A post hoc analysis of immunoglobulin levels in patients with relapsing multiple sclerosis treated with ozanimod in phase 3 trials

Sarah Harris,[1] Harry Southworth,[2] James K. Sheffield,[1] Rachel Maddux,[1] Jeffrey A. Cohen[3]

[1]Bristol Myers Squibb, Princeton, New Jersey; [2]Data Clarity Consulting Ltd, Stockport, England; [3]Mellen Center for MS Treatment and Research, Cleveland Clinic, Cleveland, Ohio

DMT53

To request a copy of this poster:

Scan QR code via a barcode reader application

Introduction

- Ozanimod, a sphingosine 1-phosphate receptor 1 and 5 modulator, is approved in multiple countries for the treatment of adults with either relapsing forms of multiple sclerosis (RMS) or moderately to severely active ulcerative colitis[1,2]
- The mechanism of action of ozanimod in multiple sclerosis involves reduction of lymphocyte egress from secondary lymphoid organs,[3] which results in dose-dependent decreases in absolute lymphocyte count (ALC), specifically circulating $CD19^+$ B- and $CD3^+$ T-cells[4]
- Because B and T cells are required for immunoglobulin (Ig) production,[5] there is a need to study the effect of ozanimod on circulating Ig levels
- Objectives: To evaluate whether ozanimod treatment results in changes in Ig levels (IgA, IgG, and IgM) and to assess relationships between Ig changes and pharmacodynamic, clinical, and imaging outcomes in patients with RMS

Methods

Study design

- Exploratory, post hoc analysis of the phase 3 RMS trials SUNBEAM[6] (NCT02294058; ≥ 12 months) and RADIANCE[7] (NCT02047734; 24 months), which randomized patients to daily ozanimod 0.46 or 0.92 mg or weekly intramuscular interferon (IFN) ß-1a 30 µg

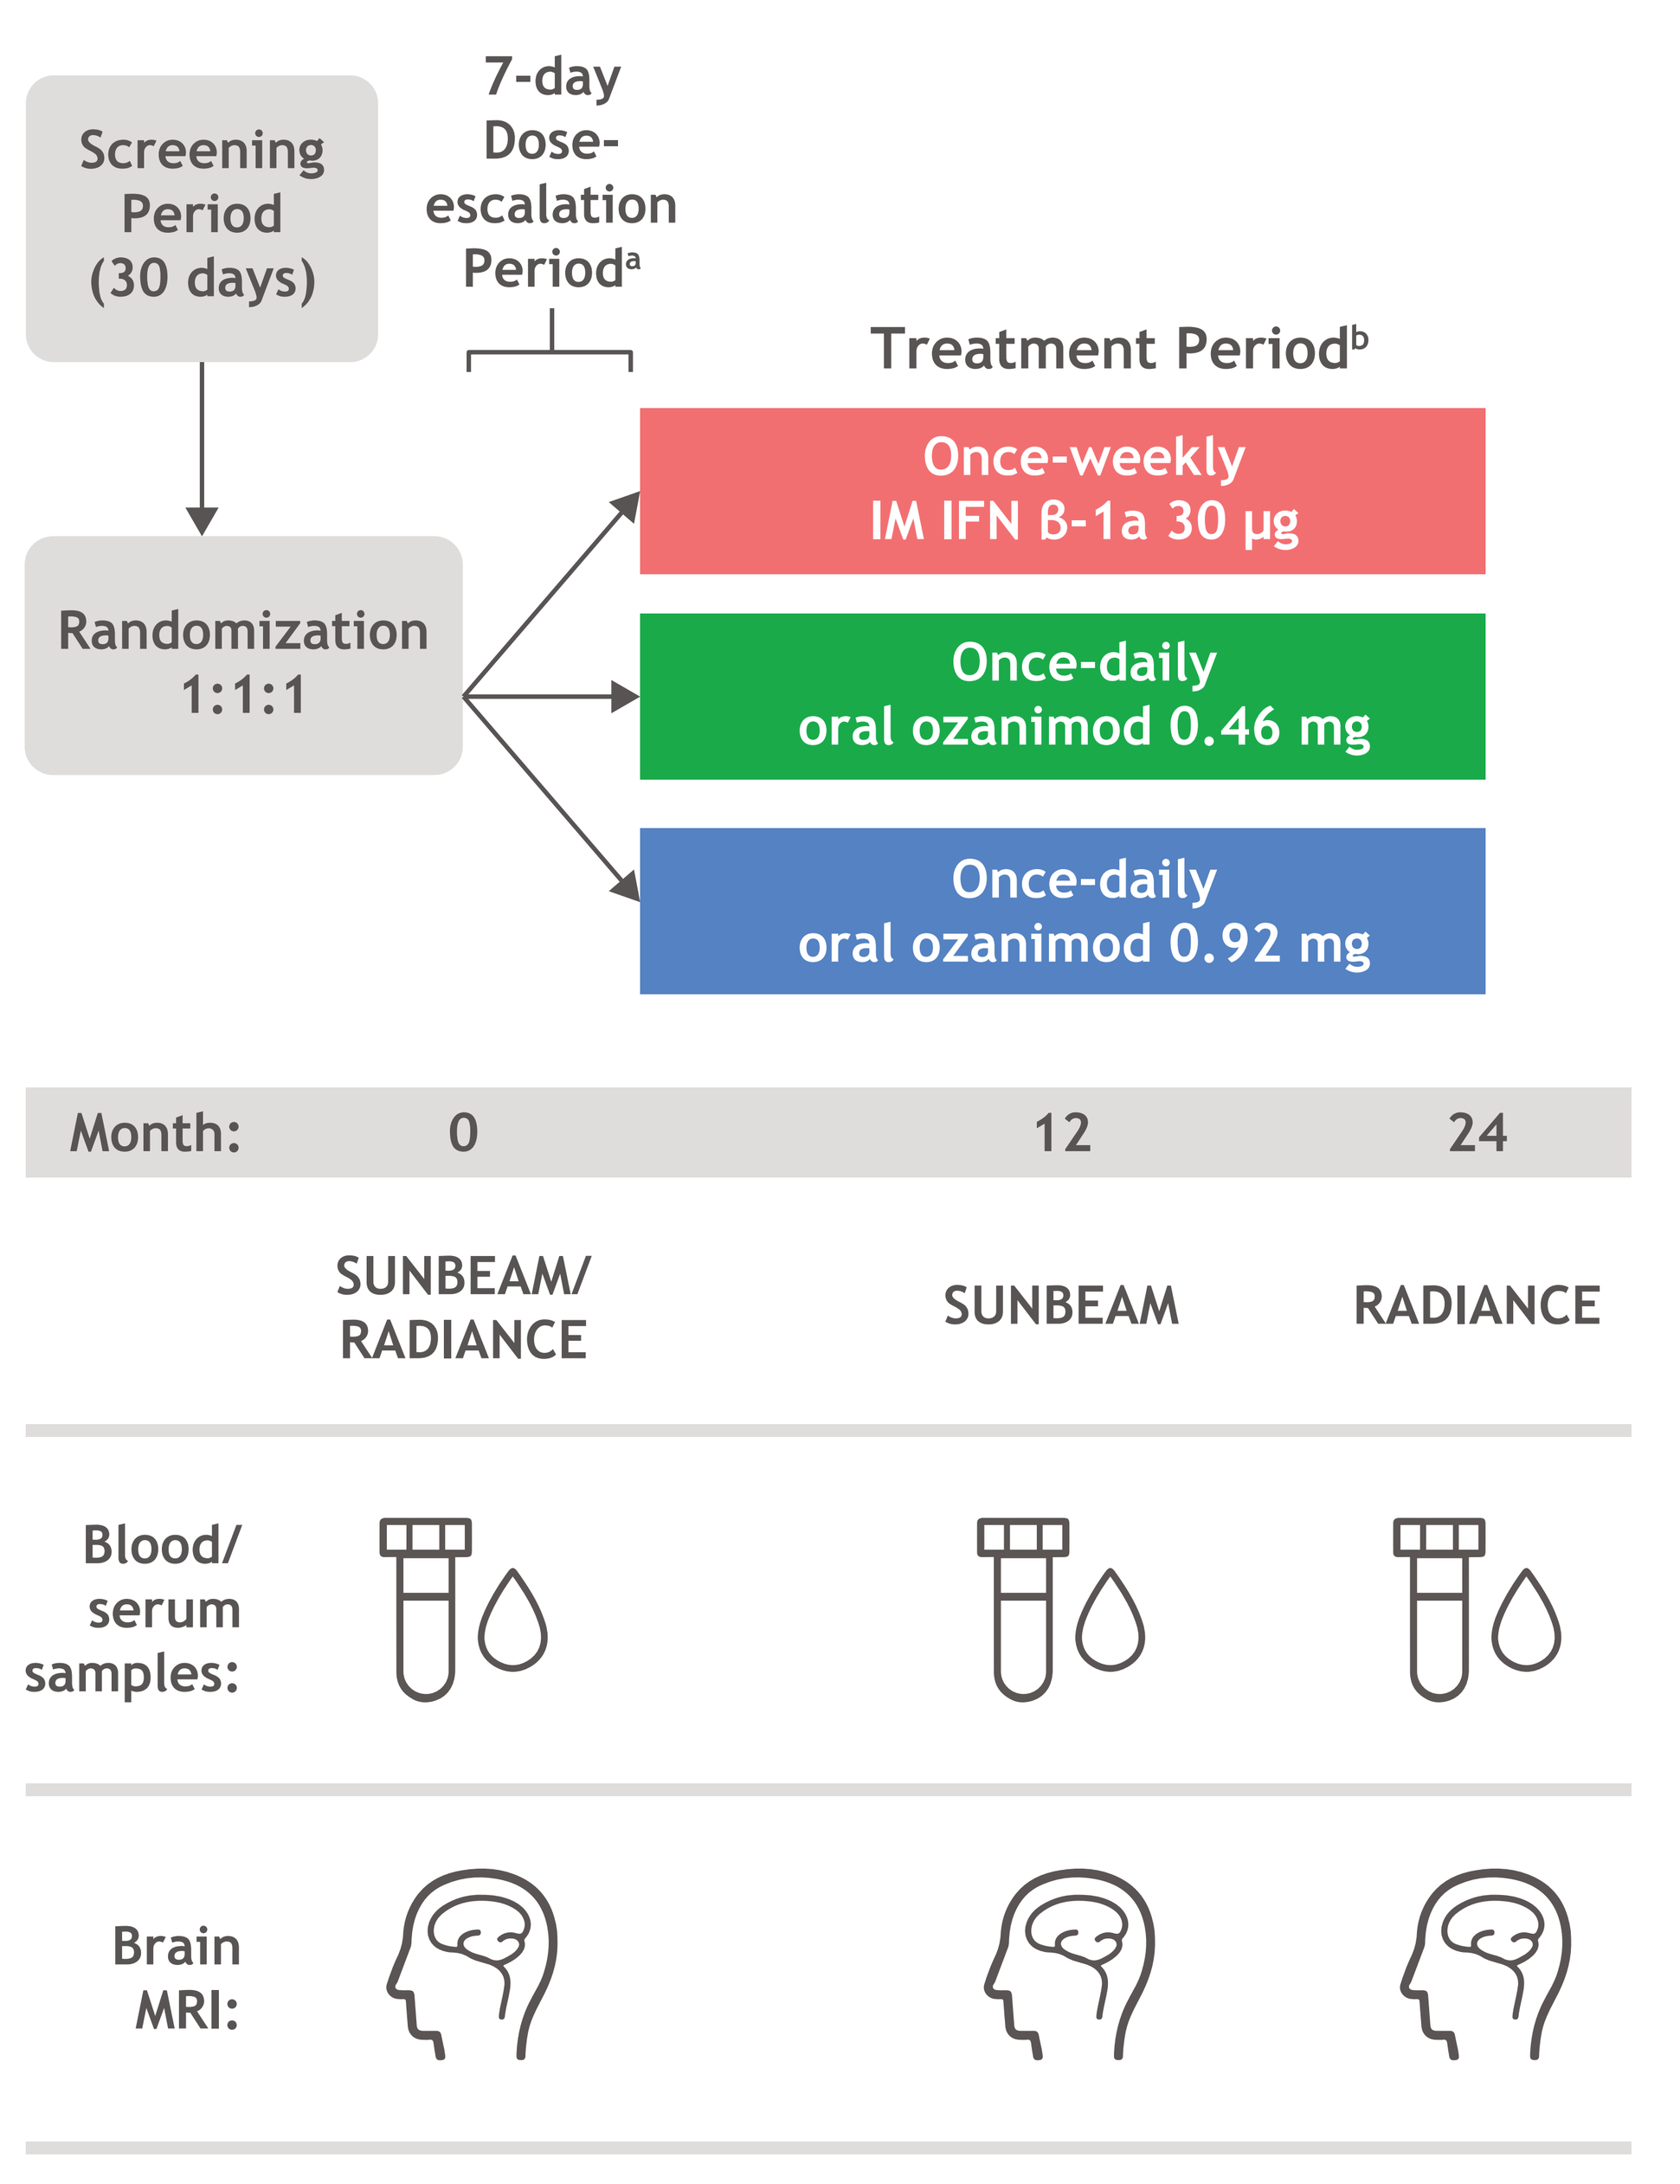

Screening Period (30 days)
7-day Dose-escalation Period[a]
Treatment Period[b]
Randomization 1:1:1
Once-weekly IM IFN ß-1a 30 µg
Once-daily oral ozanimod 0.46 mg
Once-daily oral ozanimod 0.92 mg
Month: 0 12 24
SUNBEAM/ RADIANCE
SUNBEAM
RADIANCE
Blood/ serum samples:
Brain MRI:

[a]Patients received ozanimod 0.23 mg/d on days 1-4, ozanimod 0.46 mg/d on days 5-7, and the assigned dose of ozanimod 0.46 or 0.92 mg/d starting on day 8 of ozanimod treatment. [b]SUNBEAM ended when the last patient enrolled completed 12 months, and RADIANCE had a duration of 24 months. HCl, hydrochloride; IM, intramuscular.

Assessments

- Serum IgA, IgG, and IgM levels and blood ALC were quantified from samples obtained at baseline and month 12 (SUNBEAM)[6] or month 24 (RADIANCE)[7]
- Annualized relapse rate (ARR, the primary endpoint of both studies) was calculated from confirmed, protocol-defined relapses
- Number of gadolinium-enhancing (GdE) lesions (a key secondary endpoint of both studies) was assessed from MRI

Statistical analysis

- Relationships between median proportion of baseline Ig levels at month 12 or month 24 and median proportion of baseline ALC, expected ARR, and expected mean number of GdE lesions were analyzed from 1000 bootstrap samples
- Treatment groups were compared using robust linear models

Results

- Ozanimod was associated with dose-dependent median reductions from baseline in Ig levels and ALC at months 12 and 24
- IFN ß-1a was associated with small median increases from baseline in IgA and IgG and a small median decrease in ALC

Median proportion of baseline ALC and Igs at 12 and 24 months of treatment[a]

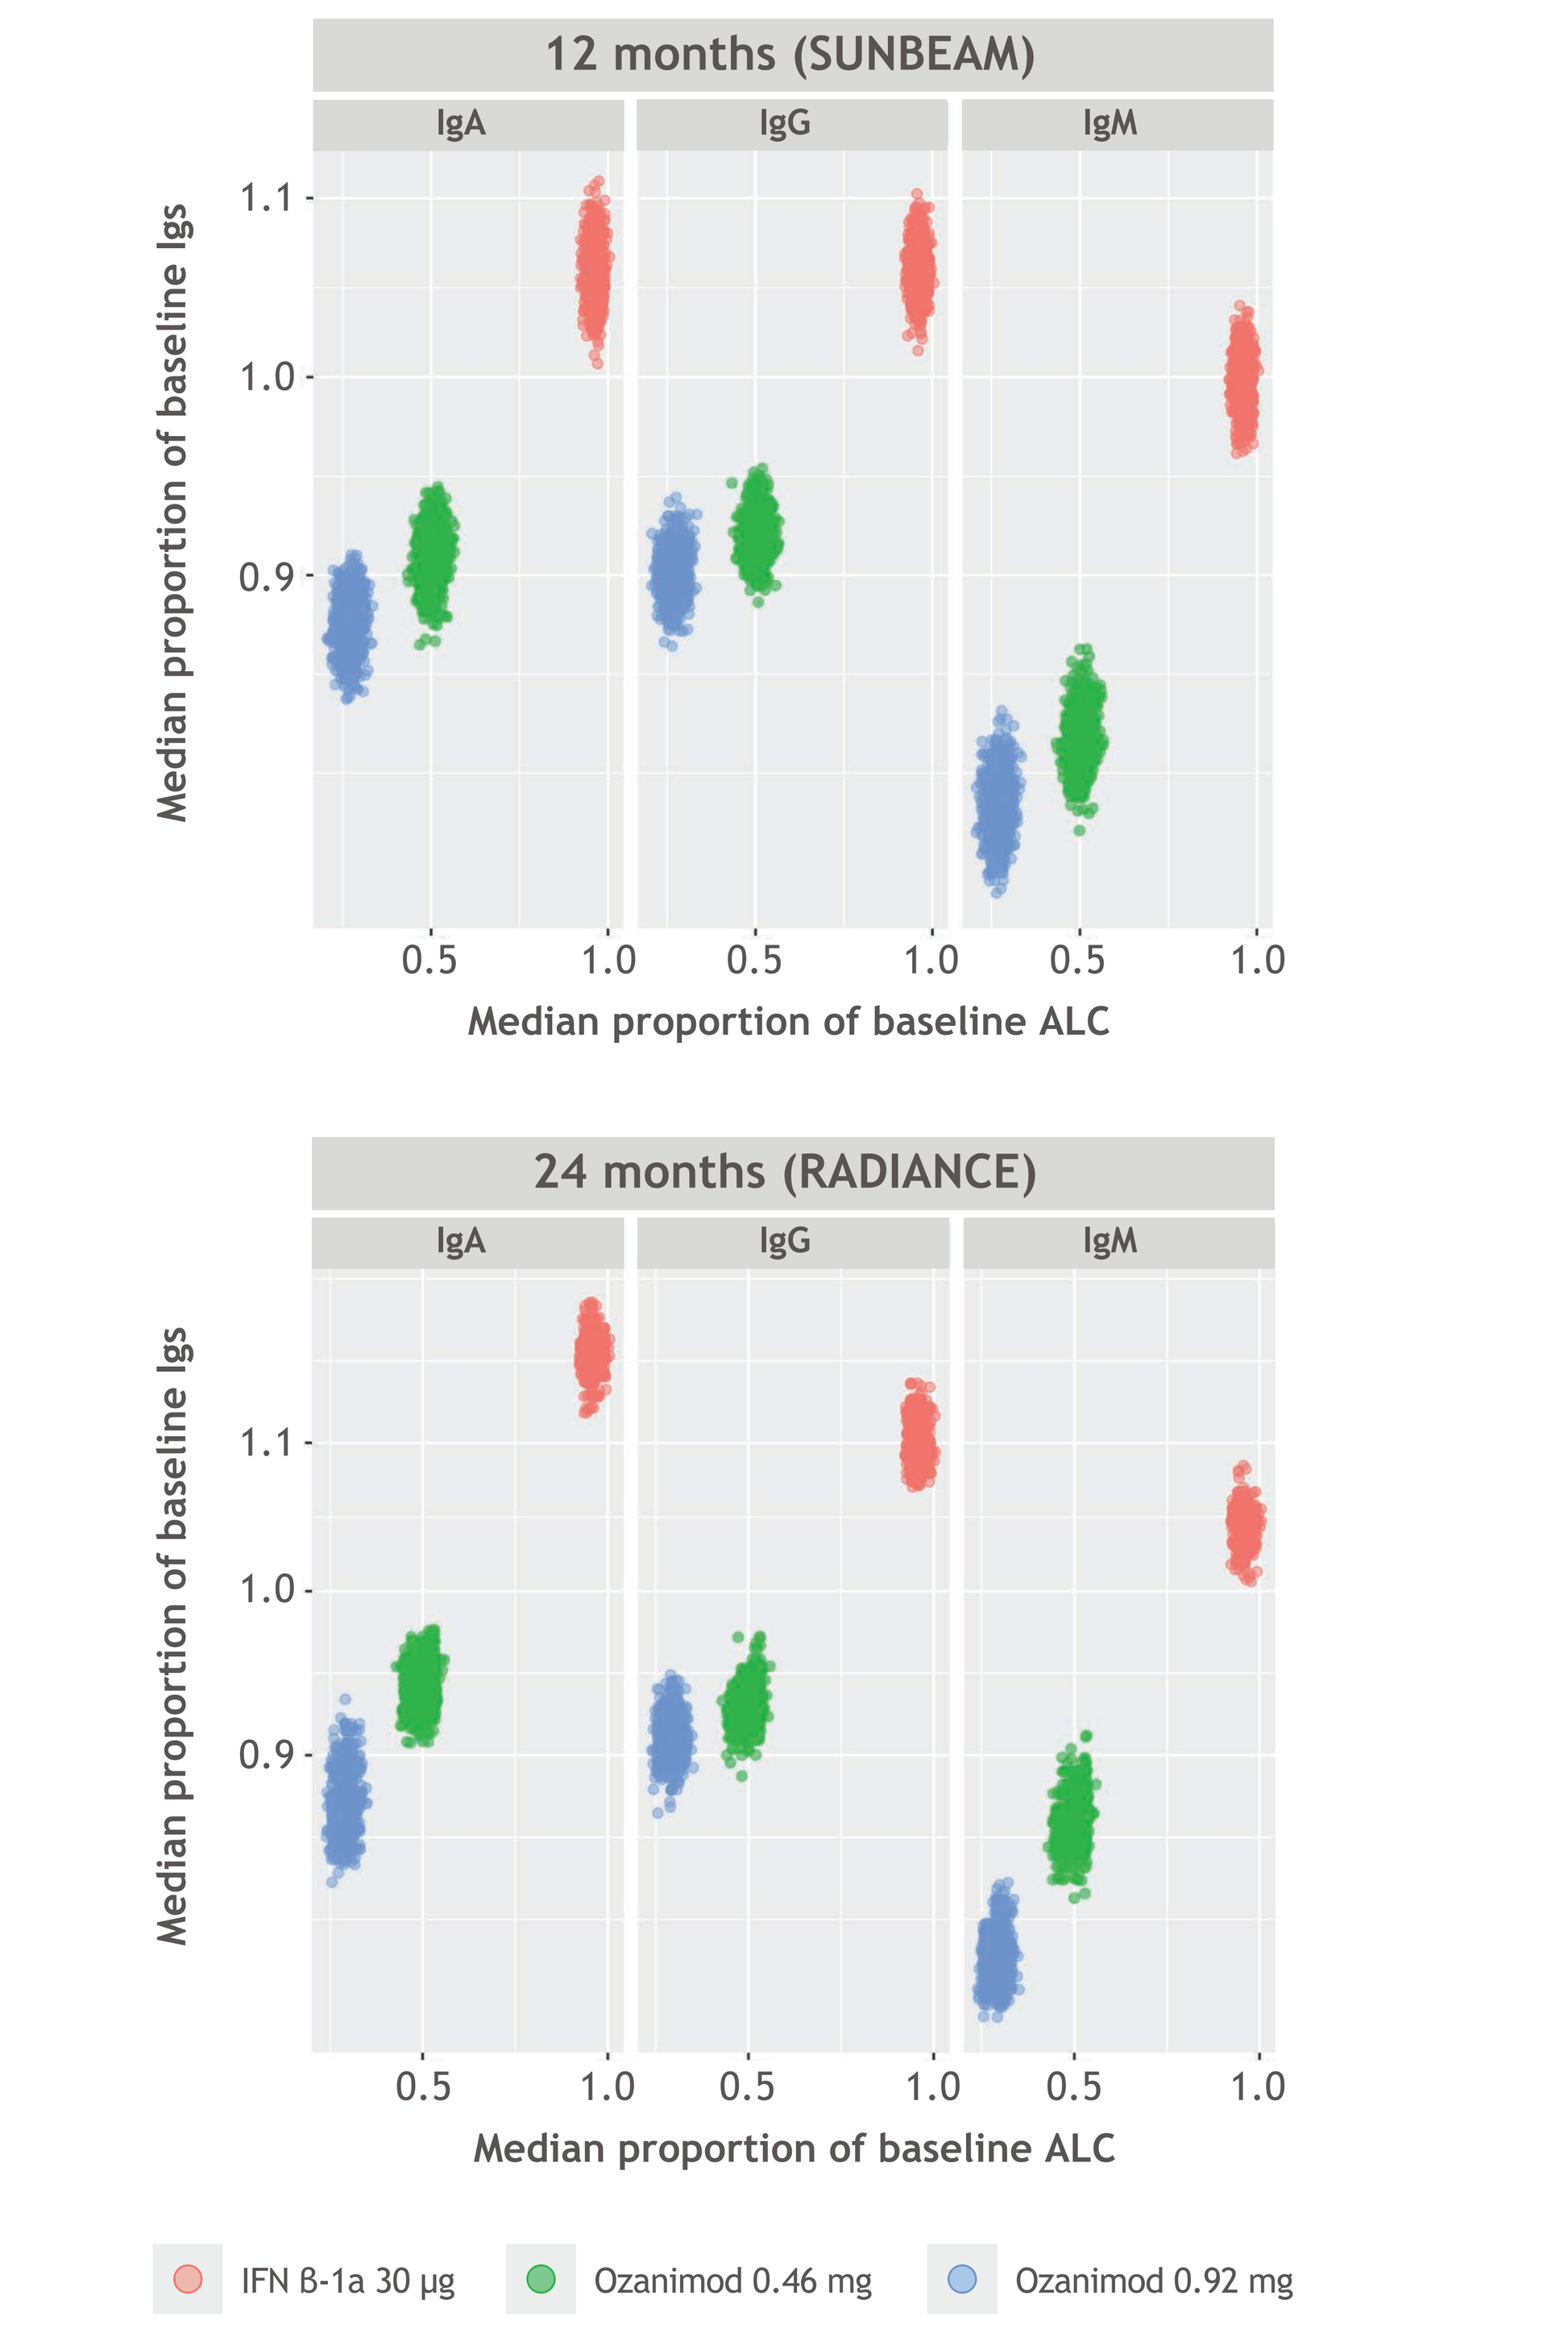

12 months (SUNBEAM)
24 months (RADIANCE)
IgA
IgG
IgM
Median proportion of baseline Igs
Median proportion of baseline ALC
IFN ß-1a 30 µg
Ozanimod 0.46 mg
Ozanimod 0.92 mg

[a]Relationship between median proportion of baseline ALC and Ig based on bootstrap sampling. Individual dots on each plot represent the individual simulations from the bootstrap procedure.

- In both trials, greater median reductions in IgA, IgG, or IgM were associated with lower expected ARRs and expected mean number of GdE lesions
 - The greatest median reduction from baseline in Ig levels was observed in the ozanimod 0.92 mg group, which also had the lowest expected ARR and expected mean number of GdE lesions

Median observed Ig values remained within the normal range for all treatment groups (for adults: IgA, 0.8-3.0 g/L; IgG, 6.0-16.0 g/L; IgM, 0.4-2.5g/L[8])

Dose-dependent decreases from baseline, and relative to IFN ß-1a, were observed with ozanimod for all Igs; the treatment effects of ozanimod 0.46 and 0.92 mg on all Igs were significantly different from IFN ß-1a at 12 and 24 months (all $P < 0.0001$)

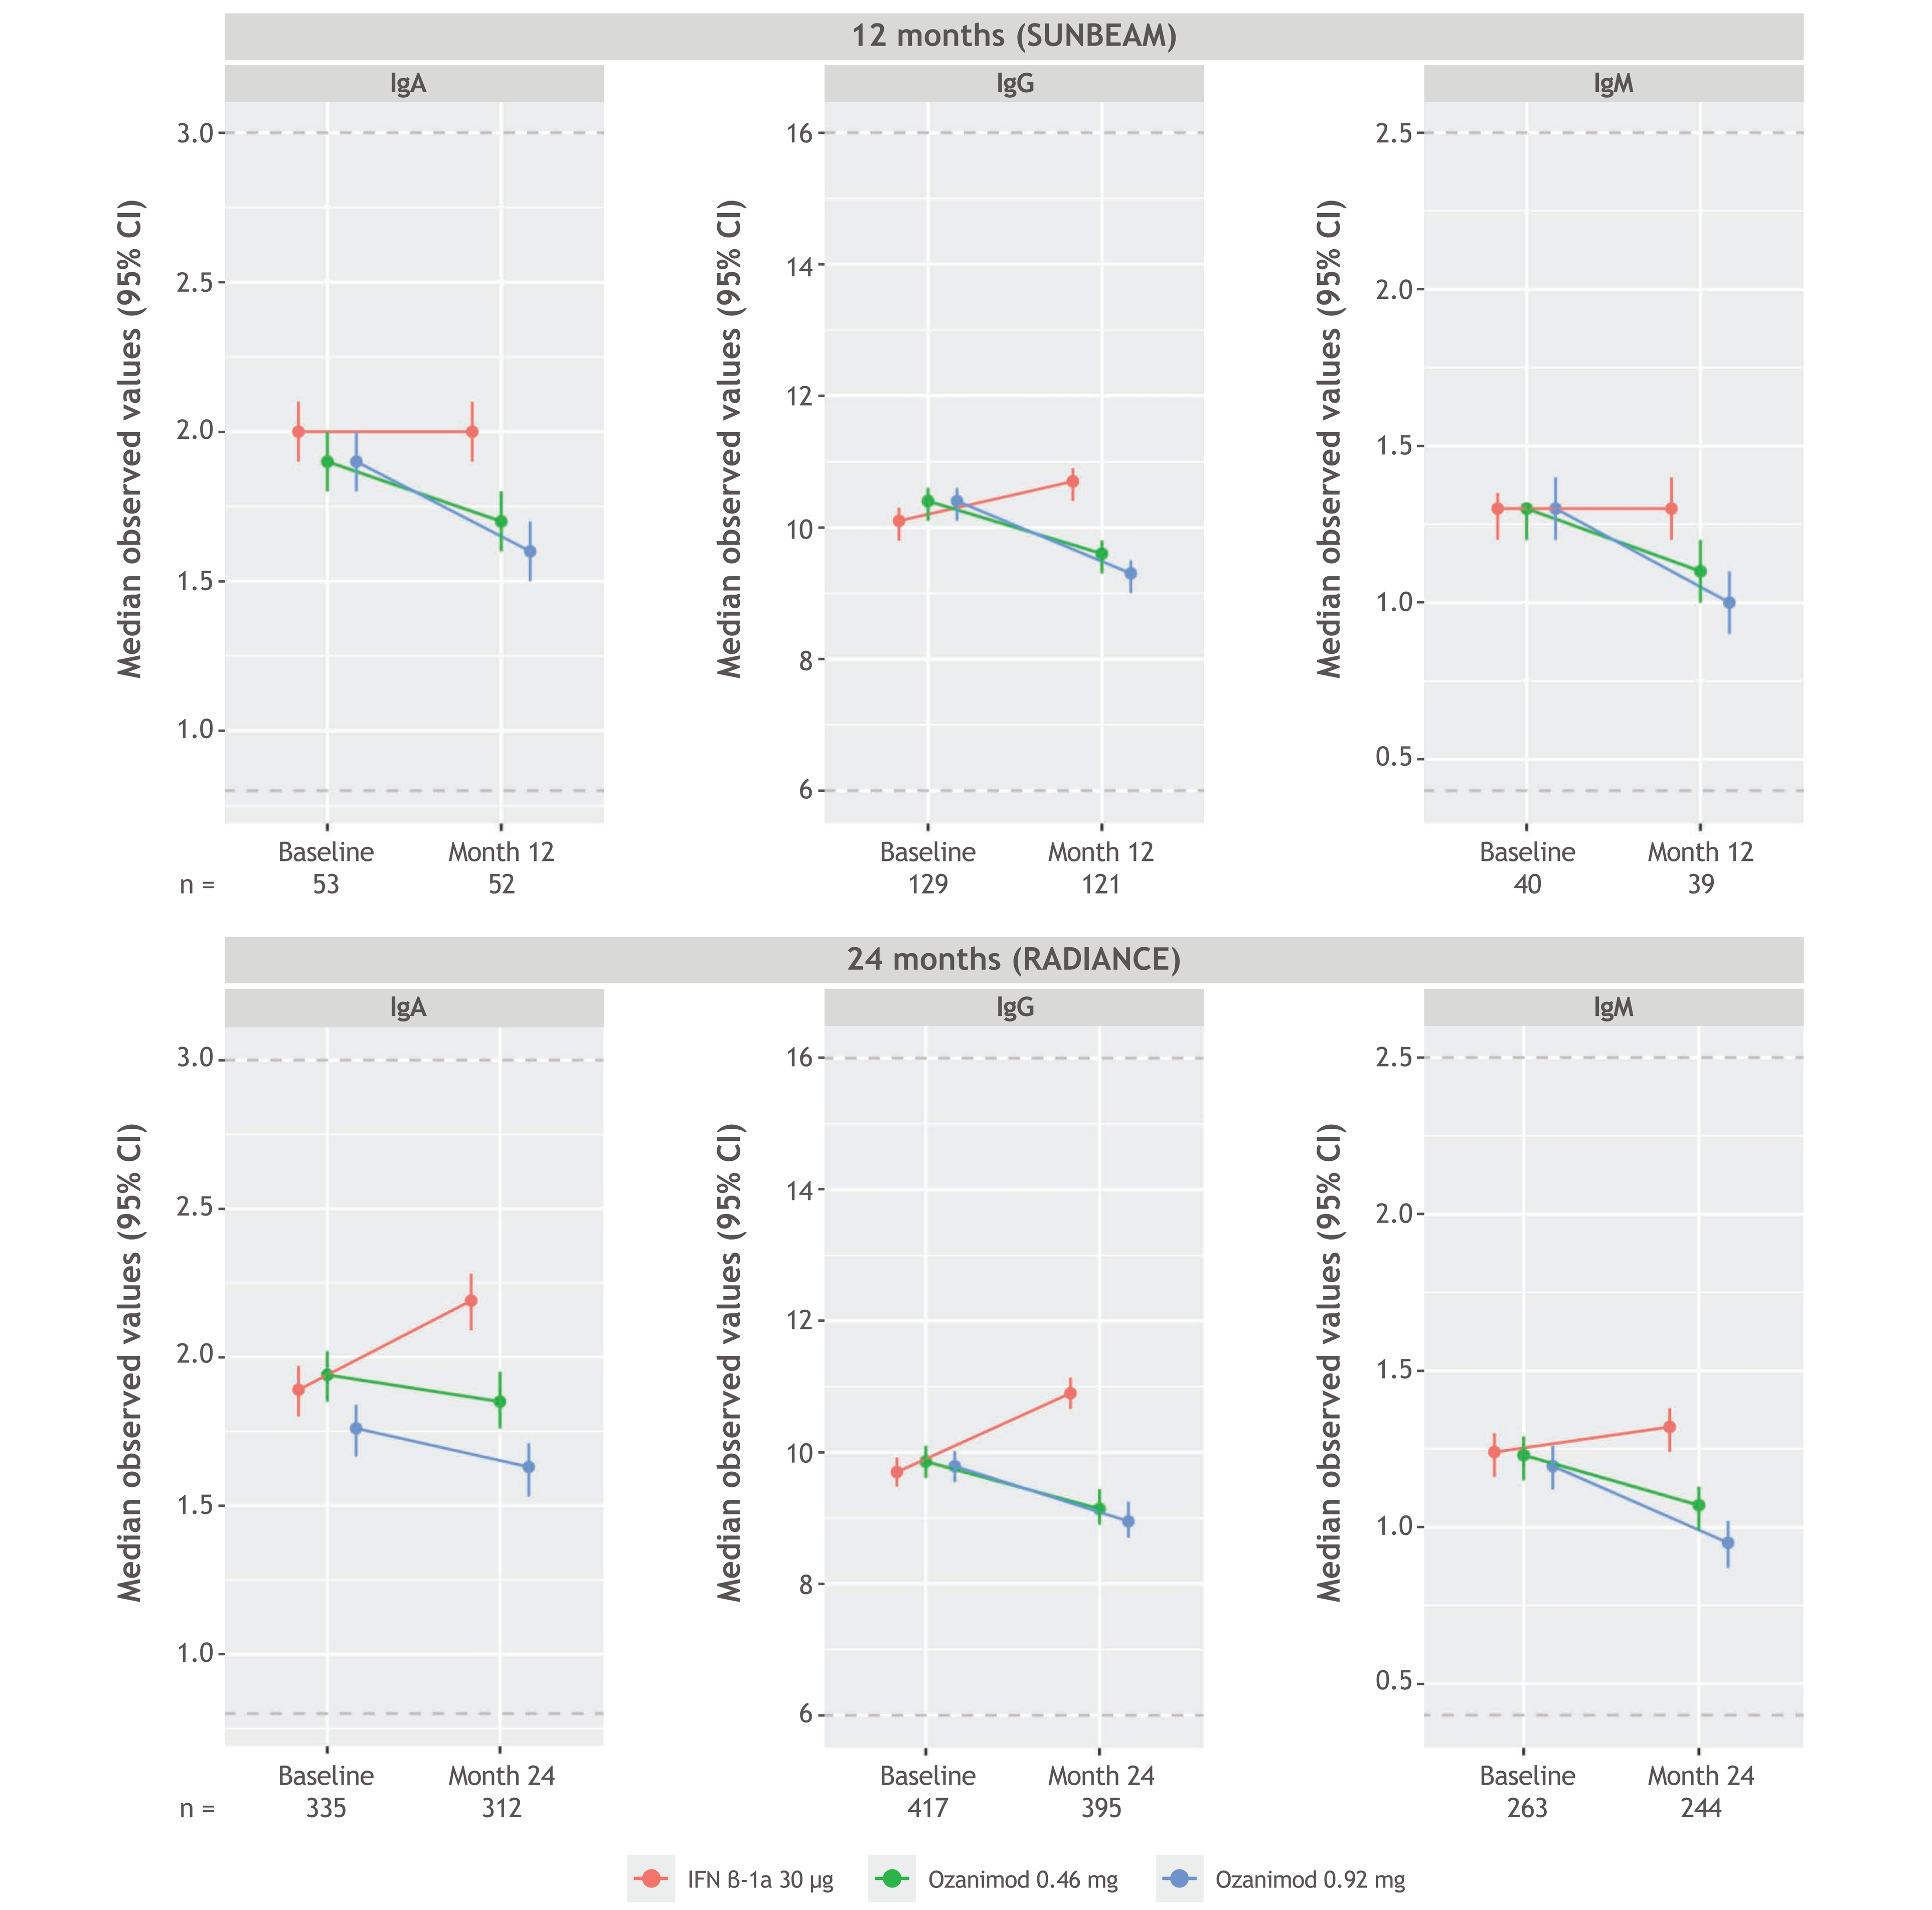

12 months (SUNBEAM)
IgA
IgG
IgM
Median observed values (95% CI)
Baseline
Month 12
n =
53
52
129
121
40
39
24 months (RADIANCE)
Month 24
335
312
417
395
263
244
IFN ß-1a 30 µg
Ozanimod 0.46 mg
Ozanimod 0.92 mg

The horizontal location is shifted slightly between groups to avoid overplotting.
The dotted lines represent the normal Ig range for adults.

Relationship between expected ARR and median proportion of baseline IgA (A), IgG (B), and IgM (C)[a]

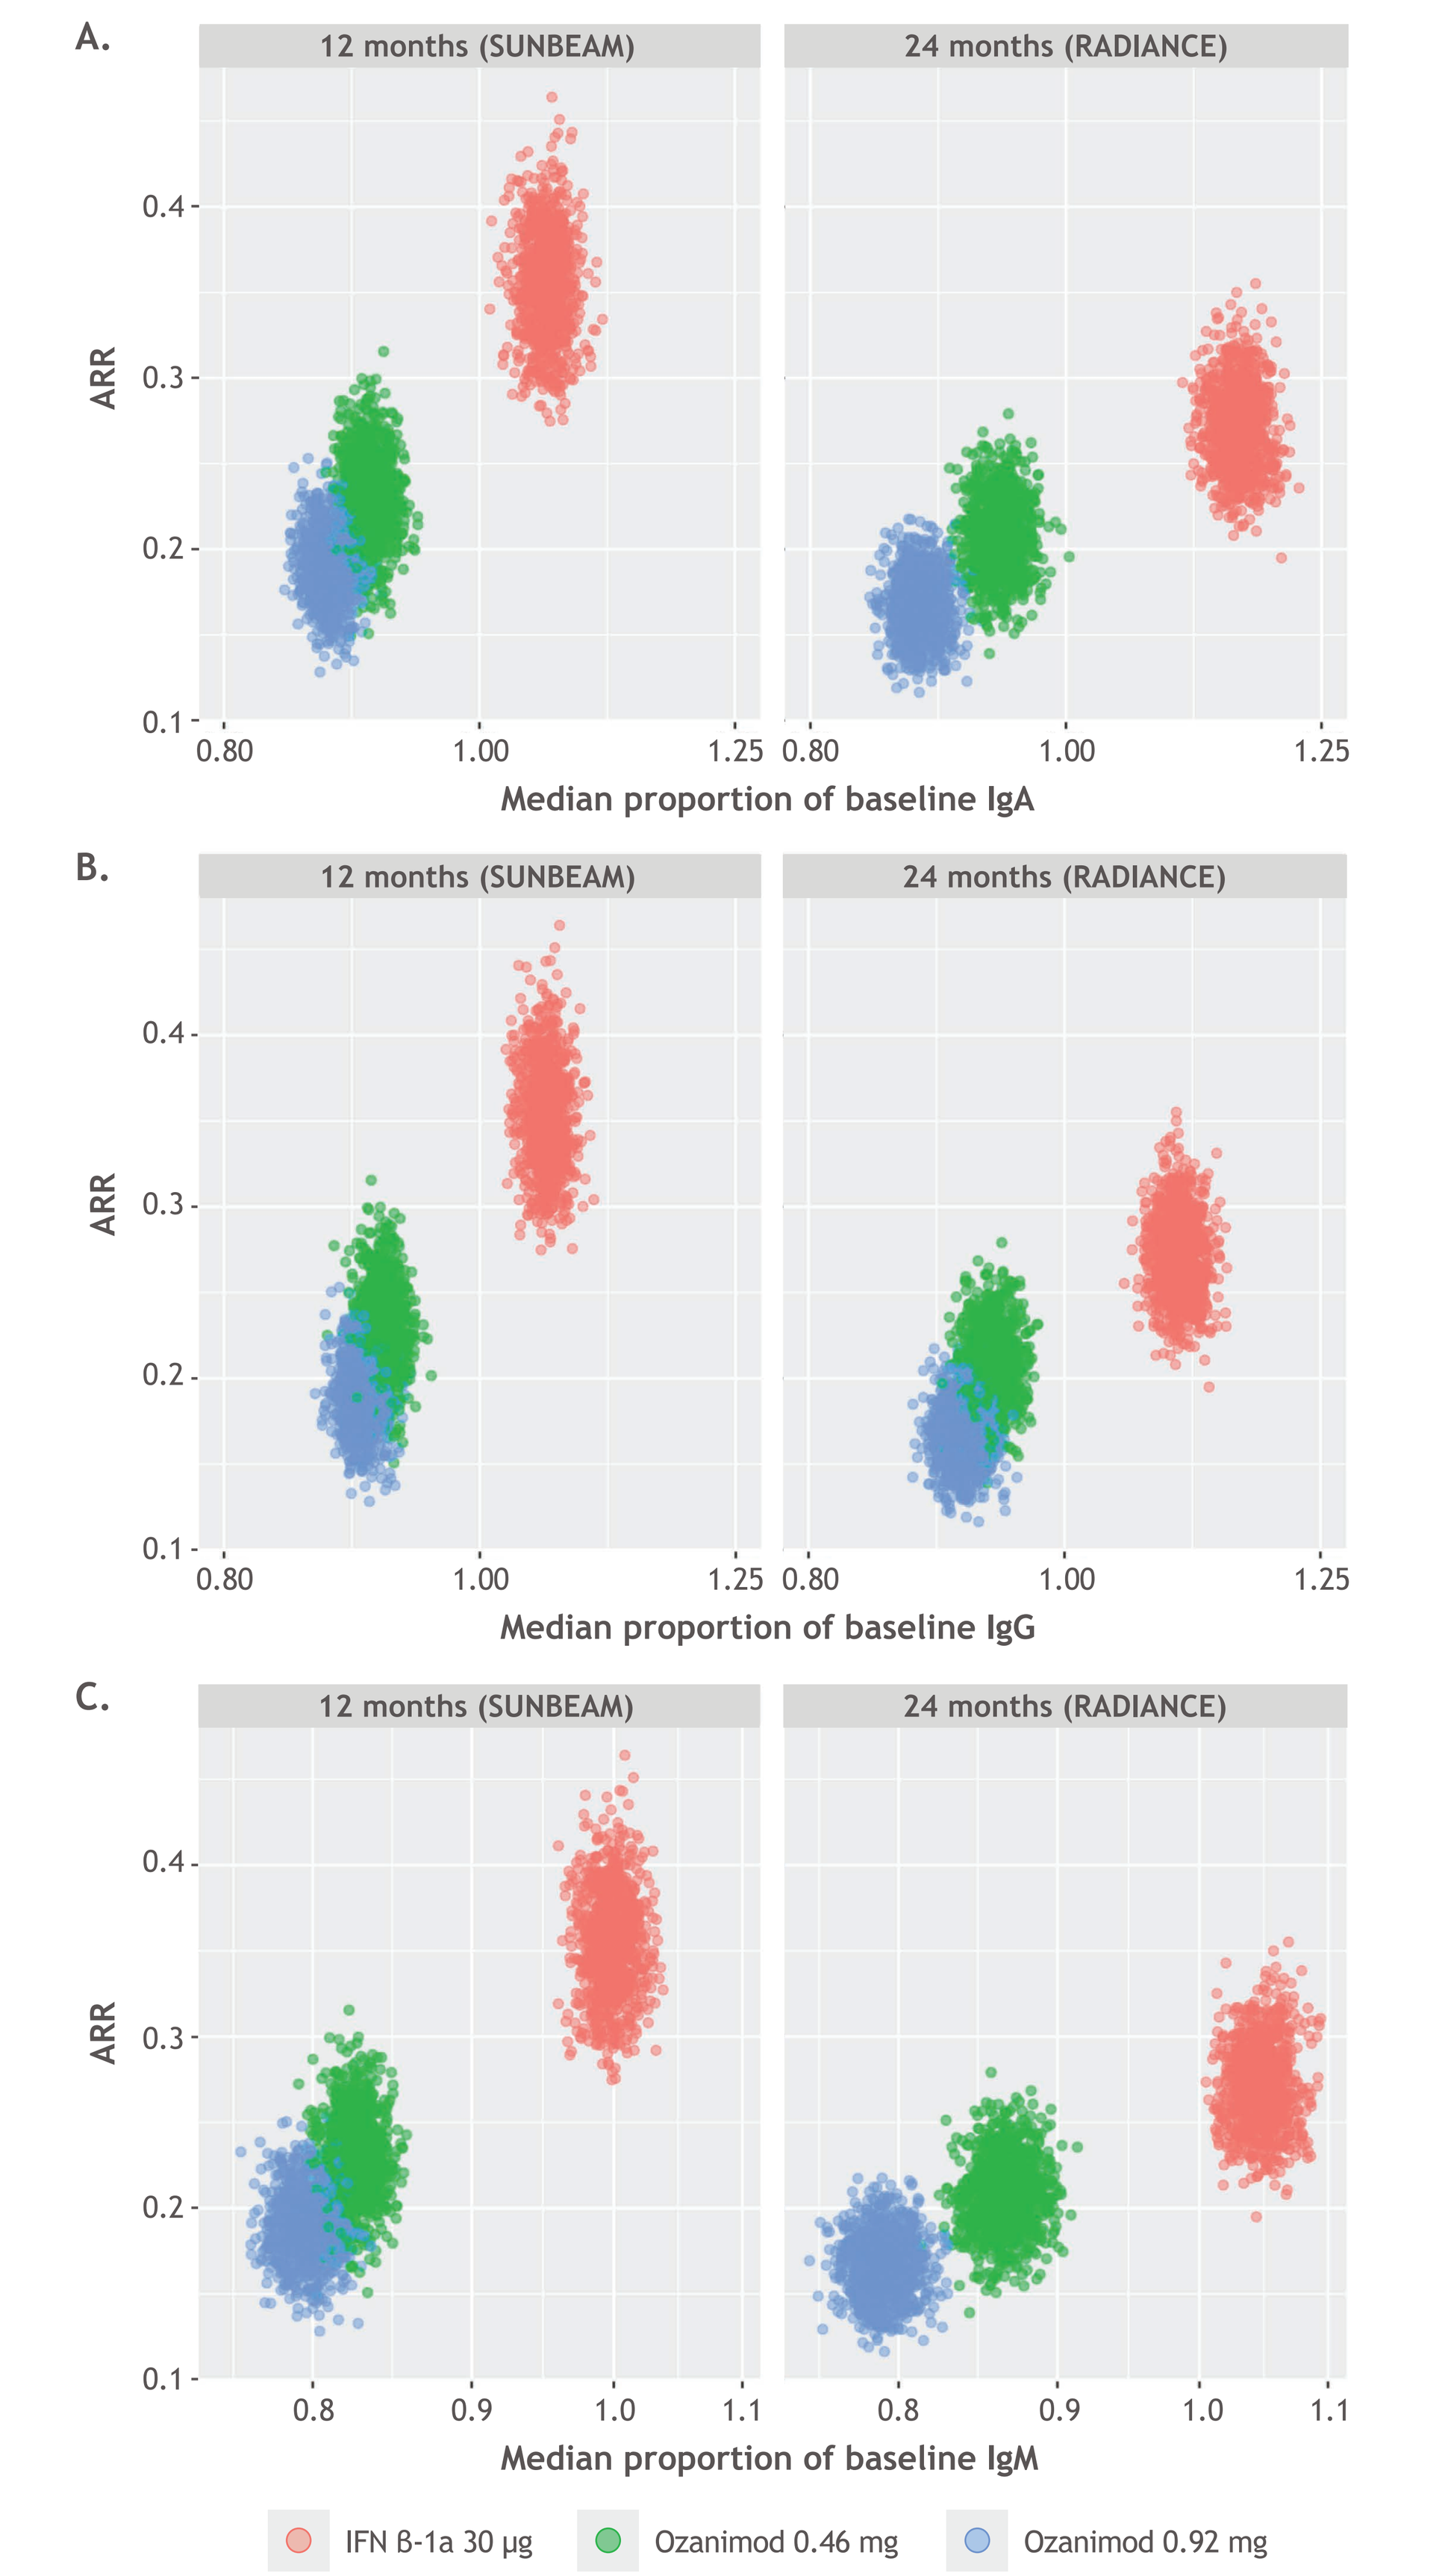

A.
B.
C.
12 months (SUNBEAM)
24 months (RADIANCE)
ARR
Median proportion of baseline IgA
Median proportion of baseline IgG
Median proportion of baseline IgM
IFN ß-1a 30 µg
Ozanimod 0.46 mg
Ozanimod 0.92 mg

[a]Relationship between ARR and median proportion of baseline Ig based on bootstrap sampling. Individual dots on each plot represent the individual simulations from the bootstrap procedure.

Relationship between expected mean number of GdE lesions and median proportion of baseline IgA (A), IgG (B), and IgM (C)[a]

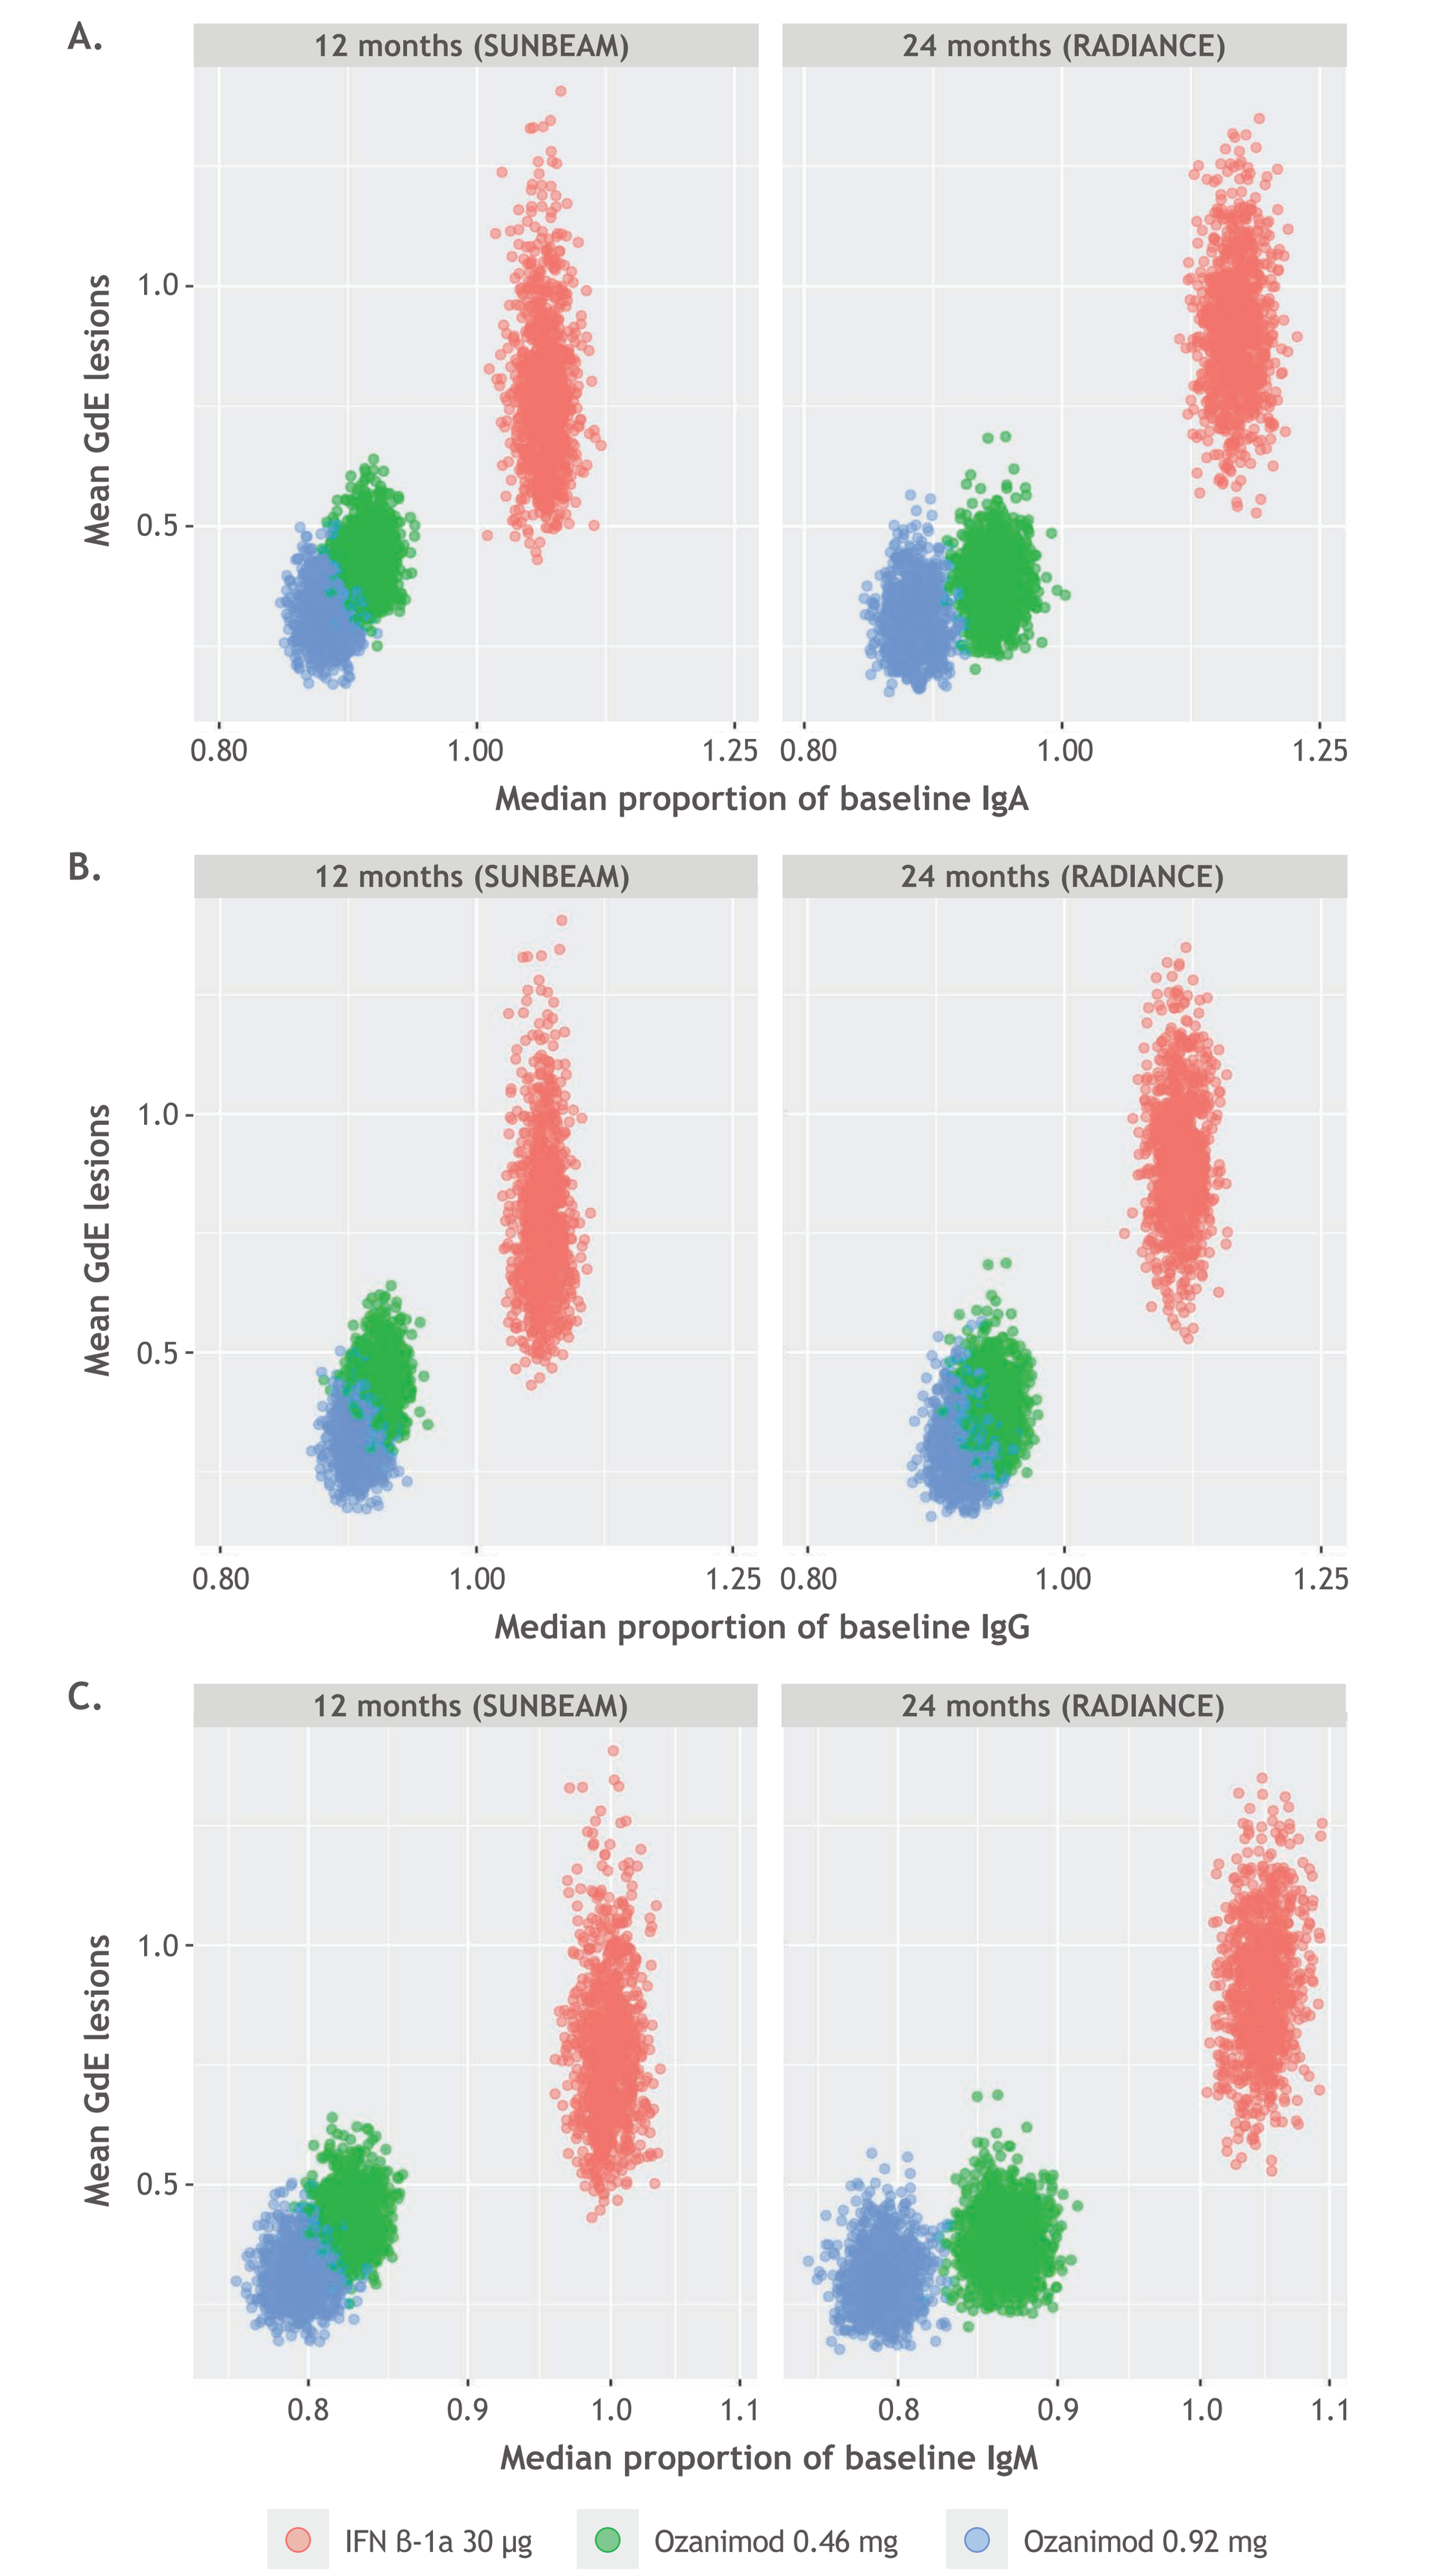

A.
B.
C.
12 months (SUNBEAM)
24 months (RADIANCE)
Mean GdE lesions
Median proportion of baseline IgA
Median proportion of baseline IgG
Median proportion of baseline IgM
IFN ß-1a 30 µg
Ozanimod 0.46 mg
Ozanimod 0.92 mg

[a]Relationship between GdE lesions and median proportion of baseline Ig based on bootstrap sampling. Individual dots on each plot represent the individual simulations from the bootstrap procedure.

Conclusions

- In phase 3 trials, ozanimod treatment resulted in dose-dependent decreases in median circulating IgA, IgG, and IgM levels in patients with RMS; however, median Ig levels were maintained within normal ranges
- Greater reductions in Ig levels were associated with lower expected ARR and fewer expected GdE lesions
- These findings suggest a potential relationship between the pharmacodynamic effect of ozanimod and relapses and MRI findings of active disease

References

1. ZEPOSIA® (ozanimod) [package insert]. Princeton, NJ, USA: Bristol Myers Squibb; December 2021. **2.** ZEPOSIA® (ozanimod) [summary of product characteristics]. Utrecht, Netherlands: Celgene Distribution BV; December 2021. **3.** Scott FL, et al. *Br J Pharmacol* 2016;173:1778-1792. **4.** Harris S, et al. *Neurol Neurimmunol Neurinflamm* 2020;7:e839. **5.** Johnson AB, et al. *Molecular Biology of the Cell*. 4th ed. New York: Garland Science; 2002. **6.** Cohen JA, et al. *Lancet Neurol* 2019;18:1021-1033. **7.** Comi G, et al. *Lancet Neurol* 2019;18:1009-1020. **8.** NHS, Oxford University Hospitals. Immunology Laboratory, Diagnostic Tests: Immunoglobulins. https://www.ouh.nhs.uk/immunology/diagnostic-tests/tests-catalogue/immunoglobulins.aspx. Accessed January 20, 2022.

Acknowledgments

- The patients and families who made these studies possible
- The clinical study teams who participated
- The SUNBEAM and RADIANCE studies were supported by Celgene International II
- All authors contributed to and approved the poster; writing and editorial assistance was provided by Emily C. Bruggeman, PhD, and Jessica D. Herr, PharmD, of Peloton Advantage, LLC, an OPEN Health company, and funded by Bristol Myers Squibb

Disclosures

- **SH, JKS:** Employees and shareholders of Bristol Myers Squibb
- **RM:** Employee of Bristol Myers Squibb
- **HS:** Compensation from Bristol Myers Squibb, F2G, GlaxoSmithKline, CellCentric, and Gossamer Bio
- **JAC:** Personal compensation for consulting for Biogen, Bristol Myers Squibb, Convelo, Genentech, Janssen, NervGen, Novartis, and PSI; speaking for H3 Communications; and serving as an Editor of *Multiple Sclerosis Journal*

Presented at the 2022 Annual Meeting of the Consortium of Multiple Sclerosis Centers; June 1-4, 2022; National Harbor, Maryland

Previously presented at ACTRIMS Forum 2022; February 24-26, 2022; West Palm Beach, Florida

Email: sarah.harris@bms.com

This poster may not be reproduced without written permission from the authors.