Short- and long-term predictors of relapses or disability worsening in patients with multiple sclerosis in the phase 3 FREEDOMS and FREEDOMS II studies

Aaron Boster,[1] Pavle Repovic,[2] Shannon Ritter,[3] Davorka Tomic,[4] Xiangyi Meng,[3] Daniela Piani Meier,[4] Fernanda Boulos,[3] Frederik Barkhof,[5] Till Sprenger[6]

[1]OhioHealth Neurological Physicians, Columbus, OH, United States; [2]Multiple Sclerosis Center, Swedish Neuroscience Institute, Seattle, WA, United States; [3]Novartis Pharmaceuticals Corporation, East Hanover, NJ, United States; [4]Novartis Pharma AG, Basel, Switzerland; [5]Department of Radiology and Nuclear Medicine, VU University Medical Center, Amsterdam, Netherlands; [6]Medical Image Analysis Center and Department of Neurology, University Hospital Basel, Basel, Switzerland and Department of Neurology, DKD HELIOS Klinik, Wiesbaden, Germany

DX55

Introduction

- Evidence suggests that long-term clinical outcomes in multiple sclerosis (MS) may be predicted by the treatment and disease histories of patients.[1-3]
- Patients with MS at risk of suboptimal response to first-line therapy may need to have their treatment modified to slow the accumulation of inflammation and neurodegeneration and to maximize therapeutic benefit in the long term.[3]
- Several studies investigated the value of measuring inflammatory magnetic resonance imaging (MRI) disease activity or clinical parameters or different combinations of these parameters early in MS to predict clinical outcomes.[1-3]

Objective

- Assess whether parameters at baseline and during the first year of the 2-year FREEDOMS[4] and FREEDOMS II[5] trials combined predict relapses, an Expanded Disability Status Scale (EDSS) score >6, or 6-month confirmed disability progression (6MCDP) in the short term and long term during the trial extensions.

Methods

Study design

- FREEDOMS and FREEDOMS II were 2-year, double-blind, randomized, phase 3 studies of fingolimod versus placebo.[4,5]
 - Enrolled patients were randomized 1:1:1 to receive fingolimod 0.5 mg or 1.25 mg, or placebo once daily.
- After 2 years, patients randomized to placebo were re-randomized to fingolimod 0.5 mg or 1.25 mg on entering the extension phase;[6-8] patients receiving 1.25 mg were then switched to the 0.5 mg dose.
- All patients then received fingolimod 0.5 mg in an ongoing open-label extension[9,10] (**Figure 1**).

Analyses

- This was a *post hoc* analysis of FREEDOMS and FREEDOMS II and their respective extension phases over 48 months (M) of treatment.
- Unadjusted logistic regression assessed which patient variables or disease variables at baseline or between baseline and M12 predicted relapses during M12-24 and M12-48, or predicted disability (EDSS score >6 or 6MCDP) during M12-48.
 - Relapses were defined as those accompanied by an increase in EDSS score of ≥0.5 points, or an increase of 1 point in two different functional systems (FS), or of 2 points in one FS (excluding bowel/bladder or cerebral FS).
 - 6MCDP was defined as an increase in EDSS score from baseline confirmed at 6M of: 1.5 points if baseline score was 0; 1.0 point if baseline score was 1.0-5.0; or 0.5 points if baseline score was ≥5.5.

Figure 1. Study design and analysis summary

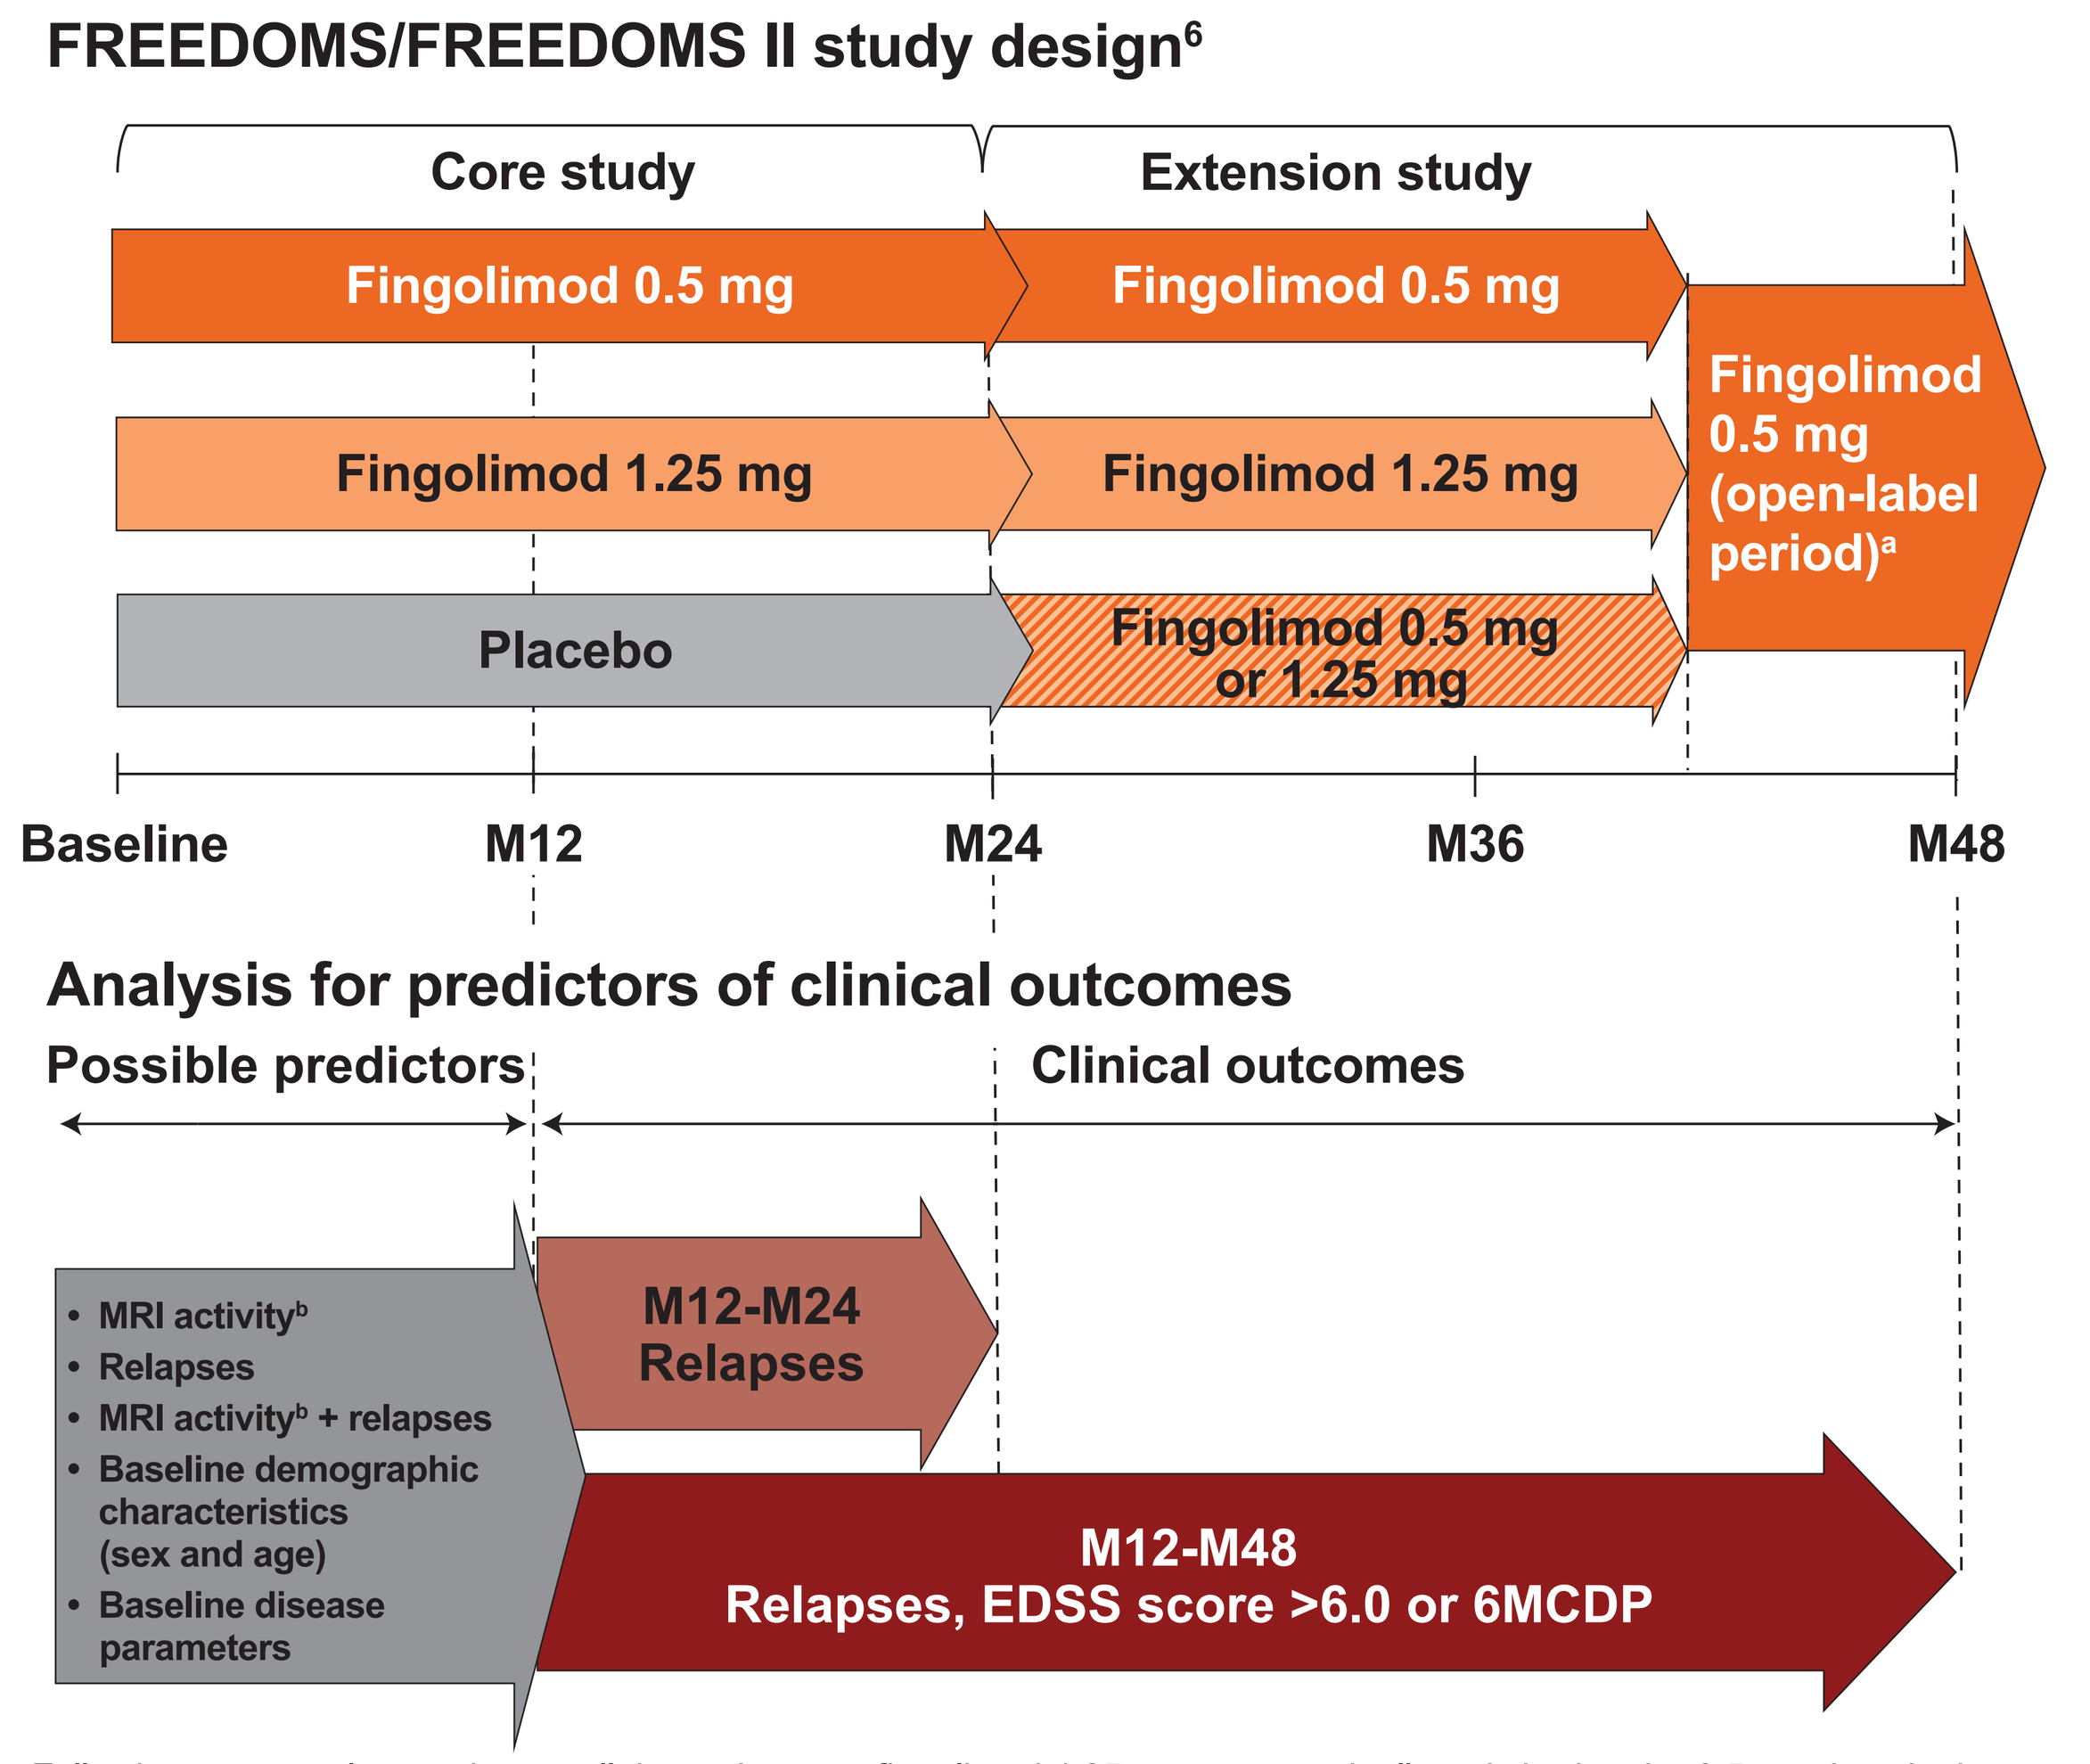

FREEDOMS/FREEDOMS II study design[6]
Core study
Extension study
Fingolimod 0.5 mg
Fingolimod 0.5 mg
Fingolimod 1.25 mg
Fingolimod 1.25 mg
Placebo
Fingolimod 0.5 mg or 1.25 mg
Fingolimod 0.5 mg (open-label period)[a]
Baseline
M12
M24
M36
M48
Analysis for predictors of clinical outcomes
Possible predictors
Clinical outcomes
MRI activity[b]
Relapses
MRI activity[b] + relapses
Baseline demographic characteristics (sex and age)
Baseline disease parameters
M12-M24 Relapses
M12-M48 Relapses, EDSS score >6.0 or 6MCDP

[a]Following a protocol amendment, all the patients on fingolimod 1.25 mg were gradually switched to the 0.5 mg dose in the extension phase.
[b]MRI lesion activity is defined as ≥1 T1 Gd+ or ≥2 T2 lesions.
Gd+, gadolinium-enhancing.

Figure 2. Odds ratios for prediction of relapse activity during the short term (M12-M24) and long term (M12-M48) of FREEDOMS/FREEDOMS II and the extension phases

Parameter	M12-M24 n	M12-M24 Odds ratio (95% CI)	M12-M24 p-value	M12-M48 n	M12-M48 Odds ratio (95% CI)	M12-M48 p-value
Sex[a]	1971	0.921 (0.700-1.211)	0.5564	1162	0.920 (0.684-1.236)	0.5780
Age at core baseline (years)	1971	0.981 (0.967-0.994)	0.0054	1162	0.991 (0.976-1.006)	0.2461
Duration of MS since diagnosis (years)	1971	1.020 (0.999-1.040)	0.0586	1162	1.028 (1.004-1.052)	0.0208
Previous treatment for MS	1971	1.608 (1.244-2.079)	0.0003	1162	1.627 (1.234-2.145)	0.0006
Number of relapses in 2 years before baseline	1968	1.300 (1.116-1.515)	0.0008	1162	1.312 (1.107-1.555)	0.0018
EDSS score at baseline	1971	1.188 (1.085-1.300)	0.0002	1162	1.316 (1.187-1.459)	<0.0001
Number of Gd+ lesions at baseline	1956	1.031 (1.005-1.059)	0.0218	1151	1.016 (0.978-1.055)	0.4184
T1 hypointense lesion volume at baseline (cm^3)	1955	1.038 (1.001-1.076)	0.0466	1150	1.059 (1.017-1.103)	0.0054
Number of confirmed relapses (M0-M12)	1971	2.475 (2.073-2.954)	<0.0001	1162	3.215 (2.485-4.159)	<0.0001
EDSS score change (M0-M12)	1968	1.209 (1.070-1.366)	0.0023	1162	1.221 (1.054-1.414)	0.0077
T2 lesion volume at baseline (cm^3)	1956	1.022 (1.008-1.036)	0.0024	1151	1.028 (1.012-1.045)	0.0006
New T2 lesions at M12 compared with M0	1962	1.738 (1.362-2.218)	<0.0001	1156	1.538 (1.178-2.006)	0.0015
MRI lesion activity (M0-M12)	1969	1.828 (1.438-2.322)	<0.0001	1161	1.732 (1.328-2.258)	<0.0001
MRI lesion activity and ≥1 confirmed relapse (M0-M12)	1971	3.399 (2.523-4.579)	<0.0001	1162	3.468 (2.370-5.073)	<0.0001

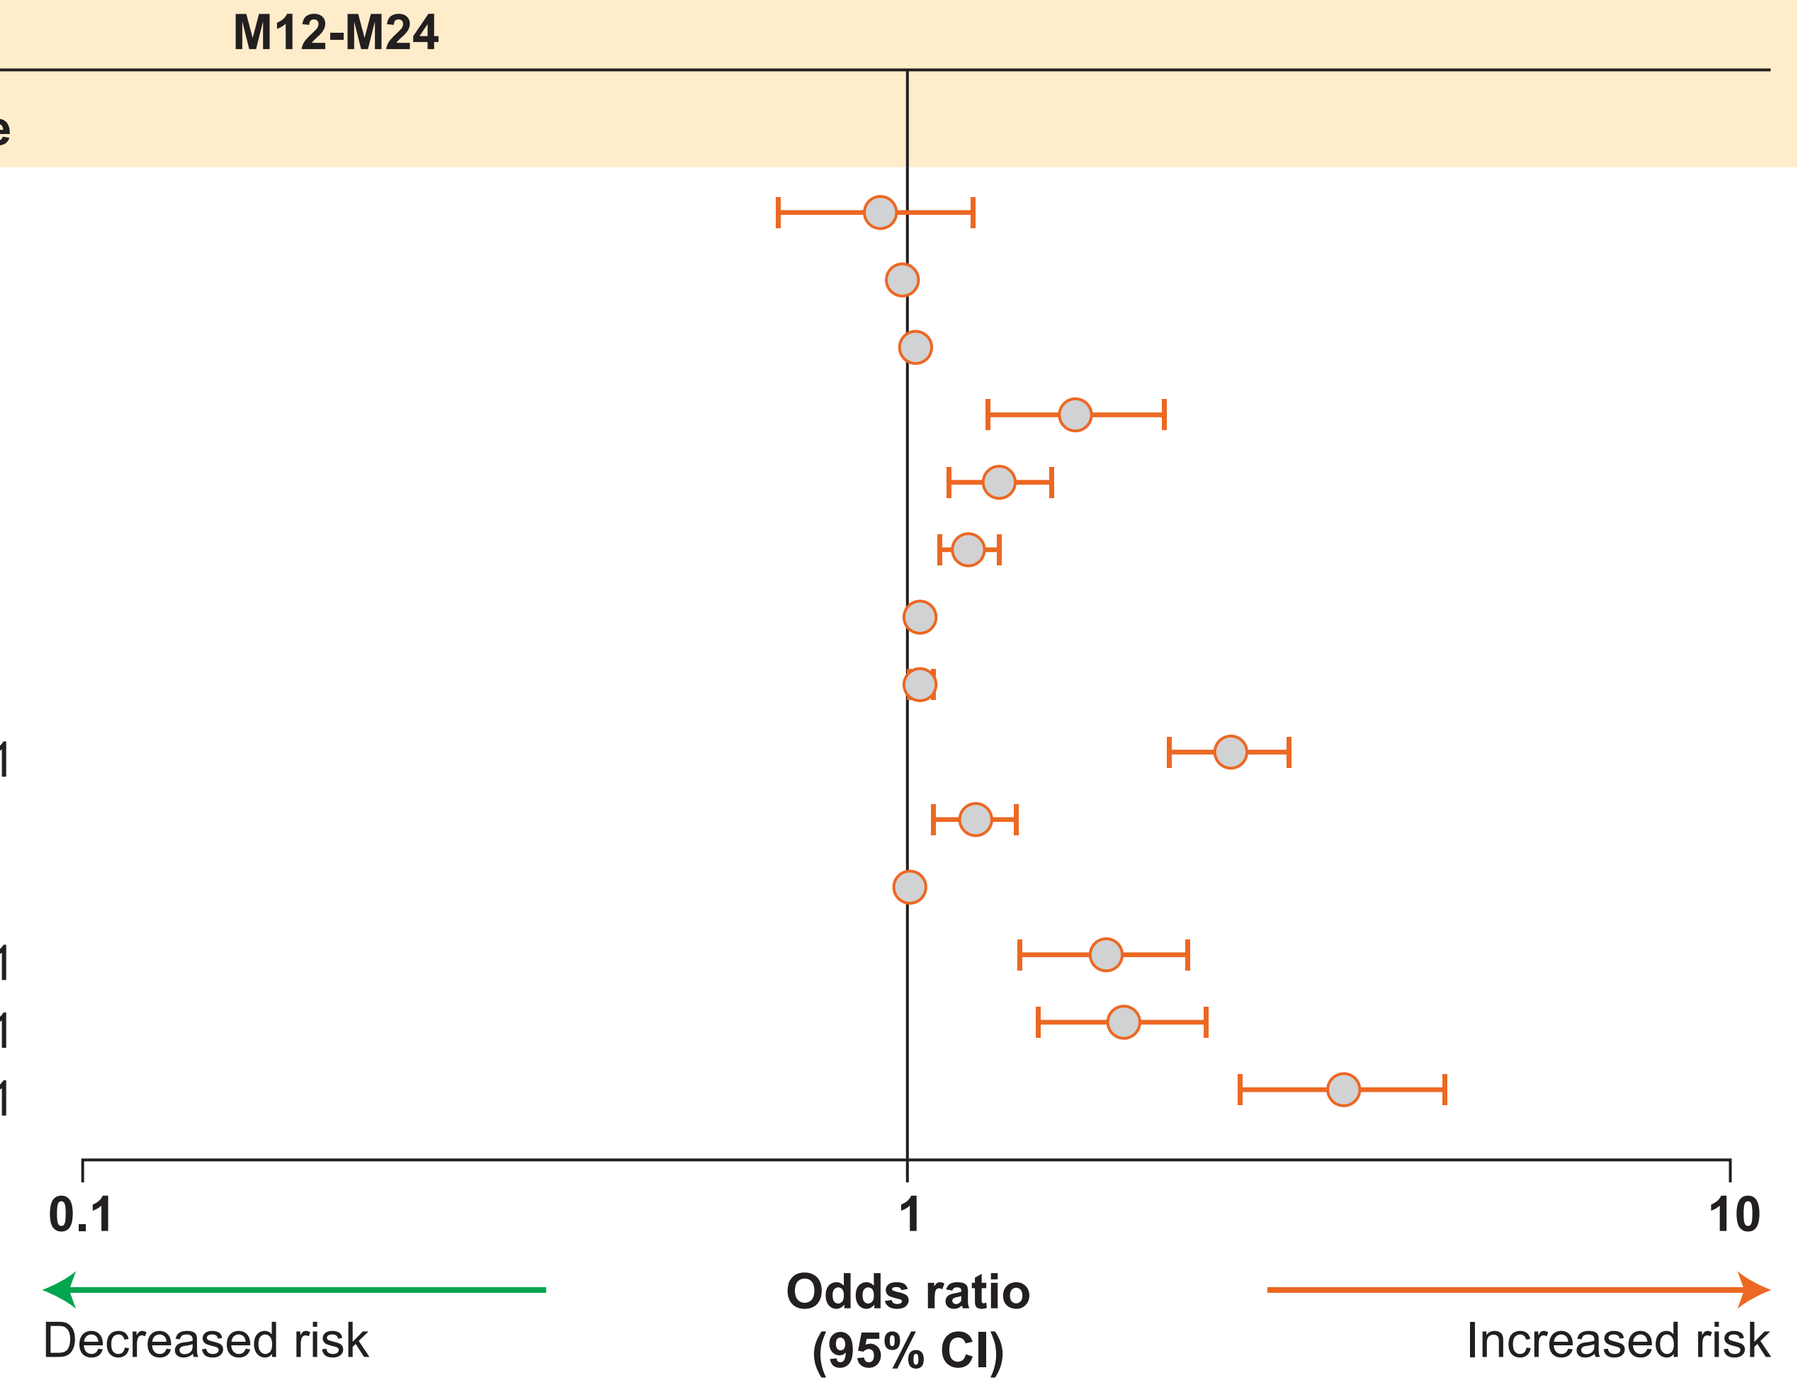

M12-M24
0.1
1
10
Decreased risk
Odds ratio (95% CI)
Increased risk

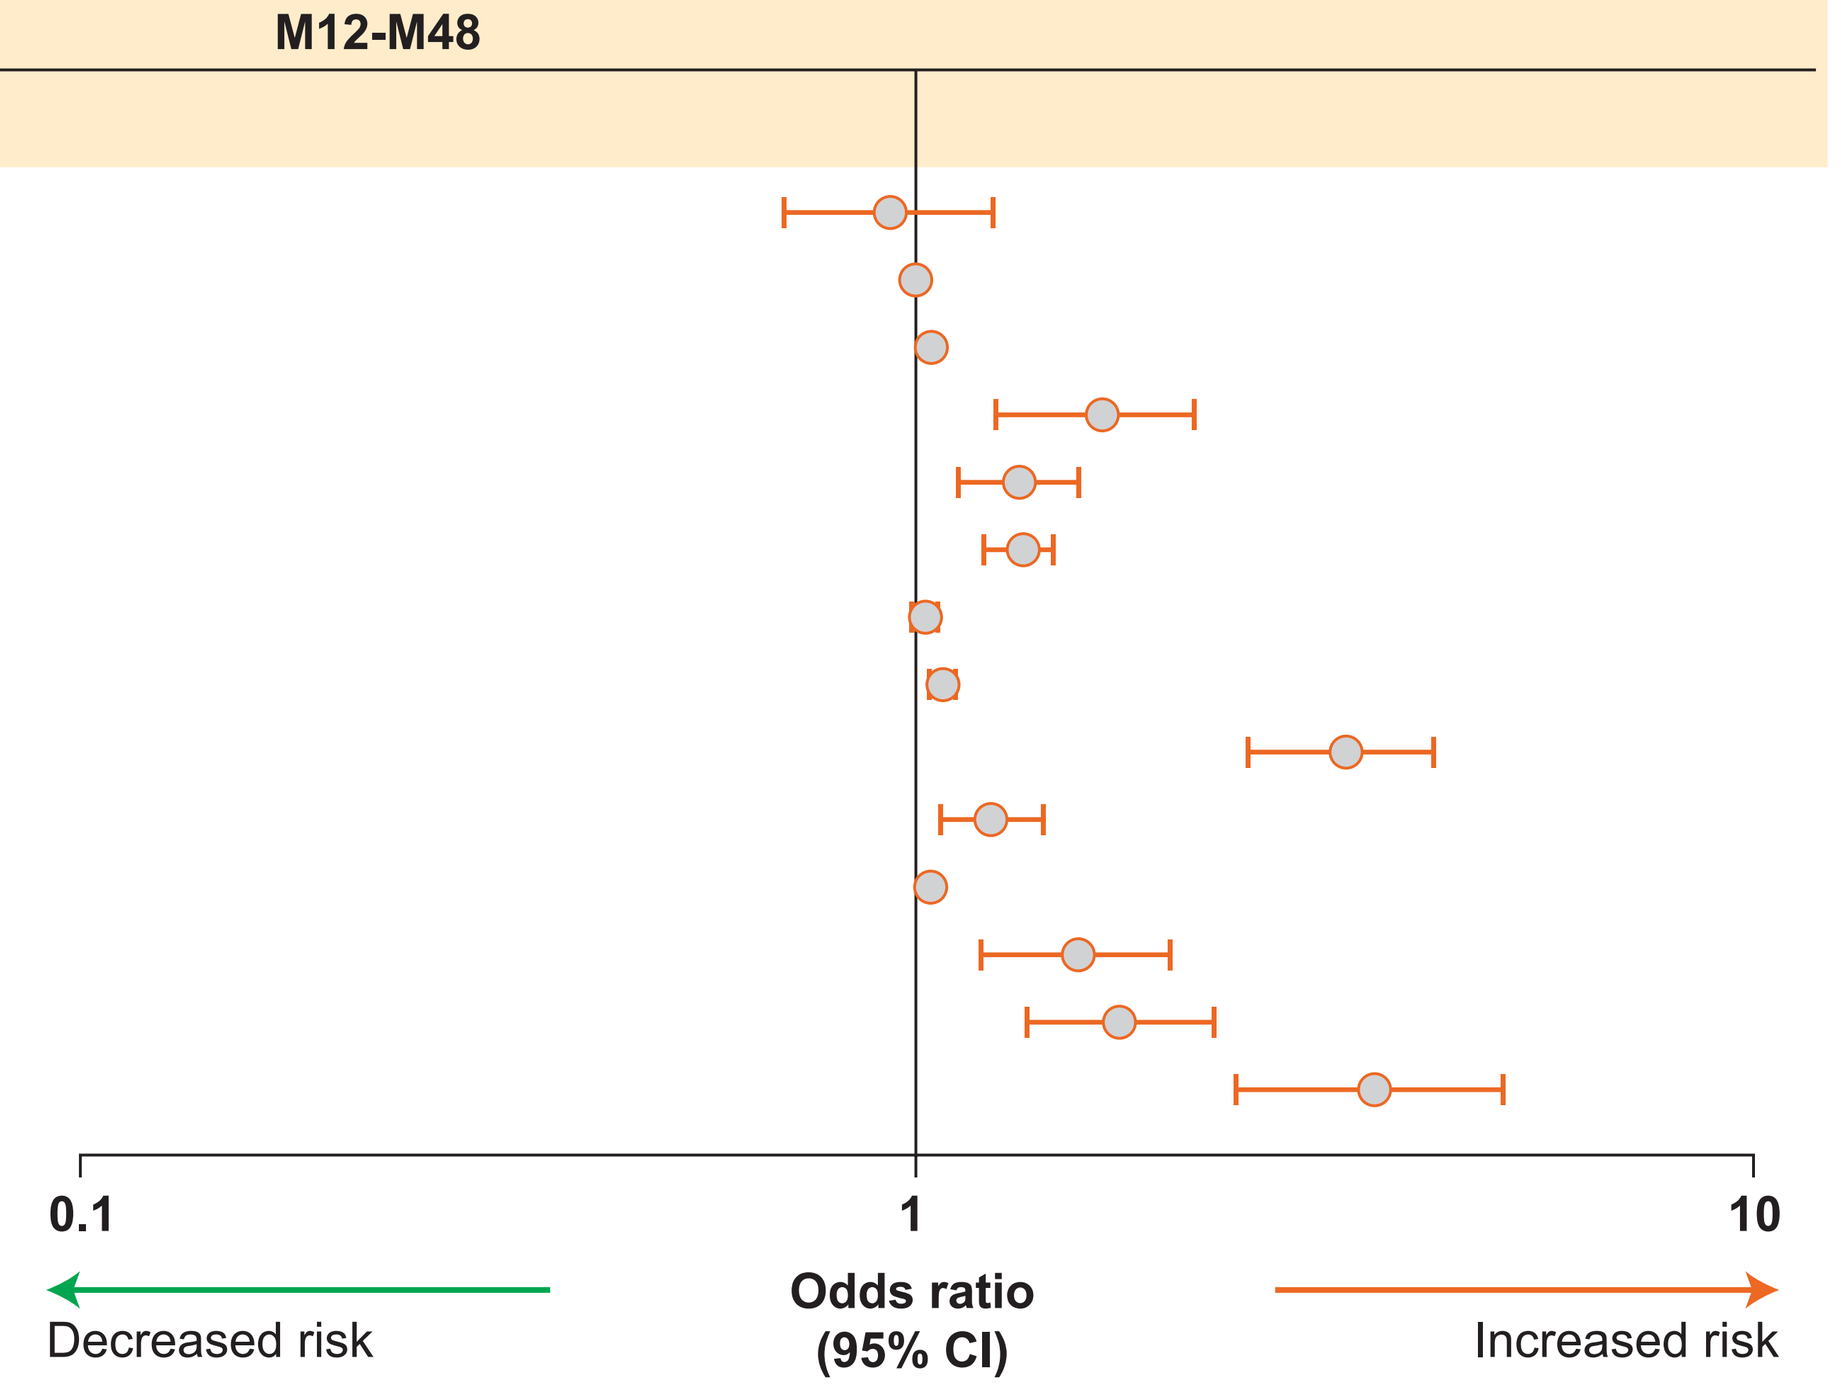

M12-M48
0.1
1
10
Decreased risk
Odds ratio (95% CI)
Increased risk

[a]An odds ratio >1 implies an increased risk in men.
Odds ratios are derived from individual regression models with the respective variable and study as predictors. For continuous variables, the odds ratio corresponds to a unit increase in the explanatory variable. MRI lesion activity is defined as ≥1 T1 Gd+ lesion or ≥2 T2 lesions at any post-baseline assessment up to M12.

Figure 3. Odd ratios for prediction of EDSS score >6 (left panel) and 6-month confirmed disability progression (right panel) in the long term (M12-M48) of FREEDOMS/FREEDOMS II and the extension phases

Parameter	EDSS >6 M12-M48 n	EDSS >6 M12-M48 Odds ratio (95% CI)	EDSS >6 M12-M48 p-value	6MCDP M12-M48 n	6MCDP M12-M48 Odds ratio (95% CI)	6MCDP M12-M48 p-value
Sex[a]	1899	1.345 (0.895-2.022)	0.1541	1162	0.897 (0.627-1.283)	0.5518
Age at core baseline (years)	1899	1.036 (1.013-1.060)	0.0021	1162	1.040 (1.020-1.060)	<0.0001
Duration of MS since diagnosis (years)	1899	1.066 (1.037-1.096)	<0.0001	1162	1.053 (1.026-1.081)	0.0001
Previous treatment for MS	1899	1.936 (1.282-2.923)	0.0017	1162	1.298 (0.934-1.805)	0.1207
Number of relapses in 2 years before baseline	1896	1.245 (0.976-1.590)	0.0782	1162	1.293 (1.056-1.582)	0.0128
EDSS score at baseline	1899	2.688 (2.257-3.200)	<0.0001	1162	1.306 (1.157-1.473)	<0.0001
Number of Gd+ lesions at baseline	1885	0.992 (0.940-1.047)	0.7683	1151	0.988 (0.938-1.040)	0.6440
T1 hypointense lesion volume at baseline (cm^3)	1884	1.116 (1.067-1.166)	<0.0001	1150	1.076 (1.029-1.125)	0.0012
Number of confirmed relapses (M0-M12)	1899	1.849 (1.447-2.361)	<0.0001	1162	1.324 (1.002-1.749)	0.0484
EDSS score change (M0-M12)	1899	1.504 (1.225-1.846)	<0.0001	1162	0.746 (0.623-0.892)	0.0014
T2 lesion volume at baseline (cm^3)	1886	1.044 (1.025-1.064)	<0.0001	1151	1.025 (1.006-1.044)	0.0083
New T2 lesions at M12 compared with M0	1892	0.791 (0.539-1.160)	0.2299	1156	0.970 (0.707-1.331)	0.8497
MRI lesion activity (M0-M12)	1898	0.897 (0.608-1.325)	0.5863	1161	1.014 (0.736-1.397)	0.9339
MRI lesion activity and ≥1 confirmed relapse (M0-M12)	1899	1.560 (0.932-2.612)	0.0904	1162	1.365 (0.854-2.183)	0.1933

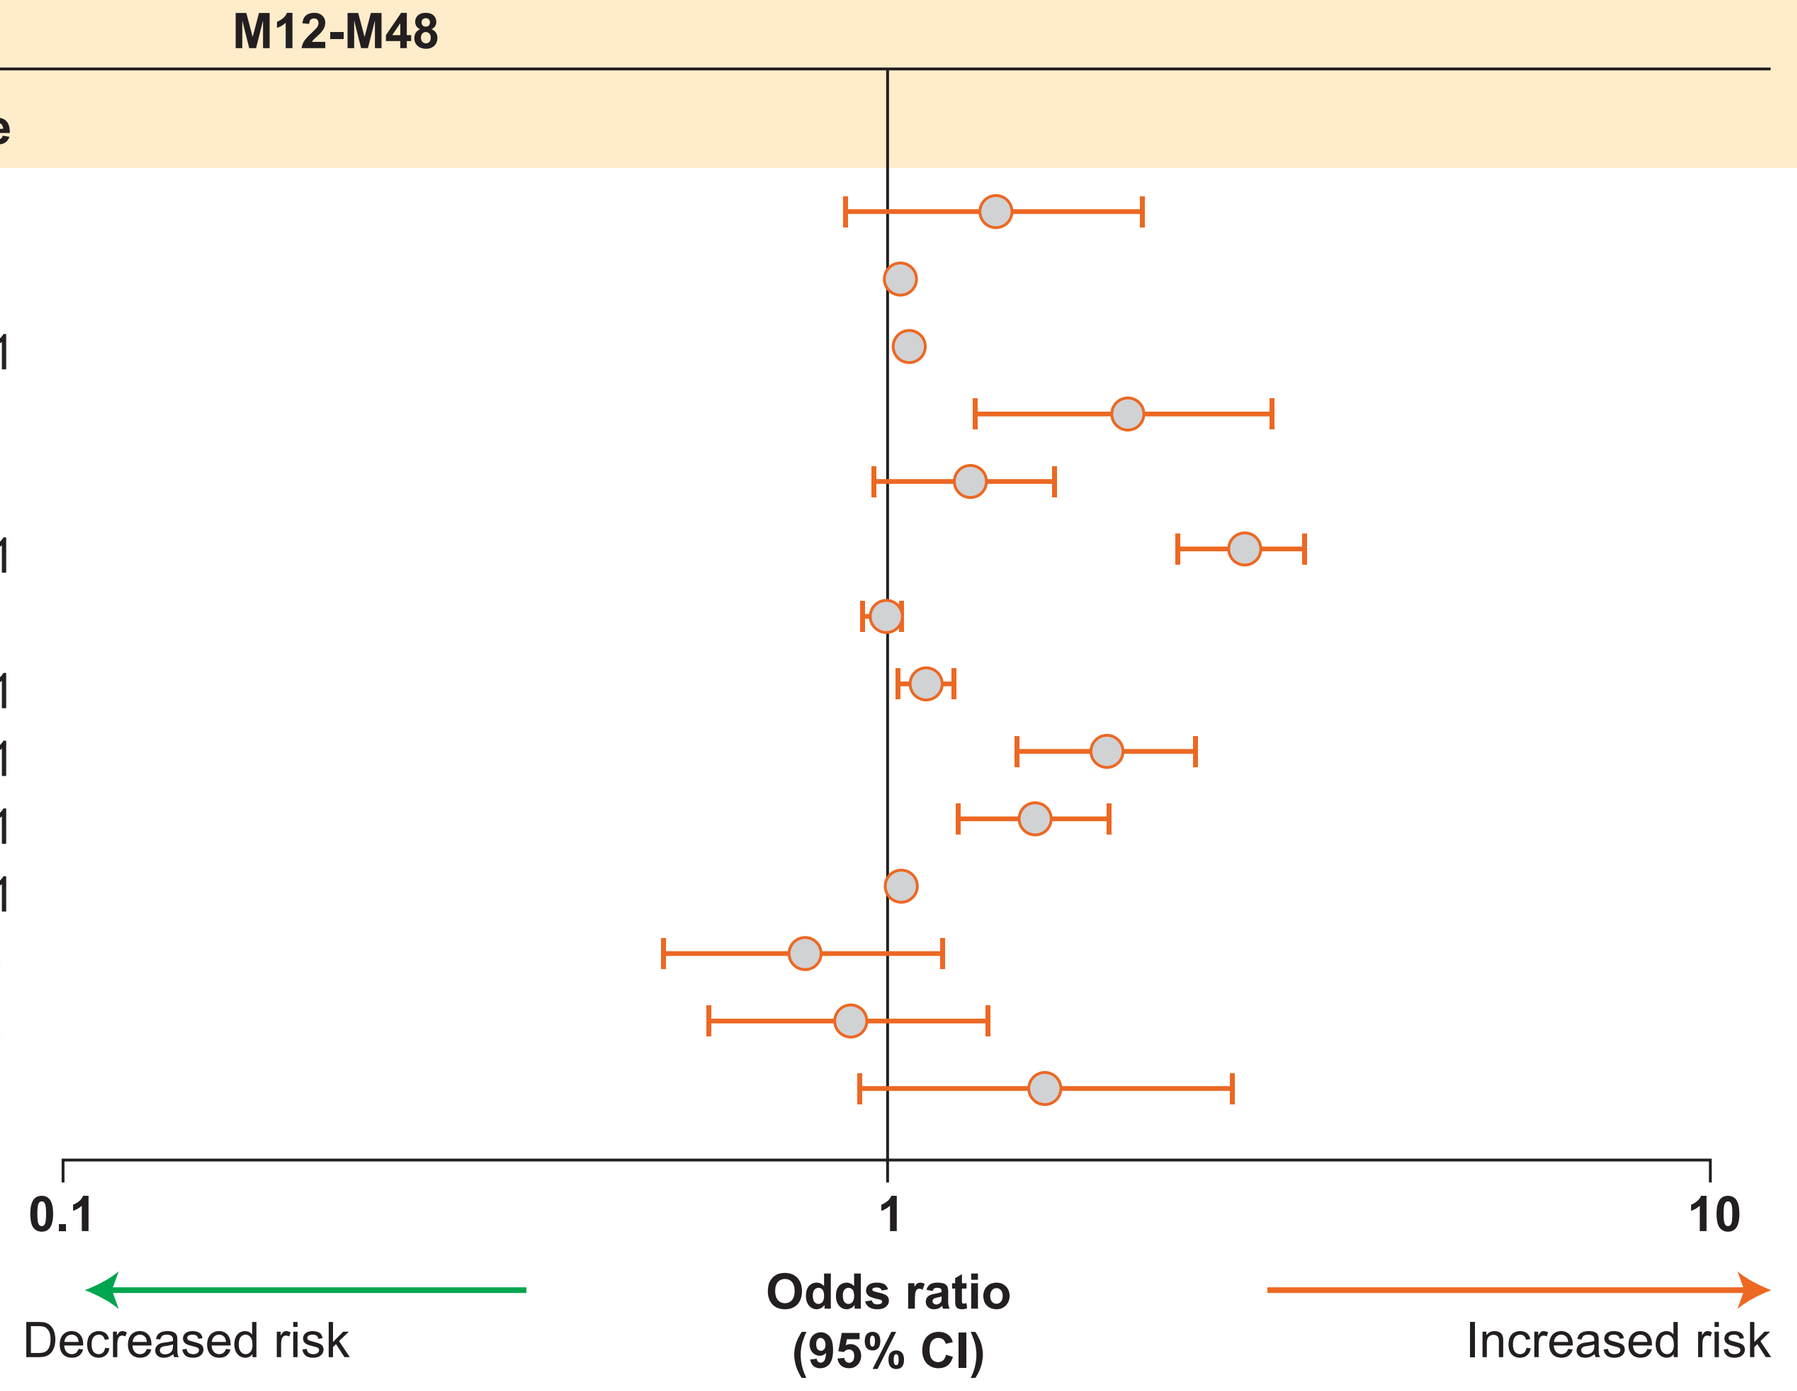

M12-M48
0.1
1
10
Decreased risk
Odds ratio (95% CI)
Increased risk

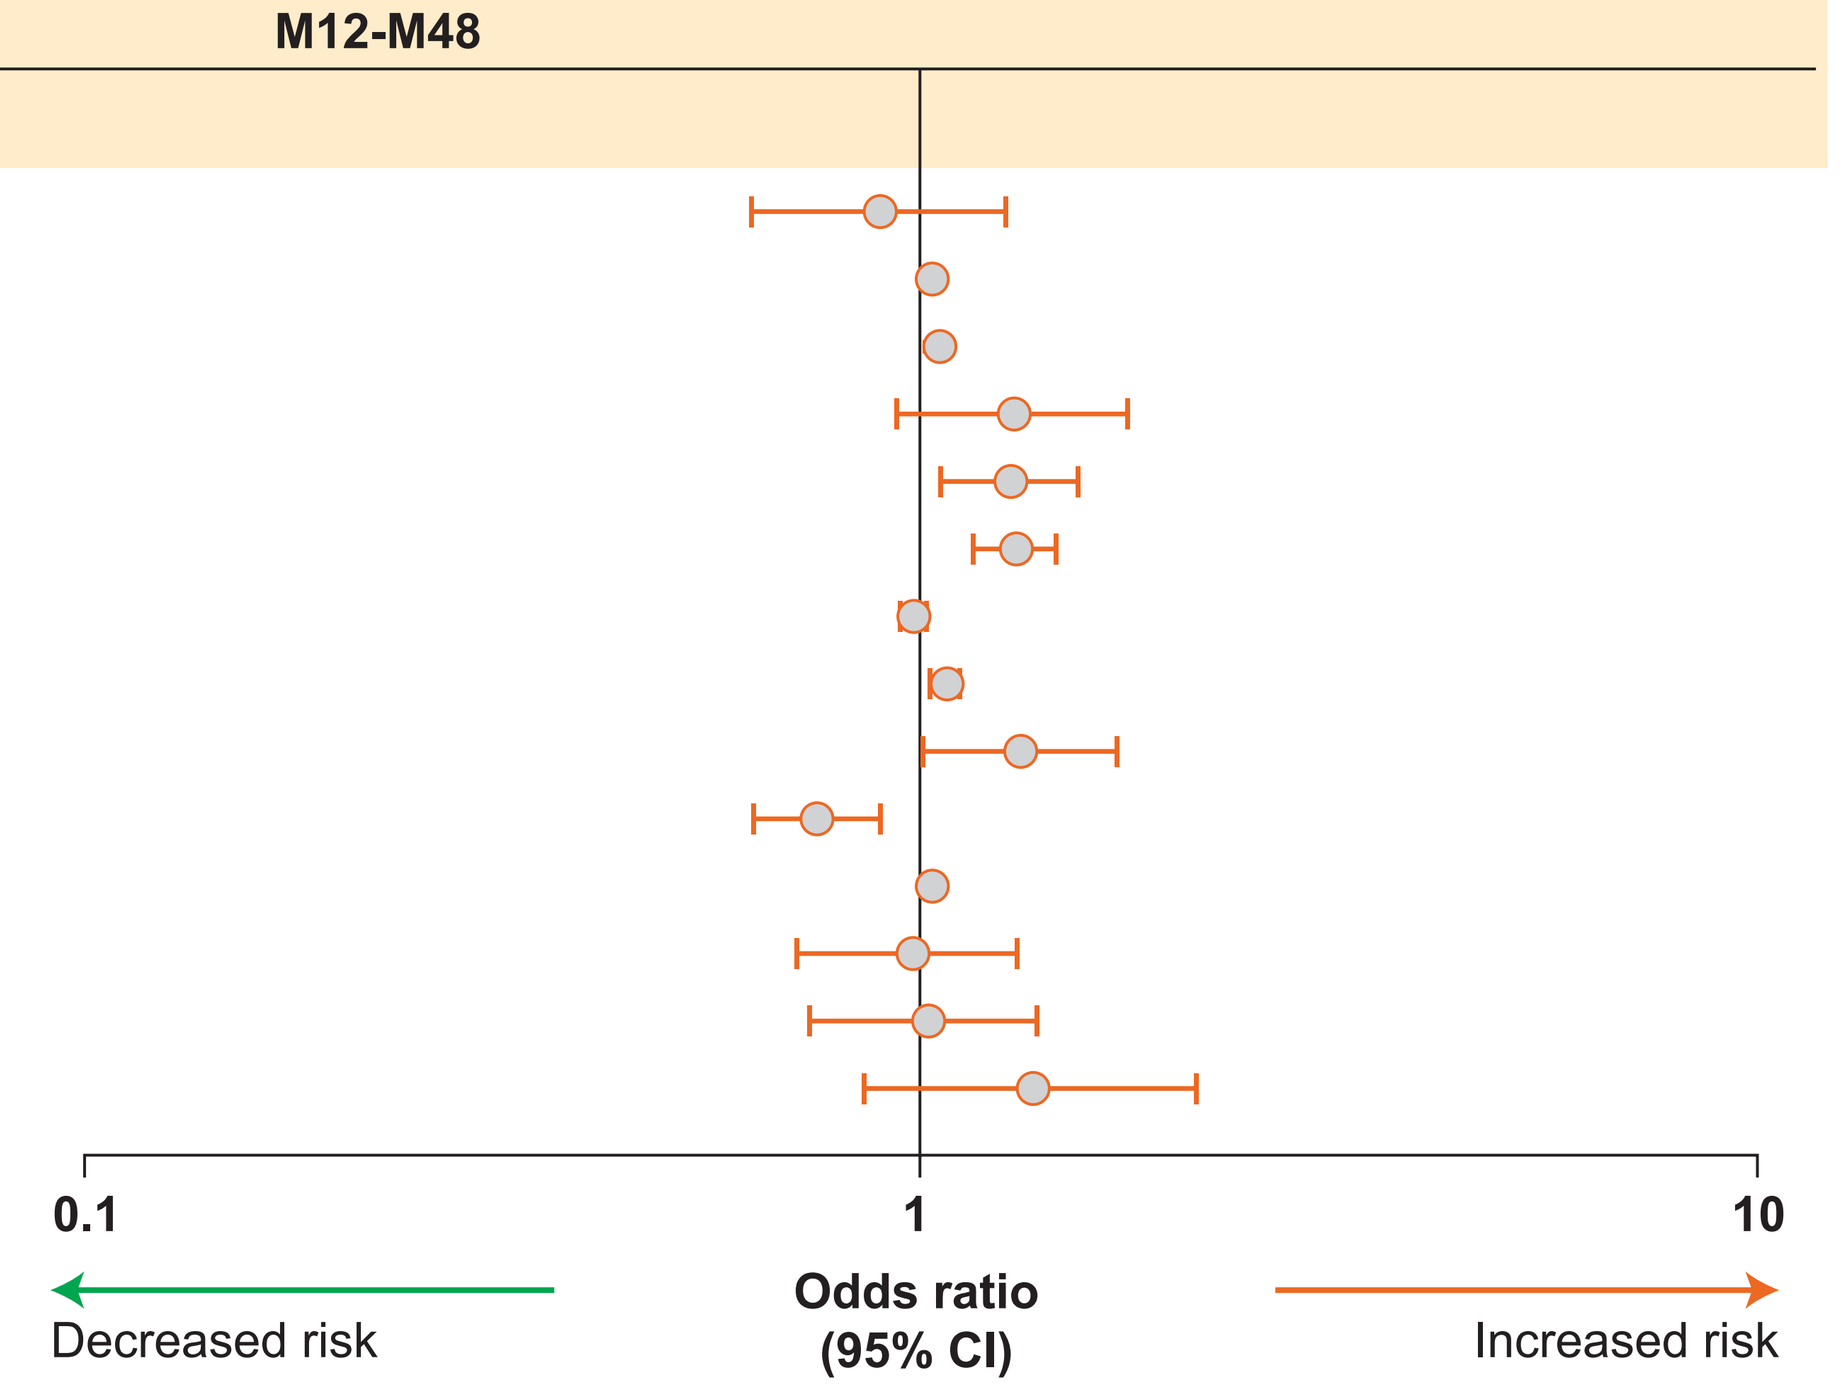

M12-M48
0.1
1
10
Decreased risk
Odds ratio (95% CI)
Increased risk

[a]An odds ratio >1 implies an increased risk in men.
Odds ratios are derived from individual regression models with the respective variable and study as predictors. For continuous variables, the odds ratio corresponds to a unit increase in the explanatory variable. MRI lesion activity is defined as ≥1 T1 Gd+ lesion or ≥2 T2 lesions at any post-baseline assessment up to M12.

- The patient or disease variables used to predict relapses, EDSS score >6 or 6MCDP were:
 - sex, age, duration of MS since diagnosis, previous treatment for MS, number of relapses in the previous 2 years, EDSS score, number of gadolinium-enhancing (Gd+) lesions, T1 hypointense lesion volume and T2 lesion volume (all at baseline)
 - number of confirmed relapses, change in EDSS score, number of new T2 lesions, MRI lesion activity (≥1 T1 Gd+ or ≥2 new T2 lesions),[11] MRI lesion activity and ≥1 confirmed relapse (all from baseline to M12).

Results

- In total, 2355 patients were randomized in FREEDOMS and FREEDOMS II (analysis sets: M12-M24, n=1971; M12-M48, n=1162).

Predictors of relapses

- Strongest predictors of relapses in both the short (M12-M24) and long (M12-M48) term were (**Figure 2**):
 - MRI lesion activity and ≥1 confirmed relapse between baseline and M12
 - number of confirmed relapses between baseline and M12
 - MRI lesion activity between baseline and M12
 - number of new T2 lesions at M12 compared with baseline
 - previous treatment for MS.
- Other predictors of relapses in both timeframes (**Figure 2**):
 - number of relapses in the 2 years before baseline
 - EDSS score at baseline
 - change in EDSS score between baseline and M12.
- Other parameters assessed were associated with a marginal change in relapse risk over these timeframes, or the association was non-significant (**Figure 2**).

Predictors of disability worsening

- Parameters that predicted disability reaching an EDSS score >6 and 6MCDP in the longer term were (**Figure 3**):
 - EDSS score at baseline
 - number of confirmed relapses between baseline and M12
 - T1 hypointense and T2 hyperintense lesion volumes at baseline
 - duration of MS at baseline
 - age at baseline.
- Other predictors of EDSS score >6 in the longer term were (**Figure 3**):
 - previous treatment for MS at baseline
 - EDSS score change between baseline and M12.
- Other predictors of 6MCDP in the longer term were (**Figure 3**):
 - number of confirmed relapses in the 2 years before baseline.

Conclusions

- In the two FREEDOMS trials, combined MRI lesion and relapse activity during year 1 and previous treatment for MS at baseline predicted relapse activity at up to 4 years.
- Longer-term predictors of disability worsening based on an EDSS score >6 or 6MCDP were baseline EDSS score, T1 hypointense lesion and T2 lesion volume, disease duration and age, and post-baseline relapse activity.
- Findings support early use of high-efficacy therapy to help reduce accumulation of irreversible disability in patients with relapsing MS.

References

1. Prosperini L et al. *Eur J Neurol.* 2009;16:1202-1209.
2. Rio J et al. *Mult Scler.* 2009;15:848-853.
3. Dobson R et al. *Neurology* 2014;82:248-254.
4. Kappos L et al. *N Engl J Med.* 2010;362:387-401.
5. Calabresi et al. *Lancet Neurol.* 2014;13:545-556.
6. Kappos L et al. *Neurology* 2015;84:1582-1591.
7. Reder AT et al. *Mult Scler.* 2013;19(Suppl. 11):510-511.
8. Vollmer T et al. *Neurology* 2013;80(Suppl.)P01.165.
9. Kappos L et al. *Mult Scler.* 2013;19(Suppl. 11):486-487.
10. Cohen J et al. *ECTRIMS Online Library* 2015;115507:P591.
11. Prosperini L et al. *Mult Scler.* 2014;20:566-576.

Acknowledgments

Editorial support was provided by Oxford PharmaGenesis, Oxford, UK, and was funded by Novartis Pharmaceuticals Corporation.

Disclosures

Aaron Boster: Acorda Therapeutics, Actelion Pharmaceuticals, Biogen Idec, CNS Therapeutics, Genzyme, Jazz Pharmaceuticals, Medtronic, Novartis Pharmaceuticals Corporation, Roche, Serono, Questcor Pharmaceuticals and Teva Pharmaceuticals. **Pavle Repovic:** Acorda Therapeutics, Biogen Idec, EMD Serono, Genzyme, Novartis Pharmaceuticals Corporation, Pfizer, Teva Pharmaceuticals (multiple sources). **Shannon Ritter, Xiangyi Meng, Fernanda Boulos:** Novartis Pharmaceuticals Corporation (employees). **Davorka Tomic, Daniela Piani Meier:** Novartis Pharma AG (employees). **Frederik Barkhof:** Biogen Idec, Bayer Schering Pharma, Genzyme, Janssen Alzheimer Immunotherapy, Lundbeck, MediciNova, Merck Serono, Novartis Pharmaceuticals Corporation, Roche, Sanofi-Aventis, Synthon (multiple sources). **Till Sprenger:** Actelion Pharmaceuticals, Allergan, ATI, Biogen Idec, ElectroCore, Novartis Pharmaceuticals Corporation, Sanofi Genzyme, Teva Pharmaceuticals.

© 2017 Novartis Pharmaceuticals Corporation.

Poster presented at the 2017 Annual Meeting of the Consortium of Multiple Sclerosis Centers, May 24-27, New Orleans, LA, United States.

This study was sponsored by Novartis Pharmaceuticals Corporation, United States.

Text: Q9b663
To: 8NOVA (86682) US only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe and Russia
+46737494608 Sweden and Europe
Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=9b663

Scan this QR code

Copies of this poster obtained through the QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address:
aaron.boster@ohiohealth.com