Poster No. DX41

Rates of comorbidities in patients with and without multiple sclerosis: a US retrospective claims database analysis

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

To download the PDF, scan the QR code or go to http://medpub-poster.merckgroup.com/CMSC2017_DX41.pdf

GET POSTER PDF

K Kresa-Reahl,[1] NC Edwards,[2] G Schneider,[3] K Stern,[3] AL Phillips[4]

[1]Providence Multiple Sclerosis Center, Portland, OR, USA; [2]Health Services Consulting Corporation, Boxborough, MA, USA; [3]Boston Health Economics, Inc., Waltham, MA, USA; [4]EMD Serono, Inc.,* Rockland, MA, USA

INTRODUCTION

- Emerging evidence suggests that comorbidities in multiple sclerosis (MS) are associated with delays in the diagnosis of MS,[1] increased rates of disability progression,[2] and decreased health-related quality of life.[3]
- Some therapies for MS may be contraindicated when certain comorbidities are present, or may increase the risk of comorbidity, resulting in difficult treatment decisions for patients and physicians.[4]
- Despite the relevance of comorbidity to MS treatment and outcomes, large population-based studies that describe the prevalence of comorbidities in patients with MS in the US are lacking.[5]

OBJECTIVE

- To compare the prevalence of comorbidities in patients with and without MS using data from a large US administrative claims database.

METHODS

- This retrospective administrative claims database study used IMS Health Real World Data Adjudicated Claims – US data from between January 1, 2011 and September 30, 2015.
 - Approximately 150 million patients with a medical benefit, and a subset of 95 million patients with both medical and pharmacy benefits, are included in the database.
- Patients with MS, aged 18–65 years, were required to have ⩾2 claims with a diagnosis of MS (International Classification of Diseases, Ninth Revision, Clinical Modification [ICD-9-CM] code: 340.xx) that were ⩾30 days apart.
- Exact matching was used to match patients with MS 1:1 to the pool of patients without MS (zero claims of MS) by age group (5-year intervals), sex, geographic region, and index year quarter.
- The index date for patients with MS was the date of the second MS claim, and for patients without MS was the 366th day of continuous enrollment. One year of continuous eligibility was required for all patients.
- The prevalence of the 10 most common comorbidities in MS (based on a systematic review of the published literature by Marrie *et al.*)[5] and of the individual comorbidities included in the Charlson Comorbidity Index (CCI) were compared between patients with and without MS.
 - Statistical significance was assessed using pairwise chi-squared tests.

RESULTS

- A total of 69,550 eligible patients with MS were matched to the pool of 3,129,573 patients without MS, resulting in 66,616 matched patients in each cohort (**Figure 1**).
- In both matched cohorts, the mean age was approximately 46 years and approximately 76% of patients were female (**Table 1**).
- The majority of matched patients had commercial insurance (97.03% with MS and 95.43% without MS; **Table 1**).
 - Although healthcare coverage by Medicaid or Medicare was uncommon, matched patients with MS used Medicare nearly twice as often as matched patients without MS (0.89% vs 0.46%, respectively). Conversely, matched patients without MS used Medicaid nearly twice as often as matched patients with MS (4.11% vs 2.07%, respectively).
- Of the 10 most common MS comorbidities, eight occurred significantly more frequently ($p<0.0001$) in patients with MS than in patients without MS (**Figure 2**).
- Of the 17 comorbidities included in the CCI, more than two-thirds (n=15; 88%) were significantly more common ($p<0.05$) in patients with MS than in those without MS (**Figure 3**).
 - The mean (standard deviation [SD]) CCI score was statistically significantly higher in patients with MS (0.46 [1.02]) than in those without MS (0.31 [0.87]) ($p<0.0001$). The median was 0 for both cohorts.

Table 1. Baseline demographic characteristics of matched cohorts of patients with and without MS

	Patients with MS (n=66,616)	Patients without MS (n=66,616)
Age, years		
Mean (SD)	45.63 (10.43)	45.59 (10.50)
Median	47	47
Age group, years, n (%)		
18–22	1188 (1.78)	1188 (1.78)
23–27	2501 (3.75)	2501 (3.75)
28–32	4913 (7.38)	4913 (7.38)
33–37	6753 (10.14)	6753 (10.14)
38–42	9071 (13.62)	9071 (13.62)
43–47	10,532 (15.81)	10,532 (15.81)
48–52	11,644 (17.48)	11,644 (17.48)
53–57	10,967 (16.46)	10,967 (16.46)
58–62	8079 (12.13)	8079 (12.13)
63–65	968 (1.45)	968 (1.45)
Female, n (%)	50,790 (76.24)	50,790 (76.24)
US region, n (%)		
Midwest	21,771 (32.68)	21,771 (32.68)
Northeast	18,382 (27.59)	18,382 (27.59)
South	20,296 (30.47)	20,296 (30.47)
West	6167 (9.26)	6167 (9.26)
Healthcare payer, n (%)[a]		
Commercial	64,640 (97.03)	63,572 (95.43)
Medicaid	1381 (2.07)	2735 (4.11)
Medicare	595 (0.89)	309 (0.46)

MS, multiple sclerosis; **SD**, standard deviation.
[a]Due to rounding, percentages do not equal 100%.

Figure 1. Matched cohort selection for the analysis of comorbidities in patients with and without MS

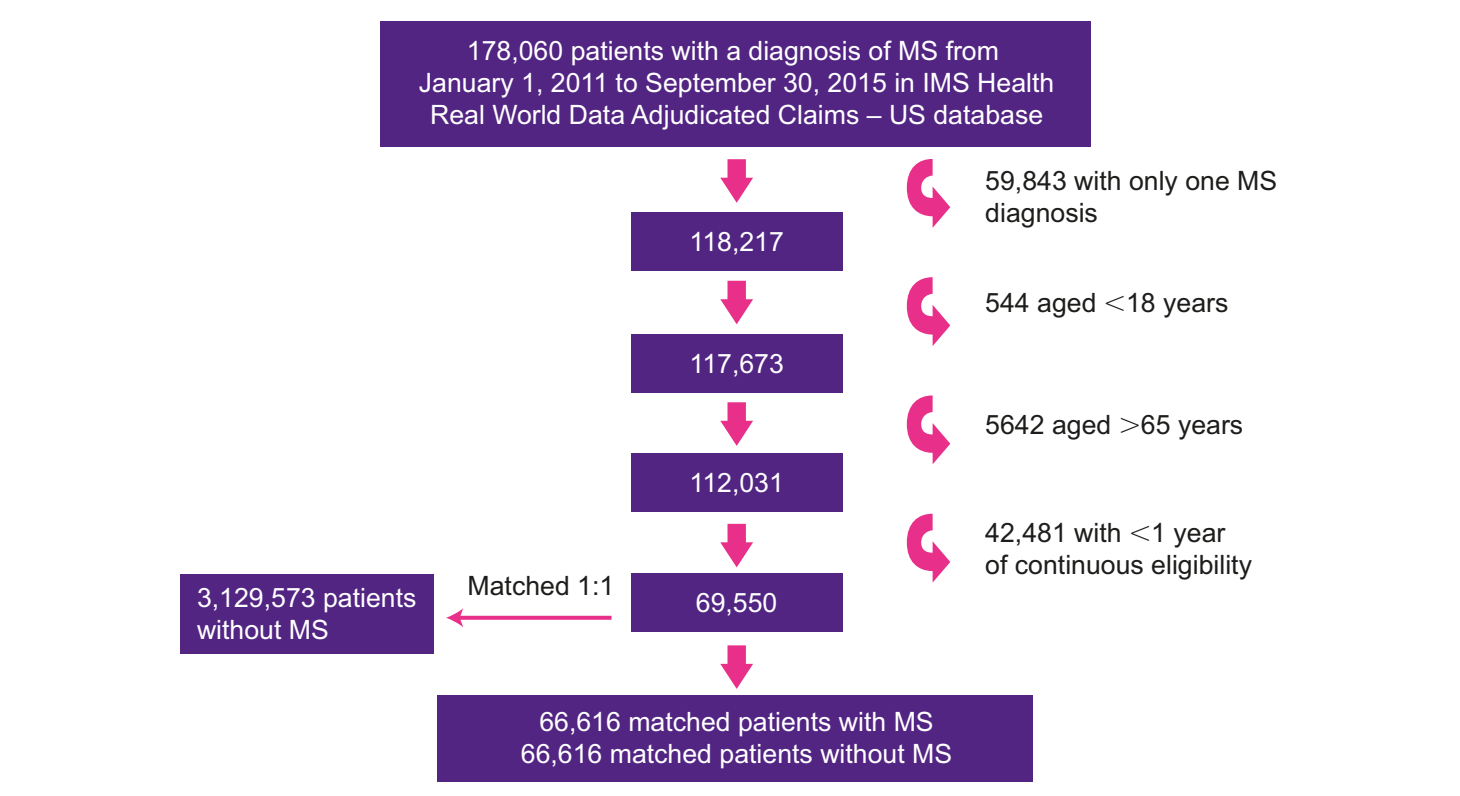
178,060 patients with a diagnosis of MS from January 1, 2011 to September 30, 2015 in IMS Health Real World Data Adjudicated Claims – US database
59,843 with only one MS diagnosis
118,217
544 aged <18 years
117,673
5642 aged >65 years
112,031
42,481 with <1 year of continuous eligibility
3,129,573 patients without MS
Matched 1:1
69,550
66,616 matched patients with MS
66,616 matched patients without MS

MS, multiple sclerosis.

- Patients with MS had a significantly lower prevalence of alcohol abuse ($p<0.05$; **Figures 2**), AIDS, mild liver disease, and moderate liver disease than patients without MS ($p<0.05$; **Figure 3**).
- No significant differences in prevalence between patients with and without MS were observed for type 1 and type 2 diabetes (**Figure 2**), diabetes without comorbidities and complications, and metastatic solid tumor (**Figure 3**).

Figure 2. Prevalence of the 10 most common comorbidities in MS in matched cohorts of patients with and without MS

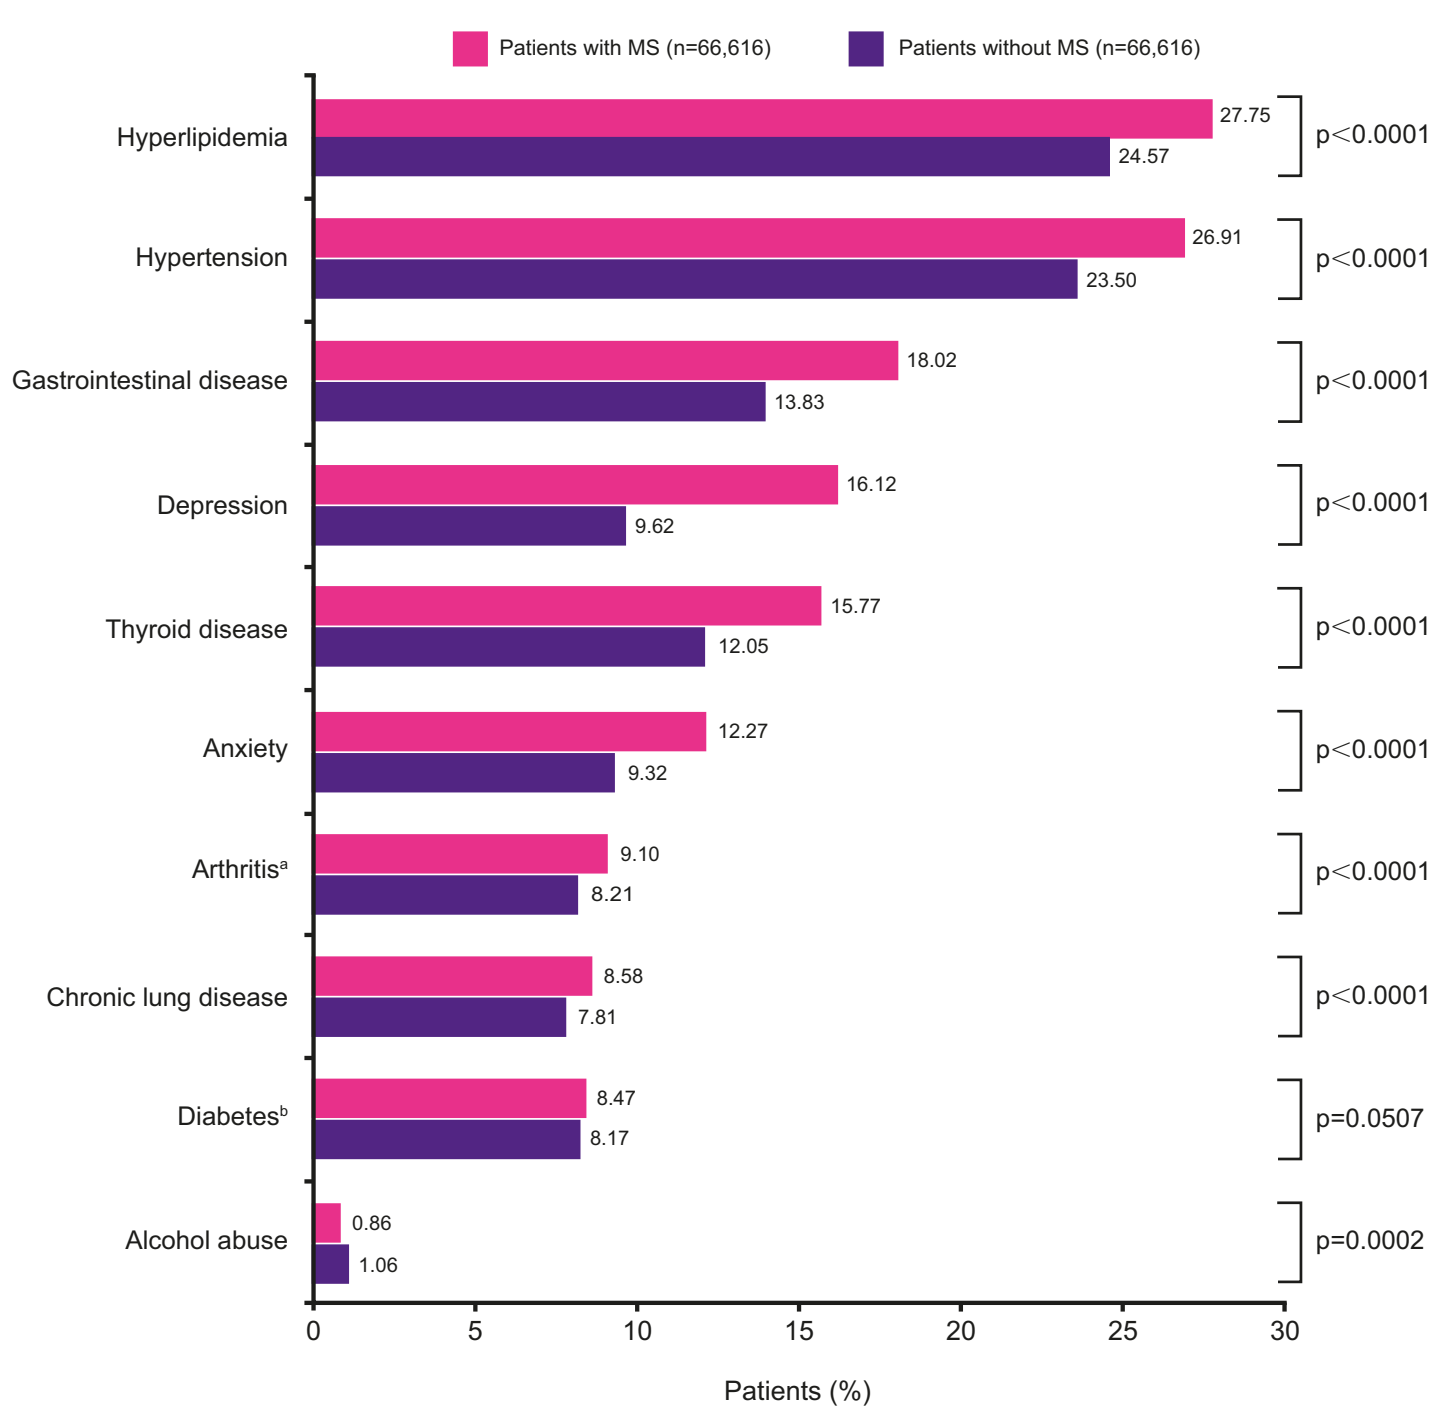
Patients with MS (n=66,616)
Patients without MS (n=66,616)
Hyperlipidemia 27.75 24.57 p<0.0001
Hypertension 26.91 23.50 p<0.0001
Gastrointestinal disease 18.02 13.83 p<0.0001
Depression 16.12 9.62 p<0.0001
Thyroid disease 15.77 12.05 p<0.0001
Anxiety 12.27 9.32 p<0.0001
Arthritis[a] 9.10 8.21 p<0.0001
Chronic lung disease 8.58 7.81 p<0.0001
Diabetes[b] 8.47 8.17 p=0.0507
Alcohol abuse 0.86 1.06 p=0.0002
0 5 10 15 20 25 30
Patients (%)

MS, multiple sclerosis.
Statistical significance was assessed using pairwise chi-squared tests.
[a]Rheumatoid arthritis or osteoarthritis.
[b]Type 1 and type 2 diabetes.

LIMITATIONS

- Claims data are not specifically collected for research purposes and missing information may limit the inferences that can be made from the data.
- The ICD-9-CM code for MS does not differentiate between different types of MS (eg, primary progressive MS, relapsing–remitting MS, or secondary progressive MS).
- Administrative claims databases provide information on patients with health insurance administered by regional health plans in the US and results may not be generalizable to patients who self-pay or patients without employer-sponsored commercial health insurance.

CONCLUSIONS

- **In this study, several comorbidities, such as hyperlipidemia, hypertension, gastrointestinal disorders, and depression, were significantly more common among patients with MS compared with those without MS.**
- **These results may better inform physicians and patients about potential comorbidities in patients with MS and may also help guide treatment decisions.**
- **Administrative claims data can be a valuable resource for investigating a variety of health service research questions.**

Figure 3. Prevalence of comorbidities included in the CCI in matched cohorts of patients with and without MS

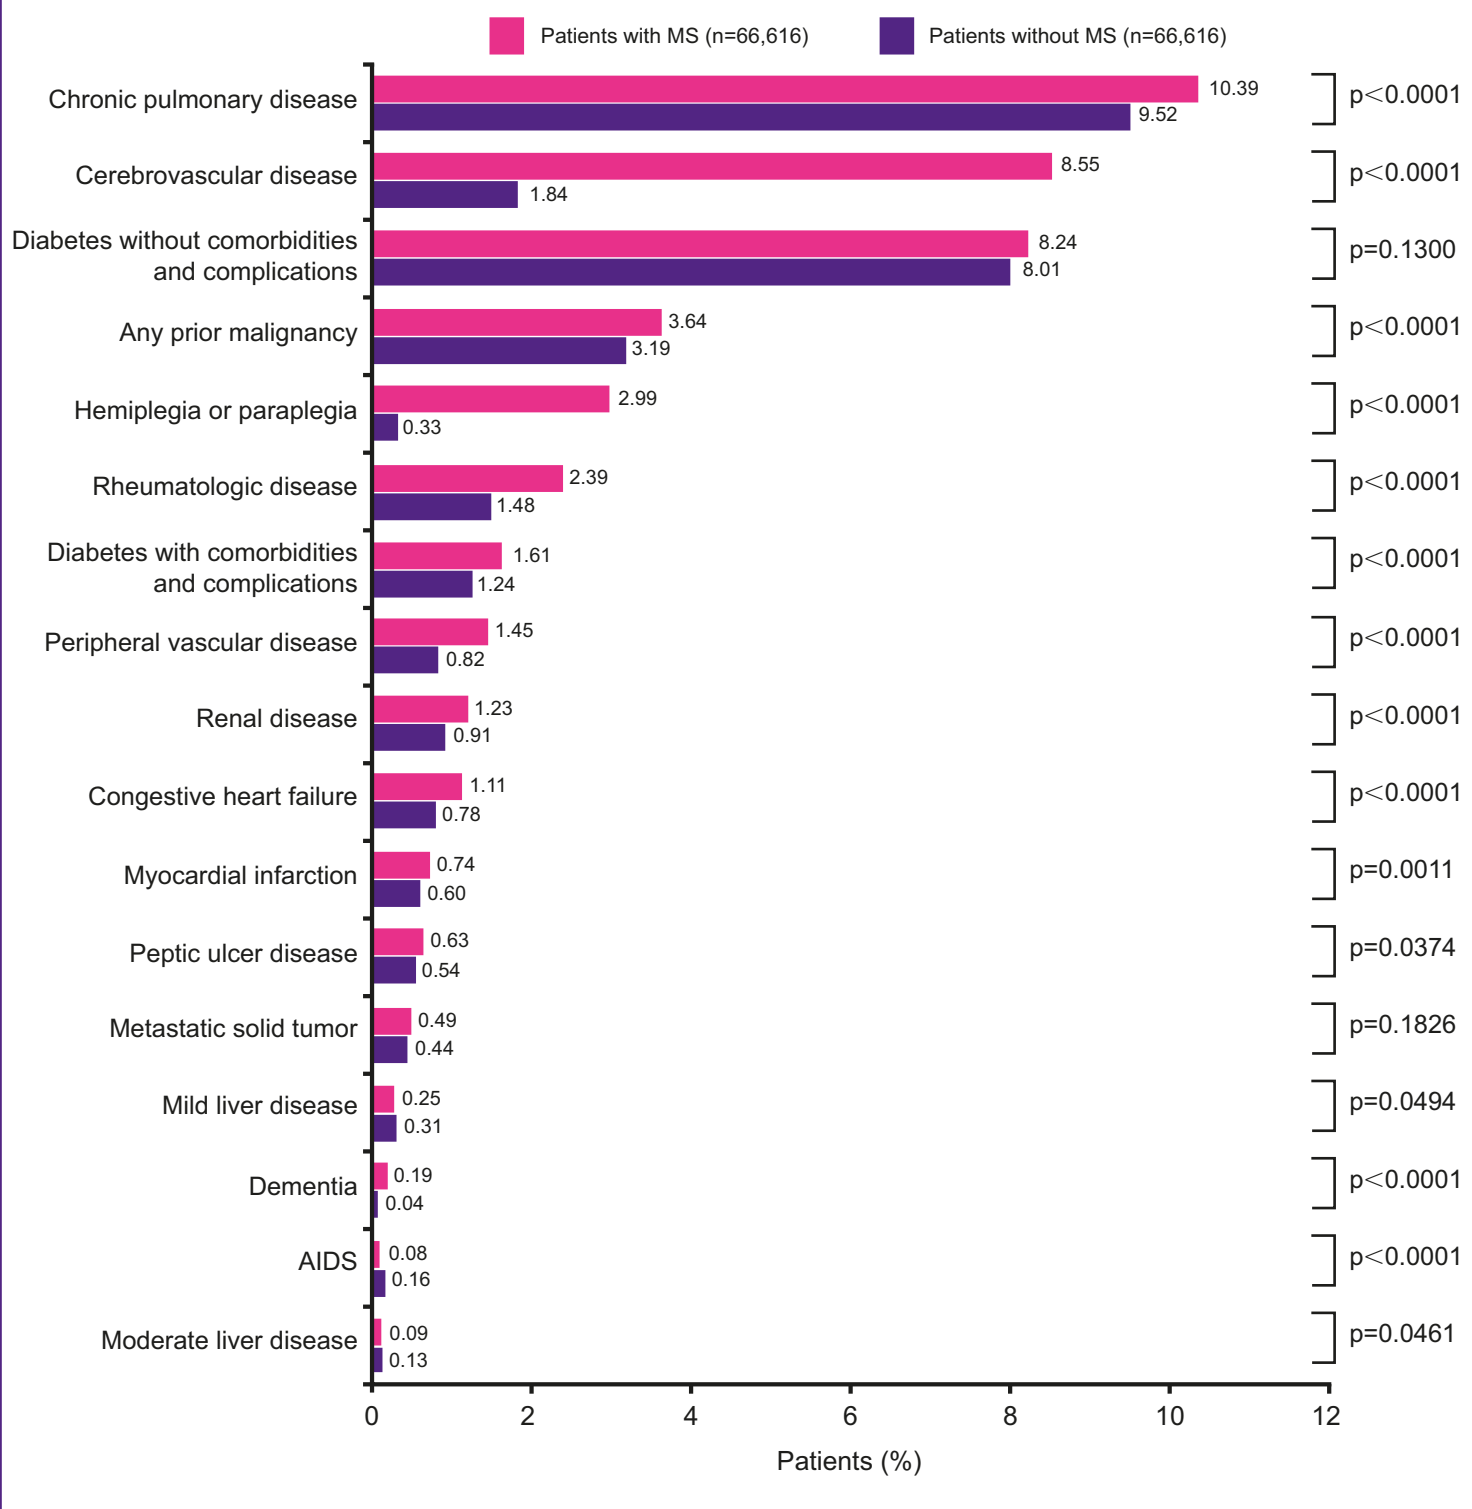
Patients with MS (n=66,616)
Patients without MS (n=66,616)
Chronic pulmonary disease 10.39 9.52 p<0.0001
Cerebrovascular disease 8.55 1.84 p<0.0001
Diabetes without comorbidities and complications 8.24 8.01 p=0.1300
Any prior malignancy 3.64 3.19 p<0.0001
Hemiplegia or paraplegia 2.99 0.33 p<0.0001
Rheumatologic disease 2.39 1.48 p<0.0001
Diabetes with comorbidities and complications 1.61 1.24 p<0.0001
Peripheral vascular disease 1.45 0.82 p<0.0001
Renal disease 1.23 0.91 p<0.0001
Congestive heart failure 1.11 0.78 p<0.0001
Myocardial infarction 0.74 0.60 p=0.0011
Peptic ulcer disease 0.63 0.54 p=0.0374
Metastatic solid tumor 0.49 0.44 p=0.1826
Mild liver disease 0.25 0.31 p=0.0494
Dementia 0.19 0.04 p<0.0001
AIDS 0.08 0.16 p<0.0001
Moderate liver disease 0.09 0.13 p=0.0461
0 2 4 6 8 10 12
Patients (%)

Statistical significance was assessed using pairwise chi-squared tests.
AIDS, acquired immune deficiency syndrome; **CCI**, Charlson Comorbidity Index; **MS**, multiple sclerosis.

REFERENCES

1. Marrie RA, *et al*. *Neurology* 2009;72:117–24.
2. Marrie RA, *et al*. *Neurology* 2010;74:1041–7.
3. Warren SA, *et al*. *Int J MS Care* 2009;11:6–16.
4. Cohen JA. *Arch Neurol* 2009;66:821–8.
5. Marrie RA, *et al*. *Mult Scler* 2015;21:263–81.

ACKNOWLEDGMENTS

The study was sponsored by EMD Serono Inc.,* Rockland, MA, USA. Medical writing and editorial assistance in the preparation of this poster was provided by Laura Yee, Caudex, New York, NY, USA and funded by EMD Serono, Inc.,* Rockland, MA, USA.

DISCLOSURES

KK-R has received consulting fees from Biogen and EMD Serono, Inc.*; speakers fees and honoraria from Biogen, EMD Serono, Inc.,* Genzyme, Mallinckrodt, Novartis, and Teva; and grant/research support from Biogen, Genentech, Genzyme, Mallinckrodt, and Novartis.

NCE is an employee of Health Services Consulting Corporation. Health Services Consulting Corporation received funding from EMD Serono, Inc.,* for this study.

GS and **KS** are employees of Boston Health Economics, Inc. (BHE). BHE received consulting fees from EMD Serono, Inc.*

ALP is an employee of EMD Serono, Inc.,* Rockland, MA, USA.

The authors received no funding for their participation in the development of this poster.

*A business of Merck KGaA, Darmstadt, Germany.

Presented at the 31st Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC); May 24–27, 2017; New Orleans, LA, USA