Presented at CMSC Annual Meeting 2022 • June 1-4, 2022 • National Harbor, MD

DMT51

Identifying signs of progression in people diagnosed with multiple sclerosis using administrative health care claims data

Regina Berkovich,[1] Wing Chow,[2] Qiujun (Samantha) Shao,[2] Magdaliz Gorritz,[3] Zifan Zhou,[3] Hsiu-Ching Chang,[3] Rolin L. Wade,[3]

[1]Los Angeles County USC General Hospital and Clinic; [2]Novartis Pharmaceuticals Corporation; [3]IQVIA, Inc.

Disclosures

RB received honoraria for speaking engagements, serving at scientific advisory boards and consulting activities from: Alexion, Bayer, Biogen, Celgene, Genentech, Mallinckrodt, Merck-Serono, Novartis, Sanofi.

WC and QS are employees of Novartis

MG, ZZ, HC and RW are employees of IQVIA

SUMMARY

1 **A claims-based methodology was developed and has shown to be predictive of MS progression in the absence of distinct clinical markers of progression in claims data.**

2 **49.7% of MS patients were identified by the claims-based algorithm to have signs for MS progression.**

3 **Significant indicators of MS progression included older age at onset, being female, duration of disease, comorbid conditions, higher disability level, and pre-index use of medications to treat MS-related conditions.**

Text : Qc5473
To : 8NOVA (86682) US Only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe & Russia
+46737494608 Sweden, Europe

INTRODUCTION

- Relapsing-remitting MS (RRMS) is the most common form of MS, which accounts for ~85% of MS patients at onset. The majority of patients with RRMS convert to secondary progressive MS (SPMS) over time.[1,2]
- With the advent of disease modifying therapies (DMTs) for progressive forms of MS, identification of patients with progressive disease is a critical step in real-world effectiveness assessments, yet data are limited as most studies apply relapse proxies in administrative claims data, which lack distinct indicators for progression.[3-12]

OBJECTIVE

- The study aimed to develop a claims-based algorithm using a U.S.-based administrative claims database to identify and characterize patients with signs of MS progression

METHODS

STUDY DESIGN

- A retrospective cohort analysis was performed using the IQVIA PharMetrics® Plus database with the study time period that spanned from January 1, 2012-June 30, 2020.
- Patients with ≥2 outpatient or one inpatient claim with a diagnosis of MS (ICD-9 CM code 340 or ICD-10 CM code G35) between July 1, 2012 and December 31, 2019 were included in the study. The first MS diagnosis = cohort entry date.
- Patients were required to have ≥6 months of continuous enrollment in a health plan with medical and pharmacy benefits prior to and after the cohort entry date and were ≥18 years of age on the date of cohort entry.
- MS disability level was assessed using a published claims-based disability score based on evidence of EDSS-related symptoms, durable medical equipment (DME) use, and incontinence-related surgical procedures that occurred outside of relapses during each 6-month time interval from 6 months prior to cohort entry until the end of follow-up.[13]

Figure 1. Examples of progressive and non-progressive MS

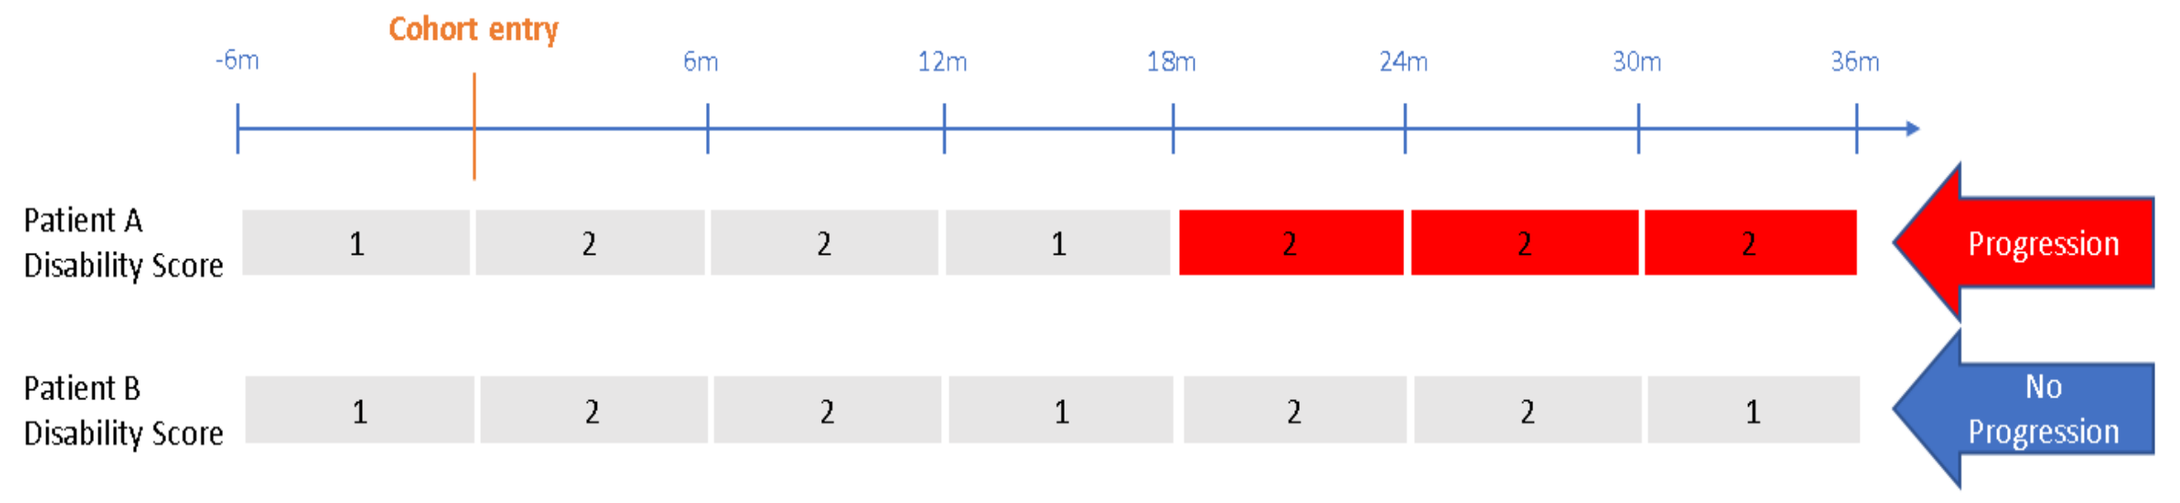

Cohort entry
-6m
6m
12m
18m
24m
30m
36m
Patient A Disability Score
1 2 2 1 2 2 2
Progression
Patient B Disability Score
1 2 2 1 2 2 1
No Progression

Note: Disability score 1=Mild, 2=Moderate, 3=Severe

MS PROGRESSION ALGORITHM

- Disability level was reset at the start of each 6-month interval. Based on a neurologist expert opinion, certain symptoms and DMEs were considered irreversible (incontinence, spasm, muscle weakness, cognitive impairment, dysphagia, partial/full paralysis, visual function, muscle contracture, nystagmus, wheelchair dependence, constant catheterization, bed confinement, bowel incontinence).
- Progressive disease was defined as:
 - Having evidence of an irreversible symptom or DME; or
 - Having an increase in MS disability level that was maintained for ≥3 consecutive 6-month time intervals.
- The index date for patients with progressive disease was defined as the date of the first claim that indicated progression.
- Patients with probable progression were identified. The index date was defined as the date of the first sign of progression. Patients who did not meet the criteria for progressive disease were assigned a pseudo index date based on the distribution of time from cohort entry to progression in the progressed patients.

STATISTICAL ANALYSIS

- A logistic regression model was developed to identify independent indicators of disease progression. Independent variables included all pre-index variables that had a standardized difference of ≥10% between progression and non-progressed patients, and variables that deemed clinically relevant.

RESULTS

- A total of 81,647 patients were included in the analysis, among which, 40,610 (49.7%) and 41,037 (50.3%) patients were classified as progressed and non-progressive, respectively.

UNADJUSTED DEMOGRAPHIC and CLINICAL CHARACTERISTICS

- Over 80% of progressed patients entered the study prior to 2015 compared to 75.2% of non-progressed patients possibly indicating that patients with progressive disease had MS for a longer period.
- Patients with progressive disease had a higher comorbidity burden (mean CCI: 0.7 [1.3] vs. 0.3 [0.8]; p<0.0001)
- More progressed patients had ≥ one relapse in the 6-month pre-index period and the mean (SD) number of relapses was 1.3 (0.7) and 1.2 (0.6) for progressed and non-progressive patients (Table 1).
- More progressed patients had moderate to severe disability level at baseline compared to non-progressive patients (Table 1).

Table 1. Unadjusted demographic and clinical characteristics by progression status

Demographic and Clinical Characteristics*	Progressed N=40,610	Non-progressive N=41,037	Std diff**
Age at cohort entry, mean (SD)	50 (11.7)	46 (11.6)	-0.3192
Age at index, mean (SD)	51 (11.7)	47 (11.7)	-0.3513
Female (n, %)	31,050 (76.5%)	30,403 (74.1%)	0.055
Time (days) from cohort entry to index date, mean (SD)	993 (759)	707 (612)	-0.416
Comorbidities (n, %)			
Chronic pain/fibromyalgia	5,585 (13.8%)	2,668 (6.5%)	-0.2421
Depression	7,397 (18.2%)	4,602 (11.2%)	-0.1099
Dyslipidemia	9,317 (22.9%)	6,940 (16.9%)	-0.1015
Hypertension	11,112 (27.4%)	7,459 (18.2%)	-0.1300
Osteoarthritis	16,875 (41.6%)	11,333 (27.6%)	-0.1986
Sleep disorders	4,723 (11.6%)	3,027 (7.4%)	-0.1482
Thyroid disease	4,649 (11.5%)	3,432 (8.4%)	-0.1514
UTI	9,571 (23.6%)	5,061 (12.3%)	-0.2959
No Prior DMT used (n,%):	21,014 (51.8%)	20,224 (49.3%)	-0.0493
Patients with MS relapses[1] (n,%):	6,945 (17.1%)	5,363 (13.1%)	-0.1129
Baseline disability level including index date[2] (n,%):			
No EDSS-related symptoms or DME use	0 (0.00%)	30,052 (73.2%)	2.8584
Mild	2,465 (6.07%)	4,983 (12.1%)	
Moderate	25,840 (63.6%)	5,236 (12.8%)	
Severe	12,305 (30.3%)	766 (1.9%)	

*Measured during the 6-month pre-index period (excluding index date). ** Std diff: standard difference
1 Patients were flagged as having a relapse if 1) An inpatient hospitalization with a primary diagnosis of MS (ICD-9 code of 340 or ICD-10 code of G35) OR 2) An oral or intravenous corticosteroid use within 7-days of an MS-related outpatient visit. Corticosteroid use on the same day as infused DMTs was be counted as relapse events
2 Mild – only 1 functional system with symptoms, and all symptoms must be level 1; Moderate – any functional system with symptoms at level 2 OR 2 or more functional systems with symptoms and all symptoms are level 1 OR any level 2 DME use (i.e., cane/crutch/etc., intermittent catheter codes); Severe – any functional system with symptoms at level 3 OR any level 3 DME use OR surgical procedure for incontinence None – no EDSS-related symptoms, DME use, or surgical procedures for incontinence

LOGISTIC REGRESSION RESULTS

- The strongest indicator of MS progression was baseline disability level. Patients with moderate or severe MS disability at baseline had the highest odds of progressive disease (Figure 2).
- Patients who used medications to treat MS-related symptoms and conditions, had higher odds of having progressive disease (Figure 2).

VALIDATION USING ELECTRONIC MEDICAL RECORDS DATA

- A total of 1,671 patients linked to IQVIA's Ambulatory Electronic Medical Records database (aEMR), 855 of whom were classified as progressed in PharMetrics Plus.
- Twenty-four patients in the linked dataset had evidence of progressive MS in the aEMR based on a text search of the problem list.
 - Progressive MS was defined in the aEMR as the observance of the following in the problem list: progression, progressed, progressive, SPMS or secondary progressive
- Of the 24 patients with evidence of progressive MS in the aEMR, 23 (95.8%) were flagged as having progressive disease in claims using the claims-based algorithm
- On average, time from cohort entry to progression in the aEMR was 573.7 and only 231.0 days in claims using the claims-based algorithm (Table 2).
- Overall, the algorithm had a sensitivity (i.e., true positive rate) of 95.8% and specificity (i.e., true negative rate) of 49.5%

Table 2. Progression and time to progression in the aEMR versus PharMetrics Plus (n=1,671)

	aEMR	PharMetrics Plus
Patients with evidence of progressive MS in the aEMR	24 (1.4%)	
Patients with probably progression in PharMetrics Plus		855 (51.2%)
Patients with evidence of progression in aEMR and PharMetrics Plus	23 (1.4%)	23 (1.4%)
Time from cohort entry to progression, days (mean, SD); median	573.7 (715.5); 324	231.0 (452.5); 54

aEMR: ambulatory EMR; MS: multiple sclerosis; SD: standard deviation

Figure 2. Independent indicators of disease progression

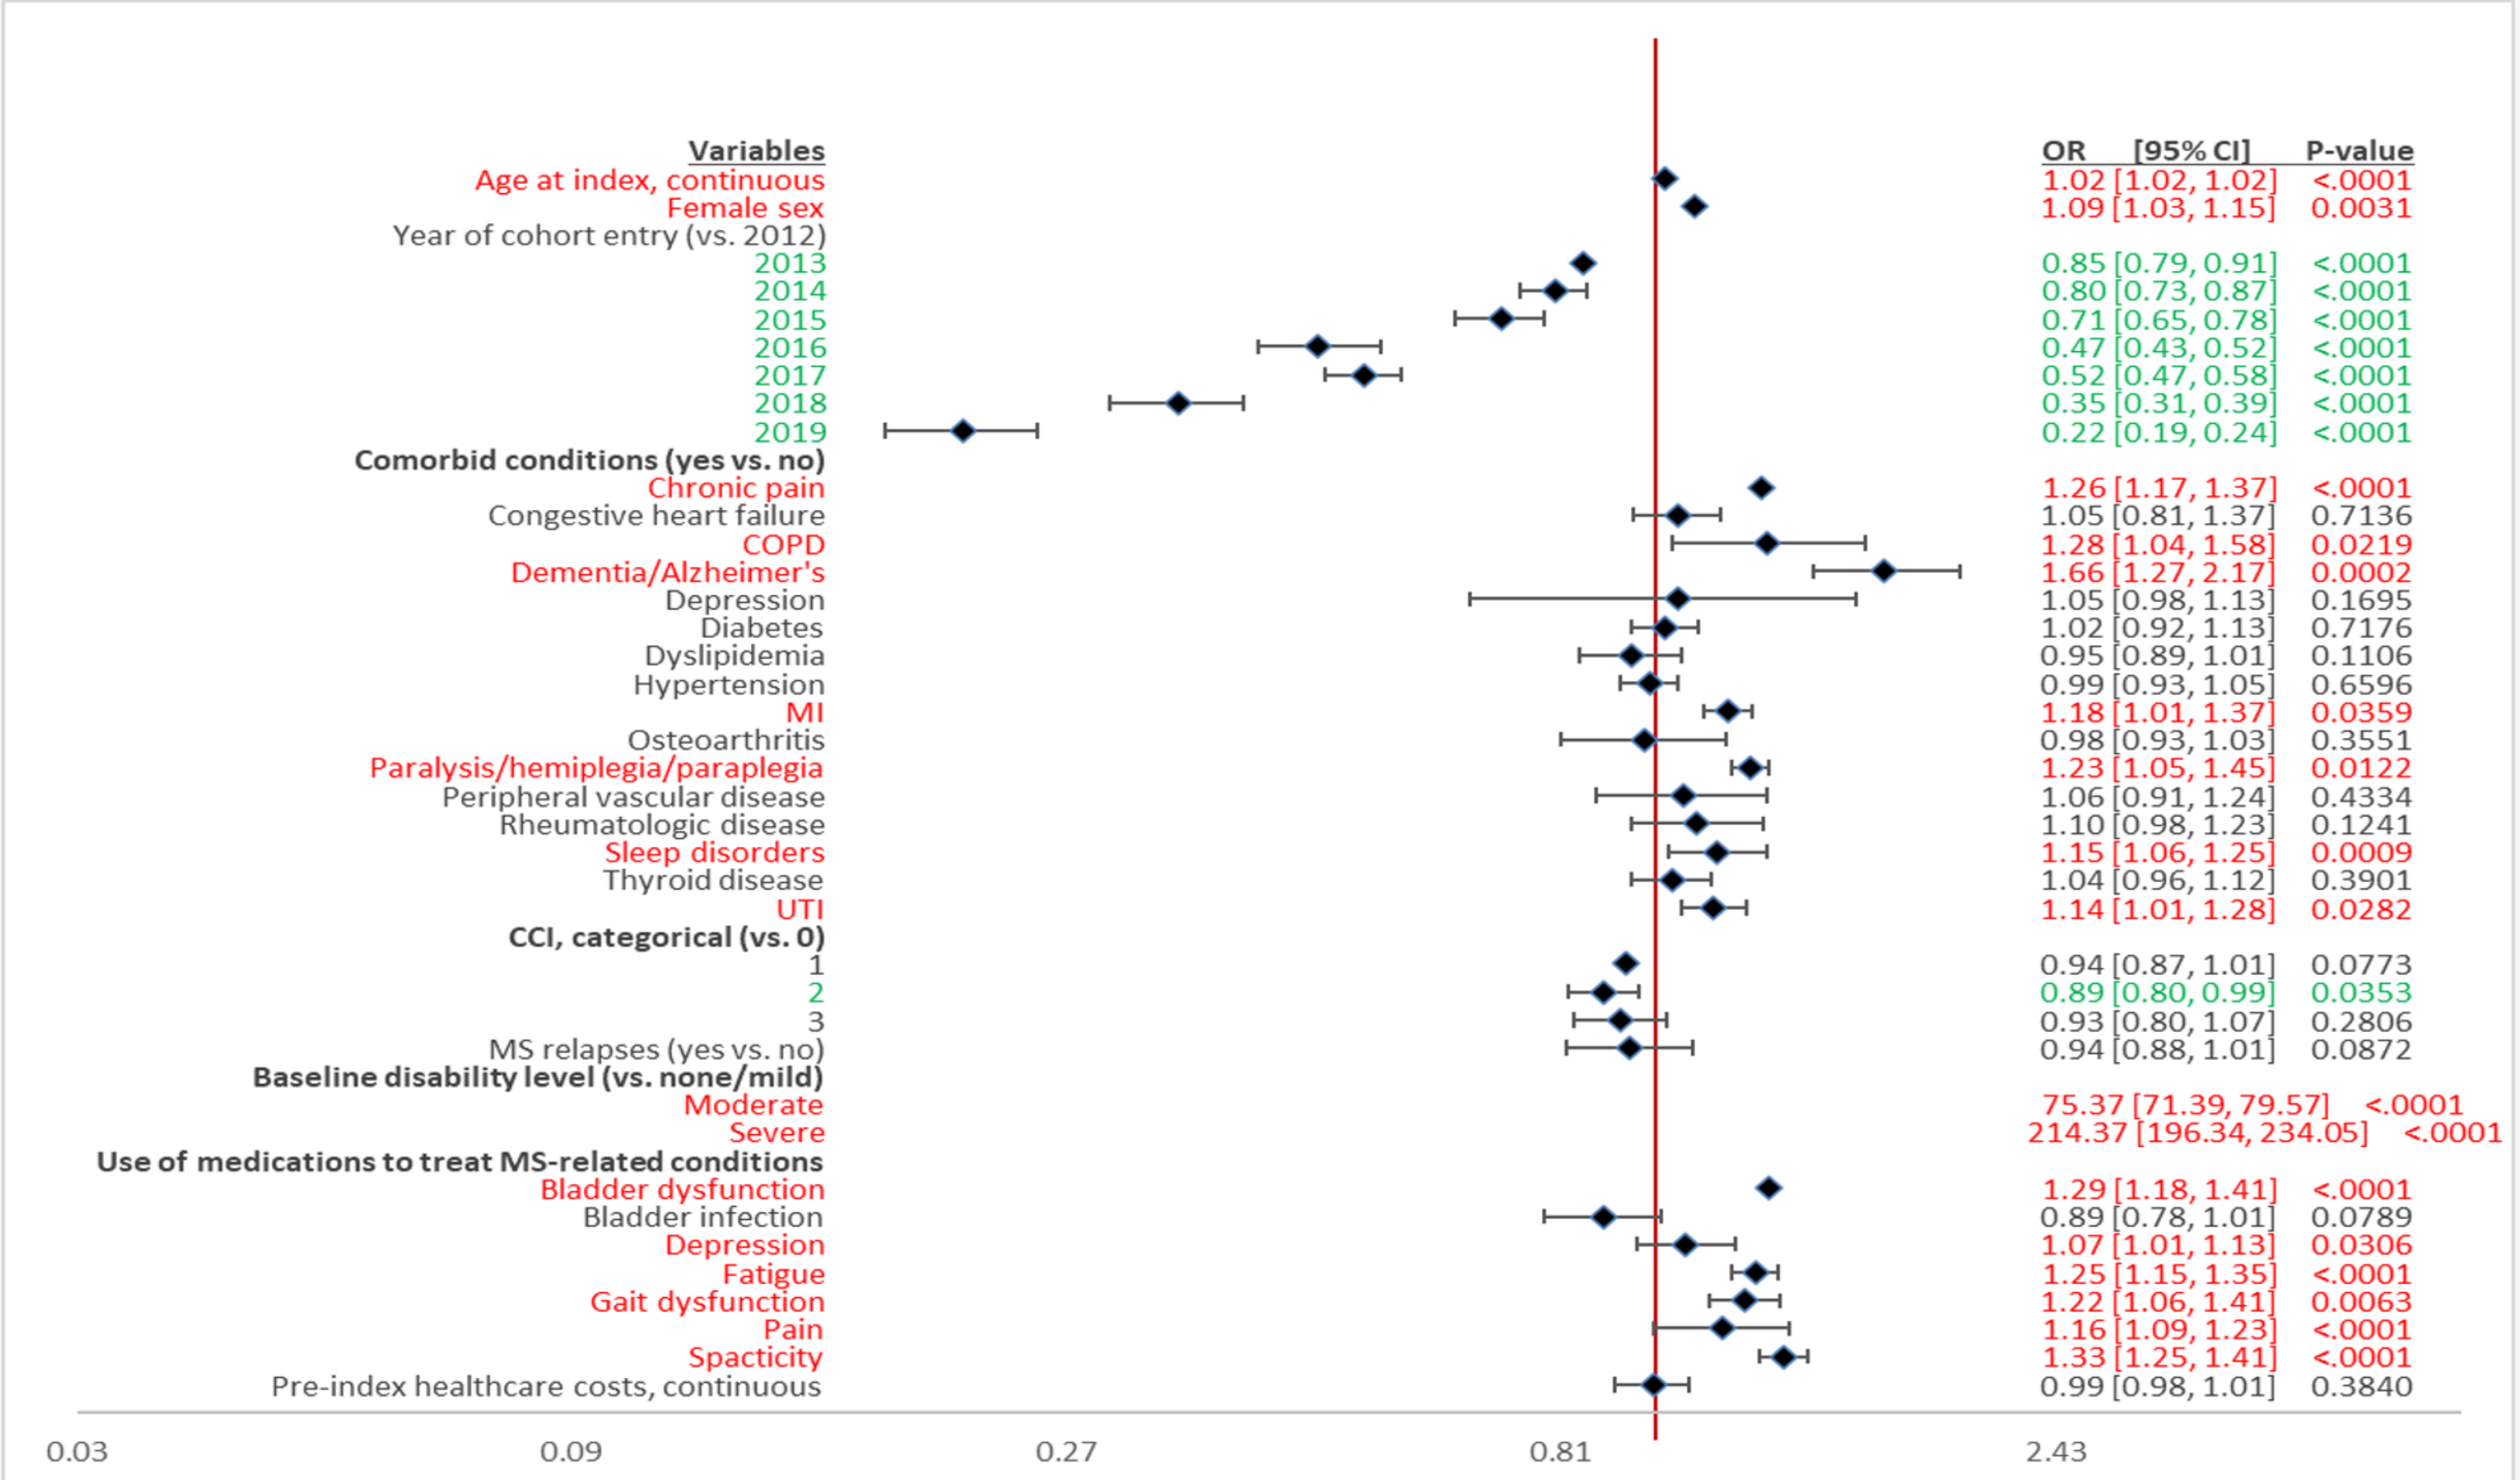

Variables
OR [95% CI] P-value
Age at index, continuous 1.02 [1.02, 1.02] <.0001
Female sex 1.09 [1.03, 1.15] 0.0031
Year of cohort entry (vs. 2012)
2013 0.85 [0.79, 0.91] <.0001
2014 0.80 [0.73, 0.87] <.0001
2015 0.71 [0.65, 0.78] <.0001
2016 0.47 [0.43, 0.52] <.0001
2017 0.52 [0.47, 0.58] <.0001
2018 0.35 [0.31, 0.39] <.0001
2019 0.22 [0.19, 0.24] <.0001
Comorbid conditions (yes vs. no)
Chronic pain 1.26 [1.17, 1.37] <.0001
Congestive heart failure 1.05 [0.81, 1.37] 0.7136
COPD 1.28 [1.04, 1.58] 0.0219
Dementia/Alzheimer's 1.66 [1.27, 2.17] 0.0002
Depression 1.05 [0.98, 1.13] 0.1695
Diabetes 1.02 [0.92, 1.13] 0.7176
Dyslipidemia 0.95 [0.89, 1.01] 0.1106
Hypertension 0.99 [0.93, 1.05] 0.6596
MI 1.18 [1.01, 1.37] 0.0359
Osteoarthritis 0.98 [0.93, 1.03] 0.3551
Paralysis/hemiplegia/paraplegia 1.23 [1.05, 1.45] 0.0122
Peripheral vascular disease 1.06 [0.91, 1.24] 0.4334
Rheumatologic disease 1.10 [0.98, 1.23] 0.1241
Sleep disorders 1.15 [1.06, 1.25] 0.0009
Thyroid disease 1.04 [0.96, 1.12] 0.3901
UTI 1.14 [1.01, 1.28] 0.0282
CCI, categorical (vs. 0)
1 0.94 [0.87, 1.01] 0.0773
2 0.89 [0.80, 0.99] 0.0353
3 0.93 [0.80, 1.07] 0.2806
MS relapses (yes vs. no) 0.94 [0.88, 1.01] 0.0872
Baseline disability level (vs. none/mild)
Moderate 75.37 [71.39, 79.57] <.0001
Severe 214.37 [196.34, 234.05] <.0001
Use of medications to treat MS-related conditions
Bladder dysfunction 1.29 [1.18, 1.41] <.0001
Bladder infection 0.89 [0.78, 1.01] 0.0789
Depression 1.07 [1.01, 1.13] 0.0306
Fatigue 1.25 [1.15, 1.35] <.0001
Gait dysfunction 1.22 [1.06, 1.41] 0.0063
Pain 1.16 [1.09, 1.23] <.0001
Spacticity 1.33 [1.25, 1.41] <.0001
Pre-index healthcare costs, continuous 0.99 [0.98, 1.01] 0.3840
0.03
0.09
0.27
0.81
2.43

- Significant variables associated with increased risk of progressive disease in red; variables associated with a decreased risk of progressive disease in green
- CCI: Charlson comorbidity index; CI: confidence interval; MI: myocardial infarction; MS: multiple sclerosis; OR: odds ratio; UTI: urinary tract infection

LIMITATIONS

- Disability level was estimated using proxies for ambulation/functional status in claims, potentially overestimating MS-related DME use, although clinically DME use unrelated to MS is very rare in MS patients.
- Disease progression is a gradual process taking place over years to decades, rather than at a single episode or point in time. Therefore, follow-up periods provided by administrative claims data may not be adequate to evaluate the development of progressive disease reliably.
- The IQVIA PharMetrics Plus is sourced from commercial insurance plans including Medicare Advantage and Part D plans, and as such do not represent patients using fee-for-service Medicare and Medicaid.
- The validation was small with only 24 patients. Furthermore, SPMS may be underreported in the EMR, which is not a gold standard to test against. Future validation of this claims-based progression algorithm is needed.

CONCLUSIONS

– This novel real-world study suggests that patients with higher pre-index disability level and comorbid conditions may be at higher risk of progression. MS relapse did not differentiate between progression and non-progression. Future efforts should explore the impact of pharmacologic interventions to optimize outcomes in MS patients with indicators suggestive of progression risk.

ACKNOWLEDGEMENTS: This study was funded by Novartis Pharmaceuticals Corporation. Programming support was provided by Guangyu Hu of IQVIA and was funded by Novartis Pharmaceuticals Corporation. This poster was developed in accordance with Good Publication Practice (GPP3) guidelines. Authors had full control of the content and made the final decision on all aspects of this poster.

REFERENCES: **1.** National Multiple Sclerosis Society. **2.** Hempel S, et al. **3.** Bergvall N, et al. **4.** Bergvall N, et al. **5.** Bergvall N, et al. **6.** Capkun G, et al. **7.** Castelli-Haley J, et al.. **8.** Halpern R, et al. **9.** Ivanova JI, et al. **10.** Johnson BH, et al. **11.** Kozma CM, et al. **12.** Tan H, et al. **13.** Berkovich R.et al.