5446

Dosing of abobotulinumtoxinA (Dysport®) injections for adults with lower limb spasticity

Salvatore Napoli,[1] Regina Berkovich,[2] Theodore Brown,[3] Ziyad Ayyoub,[4,5,6] Gustavo Suarez,[7] Philippe Picaut,[8] Peter Hedera[9]

[1]Neurology Center of New England, Foxborough, MA, USA; [2]MS Comprehensive Care Center and Research Group, University of Southern California, Los Angeles, CA, USA; [3]EvergreenHealth Multiple Sclerosis Center, Kirkland, WA, USA; [4]Rancho Los Amigos National Rehabilitation Center, Downey, CA, USA; [5]UCLA David Geffen School of Medicine at UCLA, Los Angeles, CA, USA; [6]Western University of Heath Sciences, Pomona, CA, USA; [7]Ipsen Biopharmaceuticals, Basking Ridge, NJ, USA; [8]Ipsen Pharma, Les Ulis, France; [9]Department of Neurology, Division of Movement Disorders, Vanderbilt University, Nashville, TN, USA

Scan here to view a PDF of this poster

Copies of this poster obtained through Quick Response Code are for personal use only

Background

- Spasticity is a common and frequently disabling symptom of multiple sclerosis, associated with functional impairment that can affect quality of life.[1]
- Spasticity is also common in patients who have suffered stroke or traumatic brain injury (TBI).[2,3]
- AbobotulinumtoxinA (aboBoNT-A, Dysport®) has recently received approval in the United States for the treatment of adult spasticity of any etiology.[4]
- This phase 3, double-blind, randomized trial (NCT01249404) assessed the efficacy of aboBoNT-A in treating lower limb spasticity in hemiparetic patients following stroke or TBI.
- This analysis describes aboBoNT-A doses injected in muscles that may also be involved in spasticity related to multiple sclerosis.

Methods

Study population

- Inclusion criteria included:
 - Adults (18–80 years) with spastic hemiparesis of the lower limb, at least 6 months post-stroke or -TBI.
 - Patients who had only one clinically defined stroke episode or one brain trauma at least 6 months prior to study entry.
 - Modified Ashworth Scale (MAS) score ≥2 or ≥3 in the affected gastrocnemius soleus complex (GSC) with knee extended, for toxin-naïve and non-naïve (at least 4 months since last botulinum toxin injection in affected lower limb) patients, respectively.
 - Comfortable barefoot walking speed between 0.1 m/s and 0.8 m/s at baseline measured using a 10-meter comfortable walking speed test (WST) without walking aids.
 - Spasticity angle ≥5° for the GSC of the affected leg measured by the Tardieu Scale with the knee extended.
- Exclusion criteria included:
 - Major limitation in the passive range of motion at the affected hip, knee, or ankle.
 - Physiotherapy initiated less than 4 weeks before entry or expected to be initiated during the trial.
 - Severe neurologic impairment (not associated with the stroke or brain trauma) due to underlying neuromuscular disease or any other underlying disease or condition affecting gait (e.g. multiple sclerosis).
 - Current or planned treatment with any drug that interferes directly or indirectly with neuromuscular function (e.g. aminoglycosides) within the last 4 weeks prior to study treatment.

Study design and treatment

- This was a phase 3, multicenter, prospective, double-blind, randomized, placebo-controlled, single-treatment cycle study.
- Patients were recruited from 53 centers in Australia, Belgium, the Czech Republic, France, Hungary, Italy, Poland, Portugal, Russia, Slovakia, and the United States.
- Patients were randomized into one of three treatment groups (aboBoNT-A 1000 units [U] or 1500 U, or placebo) at a ratio of 1:1:1 (**Figure 1**).
- Intramuscular injections were administered into the GSC, plus at least one additional distal or proximal muscle.
- All patients received a single treatment cycle at Week 0.
- Change from baseline in GSC muscle tone (assessed by the MAS) at Week 4 was the primary endpoint of the study.
 - Analyzed using a single mixed-effect analysis of covariance model.
- The intent-to-treat (ITT) population included all randomized patients who received at least one injection and who had a MAS score in the GSC assessed at baseline and Week 4.
- The safety population was defined as all randomized subjects who received at least one injection of study medication.

Results

Patient disposition and characteristics

- Of 456 patients who attended screening, 388 were enrolled and randomized (**Figure 1**).
- Patient characteristics were similar between groups (**Table 1**).

Change from baseline in the MAS score

- Mean standard deviation (SD) changes in the MAS score for GSC (knee extended) and the soleus muscle only (knee flexed) are shown (**Figure 2**).

Table 1. Patient characteristics.

Parameter	aboBoNT-A 1000 U (N=125)	aboBoNT-A 1500 U (N=128)	Total aboBoNT-A (N=253)	Placebo (N=128)
Age, mean (SD) years	53.2 (13.2)	53.3 (12.0)	53.3 (12.6)	51.4 (12.9)
Sex, men (%)	87 (69.6)	79 (61.7)	166 (65.6)	90 (70.3)
Ethnicity, n (%)				
Hispanic	14 (11.2)	11 (8.6)	25 (9.9)	11 (8.6)
Non-Hispanic	111 (88.8)	117 (91.4)	228 (90.1)	117 (91.4)
BMI, mean (SD) kg/m²	27.3 (5.0)	27.3 (4.1)	27.3 (4.6)	27.4 (5.2)
Cause of spasticity, n (%)				
Stroke	109 (87.2)	116 (90.6)	225 (88.9)	106 (82.8)
Traumatic brain injury	16 (12.8)	12 (9.4)	28 (11.1)	22 (17.2)
Time since stroke, median (range) years	2.7 (0.6, 27.3)	2.6 (0.6, 30.8)	2.7 (0.6, 30.8)	3.0 (0.5, 16.6)
Time since traumatic brain injury, median (range) years	4.2 (1, 28)	7.5 (2, 19)	5.2 (1, 28)	5.8 (1, 55)
Treatment-naïve, n (%)				
Total	82 (65.6)	80 (62.5)	162 (64.0)	81 (63.3)
US centers	17 (77.3)	13 (61.9)	30 (68.9)	15 (62.5)
Centers in other countries	65 (63.1)	67 (62.6)	132 (62.9)	66 (63.5)
Not treatment-naïve, n (%)				
Total	43 (34.4)	48 (37.5)	91 (36.0)	47 (36.7)
US centers	5 (22.7)	8 (38.1)	13 (30.2)	9 (37.5)
Centers in other countries	38 (36.9)	40 (37.4)	78 (37.1)	38 (36.5)

AboBoNT-A, abobotulinumtoxinA; BMI, body mass index; SD, standard deviation; U, units; US, United States.

Figure 1. Study design and patient disposition.

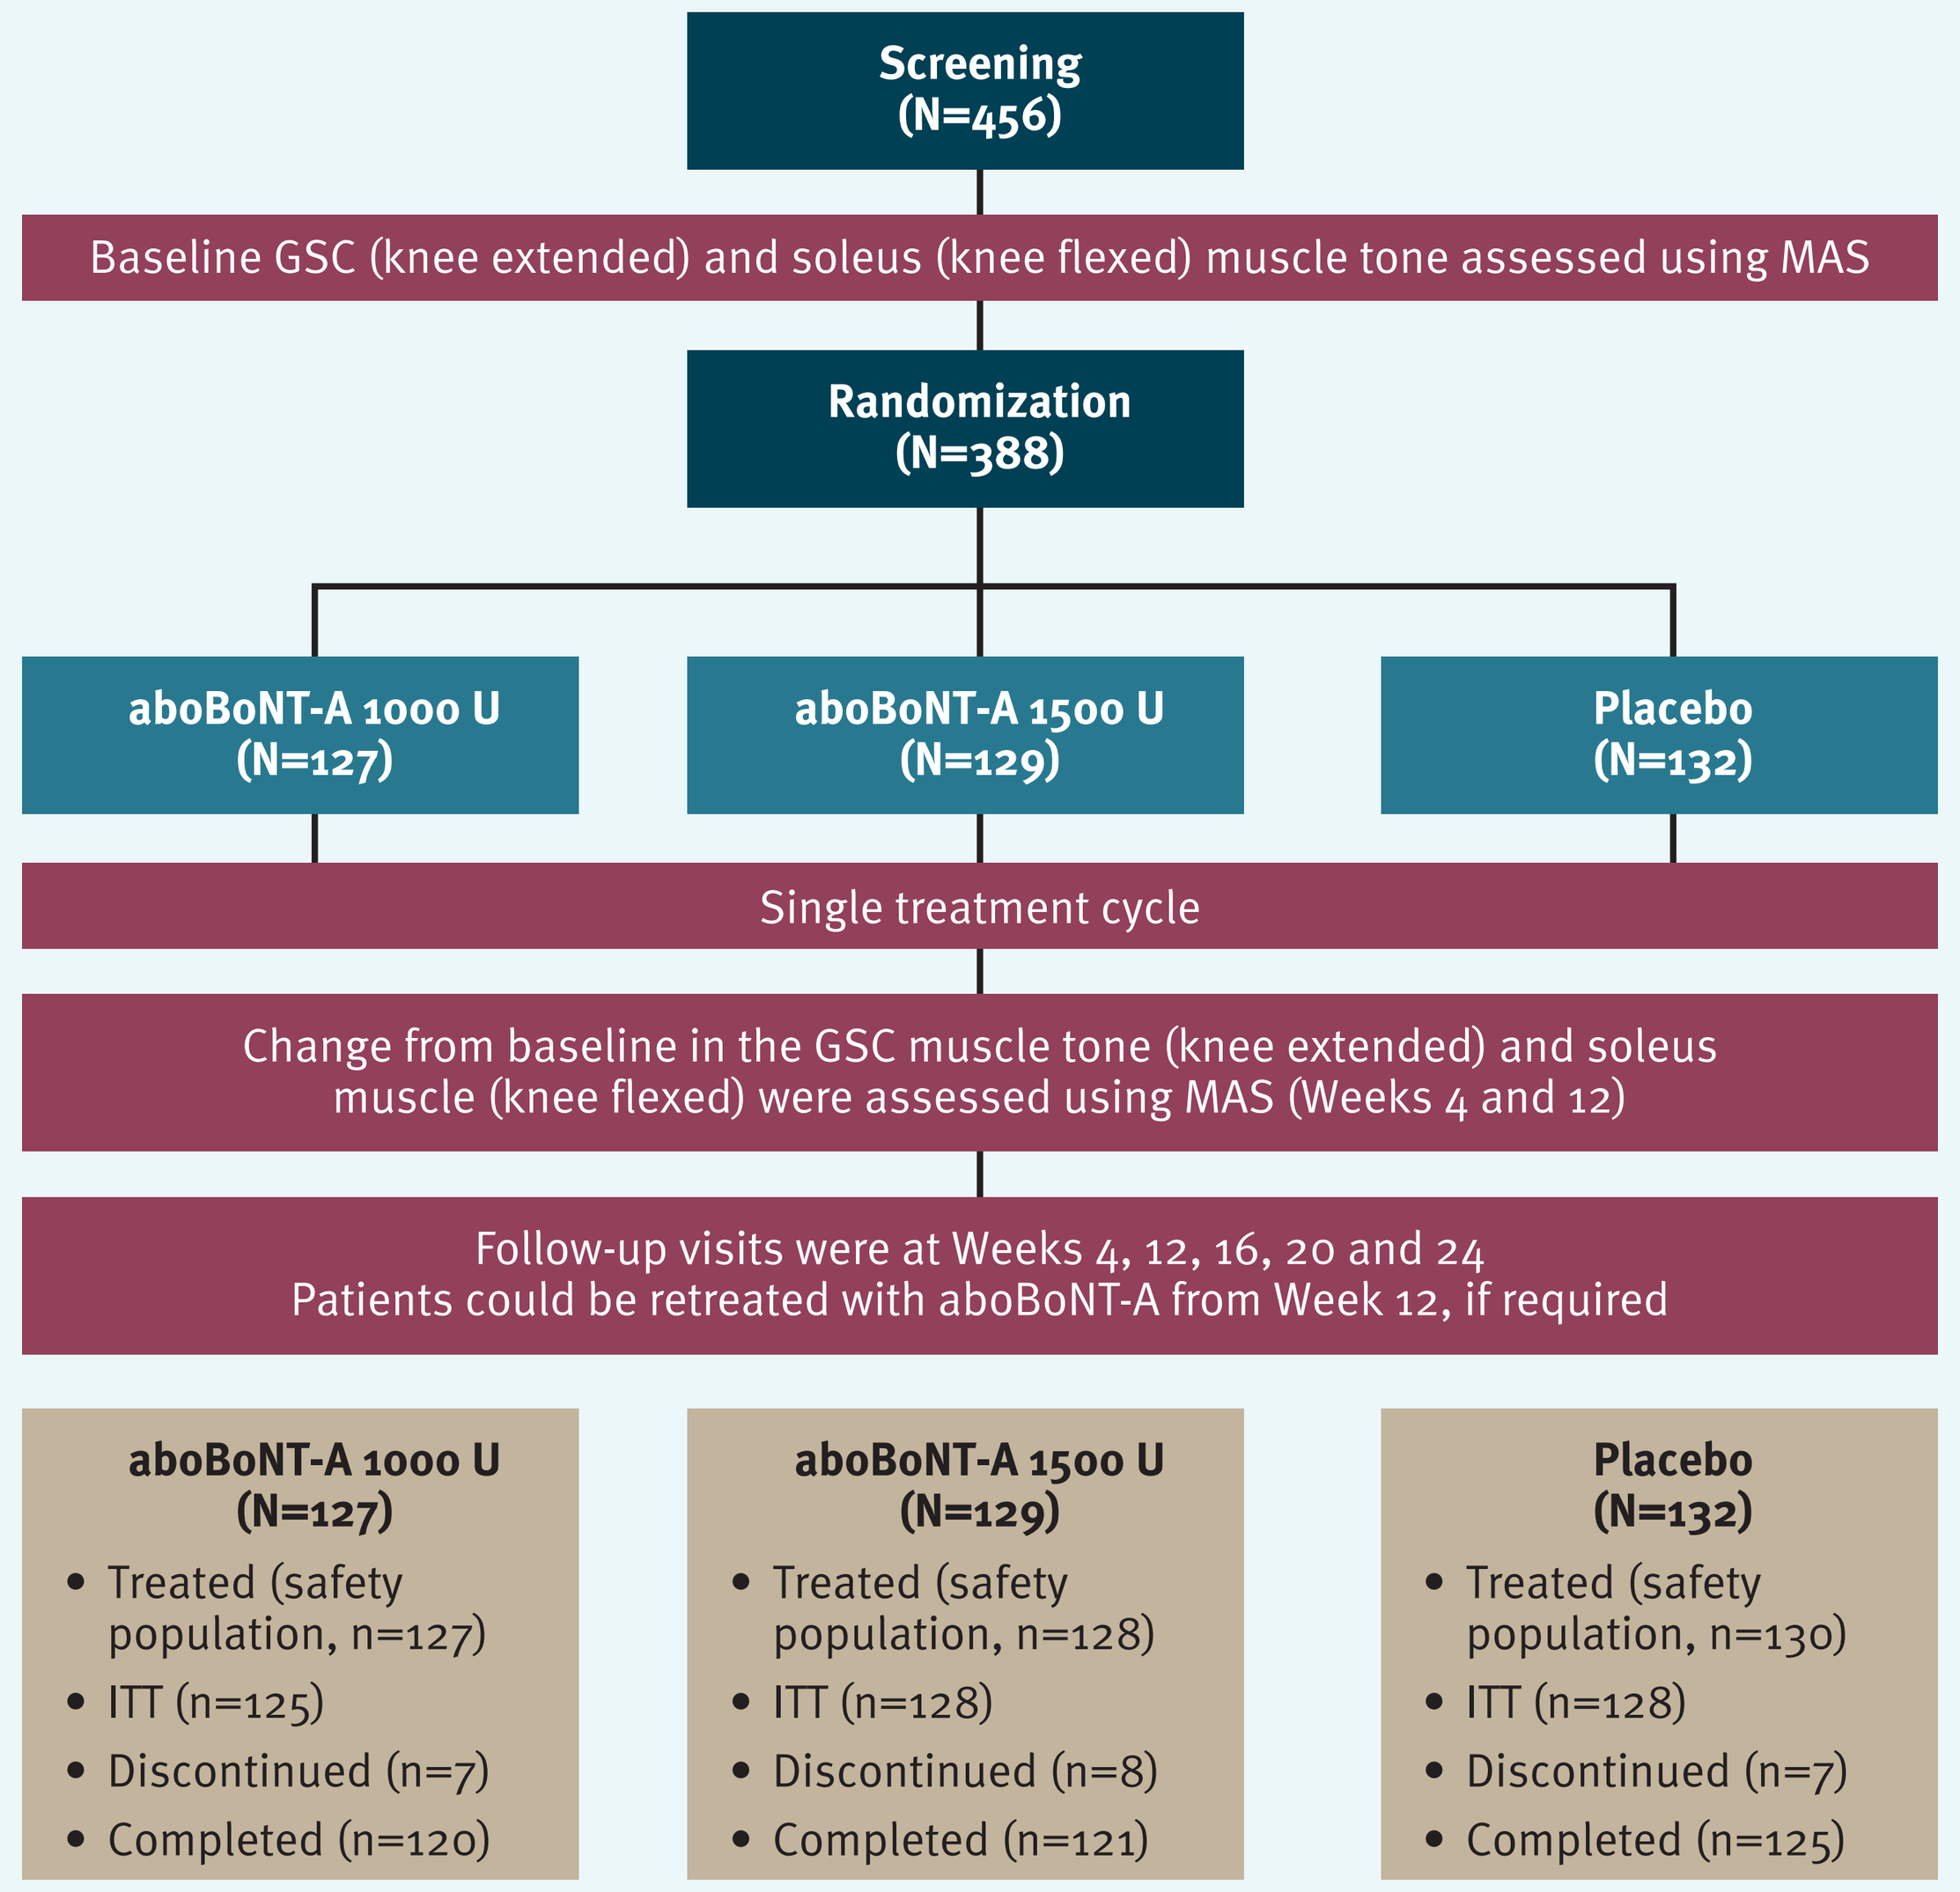

Screening (N=456)
Baseline GSC (knee extended) and soleus (knee flexed) muscle tone assessed using MAS
Randomization (N=388)
aboBoNT-A 1000 U (N=127)
aboBoNT-A 1500 U (N=129)
Placebo (N=132)
Single treatment cycle
Change from baseline in the GSC muscle tone (knee extended) and soleus muscle (knee flexed) were assessed using MAS (Weeks 4 and 12)
Follow-up visits were at Weeks 4, 12, 16, 20 and 24
Patients could be retreated with aboBoNT-A from Week 12, if required
aboBoNT-A 1000 U (N=127)
• Treated (safety population, n=127)
• ITT (n=125)
• Discontinued (n=7)
• Completed (n=120)
aboBoNT-A 1500 U (N=129)
• Treated (safety population, n=128)
• ITT (n=128)
• Discontinued (n=8)
• Completed (n=121)
Placebo (N=132)
• Treated (safety population, n=130)
• ITT (n=128)
• Discontinued (n=7)
• Completed (n=125)

AboBoNT-A, abobotulinumtoxinA; GSC, gastrocnemius soleus complex; ITT, intent-to-treat population; MAS, Modified Ashworth Scale; U, units.

Figure 2. Change from baseline in Modified Ashworth Scale score.

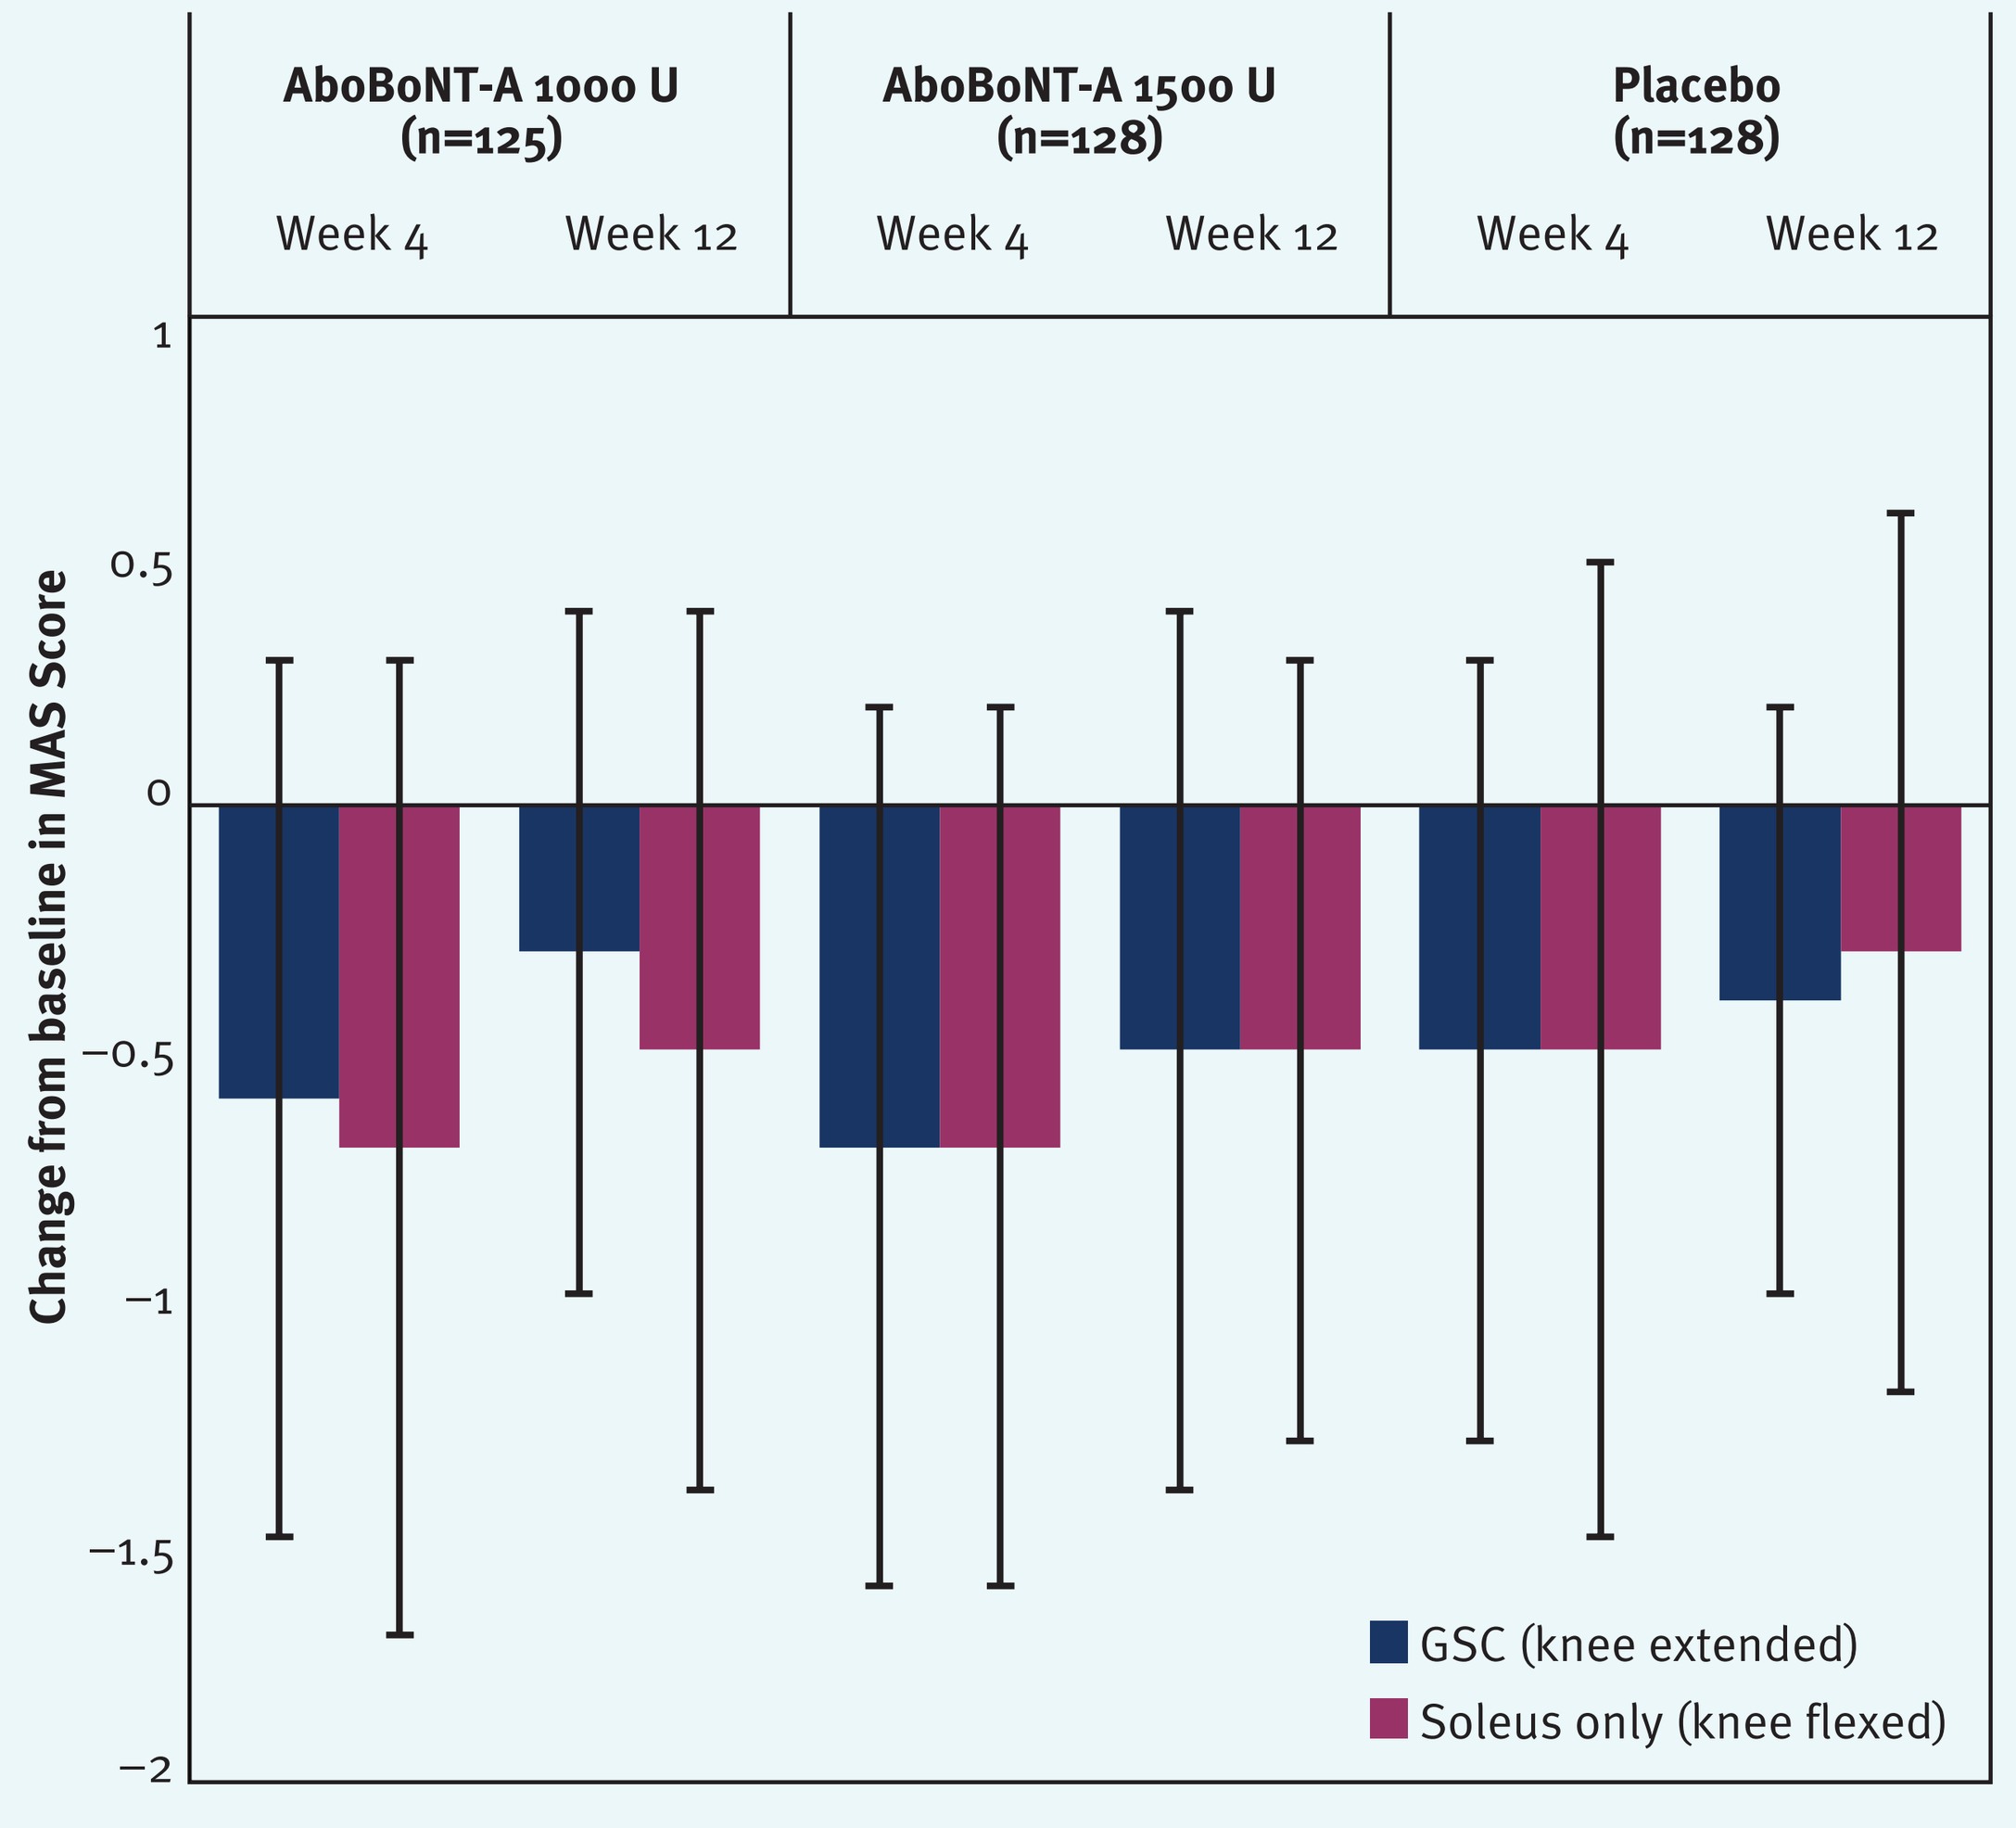

AboBoNT-A 1000 U (n=125)
AboBoNT-A 1500 U (n=128)
Placebo (n=128)
Week 4
Week 12
Week 4
Week 12
Week 4
Week 12
Change from baseline in MAS Score
1
0.5
0
−0.5
−1
−1.5
−2
GSC (knee extended)
Soleus only (knee flexed)

Error bars show standard deviation.
GSC, gastrocnemius soleus complex; MAS, Modified Ashworth Scale; U, units

Table 2. Dose ranges injected per muscle for each aboBoNT-A treatment group.

	Dose ranges injected per muscle	
	aboBoNT-A 1000 U (N=127)	**aboBoNT-A 1500 U (N=128)**
Distal muscles, data shown as units (number of patients)		
Flexor digitorum brevis	53–133 (34)	50–300 (38)
Flexor digitorum longus	67–267 (90)	40–400 (88)
Flexor hallucis brevis	67–133 (20)	50–200 (19)
Flexor hallucis longus	53–267 (46)	60–300 (44)
Lateral gastrocnemius	67–200 (108)	100–300 (105)
Medial gastrocnemius	67–200 (125)	0–300 (127)
Soleus	333–333 (127)	0–500 (128)
Tibialis posterior	67–467 (95)	100–700 (91)
Proximal muscles, data shown as units (number of patients)		
Adductor magnus	133–267 (8)	200–400 (7)
Gluteus maximus	67–133 (2)	100–400 (5)
Gracilis	67–133 (3)	100–200 (6)
Hamstrings	67–333 (16)	100–550 (24)
Rectus femoris	67–467 (63)	100–700 (50)

aboBoNT-A, abobotulinumtoxinA; U, units.

Duration of effect

- The majority of patients required retreatment 12 weeks after the first aboBoNT-A injection.

Dose ranges injected per muscle

- Dose ranges injected per muscle (safety population) are shown for distal and proximal muscles (**Table 2**) for aboBoNT-A 1000 U vs. 1500 U, respectively, and included:
 - 67–267 vs. 40–400 U administered to the flexor digitorum longus
 - 67–133 vs. 50–200 U administered to the flexor hallucis brevis
 - 67–200 vs. 100–300 U administered to the lateral gastrocnemius
 - 67–200 vs. 0–300 U administered to the medial gastrocnemius
 - 333–333 vs. 0–500 U administered to the soleus
 - 67–467 vs. 100–700 U administered to the tibialis posterior.
- The dose range given to patients in any single distal or proximal muscle in the aboBoNT-A 1000 U group was 53–467 U; patients in the aboBoNT-A 1500 U group received between 0 and 700 U in any single muscle.
- The total dose range given in the aboBoNT-A 1000 U group was 867–1000 U; patients in the aboBoNT-A 1500 U group received between 800 and 1500 U.

Safety

- Both doses of aboBoNT-A were well tolerated.
- The most frequent adverse events were falls, pain in extremity, and muscular weakness (which was localized weakness in the injected muscles in the placebo and aboBoNT-A 1000 U groups).
- The incidence of muscular weakness was higher in the aboBoNT-A 1500 U group (6.3%) than in the aboBoNT-A 1000 U (2.4%) or placebo groups (3.1%).
- The development of binding or neutralizing antibodies was not detected following aboBoNT-A injection.

Conclusions

- The information on dosing for lower limb spasticity in patients with stroke or TBI may be considered for dosing in patients with lower limb spasticity due to multiple sclerosis.
- The results of this phase 3 randomized study demonstrate the efficacy of aboBoNT-A (approved in the US for any etiology) in patients with lower limb spasticity.
- Safety was consistent with the known profile of aboBoNT-A.

References

1. Patejdl *et al. Autoimm Rev* 2017;9:925–36.
2. Gracies. *Muscle Nerve* 2005;31:535–51.
3. Gracies. *Muscle Nerve* 2005;31:552–71.
4. FDA 2017. https://www.accessdata.fda.gov/drugsatfda_docs/label/2017/125274s109lbl.pdf

Acknowledgments

The authors thank the investigators and patients participating in this study. The authors also thank Watermeadow Medical for medical writing support in preparing this poster.

Conflicts of interest

SN is a consultant for: Acorda Therapeutics; Biogen; Novartis; Pfizer; Questcor Pharmaceuticals; Sanofi Genzyme; Serono; and Teva Neuroscience. RB is a consultant for Bayer AG and Mallinckrodt Pharmaceuticals, and is a consultant and member of a speakers' bureau for: Acorda Therapeutics; Avanir Pharmaceuticals, Inc; Biogen; Novartis AG; Sanofi Genzyme; and Teva Pharmaceutical Industries Ltd. ZA is a consultant, member of a speakers' bureau, and a recipient of research grants from Ipsen, Allergan, Merz and US WorldMeds. TB is the recipient of a research grant from Astellas, Merck and Biogen, and is a consultant for Ipsen, Biogen, Sanofi Genzyme and Teva. GS is an employee of Ipsen. PP has served as an expert medical witness for Patrick, Beard, Schulman, and Jacoway, PC and The Talaska Law Firm, PLLC, and is an employee of Ipsen. PH is a recipient of royalties from Elsevier Publishing and is a member of a speakers' bureau for Teva and Ipsen.

Presented at the Annual Meeting of the Consortium of Multiple Sclerosis Centers, 30 May – 2 June 2018, Nashville, TN, USA.

This study was sponsored by Ipsen Pharma SAS