Short- and long-term predictors of relapses or disability worsening in patients with multiple sclerosis in the phase 3 TRANSFORMS study

Aaron Boster,[1]
Pavle Repovic,[2] Xiangyi Meng,[3]
Daniela Piani Meier,[4]
Fernanda Boulos,[3] Till Sprenger,[5]
Frederik Barkhof[6]

[1]OhioHealth Neurological Physicians, Columbus, OH, United States; [2]Multiple Sclerosis Center, Swedish Neuroscience Institute, Seattle, WA, United States; [3]Novartis Pharmaceuticals Corporation, East Hanover, NJ, United States; [4]Novartis Pharma AG, Basel, Switzerland; [5]Medical Image Analysis Center and Department of Neurology, University Hospital Basel, Basel, Switzerland and DKD HELIOS Klinik, Wiesbaden, Germany; [6]Department of Radiology and Nuclear Medicine, VU University Medical Center, Amsterdam, Netherlands

DX56

Introduction

- Evidence suggests that long-term clinical outcomes in multiple sclerosis (MS) may be predicted by the treatment and disease histories of patients.[1-3]
- Patients with MS at risk of suboptimal response to first-line therapy may need to have their treatment modified to slow accumulation of inflammation and neurodegeneration and to maximize therapeutic benefit in the long term.[3]
- Several studies investigated the value of measuring inflammatory magnetic resonance imaging (MRI) disease activity or clinical parameters or combinations of these parameters early in MS to predict clinical outcomes.[1-3]

Objective

- Assess whether parameters at baseline and during the 1-year TRANSFORMS study[4] predict relapses, an Expanded Disability Status Scale (EDSS) score >6, or 6-month confirmed disability progression (6MCDP) in the short term and long term during the trial's extension phases.

Methods

Study design

- TRANSFORMS was a 1-year, double-blind, randomized, phase 3 study of fingolimod compared with intramuscular (IM) interferon (IFN) β-1a.[4]
 - Enrolled patients with relapsing MS were randomized 1:1:1 to receive fingolimod 0.5 mg or 1.25 mg once daily, or IFNβ-1a IM 30 μg once weekly.
- At 1 year, patients randomized to IFNβ-1a IM were re-randomized to fingolimod 0.5 mg or 1.25 mg on entering a 1-year extension phase.[5,6]
- All patients then received fingolimod 0.5 mg in an ongoing open-label extension[7,8] (**Figure 1**).

Analyses

- This was a *post hoc* analysis of TRANSFORMS and its extension over 48 months (M).
- Unadjusted logistic regression analyses assessed which patient variables or disease variables at baseline or between baseline and M12 predicted relapses during M12-24 and M12-48, or predicted disability (EDSS score >6 or 6MCDP) during M12-48.
 - Relapses were confirmed when accompanied by an increase in EDSS score of ≥0.5 points, or an increase of 1 point in two different functional systems (FS), or of 2 points in one FS (excluding bowel/bladder or cerebral FS).
 - 6MCDP was defined as an increase in EDSS score from baseline confirmed at 6M of: 1.5 points if baseline score was 0; 1.0 point if baseline score was 1.0-5.0; or 0.5 points if baseline score was ≥5.5.

Figure 1. Study design and analysis summary

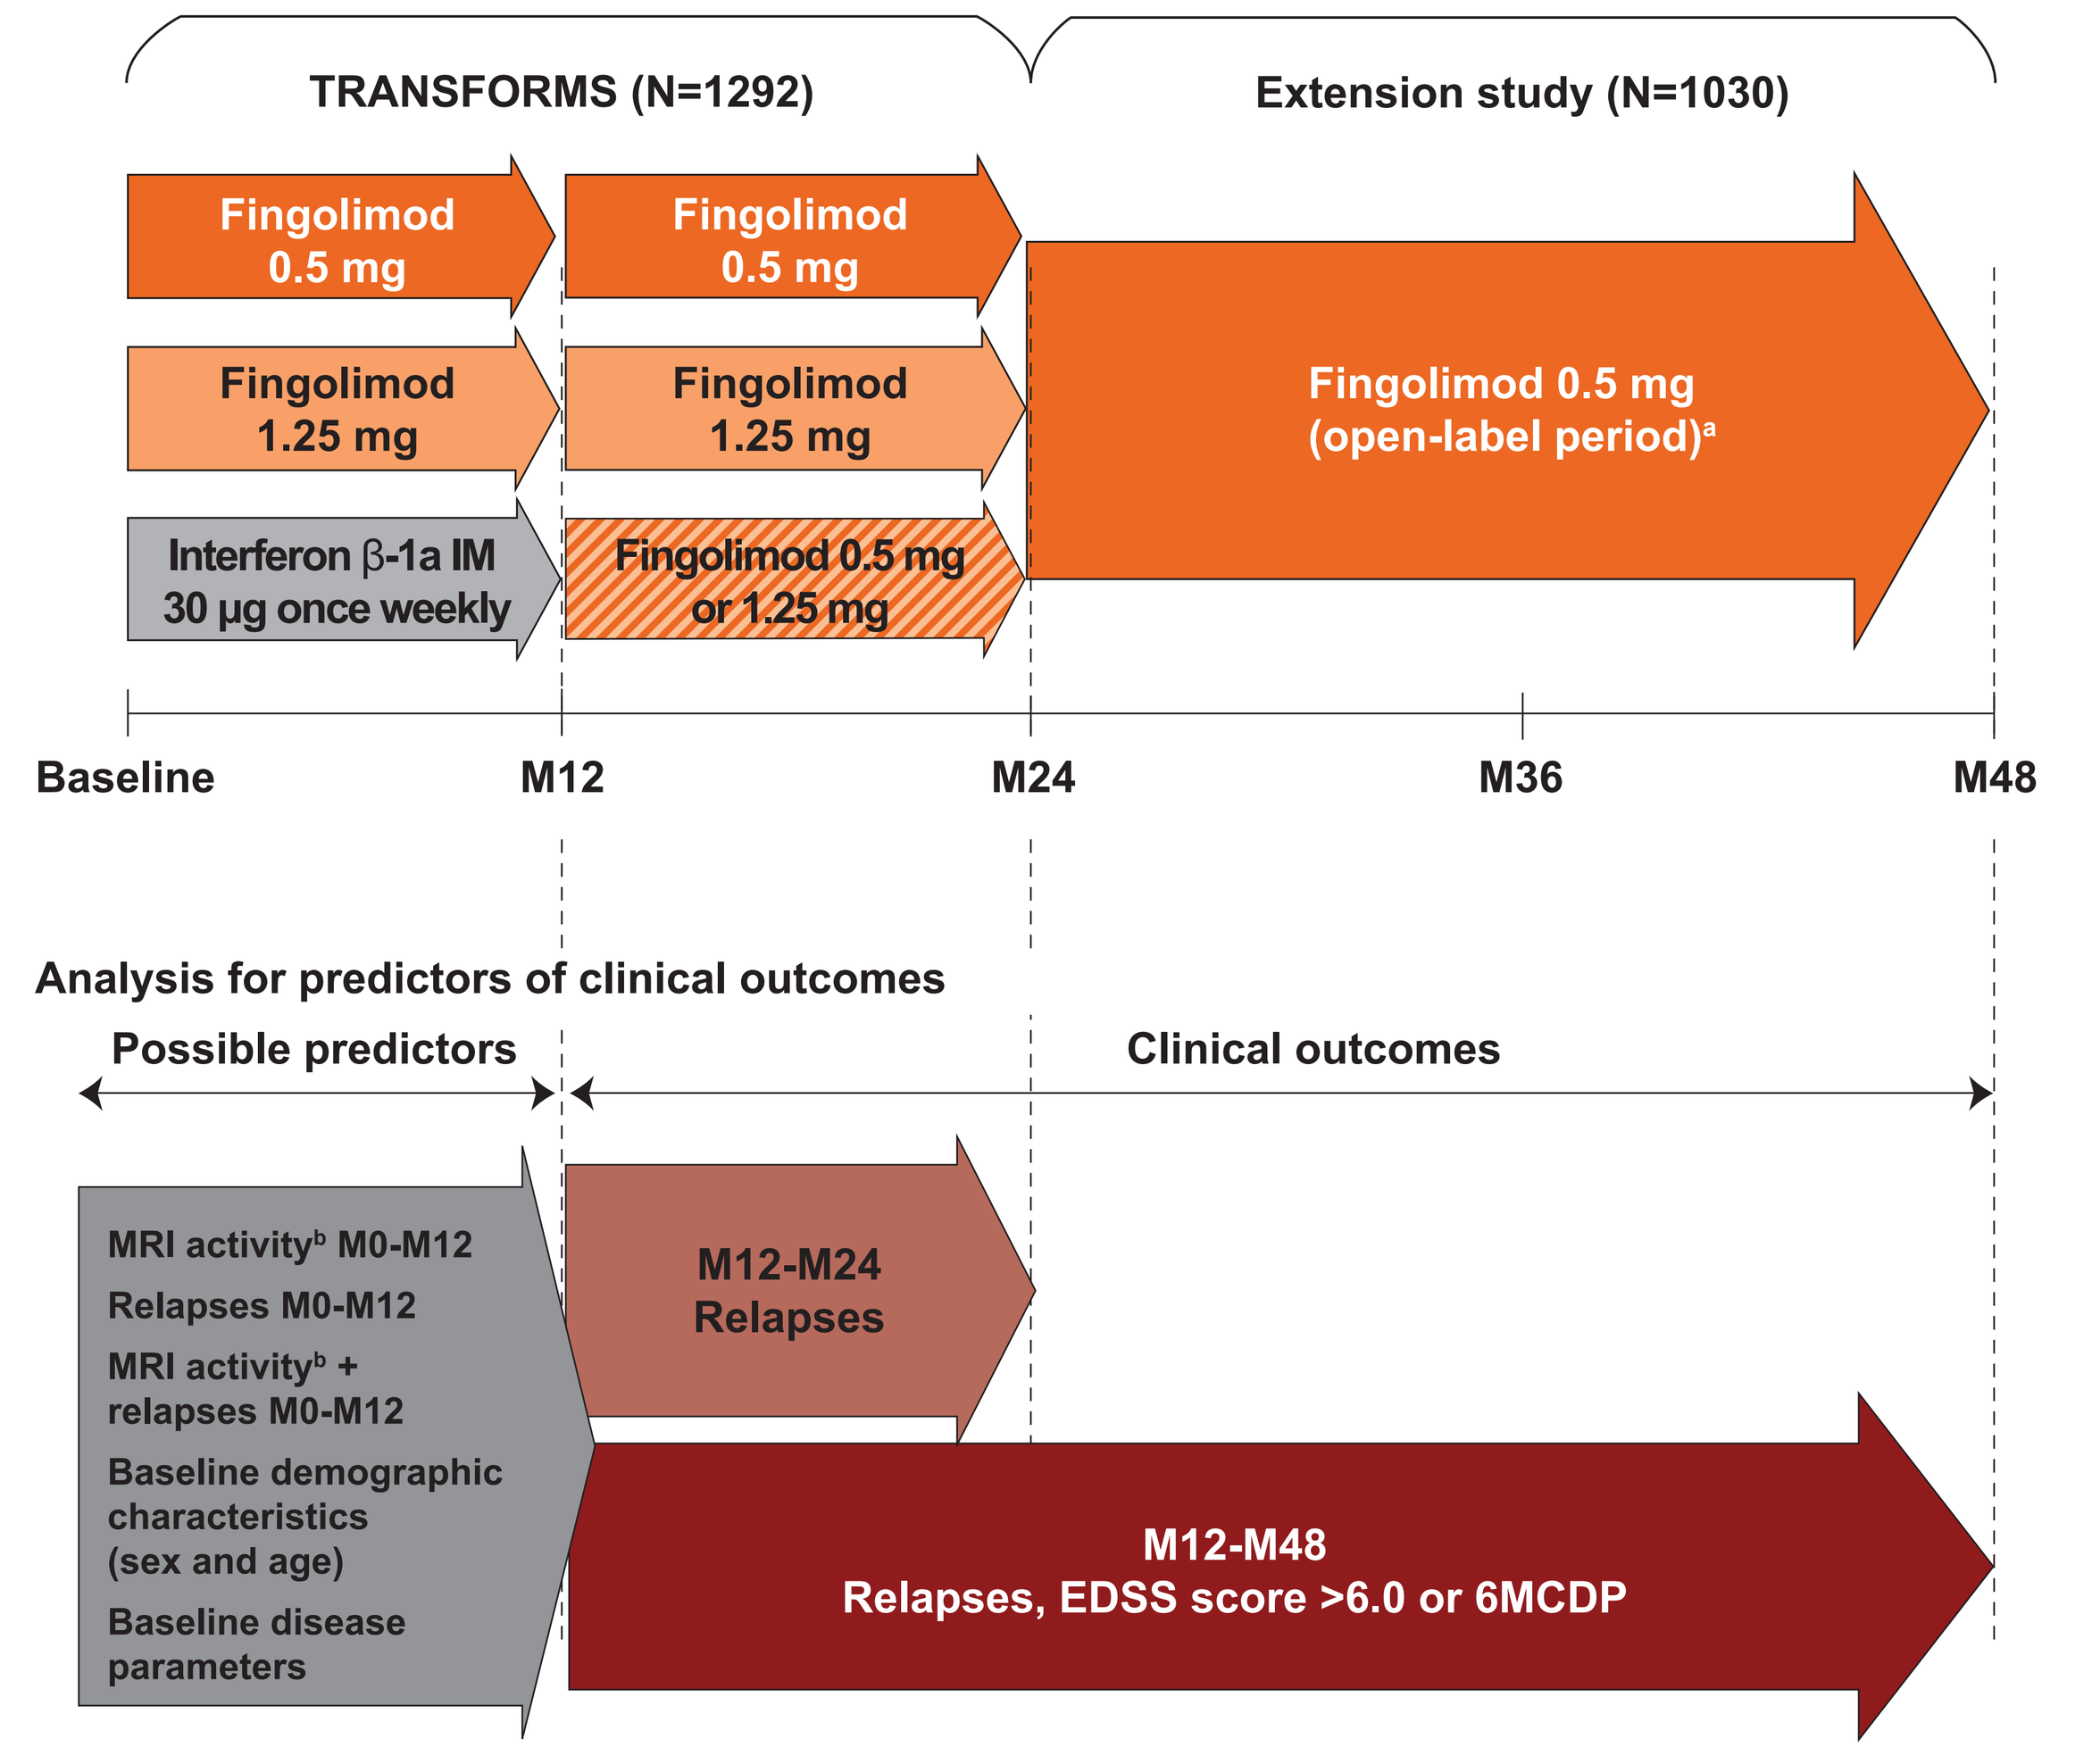
TRANSFORMS (N=1292)
Extension study (N=1030)
Fingolimod 0.5 mg
Fingolimod 0.5 mg
Fingolimod 1.25 mg
Fingolimod 1.25 mg
Interferon β-1a IM 30 μg once weekly
Fingolimod 0.5 mg or 1.25 mg
Fingolimod 0.5 mg (open-label period)a
Baseline
M12
M24
M36
M48
Analysis for predictors of clinical outcomes
Possible predictors
Clinical outcomes
MRI activityb M0-M12
Relapses M0-M12
MRI activityb + relapses M0-M12
Baseline demographic characteristics (sex and age)
Baseline disease parameters
M12-M24 Relapses
M12-M48 Relapses, EDSS score >6.0 or 6MCDP

[a]Following a protocol amendment, and depending on the terms of approval in different countries, all patients receiving fingolimod 1.25 mg/day were switched to fingolimod 0.5 mg/day; the timing of this switch varied among patients.
[b]MRI activity is defined as ≥1 T1 Gd+ or ≥2 T2 lesions.
Gd+, gadolinium-enhancing.

Figure 2. Odds ratios for prediction of relapse activity during the short term (M12-M24) and long term (M12-M48) of TRANSFORMS and its extension phases

Parameter	M12-M24 n	M12-M24 Odds ratio (95% CI)	M12-M24 p-value	M12-M48 n	M12-M48 Odds ratio (95% CI)	M12-M48 p-value
Sex[a]	973	0.895 (0.602-1.330)	0.5832	778	0.851 (0.597-1.213)	0.3719
Age at core baseline (years)	973	0.996 (0.975-1.018)	0.7341	778	0.989 (0.970-1.009)	0.2859
Duration of MS since diagnosis (years)	973	1.022 (0.987-1.057)	0.2176	778	1.038 (1.006-1.071)	0.0189
Previous treatment for MS	973	1.382 (0.944-2.022)	0.0958	778	1.791 (1.269-2.528)	0.0009
Number of relapses in 2 years before baseline	972	1.428 (1.109-1.839)	0.0058	778	1.325 (1.060-1.656)	0.0134
EDSS score at baseline	973	1.237 (1.075-1.423)	0.0030	778	1.183 (1.040-1.346)	0.0104
Number of Gd+ lesions at baseline	958	1.031 (0.992-1.073)	0.1239	767	1.042 (1.001-1.085)	0.0471
T1 hypointense lesion volume at baseline (cm³)	964	1.002 (0.934-1.074)	0.9632	771	1.028 (0.971-1.089)	0.3472
Number of confirmed relapses (M0-M12)	973	2.240 (1.751-2.866)	<0.0001	778	2.297 (1.753-3.009)	<0.0001
EDSS score change (M0-M12)	965	1.089 (0.882-1.344)	0.4269	772	0.996 (0.821-1.208)	0.9657
T2 lesion volume at baseline (cm³)	964	1.018 (0.990-1.046)	0.2092	771	1.024 (0.998-1.050)	0.0682
New T2 lesions at M12 compared with M0	965	1.224 (0.843-1.777)	0.2872	774	1.548 (1.108-2.162)	0.0104
MRI lesion activity (M0-M12)	968	1.336 (0.914-1.952)	0.1345	775	1.789 (1.274-2.511)	0.0008
MRI lesion activity and ≥1 confirmed relapse (M0-M12)	973	2.776 (1.649-4.674)	0.0001	778	3.703 (2.158-6.356)	<0.0001

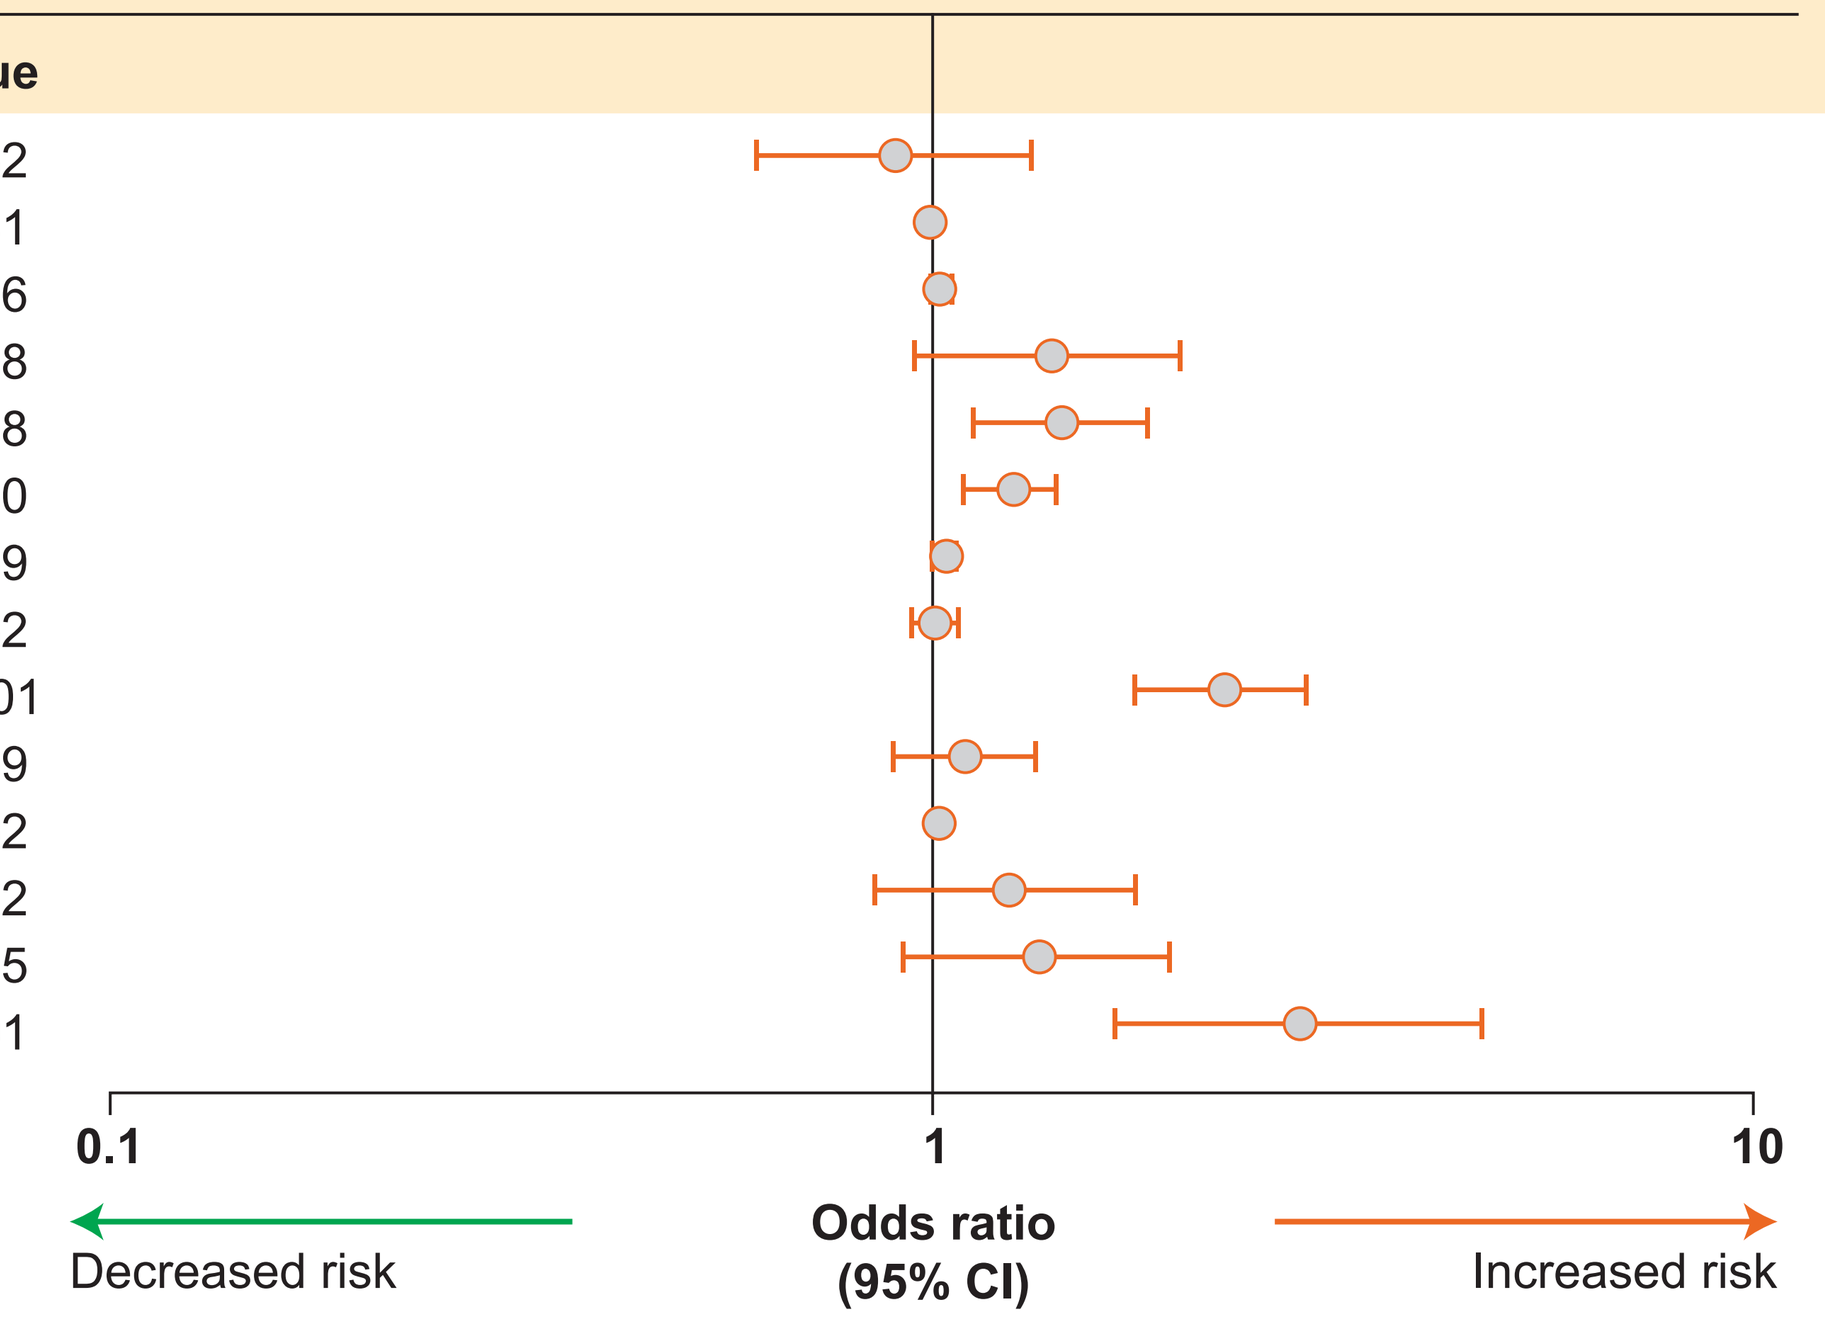
0.1
1
10
Decreased risk
Odds ratio (95% CI)
Increased risk

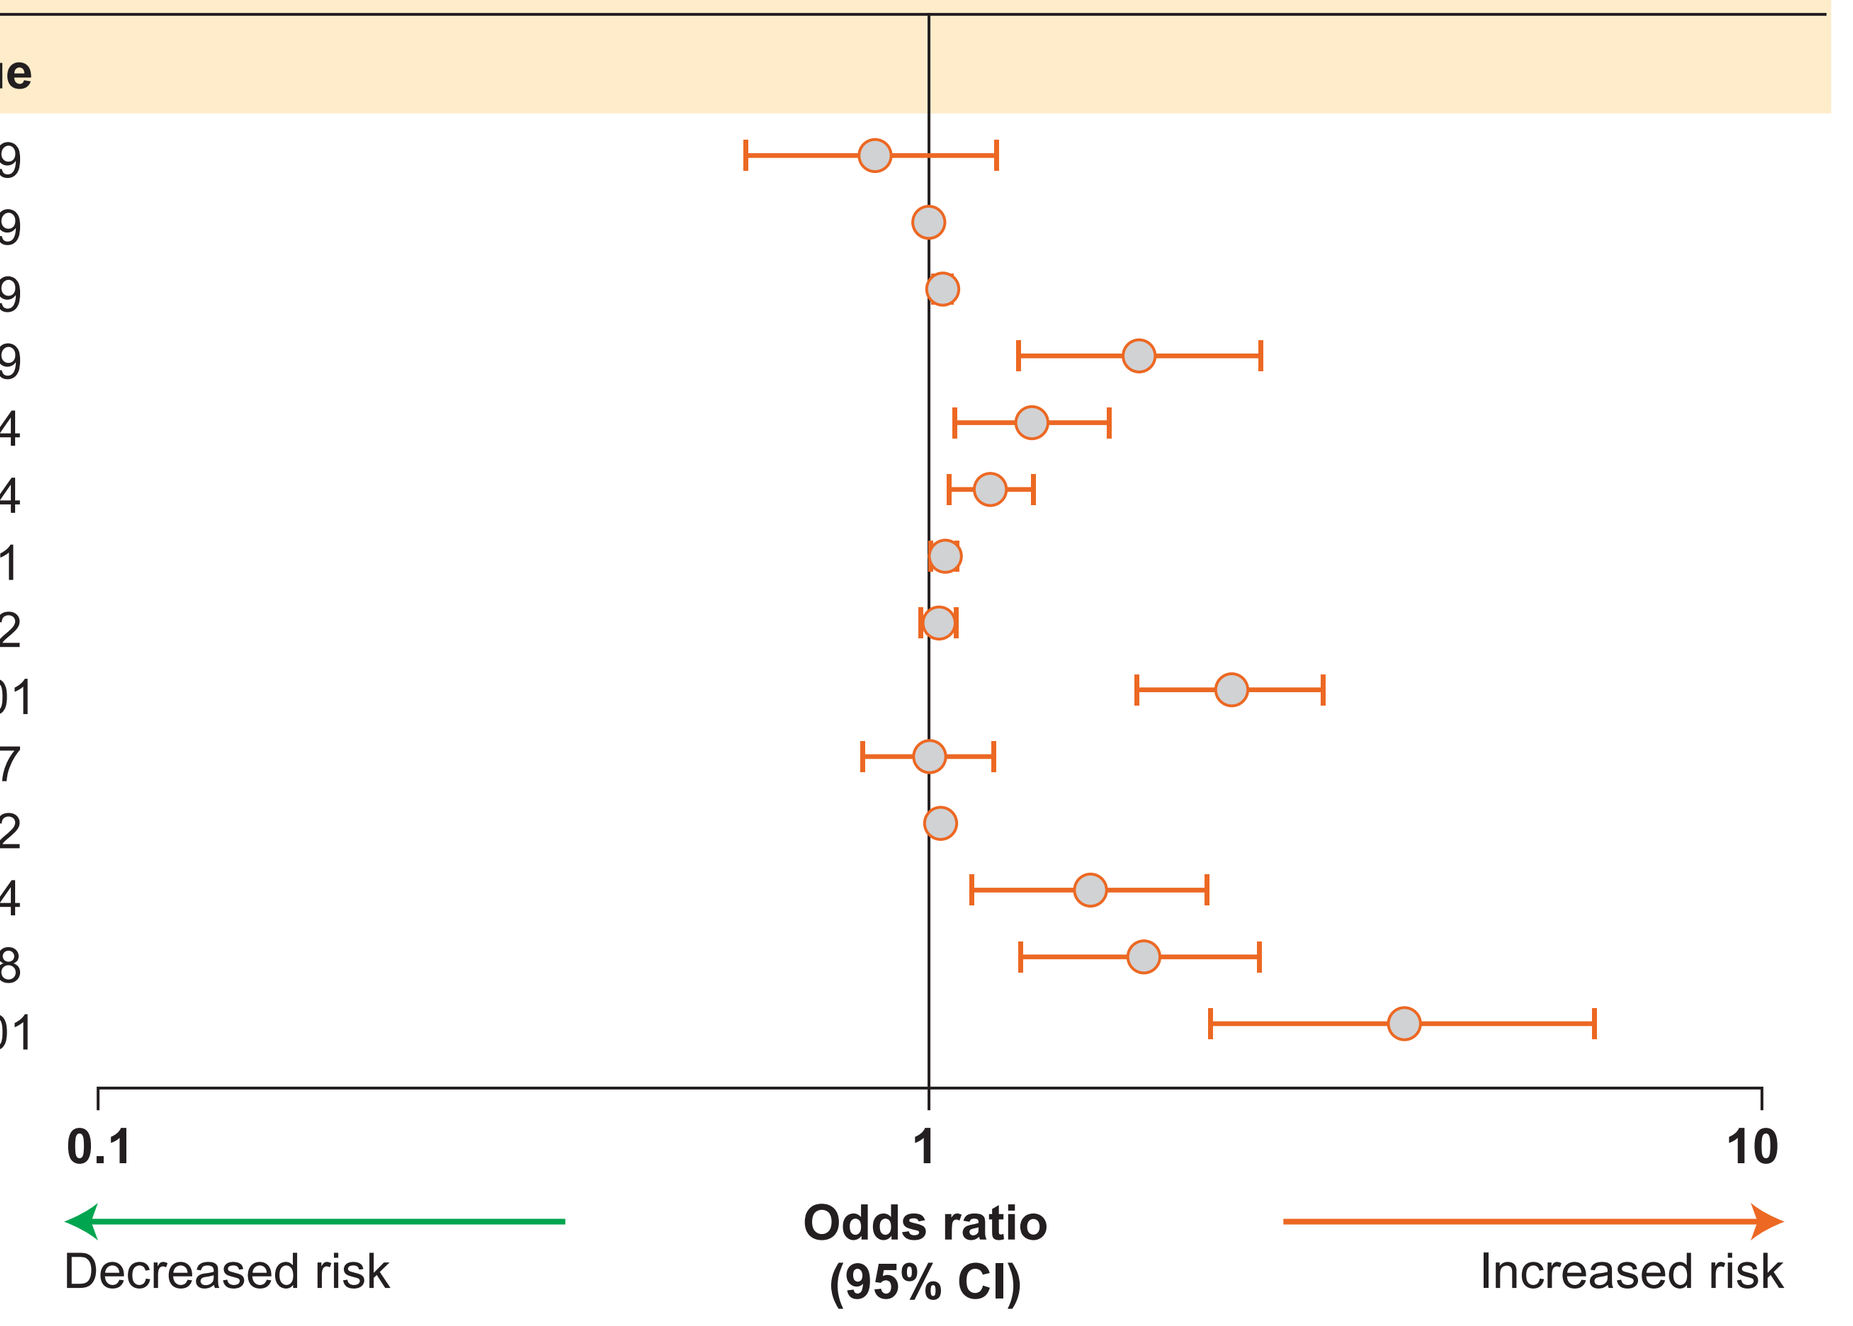
0.1
1
10
Decreased risk
Odds ratio (95% CI)
Increased risk

[a]An odds ratio >1 implies an increased risk in men.
Odds ratios are derived from individual regression models with the respective variable and study as predictors. For continuous variables, the odds ratio corresponds to a unit increase in the explanatory variable. MRI lesion activity is defined as ≥1 T1 Gd+ lesion or ≥2 T2 lesions at any post-baseline assessment up to M12.

Figure 3. Odds ratios for prediction of EDSS score >6 (left panel) and 6-month confirmed disability progression (right panel) in the long term (M12-M48) of TRANSFORMS and its extension phases

Parameter	EDSS >6 M12-M48 n	EDSS >6 M12-M48 Odds ratio (95% CI)	EDSS >6 M12-M48 p-value	6MCDP M12-M48 n	6MCDP M12-M48 Odds ratio (95% CI)	6MCDP M12-M48 p-value
Sex[a]	955	1.037 (0.627-1.714)	0.8888	772	1.253 (0.848-1.853)	0.2580
Age at core baseline (years)	955	1.052 (1.022-1.083)	0.0007	772	1.033 (1.010-1.057)	0.0050
Duration of MS since diagnosis (years)	955	1.056 (1.015-1.098)	0.0065	772	1.045 (1.009-1.082)	0.0131
Previous treatment for MS	955	2.348 (1.369-4.026)	0.0019	772	1.892 (1.267-2.826)	0.0018
Number of relapses in 2 years before baseline	954	1.517 (1.090-2.112)	0.0136	772	0.867 (0.673-1.118)	0.2718
EDSS score at baseline	955	2.669 (2.158-3.300)	<0.0001	772	1.440 (1.244-1.668)	<0.0001
Number of Gd+ lesions at baseline	940	0.896 (0.768-1.046)	0.1638	761	0.799 (0.677-0.944)	0.0082
T1 hypointense lesion volume at baseline (cm³)	946	1.103 (1.035-1.175)	0.0024	765	1.011 (0.944-1.082)	0.7553
Number of confirmed relapses (M0-M12)	955	1.889 (1.408-2.535)	<0.0001	772	1.199 (0.884-1.627)	0.2422
EDSS score change (M0-M12)	955	1.984 (1.514-2.601)	<0.0001	772	0.893 (0.715-1.114)	0.3147
T2 lesion volume at baseline (cm³)	946	1.019 (0.984-1.056)	0.2919	765	0.993 (0.961-1.027)	0.6892
New T2 lesions at M12 compared with M0	948	0.864 (0.531-1.407)	0.5565	768	1.119 (0.765-1.636)	0.5614
MRI lesion activity (M0-M12)	951	1.383 (0.845-2.266)	0.1974	769	1.354 (0.915-2.003)	0.1299
MRI lesion activity and ≥1 confirmed relapse (M0-M12)	955	2.027 (1.021-4.023)	0.0434	772	1.773 (0.955-3.292)	0.0699

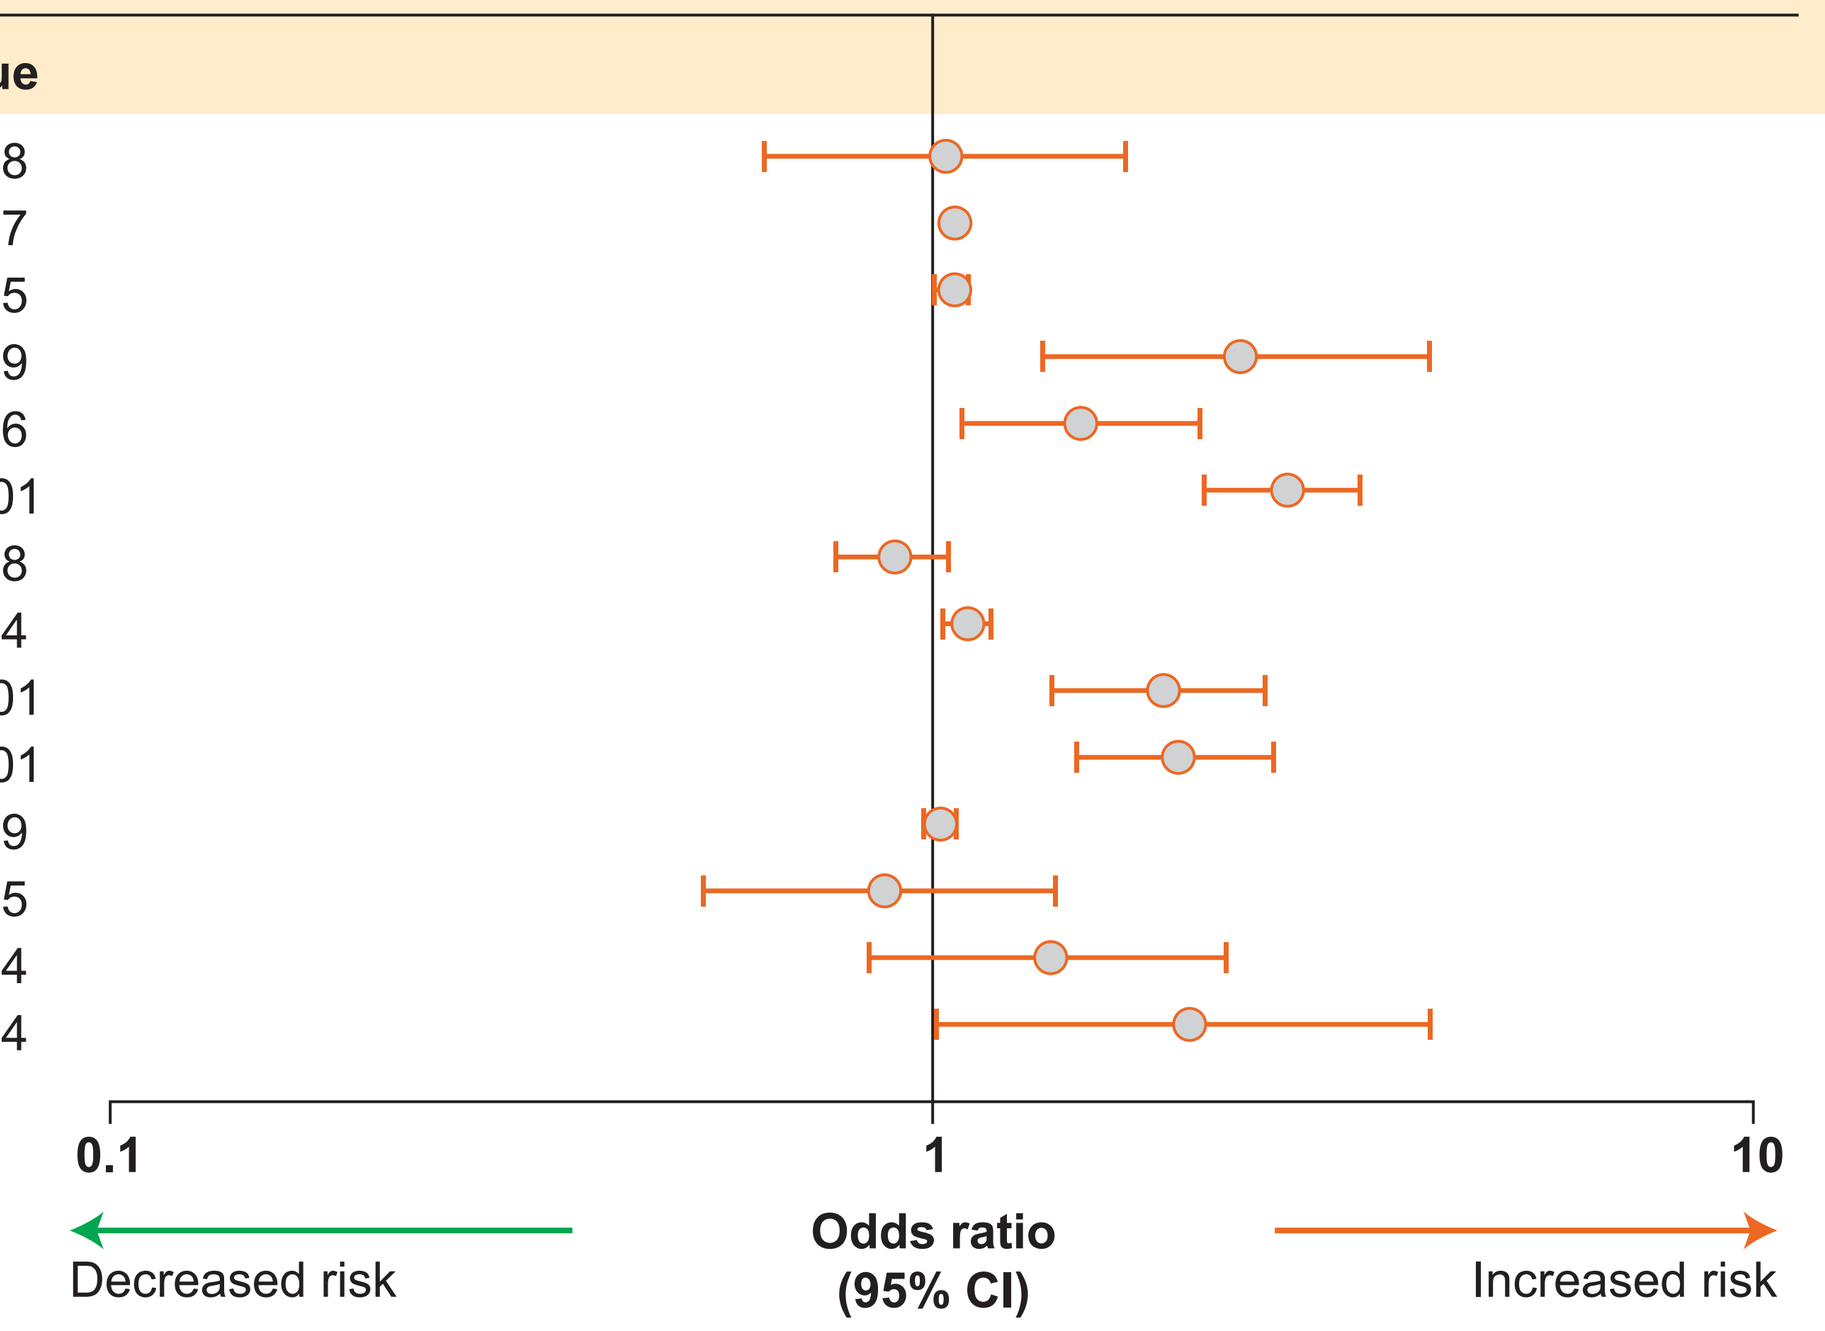
0.1
1
10
Decreased risk
Odds ratio (95% CI)
Increased risk

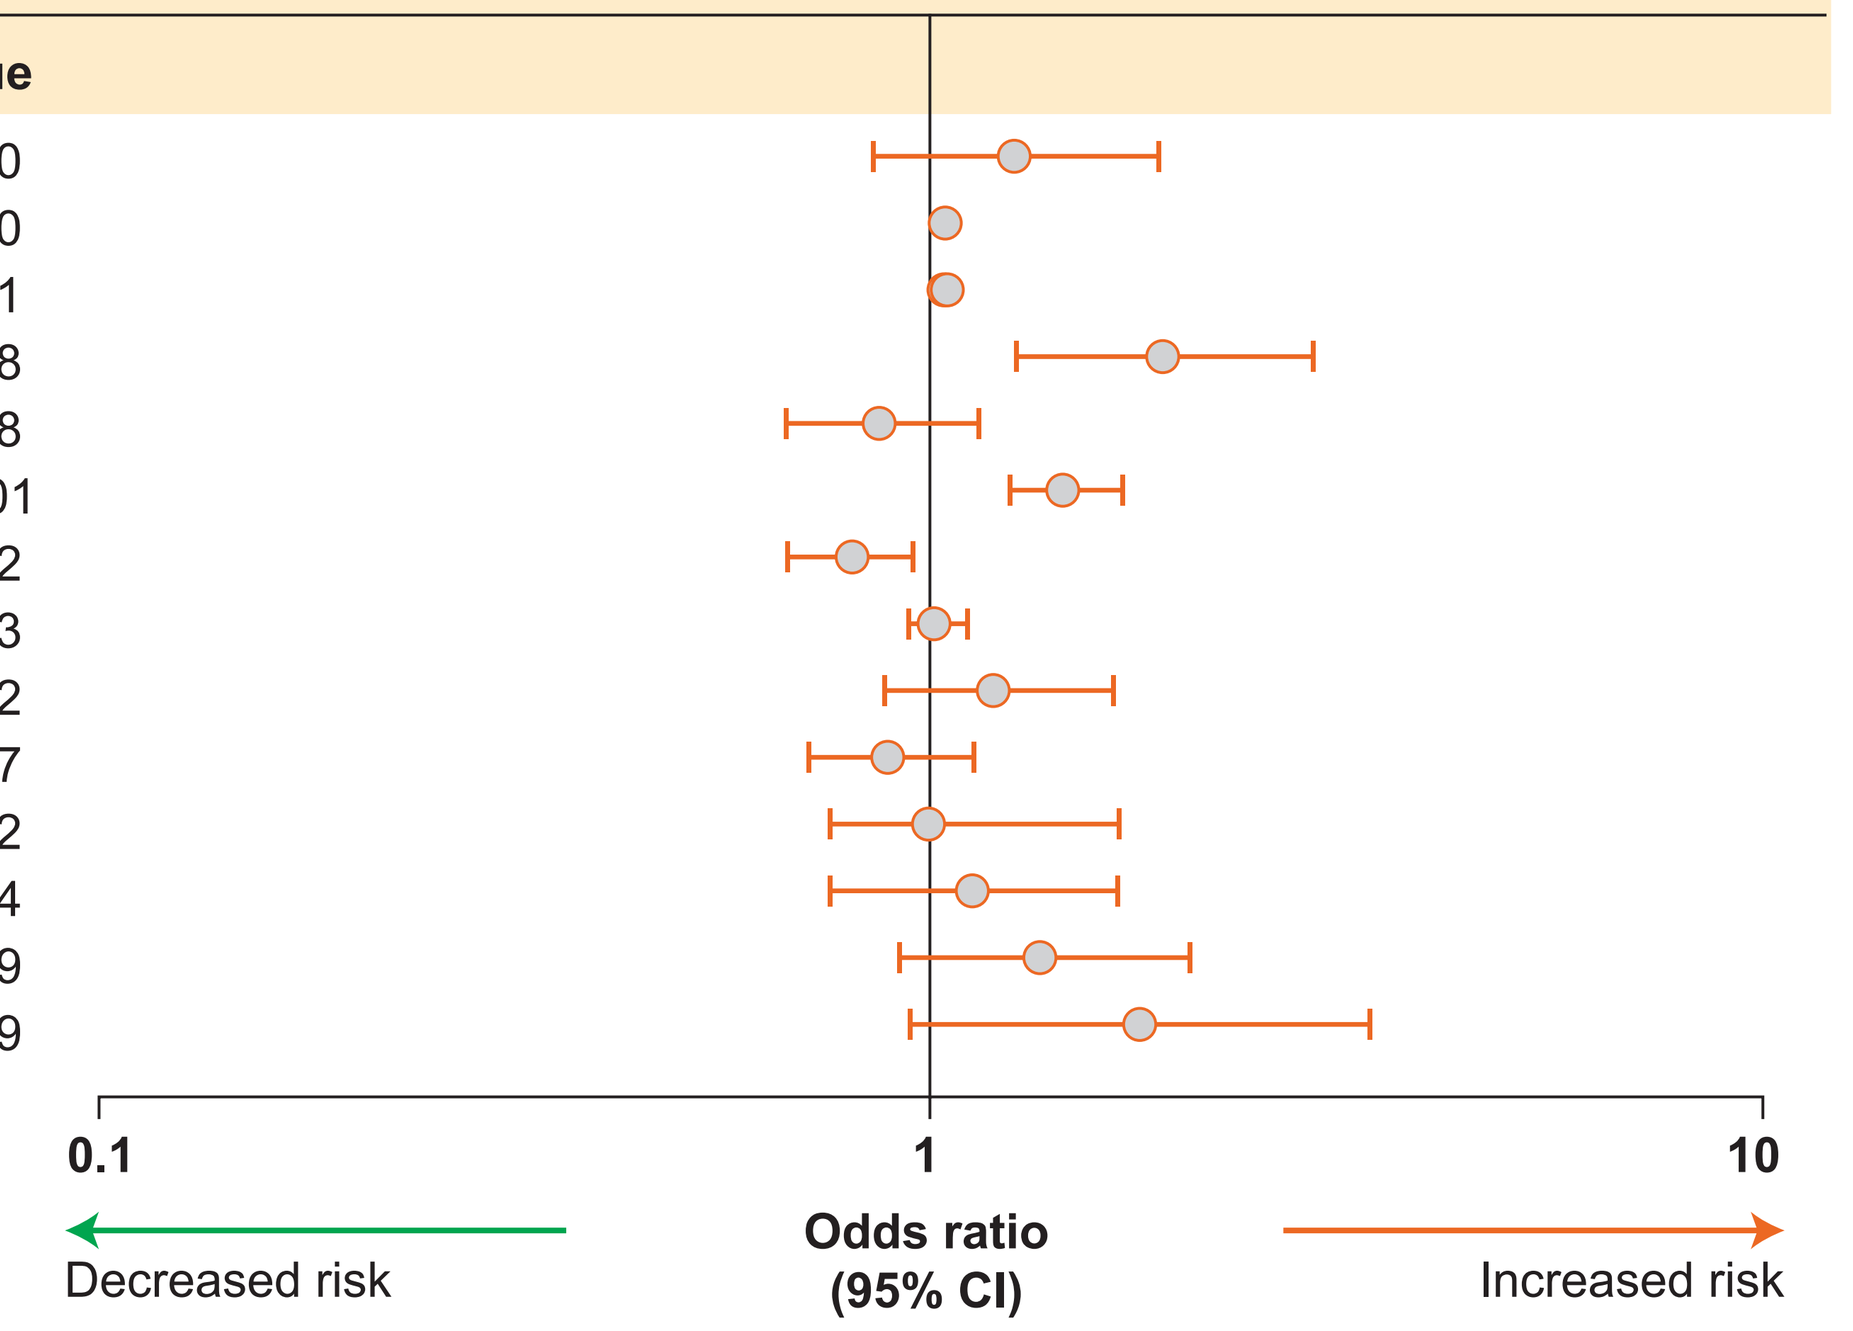
0.1
1
10
Decreased risk
Odds ratio (95% CI)
Increased risk

[a]An odds ratio >1 implies an increased risk in men.
Odds ratios are derived from individual regression models with the respective variable and study as predictors. For continuous variables, the odds ratio corresponds to a unit increase in the explanatory variable. MRI lesion activity is defined as ≥1 T1 Gd+ lesion or ≥2 T2 lesions at any post-baseline assessment up to M12.

- The patient or disease variables used to predict relapses, EDSS score >6 or 6MCDP were:
 - sex, age, duration of MS since diagnosis, previous treatment for MS, number of relapses in the previous 2 years, EDSS score, number of gadolinium-enhancing (Gd+) lesions, T1 hypointense lesion volume and T2 lesion volume (all at baseline)
 - number of confirmed relapses, change in EDSS score, number of new T2 lesions, MRI lesion activity (≥1 T1 Gd+ or ≥2 new T2 lesions),[9] MRI lesion activity and ≥1 confirmed relapse (all from baseline to M12).

Results

- In total, 1292 patients were randomized in TRANSFORMS (analysis sets: M12-M24, n=973; M12-M48, n=778).

Predictors of relapses

- Strongest predictors of relapses in both the short (M12-M24) and long (M12-M48) term were (**Figure 2**):
 - MRI lesion activity and ≥1 confirmed relapse between baseline and M12
 - number of confirmed relapses between baseline and M12.
- Other strong predictors of relapse activity in the long term were (**Figure 2**):
 - previous treatment for MS
 - MRI lesion activity between baseline and M12
 - number of new T2 lesions at M12 compared with baseline.
- Other predictors of relapses in both timeframes were (**Figure 2**):
 - number of relapses in the 2 years before baseline
 - EDSS score at baseline.

Predictors of disability worsening

- Baseline parameters that predicted disability reaching an EDSS score >6 and 6MCDP in the longer term were (**Figure 3**):
 - EDSS score
 - duration of MS
 - age
 - previous treatment for MS.
- Other predictors of an EDSS score >6 in the longer term were (**Figure 3**):
 - EDSS score change between baseline and M12
 - number of relapses in the 2 years before baseline
 - number of confirmed relapses between baseline and M12
 - T1 hypointense lesion volume at baseline
 - MRI lesion activity and ≥1 confirmed relapse between baseline and M12.

Conclusions

- Combined MRI lesion and relapse activity in the 1-year TRANSFORMS study was the strongest predictor of relapses at up to 4 years.
- Longer-term predictors of disability worsening based on an EDSS score >6 or on 6MCDP were EDSS score, previous treatment, disease duration and age at baseline.

References

1. Prosperini L et al. *Eur J Neurol.* 2009;16:1202-1209.
2. Rio J et al. *Mult Scler.* 2009;15:848-853.
3. Dobson R et al. *Neurology* 2014;82:248-254.
4. Cohen JA et al. *New Engl J Med.* 2010;362:402-415.
5. Cohen JA et al. *J Neurol Neurosurg Psychiatry* 2016;87:468-475.
6. Khatri B et al. *Lancet Neurol.* 2011;10:520-529.
7. Kappos L et al. *Mult Scler.* 2013;19(Suppl. 11):486-487.
8. Cohen J et al. *ECTRIMS Online Library* 2015;115507:P591.
9. Prosperini L et al. *Mult Scler.* 2014;20:566-576.

Acknowledgments

Editorial support was provided by Oxford PharmaGenesis, Oxford, UK, and was funded by Novartis Pharmaceuticals Corporation.

Disclosures

Aaron Boster: Acorda Therapeutics, Actelion Pharmaceuticals, Biogen Idec, CNS Therapeutics, Genzyme, Jazz Pharmaceuticals, Medtronic, Novartis Pharmaceuticals Corporation, Roche, Serono, Questcor Pharmaceuticals and Teva Pharmaceuticals. **Pavle Repovic:** Acorda Therapeutics, Biogen Idec, EMD Serono, Genzyme, Novartis Pharmaceuticals Corporation, Pfizer, Teva Pharmaceuticals (multiple sources). **Xiangyi Meng, Fernanda Boulos:** Novartis Pharmaceuticals Corporation (employees). **Daniela Piani Meier:** Novartis Pharma AG (employee). **Till Sprenger:** Actelion Pharmaceuticals, Allergan, ATI, Biogen Idec, ElectroCore, Novartis Pharmaceuticals Corporation, Sanofi Genzyme, Teva Pharmaceuticals. **Frederik Barkhof:** Biogen Idec, Bayer Schering Pharma, Genzyme, Janssen Alzheimer Immunotherapy, Lundbeck, MediciNova, Merck Serono, Novartis Pharmaceuticals Corporation, Roche, Sanofi-Aventis, Synthon (multiple sources).

© 2017 Novartis Pharmaceuticals Corporation.

Poster presented at the 2017 Annual Meeting of the Consortium of Multiple Sclerosis Centers, May 24-27, New Orleans, LA, United States.

This study was sponsored by Novartis Pharmaceuticals Corporation, United States.

Text: Qe1f3e
To: 8NOVA (86682) US only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe and Russia
+46737494608 Sweden and Europe
Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=e1f3e

Scan this QR code

Copies of this poster obtained through the QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address:
aaron.boster@ohiohealth.com