Efficacy Outcomes in Patients Randomized to Fingolimod or Injectable Disease-Modifying Therapies in PREFER*MS*: Effect of Previous Treatment Cycles

Samuel F Hunter[1], Florian P Thomas[2,3], Xiangyi Meng[4], Lesley Schofield[4], Scott Kolodny[4], Nadia Tenenbaum[4], Bruce AC Cree[5], on behalf of the PREFER*MS* investigators

[1]Advanced Neurosciences Institute, Franklin, TN, USA; [2]Hackensack Meridian School of Medicine at Seton Hall University, South Orange, NJ, USA; [3]Hackensack University Medical Center, Hackensack, NJ, USA; [4]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [5]UCSF Weill Institute for Neurosciences, Department of Neurology, University of California San Francisco, San Francisco, CA, USA

DX52

Introduction

- Fingolimod 0.5 mg is a once-daily, oral therapy for relapsing forms of multiple sclerosis (RMS),[1] approved for first-line use in many countries
 - New American Academy of Neurology guidelines suggest high-efficacy therapies such as fingolimod should be considered as a first-line therapy for patients with active disease[2]
- In clinical practice, fingolimod is often prescribed as a second-line therapy following one or more injectable disease-modifying therapies (iDMTs), such as glatiramer acetate, interferon β-1a or interferon β-1b
- PREFER*MS* was a 12-month, Phase 4, active-controlled, open-label, multicenter study that compared treatment retention and clinical outcomes with fingolimod and iDMTs in patients with RMS[2,3]
 - Just over half the patients in PREFER*MS* had been treated with one class of iDMT; the remainder were treatment-naïve
 - Patients were randomized to fingolimod 0.5 mg or an iDMT, and allowed a single switch to an alternative, on-study treatment
 - For those randomized to an iDMT, efficacy outcomes improved following switch to fingolimod[4,5]
- We examined the number of iDMTs received before initiating fingolimod on efficacy outcomes in patients with RMS

Objective

- Assess impact of number of previous cycles of iDMT (two, one or none) on response to fingolimod in PREFER*MS*

Methods

Study design

- PREFER*MS* was conducted at 117 sites in the USA (**Figure 1**)
- Primary endpoint was retention on randomized treatment over 12 months
- Secondary endpoints included radiological outcomes, such as brain atrophy and lesion counts
- Enrolled patients were treatment-naïve or had previously received one class of iDMT (glatiramer acetate, interferon β-1a or interferon β-1b)
- Patients were randomized (1:1) to fingolimod 0.5 mg or a preselected iDMT, and observed quarterly over 12 months
 - Patients previously treated with an iDMT received an alternative iDMT class if randomized to iDMT
- One treatment switch was allowed for any reason after 3 months, although patients could switch earlier for efficacy or safety reasons

Figure 1. PREFER*MS* study design

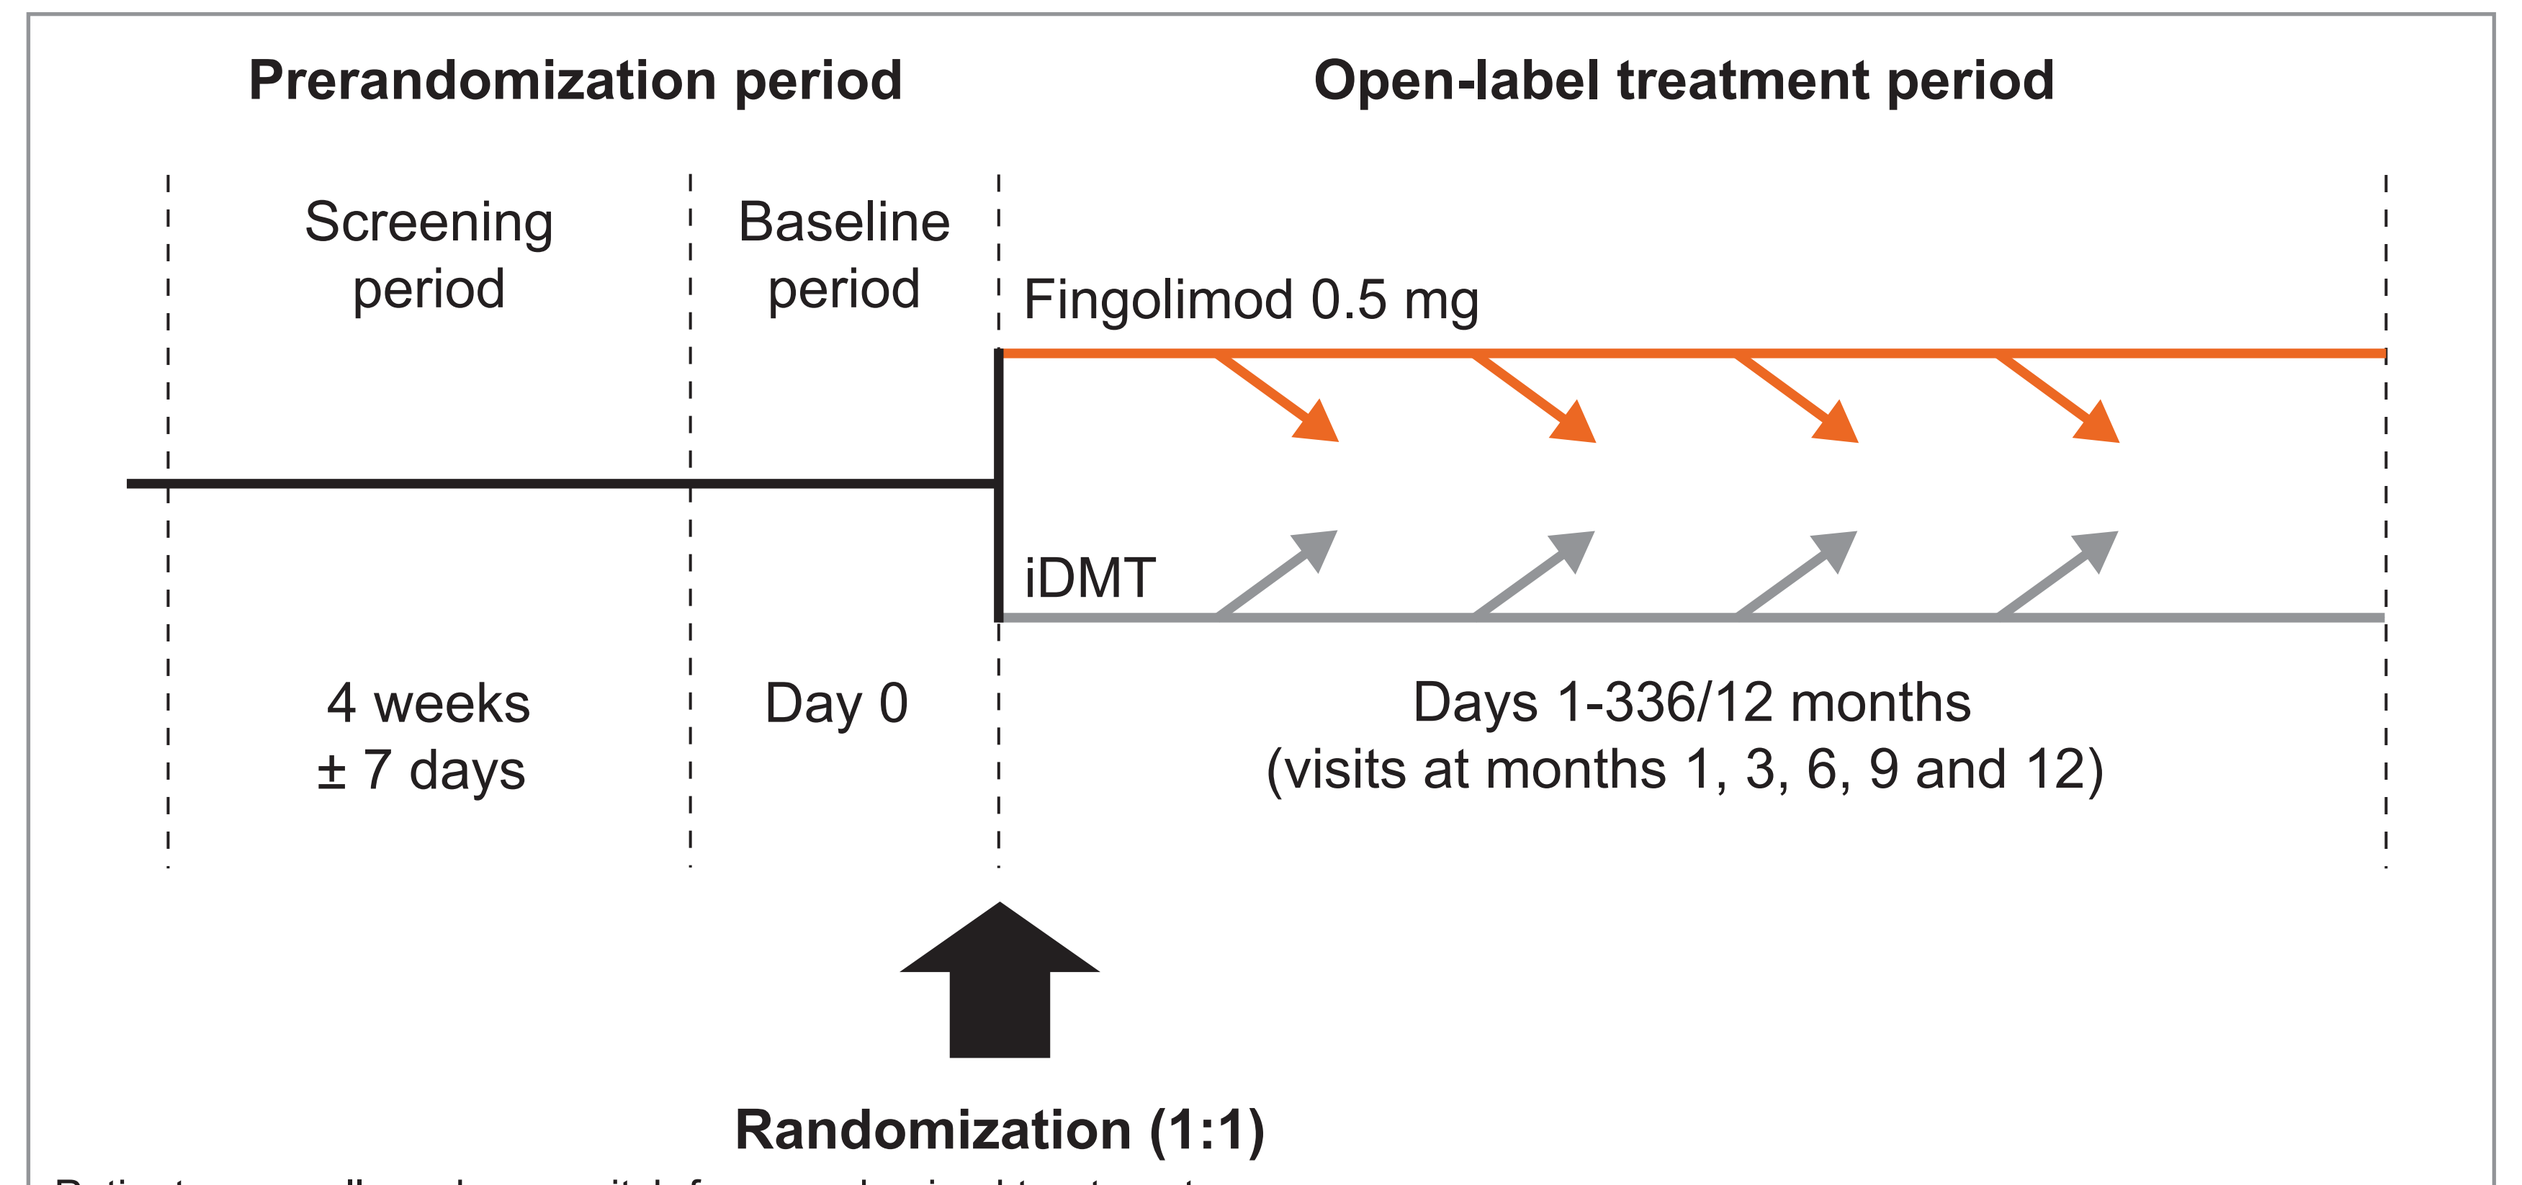
Prerandomization period
Open-label treatment period
Screening period
Baseline period
Fingolimod 0.5 mg
iDMT
4 weeks ± 7 days
Day 0
Days 1-336/12 months (visits at months 1, 3, 6, 9 and 12)
Randomization (1:1)

Patients were allowed one switch from randomized treatment
Reason for switch ≤3 months: safety or efficacy
Reason for switch at 3-12 months: safety, efficacy, tolerability or convenience
iDMT, injectable disease-modifying therapy

Analyses

- Three patient subgroups were analyzed post hoc according to iDMT treatment history:
 - **Group A (two iDMTs):** patients who had received one iDMT before PREFER*MS*, were randomized to a new iDMT and subsequently switched to fingolimod (n=155). Data for this subgroup are reported for two time points:
 - Last study visit before switching to fingolimod (ie pre-fingolimod, after two previous iDMTs)
 - End of study following the switch to fingolimod (ie post-fingolimod, after two previous iDMTs)
 - **Group B (one iDMT):** patients who had received one iDMT before PREFER*MS* and were randomized to fingolimod (ie after one previous iDMT cycle; n=233)
 - **Group C (no iDMT):** treatment-naïve patients who were randomized to fingolimod (ie no previous iDMT cycles; n=213)
 - Data from groups B and C were reported for last study visit at which patients received fingolimod
- Outcomes evaluated were:
 - exposure-adjusted percentage brain volume loss (BVL) from baseline
 - number of new gadolinium-enhancing (Gd+) lesions
 - change in overall Gd+ lesion count from baseline
- All outcomes are presented as means with 95% confidence intervals (CI)
- Effect of switching to fingolimod following two iDMT cycles was investigated in group A (two iDMTs) by assessing efficacy outcomes at the pre-fingolimod versus post-fingolimod time points, and compared with post-fingolimod outcomes in group B (one iDMT) and group C (no iDMT)
- Comparisons were for hypothesis generation only; PREFER*MS* was not powered to detect treatment effects in subgroups of patients

Results

- In total, 875 patients were randomized, and 861 (98.4%) were included in the full analysis set (fingolimod, n=433; iDMTs, n=428)
 - At baseline, 53.8% of patients (n=471) were treatment-naïve; 46.2% (n=404) had previously received one class of iDMT
 - Most patients randomized to an iDMT subsequently switched to fingolimod (58.5%); few patients receiving fingolimod switched to an iDMT (6.2%)
- Demographic and baseline characteristics were generally similar in the three groups (**Table 1**)
 - Compared with previously treated patients (groups A and B), treatment-naïve patients (group C):
 - were younger and more likely to be men
 - had been diagnosed with multiple sclerosis more recently
 - had fewer relapses in the 2 years before enrollment
 - had more Gd+ lesions (**Table 1**)
- In pre-fingolimod group A (previously received two iDMTs), mean (95% CI) BVL from baseline was –0.90% (–1.36, –0.44; **Figure 2**)

Table 1. PREFER*MS* patient demographic and baseline characteristics

Characteristic	Group A (two iDMTs, n=155)	Group B (one iDMT, n=223)	Group C (no iDMT, n=213)
Age, years	42.0 (10.4)	43.4 (10.8)	39.5 (10.6)
Sex, n (%)			
Male	39 (25.2)	57 (25.6)	68 (31.9)
Female	116 (74.8)	166 (74.4)	145 (68.1)
Race, n (%)			
Caucasian	131 (84.5)	188 (84.3)	167 (78.4)
Black	22 (14.2)	31 (13.9)	38 (17.8)
Asian	0	0	1 (0.5)
Native American	0	0	1 (0.5)
Pacific Islander	1 (0.6)	0	0
Other	1 (0.6)	4 (1.8)	6 (2.8)
Height, cm	167.2 (10.3)	167.8 (8.3)	169.1 (9.6)
Weight, kg	83.5 (21.2)	81.7 (19.9)	84.2 (20.3)
BMI, kg/m²	30.0 (7.9)	28.9 (6.4)	29.5 (7.0)
Duration of MS since diagnosis, years	n=155 6.0 (5.8)	n=222 7.0 (7.1)	n=212 1.7 (4.9)
Duration of MS since first symptoms, years	n=155 7.9 (6.8)	n=222 9.6 (8.9)	n=212 4.8 (6.6)
Number of relapses in the past year	n=155 0.7 (0.8)	n=222 0.7 (1.0)	n=208 0.5 (0.9)
Number of relapses in the past 2 years	n=155 1.1 (1.3)	n=222 1.2 (1.6)	n=208 0.6 (1.3)
Normalized brain volume, cm³	n=149 1501.5 (99.8)	n=221 1509.6 (83.8)	n=210 1533.8 (82.4)
Number of Gd+ lesions	n=150 0.6 (2.0)	n=220 0.6 (2.1)	n=209 1.6 (4.8)

Randomized set. Data are mean (SD) unless stated otherwise
BMI, body mass index; Gd+, gadolinium-enhancing; iDMT, injectable disease-modifying therapy; MS, multiple sclerosis; SD, standard deviation

- Mean (95% CI) BVL from baseline was smaller in all groups after receiving fingolimod treatment (**Figure 2**)
 - Post-fingolimod group A (previously received two iDMTs): –0.42% (–0.62, –0.21)
 - Group B (previously received one iDMT): –0.37% (–0.52, –0.21)
 - Group C (previously received no iDMT): –0.61% (–0.77, –0.45)
- In pre-fingolimod group A (previously received two iDMTs), mean (95% CI) number of new Gd+ lesions was 1.46 (0.32, 2.61; **Figure 3**)
 - There were fewer new Gd+ lesions in all groups after receiving fingolimod treatment (**Figure 3**)
 - Post-fingolimod group A (previously received two iDMTs): 0.54 (0.15, 0.93)
 - Group B (previously received one iDMT): 0.13 (0.04, 0.22)
 - Group C (previously received no iDMT): 0.19 (0.05, 0.33)
- In pre-fingolimod group A (previously received two iDMTs), change in overall Gd+ lesion count showed no reduction from baseline: mean (95% CI) 0.04 (−1.24, 1.32; **Figure 4**)

Figure 2. Effect of iDMT cycles on BVL in PREFER*MS*

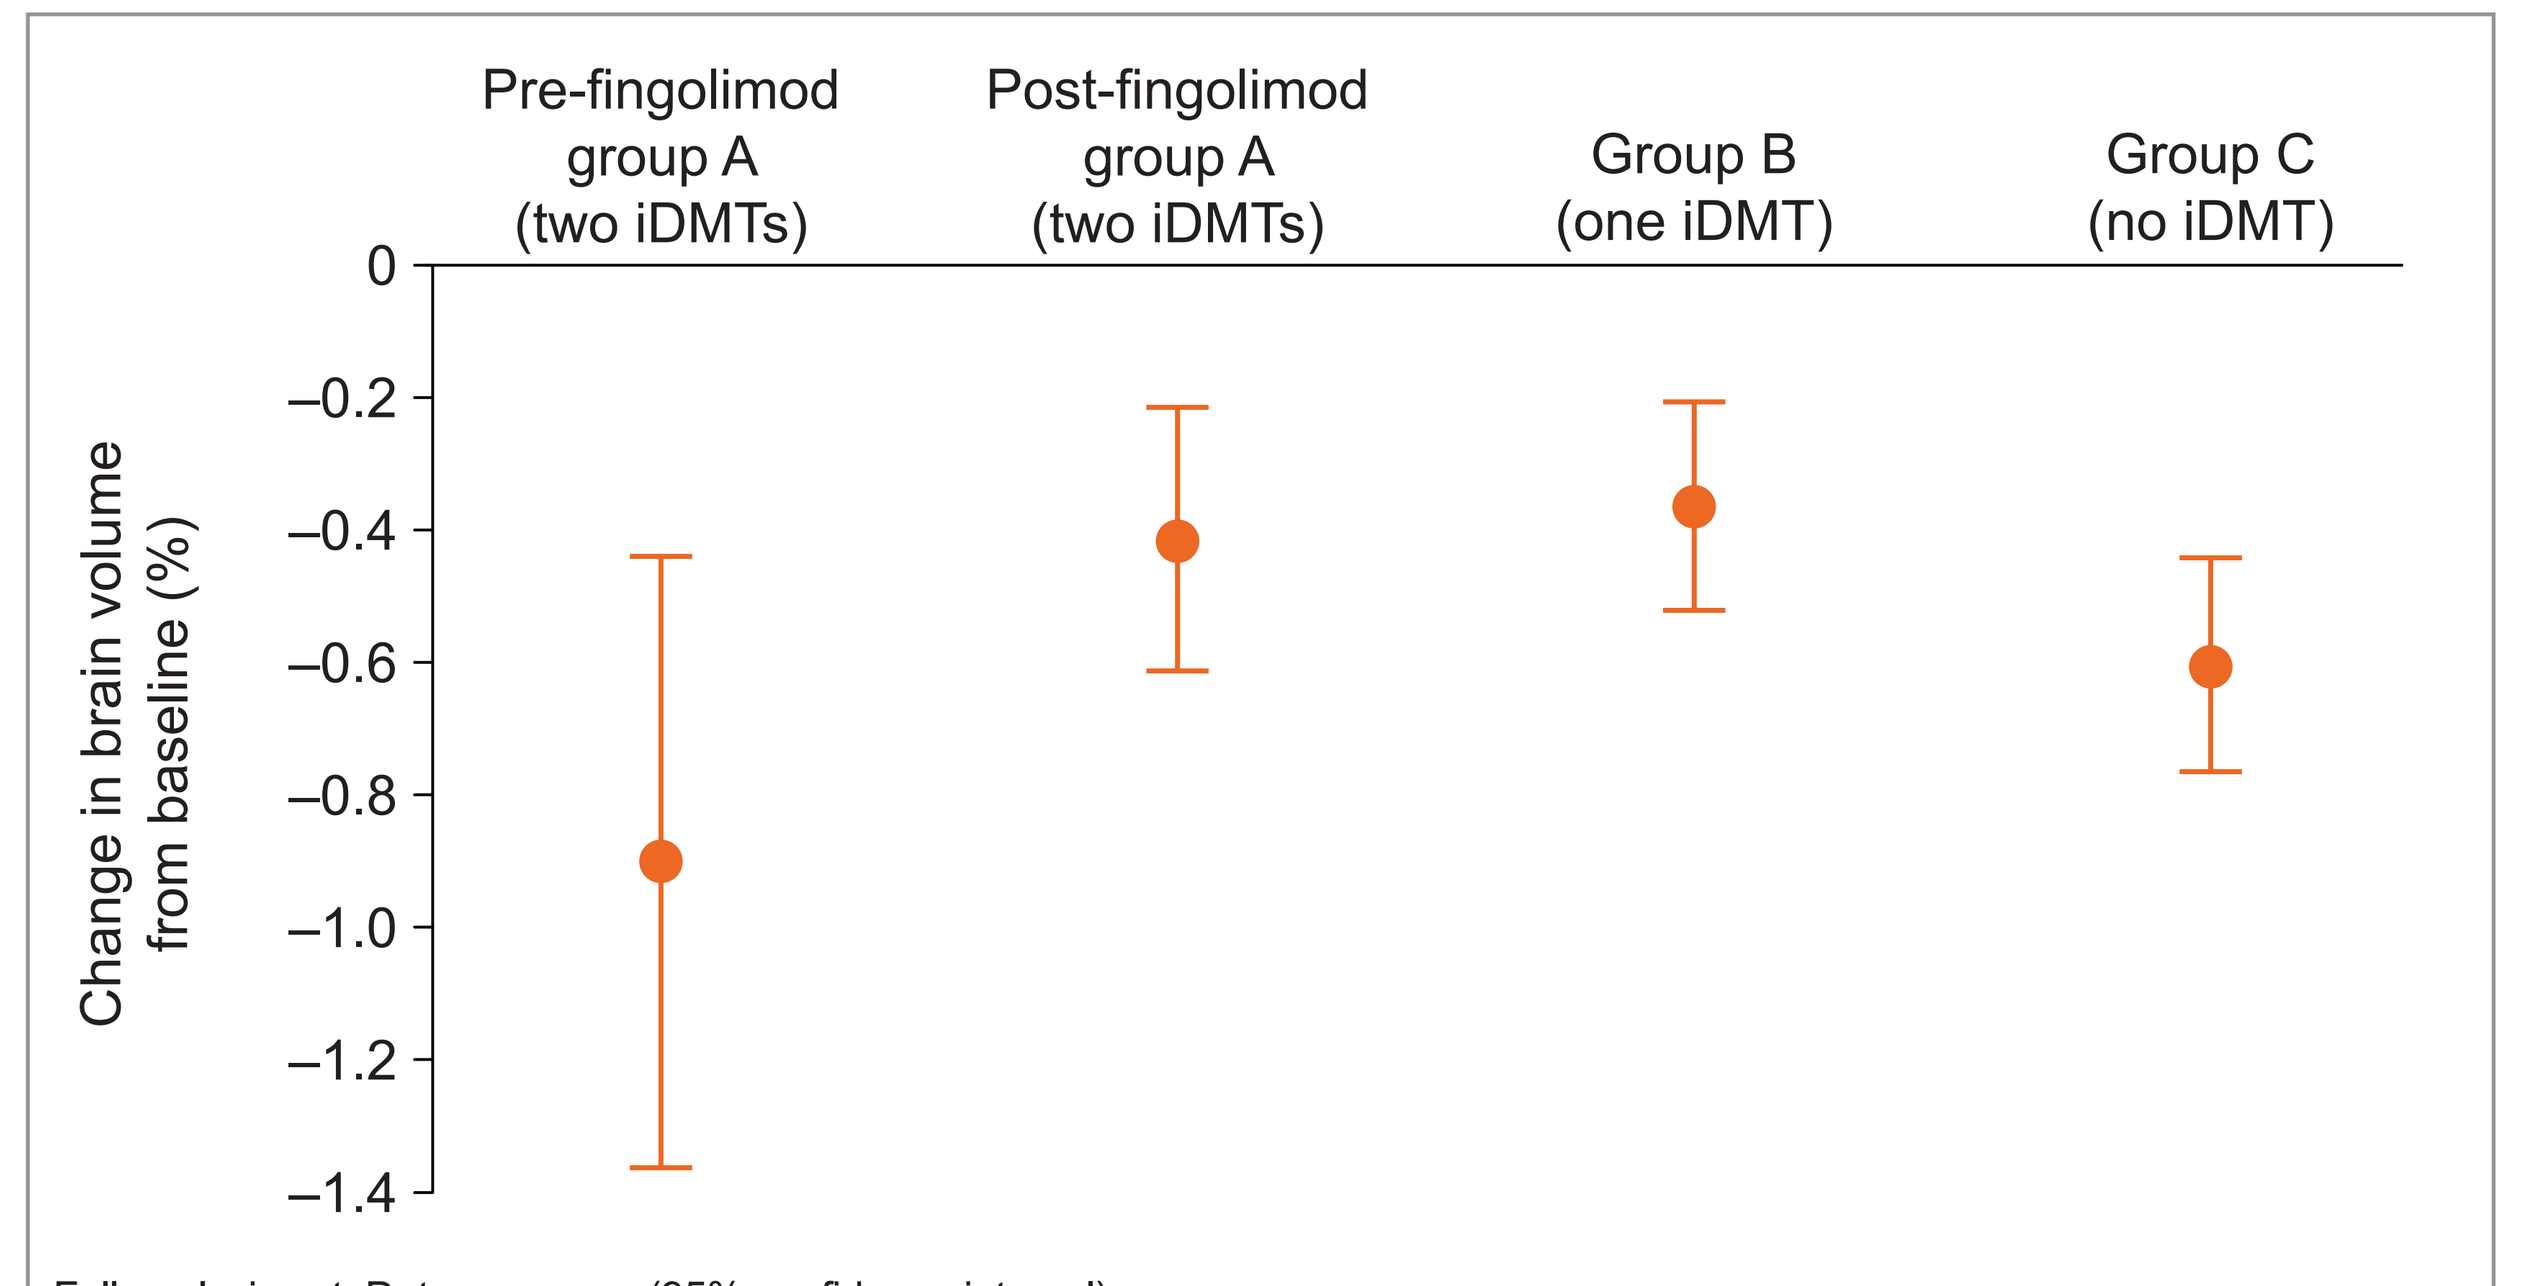
Change in brain volume from baseline (%)
0
–0.2
–0.4
–0.6
–0.8
–1.0
–1.2
–1.4
Pre-fingolimod group A (two iDMTs)
Post-fingolimod group A (two iDMTs)
Group B (one iDMT)
Group C (no iDMT)

Full analysis set. Data are mean (95% confidence interval)
BVL, brain volume loss; iDMT, injectable disease-modifying therapy

Figure 3. Effect of iDMT cycles on new Gd+ lesions in PREFER*MS*

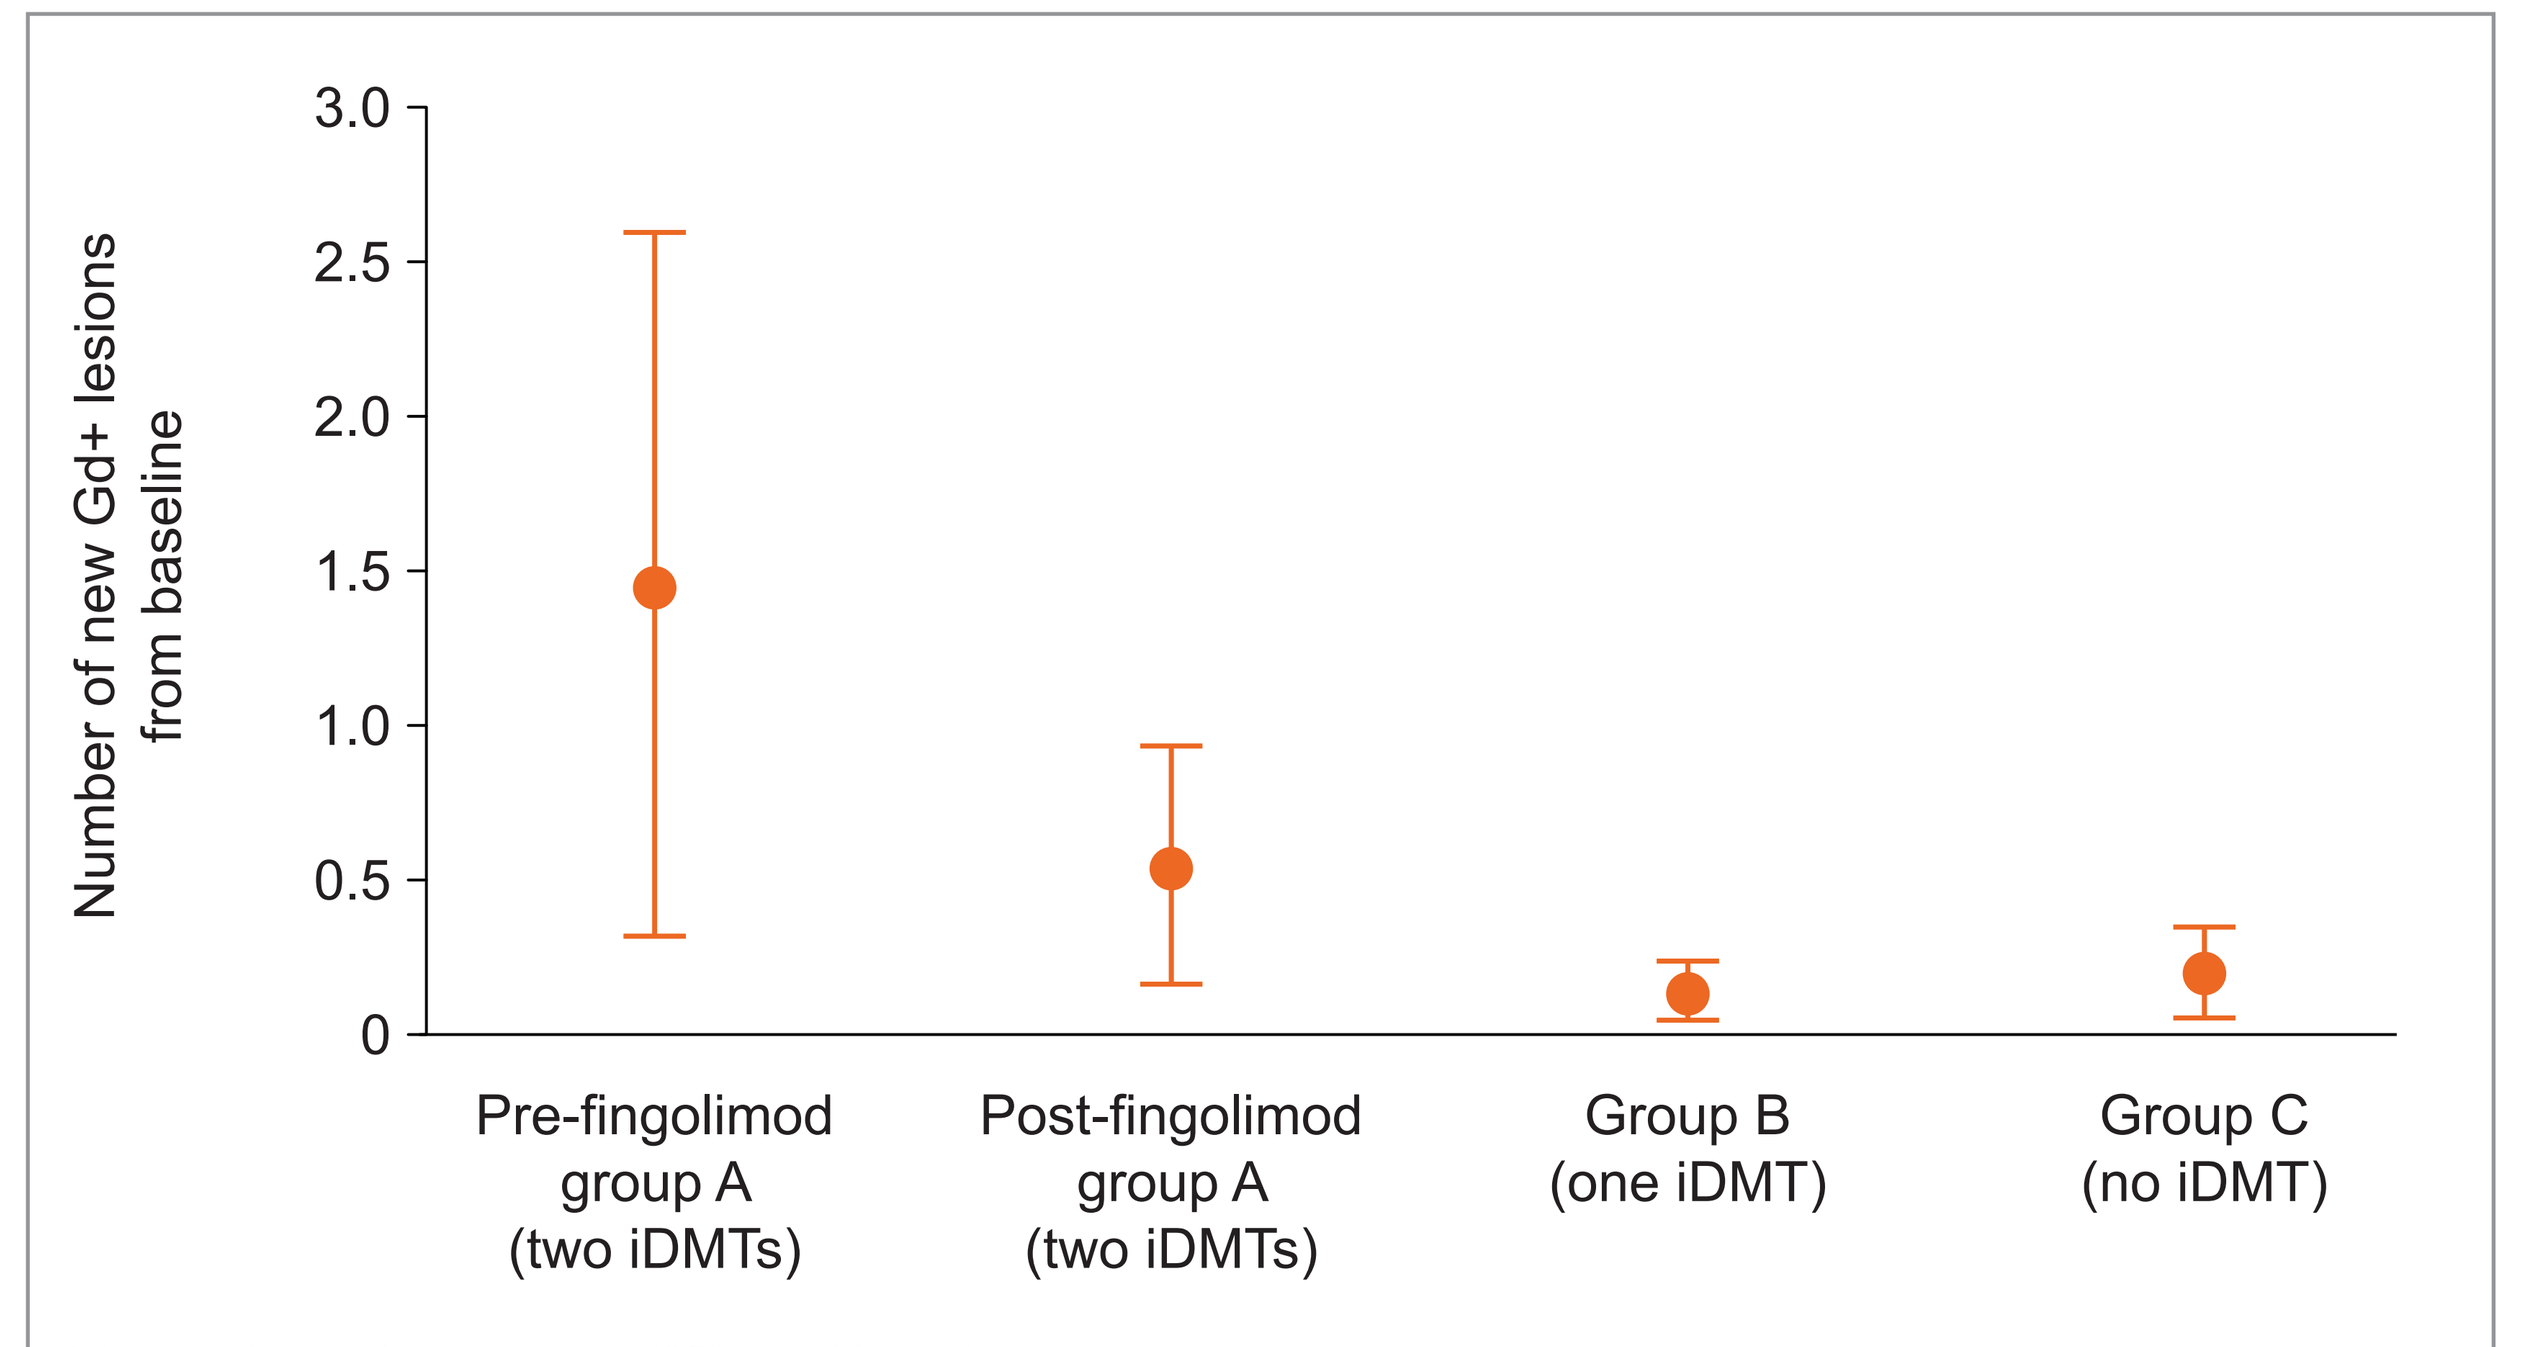
Number of new Gd+ lesions from baseline
3.0
2.5
2.0
1.5
1.0
0.5
0
Pre-fingolimod group A (two iDMTs)
Post-fingolimod group A (two iDMTs)
Group B (one iDMT)
Group C (no iDMT)

Full analysis set. Data are mean (95% confidence interval)
Gd+, gadolinium-enhancing; iDMT, injectable disease-modifying therapy

Figure 4. Effect of iDMT cycles on the overall change in Gd+ lesions in PREFER*MS*

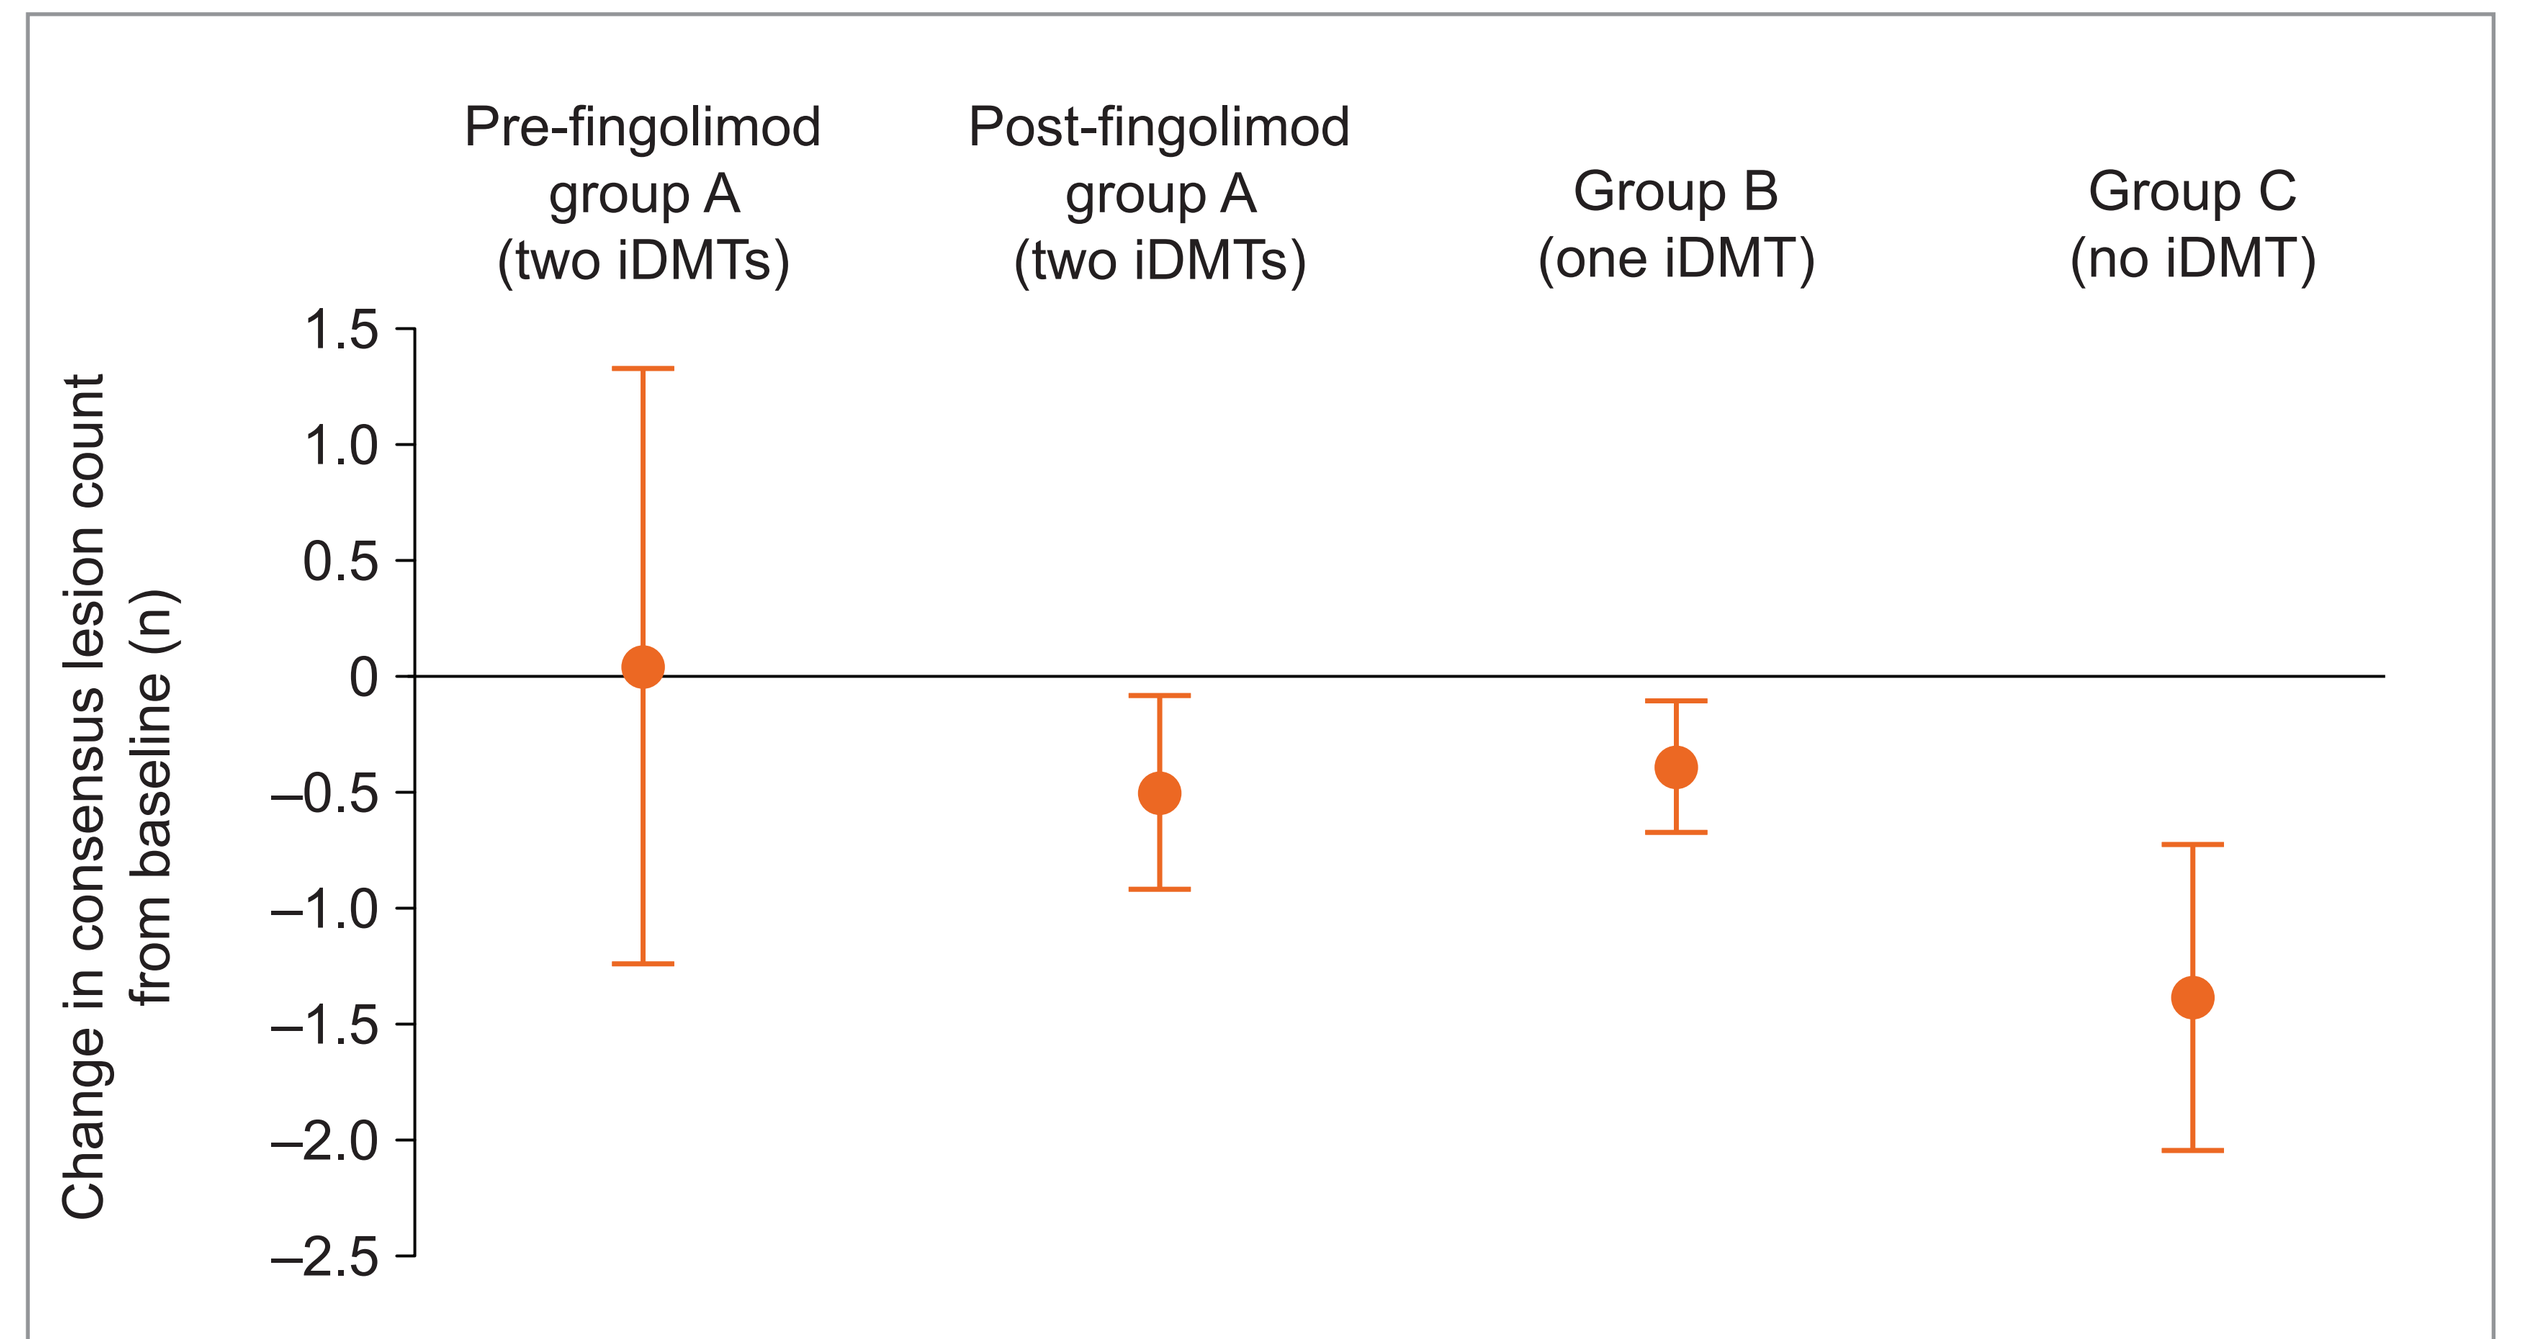
Change in consensus lesion count from baseline (n)
1.5
1.0
0.5
0
–0.5
–1.0
–1.5
–2.0
–2.5
Pre-fingolimod group A (two iDMTs)
Post-fingolimod group A (two iDMTs)
Group B (one iDMT)
Group C (no iDMT)

Full analysis set. Data are mean (95% confidence interval)
Gd+, gadolinium-enhancing; iDMT, injectable disease-modifying therapy

- Overall Gd+ lesion count was lower in all groups after receiving fingolimod (**Figure 4**)
 - Post-fingolimod group A (previously received two iDMTs): –0.49 (−0.90, −0.08)
 - Group B (previously received one iDMT): –0.39 (−0.67, −0.10)
 - Group C (previously received no iDMT): –1.38 (−2.04, −0.72)
- In pre-fingolimod group A (previously received two iDMTs), mean (95% CI) annualized relapse rate (ARR) was 0.25 (0.16, 0.38; **Figure 5**)
 - ARR was similar in all groups after receiving fingolimod (**Figure 5**)
 - Post-fingolimod group A (previously received two iDMTs): 0.16 (0.09, 0.28)
 - Group B (previously received one iDMT cycle): 0.19 (0.13, 0.29)
 - Group C (previously received no iDMT): 0.25 (0.17, 0.36)

Figure 5. Effect of iDMT cycles on ARR in PREFER*MS*

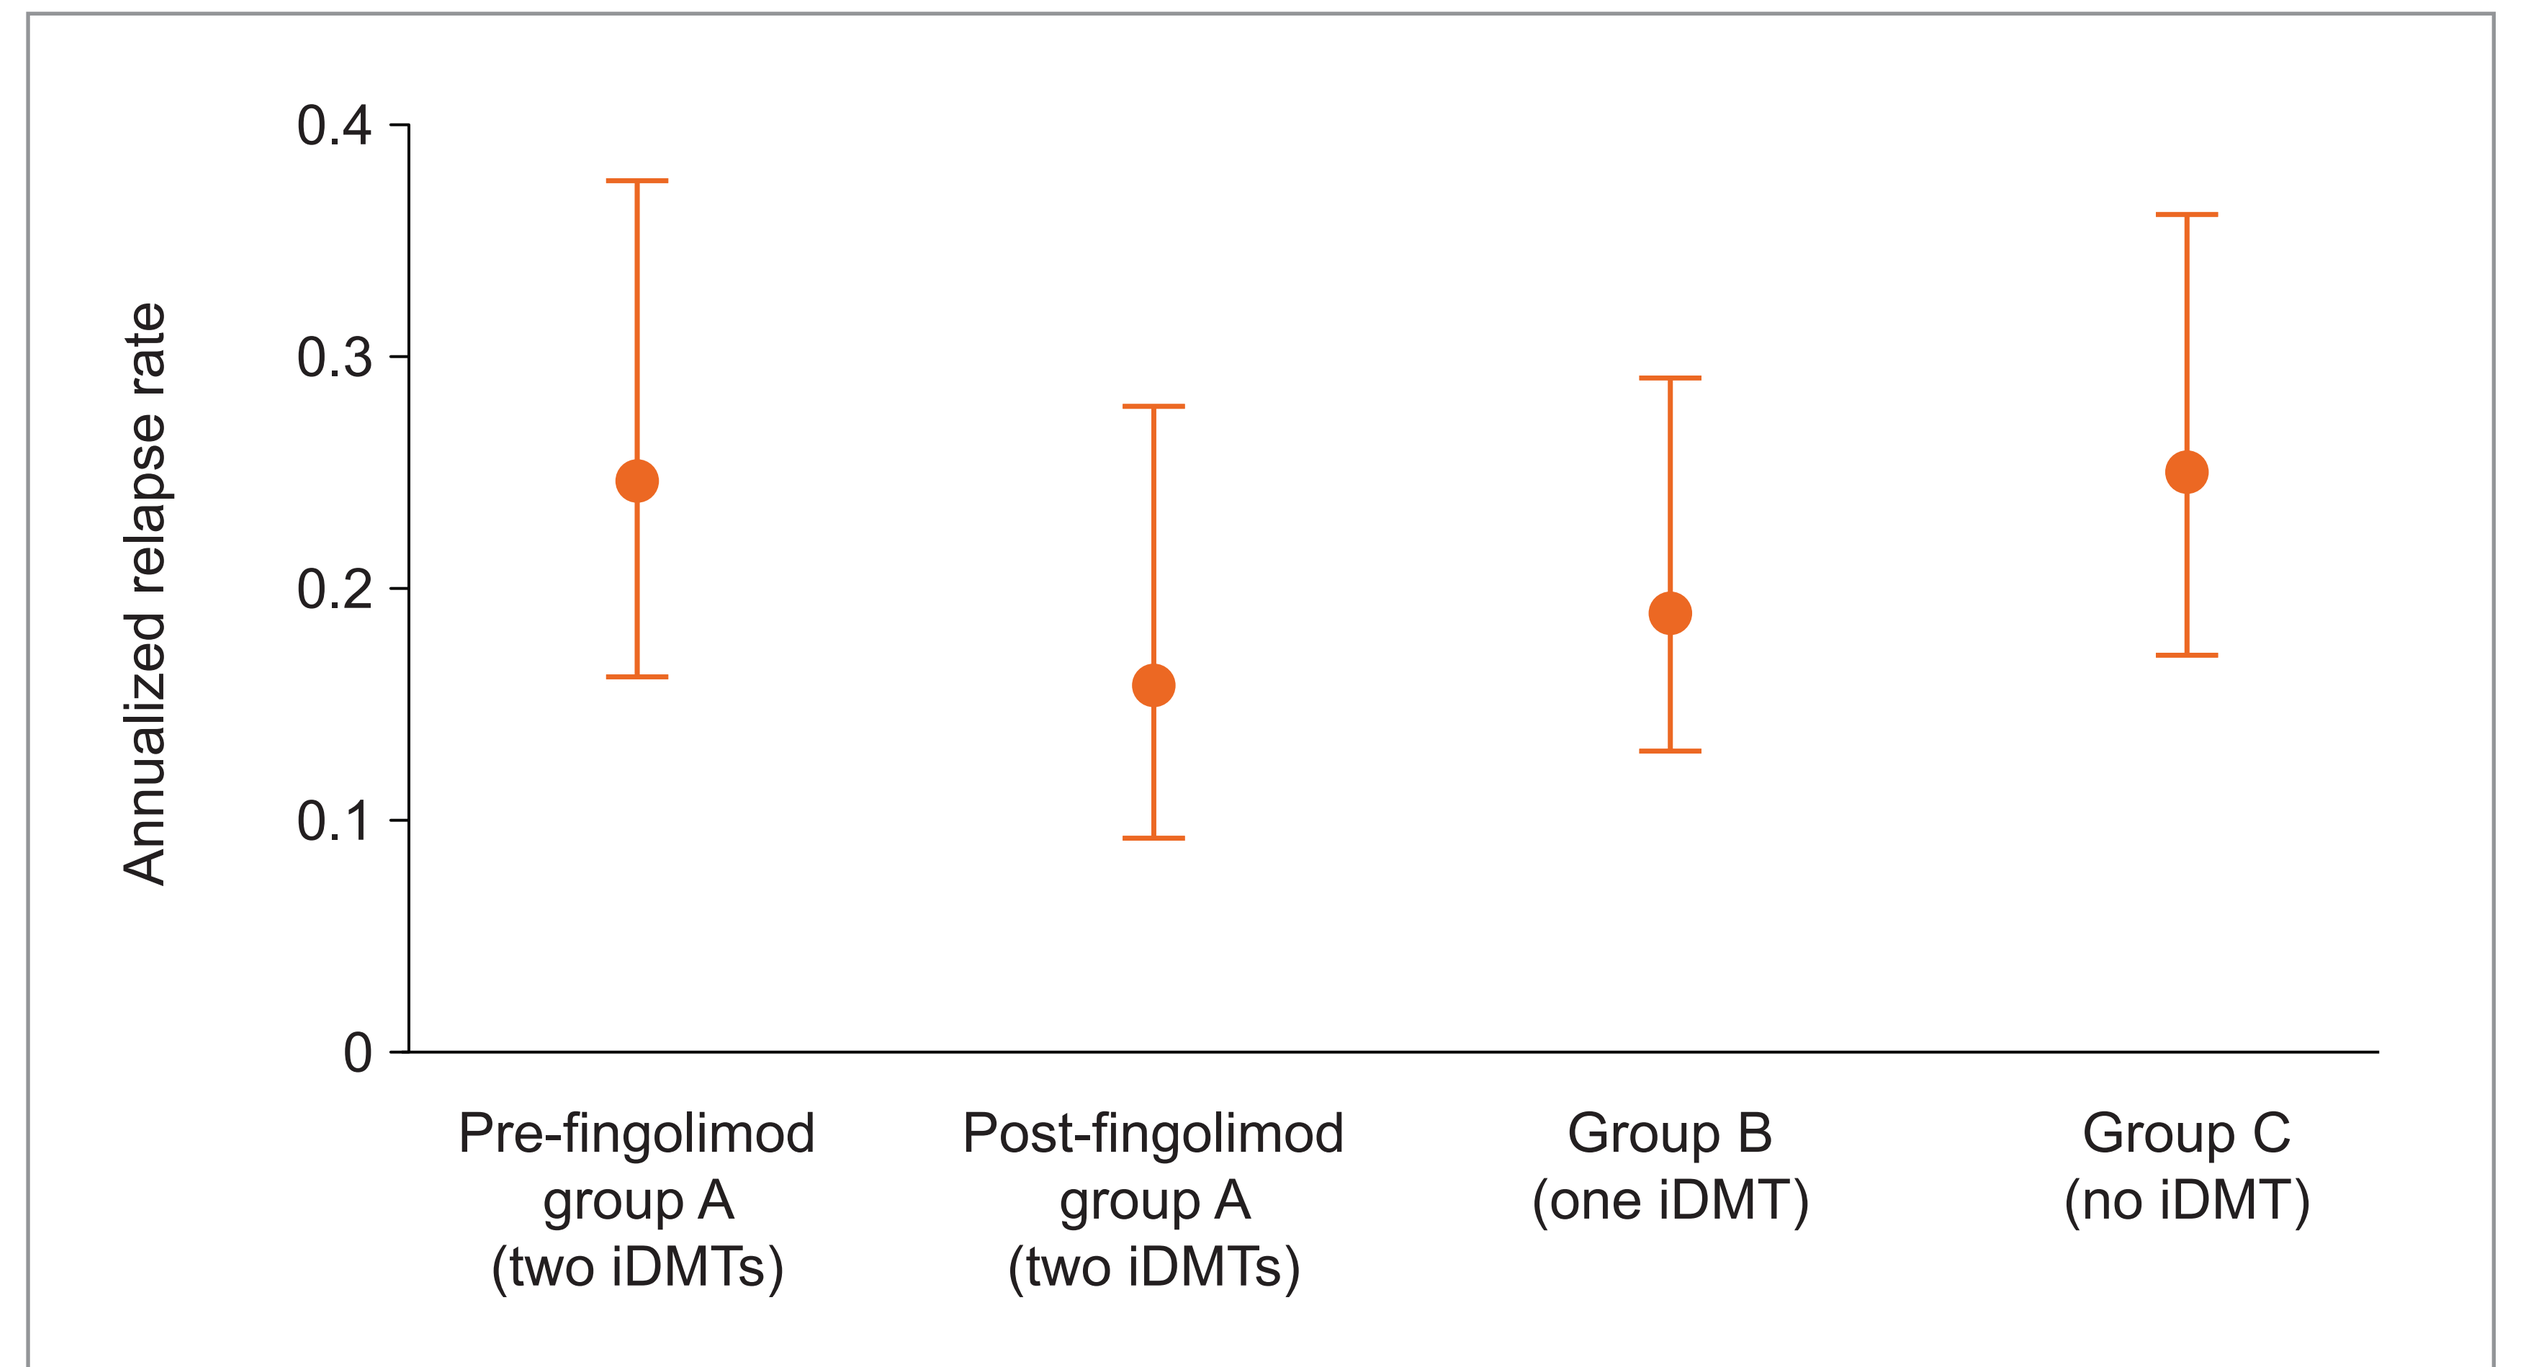
Annualized relapse rate
0.4
0.3
0.2
0.1
0
Pre-fingolimod group A (two iDMTs)
Post-fingolimod group A (two iDMTs)
Group B (one iDMT)
Group C (no iDMT)

Full analysis set. Data are mean (95% confidence interval)
ARR, annualized relapse rate; iDMT, injectable disease-modifying therapy

Conclusions

- Outcomes were favorable in patients with two previous iDMTs, following fingolimod treatment
 - Similar data were obtained for patients with one or no previous iDMT
- These data suggest that benefits may be seen with fingolimod irrespective of number of previous iDMTs
- Understanding the impact of previous treatments could help inform treatment decision-making in early stages of MS

References

1. Novartis Pharmaceuticals Corporation. Prescribing information. Gilenya® 2016. Available from: https://www.pharma.us.novartis.com/product/pi/pdf/gilenya.pdf (Accessed March 21, 2018).
2. Rae-Grant A, *et al. Neurology* 2018;90:777-788.
3. Cree BAC, *et al. Ther Adv Neurol Disord* 2018;in press.
4. Hunter SF, *et al. ECTRIMS Online Library* 2017;200820.
5. Hunter SF, *et al. ECTRIMS Online Library* 2017;199683.

Acknowledgments

Editorial support was provided by Oxford PharmaGenesis, Oxford, UK, which was funded by Novartis Pharmaceuticals Corporation. The final responsibility for the content lies with the authors

Disclosures

Samuel F Hunter has received consulting fees and/or research support from, and/or served on speakers' bureaux for Acorda Therapeutics, Avanir, Bayer HealthCare Pharmaceuticals, Novartis, Osmotica, Roche/Genentech, Sanofi Genzyme and Teva Neuroscience. **Florian P Thomas** has served as a speaker and/or consultant for Acorda Therapeutics, Genzyme and Teva Neuroscience. **Xiangyi Meng**, **Lesley Schofield**, **Scott Kolodny** and **Nadia Tenenbaum** are employees of, and received salary from, Novartis Pharmaceuticals Corporation. **Bruce AC Cree** has received consulting fees from AbbVie, Biogen, EMD Serono, GeNeuro, Novartis and Sanofi Genzyme

© 2018 Novartis Pharmaceuticals Corporation

Poster presented at the 2018 Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC), May 30-June 2, Nashville, TN, USA

This study was sponsored by Novartis Pharmaceuticals Corporation, USA

DOWNLOAD THIS POSTER AND VIEW VIDEO OF PREFER*MS* PRIMARY DATA AND OF PREFER*MS* TREAMENT SWITCH DATA

Scan QR code to download this poster and view videos of PREFERMS primary data.
Also available at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=ad0a5

And via Text Message (SMS)
Text: **Qad0a5**
To: **8NOVA (86682)** US only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe and Russia
+46737494608 Sweden and Europe

Scan to view a video presentation

Copies of this poster obtained through the QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address:
sfhunter@neurosci.us