@Home visit, baseline assessment and cardiac history are reviewed, EKG recording is repeated, and the fundus and macula are examined.
- FDO begins when fingolimod 0.5 mg is administered; HR and BP are recorded at baseline and at hourly intervals, and the patient is monitored for adverse events (AEs), including any that may be bradycardia-related.
- Patients are encouraged to report all symptoms during FDO.
- At 6 hours post-administration, EKG, HR and BP are recorded, and discharge evaluation is undertaken. Monitoring is extended until recovery if discharge criteria are not met.
- For monitoring beyond 10 hours, or if new-onset second-degree (or higher) atrioventricular (AV) block occurs, patients are transferred to an emergency room for continuous, precautionary EKG monitoring overnight.

Safety

- Retrospective safety data were collected anonymously from Gilenya@Home patient records.
- Quorum Review Institutional Review Board exempted the analysis from ethical approval (Quorum Review, Seattle, WA, United States).

Figure 1. Overview of the Gilenya@Home FDO procedure

Baseline assessment before treatment initiation

- Resting EKG is recorded
- Stipulated fingolimod label precautions and contraindications are checked
- Fingolimod should not be initiated at home if the patient:
 - has a prolonged QTc interval (men, >450 ms; women, >470 ms)
 - is at additional risk of QT prolongation
 - is concomitantly receiving drugs that can prolong QT interval or slow HR or AV conduction

Assessment at home, treatment initiation and the FDO procedure

- On the day fingolimod treatment initiation is scheduled, but before any drug is administered:
 - baseline assessment results are reviewed
 - relevant cardiac history is confirmed
 - an EKG is recorded, and if required, reviewed immediately (electronically) by a cardiologist
 - the fundus and macula are examined using a portable optical coherence tomography machine
- If assessments confirm that treatment can be initiated, a single 0.5 mg dose of fingolimod is administered
- Vital signs (HR and BP) are recorded at baseline and hourly for 6 hours[a] with monitoring for symptoms of bradycardia[b]
- After 6 hours, an EKG is recorded, HR and BP are measured and a discharge evaluation is undertaken

Discharge criteria

- Patients meet the discharge criteria and are discharged from FDO if:
 - they have been observed for ≥6 hours
 - their HR is >45 bpm and has passed its nadir
 - they have no symptoms relating to a decreased HR
- If any of these criteria are not met, the at-home FDO procedure continues until recovery
- If the criteria are not met at 10 hours, the patient is transferred to the ER for continuous EKG monitoring overnight
 - The patient is also transferred to the ER if a prolonged QTc interval is recorded, or there is new onset second-degree (or higher) AV block post-FDO
- The discharge process is completed by returning a 'Vital signs flow sheet' and a 'Treating physician report' to the patient's prescribing neurologist

[a]Vital signs can be recorded more frequently, and for up to 10 hours if needed.
[b]Syncope, near syncope, loss of consciousness, nausea, emesis, chest pain or shortness of breath.
ER, emergency room; QTc, corrected QT.

Table 1. Contraindications to fingolimod FDO in any setting and additional contraindications specific to the Gilenya@Home program

Fingolimod contraindications in any setting	
Factors that contraindicate initiation of fingolimod in any setting	Patients who, in the preceding 6 months, have experienced any of the following: • myocardial infarction • unstable angina • stroke • transient ischemic attack • decompensated heart failure requiring hospitalization • class III/IV heart failure
	Patients with a history or presence of the following: • Mobitz type II second- or third-degree AV block • sick sinus syndrome (unless the patient has a pacemaker) • baseline QTc interval ≥500 ms
	Patients receiving class Ia or class III anti-arrhythmic drugs
Additional fingolimod contraindications specific to the Gilenya@Home program	
Factors that contraindicate at-home fingolimod initiation, owing to the need for overnight EKG monitoring in a medical facility	Patients who tolerate bradycardia poorly or who may experience serious heart rhythm disturbances, including the following: • ischemic heart disease • history of myocardial infarction • congestive heart failure • history of cardiac arrest • cerebrovascular disease • uncontrolled hypertension • history of symptomatic bradycardia • history of recurrent syncope • severe untreated sleep apnea • AV block • sinoatrial heart block

Patient satisfaction with the Gilenya@Home program

- A patient-satisfaction survey was administered to all patients at the end of FDO under the Gilenya@Home program. Patients were asked to return the survey by mail or by email.
- The survey comprised 13 questions, covering four main areas of patient satisfaction (**Figure 2**).
- Patients answered the survey using a five-point Likert scale, rating their experience as either 'very good', 'good', 'fair', 'poor' or 'very poor'.

Data analyses

- All safety, AE and survey satisfaction data are reported descriptively.

Figure 2. Survey results for patient satisfaction with the Gilenya@Home program (N=1067)

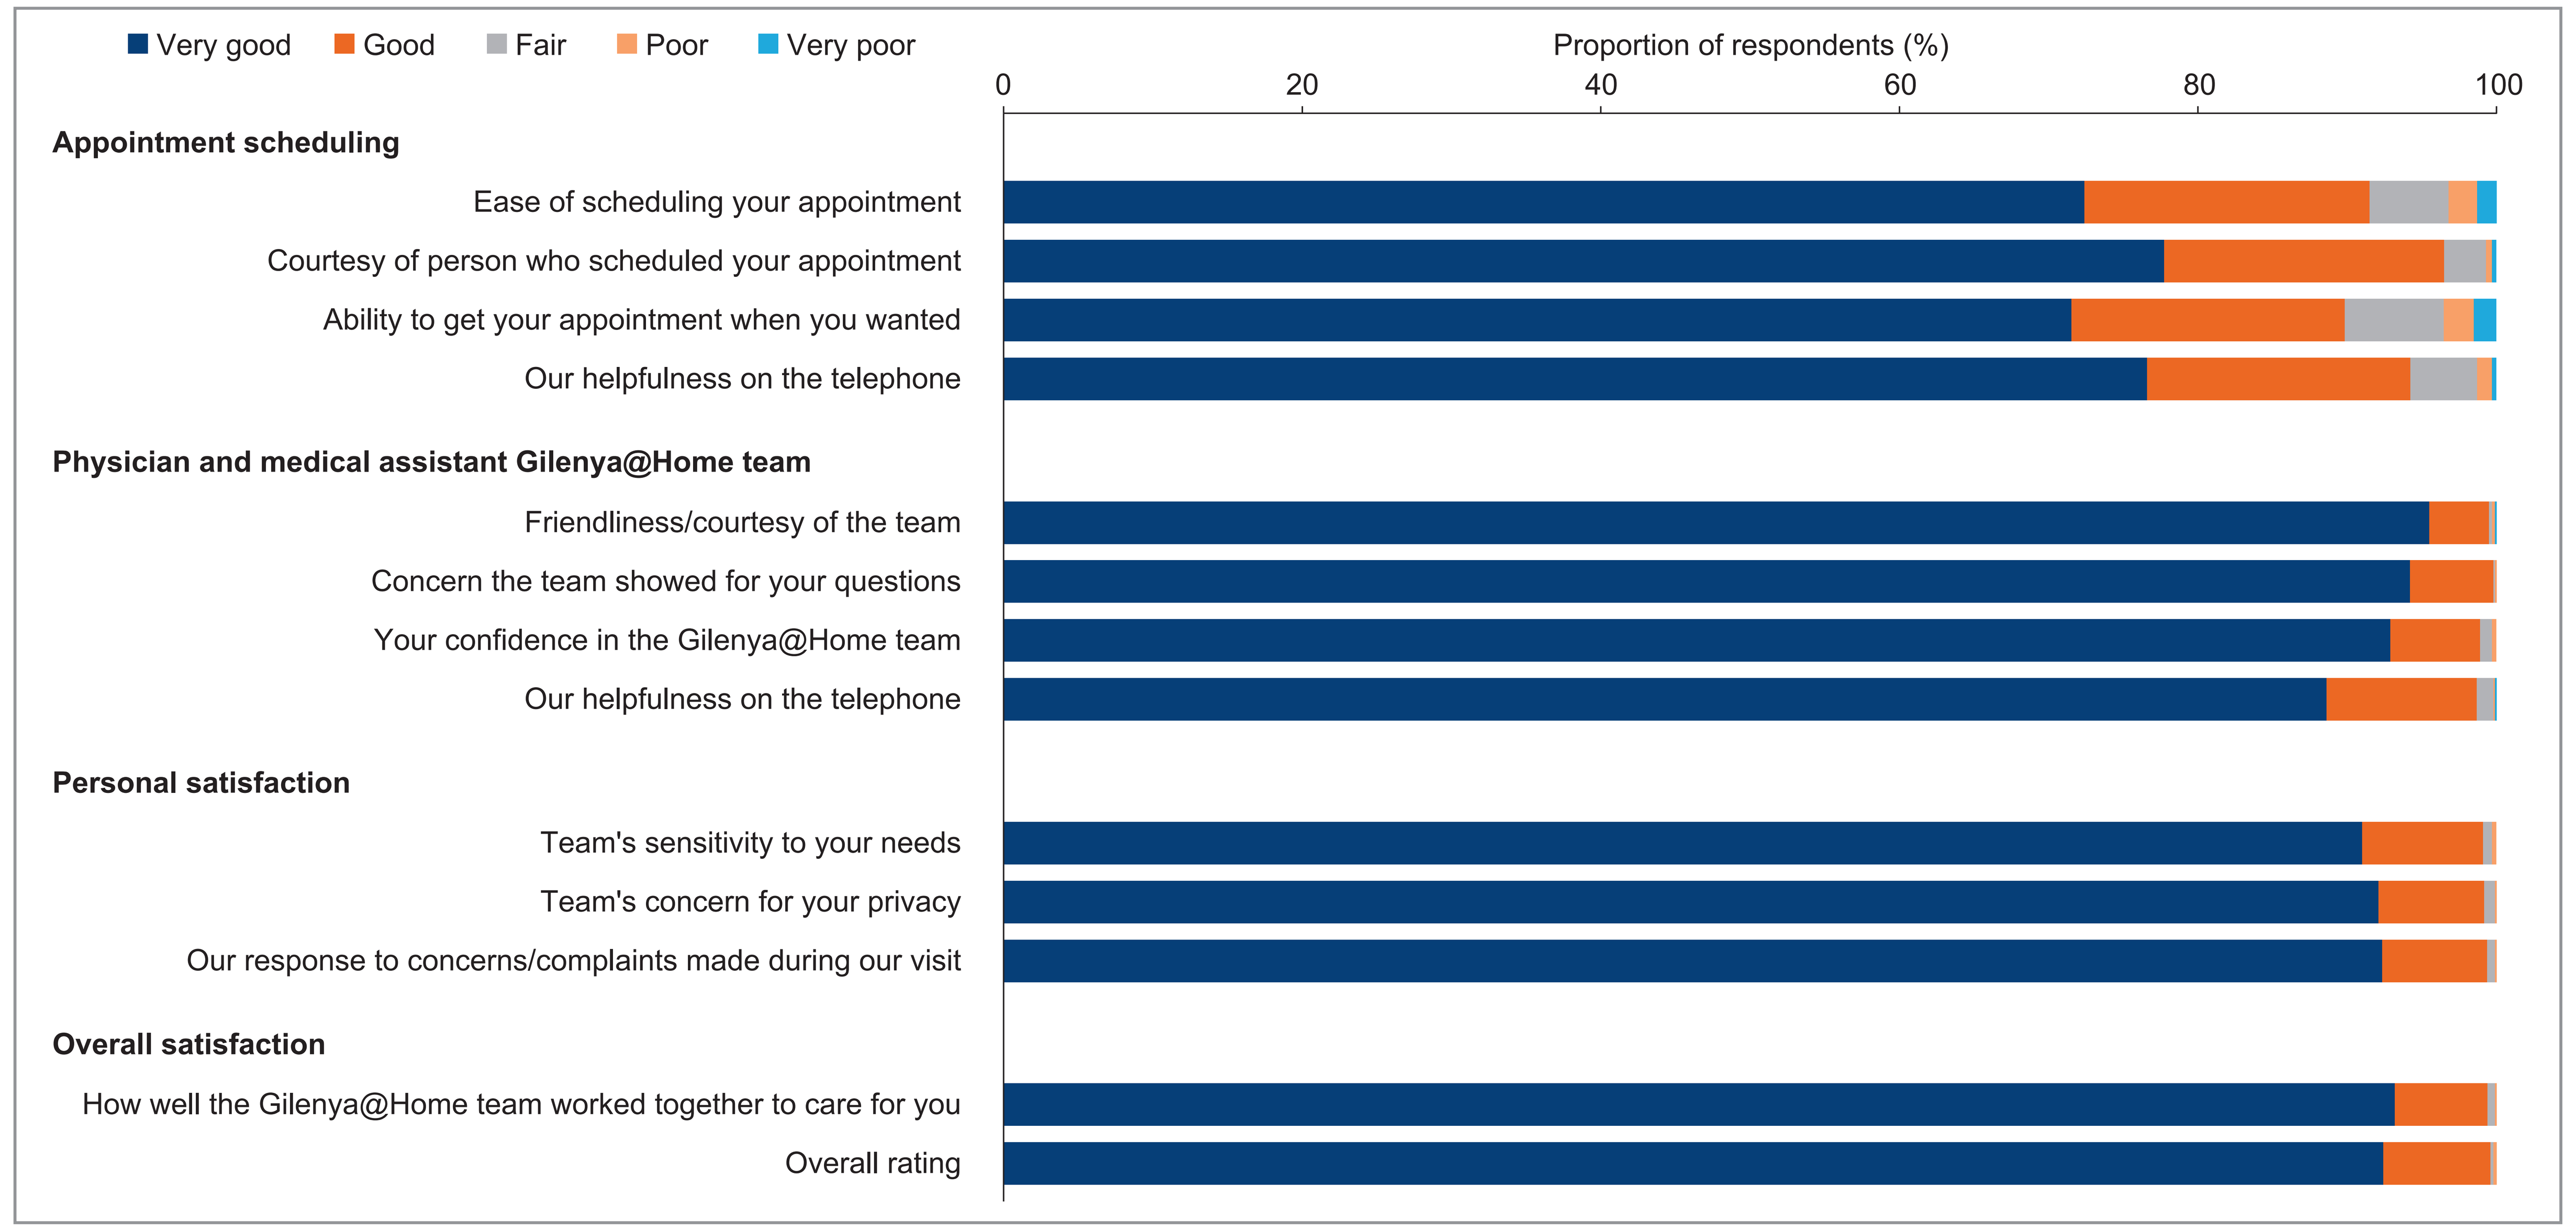
Very good
Good
Fair
Poor
Very poor
Proportion of respondents (%)
0
20
40
60
80
100
Appointment scheduling
Ease of scheduling your appointment
Courtesy of person who scheduled your appointment
Ability to get your appointment when you wanted
Our helpfulness on the telephone
Physician and medical assistant Gilenya@Home team
Friendliness/courtesy of the team
Concern the team showed for your questions
Your confidence in the Gilenya@Home team
Our helpfulness on the telephone
Personal satisfaction
Team's sensitivity to your needs
Team's concern for your privacy
Our response to concerns/complaints made during our visit
Overall satisfaction
How well the Gilenya@Home team worked together to care for you
Overall rating

Results

Cardiac safety

- Safety data were collected (October 2014 to April 2015) from 511 patients, including 354 women (69.3%).
- Most patients were monitored for only 6 hours; mean baseline HR, reduction in HR during FDO and extended monitoring data are shown in **Table 2**.
- In 25 patients monitored for up to 10 hours, the mean (SD) reduction in HR from baseline was 15.5 (8.9) bpm at 6 hours and 7.0 (11.6) bpm at 8 hours.
- No second-degree or complete AV block was observed.
- Mean maximum reduction in HR at 6 hours, the proportion of patients monitored for more than 6 hours and rates of symptomatic bradycardia were similar to values reported from the pooled phase 3 clinical trials of fingolimod 0.5 mg (**Table 2**).[5]

Adverse events

- 154 (30.1%) patients reported AEs; most frequent AEs were:
 - dizziness, n=31 (6.1%; 4 with bradycardia)
 - fatigue or somnolence, n=31 (6.1%)
 - nervousness or anxiety, n=11 (2.2%)
 - palpitations, n=3 (0.6%; none with bradycardia).

Table 2. Cardiovascular effects experienced by patients during Gilenya@Home FDO

Event	Gilenya@Home program (N=511)	Pooled phase 3 trials (fingolimod 0.5 mg)[5] (N=1212)
HR		
Baseline sitting HR, mean (SD), bpm	73.7 (11.7)	73.0 (9.7)
Maximum reduction at 6 hours, mean (SD), bpm	9.4 (9.4)	8.1 (8.1)
Monitoring for more than 6 hours,[a] n (%)	61 (11.9)	157 (13.0)
Monitoring for up to 10 hours,[b] n (%)	25 (4.9)	NR
Symptomatic bradycardia, n (%)	4 (0.8)	7 (0.6)
AV conduction abnormalities		
First-degree AV block, n (%)	9 (1.8)	56 (4.7)
Second-degree or complete AV block, n (%)	0 (0)	2 (0.2)[c]

[a]Patients are encouraged to report any symptoms experienced during Gilenya@Home, resulting in reported AE rates higher than those typically seen in clinical settings. Thus, precautionary extended monitoring can occur more often with Gilenya@Home than in medical facilities.
[b]Monitoring was extended if a patient's HR had not reached nadir by 6 hours.
[c]Both events were Wenckebach (Mobitz type I) second-degree AV block.
NR, not reported.

- Two patients (0.4%) were transferred to an emergency room for continuous EKG monitoring owing to symptomatic bradycardia (dizziness or lightheadedness):
 - both received standard care until HR normalized
 - both were released the next day and continued fingolimod treatment.

Patient satisfaction with the Gilenya@Home program

- Responses to satisfaction surveys were collected from October 2014 to May 2016.
 - Of 2951 patients completing FDO under Gilenya@Home, 1067 (36%) returned a completed survey.
- For all aspects of Gilenya@Home evaluated, at least 89% of patients rated satisfaction as 'very good' or 'good' (**Figure 2**).

Conclusions

- Based on patient survey responses, Gilenya@Home offers patients with relapsing forms of MS a convenient alternative to initiating fingolimod in a medical facility.
- Cardiac monitoring and safety data indicate that fingolimod initiation under Gilenya@Home is associated with a good safety profile.
- Most patients are eligible for Gilenya@Home and, based on responses to the patient survey, most have high levels of satisfaction with the program.

References

1. Novartis Pharmaceuticals Corporation. Prescribing information – Gilenya®. 2016. Available from: https://www.pharma.us.novartis.com/product/pi/pdf/gilenya.pdf (Accessed February 28, 2017).
2. Brinkmann V et al. *Pharmacol Ther.* 2007;115:84-105.
3. Means CK et al. *Cardiovasc Res.* 2009;82:193-200.
4. Camm J et al. *Am Heart J.* 2014;168:632-644.
5. DiMarco J et al. *Mult Scler Relat Disord.* 2014;3:629-638.

Acknowledgments

Editorial support was provided by Oxford PharmaGenesis, Oxford, UK, and was funded by Novartis Pharmaceuticals Corporation. The authors also acknowledge Ashish Pradhan, formerly of Novartis Pharmaceuticals Corporation, for his support in the development of this poster.

Disclosures

John Osborne: Novartis Pharmaceutical Corporation (Medical Director of the Gilenya@Home program, speaker for Novartis professional and consumer programs). **Jamie L Weiss, Xiangyi Meng** and **Brandon Brown:** Novartis Pharmaceuticals Corporation (employees).

© 2017 Novartis Pharmaceuticals Corporation.

Poster presented at the 2017 Annual Meeting of the Consortium of Multiple Sclerosis Centers, May 24-27, New Orleans, LA, United States.

This study was sponsored by Novartis Pharmaceuticals Corporation, United States.

Scan this QR code

Text: Q3367c
To: 8NOVA (86682) US only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe and Russia
+46737494608 Sweden and Europe
Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=3367c

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address:
josborne@sothcardiology.com