DX31

The Neurology Group
Neurologists - EEG, EMG, NCV

Stevens-Johnson Syndrome Associated with Daclizumab Use- Report of Two Cases

Ronald Bailey, M.D.[1], Marco Pasco, M.PH[1], and Shard Yaqoot, R.N.[2]
[1]The Neurology Group, Pomona, CA, [2]CA Specialty Pharmacy, Whittier, CA

CALIFORNIA SPECIALTY™ PHARMACY
Discover the difference of an independent specialty pharmacy.™

OBJECTIVES

To report a potentially lethal systemic dermatologic complication associated with Daclizumab (DAC) use.

BACKGROUND

- 11% of patients using DAC report rash;
- Autoimmune disorders including hepatitis associated with liver failure have been reported.
- Stevens-Johnson (SJS) has not been reported with use of DAC
- SJS together with toxic epidermal necrolysis (TEN) form a spectrum of dermatologic disease

METHODS

Two cases of SJS associated with the use of DAC were evaluated.

RESULTS

Case 1. A 52 year old black woman with MS complained of flu-like symptoms and had low grade pyrexia after 5 injections of DAC. She was taking no other medications. REMS blood work was normal up until the event. Her myelopathy worsened and EDSS score deteriorated from 2.5 to 4.5. She developed a confluent macular rash over her face, torso, back, and limbs. The rash was pruritic.

Fig. 1: Confluent macular rash over the back of patient following 5th. Injection of DAC associated with intense pruritis

After several days the patient developed pronounced alopecia and blister-like bullous lesions on her skin that formed raw areas with intense pain. Cystic lesions were also present in the oropharynx.

Fig. 2: Severe alopecia because of DAC use

Fig.3: Rupture of large bullous cystic lesion in the palm of the hand

Fever persisted with temperature spikes to 103 degrees, WBC count rose to 18.6 with shift, and a sedimentation rate was 84. She declined hospitalization but was treated with high dose steroids and antihistamines. Over several weeks blood labs normalized and with resolution of leukocytosis. The patient's skin exfoliated, and alopecia gradually resolved over weeks. No super infections occurred. Several skin biopsies were performed and revealed spongiotic and interface dermatitis with histologic features consistent with drug reaction.

Case 1

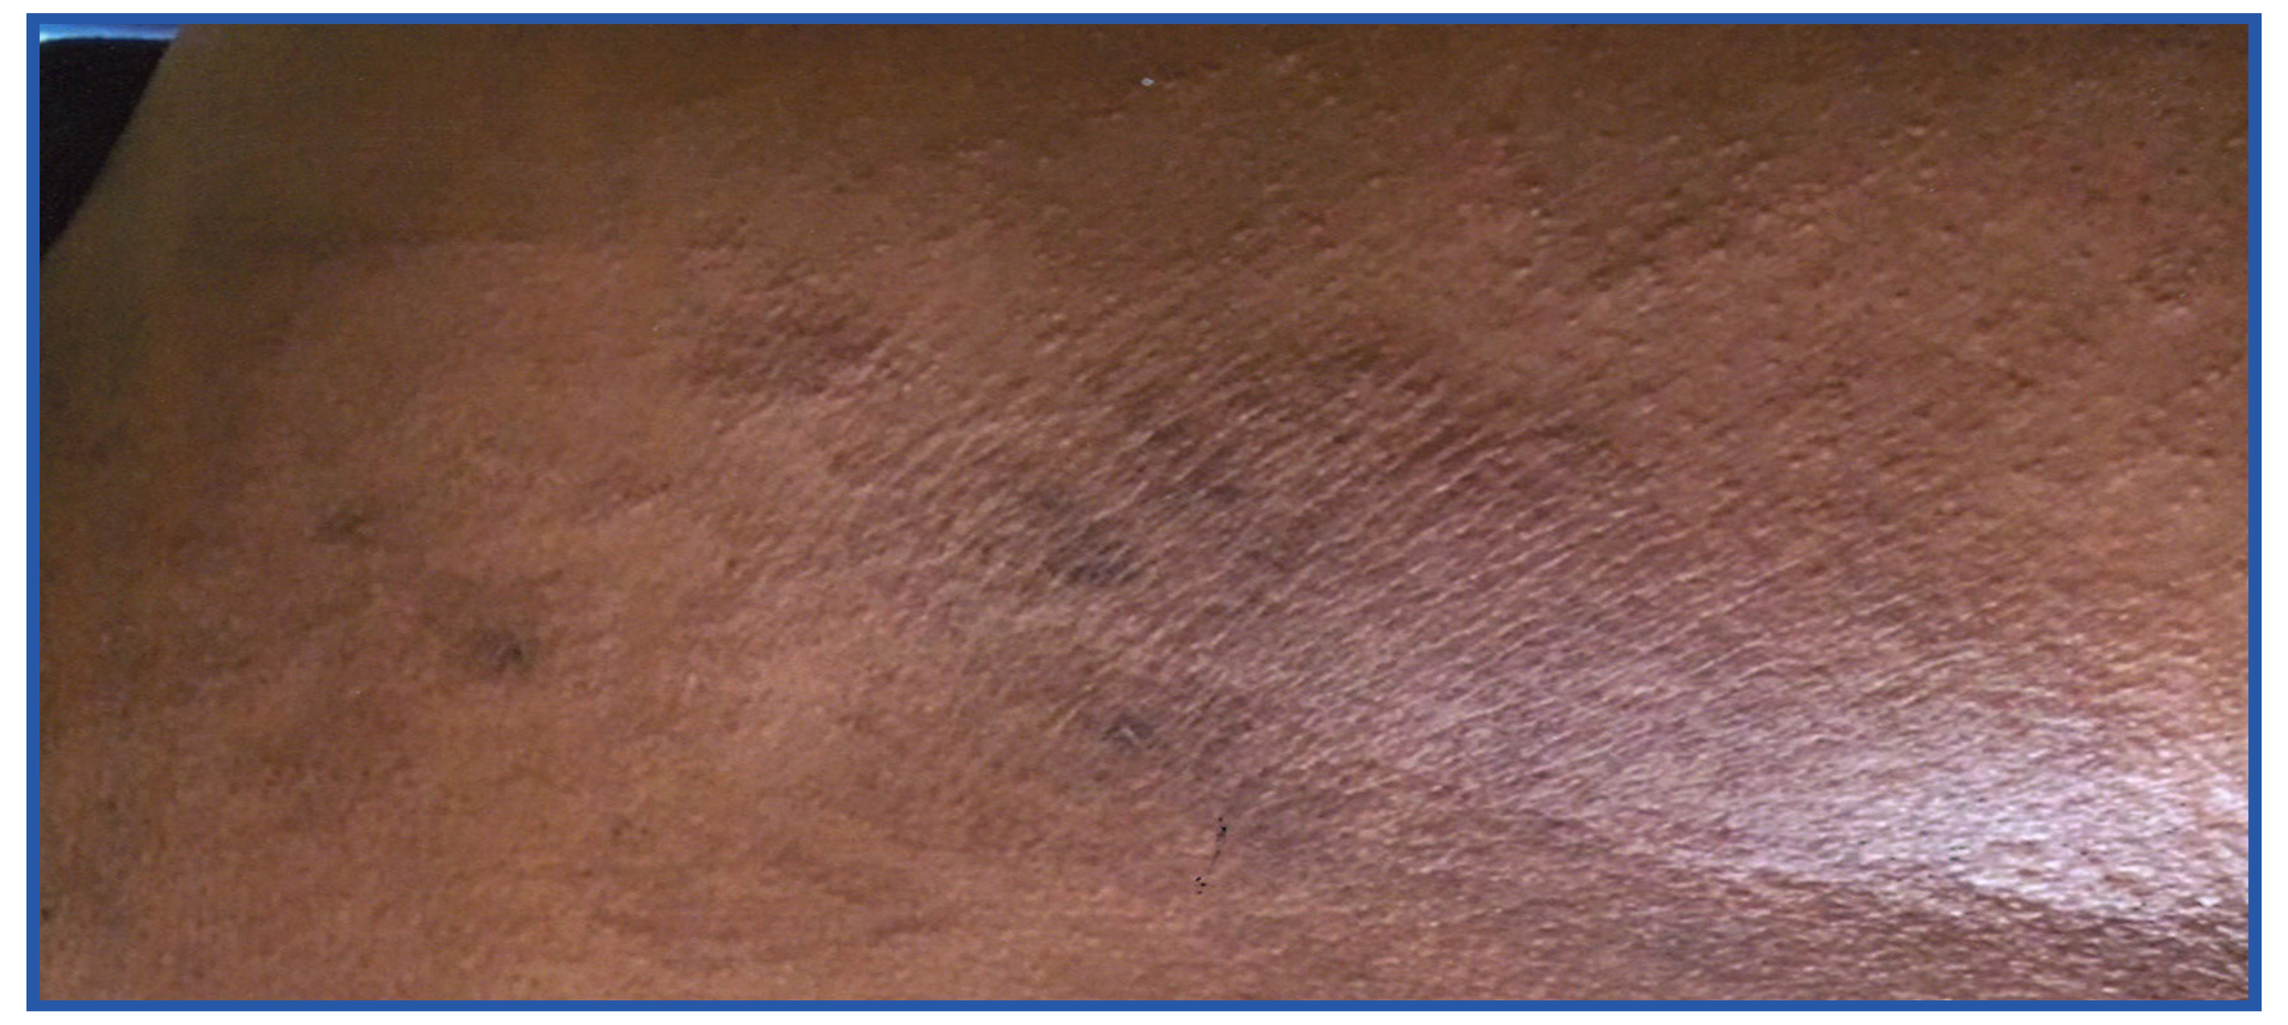

Fig. 1

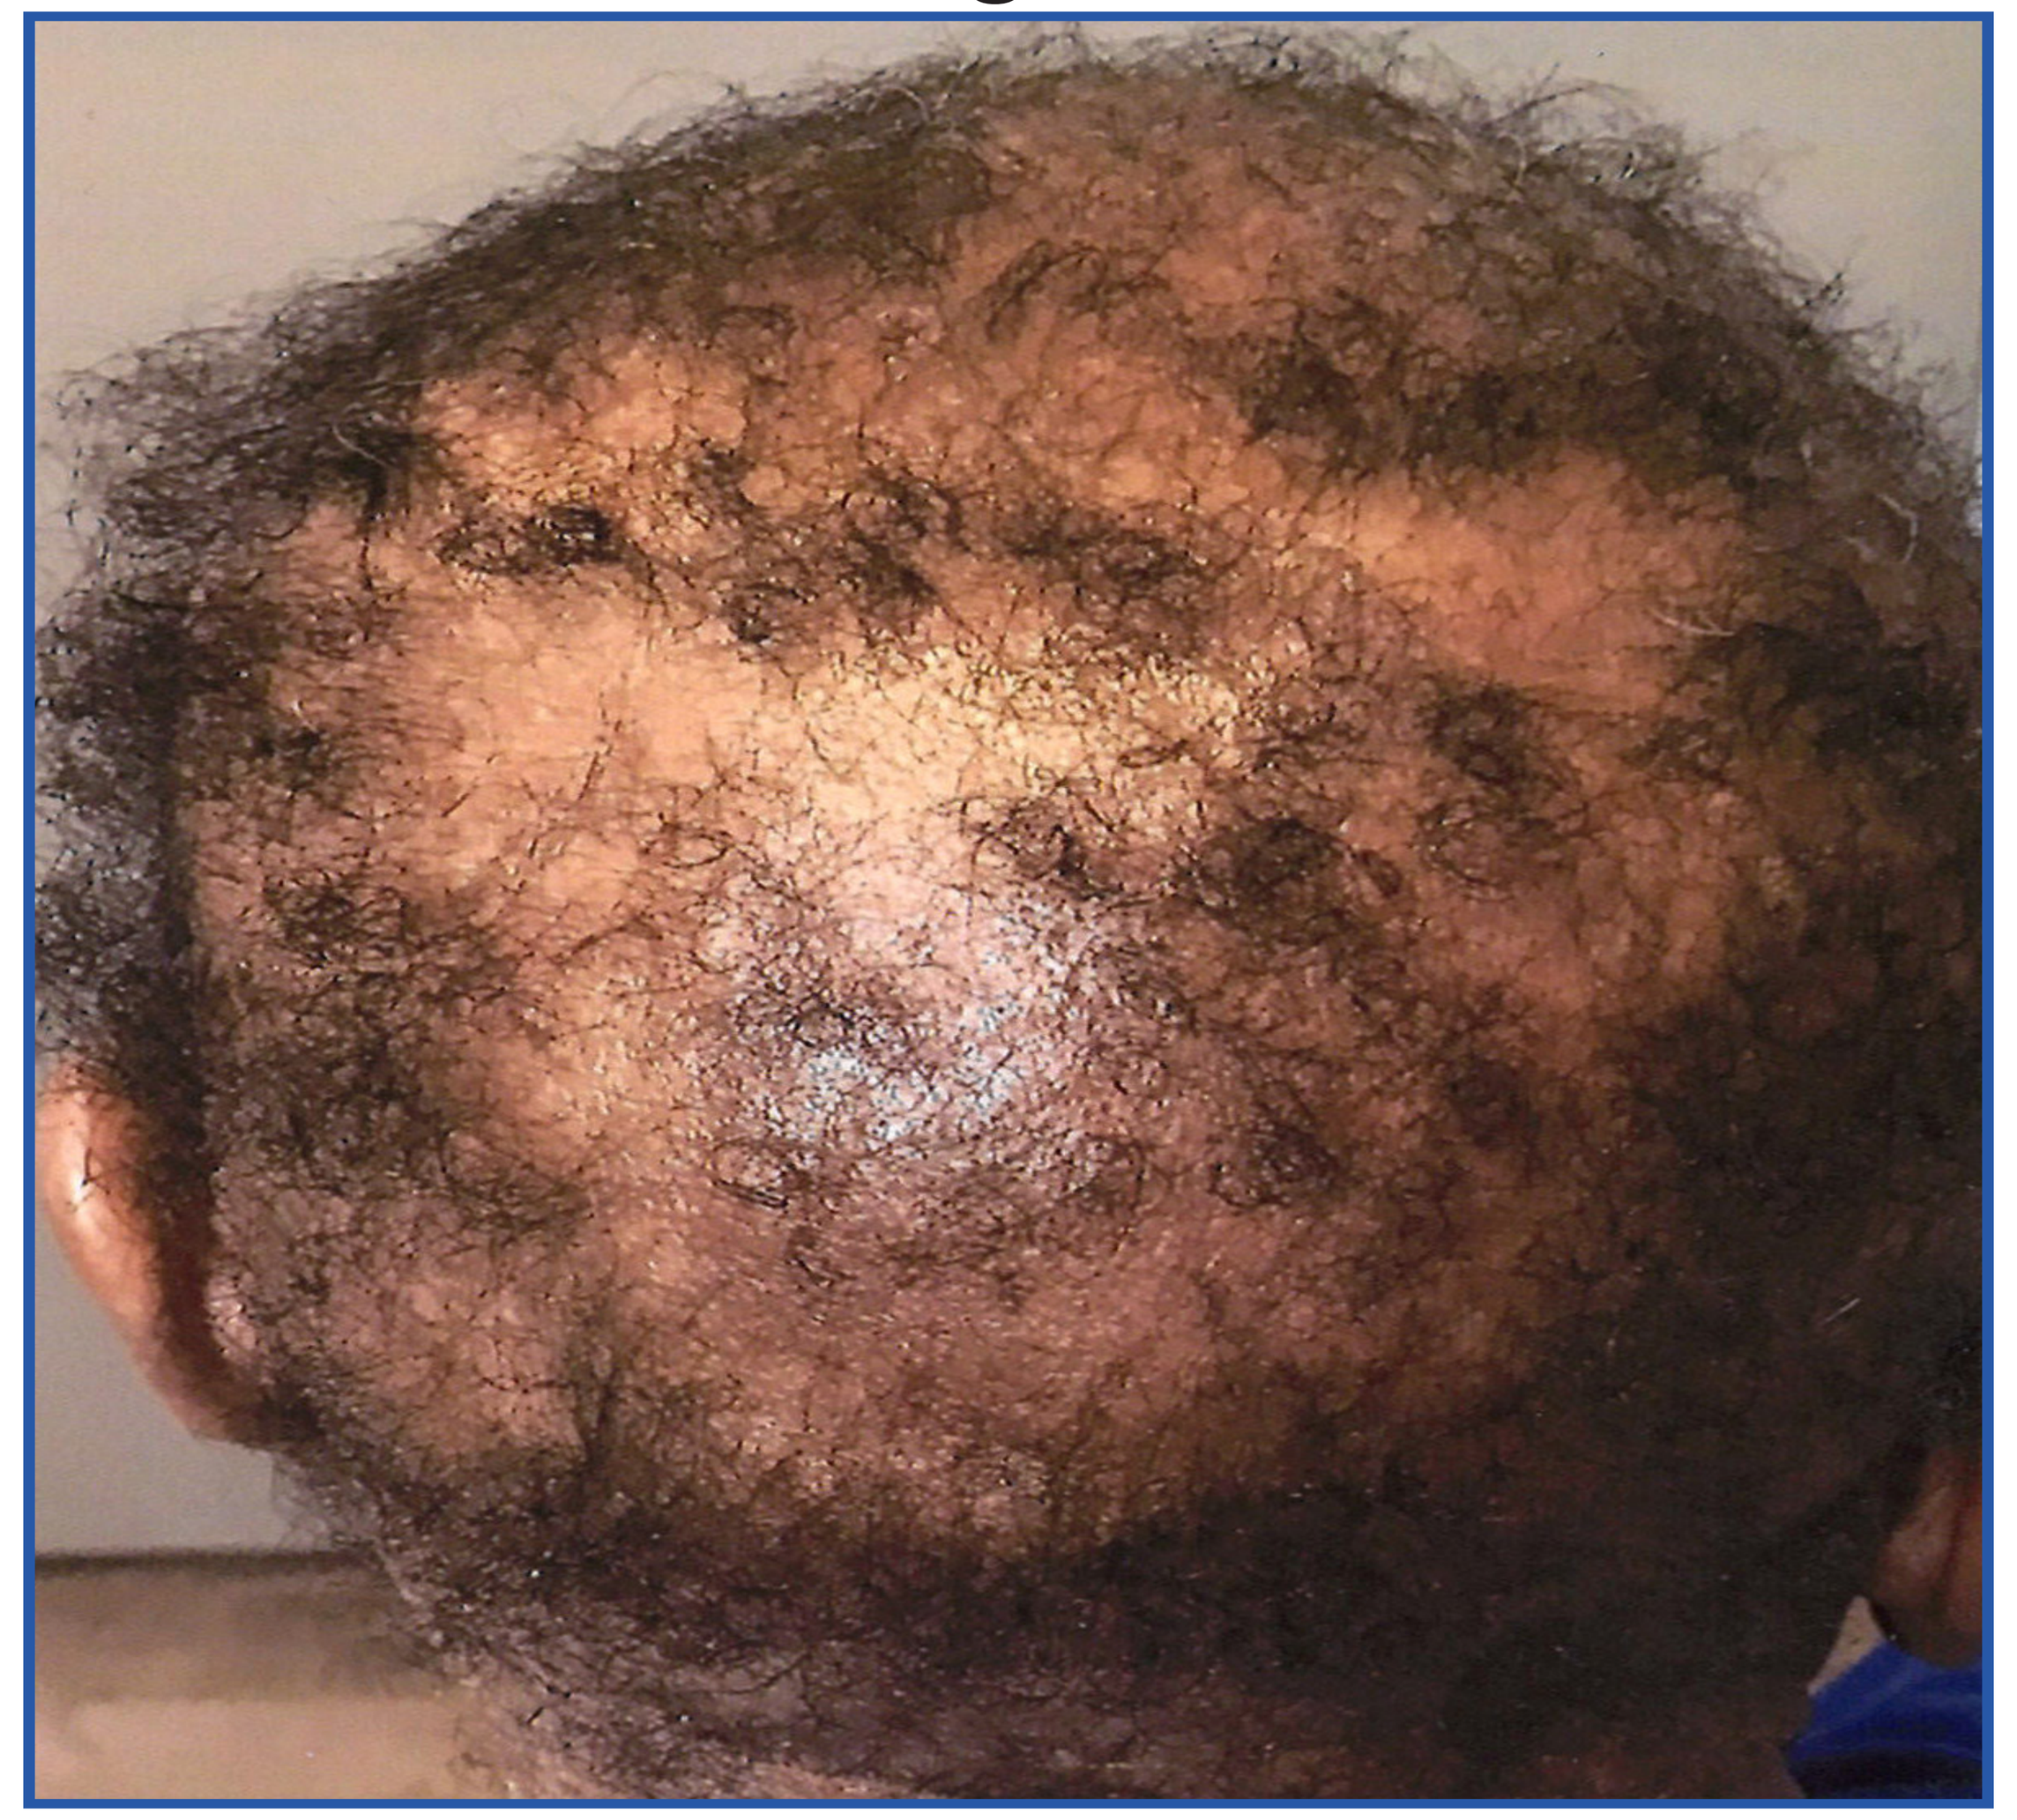

Fig. 2

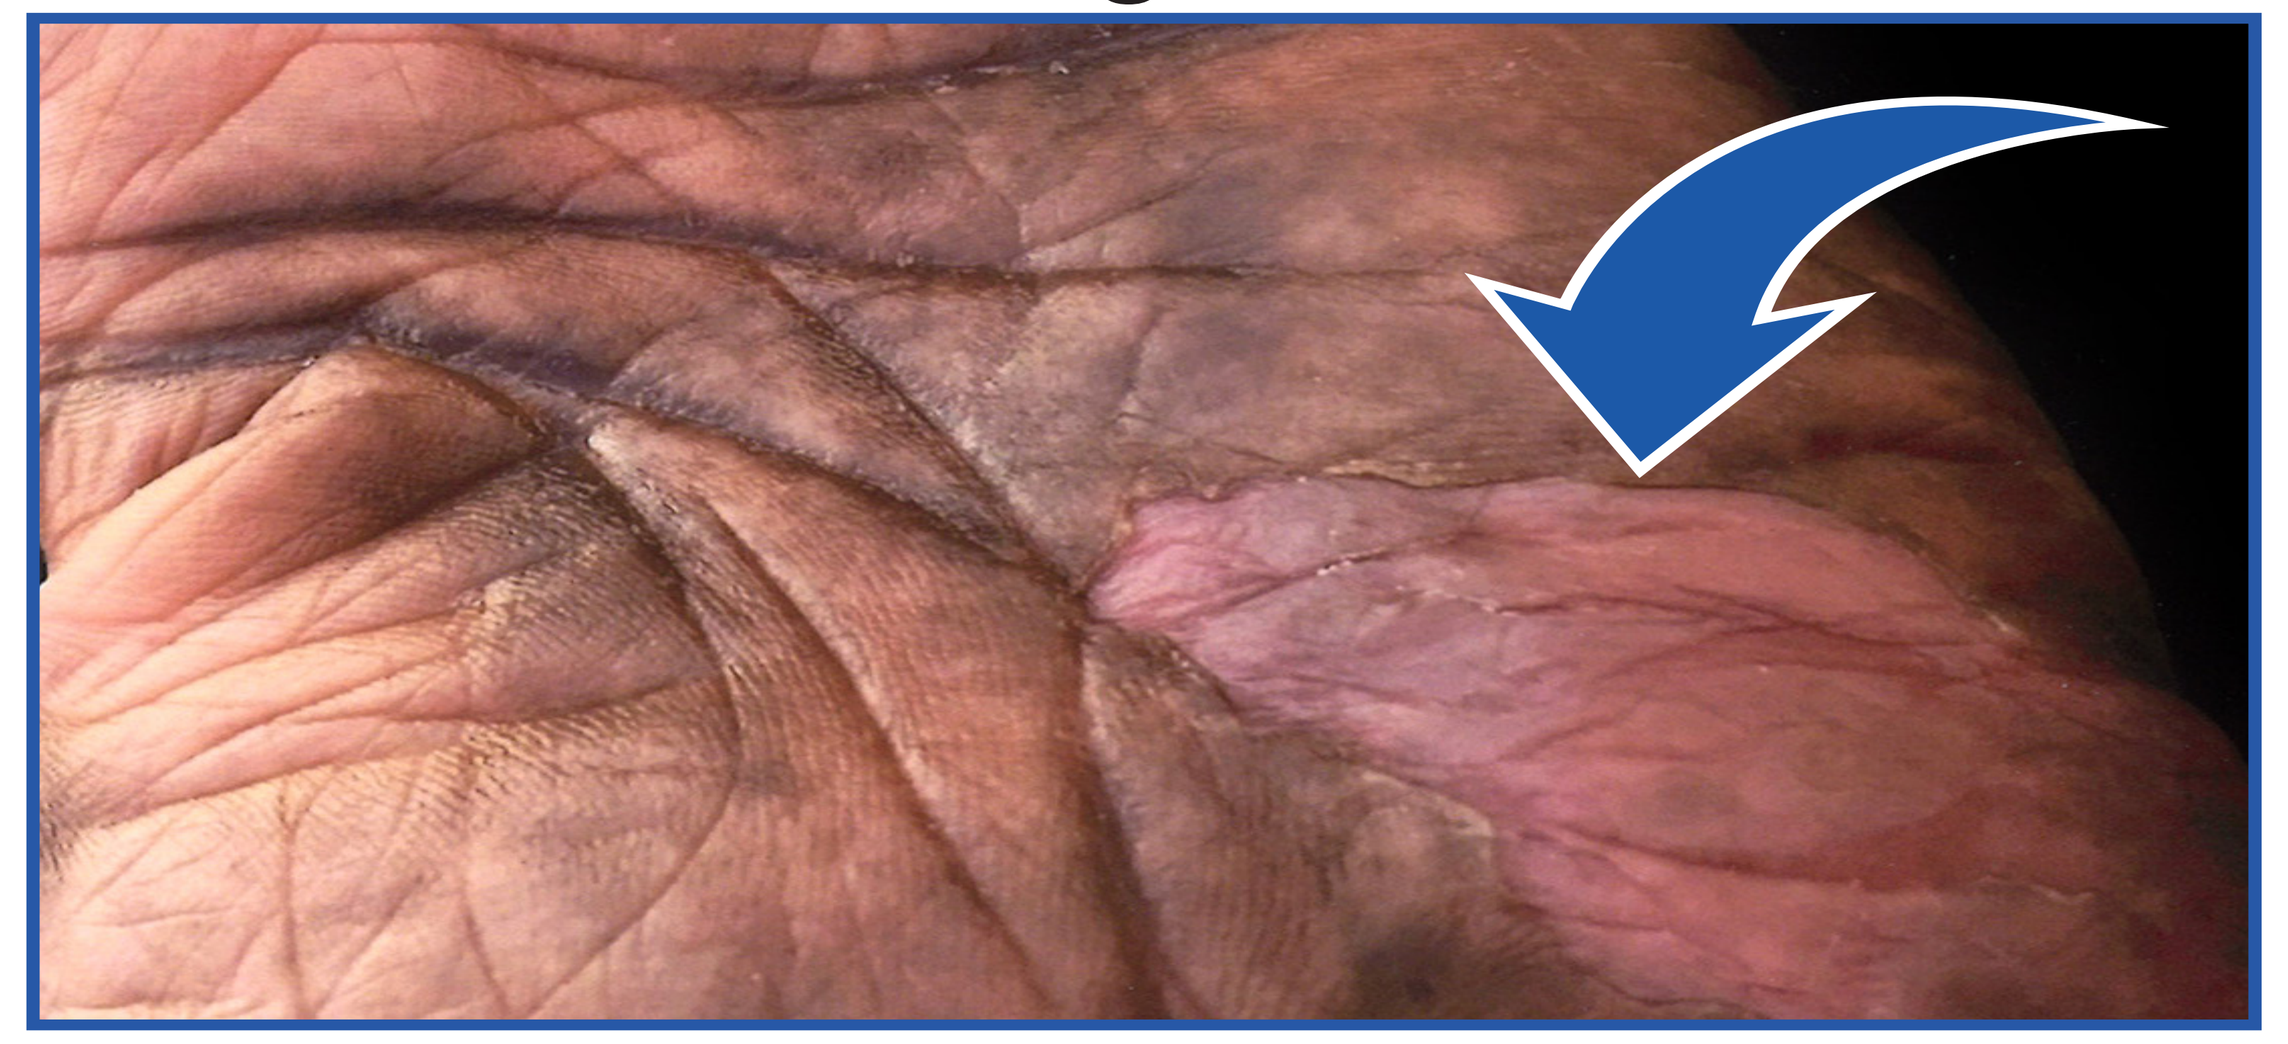

Fig. 3

Case 2

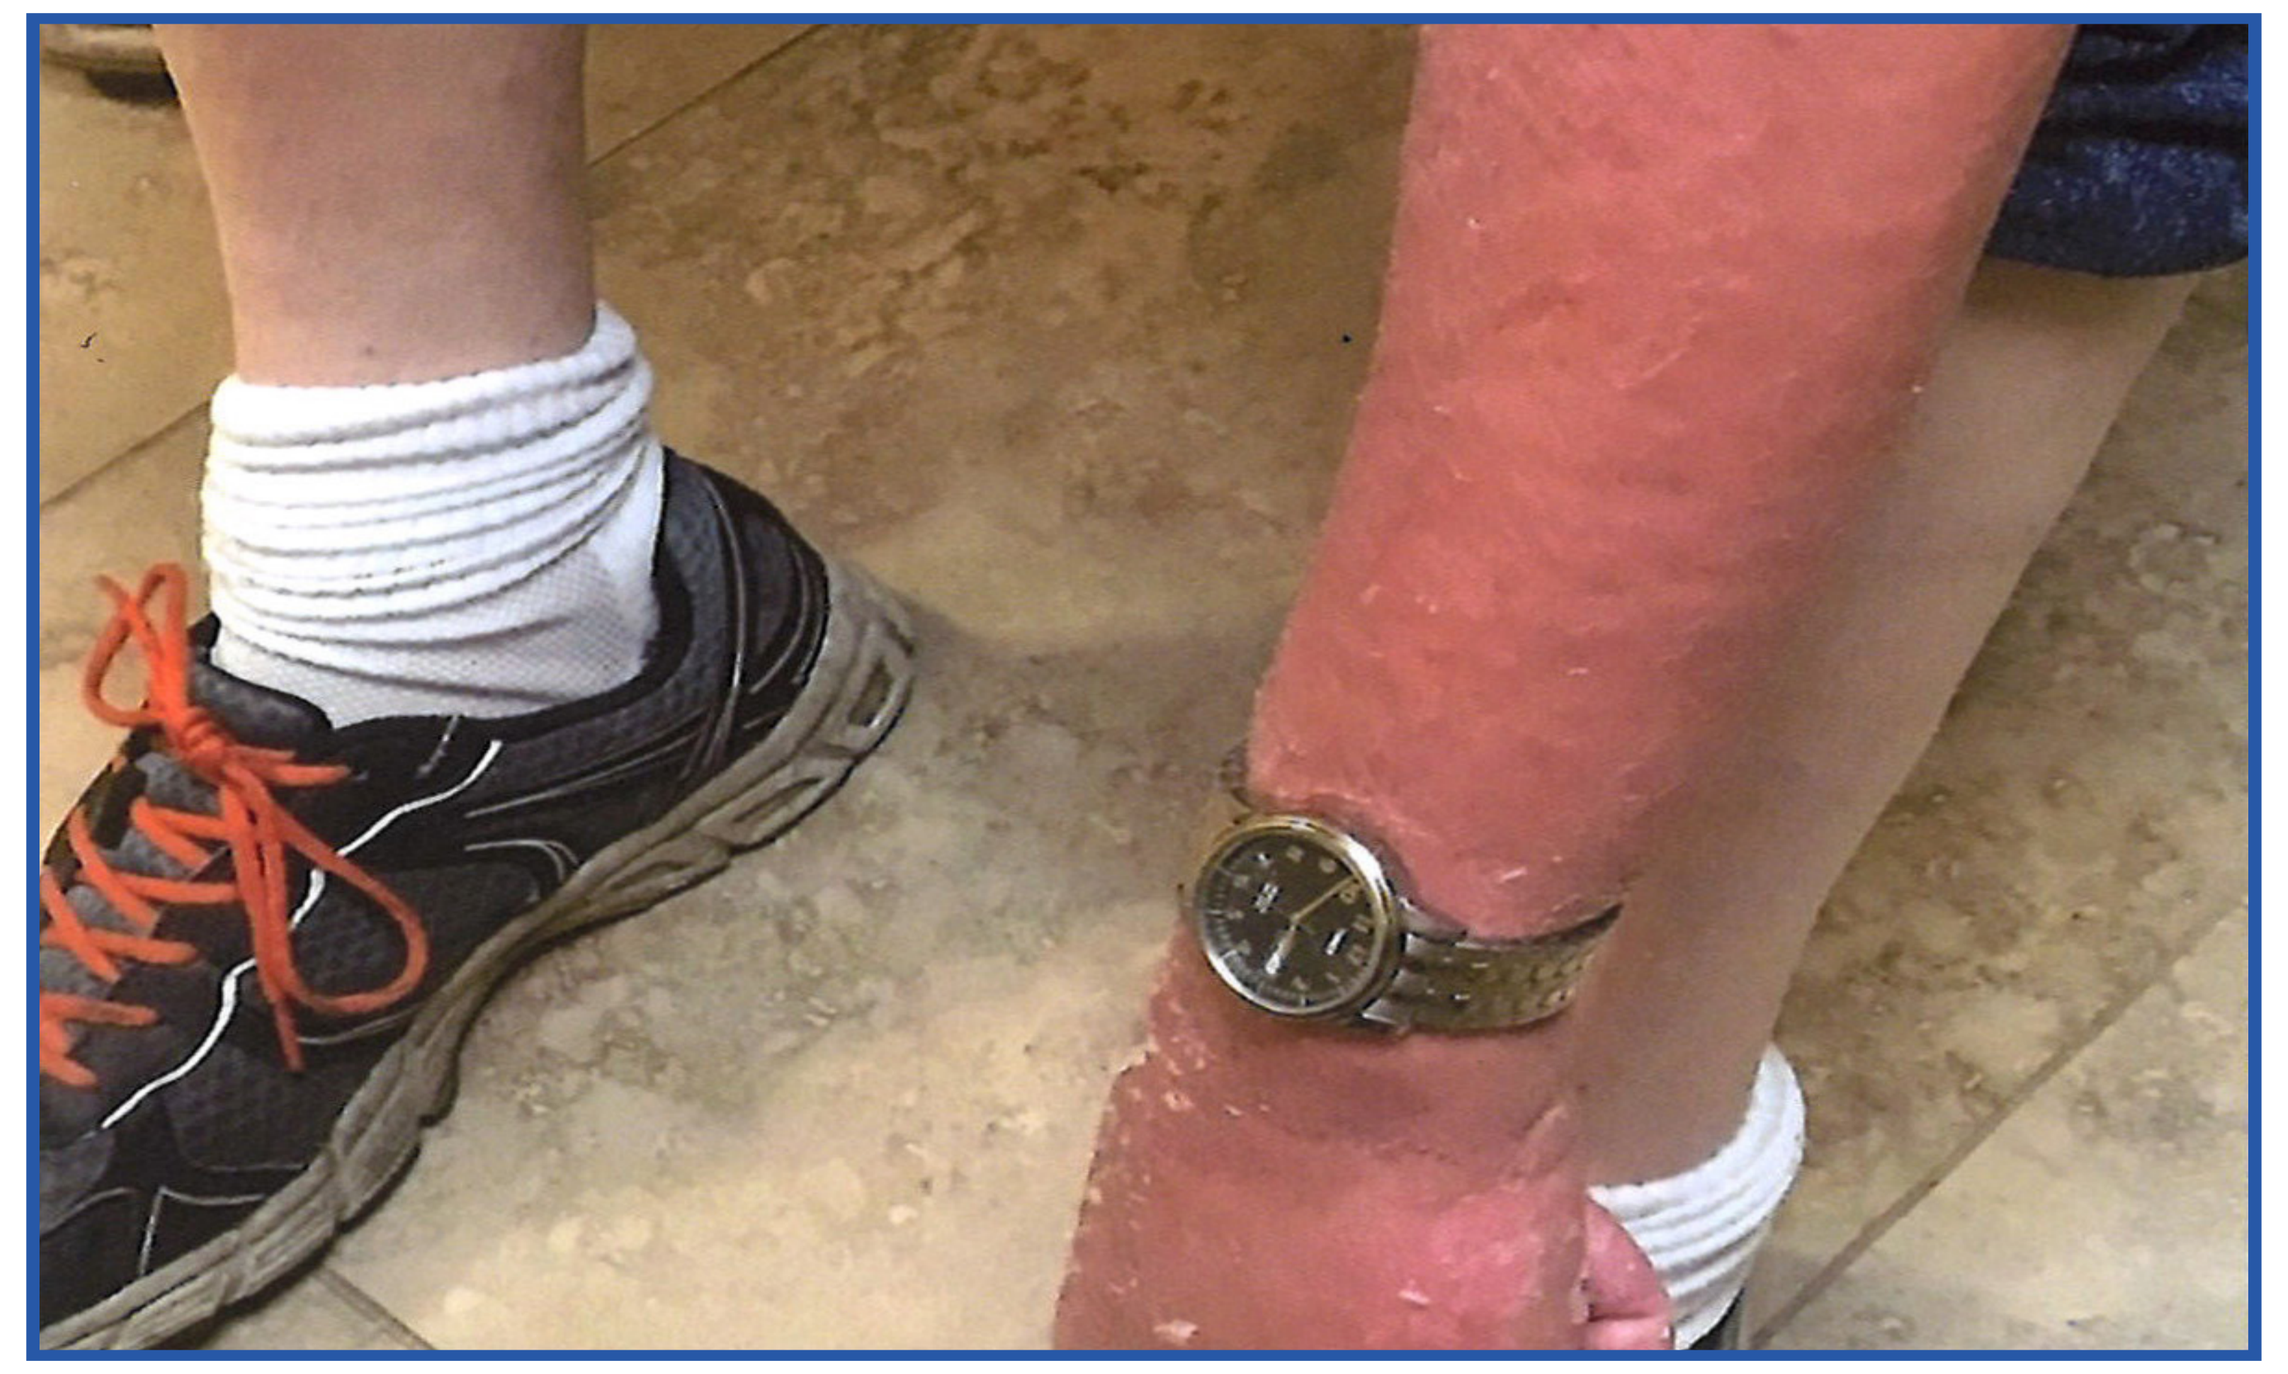

Fig. 4

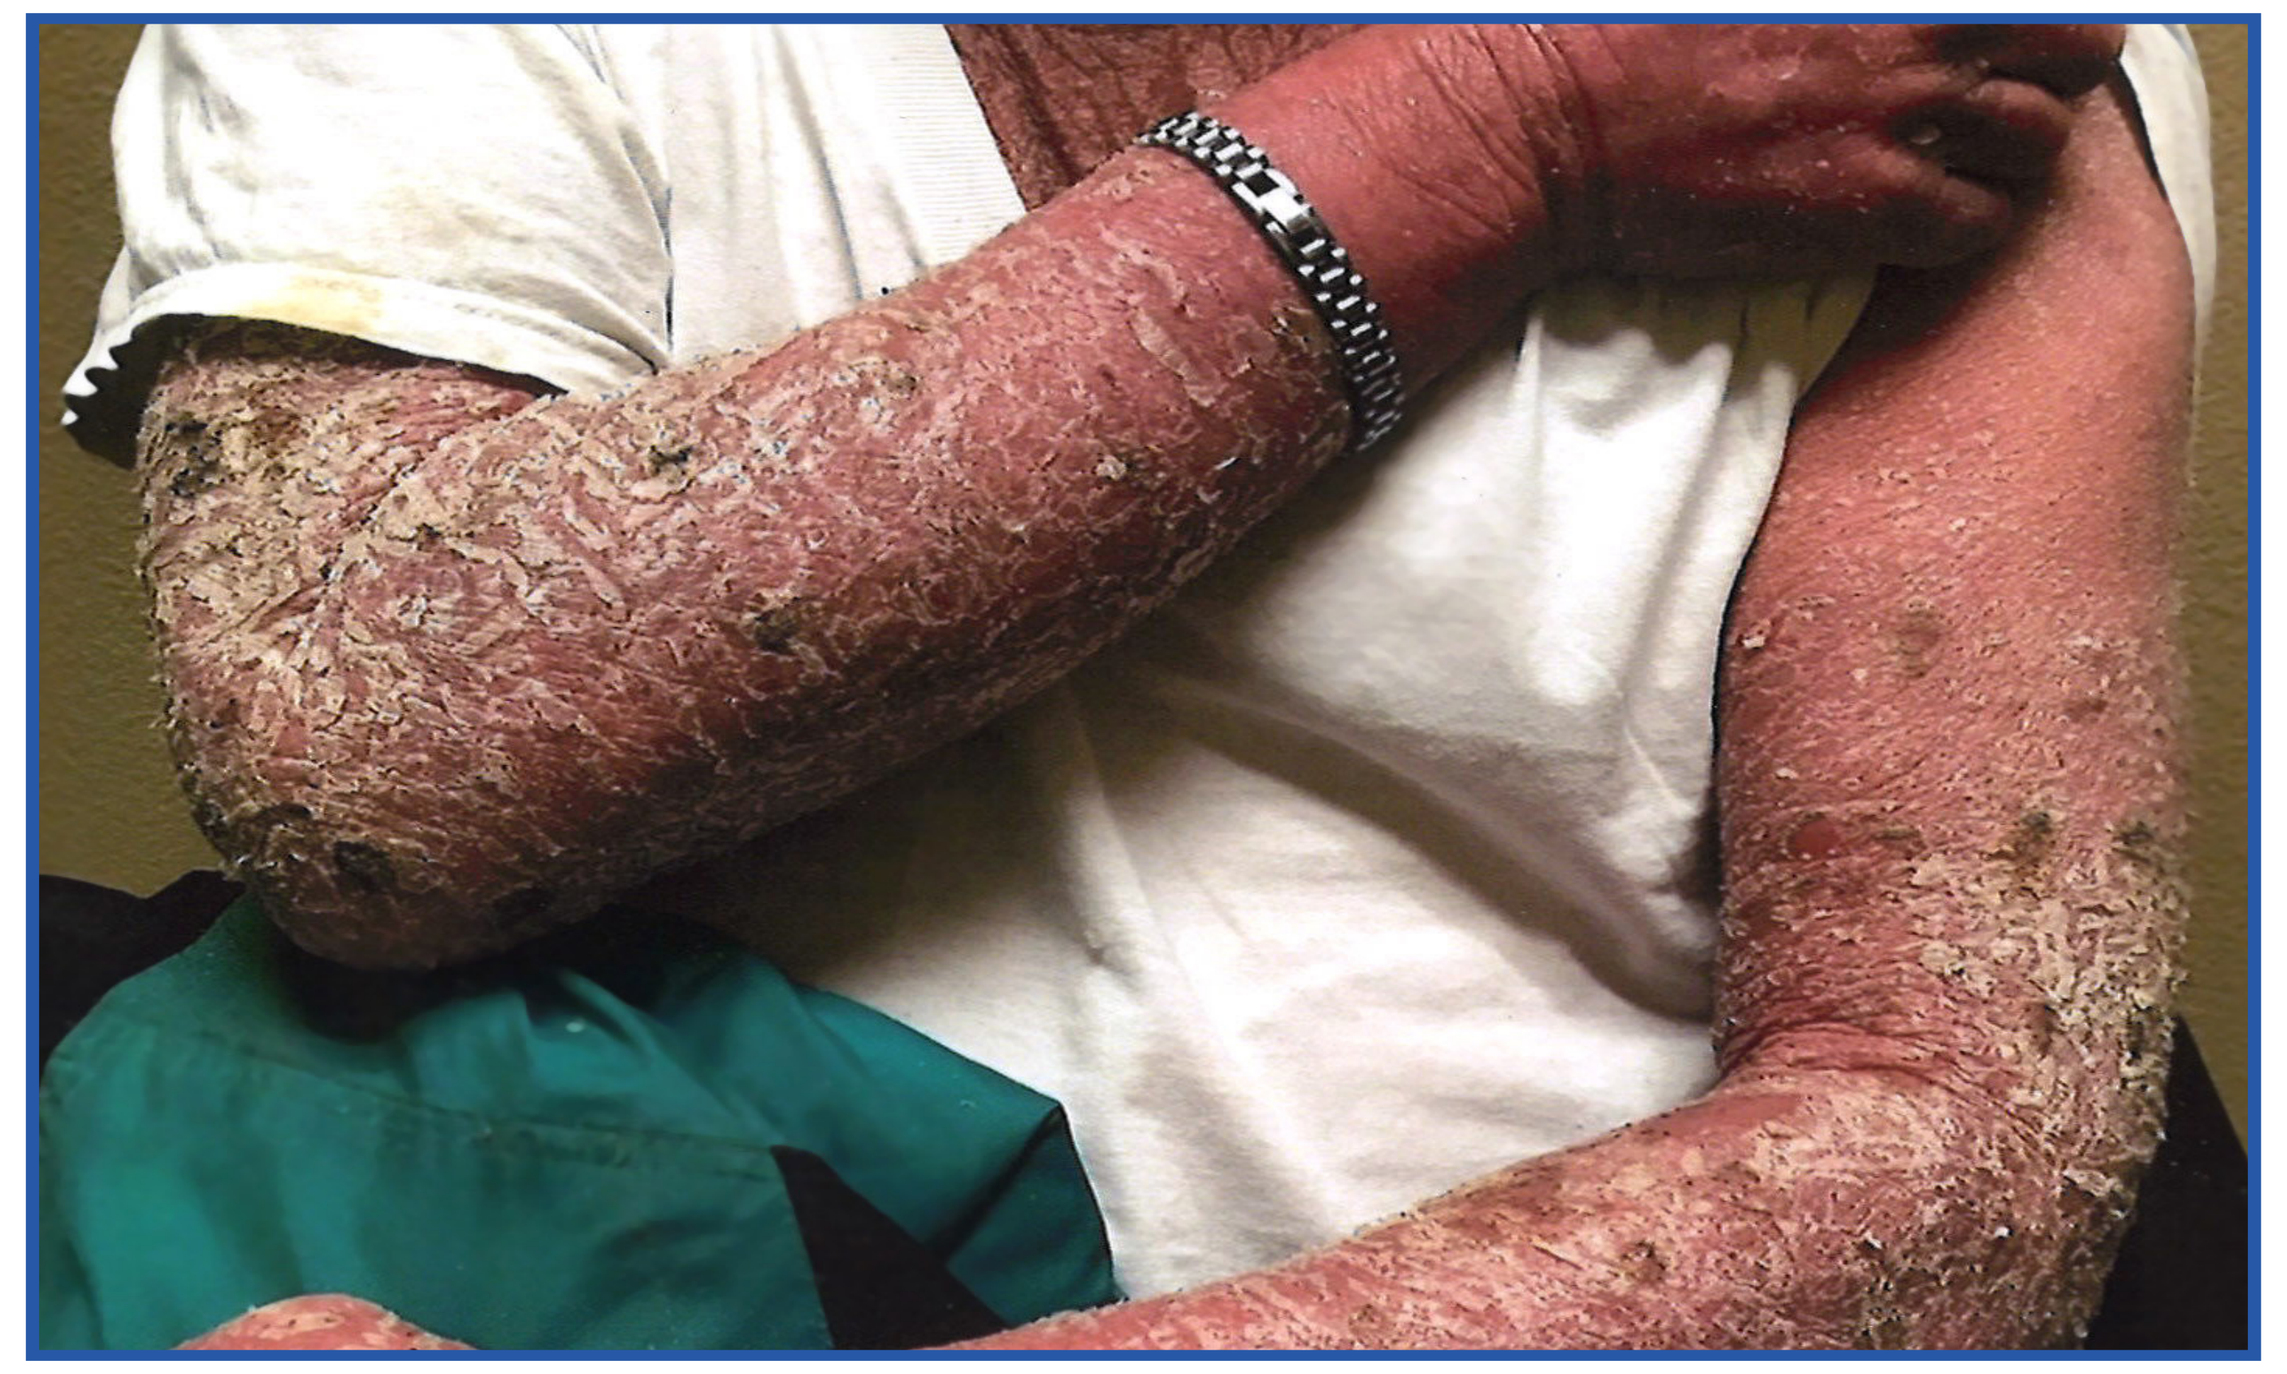

Fig. 7

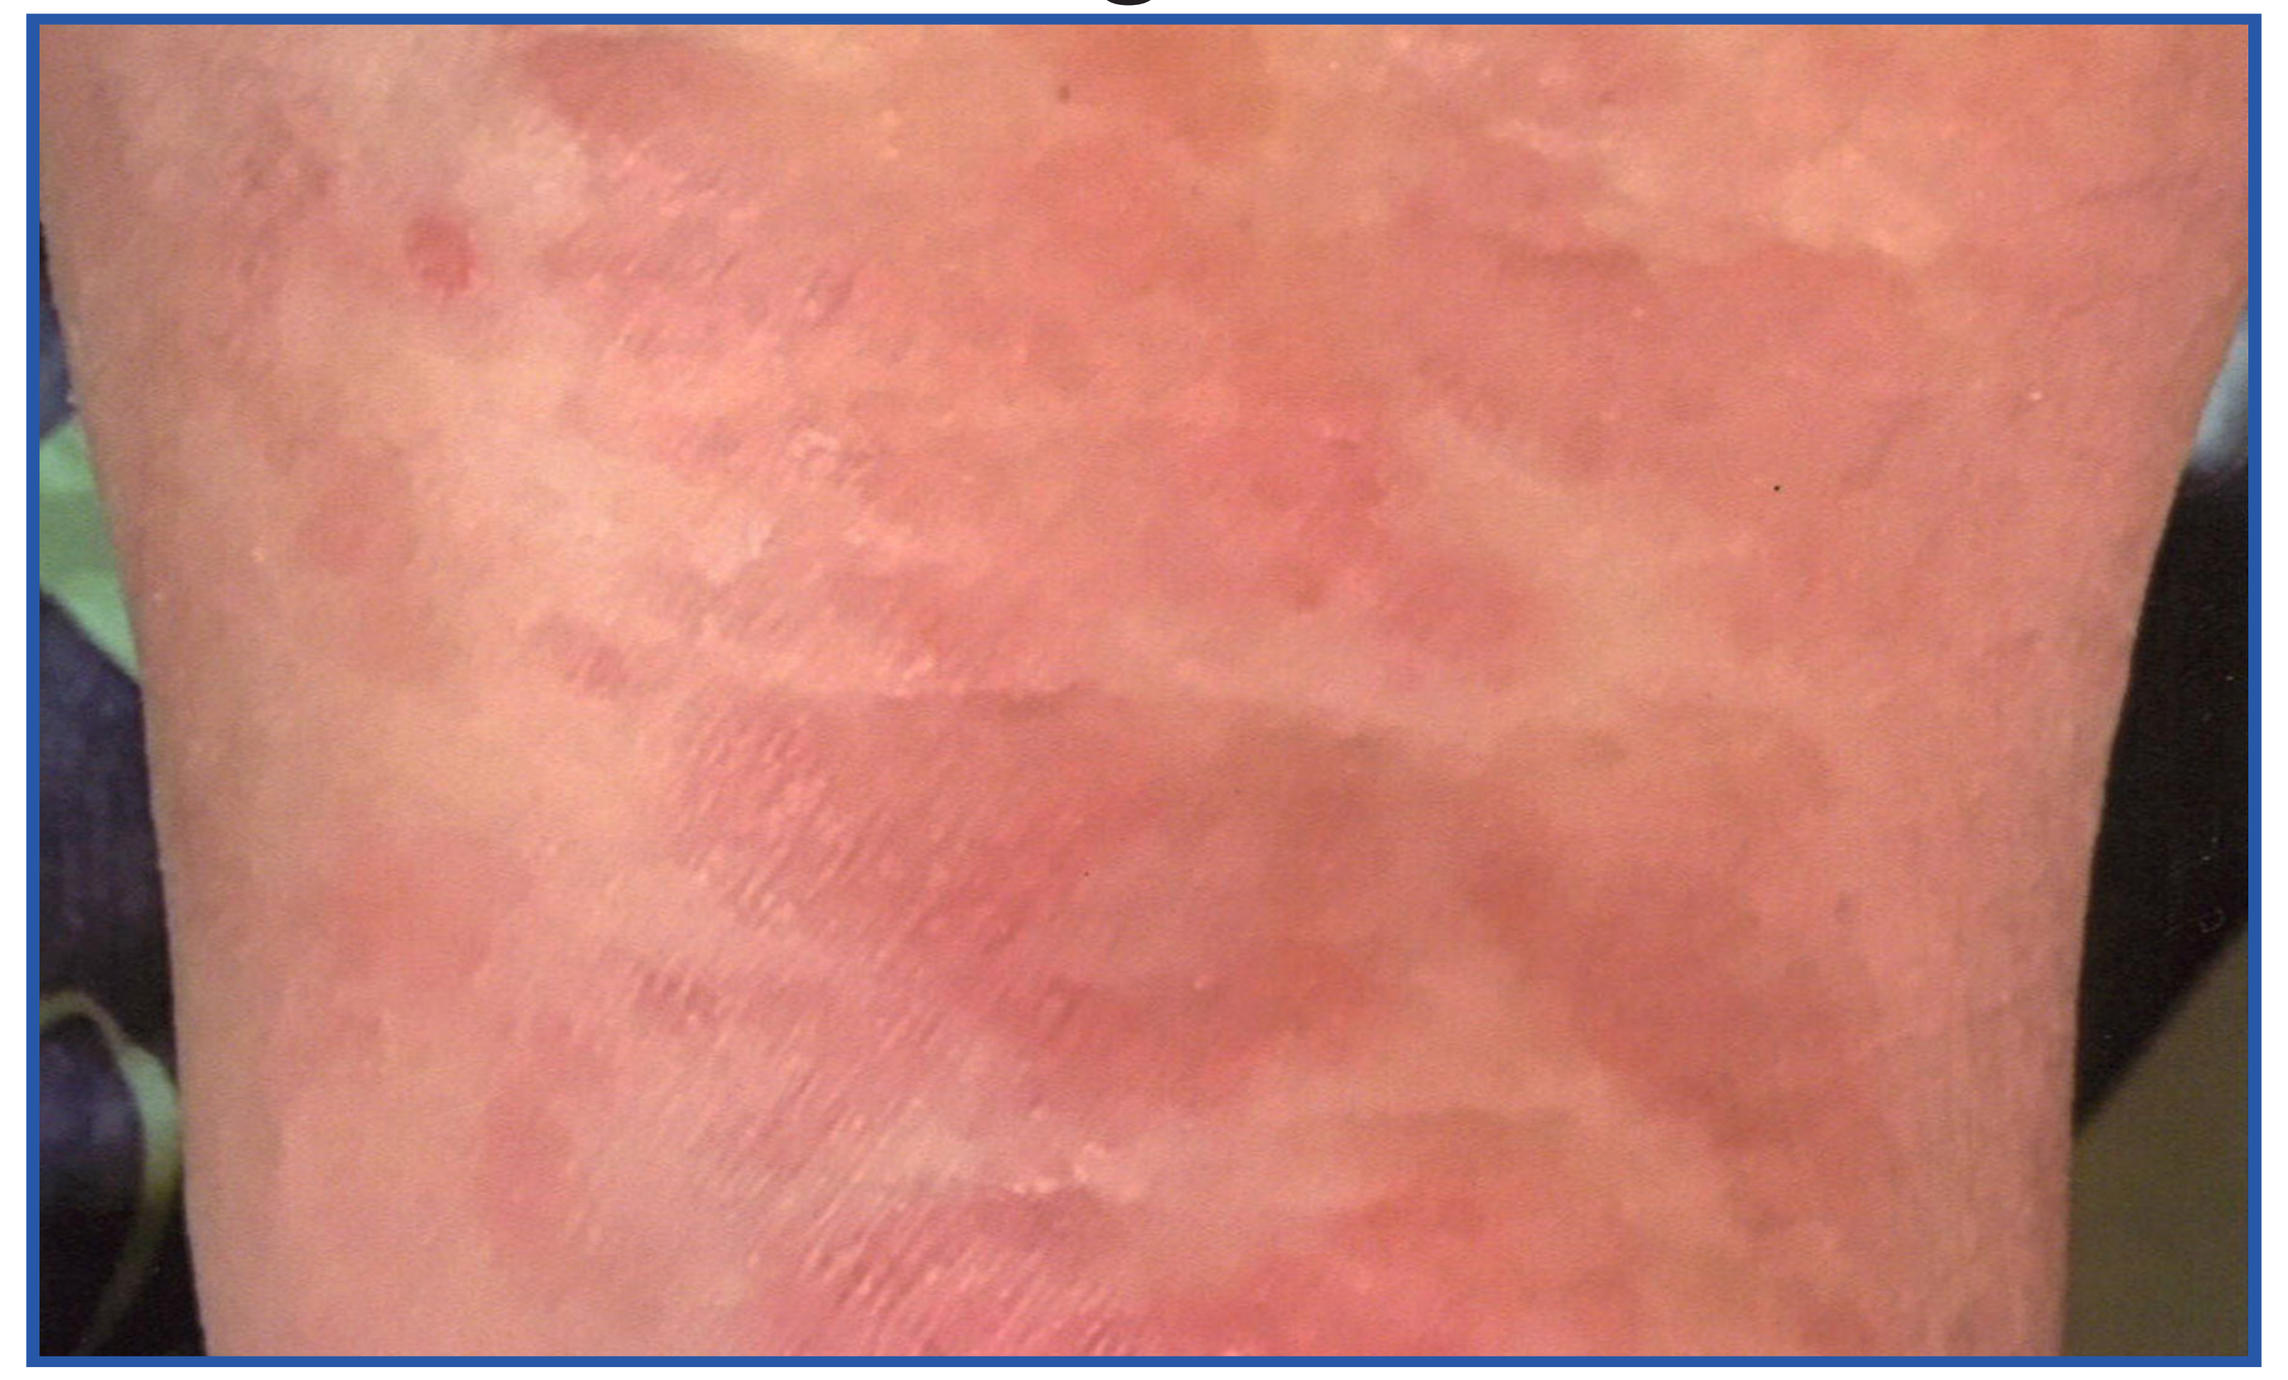

Fig. 5

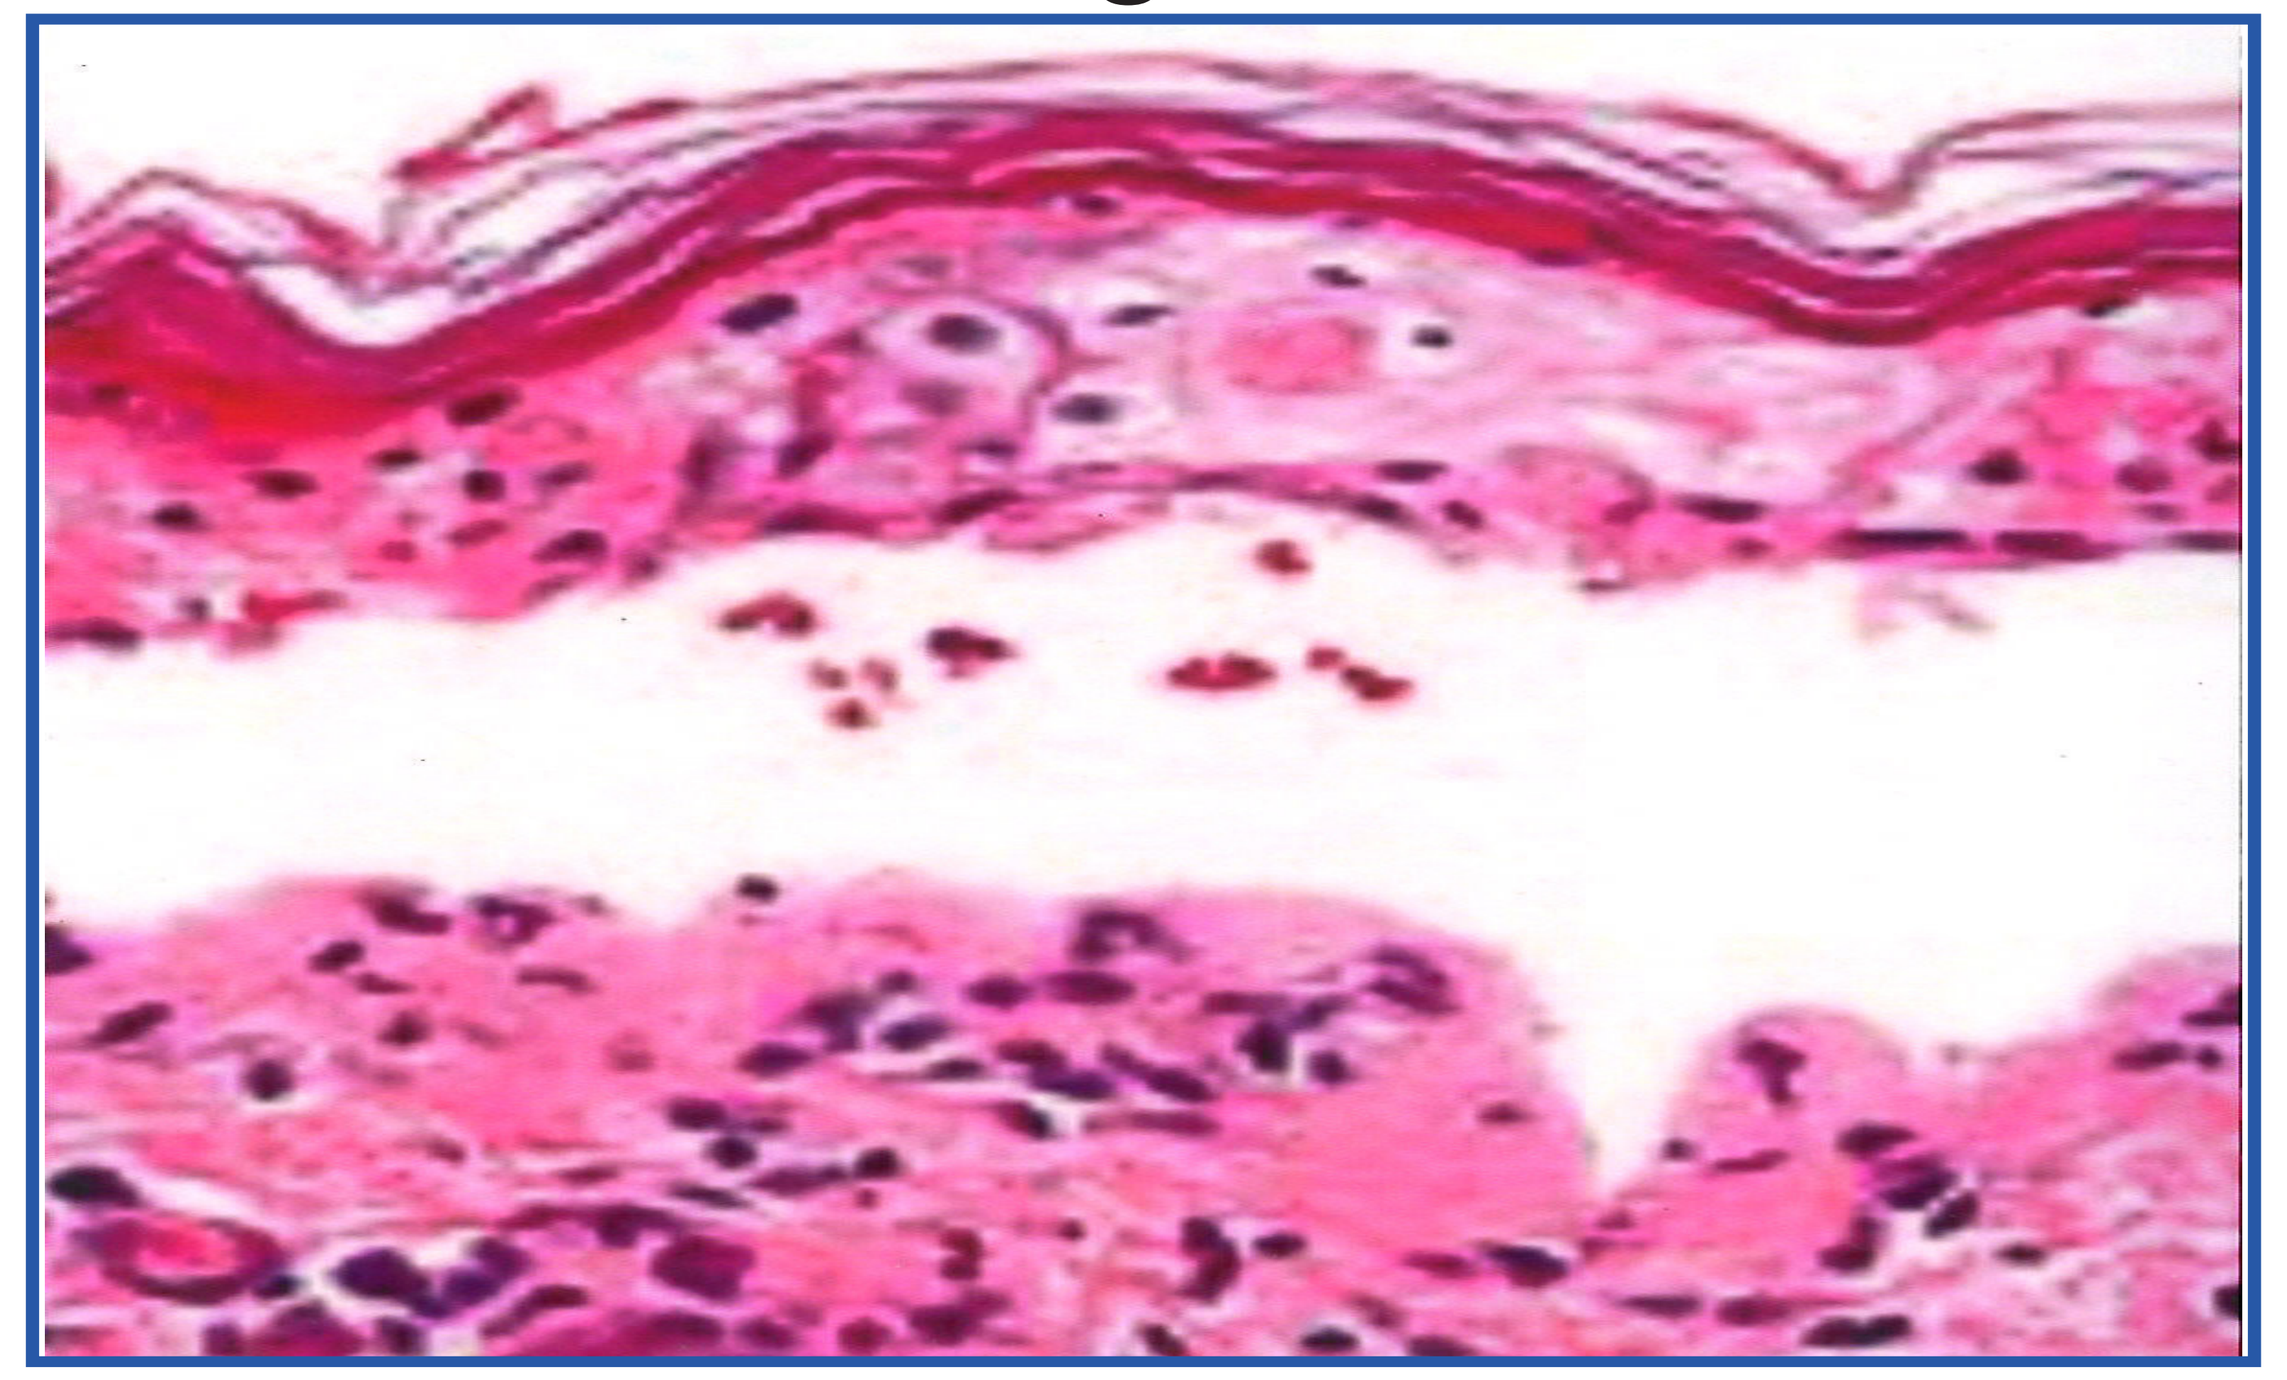

Fig. 8

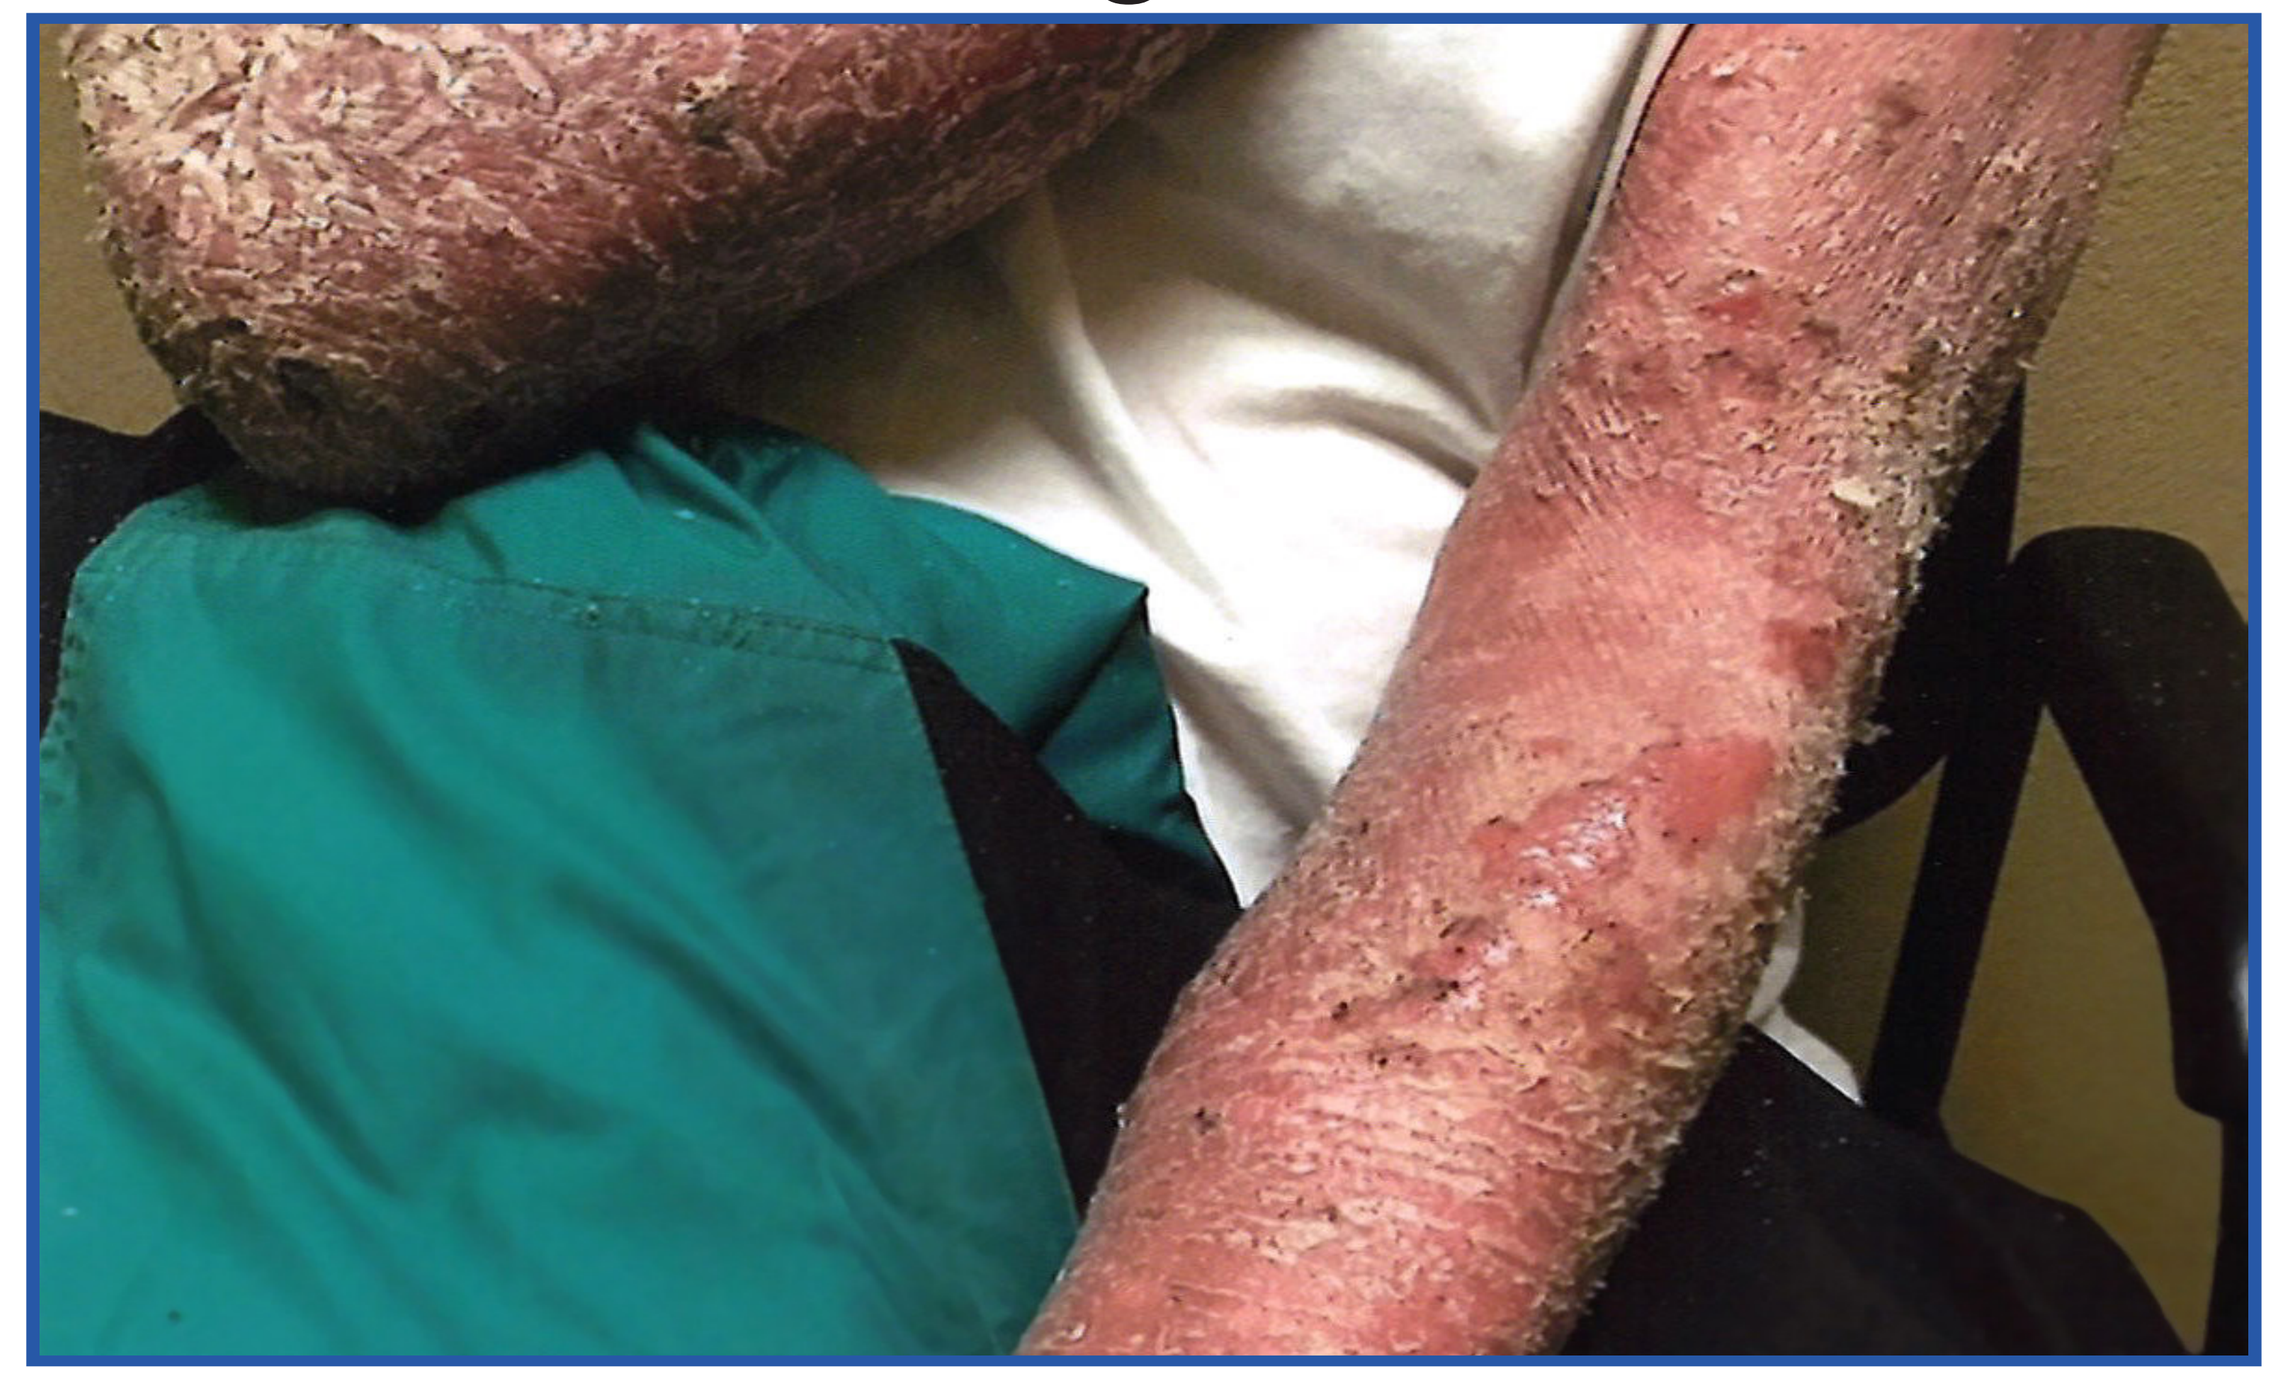

Fig. 6

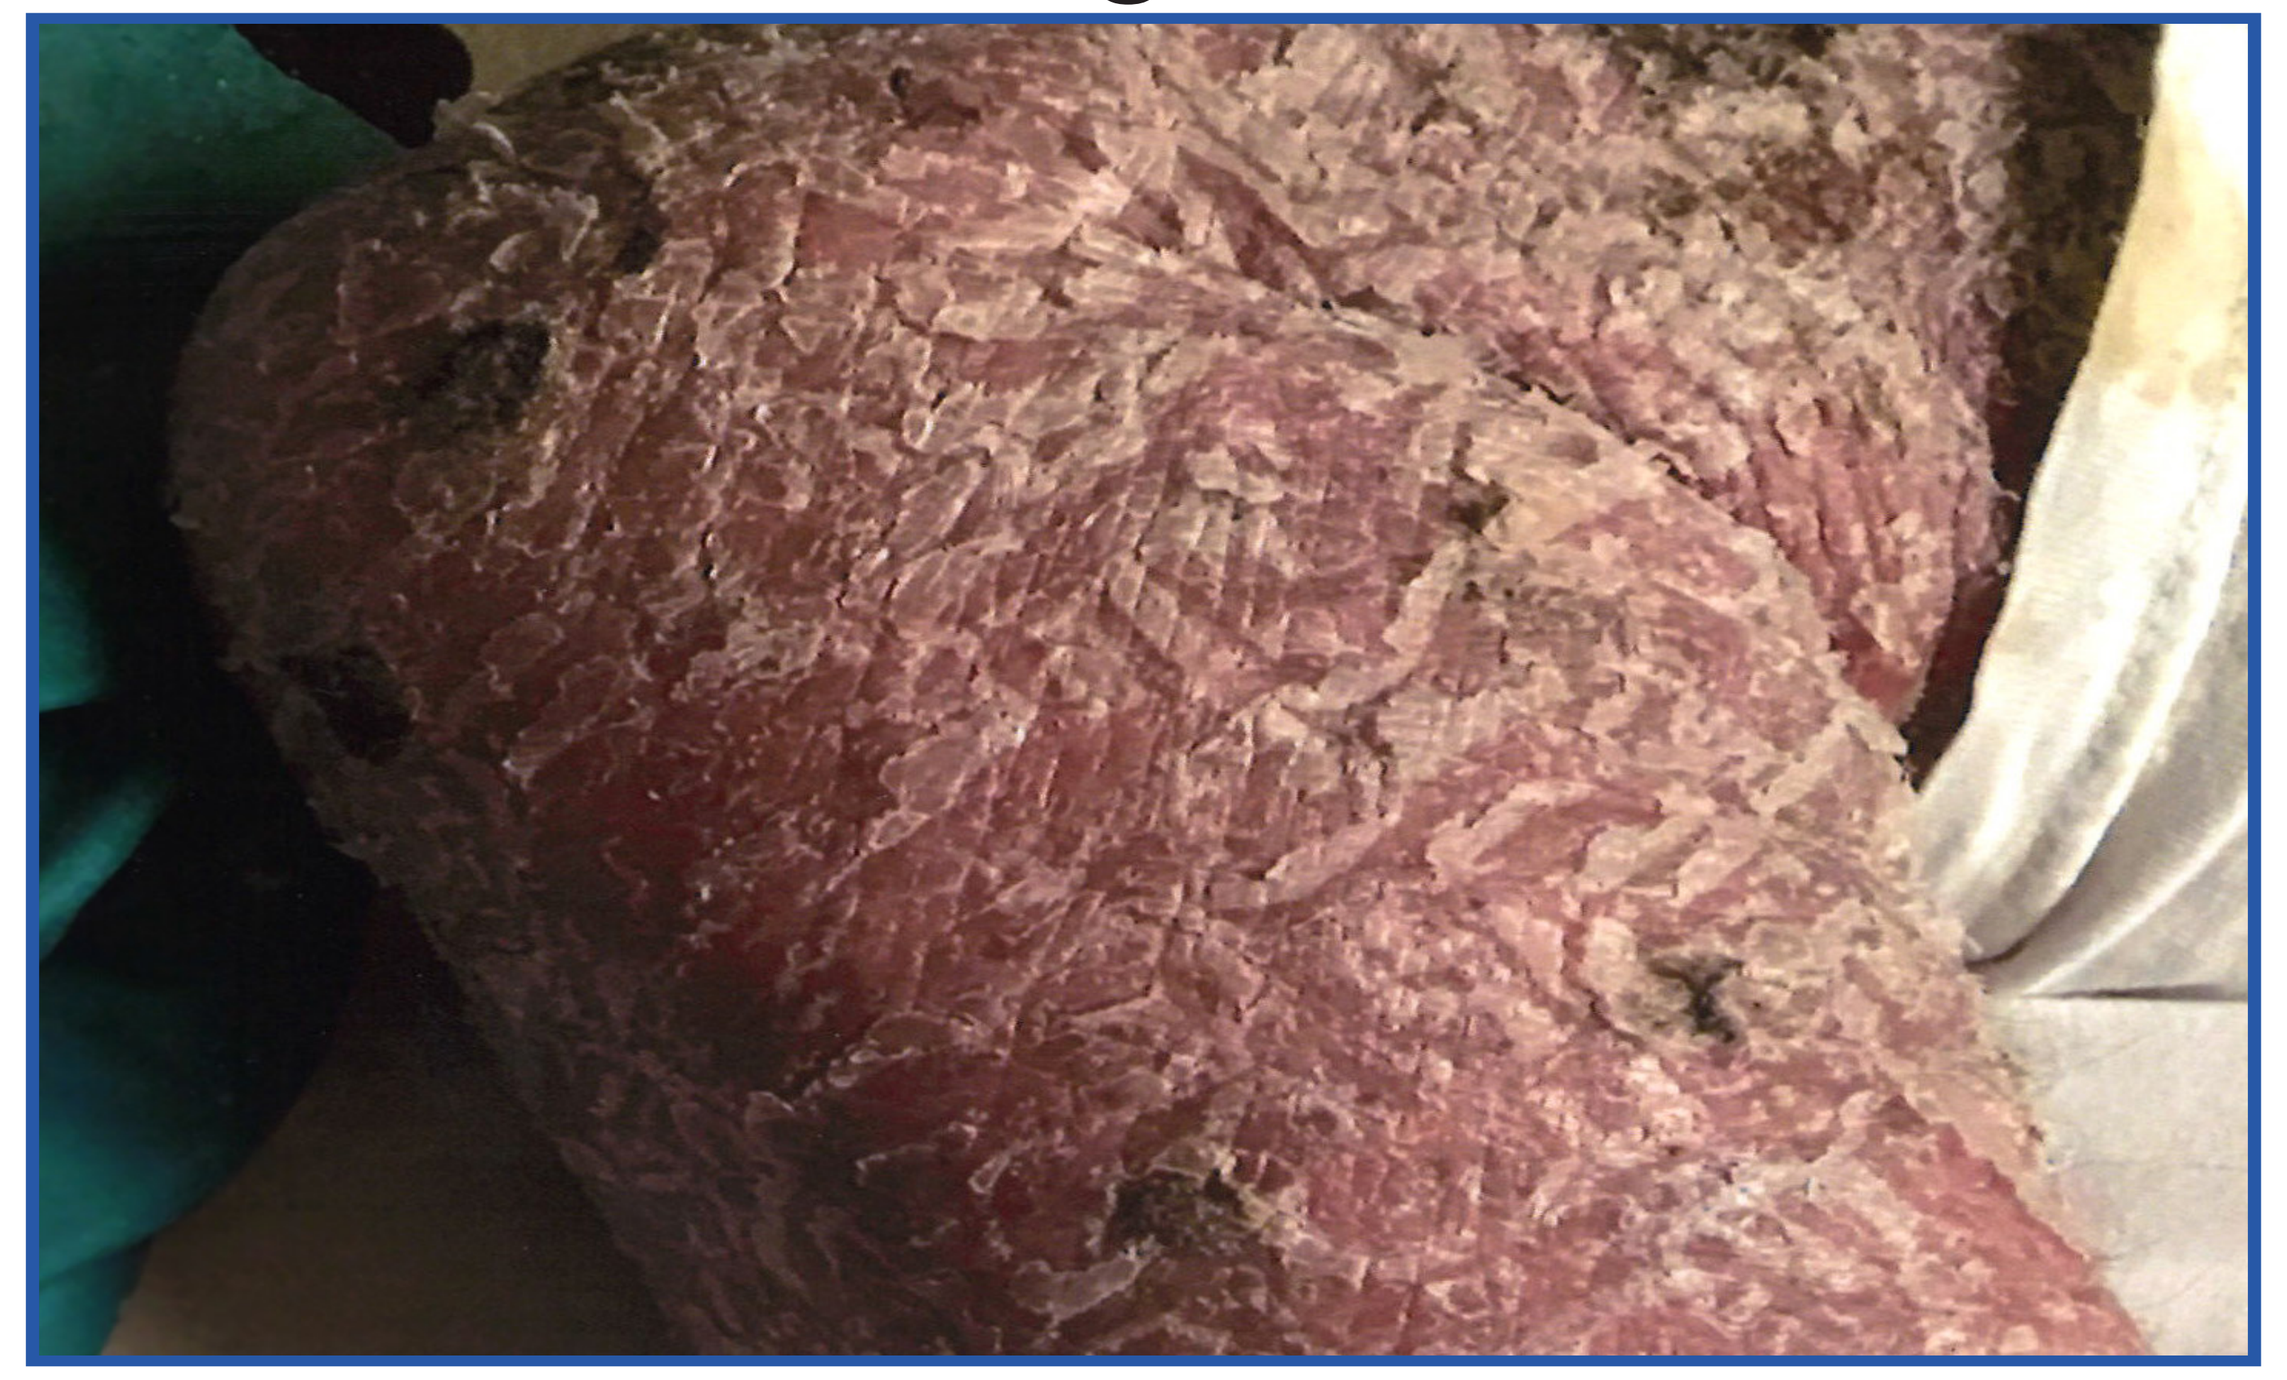

Fig. 9

RESULTS

Case 2. After 6 months of DAC, a 68 year old Caucasian man developed the insidious onset of a diffuse macular rash associated with pruritis, which appeared initially to be in a photo sensitive skin area but eventually generalized.

Fig. 4: Initial insidious development of what appeared to be a 'photo sensitive' areas of diffuse macular rash associated with pruritis following 6th. Injection of DAC

Fig. 5: Macular rash on forearm

REMS blood work was normal up until the event. He was initially treated with antihistamines. Over several days the rash appeared to be generalizing and eventually involved at least 30% of his body. Repeated temperature spikes of up to 103 degrees were recorded. Over several days the macular rash evolved into intense painful blisters with induration. Headache, malaise, and arthralgia were noted. WBC count was initially 23.5 with shift and sedimentation rate was 77. His skin eventually exfoliated.

Fig. 6: bullous eruption following macular rash that was associated with intense pain, induration, and secondary infection

Fig. 7: Generalization of bullous eruption with subsequent exfoliation

He was hospitalized and treated aggressively with steroids and antihistamines. Skin biopsy had revealed epidermal necrosis with inflammatory change. After a period of several weeks, the patient improved.

Fig. 8: H&E Stain. Skin biopsy showing full-thickness epidermal necrosis with basketweave-like stratum corneum and separation of the dermis and epidermis; minimal inflammatory change is noted.

Fig. 9: Residual dermatologic deficit with eventual complete exfoliation

CONCLUSION

This is the first report of SJS associated with DAC use. The cases suggest that vigilance is mandatory when using DAC since more serious and potentially lethal dermatologic complications can occur.

DISCLOSURE

The authors have nothing to disclose.