Poster No. DX30

Durable efficacy of cladribine tablets in patients with multiple sclerosis: analysis of relapse rates and relapse-free patients in the CLARITY and CLARITY extension studies

G Giovannoni,[1] G Comi,[2] S Cook,[3] P Rieckmann,[4] K Rammohan,[5] P Soelberg-Sorensen,[6] P Vermersch,[7] C Hicking,[8] AK Adeniji,[9] F Dangond[9]

[1]Queen Mary University of London, Blizard Institute, Barts and The London School of Medicine and Dentistry, London, UK; [2]Department of Neurology and Institute of Experimental Neurology, Università Vita-Salute San Raffaele, Ospedale San Raffaele, Milan, Italy; [3]Rutgers, The State University of New Jersey, New Jersey Medical School, Newark, NJ, USA; [4]Neurologische Klinik, Akademisches Krankenhaus Sozialstiftung Bamberg, Germany; [5]Ohio State University Hospital, Columbus, OH, USA; [6]Danish MS Center, Department of Neurology, Copenhagen University Hospital, Copenhagen, Denmark; [7]Université de Lille, CHU Lille, LIRIC-INSERM U995, FHU Imminent, Lille, France; [8]Merck KGaA, Darmstadt, Germany; [9]EMD Serono, Inc.,* Billerica, MA, USA

INTRODUCTION

- In the CLARITY study, cladribine tablets, given annually for 2 years in short-duration courses (3.5 mg/kg and 5.25 mg/kg of bodyweight, cumulative dose) significantly improved clinical (relapses and disability progression) and magnetic resonance imaging (MRI) outcomes in patients with relapsing multiple sclerosis.[1]
- The CLARITY Extension (EXT) study compared the safety and efficacy of 2 years' additional cladribine tablet treatment versus no additional treatment and the effect of switching from two courses of placebo to two courses of cladribine tablets (**Figure 1**).

Figure 1. CLARITY Extension study design

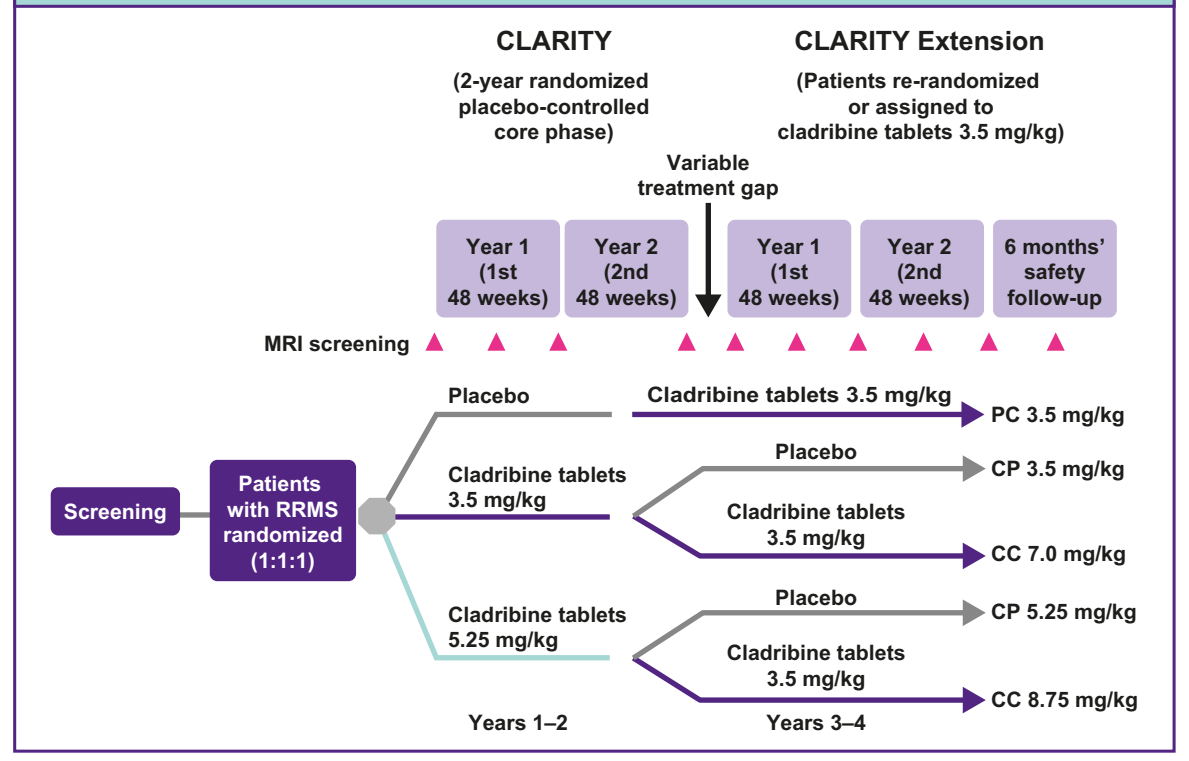
CLARITY
(2-year randomized placebo-controlled core phase)
CLARITY Extension
(Patients re-randomized or assigned to cladribine tablets 3.5 mg/kg)
Variable treatment gap
Year 1 (1st 48 weeks)
Year 2 (2nd 48 weeks)
Year 1 (1st 48 weeks)
Year 2 (2nd 48 weeks)
6 months' safety follow-up
MRI screening
Screening
Patients with RRMS randomized (1:1:1)
Placebo
Cladribine tablets 3.5 mg/kg
Cladribine tablets 5.25 mg/kg
Cladribine tablets 3.5 mg/kg
PC 3.5 mg/kg
Placebo
CP 3.5 mg/kg
Cladribine tablets 3.5 mg/kg
CC 7.0 mg/kg
Placebo
CP 5.25 mg/kg
Cladribine tablets 3.5 mg/kg
CC 8.75 mg/kg
Years 1–2
Years 3–4

MRI, magnetic resonance imaging; **RRMS**, relapsing–remitting multiple sclerosis.

OBJECTIVE

- To assess efficacy outcomes in CLARITY EXT and compare with those obtained in the same patient groups during CLARITY.

METHODS

- Placebo recipients in CLARITY were assigned to cladribine tablets 3.5 mg/kg in CLARITY EXT and all other patients were re-randomized 2:1 to cladribine tablets 3.5 mg/kg or placebo for 2 years (Table 1).

Table 1. Treatment group allocation in CLARITY Extension

		Treatment received in CLARITY Extension	
		Placebo	Cladribine tablets 3.5 mg/kg
Treatment received in CLARITY	Placebo	N/A	PC 3.5 mg/kg
	Cladribine tablets 3.5 mg/kg	CP 3.5 mg/kg	CC 7.0 mg/kg
	Cladribine tablets 5.25 mg/kg	CP 5.25 mg/kg	CC 8.75 mg/kg

N/A, not applicable.

- There was a variable treatment gap between the final CLARITY and first CLARITY EXT visits, with an overall median duration of ~41 weeks.
- After CLARITY EXT, patients entered a 6-month safety follow-up.
- Results are presented for annualized relapse rates (ARRs), proportion of patients who qualified as relapse free, and mean numbers of new T1 gadolinium-enhancing (Gd+) lesions.
- Efficacy objectives are exploratory; all determinations of significance are nominal.

RESULTS

Patients

- 806 patients were randomized/assigned to treatment in CLARITY EXT (**Table 2**).
- The treatment gap between CLARITY and CLARITY EXT varied within each group, but was distributed similarly across groups (**Table 2**).

Table 2. Baseline demographics and disease characteristics in CLARITY Extension

	CP 3.5 mg/kg (n=98)	CP 5.25 mg/kg (n=92)	CC 7 mg/kg (n=186)	CC 8.75 mg/kg (n=186)	PC 3.5 mg/kg (n=244)
Age, years	40.7 (10.7)	40.8 (9.6)	40.6 (10.5)	41.4 (10.1)	41.6 (9.6)
Female, n (%)	67 (68.4)	59 (64.1)	124 (66.7)	125 (67.2)	156 (63.9)
White, n (%)	96 (98.0)	90 (97.8)	181 (97.3)	180 (96.8)	240 (98.4)
Weight, kg	67.93 (14.89)	70.53 (15.16)	68.91 (14.09)	68.56 (14.01)	70.68 (15.56)
DMD use between CLARITY and CLARITY EXT, n (%)	2 (2.0)	0	0	0	4 (1.6)
Relapsed between CLARITY and CLARITY EXT, n (%)	9 (9.2)	8 (8.7)	17 (9.1)	18 (9.7)	46 (18.9)
Disease duration,[a] years	10.1 (6.7)	12.3 (8.0)	10.4 (7.1)	11.9 (7.9)	10.8 (6.8)
Median EDSS score (min; max)	2.5 (0.0; 6.5)	2.5 (0.0; 6.5)	2.5 (0.0; 6.5)	2.5 (0.0; 6.5)	3.0 (0.0; 6.5)
Median gap between studies (min; max), weeks	41.3 (0.1; 116.0)	43.1 (0.3; 112.9)	41.4 (0.4; 115.3)	39.5 (0.9; 111.0)	39.7 (0.3; 118.0)

Data are mean (SD), unless otherwise stated.
[a]Time from first attack.
DMD, disease-modifying drug; **EDSS**, Expanded Disability Status Scale.

ARR

- ARR was significantly lower in CLARITY EXT than CLARITY for patients in the PC 3.5 mg/kg group (0.26 vs 0.10, p<0.0001; **Figure 2**).
- There were no significant differences in ARR in CLARITY and CLARITY EXT in any of the other groups (**Figure 2**).

Proportion of patients who qualified as relapse free

- Overall, the proportion of patients who were reported as relapse free was high (>75%) across all groups in CLARITY EXT (**Figure 3**).
- Comparing groups between CLARITY and CLARITY EXT, there were no differences between study phases in the proportions of patients who remained relapse free, except in the PC 3.5 mg/kg group (58.0% vs 79.6%, p<0.0001; **Figure 3**).

Mean numbers of new T1 Gd+ lesions

- The mean number of new T1 Gd+ lesions was low in all treatment groups (**Figure 4**), but significant differences were seen between CLARITY and CLARITY EXT for the CP 3.5-, CC 7.0- and PC 3.5-mg/kg groups, but not the CC 8.75-mg/kg group.
- Patients who received placebo in CLARITY (PC 3.5 mg/kg) showed the highest mean number of new T1 Gd+ lesions (0.68).
 - Compared with CLARITY, these patients experienced a 90.4% relative reduction in new T1 Gd+ after receiving cladribine tablets 3.5 mg/kg during CLARITY EXT (0.07, p<0.001).

Figure 2. ARR by treatment group, for CLARITY and CLARITY Extension

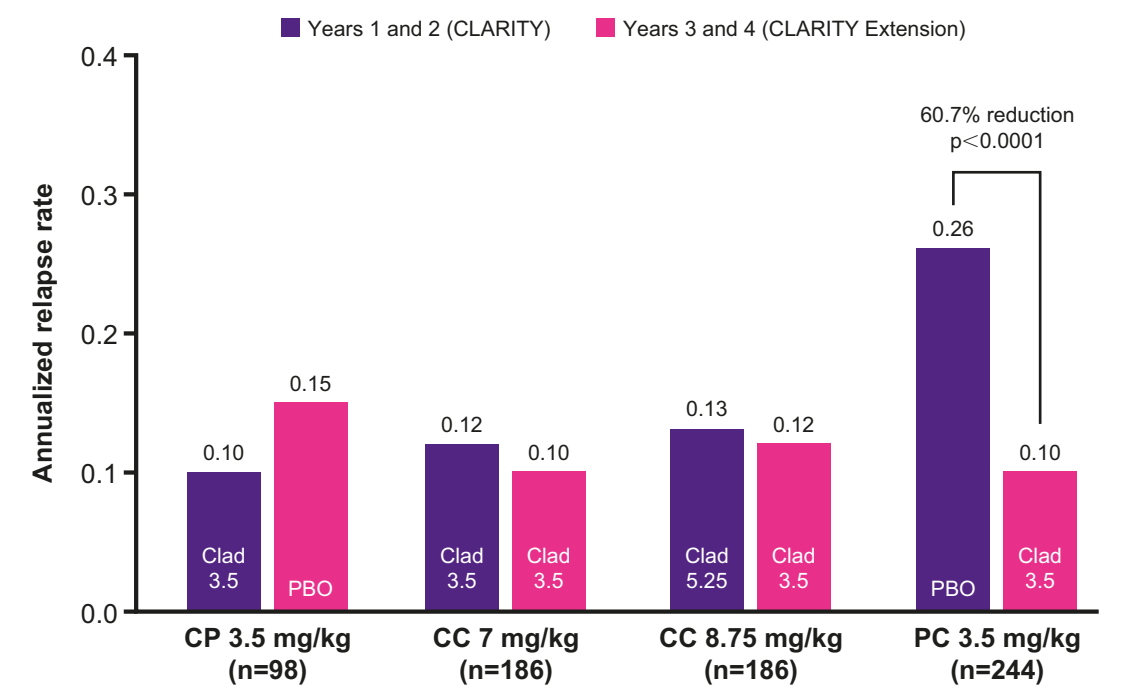
Years 1 and 2 (CLARITY)
Years 3 and 4 (CLARITY Extension)
Annualized relapse rate
0.4
0.3
0.2
0.1
0.0
60.7% reduction
p<0.0001
0.10
0.15
0.12
0.10
0.13
0.12
0.26
0.10
Clad 3.5
PBO
Clad 3.5
Clad 3.5
Clad 5.25
Clad 3.5
PBO
Clad 3.5
CP 3.5 mg/kg (n=98)
CC 7 mg/kg (n=186)
CC 8.75 mg/kg (n=186)
PC 3.5 mg/kg (n=244)

Clad, cladribine tablets; **PBO**, placebo.

Figure 3. Proportion of patients who remained relapse free[a] in CLARITY Extension irrespective of their treatment in CLARITY

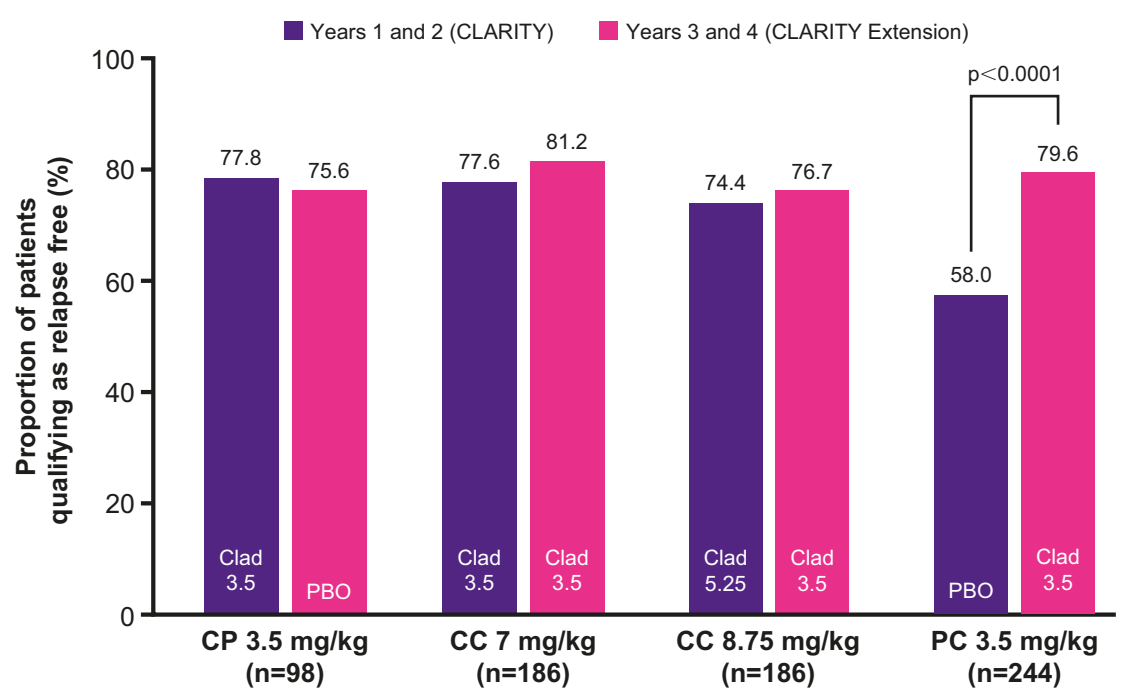
Years 1 and 2 (CLARITY)
Years 3 and 4 (CLARITY Extension)
Proportion of patients qualifying as relapse free (%)
100
80
60
40
20
0
p<0.0001
77.8
75.6
77.6
81.2
74.4
76.7
58.0
79.6
Clad 3.5
PBO
Clad 3.5
Clad 3.5
Clad 5.25
Clad 3.5
PBO
Clad 3.5
CP 3.5 mg/kg (n=98)
CC 7 mg/kg (n=186)
CC 8.75 mg/kg (n=186)
PC 3.5 mg/kg (n=244)

Clad, cladribine tablets; **PBO**, placebo.
[a]Denotes qualifying relapses.

Figure 4. Mean per-scan number of new T1 Gd+ lesions in patients in CLARITY Extension

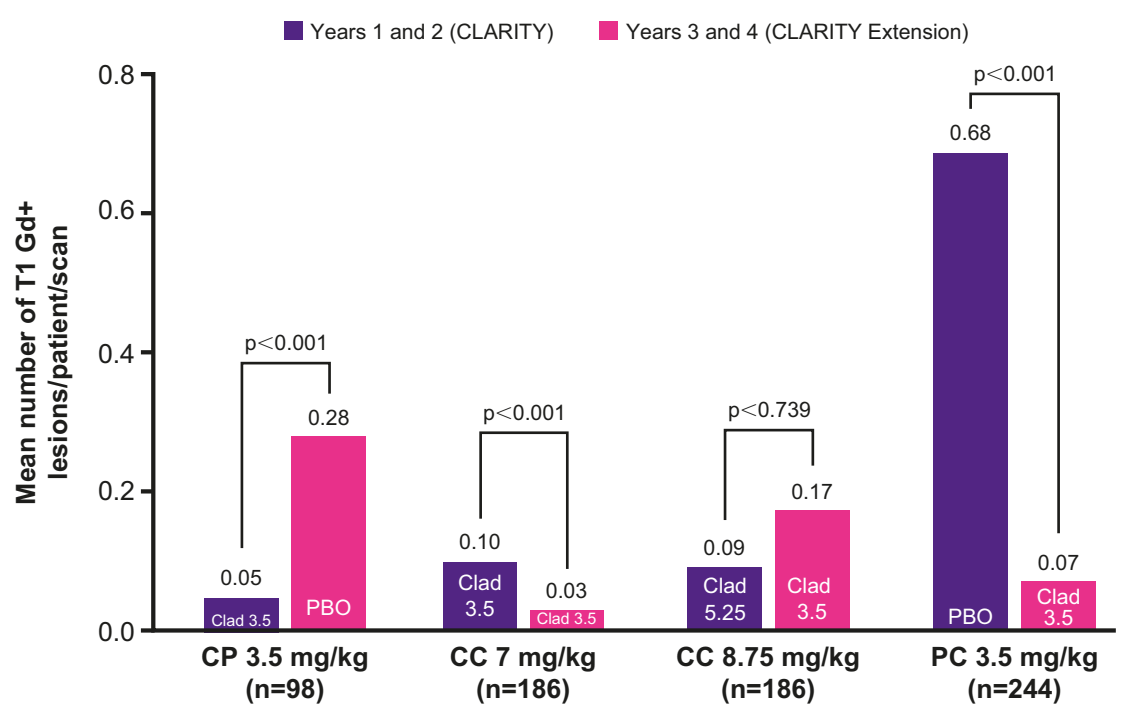
Years 1 and 2 (CLARITY)
Years 3 and 4 (CLARITY Extension)
Mean number of T1 Gd+ lesions/patient/scan
0.8
0.6
0.4
0.2
0.0
p<0.001
p<0.001
p<0.739
p<0.001
0.05
0.28
0.10
0.03
0.09
0.17
0.68
0.07
Clad 3.5
PBO
Clad 3.5
Clad 3.5
Clad 5.25
Clad 3.5
PBO
Clad 3.5
CP 3.5 mg/kg (n=98)
CC 7 mg/kg (n=186)
CC 8.75 mg/kg (n=186)
PC 3.5 mg/kg (n=244)

Clad, cladribine tablets; **Gd+**, gadolinium-enhancing; **PBO**, placebo.

- Patients who received cladribine tablets 3.5 mg/kg in CLARITY and 3.5 mg/kg in CLARITY EXT (CC 7 mg/kg) showed a low mean number of new T1 Gd+ lesions during CLARITY (0.10), and a small but significant reduction in CLARITY EXT (0.03, p<0.001).
- Patients who received cladribine tablets 5.25 mg/kg in CLARITY and 3.5 mg/kg in CLARITY EXT (CC 8.75 mg/kg) also showed a low mean number of new T1 Gd+ lesions during CLARITY (0.09) with no significant change during CLARITY EXT.
- Patients who received cladribine tablets 3.5 mg/kg in CLARITY and placebo in CLARITY EXT (CP 3.5 mg/kg) showed a low mean number of T1 Gd+ lesions during CLARITY (0.05) and a significant increase during CLARITY EXT (CP 3.5 mg/kg=0.28, p<0.001).
 - Despite this increase, the value was >2-fold lower than in patients treated with placebo in CLARITY (PC 3.5-mg/kg=0.68)
- Analysis of the distribution of T1 Gd+ lesions showed that the increase in mean T1 Gd+ counts in the CP 3.5-mg/kg group was associated with a subgroup of patients (11.6%) in whom the mean number of new T1 Gd+ lesions was ⩾ 1.0 (**Table 3**).

Table 3. Mean number of T1 Gd+ lesions/patient/scan by category, ITT analysis

	CP 3.5 mg/kg (n=98)	CC 7 mg/kg (n=186)	CC 8.75 mg/kg (n=186)	PC 3.5 mg/kg (n=244)
N (missing)	95 (3)	178 (8)	180 (6)	236 (8)
Mean number of new T1 Gd+ lesions per subject per scan				
<1.0, n (%)	84 (88.4)	178 (100)	175 (97.2)	234 (99.2)
≥1.0, n (%)	11 (11.6)	0 (0.0)	5 (2.9)	2 (0.8)

Gd+, gadolinium-enhancing; **ITT**, intent-to-treat.

Treatment gap between CLARITY and CLARITY EXT

- The mean per-scan number of new T1 Gd+ lesions was also analyzed by gap duration to assess if there was evidence of higher MRI activation in patients with a longer duration since the last dose of cladribine tablets.
 - 10.7% of patients experienced a gap ⩽4 weeks
 - 44.8% had a gap of >4 to ⩽43 weeks
 - 44.5% had a gap of >43 weeks.
- For patients with a gap of ⩽4 weeks, the time since the start of CLARITY was around 4.5 years versus 6.5 years for those who experienced a gap duration >43 weeks.
- In the CP 3.5-mg/kg group, T1 Gd+ MRI activity was greatest in patients who experienced the longest treatment gap.
 - The mean number of new T1 Gd+ lesions was higher among patients with a gap duration >43 weeks (0.39) than among those with a gap duration between 4 and 43 weeks (0.20), or ⩽4 weeks (0.19).
 - This pattern of results was not seen in the CP 5.25-mg/kg group.

CONCLUSIONS

- **Comparing CLARITY versus CLARITY EXT demonstrates the durable clinical benefits of cladribine tablets that persisted even in patients who received cladribine tablets in CLARITY and then placebo in CLARITY EXT.**
- **Patients who received placebo in CLARITY and then switched to cladribine tablets in CLARITY EXT experienced a significantly reduced ARR and increased proportion of relapse-free patients.**
- **In patients who received cladribine tablets in CLARITY, further treatment in CLARITY EXT did not bring additional efficacy.**
- **T1 Gd+ MRI activity was low in the majority of patients in each group at the end of CLARITY EXT, with patients who received placebo in CLARITY and cladribine tablets 3.5 mg/kg in CLARITY EXT experiencing a significant reduction in new T1 Gd+ lesions.**
- **A subgroup of patients who experienced a prolonged treatment gap showed a slight increase in T1 Gd+ activity but this appeared to have no impact on the persistent clinical benefits of cladribine tablets.**

REFERENCES

1. Giovannoni G, et al. *N Engl J Med* 2010;362:416–426.

ACKNOWLEDGMENTS

This study was sponsored by EMD Serono, Inc.* (in the USA) and Merck Serono SA – Geneva, an affiliate of Merck KGaA Darmstadt, Germany (ROW). The authors would like to thank patients and their families, investigators, co-investigators, and the study teams at each of the participating centers and at Merck KGaA, Darmstadt, Germany. Medical writing assistance was provided by Ash Dunne of inScience Communications, Springer Healthcare, Chester, UK, and was funded by Merck KGaA, Darmstadt, Germany.

DISCLOSURES

GG serves on advisory boards for Merck, Biogen Idec, and Vertex Pharmaceuticals; has received speaker honoraria and consulting fees from Bayer Schering Pharma, FivePrime, GlaxoSmithKline, GW Pharma, Merck, Biogen Idec, Pfizer Inc, Protein Discovery Laboratories, Teva Pharmaceutical Ind. Ltd, Sanofi-Aventis, UCB, Vertex Pharmaceuticals, Genzyme Corporation, Ironwood, and Novartis; serves on the Merck speakers bureau; and received research support unrelated to this study from Biogen Idec, Merck, Novartis, and Ironwood. **GC** has received consulting fees from Novartis, Teva Pharmaceutical Ind. Ltd., Sanofi-Aventis, Merck, and Bayer Schering; lecture fees from Novartis, Teva Pharmaceutical Ind. Ltd., Sanofi-Aventis, Merck Serono, Biogen Dompè, Bayer Schering, and Serono Symposia International Foundation; and trial grant support from Novartis, Teva Pharmaceutical Ind. Ltd., Sanofi-Aventis, Merck, Biogen Dompè, and Bayer Schering. **SC** has received honoraria for lectures/consultations from Merck Serono, Bayer HealthCare, Sanofi-Aventis, Neurology Reviews, Biogen Idec, Teva Pharmaceuticals, and Actinobac Biomed Inc.; has served on advisory boards for Bayer HealthCare, Merck, Actinobac Biomed, Teva Pharmaceuticals, and Biogen Idec; and received grant support from Bayer HealthCare. **PR** has received honoraria for lectures/steering committee meetings from Merck, Biogen Idec, Bayer Schering Pharma, Boehringer-Ingelheim, Sanofi-Aventis, Genzyme, Novartis, Teva Pharmaceutical Industries, and Serono Symposia International Foundation. **KR** has received honoraria for lectures and steering committee meetings from EMD Serono, Biogen Idec, Sanofi-Aventis, Genzyme, Novartis, Teva Neurosciences, Acorda and Roche/Genentech. **PS-S** has served on advisory boards for Biogen Idec, Merck, Novartis, Genmab, Teva, Elan, and GSK; on steering committees or independent data monitoring boards in trials sponsored by Merck, Genmab, Teva, GSK, and Bayer Schering; has served as Editor-in-Chief of the European Journal of Neurology, is currently editorial board member for Multiple Sclerosis Journal, European Journal of Neurology, and Therapeutic Advances in Neurological Disorders; has received speaker honoraria from Biogen Idec, Merck Serono, Teva, Bayer Schering, Sanofi-Aventis, Genzyme, and Novartis; and has received payment for writing/reviewing manuscripts from IBI Consulting, a division of Informa plc. His department has received research support from Biogen Idec, Bayer Schering, Merck Serono, Teva, Baxter, Sanofi-Aventis, BioMS, Novartis, Bayer, RoFAR, Roche, Genzyme, the Danish Multiple Sclerosis Society, the Danish Medical Research Council, the European Union Sixth Framework Programme: Life sciences, Genomics and Biotechnology for health. **PV** has received honoraria and consulting fees from Biogen Idec, Sanofi, Bayer, Novartis, Merck, GSK, and Almirall; and research support from Biogen Idec, Sanofi, Bayer, and Merck. **CH** is an employee of Merck KGaA, Darmstadt, Germany. **AKA** and **FD** are employees of EMD Serono, Inc.,* Billerica, MA, USA.

Cladribine tablets is currently under clinical investigation and has not yet been approved by any regulatory authority. Status: March 2017.

The CLARITY study: NCT00213135, the CLARITY EXT study: NCT00641537.

*A business of Merck KGaA, Darmstadt, Germany.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

GET POSTER PDF

Presented at the 31st Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC); May 24–27, 2017; New Orleans, LA, USA