Poster No. DX33

Benefits of cladribine tablets on relapse rates and disability progression in patients with multiple sclerosis: analysis of pooled data from the CLARITY and ONWARD studies

G Giovannoni,[1] X Montalban,[2] C Hicking,[3] F Dangond[4]

[1]Queen Mary University of London, Blizard Institute, Barts and The London School of Medicine and Dentistry, London, UK;
[2]Department of Neurology-Neuroimmunology, Hospital Universitari Vall d'Hebron, Barcelona, Spain; [3]Merck KGaA, Darmstadt, Germany; [4]EMD Serono Inc.,* Billerica, MA, USA

INTRODUCTION

- The CLARITY study evaluated the efficacy of cladribine tablets (3.5 and 5.25 mg/kg of bodyweight, cumulative dose) given in short courses annually for 2 years to patients with relapsing multiple sclerosis (RMS; **Figure 1**).[1]
- The ONWARD study evaluated the safety and tolerability of cladribine tablets (3.5 mg/kg and 5.25 mg/kg of bodyweight, cumulative dose) given in short courses annually for 2 years as an add-on to interferon (IFN)-β therapy in patients with RMS who experienced at least one relapse while on IFN-β (**Figure 1**). Treatment with cladribine tablets 5.25 mg/kg was discontinued after an early protocol amendment.[2]
 - Efficacy was explored as a secondary objective of the ONWARD study.[2]

Figure 1. CLARITY and ONWARD study designs

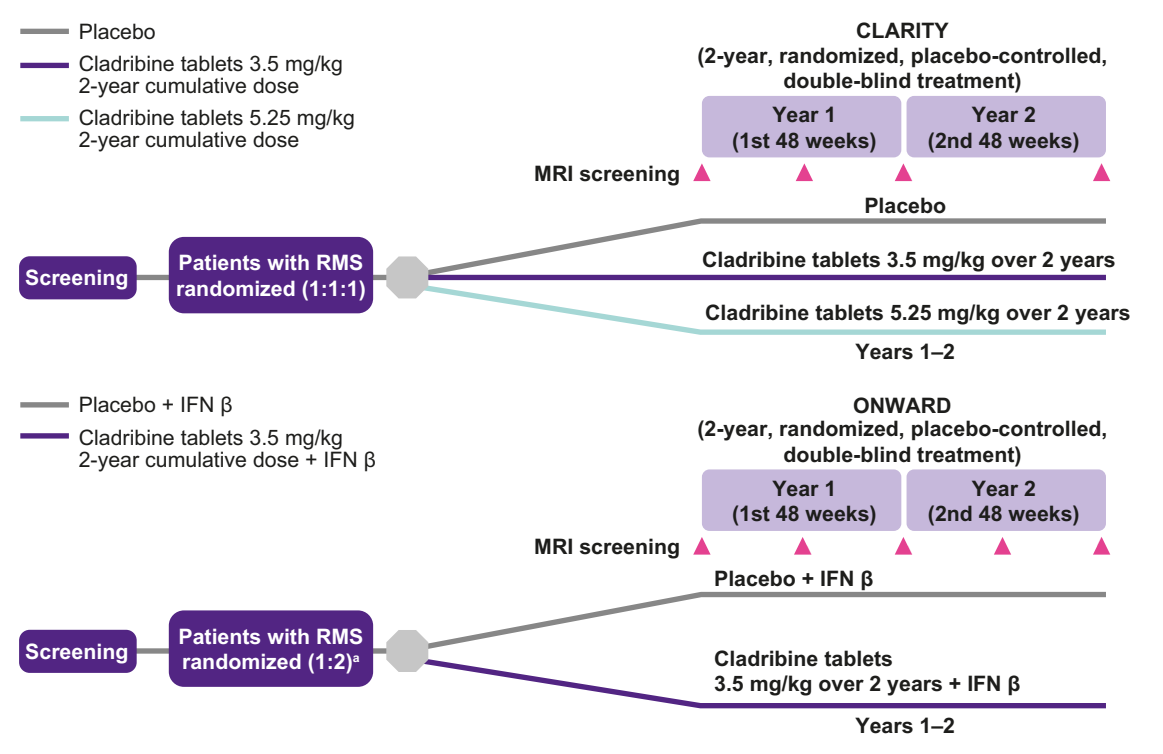

Placebo
Cladribine tablets 3.5 mg/kg 2-year cumulative dose
Cladribine tablets 5.25 mg/kg 2-year cumulative dose
CLARITY (2-year, randomized, placebo-controlled, double-blind treatment)
Year 1 (1st 48 weeks)
Year 2 (2nd 48 weeks)
MRI screening
Screening
Patients with RMS randomized (1:1:1)
Placebo
Cladribine tablets 3.5 mg/kg over 2 years
Cladribine tablets 5.25 mg/kg over 2 years
Years 1–2
Placebo + IFN β
Cladribine tablets 3.5 mg/kg 2-year cumulative dose + IFN β
ONWARD (2-year, randomized, placebo-controlled, double-blind treatment)
Year 1 (1st 48 weeks)
Year 2 (2nd 48 weeks)
MRI screening
Screening
Patients with RMS randomized (1:2)a
Placebo + IFN β
Cladribine tablets 3.5 mg/kg over 2 years + IFN β
Years 1–2

IFN, interferon; **MRI**, magnetic resonance imaging; **RMS**, relapsing multiple sclerosis.
[a]Prior to an early protocol amendment, the ONWARD study included randomization to treatment with cladribine tablets 5.25 mg/kg (cumulative dose over 2 years) + IFN β.

- Treatment with cladribine tablets in CLARITY significantly improved clinical outcomes (relapses and disability progression) versus placebo.[1]
 - Among patients who received either cladribine tablets 3.5 mg/kg or 5.25 mg/kg, the annualized relapse rate (ARR) was significantly lower than in the placebo group (0.14 and 0.15, respectively, 0.33; $p<0.001$ for each comparison).[1]
 - There was also a lower risk of 3-month sustained disability progression (hazard ratio for the 3.5 mg/kg group, 0.67; 95% confidence interval [CI], 0.48–0.93; p=0.02; and hazard ratio for the 5.25 mg/kg group, 0.69; 95% CI, 0.49–0.96; p=0.03).[1]
- In ONWARD, significant reductions in relapse rates were also seen with cladribine tablets vs placebo.[2]
- Combining efficacy data from the double-blind periods of the CLARITY and ONWARD studies allows the effect of 2 years' treatment with cladribine tablets on relapse rates and disability progression to be assessed.

OBJECTIVE

- To summarize the effects of cladribine tablets on relapse rates and disability progression in pooled analyses of the CLARITY and ONWARD studies, including patient subgroups.

METHODS

- Inclusion criteria for each study are shown in **Table 1**.
- Data from the 2-year, double-blind periods of the CLARITY and ONWARD studies were used to summarize the efficacy of cladribine tablets vs placebo in patients with RMS in the intent-to-treat (ITT) population (all patients randomly allocated to cladribine tablets, or placebo), and in subgroups defined by baseline characteristics.
- This *post hoc* pooled analysis includes 17 patients who received cladribine tablets 5.25 mg/kg under the original ONWARD protocol.
- Further outcomes from this pooled analysis are described in other presentations at this meeting (posters DX31 and DX32).

Table 1. Inclusion criteria for CLARITY and ONWARD

Inclusion criteria	CLARITY	ONWARD
Age	18−65 years	18−55 years
MS stage	RRMS	RRMS or relapsing SPMS
DMD use for MS at study entry	Washout of DMDs (for 3 months) prior to study entry was required	Treatment with IFN for ⩾48 weeks before screening was required
Relapses	⩾1 relapses within 12 months before study. No relapse within 28 days of screening	⩾1 relapse while receiving IFN but otherwise clinically stable in the 28 days before screening
EDSS score	0−⩽5.5	1−⩽5.5

DMD, disease-modifying drug; **EDSS**, Expanded Disability Status Scale; **IFN**, interferon; **RRMS**, relapsing-remitting multiple sclerosis; **SPMS**, secondary progressive multiple sclerosis.

Annualized relapse rate

- Relative risk (RR) and 95% CI were estimated for qualifying ARR in patients treated with cladribine tablets or placebo, in the ITT population and in patient subgroups, using a Poisson regression model with fixed effects for treatment group and study, the number of relapses in previous year as covariate and with the log of time on study as the offset variable.
 - p values were based on the Wald Chi-square test.

3- and 6-month confirmed disability progression

- Hazard ratios and 95% CIs for time to 3- or 6-month confirmed Expanded Disability Status Scale (EDSS) progression with cladribine tablets and placebo were calculated for the ITT population and for patient subgroups from a Cox proportional hazard model adjusted for study effect and with the EDSS score at baseline as covariate.

Subgroup analyses

- Endpoints:
 - ARR
 - 3-month confirmed EDSS progression
 - 6-month confirmed EDSS progression.
- Subgroups:
 - patients with no relapses or ⩾1 relapse in the previous year
 - patients with no T1 gadolinium-enhancing (Gd+) lesions or ⩾1 T1 Gd+ lesions at baseline
 - patients with <9 or ⩾9 T2 lesions at baseline
 - patients with no prior use of disease-modifying drugs (DMDs) or prior use of DMDs before baseline
 - males or females
 - patients aged ⩽40 or >40 years
 - patients with EDSS score ⩽3.0 or ⩾3.5 at baseline
 - patients with or without evidence of high disease activity (HDA) at baseline. HDA was defined as patients with ⩾2 relapses in the previous year (regardless of previous treatment status), or patients with ⩾1 relapse in the previous year while on DMD therapy and ⩾1 T1 Gd+ lesion or 9 T2 lesions. This was selected as a representative definition of HDA as there is no consensus on this measure at present.

RESULTS

Patients

- There were 1540 patients in total (**Table 2**; placebo, n=494, cladribine tablets 3.5 mg/kg, n=573, cladribine tablets 5.25 mg/kg, n=473).

Annualized relapse rates

- In the ITT population, the mean ARR (**Table 3**) in patients treated with cladribine tablets 3.5 and 5.25 mg/kg represented a 57% and a 59% reduction, respectively, compared with the mean ARR of the placebo group (RR ratio 0.43, 95% CI 0.35–0.52, $p<0.0001$, and 0.41, 95% CI 0.33–0.51, $p<0.0001$, respectively).

Table 2. Demographics and baseline characteristics of patients randomized to placebo or cladribine tablets 3.5 or 5.25 mg/kg in the double-blind treatment periods of CLARITY and ONWARD

Parameter	Placebo (n=494)	Cladribine tablets 3.5 mg/kg (n=573)	Cladribine tablets 5.25 mg/kg (n=473)
Age, years			
Mean (SD)	38.9 (9.9)	38.1 (10.3)	39.1 (9.9)
Median	39.0	38.0	40.0
Min; max	18; 64	18; 65	18; 65
Patients aged:			
⩽40 years, n (%)	277 (56.1)	340 (59.3)	253 (53.5)
>40 years, n (%)	217 (43.9)	233 (40.7)	220 (46.5)
Sex			
Male, n (%)	164 (33.2)	181 (31.6)	148 (31.3)
Female, n (%)	330 (66.8)	392 (68.4)	325 (68.7)
Disease duration, years			
Mean (SD)	5.5 (5.6)	5.2 (5.5)	5.1 (5.2)
Median	3.9	3.5	3.5
Min; max	0.1; 36.3	0.1; 38.1	0.0; 31.8
No prior DMD use, n (%)	305 (61.7)	323 (56.4)	320 (67.7)
EDSS score at baseline			
Mean (SD)	3.0 (1.3)	2.8 (1.2)	3.0 (1.4)
Median	3.0	2.5	3.0
Min; max	0.0; 5.5	0.0; 6.0	0.0; 5.5
Patients with EDSS score:			
⩽3, n (%)	292 (59.1)	361 (63.0)	260 (55.0)
⩾3.5, n (%)	202 (40.9)	212 (37.0)	213 (45.0)

DMD, disease-modifying drug; **EDSS**, Expanded Disability Status Scale; **Gd+**, gadolinium-enhancing; **SD** standard deviation.

Table 3. RR ratios for mean ARR and hazard ratios for time to 3- and 6-month confirmed EDSS progression in the ITT population in CLARITY and ONWARD

	Placebo (n=494)	Cladribine tablets 3.5 mg/kg (n=573)	Cladribine tablets 5.25 mg/kg (n=473)
Qualifying relapses, ITT population			
Number of qualifying relapses, mean (SD)	0.58 (0.92)	0.25 (0.58)	0.25 (0.57)
ARR (adjusted)	0.33	0.14	0.14
95% CI	0.30; 0.38	0.12; 0.17	0.11; 0.17
% reduction (cladribine tablets/placebo)		57.2	59.0
95% CI		47.63; 64.99	49.11; 66.98
Relative risk ratio		0.43	0.41
95% CI		0.35; 0.52	0.33; 0.51
p value		<0.0001	<0.0001
Time to 3-month confirmed EDSS progression, ITT population			
Subjects with event, n (%)	106 (21.5)	84 (14.7)	70 (14.8)
K-M estimate at last event	77.1	84.7	84.3
95% CI	73.0; 80.7	81.4; 87.5	80.6; 87.4
Hazard ratio (cladribine tablets/placebo)		0.64	0.63
95% CI		0.48; 0.86	0.47; 0.86
p value		0.0028	0.0033
Time to 6-month confirmed EDSS progression, ITT population			
Subjects with event, n (%)	75 (15.2)	56 (9.8)	54 (11.4)
K-M estimate at last event	84.0	89.8	87.9
95% CI	80.3; 87.0	87.0; 92.1	84.5; 90.6
Hazard ratio (cladribine tablets/placebo)		0.61	0.70
95% CI		0.43; 0.87	0.49; 0.99
p value		0.0058	0.0446

ARR, annualized relapse rate; **CI**, confidence interval; **EDSS**, Expanded Disability Status Scale; **ITT**, intent-to-treat; **K-M**, Kaplan-Meier; **SD** standard deviation.

- Consistent findings were seen in the subgroups, including the HDA subgroup; each showed a significantly lower RR ratio with cladribine tablets 3.5 mg/kg than placebo ($p<0.001$ for each comparison, **Figure 2**). The only exception to this was the subgroup patients with no relapses in the previous year, in which patient numbers were too low for analysis. Similar results were seen in subgroups of patients treated with cladribine tablets 5.25 mg/kg (data not shown).

Figure 2. Effects of cladribine tablets 3.5 mg/kg versus placebo on ARR in patient subgroups in CLARITY and ONWARD

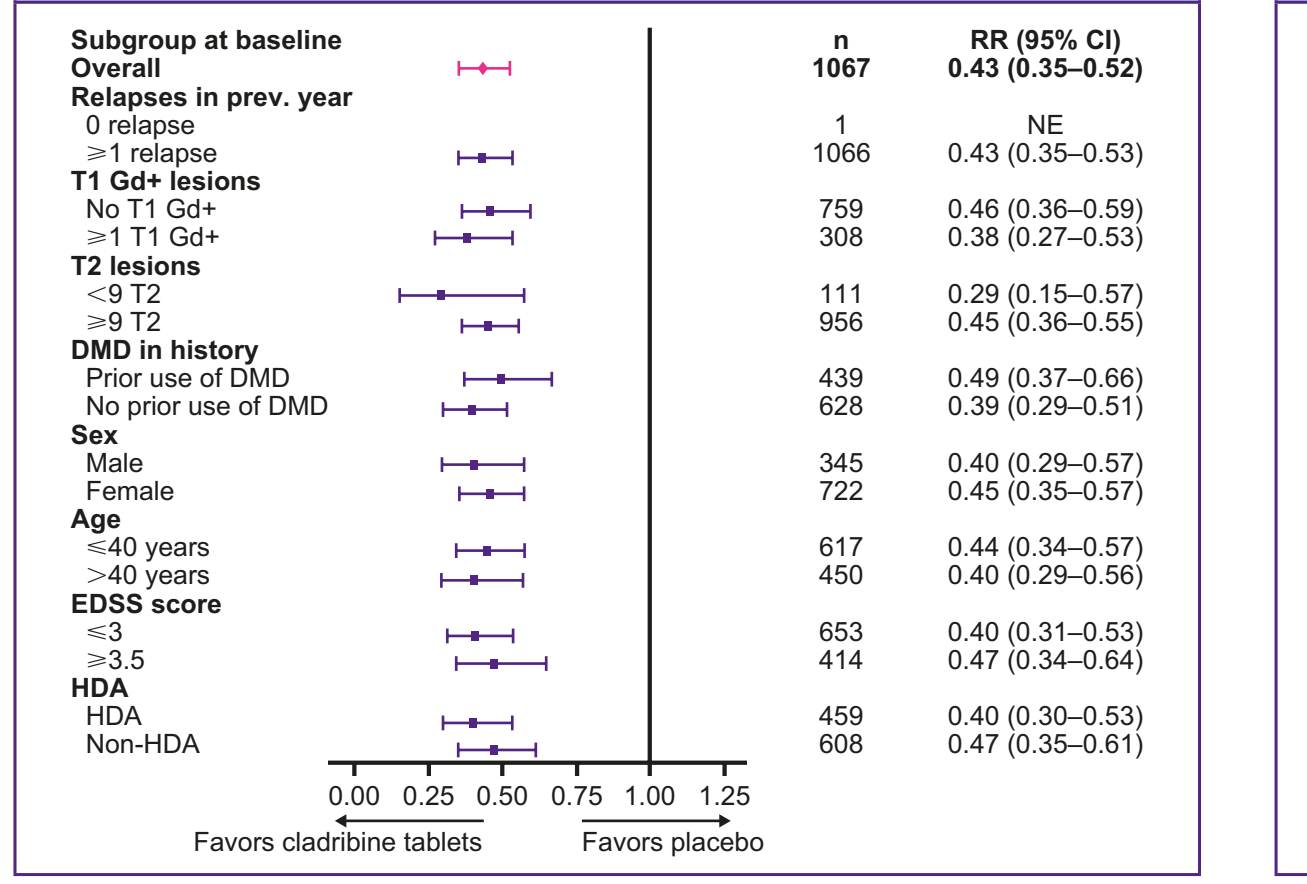

Subgroup at baseline | n | RR (95% CI)
Overall | 1067 | 0.43 (0.35–0.52)
Relapses in prev. year
0 relapse | 1 | NE
⩾1 relapse | 1066 | 0.43 (0.35–0.53)
T1 Gd+ lesions
No T1 Gd+ | 759 | 0.46 (0.36–0.59)
⩾1 T1 Gd+ | 308 | 0.38 (0.27–0.53)
T2 lesions
<9 T2 | 111 | 0.29 (0.15–0.57)
⩾9 T2 | 956 | 0.45 (0.36–0.55)
DMD in history
Prior use of DMD | 439 | 0.49 (0.37–0.66)
No prior use of DMD | 628 | 0.39 (0.29–0.51)
Sex
Male | 345 | 0.40 (0.29–0.57)
Female | 722 | 0.45 (0.35–0.57)
Age
⩽40 years | 617 | 0.44 (0.34–0.57)
>40 years | 450 | 0.40 (0.29–0.56)
EDSS score
⩽3 | 653 | 0.40 (0.31–0.53)
⩾3.5 | 414 | 0.47 (0.34–0.64)
HDA
HDA | 459 | 0.40 (0.30–0.53)
Non-HDA | 608 | 0.47 (0.35–0.61)
0.00 0.25 0.50 0.75 1.00 1.25
Favors cladribine tablets
Favors placebo

ARR, annualized relapse rate; **CI**, confidence interval; **DMD**, disease-modifying drug; **EDSS**, Expanded Disability Status Scale; **Gd+**, gadolinium-enhancing; **HDA**, high disease activity (⩾2 relapses in previous year regardless of treatment status, or ⩾1 relapse in the previous year while on DMD therapy and ⩾1 T1 Gd+ lesion or 9 T2 lesions); **NE**, not estimable; **RR**, relative risk.

Time to 3-month EDSS progression

- In the ITT population, time to 3-month confirmed EDSS progression was significantly reduced with cladribine tablets 3.5 mg/kg and 5.25 mg/kg compared with placebo (hazard ratio 0.64, 95% CI 0.48–0.86, p=0.0028; and 0.63, 95% CI 0.47–0.86, p=0.0033, respectively, **Table 3**).
- Consistent findings were seen in a majority of the subgroups analyzed, including the HDA subgroup (**Figure 3**). Similar results were also seen in subgroups of patients treated with cladribine tablets 5.25 mg/kg (data not shown).

Figure 3. Effects of cladribine tablets 3.5 mg/kg versus placebo on time to 3-month confirmed EDSS progression in patient subgroups

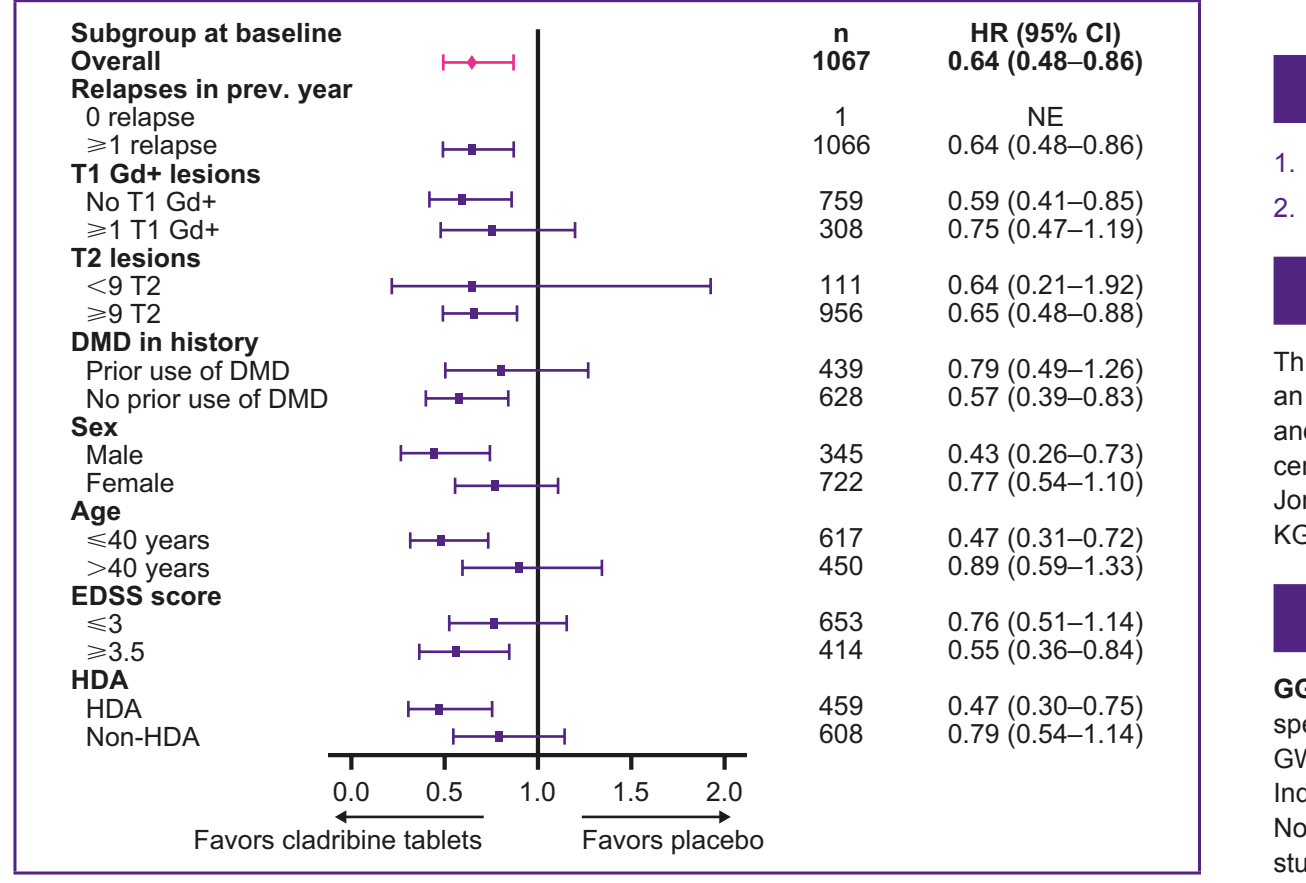

Subgroup at baseline | n | HR (95% CI)
Overall | 1067 | 0.64 (0.48–0.86)
Relapses in prev. year
0 relapse | 1 | NE
⩾1 relapse | 1066 | 0.64 (0.48–0.86)
T1 Gd+ lesions
No T1 Gd+ | 759 | 0.59 (0.41–0.85)
⩾1 T1 Gd+ | 308 | 0.75 (0.47–1.19)
T2 lesions
<9 T2 | 111 | 0.64 (0.21–1.92)
⩾9 T2 | 956 | 0.65 (0.48–0.88)
DMD in history
Prior use of DMD | 439 | 0.79 (0.49–1.26)
No prior use of DMD | 628 | 0.57 (0.39–0.83)
Sex
Male | 345 | 0.43 (0.26–0.73)
Female | 722 | 0.77 (0.54–1.10)
Age
⩽40 years | 617 | 0.47 (0.31–0.72)
>40 years | 450 | 0.89 (0.59–1.33)
EDSS score
⩽3 | 653 | 0.76 (0.51–1.14)
⩾3.5 | 414 | 0.55 (0.36–0.84)
HDA
HDA | 459 | 0.47 (0.30–0.75)
Non-HDA | 608 | 0.79 (0.54–1.14)
0.0 0.5 1.0 1.5 2.0
Favors cladribine tablets
Favors placebo

CI, confidence interval; **DMD**, disease-modifying drug; **EDSS**, Expanded Disability Status Scale; **Gd+**, gadolinium-enhancing; **HDA**, high disease activity (⩾2 relapses in previous year regardless of treatment status, or ⩾1 relapse in the previous year while on DMD therapy and ⩾1 T1 Gd+ lesion or 9 T2 lesions); **HR**, hazard ratio; **NE**, not estimable.

Time to 6-month EDSS progression

- In the ITT population, time to 6-month confirmed EDSS progression was significantly reduced with cladribine tablets 3.5 mg/kg and 5.25 mg/kg compared with placebo (hazard ratio 0.61, 95% CI 0.43–0.87, p=0.0058; and 0.70, 95%CI 0.49–0.99, p=0.0446, respectively, **Table 3**).
- Consistent findings were seen in the majority of the subgroups analysed, including the HDA subgroup (**Figure 4**). Similar results were seen in subgroups of patients treated with cladribine tablets 5.25 mg/kg (data not shown).

Figure 4. Effects of cladribine tablets 3.5 mg/kg versus placebo on time to 6-month confirmed EDSS progression in patient subgroups

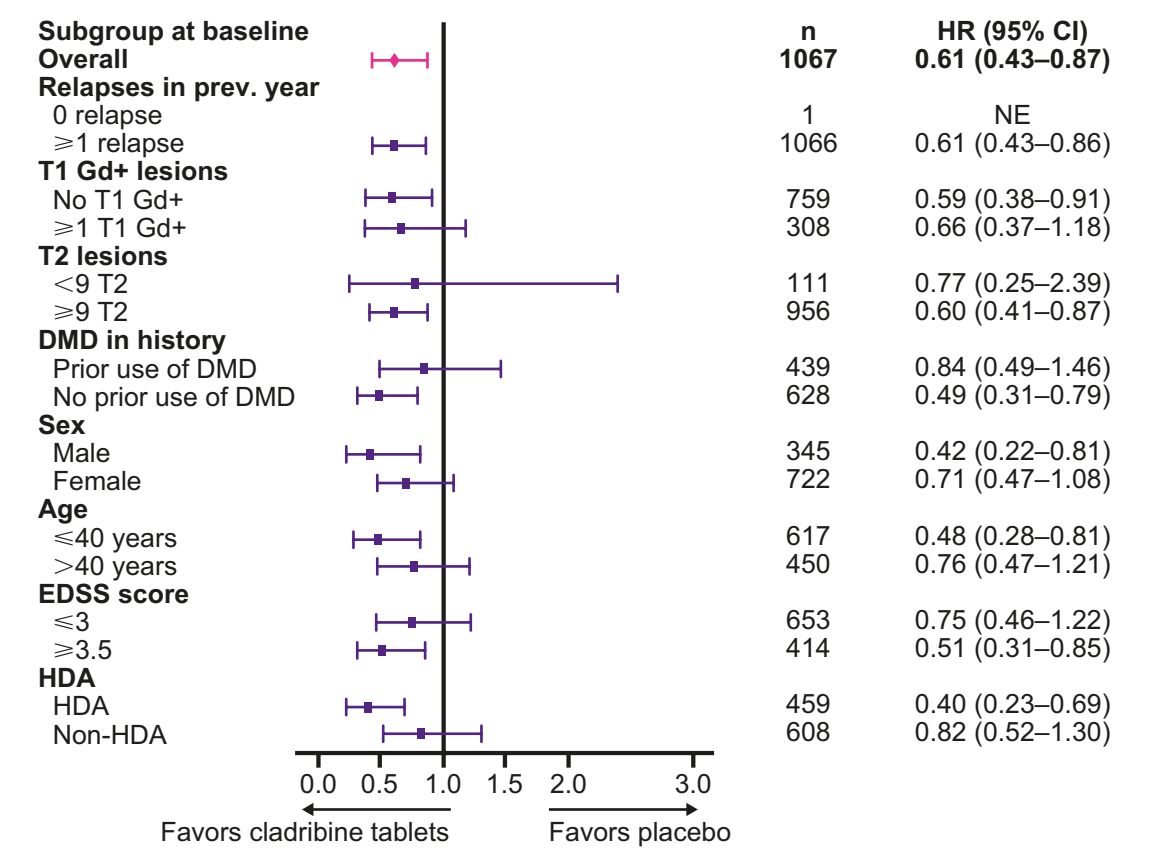

Subgroup at baseline | n | HR (95% CI)
Overall | 1067 | 0.61 (0.43–0.87)
Relapses in prev. year
0 relapse | 1 | NE
⩾1 relapse | 1066 | 0.61 (0.43–0.86)
T1 Gd+ lesions
No T1 Gd+ | 759 | 0.59 (0.38–0.91)
⩾1 T1 Gd+ | 308 | 0.66 (0.37–1.18)
T2 lesions
<9 T2 | 111 | 0.77 (0.25–2.39)
⩾9 T2 | 956 | 0.60 (0.41–0.87)
DMD in history
Prior use of DMD | 439 | 0.84 (0.49–1.46)
No prior use of DMD | 628 | 0.49 (0.31–0.79)
Sex
Male | 345 | 0.42 (0.22–0.81)
Female | 722 | 0.71 (0.47–1.08)
Age
⩽40 years | 617 | 0.48 (0.28–0.81)
>40 years | 450 | 0.76 (0.47–1.21)
EDSS score
⩽3 | 653 | 0.75 (0.46–1.22)
⩾3.5 | 414 | 0.51 (0.31–0.85)
HDA
HDA | 459 | 0.40 (0.23–0.69)
Non-HDA | 608 | 0.82 (0.52–1.30)
0.0 0.5 1.0 1.5 2.0 3.0
Favors cladribine tablets
Favors placebo

CI, confidence interval; **DMD**, disease-modifying drug; **EDSS**, Expanded Disability Status Scale; **Gd+**, gadolinium-enhancing; **HDA**, high disease activity (⩾2 relapses in previous year regardless of treatment status, or ⩾1 relapse in the previous year while on DMD therapy and ⩾1 T1 Gd+ lesion or 9 T2 lesions); **HR**, hazard ratio; **NE**, not estimable.

CONCLUSIONS

- **Analysis of pooled data from CLARITY and ONWARD showed that cladribine tablets significantly decreased ARR and reduced the risk for 3- and 6-month confirmed disability progression vs placebo in patients with active RMS.**
 - **The benefits on relapse outcomes were consistent across all patient subgroups. Point estimates for EDSS progression also indicated consistent effect. Benefits were seen across a range of patient subgroups, including patients with clinical and MRI indications of high disease activity.**

REFERENCES

1. Giovannoni G, *et al. N Engl J Med* 2010;362:416-26.
2. Montalban X, *et al. Neurology* 2013;80:P07-09 [Abstract].

ACKNOWLEDGMENTS

This study was sponsored by EMD Serono, Inc.* (in the USA) and Merck Serono SA – Geneva, an affiliate of Merck KGaA Darmstadt, Germany (ROW). The authors would like to thank patients and their families, investigators, co-investigators, and the study teams at each of the participating centers and at Merck KGaA, Darmstadt, Germany. Medical writing assistance was provided by Phil Jones of inScience Communications, Springer Healthcare, Chester, UK, and was funded by Merck KGaA, Darmstadt, Germany.

DISCLOSURES

GG serves on advisory boards for Merck, Biogen Idec, and Vertex Pharmaceuticals; has received speaker honoraria and consulting fees from Bayer Schering Pharma, FivePrime, GlaxoSmithKline, GW Pharma, Merck, Biogen Idec, Pfizer Inc, Protein Discovery Laboratories, Teva Pharmaceutical Industries Ltd, Sanofi-Aventis, UCB, Vertex Pharmaceuticals, Genzyme Corporation, Ironwood, and Novartis; serves on the Merck speakers bureau; and received research support unrelated to this study from Biogen Idec, Merck, Novartis, and Ironwood. **XM** has received speaker honoraria and travel expenses for scientific meetings, steering committee member, and advisory board member of clinical trials for Bayer Schering Pharma, Biogen Idec, EMD Serono, Genentech, Genzyme, Novartis, Roche, Sanofi-Aventis, Teva Pharmaceuticals, and Almirall. **CH** is an employee of Merck KGaA, Darmstadt, Germany. **FD** is an employee of EMD Serono, Inc.,* Billerica, MA, USA.

Cladribine tablets is currently under clinical investigation and has not yet been approved by any regulatory authority. Status: March 2017.

The CLARITY study: NCT00213135, the ONWARD study NCT00436826.

*A business of Merck KGaA, Darmstadt, Germany.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

GET POSTER PDF

Presented at the 31st Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC); May 24–27, 2017; New Orleans, LA, USA