Prespecified Subgroup Analyses of Ocrelizumab Efficacy in Patients With Primary Progressive Multiple Sclerosis From the Phase III ORATORIO Study

JS Wolinsky,[1] X Montalban,[2,3] SL Hauser,[4] L Kappos,[5] L Julian,[6] M Manfrini,[7] S Belachew,[7] F Model,[7] S Hubeaux,[7] A Bar-Or[8]

[1]McGovern Medical School, UTHealth, Houston, TX, USA; [2]Division of Neurology, University of Toronto, Toronto, ON, Canada; [3]Vall d'Hebron University Hospital, Barcelona, Spain; [4]University of California, San Francisco, San Francisco, CA, USA; [5]University Hospital Basel, University of Basel, Basel, Switzerland; [6]Genentech, Inc., South San Francisco, CA, USA; [7]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [8]University of Pennsylvania, Philadelphia, PA, USA

BACKGROUND

- Ocrelizumab (OCR) is a humanized monoclonal antibody that selectively depletes CD20+ B cells
- OCR has shown superior efficacy vs. placebo (PBO) in patients with primary progressive multiple sclerosis (PPMS) in the Phase III ORATORIO study (NCT01194570)[1]
 - — The proportion of patients with 12 week-confirmed disability progression (12W-CDP; primary endpoint), as measured by Expanded Disability Status Scale (EDSS) score, was lower in patients receiving OCR (32.9%) vs. PBO (39.3%; hazard ratio, 0.76 [95% CI: 0.59–0.98; p=0.03])

OBJECTIVE

- To evaluate the effects of OCR vs PBO on clinical and imaging outcomes in prespecified subgroups of patients with PPMS from the ORATORIO study

METHODS

Study Design

- ORATORIO study design has been reported previously (**Figure 1**)[1]
- Patients were randomized (2:1) to receive OCR 600 mg (given as two 300 mg intravenous infusions 14 days apart) or corresponding PBO every 24 weeks
 - — ORATORIO was event driven: treatment was administered for a minimum of five doses (120 weeks) and until approximately 253 events of 12W-CDP were observed
 - — 12/24W-CDP was defined as an increase in EDSS score of ≥1.0 points from baseline EDSS sustained for ≥12/24 weeks if the baseline score was ≤5.5, or an increase of ≥0.5 points sustained for ≥12/24 weeks if the baseline score was >5.5

Statistical Analyses

- The treatment effect of OCR on 12W-CDP was analyzed in prespecified, baseline characteristic based subgroups:
 - — Region, age, sex, body mass index, body weight, prior disease-modifying therapy (DMT; excludes corticosteroids), baseline EDSS score, duration of multiple sclerosis (MS) since symptom onset and baseline T1 gadolinium-enhancing (Gd+) lesions
- **Additional analyses of disability, MRI and relapse outcomes were performed for subgroup comparisons that showed a trend for differences in treatment effect on 12W-CDP (nominal interaction p<0.3 on the primary endpoint)**
 - — **Disability outcomes analyzed by subgroups:** 12/24W-CDP, and12/24W-confirmed ≥20% increase in timed 25-foot walk (12/24W-T25FW) and 12/24W-confirmed ≥20% increase in timed 9-hole peg test (12/24W-9HPT)
 - — **MRI outcomes analyzed by subgroups:** total T2 lesion volume change (baseline to Week 120) and total brain volume change (Weeks 24-120)
 - — **Relapse outcomes analyzed by subgroups:** annualized relapse rate (ARR)
 - — Analyses used Cox regression, negative-binomial regression or mixed models of repeated measures (**Table 1**)
- The study was not powered to demonstrate efficacy within subgroups or differences between subgroups; analyses should be interpreted with caution

For Figure 1 and Table 1, please scan here

RESULTS

Patient Demographic and Baseline Characteristics

- Data from 488 OCR recipients and 244 PBO recipients have been reported previously[1]

12W-CDP Analyzed by Prespecified Subgroups

- None of the prespecified subgroup comparisons showed a statistically significant (all interaction p values >0.05) difference in 12W-CDP treatment effect (**Figure 2**)
 - — Trends (p<0.3) for differences in treatment effect between subgroups were observed for sex, presence of T1 Gd+ lesions at baseline and age
 - — Disability, MRI and relapse outcomes were further analyzed by these subgroups

Sex Subgroup Analyses

- Across disability, relapse and MRI outcomes, OCR recipients (including females) received at least numerical benefit, relative to those receiving PBO
- Disability and relapse:
 - — A trend for a greater magnitude of OCR treatment effect in male patients vs. female patients was observed for 12W-CDP, and 12W-T25FW and 12W-9HPT (**Figure 3a**)
 - — Analyses of 24W-CDP were generally comparable with 12W-CDP (**Figure 3b**)
 - — There was no sex-based trends observed on ARR (**Figure 3c**)
- MRI:
 - — There were no sex-based trends observed on the change in T2 lesion volume from baseline (**Figure 3d**) or total brain volume (**Figure 3e**)

Baseline T1 Gd+ Lesion Subgroup Analyses

- Across disability, relapse and MRI outcomes, OCR recipients (including those without baseline T1 Gd+ lesions) received at least numerical benefit, relative to those receiving PBO
- Disability and relapse:
 - — A trend for a greater magnitude of OCR treatment effect in patients with baseline T1 Gd+ lesions vs. without was observed for 12W-CDP and 12W-9HPT (**Figure 4a**); comparable observations were made for 24W-CDP and 24W-9HPT (**Figure 4b**)
- MRI:
 - — A trend for a greater magnitude of OCR treatment effect in patients with baseline T1 Gd+ lesions vs. without was observed for T2 lesion volume (**Figure 4d**)
 - — There was no observed subgroup trend in the absolute change in brain volume (**Figure 4e**)

Age Subgroup Analyses

- Across disability, relapse and MRI outcomes, OCR recipients (including those >45 years) received at least numerical benefit, relative to those receiving PBO
- Disability and relapse:
 - — A trend for a greater magnitude of OCR treatment effect in patients aged ≤45 years vs. >45 years for 12W-CDP and ≥20% increases in 12W-9HPT and 12W-T25FW were observed (**Figure 5a**); comparable observations were made for 24-week confirmed data (**Figure 5b**)
 - — A trend for a greater magnitude of treatment effect on ARR was observed in patients aged ≤45 years vs. >45 years (**Figure 5c**)
- MRI:
 - — A trend for a greater magnitude of treatment effect in terms of T2 lesion volume (**Figure 5d**) and total brain volume change (**Figure 5e**) was observed in patients aged ≤45 years vs. >45 years
- Age subgroup analyses should be viewed in the context of the inverse correlation between age and acute MRI activity (**Figure 6**)

Figure 2. 12W-CDP analyzed by prespecified subgroups

Baseline risk factors	PBO (n=244) n (events)	OCR (n=488) n (events)	HR (95% CI)	p interact.
All patients	244 (96)	487 (160)	0.76 (0.59, 0.98)	NA
Baseline weight, <75 kg	142 (53)	290 (93)	0.76 (0.54, 1.07)	0.92
Baseline weight, ≥75 kg	101 (43)	195 (67)	0.76 (0.52, 1.12)	
Duration since MS symptom onset				
≤3 years	53 (24)	79 (25)	0.63 (0.36, 1.12)	0.68
>3 to ≤5 years	52 (20)	111 (39)	0.92 (0.53, 1.58)	
>5 to ≤10 years	96 (34)	202 (60)	0.83 (0.54, 1.28)	
>10 years	36 (15)	81 (30)	0.63 (0.33, 1.19)	
Baseline EDSS score, ≤5.5	163 (61)	348 (100)	0.73 (0.53, 1.00)	0.66
Baseline EDSS score, >5.5	81 (35)	139 (60)	0.84 (0.55, 1.28)	
Prior MS DMT,[a] yes	30 (15)	55 (18)	0.65 (0.32, 1.31)	0.53
Prior MS DMT,[a] no	214 (81)	432 (142)	0.79 (0.60, 1.04)	
Region, ROW	210 (84)	420 (145)	0.79 (0.60, 1.03)	0.41
Region, USA	34 (12)	67 (15)	0.55 (0.26, 1.18)	
Body mass index, <25 kg/m²	139 (57)	289 (91)	0.68 (0.48, 0.94)	0.31
Body mass index, ≥25 kg/m²	103 (39)	196 (69)	0.89 (0.60, 1.33)	
Age group, ≤45 years	118 (49)	230 (71)	0.64 (0.45, 0.92)	0.23
Age group, >45 years	126 (47)	257 (89)	0.88 (0.62, 1.26)	
Baseline T1 Gd+ lesions, yes	60 (27)	133 (43)	0.65 (0.40, 1.06)	0.21
Baseline T1 Gd+ lesions, no	183 (68)	350 (115)	0.84 (0.62, 1.13)	
Sex, Female	124 (44)	236 (85)	0.94 (0.66, 1.36)	0.10
Sex, Male	120 (52)	251 (75)	0.61 (0.43, 0.88)	

Favours OCR ← → Favours PBO; Nominal interaction p<0.3; Hazard ratio (95% CI) axis: 0.25, 0.5, 1.0, 2.0

[a]Excluding corticosteroids.
12W-CDP, 12-week confirmed disability progression; DMT, disease-modifying therapy; EDSS, Expanded Disability Status Scale; Gd+, gadolinium-enhancing; HR, hazard ratio; MS, multiple sclerosis; NA, not applicable; OCR, ocrelizumab; PBO, placebo; ROW, rest of World.

Figure 3. Sex subgroup analyses

a. 12-week / **b. 24-week**

Endpoint	Subgroup	PBO (n=244) n (events)	OCR (n=488) n (events)	HR (95% CI)	p interact.
a. 12-week CDP	Male	120 (52)	251 (75)	0.61 (0.43, 0.88)	0.10
	Female	124 (44)	236 (85)	0.94 (0.66, 1.36)	
9HPT	Male	120 (40)	251 (51)	0.54 (0.36, 0.82)	0.94
	Female	124 (26)	237 (32)	0.56 (0.33, 0.95)	
T25FW	Male	120 (76)	251 (121)	0.69 (0.52, 0.92)	0.33
	Female	124 (69)	237 (117)	0.82 (0.61, 1.11)	
b. 24-week CDP	Male	120 (46)	251 (68)	0.64 (0.44, 0.93)	0.21
	Female	124 (41)	236 (76)	0.89 (0.61, 1.31)	
9HPT	Male	120 (32)	251 (43)	0.60 (0.38, 0.94)	0.55
	Female	124 (25)	237 (26)	0.48 (0.28, 0.84)	
T25FW	Male	120 (70)	251 (103)	0.63 (0.47, 0.86)	0.15
	Female	124 (57)	237 (99)	0.86 (0.62, 1.19)	

Favours OCR ← → Favours PBO; Hazard ratio (95% CI) axis: 0.25, 0.5, 1.0, 2.0

c. ARR

Subgroup	PBO (n=244) n (relapse)	OCR (n=488) n (relapse)	RR (95% CI)	p interact.
Male	120 (21)	251 (15)	0.32 (0.13, 0.77)	0.75
Female	124 (15)	237 (12)	0.39 (0.17, 0.91)	

Rate ratio (95% CI) axis: 0.1, 0.2, 0.4, 0.8, 1.0

d. N/E T2 lesion volume[a]

Subgroup	PBO (n=244) n (change)	OCR (n=488) n (change)	Change (95% CI)	p interact.
Male	89 (1.07)	207 (0.96)	0.90 (0.87, 0.93)	0.87
Female	94 (1.07)	193 (0.96)	0.90 (0.87, 0.94)	

Ratio of adjusted mean change (95% CI) axis: 0.8, 1.0

e. Total brain volume[b]

Subgroup	PBO (n=244) n (change)	OCR (n=488) n (change)	Change (95% CI)	p interact.
Male	76 (-1.15)	166 (-0.93)	0.23 (0.00, 0.45)	0.74
Female	74 (-1.03)	159 (-0.88)	0.16 (-0.08, 0.40)	

Favours PBO ← → Favours OCR; Difference of adjusted mean change (95% CI) axis: -0.1, 0.0, 0.1, 0.3, 0.5

Figure 4. Baseline T1 Gd+ lesion subgroup analyses

Endpoint	Subgroup	PBO (n=243) n (events)	OCR (n=484) n (events)	HR (95% CI)	p interact.
a. 12-week CDP	Present	60 (27)	133 (43)	0.65 (0.40, 1.06)	0.21
	Absent	183 (68)	350 (115)	0.84 (0.62, 1.13)	
9HPT	Present	60 (22)	133 (24)	0.42 (0.23, 0.76)[c]	0.18
	Absent	183 (43)	351 (58)	0.64 (0.43, 0.95)	
T25FW	Present	60 (38)	133 (65)	0.67 (0.45, 1.02)	0.58
	Absent	183 (106)	351 (170)	0.78 (0.61, 0.99)	
b. 24-week CDP	Present	60 (23)	133 (39)	0.67 (0.40, 1.14)	0.35
	Absent	183 (63)	350 (103)	0.81 (0.59, 1.10)	
9HPT	Present	60 (21)	133 (21)	0.39 (0.21, 0.72)[c]	0.15
	Absent	183 (36)	351 (47)	0.63 (0.41, 0.97)	
T25FW	Present	60 (30)	133 (53)	0.71 (0.45, 1.12)	0.83
	Absent	183 (96)	351 (146)	0.74 (0.58, 0.96)	

Favours OCR ← → Favours PBO; Hazard ratio (95% CI) axis: 0.25, 0.5, 1.0, 2.0

c. ARR

Subgroup	PBO (n=244) n (relapse)	OCR (n=488) n (relapse)	RR (95% CI)	p interact.
Present	60 (10)	133 (8)	0.27 (0.09, 0.87)	0.68
Absent	183 (26)	351 (18)	0.36 (0.17, 0.76)	

Rate ratio (95% CI) axis: 0.1, 0.3, 1.0

d. N/E T2 lesion volume[a]

Subgroup	PBO (n=244) n (change)	OCR (n=488) n (change)	Change (95% CI)	p interact.
Present	39 (1.12)	107 (0.96)	0.86 (0.82, 0.90)	0.05
Absent	144 (1.05)	291 (0.97)	0.91 (0.89, 0.94)	

Ratio of adjusted mean change (95% CI) axis: 0.8, 0.9, 1.0

e. Total brain volume[b]

Subgroup	PBO (n=244) n (change)	OCR (n=488) n (change)	Change (95% CI)	p interact.
Present	31 (-1.39)	83 (-1.21)	0.18 (-0.21, 0.57)	0.90
Absent	119 (-1.00)	241 (-0.80)	0.21 (0.03, 0.38)	

Favours PBO ← → Favours OCR; Difference of adjusted mean change (95% CI) axis: -0.3, 0.0, 0.2, 0.7

Figure 5. Age subgroup analyses

Endpoint	Subgroup	PBO (n=244) n (events)	OCR (n=488) n (events)	HR (95% CI)	p interact.
a. 12-week CDP	≤45 years	118 (49)	230 (71)	0.64 (0.45, 0.92)	0.23
	>45 years	126 (47)	257 (89)	0.88 (0.62, 1.26)	
9HPT	≤45 years	118 (38)	230 (42)	0.48 (0.31, 0.75)	0.29
	>45 years	126 (28)	258 (41)	0.67 (0.41, 1.08)	
T25FW	≤45 years	118 (72)	230 (105)	0.64 (0.47, 0.86)	0.16
	>45 years	126 (73)	258 (133)	0.85 (0.64, 1.14)	
b. 24-week CDP	≤45 years	118 (46)	230 (65)	0.61 (0.42, 0.90)	0.16
	>45 years	126 (41)	257 (79)	0.92 (0.63, 1.34)	
9HPT	≤45 years	118 (33)	230 (36)	0.48 (0.30, 0.77)	0.40
	>45 years	126 (24)	258 (33)	0.63 (0.38, 1.07)	
T25FW	≤45 years	118 (64)	230 (85)	0.58 (0.42, 0.80)	0.05
	>45 years	126 (63)	258 (117)	0.89 (0.66, 1.21)	

Favours OCR ← → Favours PBO; Hazard ratio (95% CI) axis: 0.25, 0.5, 1.0, 2.0

c. ARR

Subgroup	PBO (n=244) n (relapse)	OCR (n=488) n (relapse)	RR (95% CI)	p interact.
≤45 years	118 (22)	230 (14)	0.28 (0.12, 0.64)	0.42
>45 years	126 (14)	258 (13)	0.46 (0.19, 1.12)	

Rate ratio (95% CI) axis: 0.1, 0.2, 0.4, 0.8, 1.0, 1.6

d. N/E T2 lesion volume[a]

Subgroup	PBO (n=244) n (change)	OCR (n=488) n (change)	Change (95% CI)	p interact.
≤45 years	84 (1.10)	183 (0.96)	0.88 (0.84, 0.91)	0.04
>45 years	99 (1.06)	217 (0.97)	0.92 (0.89, 0.95)	

Ratio of adjusted mean change (95% CI) axis: 0.75, 0.85, 0.95, 1.0, 1.05, 1.15

e. Total brain volume[b]

Subgroup	PBO (n=244) n (change)	OCR (n=488) n (change)	Change (95% CI)	p interact.
≤45 years	71 (-1.26)	145 (-0.99)	0.27 (0.03, 0.52)	0.42
>45 years	79 (-0.94)	180 (-0.81)	0.13 (-0.09, 0.35)	

Favours PBO ← → Favours OCR; Difference of adjusted mean change (95% CI) axis: -0.5, 0.0, 0.5, 1.0

[a]Total relative change (baseline to Week 120); [b]Absolute change (Weeks 24-120); [c]Lower limit of confidence interval extends beyond plotted axis limit. 9HPT, 9-hole peg test; ARR, annualized relapse rate; CDP, confirmed disability progression; EDSS, Expanded Disability Status Scale; Gd+, gadolinium-enhancing; HR, hazard ratio; N/E, new/enlarging; OCR, ocrelizumab; PBO, placebo; RR, rate ratio; TBV, total brain volume; T25FW, timed 25-foot walk.

Figure 6. Age versus proportion of patients with T1 Gd+ and N/E T2 lesions (Baseline to Week 120, PBO arm)

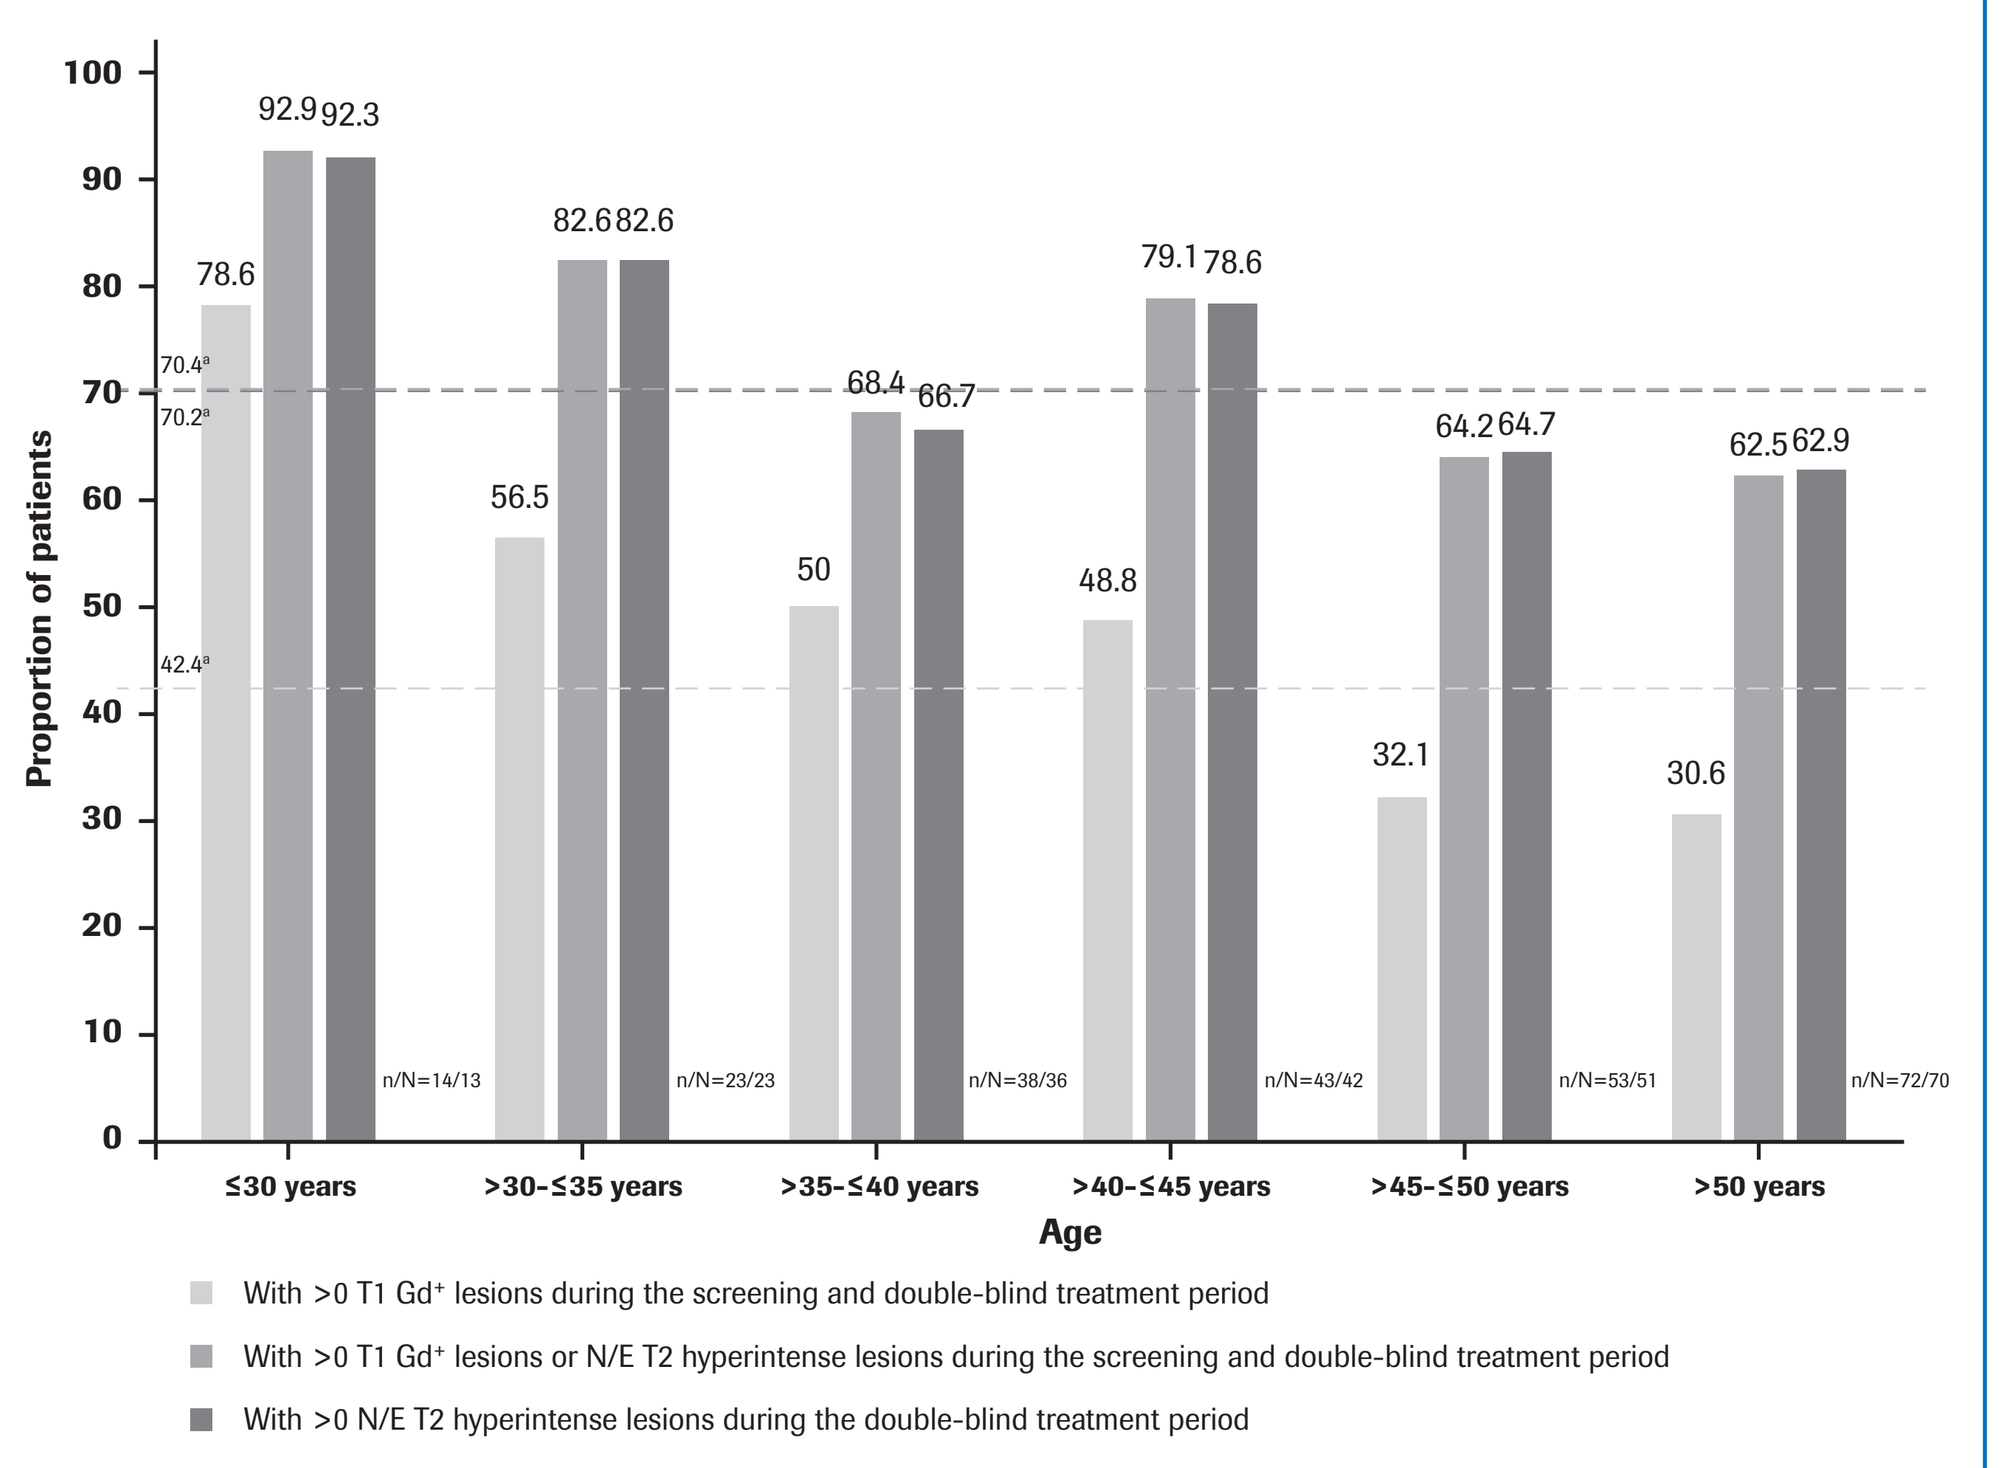

Proportion of patients
100
90
80
70
60
50
40
30
20
10
0
78.6
92.9
92.3
82.6
82.6
56.5
50
68.4
66.7
48.8
79.1
78.6
32.1
64.2
64.7
30.6
62.5
62.9
70.4[a]
70.2[a]
42.4[a]
n/N=14/13
n/N=23/23
n/N=38/36
n/N=43/42
n/N=53/51
n/N=72/70
≤30 years
>30–≤35 years
>35–≤40 years
>40–≤45 years
>45–≤50 years
>50 years
Age
With >0 T1 Gd+ lesions during the screening and double-blind treatment period
With >0 T1 Gd+ lesions or N/E T2 hyperintense lesions during the screening and double-blind treatment period
With >0 N/E T2 hyperintense lesions during the double-blind treatment period

[a]Data for overall group, n/N=243/235. N/N values are the analyzable population sample size for T1 Gd+ lesions vs. new/enlarging T2 hyperintense lesion detection in the respective age subgroups.
Gd+, gadolinium-enhancing; N/E, new/enlarging; PBO, placebo.

CONCLUSIONS

- There were no statistically significant prespecified subgroup effects on the primary endpoint, 12W-CDP
 - — Numerical differences (nominal interaction p<0.3) based on sex, baseline T1 Gd+ lesion status and age were observed
- Directionally consistent point estimates favoring ocrelizumab vs PBO were seen across all clinical and MRI endpoints in prespecified subgroups of the ORATORIO study
- A trend was observed for males to derive more benefit than female patients for 12W-CDP (driven by worse progression in male PBO recipients); male and female patients benefited from ocrelizumab on key clinical and imaging secondary/exploratory endpoints
- Although the effect of ocrelizumab was generally larger in patients with baseline T1 Gd+ lesions and/or at a younger age, older patients and those without T1 Gd+lesions at baseline also derived benefit across key endpoints
 - — Age-related subgroup differences may relate to a higher prevalence of MRI features of acute inflammatory activity in younger patients
- The study was not powered for subgroup analyses; these data should be interpreted with caution

DISCLOSURES

JS Wolinsky has served on advisory boards, data monitoring or steering committees, has consulting agreements from the following entities: AbbVie, Actelion, Alkermes, Bayer HealthCare, Biogen, Bionest, Celgene, Clene Nanomedicine, EMD Serono, Forward Pharma A/S, GeNeuro, MedDay Pharmaceuticals, Novartis Pharmaceuticals, Otsuka, PTC Therapeutics, Roche Genentech, Sanofi Genzyme, Strategic Consultants International, Takeda and Teva Pharmaceuticals; royalties are received for outlicensed monoclonal antibodies through UTHealth from Millipore Corporation. X Montalban has received speaker honoraria and travel expense reimbursement for participation in scientific meetings, been a steering committee member of clinical trials or served on advisory boards of clinical trials for Actelion, Almirall, Bayer, Biogen, F. Hoffmann-La Roche Ltd, Genzyme, Merck, Novartis, Octapharma, Receptos, Sanofi, Teva and Trophos. SL Hauser serves on the Scientific Advisory Boards for Annexon, Symbiotix, Bionure and Molecular Stethoscope. He is on the Board of Trustees for Neurona Therapeutics. SL Hauser also has received travel reimbursement and writing assistance from F. Hoffmann-La Roche Ltd for CD20-related meetings and presentations. L Kappos' institution, the University Hospital Basel, has received research support and payments that were used exclusively for research support for L Kappos' activities as principal investigator and member or chair of planning and steering committees or advisory boards for trials sponsored by Actelion, Addex, Almirall, Bayer HealthCare Pharmaceuticals, CLC Behring, F. Hoffmann-La Roche Ltd and Genentech, Inc., GeNeuro SA, Genzyme, Merck Serono, Mitsubishi Pharma, Novartis, Octapharma, Ono Pharmaceutical, Pfizer, Receptos, Sanofi, Santhera, Siemens, Teva, UCB and XenoPort; has received license fees for Neurostatus products; and has received research grants from the European Union, Gianni Rubatto Foundation, Novartis Research Foundation, Roche Research Foundation, Swiss Multiple Sclerosis Society and Swiss National Research Foundation. L Julian is an employee of Genentech, Inc., and a shareholder of F. Hoffmann-La Roche Ltd. M Manfrini is an employee of F. Hoffmann-La Roche Ltd. S Belachew is an employee and shareholder of F. Hoffmann-La Roche Ltd. F Model is an employee and shareholder of F. Hoffmann-La Roche Ltd. S Hubeaux is an employee of F. Hoffmann-La Roche Ltd. A Bar-Or has served on scientific advisory boards for Biogen, F. Hoffmann-La Roche Ltd and Genentech, Inc., GlaxoSmithKline, Guthy-Jackson/GGF, MedImmune, Merck/EMD Serono, Mitsubishi Tanabe, Ono, Receptos and Sanofi Genzyme and has received research support from Biogen, Novartis and Sanofi Genzyme.

ACKNOWLEDGMENTS

We would like to thank all patients, their families, and the investigators who participated in these trials. This research was funded by F. Hoffmann-La Roche Ltd, Basel, Switzerland. Writing and editorial assistance for this presentation was provided by Articulate Science, UK, and funded by F. Hoffmann-La Roche Ltd, Basel, Switzerland.

REFERENCES

1. Montalban X, *et al. N Engl J Med* 2017; 376:209-220.

Presented at the 2018 Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC); May 30–June 2, 2018; Nashville, TN, USA

Poster DX42