Subgroup and sensitivity analyses of treatment retention in patients participating in the PREFER*MS* study

Heidi Crayton,[1] Bruce A.C. Cree,[2] Florian P. Thomas,[3] Xiangyi Meng,[4] Lesley Schofield,[4] Fernanda Boulos,[4] Nadia Tenenbaum[4] on behalf of the PREFER*MS* investigators

[1]MS Center of Greater Washington, Vienna, VA, United States; [2]University of California San Francisco, San Francisco, CA, United States; [3]Hackensack University Medical Center, Hackensack, NJ, United States; [4]Novartis Pharmaceuticals Corporation, East Hanover, NJ, United States

DX29

Introduction

- Among patients with multiple sclerosis (MS), suboptimal adherence to injectable disease-modifying therapies (iDMTs) is common and reduces therapeutic effectiveness.[1,2]
- PREFER*MS* was the first large, randomized, prospective study comparing treatment retention in patients with relapsing forms of MS treated with fingolimod 0.5 mg/day or an iDMT.[3]
- In PREFER*MS*, treatment retention with fingolimod was significantly greater than with iDMTs.[3]

Objective

- Determine whether the greater rate of treatment retention seen with fingolimod than with iDMTs in the full analysis set (FAS) of PREFER*MS* was robust to sensitivity and subgroup analyses.

Methods

Study design

- PREFER*MS* was a 12-month, phase 4, open-label, active-controlled, randomized, multicenter study (117 sites) in the United States.
- Primary endpoint: patient retention on randomized treatment over 12 months.
- Enrolled patients with relapsing forms of MS were treatment naïve or had received no more than one class of iDMT (interferon β or glatiramer acetate).
- Patients were randomized (1:1) to fingolimod or pre-selected iDMT and followed up quarterly for 12 months.
- A single on-study treatment switch was allowed after 3 months, or earlier for efficacy or safety reasons (**Figure 1**).

Analyses

- Retention analyses: conducted during the open-label randomized treatment period.
- Sensitivity analysis: conducted in the FAS, excluded patients who switched treatment neither for safety nor efficacy reasons before day 84 (when switching for convenience became permissible), and patients who switched treatment between days 77 and 110.
- Subgroup analyses: conducted in the FAS.
- Between-group differences in retention calculated by normal approximation using continuity correction.
- Significance estimated using the Cochran–Mantel–Haenszel test, adjusted for treatment and treatment naïvety.
- The study was not powered for subgroup analyses.

Results

- 875 patients randomized; patient demographics and baseline characteristics were similar in the two treatment groups (**Table 1**).
- 861 patients included in the FAS.[3]
- Retention rate on randomized treatment in the FAS was significantly greater with fingolimod than iDMTs (**Figure 2a**).
- Mean duration of exposure to fingolimod was nearly twice that of iDMTs (301 vs 163 days).

Figure 1. PREFER*MS* study design

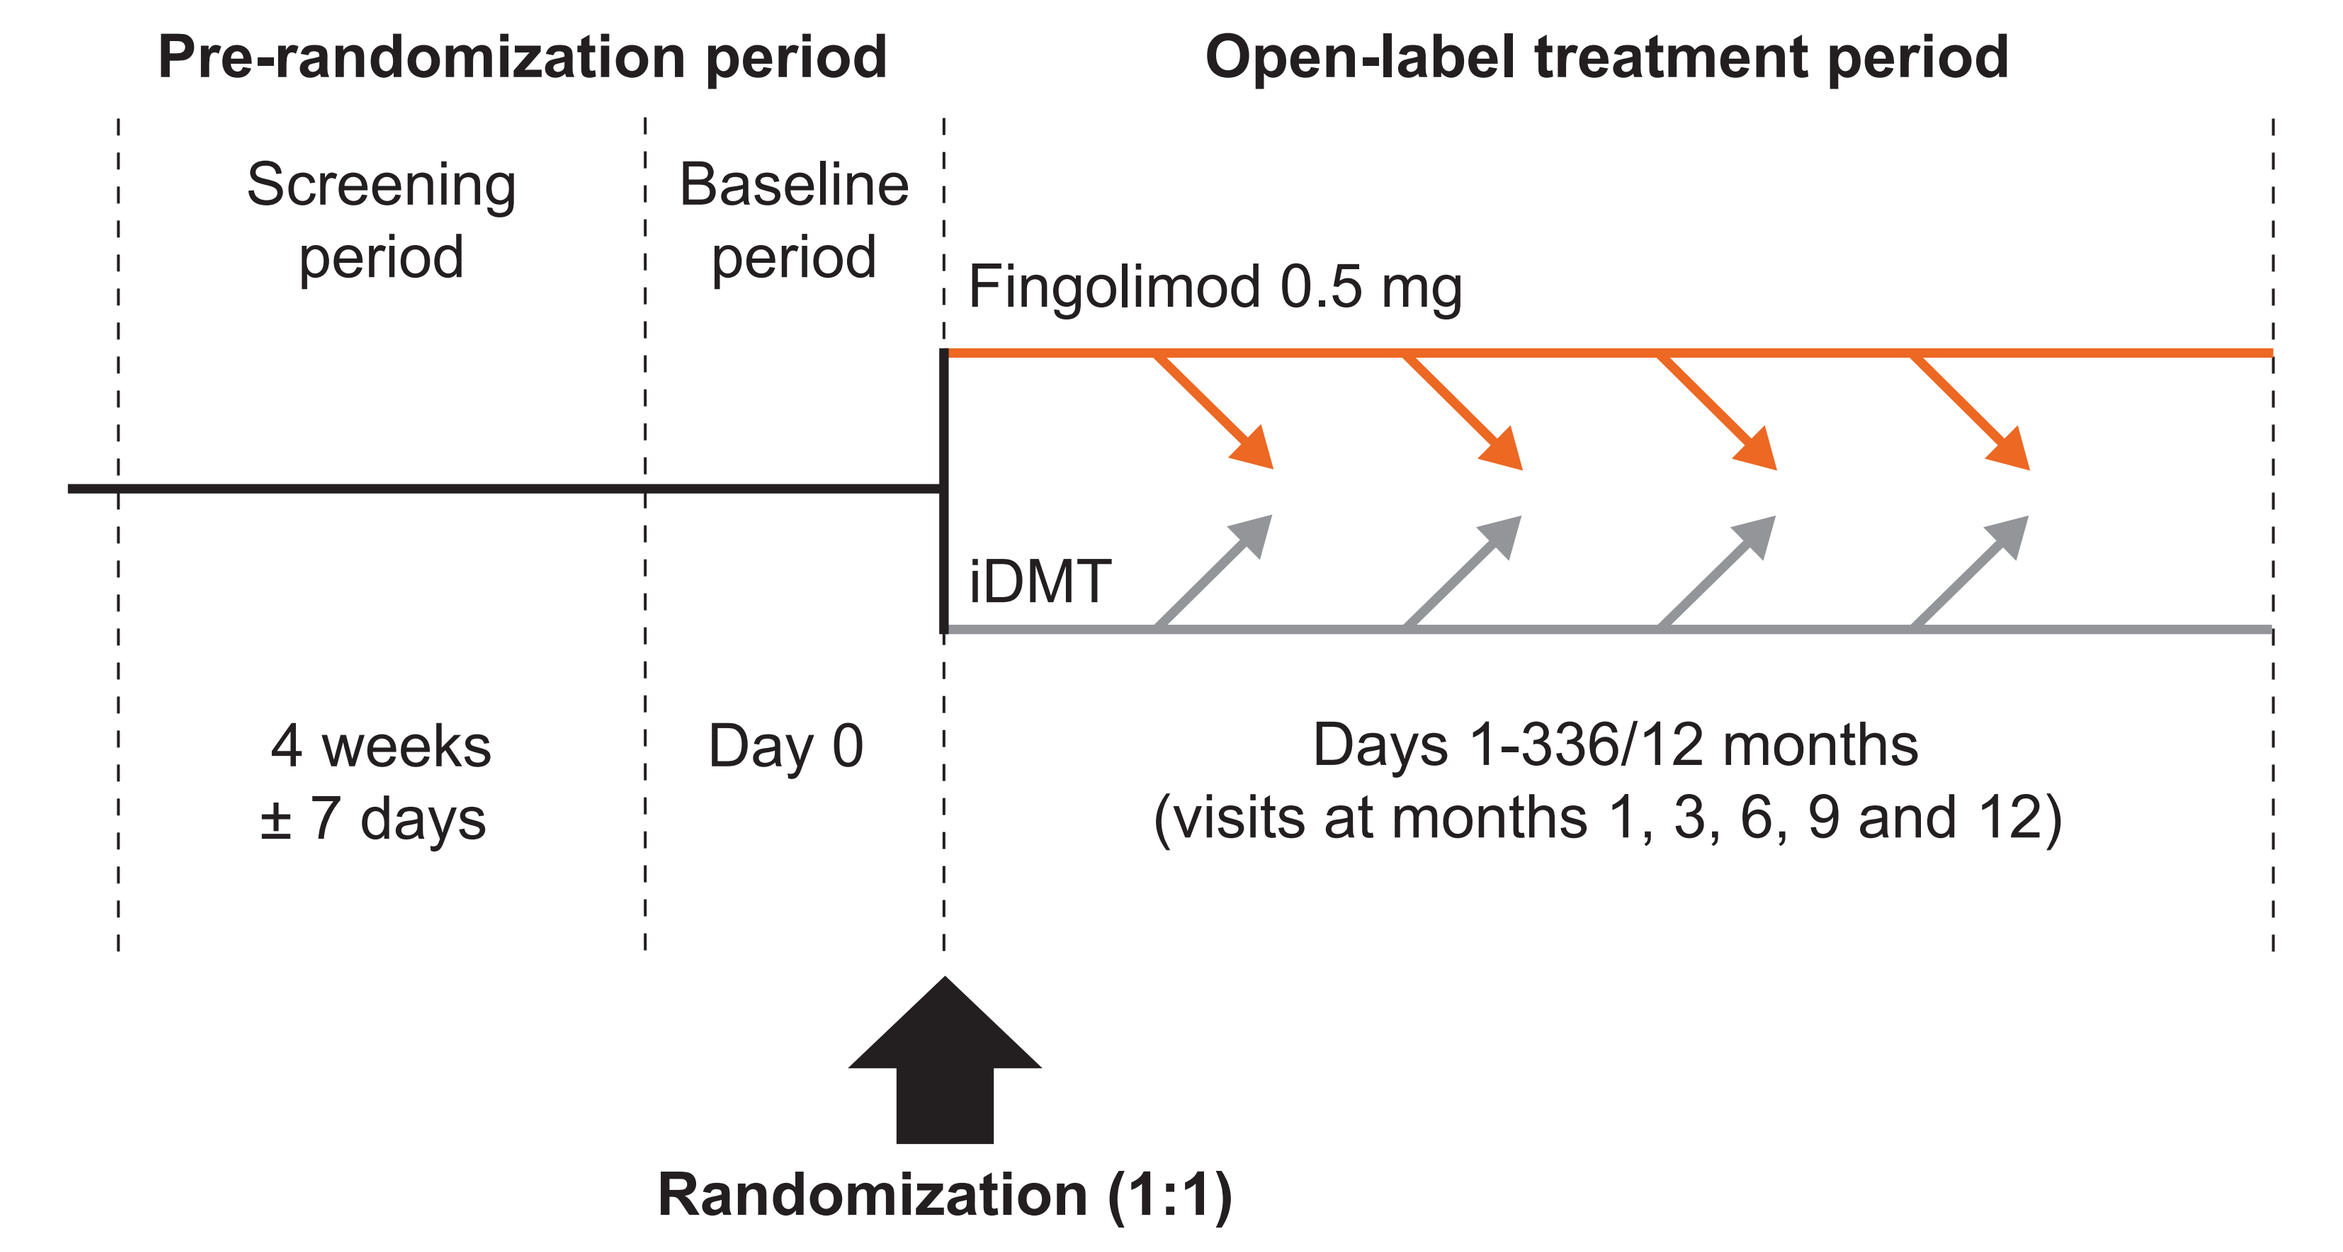

Pre-randomization period
Open-label treatment period
Screening period
Baseline period
Fingolimod 0.5 mg
iDMT
4 weeks ± 7 days
Day 0
Days 1-336/12 months (visits at months 1, 3, 6, 9 and 12)
Randomization (1:1)

Patients were allowed 1 switch from randomized treatment.
Reason for switch <3 months: safety or efficacy.
Reason for switch at 3-12 months: safety, efficacy, tolerability or convenience.

Table 1. PREFER*MS* patient demographics and baseline characteristics

Demographics and baseline characteristics[a]	Fingolimod 0.5 mg (n=436)	iDMT (n=439)	p-value
Age, years	41.5 (10.84)	41.9 (10.39)	0.6310
Sex, n (%)			
Male	125 (28.7)	110 (25.1)	0.2282
Female	311 (71.3)	329 (74.9)	
Race, n (%)			
Caucasian	355 (81.4)	355 (80.9)	
African American	69 (15.8)	72 (16.4)	
Asian	1 (0.2)	1 (0.2)	0.6553
Native American	1 (0.2)	1 (0.2)	
Pacific Islander	0	2 (0.5)	
Other	10 (2.3)	8 (1.8)	
Height, cm	168.5 (8.99)	167.5 (10.06)	0.1388
Weight, kg	82.94 (20.1)	83.56 (22.3)	0.6651
BMI, kg/m²	29.19 (6.70)	29.76 (7.55)	0.2335
Duration of MS since diagnosis, years	n=434 4.42 (6.67)	n=434 4.21 (5.94)	0.6314
Duration of MS since first symptoms, years	n=434 7.29 (8.21)	n=434 7.21 (7.66)	0.8871
Number of relapses in the past year	n=430 0.6 (0.95)	n=436 0.6 (0.94)	0.6041
Number of relapses in the past 2 years	n=430 0.9 (1.51)	n=436 0.9 (1.41)	0.6752
EDSS score	n=433 2.36 (1.56)	n=427 2.44 (1.51)	–
T2 lesion volume, cm³	n=431 7.65 (11.60)	n=415 7.44 (10.17)	–
Normalized brain volume, cm³	n=431 1521.42 (83.9)	n=412 1511.19 (90.5)	–
Number of gadolinium-enhancing lesions	n=429 1.08 (3.75)	n=414 0.85 (3.03)	–

[a]Data shown are mean (SD) unless stated otherwise.
Treatment group comparisons were made using the Cochran–Mantel–Haenszel test for categorical variables and a two-sample *t*-test for continuous variables.
BMI, body mass index; EDSS, Expanded Disability Status Scale.

Sensitivity analyses

- Retention rate in the iDMT group decreased from month 3 to month 4 (days 77-110; **Figure 2a**).
 - 126 patients switched from an iDMT to fingolimod.
 - Most common reasons patients cited for switching included flu-like symptoms (n=22), injection-site reaction (n=22) and inconvenient administration (n=21) (**Table 2**).

Table 2. Primary reasons for discontinuing randomized treatment between days 77 and 110

Reason	Randomized treatment: iDMT (n=126)	Randomized treatment: Fingolimod 0.5 mg (n=4)
Flu-like symptoms	22 (17.5)	–
Injection-site reaction	22 (17.5)	–
Inconvenient administration	21 (16.7)	–
Frequency of injection	20 (15.9)	–
Needle phobia	9 (7.1)	–
Occurrence of relapse	3 (2.4)	1 (25.0)
MRI disease activity	2 (1.6)	–
Depression	2 (1.6)	–
Hepatic side effects	1 (0.8)	3 (75.0)
Lipoatrophy	1 (0.8)	–
Spasticity	1 (0.8)	–
Other[a]	22 (17.5)	–

Data are n (%).
[a]Included the following reasons: patient opted to switch treatment, n=9; injection-site pain, sensitivity or intolerance, n=5; dissatisfaction with treatment, convenience or dislike of injections, n=4; generalized aching, feeling unwell, n=3; efficacy, n=1.
MRI, magnetic resonance imaging.

Figure 2. Retention rates in PREFER*MS*. a) Analysis in the FAS[3] and b) sensitivity analysis in the FAS excluding patients who switched treatment for reasons unrelated to efficacy or safety[a]

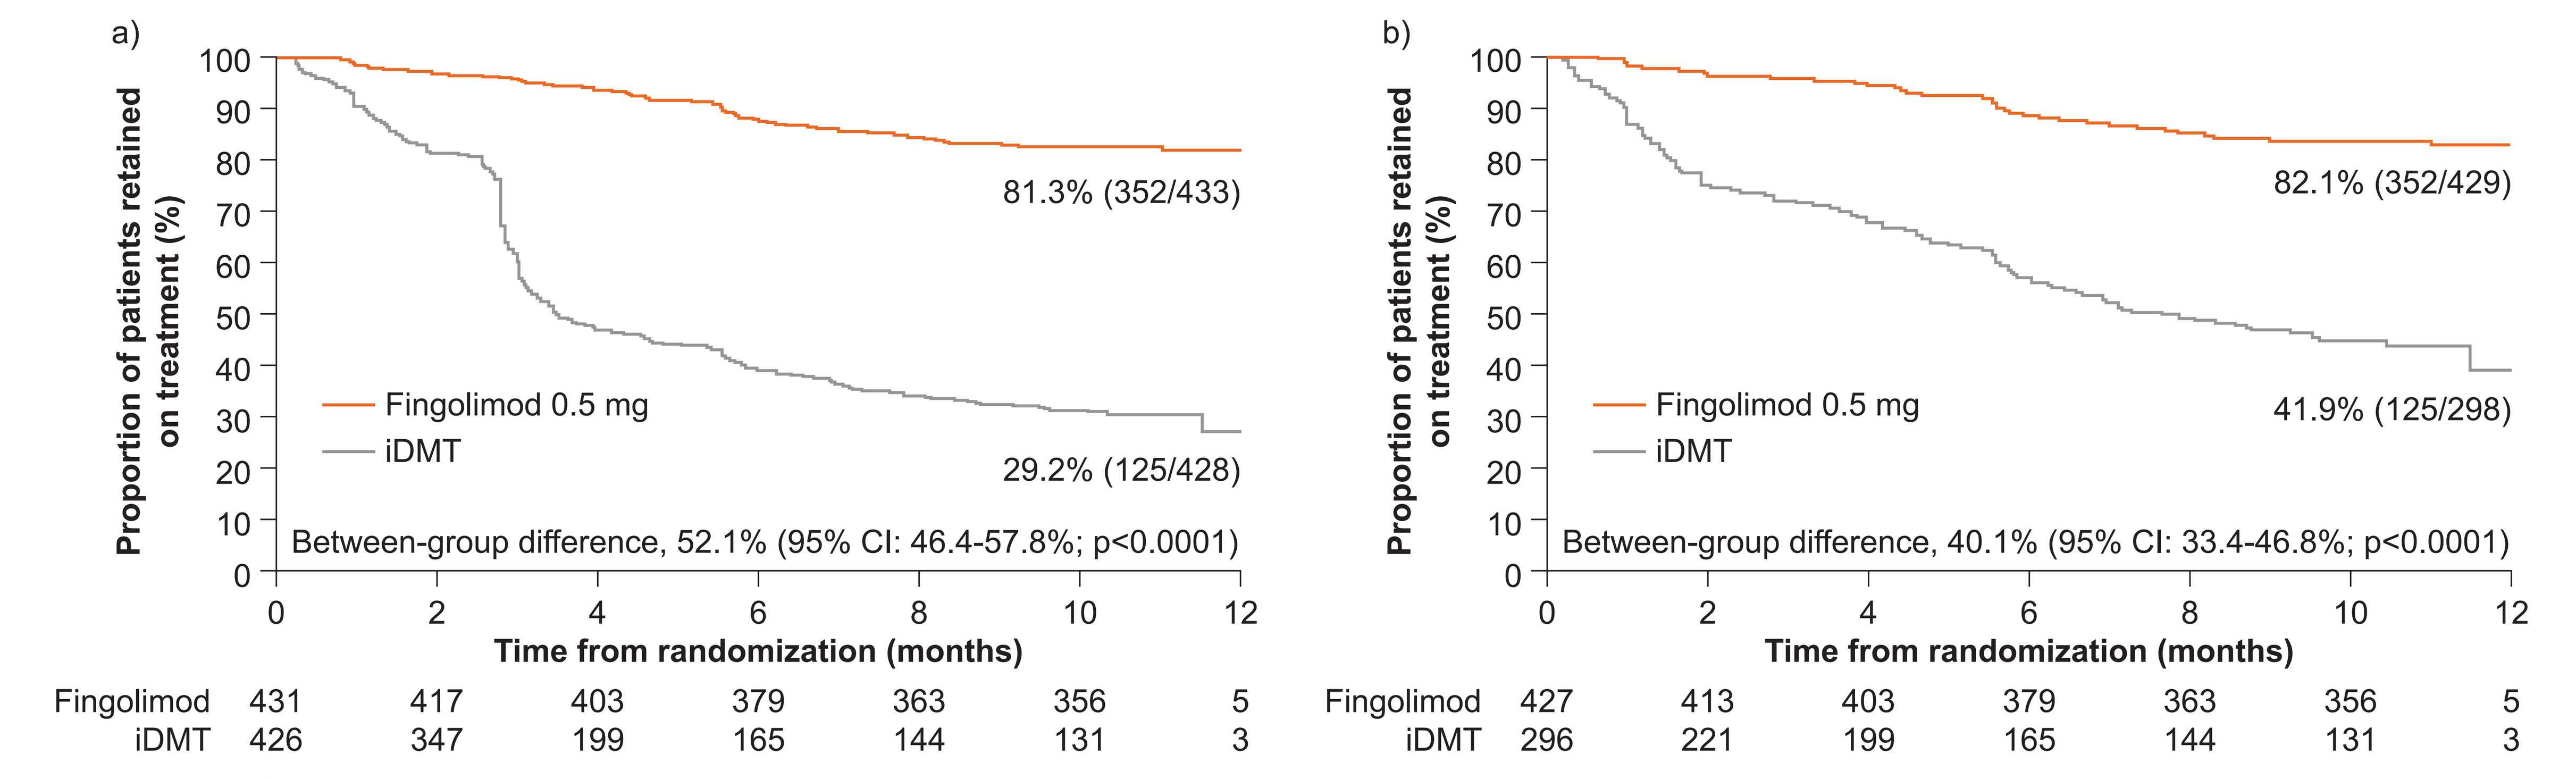

a)
Proportion of patients retained on treatment (%)
Fingolimod 0.5 mg
iDMT
81.3% (352/433)
29.2% (125/428)
Between-group difference, 52.1% (95% CI: 46.4-57.8%; p<0.0001)
Time from randomization (months)
Fingolimod 431 417 403 379 363 356 5
iDMT 426 347 199 165 144 131 3
b)
Proportion of patients retained on treatment (%)
Fingolimod 0.5 mg
iDMT
82.1% (352/429)
41.9% (125/298)
Between-group difference, 40.1% (95% CI: 33.4-46.8%; p<0.0001)
Time from randomization (months)
Fingolimod 427 413 403 379 363 356 5
iDMT 296 221 199 165 144 131 3

[a]Those who switched before day 84, and those who switched between days 77 and 110.

Figure 3. Retention rates on fingolimod and iDMTs in the FAS and by subgroup

Parameter	Fingolimod 0.5 mg, % retained (n/N)	iDMT % retained (n/N)	p-value
FAS	81.3 (352/433)	29.2 (125/428)	<0.0001
Subgroup			
Treatment naïve at enrollment	78.7 (166/211)	38.1 (72/189)	<0.0001
Previously treated	83.8 (186/222)	22.2 (53/239)	<0.0001
Age ≤40 years	83.1 (172/207)	29.8 (59/198)	<0.0001
Age >40 years	79.6 (180/226)	28.7 (66/230)	<0.0001
0-2 relapses in the 2 years before enrollment	83.0 (323/389)	29.1 (113/388)	<0.0001
≥3 relapses in the 2 years before enrollment	68.4 (26/38)	32.4 (12/37)	0.0021
Baseline EDSS score 0.0-3.5	83.9 (297/354)	29.3 (103/351)	<0.0001
Baseline EDSS score ≥4.0	69.6 (55/79)	28.9 (22/76)	<0.0001
Women	80.2 (247/308)	28.3 (91/321)	<0.0001
Men	84.0 (105/125)	31.8 (34/107)	<0.0001
African American	80.6 (54/67)	30.4 (21/69)	<0.0001
Caucasian	80.8 (286/354)	28.0 (97/347)	<0.0001
Hispanic	91.7 (22/24)	50.0 (11/22)	0.0015
Employed[a]	83.2 (228/274)	33.9 (83/245)	<0.0001
Unemployed[b]	77.9 (109/140)	21.8 (37/170)	<0.0001

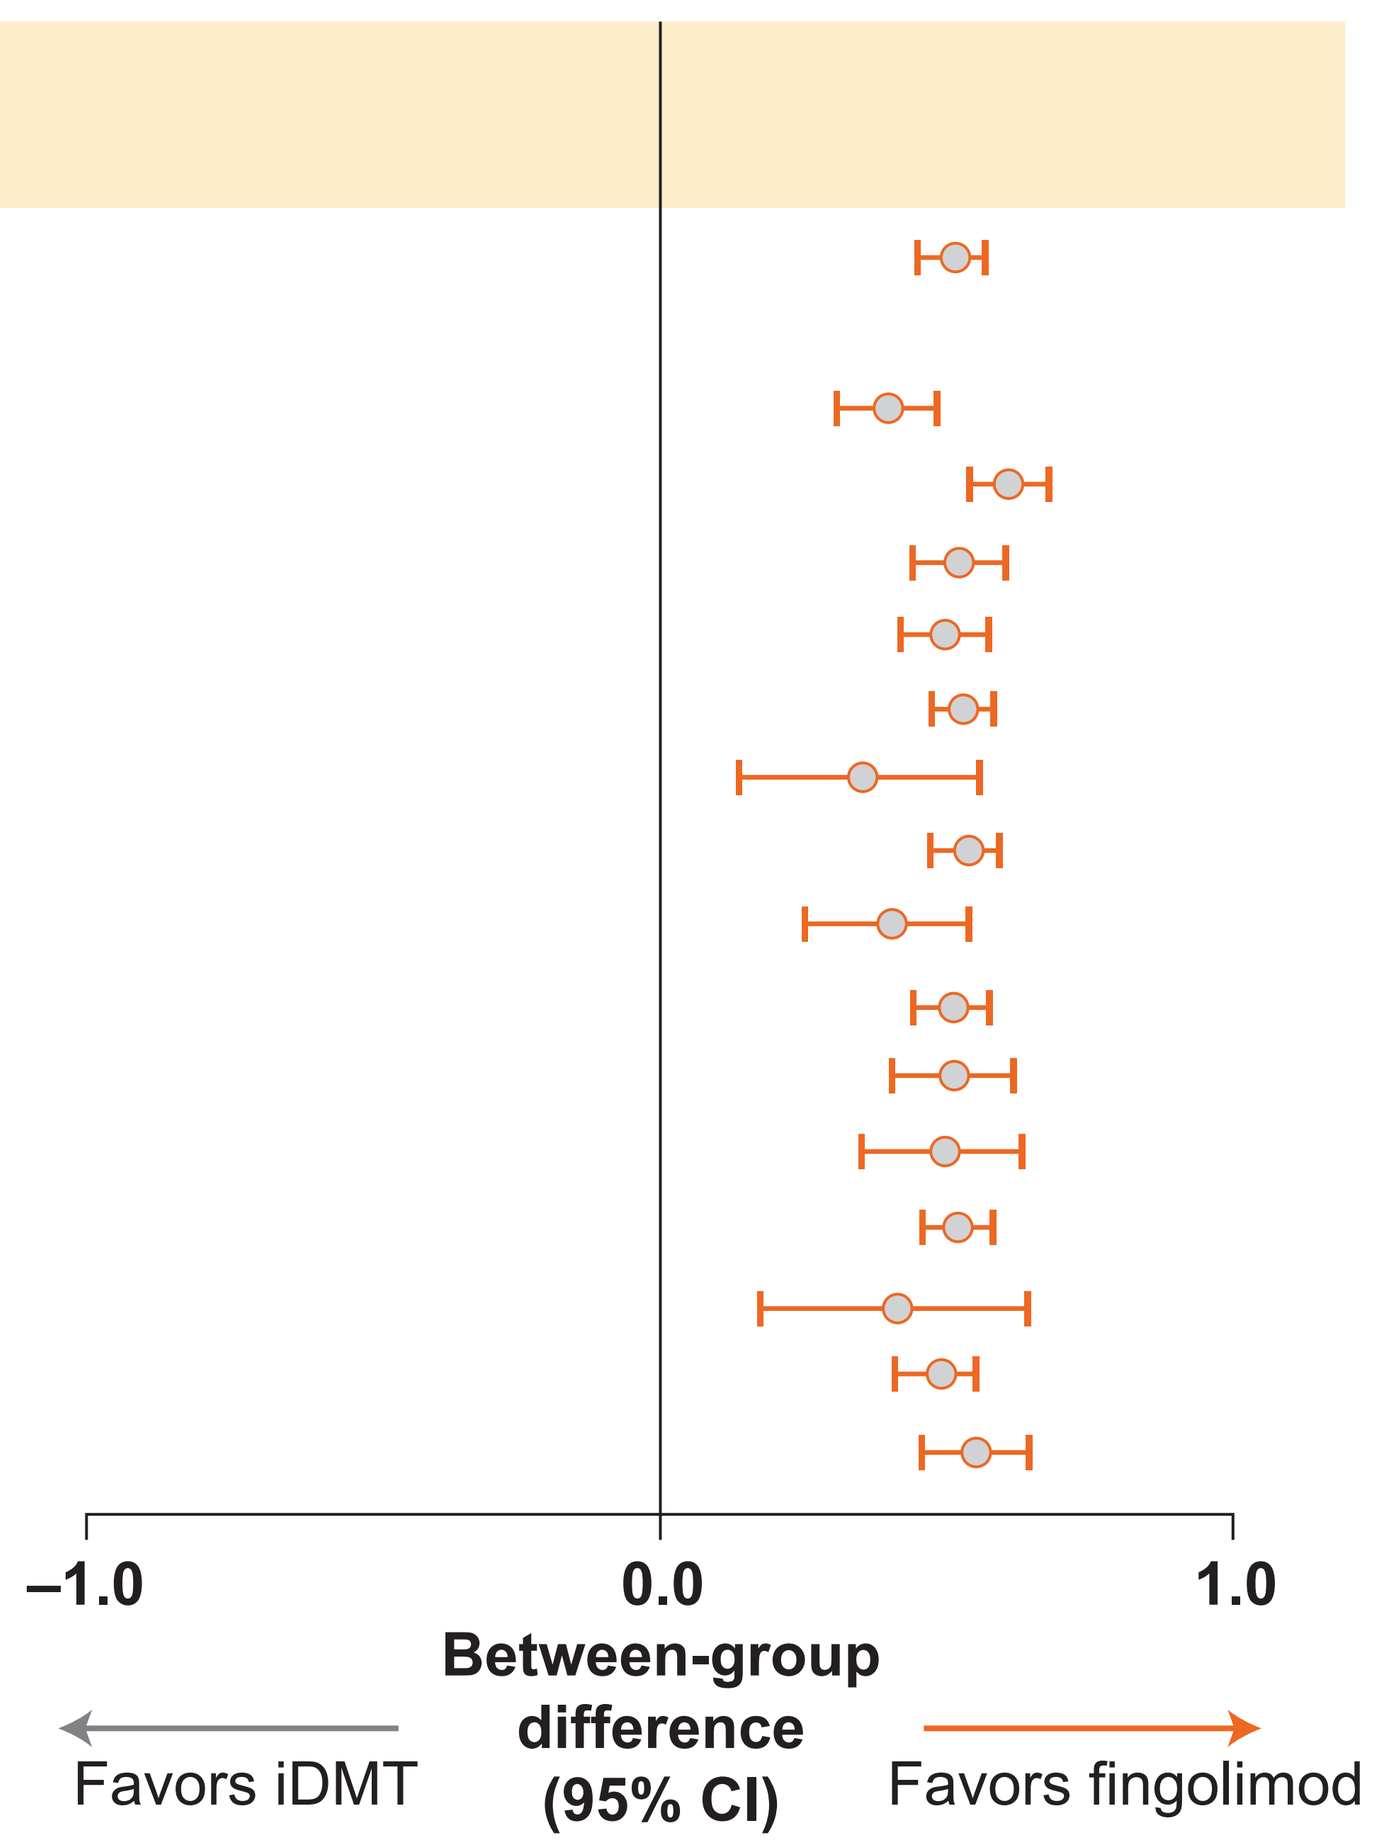

–1.0
0.0
1.0
Between-group difference (95% CI)
Favors iDMT
Favors fingolimod

[a]Including those employed full time or part time.
[b]Excluding retired individuals and those who declined to answer.

 - 4 patients switched from fingolimod to an iDMT.
 - Reasons patients cited for switching were hepatic side effects (n=3) and occurrence of relapse (n=1) (**Table 2**).
- 17 patients switched from an iDMT to fingolimod before the 3-month cut-off (day 84) for reasons unrelated to efficacy or safety.
 - Reasons included inconvenient administration (n=7), needle phobia (n=4), frequency of injections (n=3) and 'other' (n=3).
- In the sensitivity analysis, excluding these patients who switched therapy did not impact the significance of the primary endpoint (**Figure 2b**).

Subgroup analyses

- Retention rates on randomized treatment in all subgroups analyzed were greater with fingolimod than with iDMTs, regardless of age, sex, race, disease status or treatment status (**Figure 3**).

Conclusions

- The significantly greater rate of treatment retention observed with fingolimod than with iDMTs in PREFER*MS* was sustained in both sensitivity and subgroup analyses.
- If patients exhibit poor adherence to iDMTs, switching them to fingolimod, if indicated, may improve adherence.

References

1. Devonshire V et al. *Eur J Neurol.* 2011;18:69-77.
2. Patti F. *Patient Prefer Adherence* 2010;4:1-9.
3. Cree B et al. *Neurology.* 2016;86(Suppl. 16):P3.115.

Acknowledgments

Editorial support was provided by Oxford PharmaGenesis, Oxford, UK, and was funded by Novartis Pharmaceuticals Corporation.

Disclosures

Heidi Crayton: Acorda Therapeutics, Avanir, Biogen Idec, Genzyme, Novartis, Questcor and Teva Neuroscience. **Bruce A.C. Cree:** Abbvie, Biogen, EMD Serono, MedImmune, Novartis Pharmaceuticals Corporation, Sanofi-Genzyme, Shire and Teva Neuroscience. **Florian P. Thomas:** Acorda Therapeutics, Biogen, Novartis, Sanofi-Genzyme and Teva. **Xiangyi Meng, Lesley Schofield, Fernanda Boulos** and **Nadia Tenenbaum** are employees of Novartis Pharmaceuticals Corporation.

© 2017 Novartis Pharmaceuticals Corporation.

Poster presented at the 2017 Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC), May 24-27, New Orleans, LA, United States.

This study was sponsored by Novartis Pharmaceuticals Corporation, United States.

DOWNLOAD THIS POSTER AND VIEW VIDEO OF PREFER*MS* PRIMARY DATA

Scan QR code to download this poster and view video of PREFERMS primary data.
Also available at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=08cdc

And via Text Message (SMS)
Text: **Q08cdc**
To: **8NOVA (86682)** US only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe and Russia
+46737494608 Sweden and Europe

Scan to view a video presentation

Copies of this poster obtained through the QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address: msdoc1@hotmail.com