Long-term efficacy and safety, including COVID-19 infections, among ozanimod-treated patients with relapsing multiple sclerosis in the DAYBREAK open-label extension trial

DMT14

Bruce A. C. Cree,[1] Krzysztof W. Selmaj,[2] Lawrence Steinman,[3] Giancarlo Comi,[4] Amit Bar-Or,[5] Douglas L. Arnold,[6] Hans-Peter Hartung,[7] Xavier Montalbán,[8] Eva K. Havrdová,[9] James K. Sheffield,[10] Neil Minton,[10] Chun-Yen Cheng,[10] Diego Silva,[10] Ludwig Kappos,[11] Jeffrey A. Cohen[12]

[1]Weill Institute for Neurosciences, Department of Neurology, University of California San Francisco, San Francisco, California; [2]Center for Neurology, Łódź, Poland and Collegium Medicum, Department of Neurology, University of Warmia and Mazury, Olsztyn, Poland; [3]Department of Neurology and Neurological Sciences, Beckman Center for Molecular Medicine, Stanford University Medical Center, Stanford, California; [4]Vita-Salute San Raffaele University and Casa di Cura del Policlinico, Milan, Italy; [5]Center for Neuroinflammation and Experimental Therapeutics, and Department of Neurology, Perelman School of Medicine, University of Pennsylvania, Philadelphia, Pennsylvania; [6]NeuroRx Research and Montréal Neurological Institute, McGill University, Montréal, Quebec, Canada; [7]Department of Neurology, Medical Faculty, Heinrich-Heine University, Düsseldorf, Germany; Brain and Mind Centre, University of Sydney, Sydney, Australia; Department of Neurology, Medical University of Vienna, Vienna, Austria; Department of Neurology, Palacky University Olomouc, Olomouc, Czech Republic; [8]Department of Neurology-Neuroimmunology, Centre d'Esclerosi Múltiple de Catalunya (Cemcat), Hospital Universitari Vall d'Hebron, Barcelona, Spain; [9]Department of Neurology and Center for Clinical Neuroscience, First Medical Faculty, Charles University, Prague, Czech Republic; [10]Bristol Myers Squibb, Princeton, New Jersey; [11]Research Center for Clinical Neuroimmunology and Neuroscience, Departments of Medicine, Clinical Research, Biomedicine, and Biomedical Engineering, University Hospital and University of Basel, Basel, Switzerland; [12]Mellen Center for MS Treatment and Research, Cleveland Clinic, Cleveland, Ohio

To request a copy of this poster:

Scan QR code via a barcode reader application

Introduction

- Ozanimod, a sphingosine 1-phosphate receptor 1 and 5 modulator,[1] is approved in multiple countries for the treatment of adults with either relapsing forms of multiple sclerosis (RMS) or moderately to severely active ulcerative colitis[2,3]
- As of November 19, 2021
 - 5622 healthy volunteers or patients with RMS, ulcerative colitis, or Crohn's disease with 16,536.65 patient-years (PY) of exposure have received ozanimod in clinical trials
 - 9738 patients with 6455 PY of exposure were treated with ozanimod in postmarketing settings
- DAYBREAK is an ongoing open-label extension (OLE) study that enrolled patients with RMS from completed phase 1-3 trials of ozanimod[4-7]
- Objective: To report interim efficacy and safety data, including COVID-19 infections, during extended exposure to ozanimod among patients in the DAYBREAK OLE trial (NCT02576717)

Methods

Parent studies and DAYBREAK study design[8]

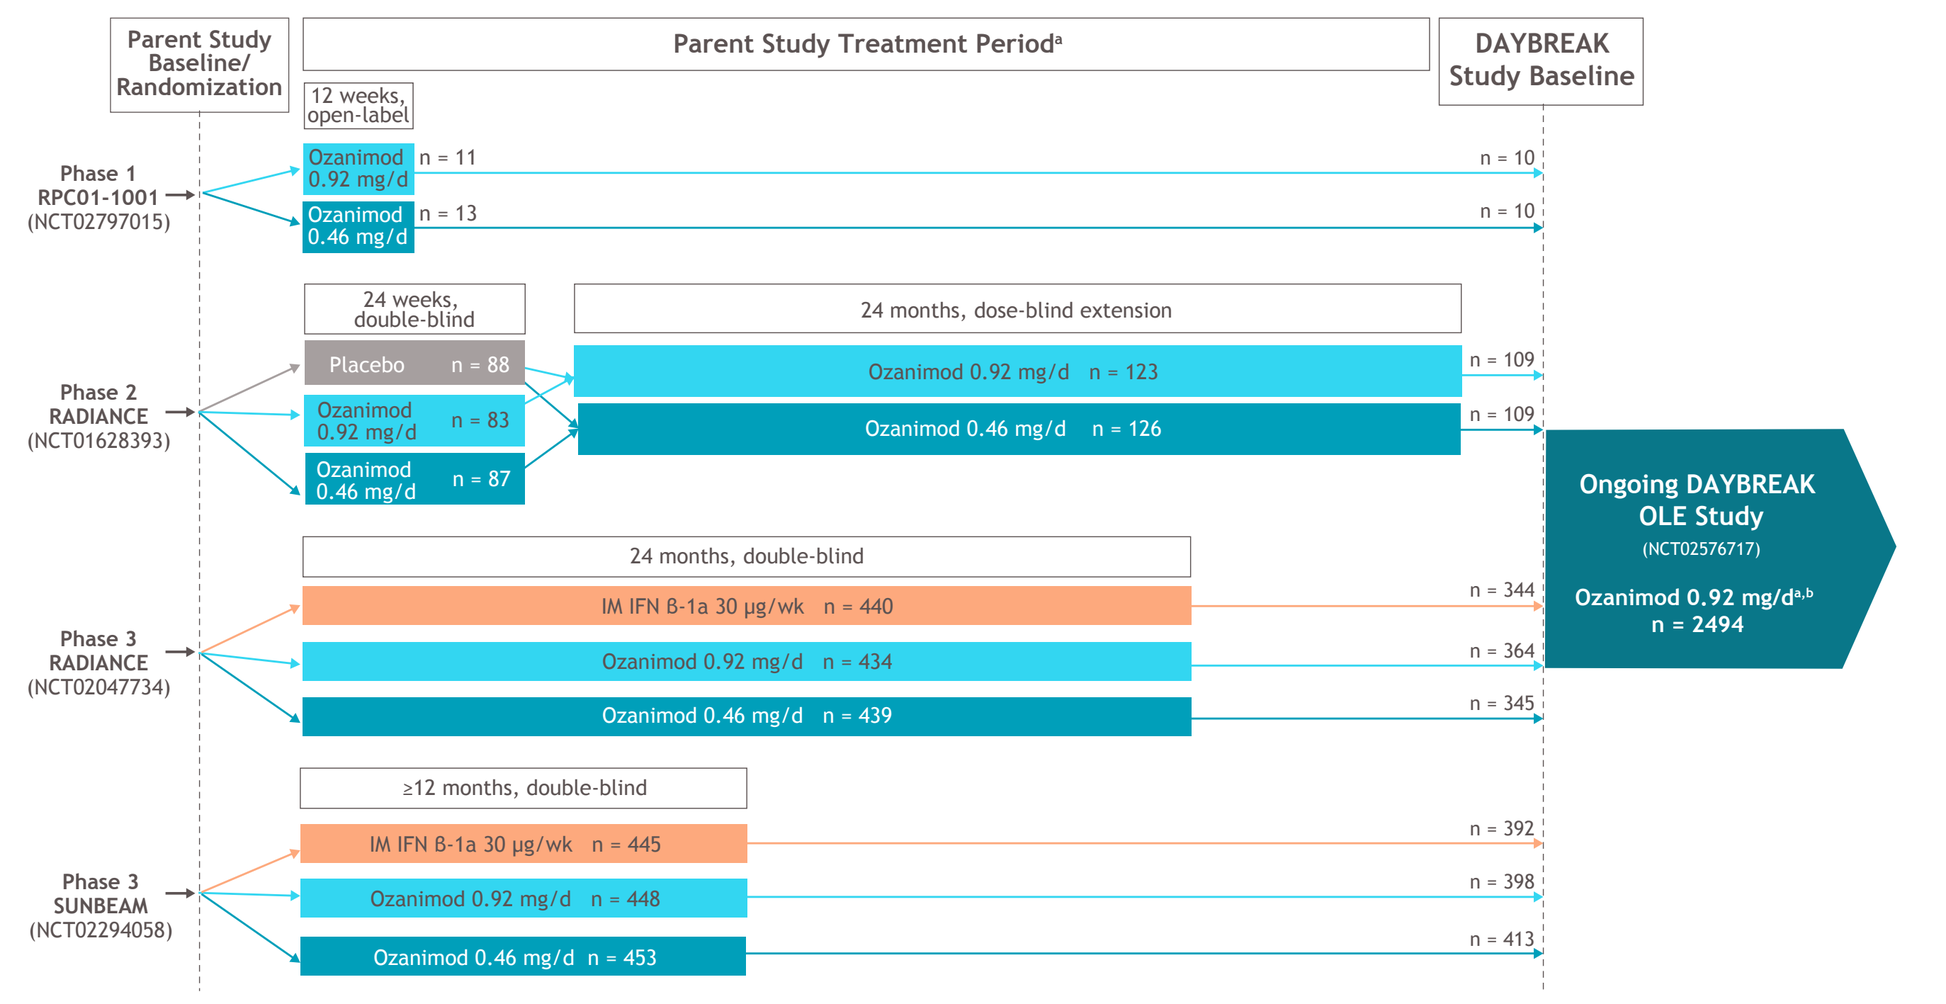

Parent Study Baseline/ Randomization
Parent Study Treatment Period[a]
DAYBREAK Study Baseline
12 weeks, open-label
Phase 1 RPC01-1001 (NCT02797015)
Ozanimod 0.92 mg/d n = 11
Ozanimod 0.46 mg/d n = 13
n = 10
n = 10
24 weeks, double-blind
24 months, dose-blind extension
Phase 2 RADIANCE (NCT01628393)
Placebo n = 88
Ozanimod 0.92 mg/d n = 83
Ozanimod 0.46 mg/d n = 87
Ozanimod 0.92 mg/d n = 123
Ozanimod 0.46 mg/d n = 126
n = 109
n = 109
Ongoing DAYBREAK OLE Study (NCT02576717)
Ozanimod 0.92 mg/d[a,b] n = 2494
24 months, double-blind
Phase 3 RADIANCE (NCT02047734)
IM IFN β-1a 30 µg/wk n = 440
Ozanimod 0.92 mg/d n = 434
Ozanimod 0.46 mg/d n = 439
n = 344
n = 364
n = 345
≥12 months, double-blind
Phase 3 SUNBEAM (NCT02294058)
IM IFN β-1a 30 µg/wk n = 445
Ozanimod 0.92 mg/d n = 448
Ozanimod 0.46 mg/d n = 453
n = 392
n = 398
n = 413

[a]In all trials, upon initiation of ozanimod, patients received 0.23 mg on days 1-4, 0.46 mg on days 5-7, and then their assigned dose of 0.46 or 0.92 mg on day 8 and thereafter. All patients entering the phase 2 dose-blinded extension period underwent dose escalation, even if treated with ozanimod in the parent trial, to maintain the blind. [b]In DAYBREAK, dose escalation was performed for all patients entering from one of the active-controlled phase 3 trials, irrespective of prior treatment assignment (to maintain the blinding in the parent trials); dose escalation was not performed for those entering from the phase 1 or 2 trials, unless the last dose of ozanimod was > 14 days before entering DAYBREAK. HCl, hydrochloride; IFN, interferon; IM, intramuscular; PO, per os (oral); QD, once daily. Figure adapted from Selmaj KW, Cohen JA, Comi G, et al. Ozanimod in relapsing multiple sclerosis: pooled safety results from the clinical development program. *Mult Scler Relat Disord*. 2021:51. https://doi.org/10.1016/j.msard.2021.102844 under Creative Commons Attribution 4.0 International License.

Analysis timeline

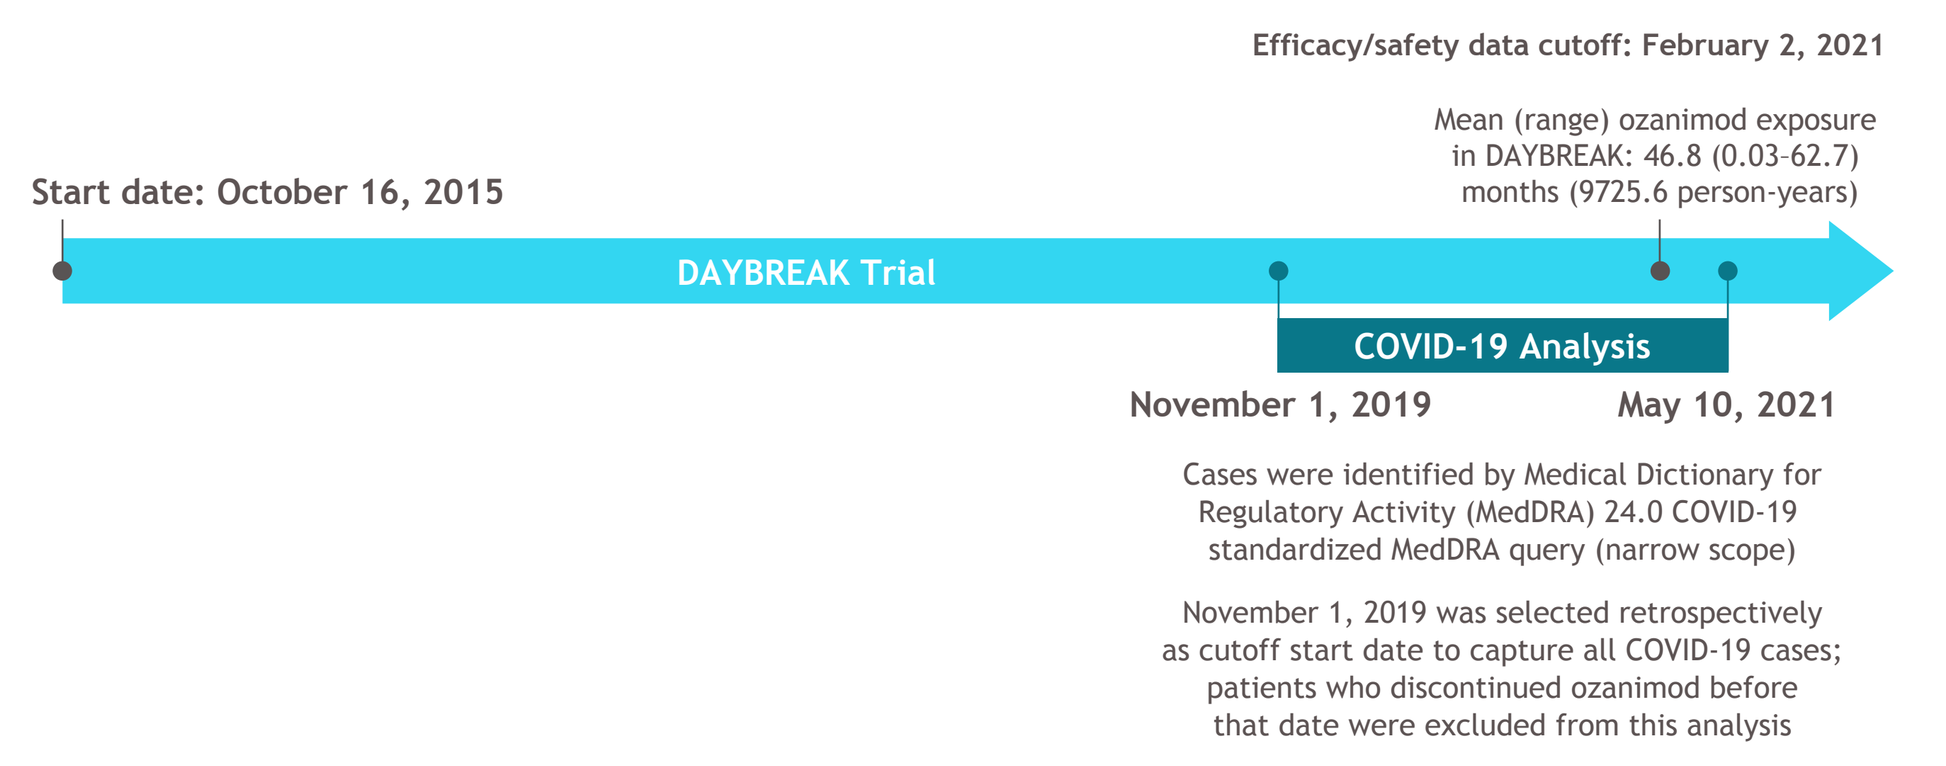

Efficacy/safety data cutoff: February 2, 2021
Mean (range) ozanimod exposure in DAYBREAK: 46.8 (0.03-62.7) months (9725.6 person-years)
Start date: October 16, 2015
DAYBREAK Trial
COVID-19 Analysis
November 1, 2019
May 10, 2021
Cases were identified by Medical Dictionary for Regulatory Activity (MedDRA) 24.0 COVID-19 standardized MedDRA query (narrow scope)
November 1, 2019 was selected retrospectively as cutoff start date to capture all COVID-19 cases; patients who discontinued ozanimod before that date were excluded from this analysis

Results

Demographics and disease characteristics at DAYBREAK baseline

Parameter	IFN β-1a 30 µg → Ozanimod 0.92 mg (n = 736)	Ozanimod 0.46 mg → Ozanimod 0.92 mg (n = 877)	Continuous Ozanimod 0.92 mg (n = 881)	All DAYBREAK (Ozanimod 0.92 mg) (N = 2494)
Age, mean (SD), years	37.4 (9.1)	38.0 (9.3)	37.6 (9.3)	37.7 (9.2)
Sex, n (%)				
Female	498 (67.7)	595 (67.8)	575 (65.3)	1668 (66.9)
Male	238 (32.3)	282 (32.2)	306 (34.7)	826 (33.1)
Race, n (%)				
White	734 (99.7)	865 (98.6)	875 (99.3)	2474 (99.2)
Black	1 (0.1)	7 (0.8)	6 (0.7)	14 (0.6)
Asian	0	3 (0.3)	0 (0)	3 (0.1)
Other	1 (0.1)	2 (0.2)	0 (0)	3 (0.1)
Region, n (%)				
Eastern Europe	672 (91.3)	791 (90.2)	783 (88.9)	2246 (90.1)
Western Europe	44 (6.0)	46 (5.2)	54 (6.1)	144 (5.8)
North America	13 (1.8)	33 (3.8)	35 (4.0)	81 (3.2)
Rest of world[a]	7 (1.0)	7 (0.8)	9 (1.0)	23 (0.9)
Age at symptom onset, mean (SD), years	29.5 (8.8)	29.7 (9.1)[b]	29.2 (8.8)[c]	29.5 (8.9)[d]
EDSS score, mean (SD)	2.5 (1.3)	2.6 (1.3)	2.6 (1.3)	2.6 (1.3)

[a]Rest of world includes New Zealand and South Africa. [b]n = 867. [c]n = 871. [d]n = 2474.
EDSS, Expanded Disability Status Scale; SD, standard deviation.

Relapses

- Ozanimod treatment demonstrated sustained efficacy with a low annualized relapse rate (ARR) and most patients remaining relapse free during DAYBREAK (75% at month 36; 71% at month 48)
 - The greatest numerical reduction in ARR from phase 3 parent trial to DAYBREAK occurred among those who switched from IFN β-1a to ozanimod 0.92 mg
 - Among those who continuously received ozanimod 0.92 mg in both a parent trial and DAYBREAK, 64% remained relapse free throughout 60 cumulative months of treatment (the last time point for which more than half of the analysis group had assessments; n = 493)

ARR in DAYBREAK, ITT population

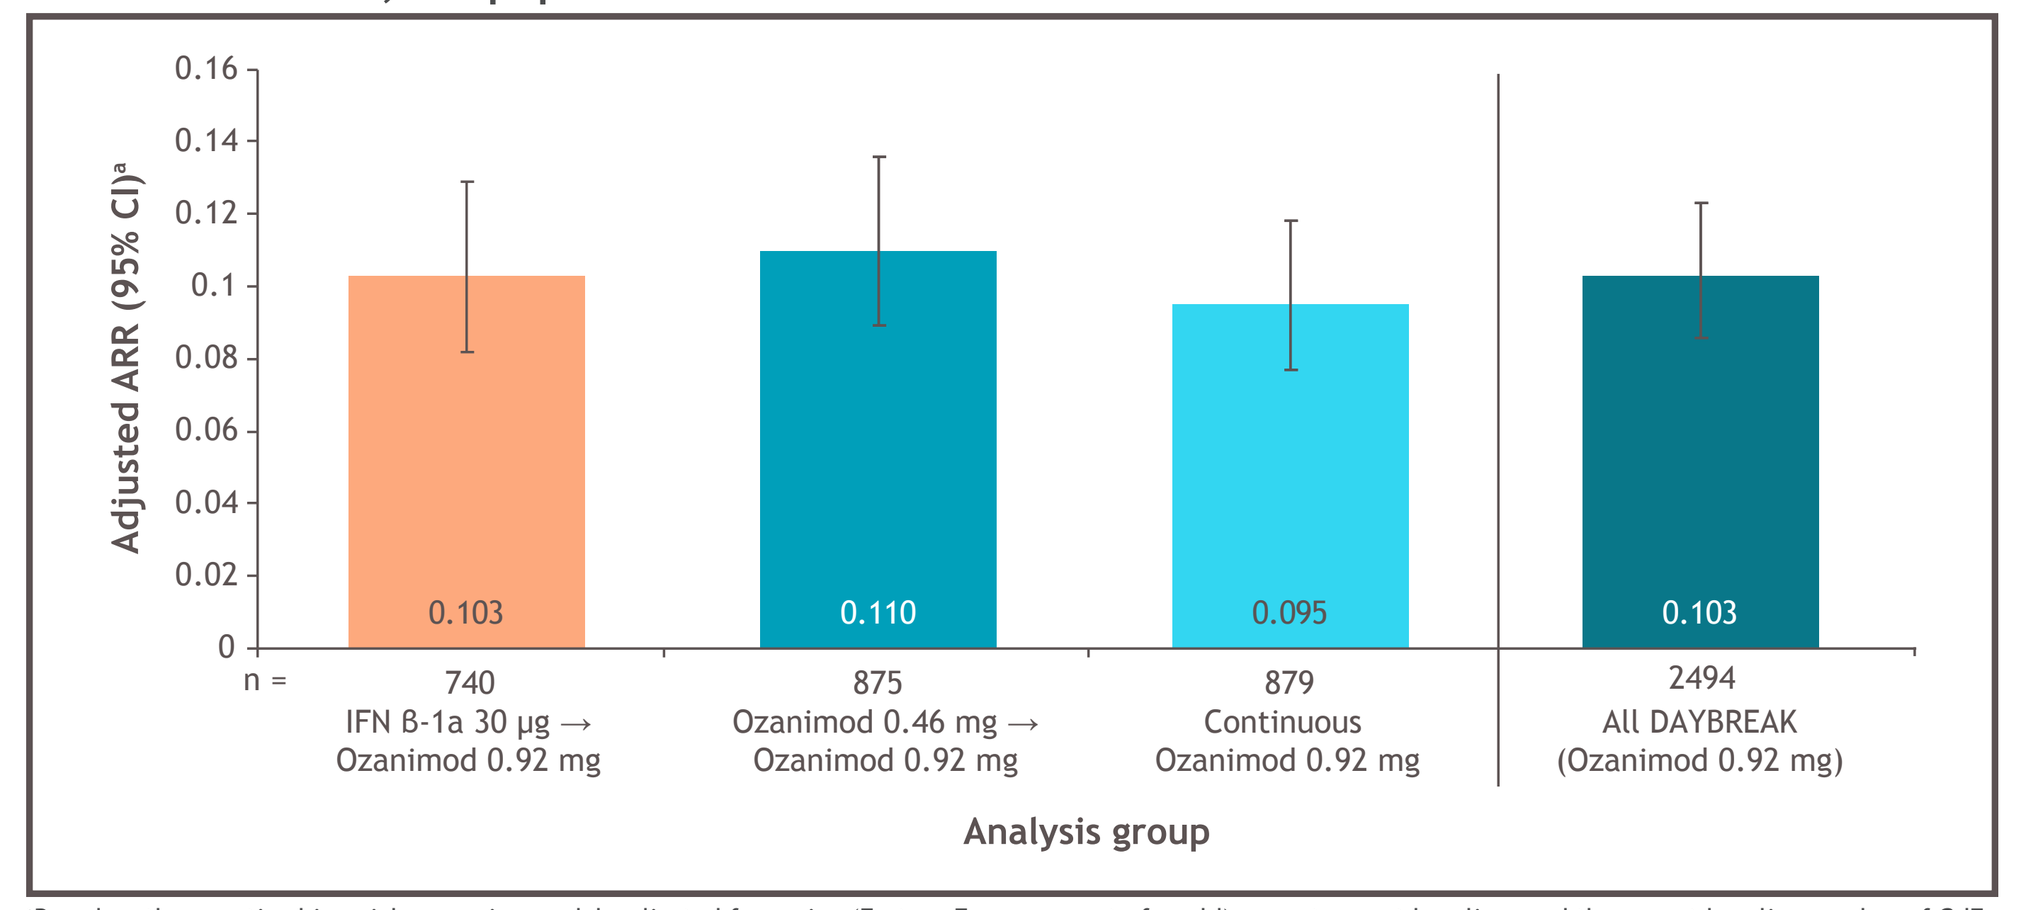

Adjusted ARR (95% CI)[a]
0.16
0.14
0.12
0.1
0.08
0.06
0.04
0.02
0
0.103
0.110
0.095
0.103
n =
740
IFN β-1a 30 µg → Ozanimod 0.92 mg
875
Ozanimod 0.46 mg → Ozanimod 0.92 mg
879
Continuous Ozanimod 0.92 mg
2494
All DAYBREAK (Ozanimod 0.92 mg)
Analysis group

[a]Based on the negative binomial regression model, adjusted for region (Eastern Europe vs rest of world), age at parent baseline, and the parent baseline number of GdE lesions. The natural log transformation of time on treatment is used as an offset term to adjust for persons having different exposure times. GdE, gadolinium-enhancing.

Kaplan-Meier curve for time to first confirmed relapse during DAYBREAK, ITT population

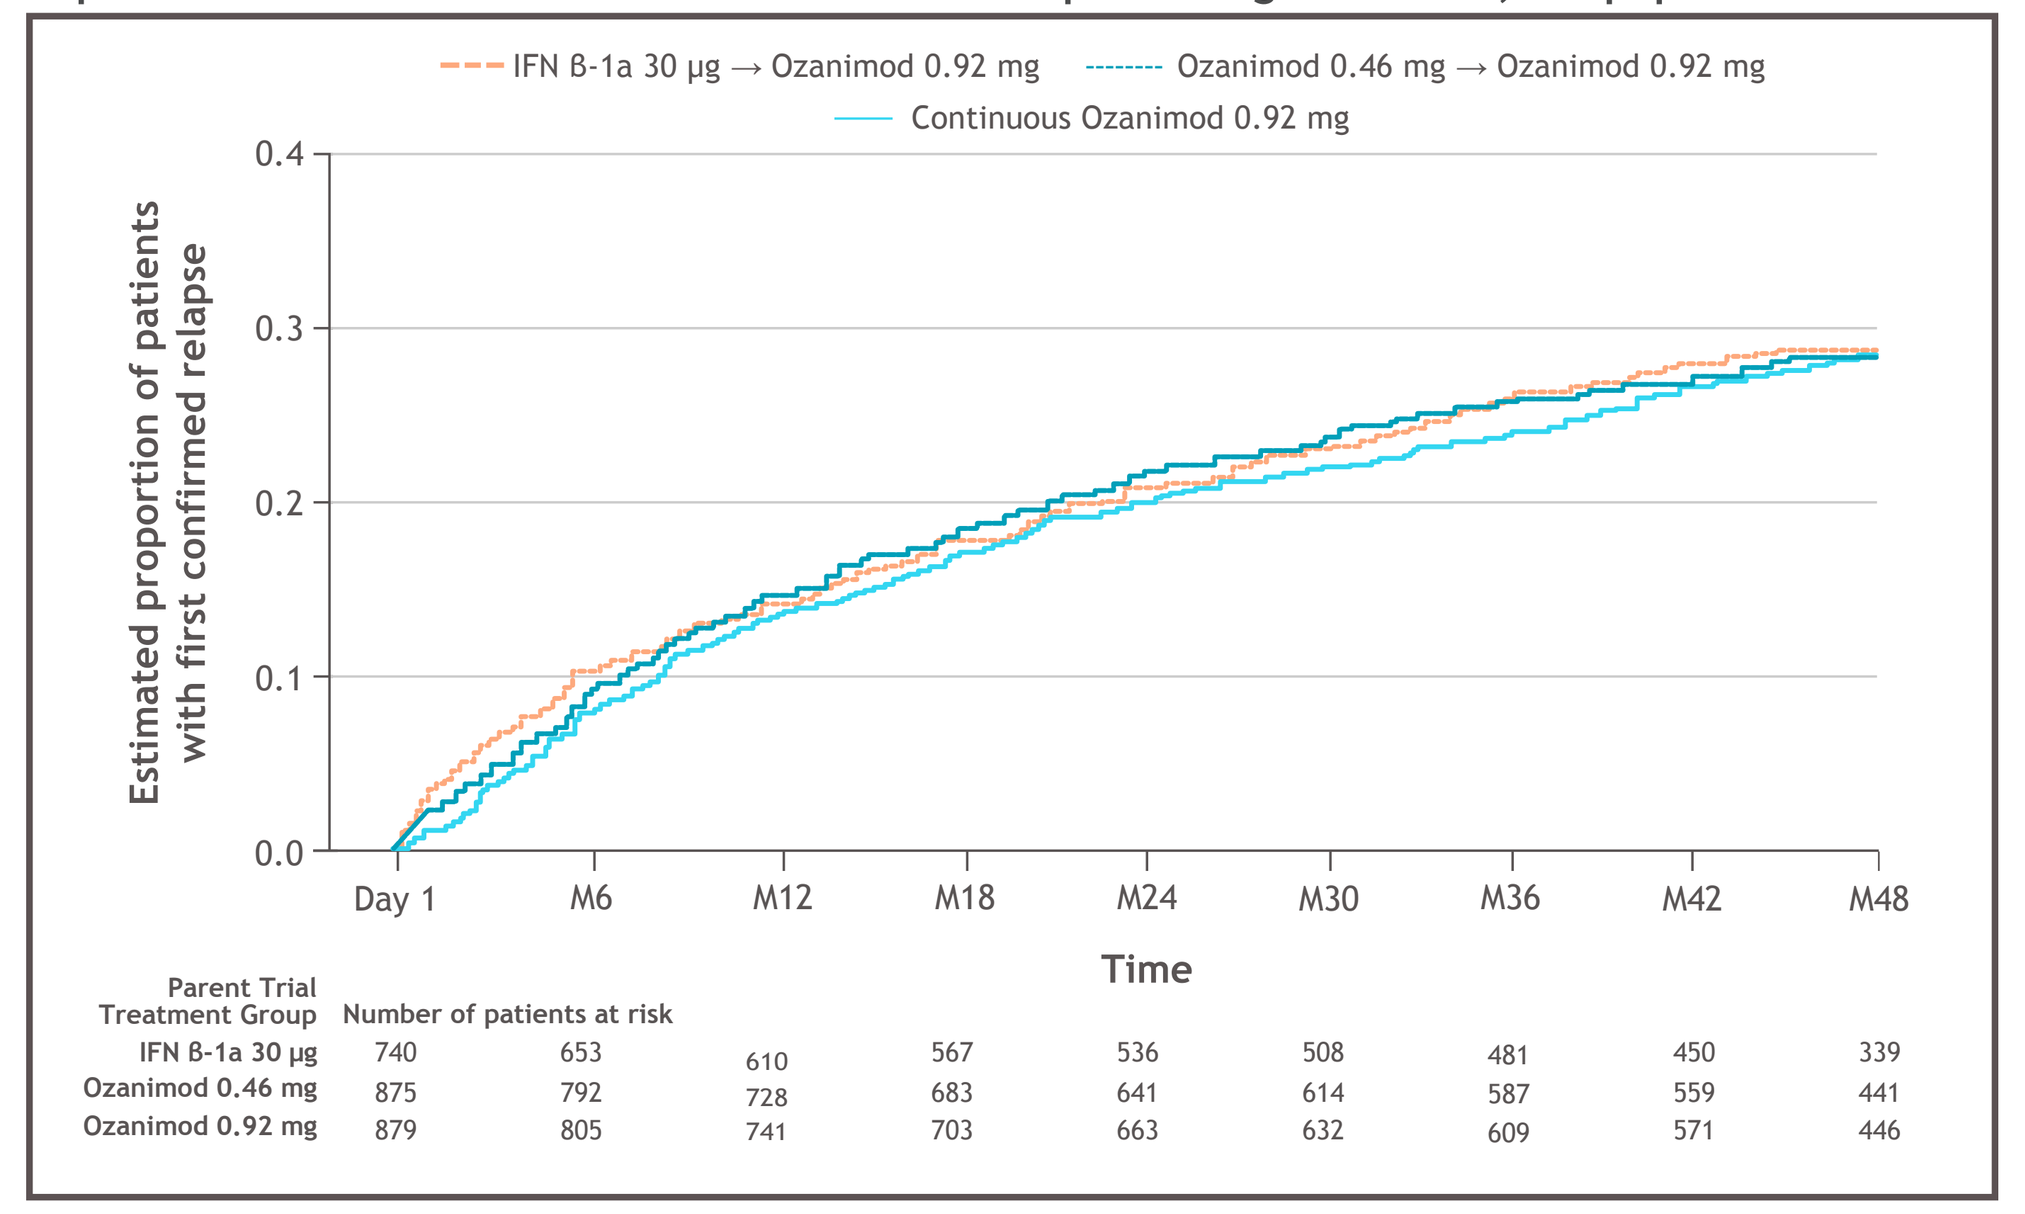

IFN β-1a 30 µg → Ozanimod 0.92 mg
Ozanimod 0.46 mg → Ozanimod 0.92 mg
Continuous Ozanimod 0.92 mg
Estimated proportion of patients with first confirmed relapse
0.4
0.3
0.2
0.1
0.0
Day 1
M6
M12
M18
M24
M30
M36
M42
M48
Time
Parent Trial Treatment Group
Number of patients at risk
IFN β-1a 30 µg 740 653 610 567 536 508 481 450 339
Ozanimod 0.46 mg 875 792 728 683 641 614 587 559 441
Ozanimod 0.92 mg 879 805 741 703 663 632 609 571 446

Other efficacy endpoints

- Adjusted mean number of new/enlarging T2 lesions per scan at month 48 of DAYBREAK was low and similar, regardless of parent trial treatment group, as was adjusted mean number of gadolinium enhancing lesions at month 48
 - Patients initially treated with IFN β-1a experienced a decline in lesion counts upon switching to ozanimod 0.92 mg in DAYBREAK
- During DAYBREAK, 346/2494 (13.9%) patients had confirmed disability progression at 3 months and 285/2494 (11.4%) had confirmed disability progression at 6 months by the data cutoff
 - Rates were similar across parent trial treatment groups

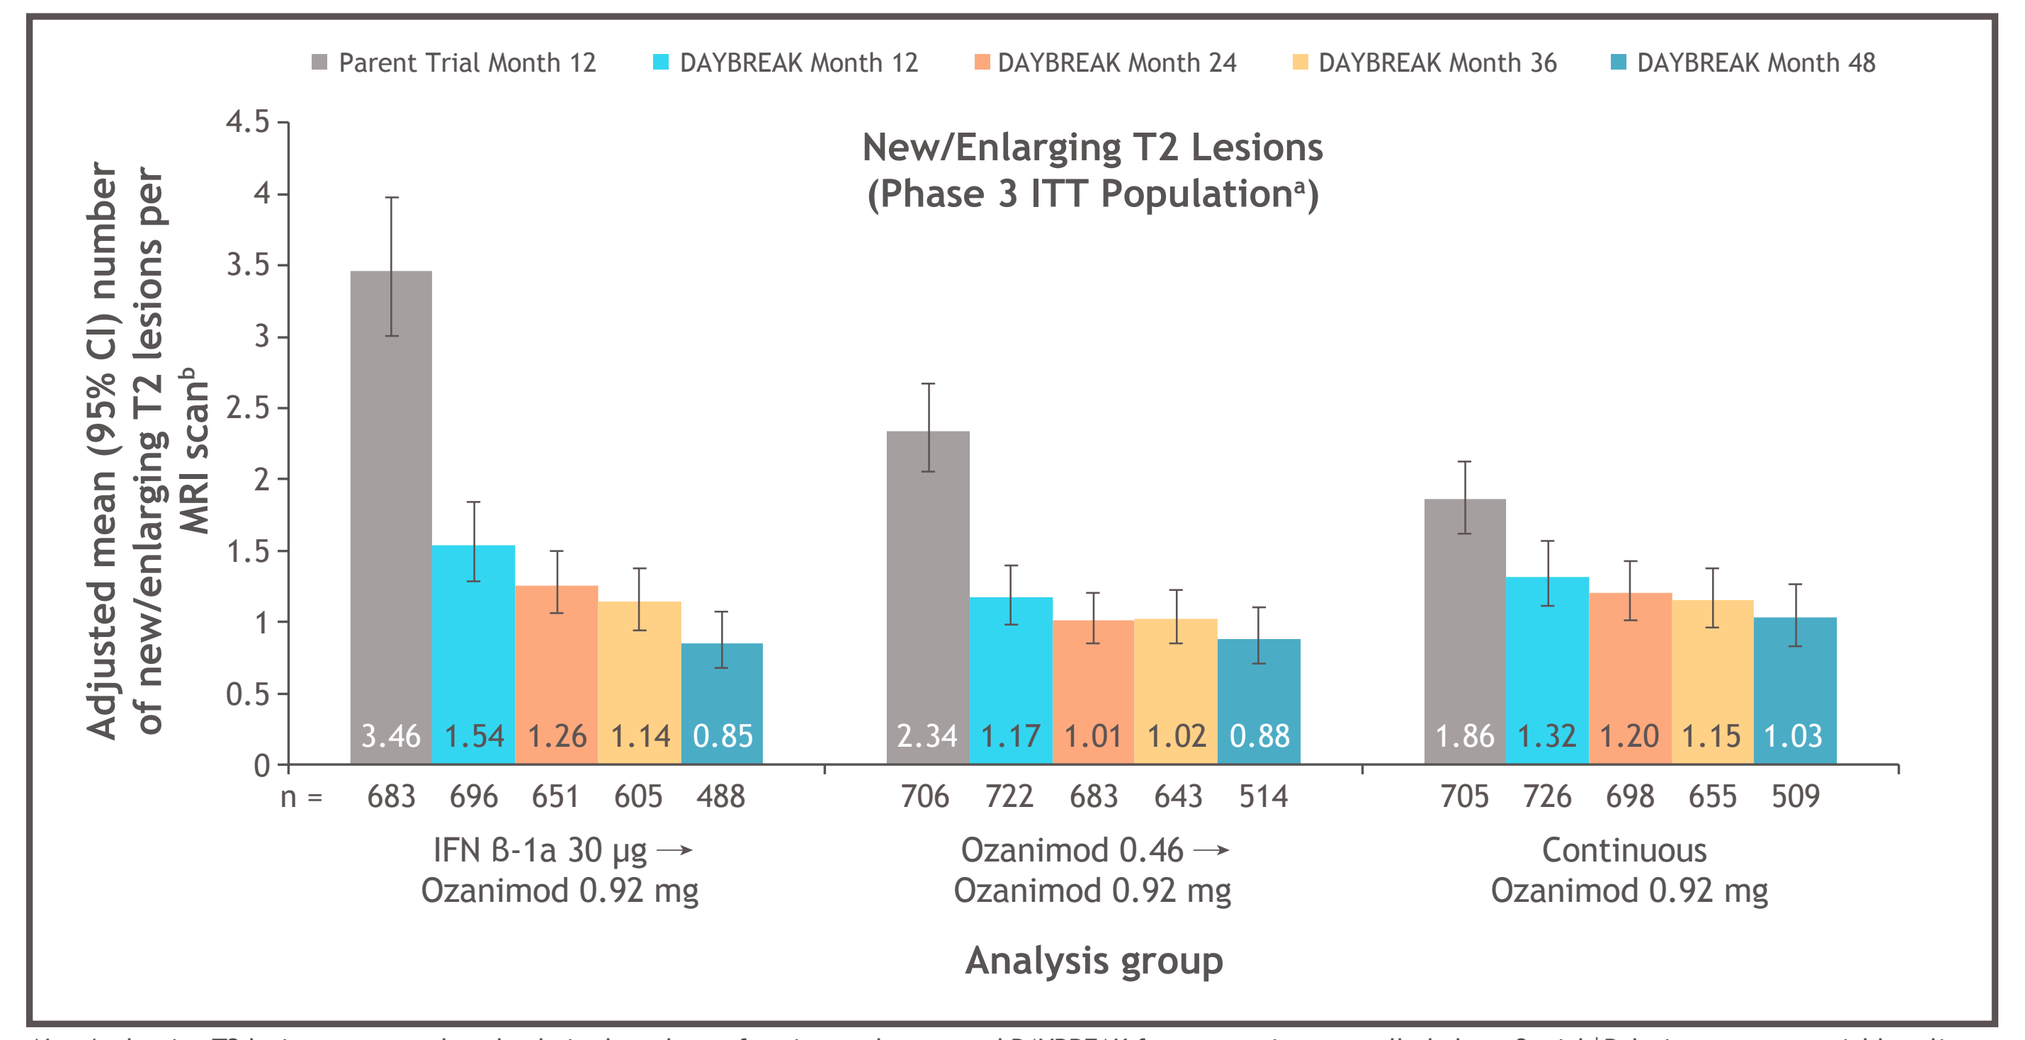

Parent Trial Month 12
DAYBREAK Month 12
DAYBREAK Month 24
DAYBREAK Month 36
DAYBREAK Month 48
New/Enlarging T2 Lesions
(Phase 3 ITT Population[a])
Adjusted mean (95% CI) number of new/enlarging T2 lesions per MRI scan[b]
3.46 1.54 1.26 1.14 0.85
2.34 1.17 1.01 1.02 0.88
1.86 1.32 1.20 1.15 1.03
n = 683 696 651 605 488
706 722 683 643 514
705 726 698 655 509
IFN β-1a 30 µg → Ozanimod 0.92 mg
Ozanimod 0.46 → Ozanimod 0.92 mg
Continuous Ozanimod 0.92 mg
Analysis group

[a]New/enlarging T2 lesions were analyzed only in the subset of patients who entered DAYBREAK from an active-controlled phase 3 trial. [b]Relative to parent trial baseline in the parent trial, and DAYBREAK baseline in DAYBREAK; based on negative binomial regression model, adjusted for study, region (Eastern Europe vs rest of world), age at baseline, and baseline number of GdE lesions, with the log of the number of postbaseline scans as an offset term in the model.

COVID-19 cases among patients taking ozanimod were largely nonserious, and 185/190 (97.4%) patients recovered while either continuing or temporarily interrupting ozanimod

Characteristics of DAYBREAK participants with ≥ 1 COVID-19 event[a]

Characteristic	COVID-19 Cases
n COVID-19 cases[a]/N total at risk (%)	190/2181 (8.7)
Median age (range), years	40 (21-60)
Age, n/N (%), years[b]	
18 - ≤ 29	18/304 (5.9)
30 - ≤ 49	127/1475 (8.6)
50 - ≤ 64	45/402 (11.2)
Sex, n/N (%)	
Female	132/1441 (9.2)
Male	58/740 (7.8)
Region, n/N (%)	
North America[c]	2/60 (3.3)
South America	0/0 (0)
Europe/Russia[d]	186/2104 (8.8)
Asia	0/0 (0)
Africa	2/10 (20.0)
Other (New Zealand)	0/7 (0)

[a]COVID-19 events occurring between November 1, 2019 through May 10, 2021 were identified by Medical Dictionary for Regulatory Activity (MedDRA) 24.0 COVID-19 standardized MedDRA query (narrow scope) among patients taking ozanimod. COVID-19 event was considered treatment-emergent if onset was reported in the time period from treatment start date to treatment end date (defined as 90 days after date of last dose or date of death, whichever occurred earlier). Only the last infection was selected for this analysis, regardless of seriousness/outcome. [b]Age of study participants with COVID-19 represents age at the start date of the reported COVID-19 adverse event. For subjects without COVID, age was derived as the age on November 1, 2019. [c]Two COVID-19 cases were reported in the United States. [d]The majority of study participants and COVID-19 cases were from Eastern Europe.

Outcomes of COVID-19 cases[a]

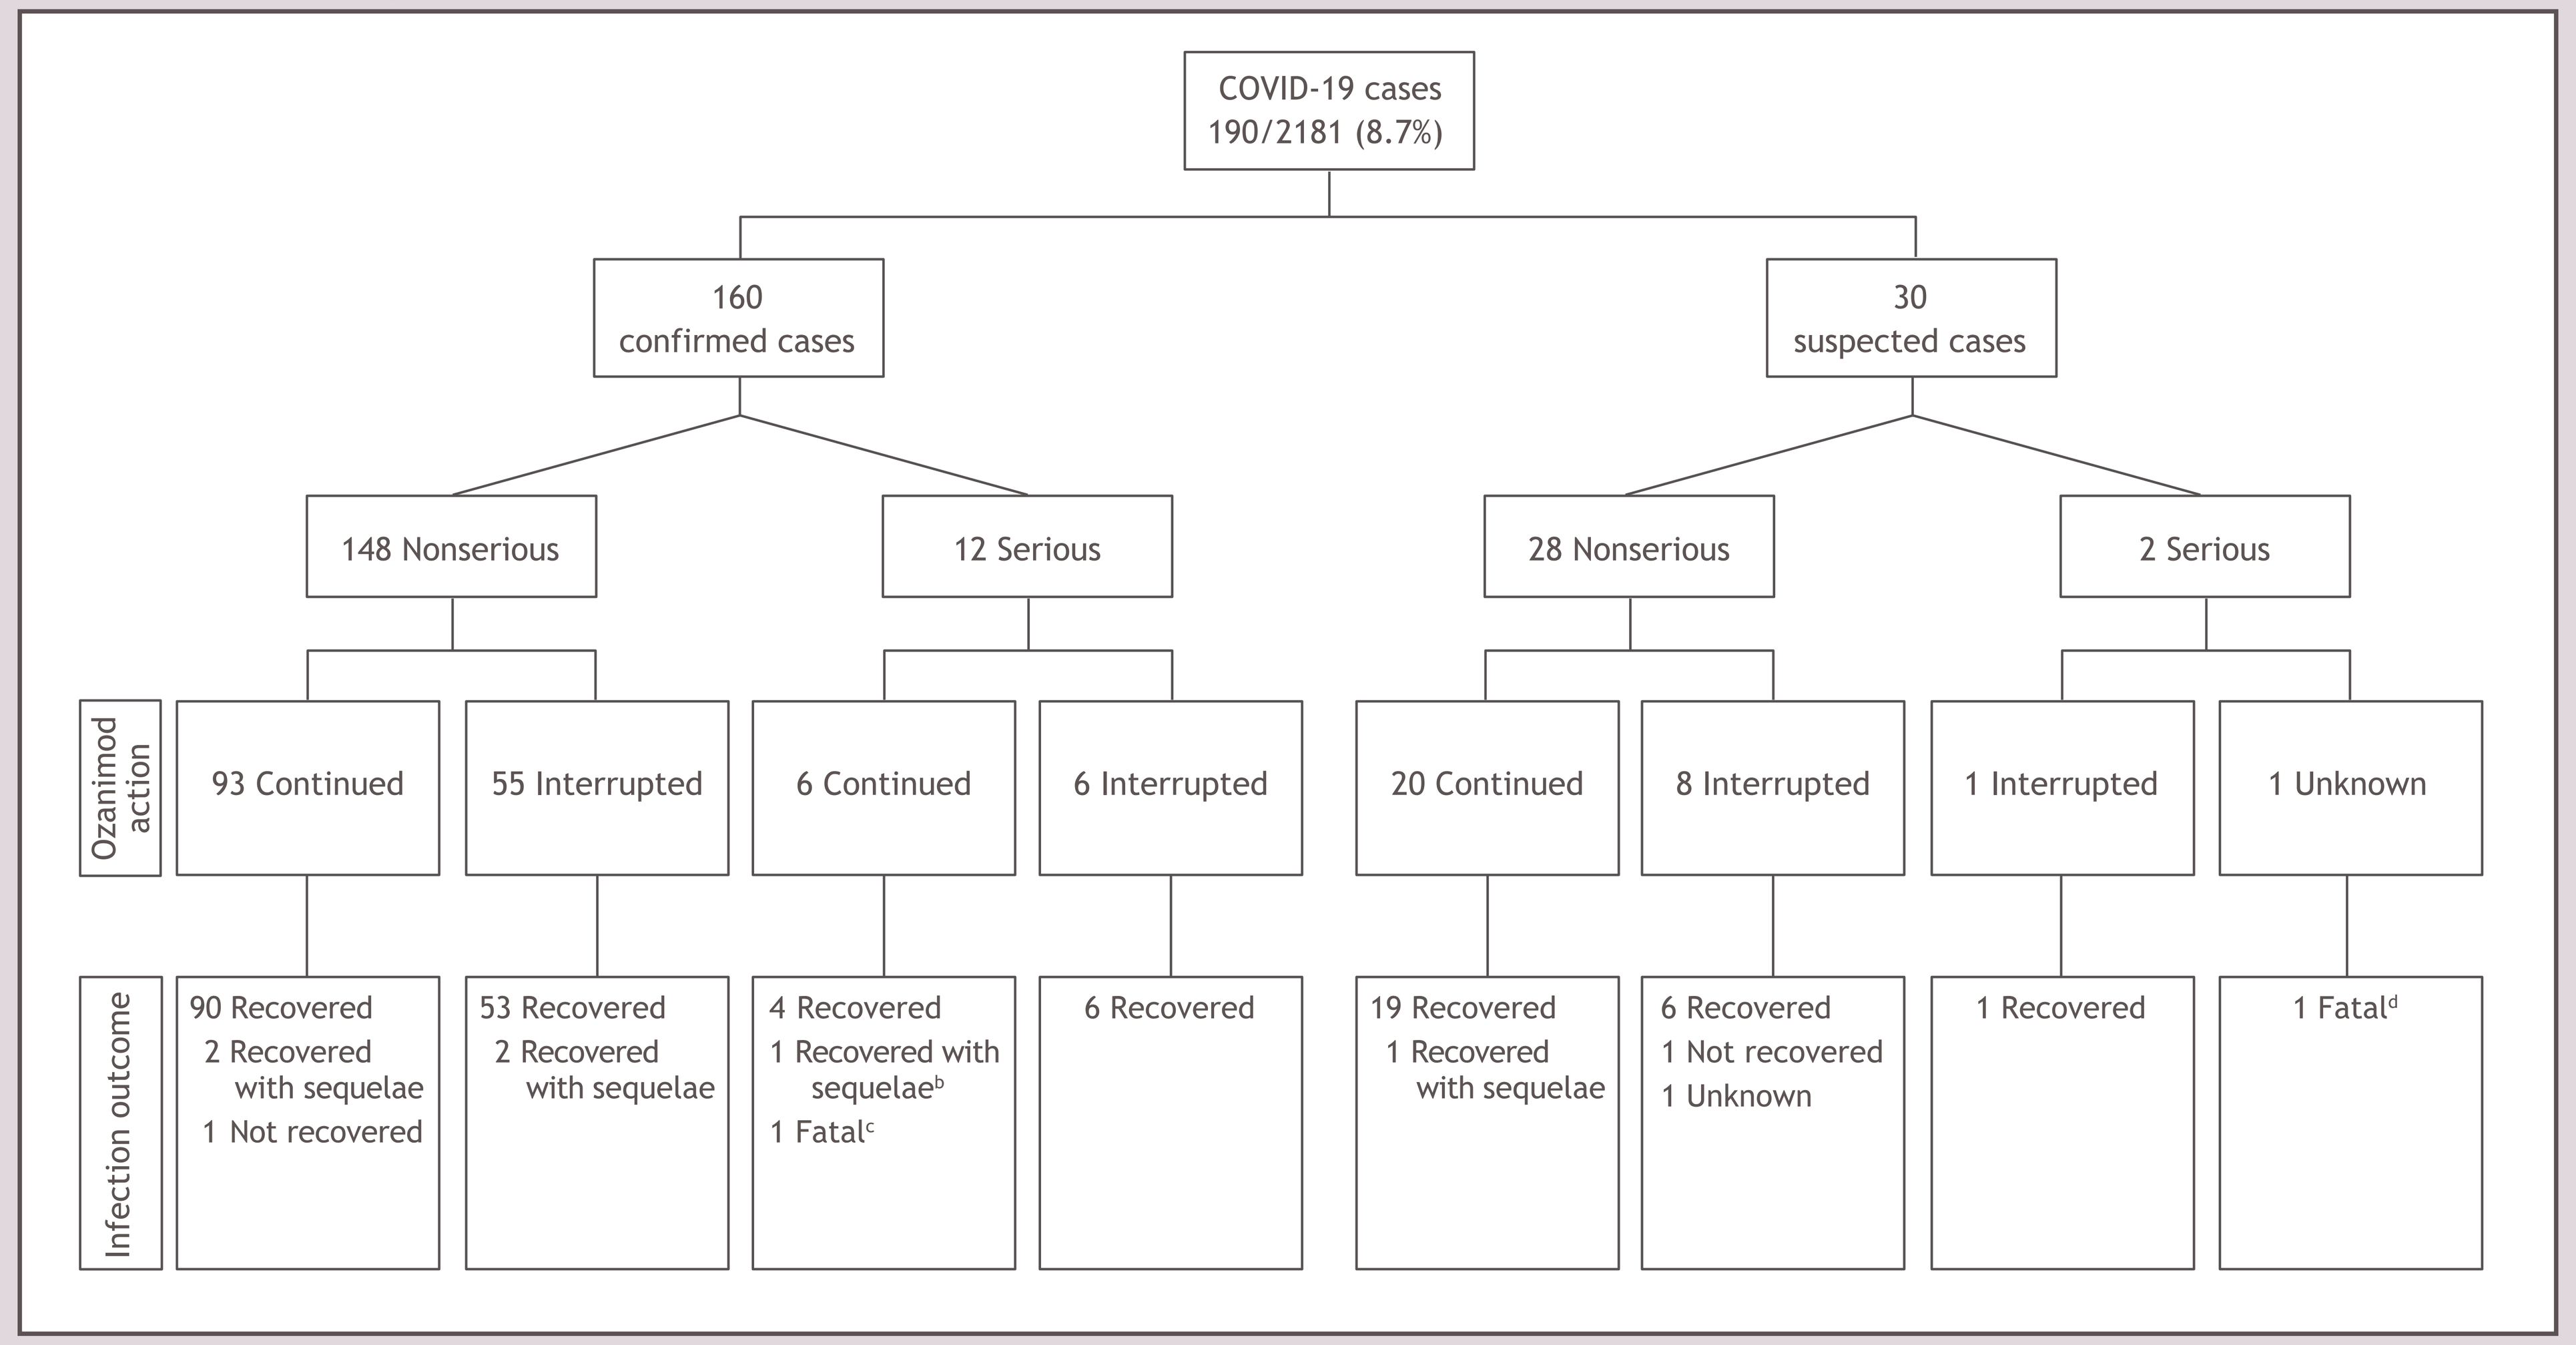

COVID-19 cases
190/2181 (8.7%)
160
confirmed cases
30
suspected cases
148 Nonserious
12 Serious
28 Nonserious
2 Serious
Ozanimod action
93 Continued
55 Interrupted
6 Continued
6 Interrupted
20 Continued
8 Interrupted
1 Interrupted
1 Unknown
Infection outcome
90 Recovered
2 Recovered with sequelae
1 Not recovered
53 Recovered
2 Recovered with sequelae
4 Recovered
1 Recovered with sequelae[b]
1 Fatal[c]
6 Recovered
19 Recovered
1 Recovered with sequelae
6 Recovered
1 Not recovered
1 Unknown
1 Recovered
1 Fatal[d]

[a]Cases were categorized as confirmed if the patient was symptomatic and COVID-19 was confirmed with a diagnostic laboratory test and/or by the investigator based on their clinical evaluation, or as suspected if the patient's diagnosis was not confirmed either with a diagnostic laboratory test or clinical evaluation. Serious adverse events were identified by the investigator based on criteria from the International Conference on Harmonisation.[9] Only the last infection was selected for this analysis, regardless of seriousness or outcome. Outcomes are at the time of reporting by the investigator. [b]One tetraplegic, cachectic patient who had swallowing difficulties developed confirmed, serious COVID-19; the patient recovered with sequelae from the initial infection but subsequently died from a lung abscess as a complication of *Pseudomonas aeruginosa* pneumonia. [c]One patient developed confirmed, serious COVID-19 immediately following a 5-day inpatient course of intravenous methylprednisolone for treatment of an MS relapse. She was readmitted for COVID-19 treatment. Her respiratory function deteriorated despite supportive care, and she died from a suspected pulmonary embolism about 2 weeks after re-admission. [d]One patient died from suspected, serious COVID-19 pneumonia.

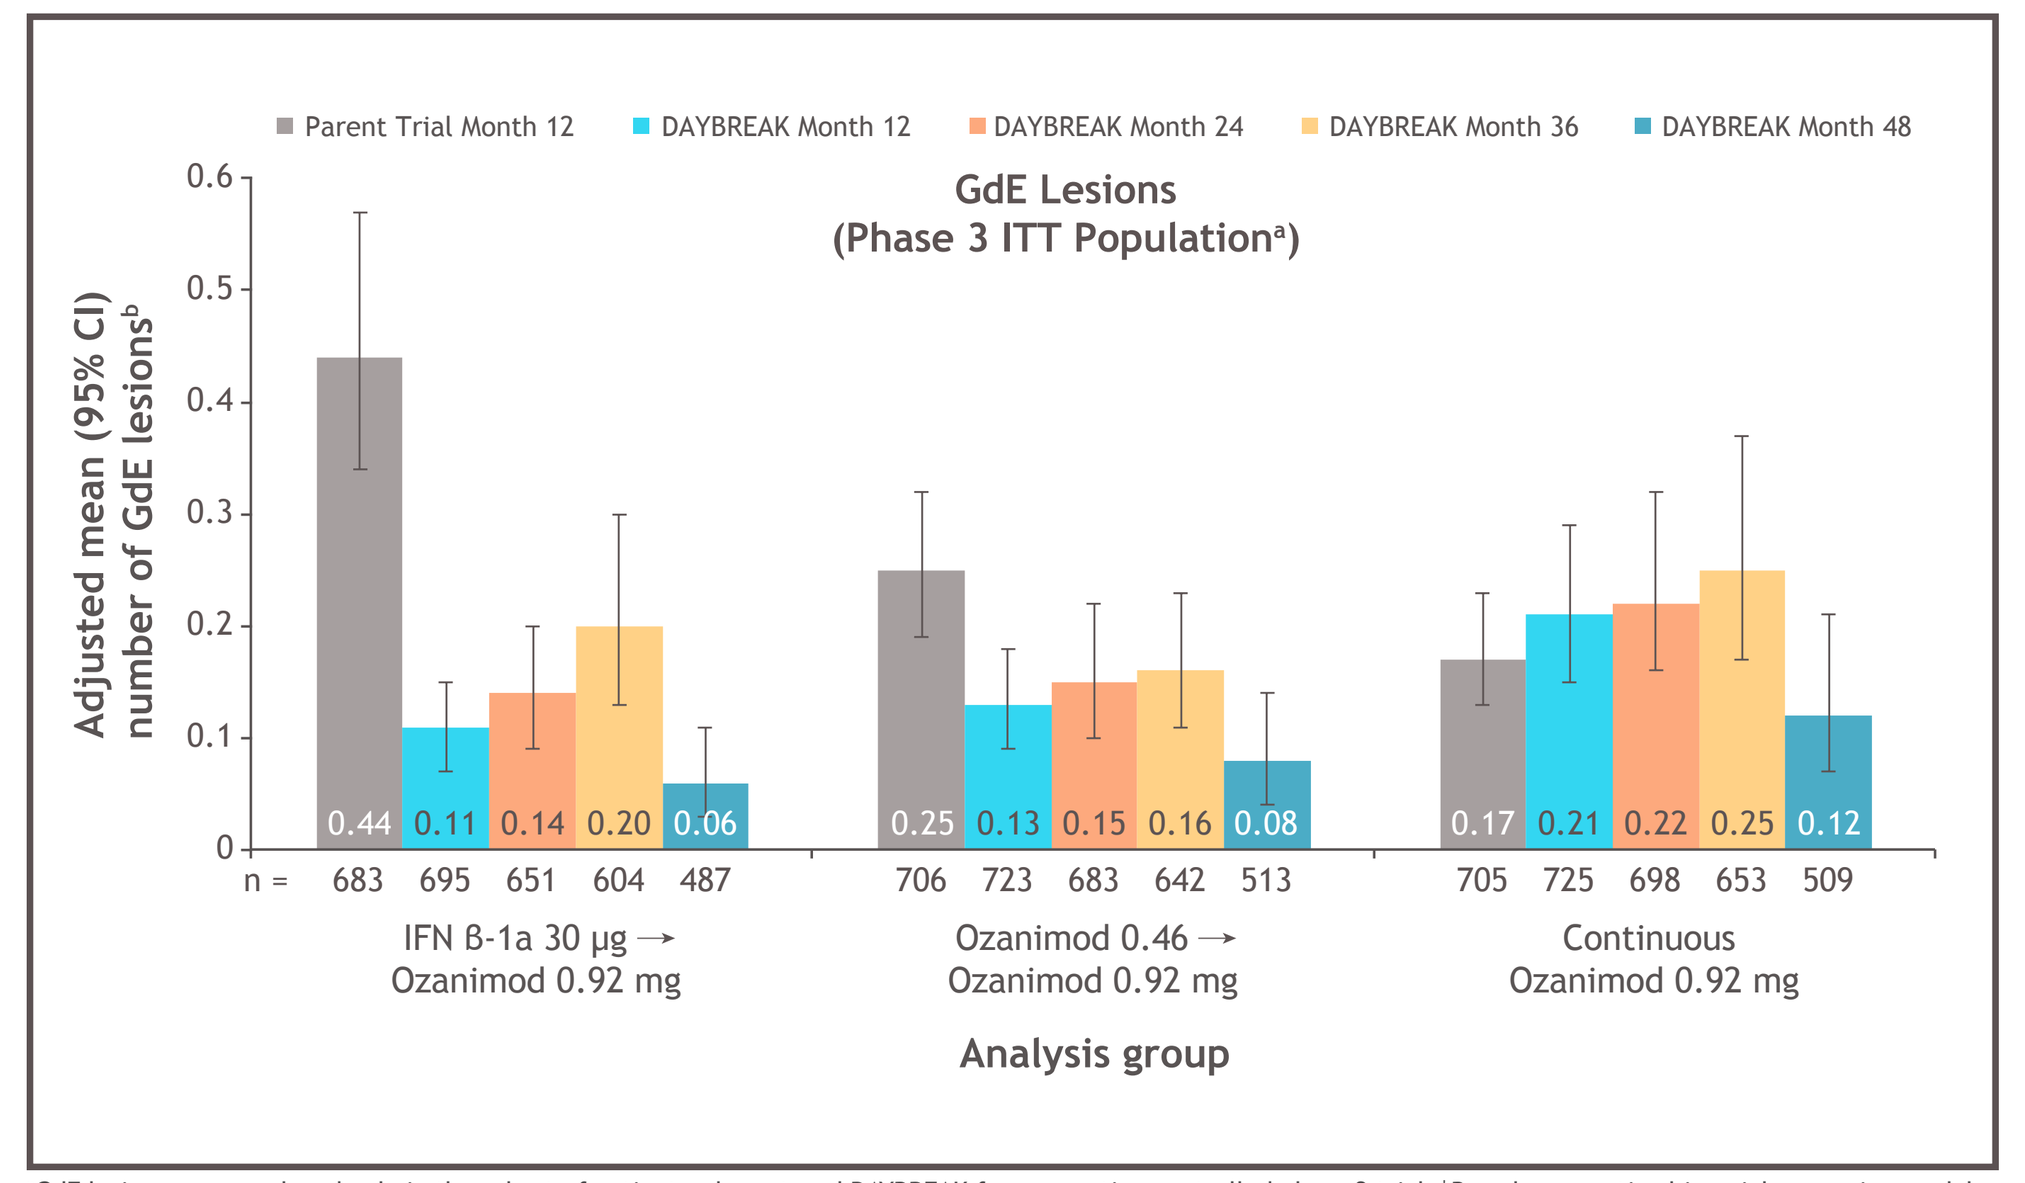

Parent Trial Month 12
DAYBREAK Month 12
DAYBREAK Month 24
DAYBREAK Month 36
DAYBREAK Month 48
GdE Lesions
(Phase 3 ITT Population[a])
Adjusted mean (95% CI) number of GdE lesions[b]
0.44 0.11 0.14 0.20 0.06
0.25 0.13 0.15 0.16 0.08
0.17 0.21 0.22 0.25 0.12
n = 683 695 651 604 487
706 723 683 642 513
705 725 698 653 509
IFN β-1a 30 µg → Ozanimod 0.92 mg
Ozanimod 0.46 → Ozanimod 0.92 mg
Continuous Ozanimod 0.92 mg
Analysis group

[a]GdE lesions were analyzed only in the subset of patients who entered DAYBREAK from an active-controlled phase 3 trial. [b]Based on negative binomial regression model, adjusted for study, region (Eastern Europe vs rest of world), age at baseline, and baseline number of GdE lesions.

Overall safety

- Similar rates of treatment-emergent adverse events (TEAEs) and serious adverse events occurred when assessed by parent trial treatment group
- The low discontinuation rate due to TEAEs (3.0%) suggests that ozanimod was well tolerated
- Prior to the February 2, 2021 data cutoff, 3 patients died from malignancies, 2 from accidents, and 1 each from community-acquired pneumonia, cerebral hemorrhage probably due to rupture of a preexisting occult aneurysm, sudden death due to unknown cause, and pulmonary embolism following leg fracture surgery
 - There were 3 additional deaths associated with COVID-19 in the separate COVID-19 analysis with a cutoff of May 10, 2021

Safety during DAYBREAK by parent trial treatment group and overall DAYBREAK population[a]

Parameter	IFN β-1a 30 µg → Ozanimod 0.92 mg (n = 736)	Ozanimod 0.46 mg → Ozanimod 0.92 mg (n = 877)	Continuous Ozanimod 0.92 mg (n = 881)	All DAYBREAK Ozanimod 0.92 mg (N = 2494) n (%)	All DAYBREAK Ozanimod 0.92 mg (N = 2494) IR/1000 PY[b] (95% CI)
Any TEAE	647 (87.9)	750 (85.5)	746 (84.7)	2143 (85.9)	693.4 (664.6-723.4)
Severe TEAEs	68 (9.2)	69 (7.9)	53 (6.0)	190 (7.6)	20.7 (18.0-23.9)
Serious TEAEs	84 (11.4)	111 (12.7)	103 (11.7)	298 (11.9)	33.3 (29.8-37.4)
TEAEs leading to permanent treatment discontinuation	27 (3.7)	24 (2.7)	24 (2.7)	75 (3.0)	7.9 (6.3-9.9)
Individual TEAEs in ≥ 5% of overall DAYBREAK population					
Nasopharyngitis	150 (20.4)	169 (19.3)	170 (19.3)	489 (19.6)	59.3 (54.2-64.8)
Headache	124 (16.8)	132 (15.1)	137 (15.6)	393 (15.8)	46.1 (41.8-50.9)
Upper RTI	88 (12.0)	94 (10.7)	96 (10.9)	278 (11.1)	31.5 (28.0-35.5)
Lymphopenia[c]	84 (11.4)	96 (10.9)	77 (8.7)	257 (10.3)	29.4 (26.0-33.3)
ALC decreased[c]	62 (8.4)	78 (8.9)	78 (8.9)	218 (8.7)	24.5 (21.5-28.0)
Back pain	60 (8.2)	74 (8.4)	71 (8.1)	205 (8.2)	22.7 (19.8-26.0)
Hypertension[d]	65 (8.8)	74 (8.4)	48 (5.4)	187 (7.5)	20.7 (17.9-23.9)
GGT increased	65 (8.8)	54 (6.2)	52 (5.9)	171 (6.9)	18.9 (16.2-21.9)
Bronchitis	35 (4.8)	59 (6.7)	51 (5.8)	145 (5.8)	15.8 (13.4-18.6)
RTI	44 (6.0)	53 (6.0)	48 (5.4)	145 (5.8)	15.9 (13.5-18.7)
UTI	38 (5.2)	54 (6.2)	51 (5.8)	143 (5.7)	15.6 (13.3-18.4)
Viral RTI	39 (5.3)	49 (5.6)	44 (5.0)	132 (5.3)	14.4 (12.1-17.0)
Depression-related TEAEs[e]	38 (5.2)	48 (5.5)	43 (4.9)	129 (5.2)	14.0 (11.8-16.6)

[a]TEAE incidence was summarized by Medical Dictionary for Regulatory Activities version 22.1 preferred term. [b]IRs per 1000 PY were calculated as follows to adjust for time on study: number of persons/person-years × 1000 for specific system organ class category or preferred term subcategory. Person-years for each category/subcategory: for a person in a particular category/subcategory, the time on study is calculated based on the date the person first has a TEAE within the category/subcategory (date of first TEAE - first dose date of study drug + 1)/365.25; for persons who do not have a TEAE in the category/subcategory, the time on study is the study duration (last date on study - first dose date of study drug +1)/365.25. [c]ALC reductions are an expected pharmacodynamic effect related to the mechanism of ozanimod; although investigators were not required to report ALC reductions as TEAEs, preferred terms of lymphopenia and ALC decreases may have been reported as TEAEs according to investigator determination. [d]Includes preferred terms of hypertension, essential hypertension, labile hypertension, systolic hypertension, and hypertensive crisis. [e]Includes preferred terms of depression, depressed mood, and depressive symptoms. ALC, absolute lymphocyte count; IR, incidence rate; PY, person-years; RTI, respiratory tract infection; UTI, urinary tract infection.

Adverse events of special interest

- 56.7% of DAYBREAK participants experienced an infection (IR 243.5/1000 PY [95% CI 231.1-256.5]), most commonly respiratory and urinary tract infections; 2.8% experienced a serious infection (IR 7.4/1000 PY [95% CI 5.8-9.3])
 - 5.6% of patients experienced opportunistic infections (IR 15.2/1000 PY [95% CI 12.9-18.0]), most commonly oral herpes (2.0%) and herpes zoster infections (1.7%); none of the herpes infections were serious or disseminated
 - There were no serious opportunistic infections at the time of the data cut; however, a case of progressive multifocal leukoencephalopathy was reported in March 2021 in a 46-year-old woman who had received ozanimod 0.92 mg for about 4 years; she discontinued ozanimod, and as of November 2021, remains stable and under investigator's care
- During the parent trials and DAYBREAK, 38 (1.4%; IR 358.7/100,000 PY [95% CI 253.9-492.4]) of 2786 patients exposed to either dose of ozanimod developed treatment-emergent malignancies
 - This included 1 malignant melanoma (0.04%; IR 9.4/100,000 PY [95% CI 0.2-52.4]), 13 nonmelanoma skin cancers (0.5%; IR 122.6/100,000 PY [95% CI 65.3-209.6]), and 24 (0.9%; IR 226.0/100,000 PY [95% CI 144.8-336.2]) noncutaneous malignancies
- During DAYBREAK, there were 9 (0.4%) TEAEs of macular edema
 - Based on review of TEAEs and optical coherence tomography images, an independent Macular Edema Review Panel confirmed 4 macular edema cases (0.2%; IR 0.4/1000 PY [95% CI 0.2-1.1]) during DAYBREAK; 3 cases were resolved and 1 was ongoing at data cutoff
- Cardiac TEAEs occurred in 69 (2.8%; IR 7.4/1000 PY [95% CI 5.8-9.3]) DAYBREAK participants, 7 of whom had serious events (3 myocardial infarctions, and 1 each of myocardial ischemia, unstable angina, bradycardia, and coronary artery stenosis)
- In the overall DAYBREAK population, ALC (based on single assessments) was < 0.2×10^9/L in 244 (9.8%) of 2488 patients
 - 30/187 patients (16.0%) who had a repeat ALC assessment within 30 days (which is 1.2% [30/2488] of those in the overall safety population who had ≥ 1 ALC test in DAYBREAK) had a consecutive ALC < 0.2×10^9/L on retest, suggesting most instances were transient

Conclusions

- In this interim analysis of the DAYBREAK OLE, ozanimod treatment demonstrated sustained control of disease activity in patients with RMS
- Clinical and radiologic disease activity remained low in patients who received continuous ozanimod 0.92 mg, and patients who switched to ozanimod 0.92 mg from either IFN β-1a or ozanimod 0.46 mg had numerically lower ARR and decreases in brain MRI lesion counts after the switch
- Ozanimod was generally well tolerated; overall safety and tolerability of ozanimod was consistent with the phase 3 trials,[6,7] with 82.4% retention at data cutoff
- Most patients with COVID-19 had nonserious infections and recovered without discontinuing ozanimod

References

1. Scott FL, et al. *Br J Pharmacol* 2016;173:1778-1792.
2. ZEPOSIA® (ozanimod) [package insert]. Princeton, NJ: Bristol Myers Squibb; December 2021.
3. ZEPOSIA® (ozanimod) [summary of product characteristics]. Utrecht, Netherlands, Celgene Distribution BV; December 2021.
4. Cohen JA, et al. *Lancet Neurol* 2016;15:373-381.
5. Cohen JA, et al. *Mult Scler* 2019;25:1255-1262.
6. Cohen JA, et al. *Lancet Neurol* 2019;18:1021-1033.
7. Comi G, et al. *Lancet Neurol* 2019;18:1009-1020.
8. Selmaj KW, et al. *Mult Scler Relat Disord* 2021:51. doi:10.1016/j.msard.2021.
9. ICH Expert Working Group. https://database.ich.org/sites/default/files/E2A_Guideline.pdf. Accessed January 24, 2022.

Acknowledgments

- The patients and families who made this study possible
- The clinical study teams who participated
- The DAYBREAK study was supported by Celgene International II
- All authors contributed to and approved the poster; writing and editorial assistance was provided by Lauren A. Cerruto and Jessica D. Herr, PharmD, of Peloton Advantage, LLC, an OPEN Health company, and funded by Bristol Myers Squibb

Disclosures

BAC: Personal compensation for consulting for Alexion, Atara, Autobahn, Avotres, Biogen, EMD Serono, Gossamer Bio, Horizon, Neuron23, Novartis, Sanofi, TG Therapeutics, and Therini, and received grant support from Genentech. **KWS:** Consulting for Biogen, Celgene, Genzyme, Merck, Novartis, Ono Pharma, Roche, Synthon, and Teva. **LS:** Consulting for AbbVie, Atreca, Celgene, Novartis, Teva, Tolerion, and EMD Serono, and research support from Atara, Biogen, and Celgene. **GC:** Compensation for consulting and/or speaking activities from Almirall, Biogen, Celgene, EXCEMED, Forward Pharma, Genzyme, Merck, Novartis, Roche, Sanofi, and Teva. **ABO:** Participated as a speaker in meetings sponsored by and received consulting fees and/or grant support from Atara Biotherapeutics, Biogen, BMS-Celgene, EMD Serono, Sanofi Genzyme, Novartis, and Roche-Genentech. **DLA:** Consulting fees from Biogen, Celgene, Frequency Therapeutics, Genentech, Merck, Novartis, Race to Erase MS, Roche, and Sanofi-Aventis, Shionogi, and Xfacto Communications, grants from Immunotec and Novartis, and an equity interest in NeuroRx. **HPH:** Personal fees for consulting, serving on steering committees, and speaking from Bayer Healthcare, Biogen, Celgene, GeNeuro, Genzyme, Merck, MedImmune, Novartis, Octapharma, Roche, Sanofi, and Teva. **XM:** Speaking honoraria and travel expenses for participation in scientific meetings and steering committee member of clinical trials or participated in advisory boards of clinical trials in the past 3 years with Actelion, Alexion, Bayer, Biogen, Bristol Myers Squibb/Celgene, EMD Serono, EXCEMED, Genzyme, Hoffmann-La Roche, Immunic, Janssen Pharmaceuticals, MedDay, Merck, Mylan, MSIF, Nervgen, NMSS, Novartis, Roche, Sanofi-Genzyme, Teva Pharmaceuticals, and TG Therapeutics. **EKH:** personal compensation for consulting and speaking for Actelion, Biogen, Celgene, Merck, Novartis, Roche, Sanofi, and Teva. **JKS, NM, CYC, DS:** Employees and shareholders of Bristol Myers Squibb. **LW:** Institutional research support: steering committee, advisory board, consultancy fees: Actelion, Bayer HealthCare, Biogen, Bristol Myers Squibb, Genzyme, Janssen, Japan Tobacco, Merck, Novartis, Roche, Sanofi, Santhera, Shionogi, and TG Therapeutics; speaker fees: Bayer HealthCare, Biogen, Merck, Novartis, Roche, and Sanofi; support of educational activities: Allergan, Bayer HealthCare, Biogen, CSL Behring, Desitin, Genzyme, Merck, Novartis, Roche, Pfizer, Sanofi, Shire, and Teva; license fees for Neurostatus products; and grants: Bayer HealthCare, Biogen, European Union, Innosuisse, Merck, Novartis, Roche, Swiss MS Society, and Swiss National Research Foundation. **JAC:** Personal compensation for consulting for Biogen, Bristol Myers Squibb, Convelo, Genentech, Janssen, NervGen, Novartis, and PSI; speaking for H3 Communications; and serving as an Editor of *Multiple Sclerosis Journal*.

Presented at the 2022 Annual Meeting of the Consortium of Multiple Sclerosis Centers; June 1-4, 2022; National Harbor, Maryland
Previously presented at ACTRIMS Forum 2022; February 24-26, 2022; West Palm Beach, Florida

Email: bruce.cree@ucsf.edu

This poster may not be reproduced without written permission from the authors.