DMT46

Real-world Efficacy and Safety After 5 Years of Dimethyl Fumarate Treatment in Black/African American and Hispanic/Latino American Patients With Multiple Sclerosis in ESTEEM

Williams MJ,[1] Amezcua L,[2] Chinea A,[3] Cohan S,[4] Okai A,[5] Okuda DT,[6] Vargas W,[7] Belviso N,[8] Bozin I,[8] Jiang X,[8] Lewin JB,[8] Lyons J,[8] Shen C,[8] England SM,[8] Grimes N[8]

[1]Joi Life Wellness MS Center, Atlanta, GA, USA; [2]University of Southern California, Keck School of Medicine, Los Angeles, CA, USA; [3]San Juan MS Center, Guaynabo, Puerto Rico; [4]Providence Portland Medical Center, Portland, OR, USA; [5]North Texas Institute of Neurology and Headache, Plano, TX, USA; [6]Neuroinnovation Program, UT Southwestern Medical Center, Dallas, TX, USA; [7]Columbia University Medical Center, New York City, NY, USA; [8]Biogen, Cambridge, MA, USA

OBJECTIVE

- To evaluate real-world effectiveness and safety of DMF in Black/AA, non-Black/non-AA, Hispanic/LA, and non-Hispanic/non-LA patients with RRMS in ESTEEM (NCT02047097).

CONCLUSIONS

- Compared with the 12 months before DMF initiation, ARR was significantly lower up to 5 years after DMF initiation in Black/AA and Hispanic/LA patients.
- The safety profile of DMF in these subgroups was consistent with the overall ESTEEM population.
- GI disorders were the most common reason for treatment discontinuation in all subgroups.
- These data demonstrate real-world treatment benefit of DMF in Black/AA and Hispanic/LA patients. Future studies will strive to increase the relative proportion of racial and ethnic minorities in clinical trials.

Consortium of Multiple Sclerosis Centers 2022 Annual Meeting
June 1-4, 2022 | National Harbor, MD

Introduction

- Delayed-release dimethyl fumarate (DMF; also known as gastro-resistant DMF) has demonstrated efficacy and a stable benefit-risk profile in studies of patients with relapsing-remitting multiple sclerosis (RRMS).[1,2]
- ESTEEM (NCT02047097) is an ongoing, Phase 4, 5-year, multinational, prospective, observational study characterizing long-term effectiveness and safety of DMF in real-world clinical practice.
- Evidence suggests that clinical course and disability outcomes associated with multiple sclerosis (MS) may vary according to ethnicity and race.[3,4]
- A low percentage of racial and ethnic minorities are included in clinical trials.[5]
 - Black/African Americans (AA) comprise 13.4% of the US population but only 5% of the clinical trial participants.
 - Hispanic/Latino Americans (LA) comprise 18.1% of the US population but only 1% of the clinical trial participants.[5]
- DMF was efficacious in a small sample of Black/AA and Hispanic/LA patients in DEFINE/CONFIRM (Phase 3)[6] and a retrospective chart review.[7]
- A previous analysis of these subgroups in ESTEEM demonstrated effectiveness during 3 years of DMF treatment;[8,9] however, data describing the long-term effects of DMF in these subgroups in the real-world setting are limited.

Methods

Study Design and Patients

- ESTEEM included patients newly prescribed DMF in routine clinical practice at ~390 sites globally.
- Key eligibility criteria:
 - Diagnosis of MS
 - Age ≥ 12 years (patients aged 12 to < 18 years excluded where enrollment of pediatric patients is prohibited)
 - Newly prescribed DMF under routine clinical care.
- The primary objective of ESTEEM was to determine the incidence, type, and pattern of serious adverse events, including serious infections, and adverse events (AEs) leading to treatment discontinuation in patients with MS treated with DMF.
- A secondary objective was to assess the effectiveness of DMF on MS relapse activity.
- In this analysis, the safety and effectiveness of DMF were evaluated in a post hoc subgroup analysis in Black/AA, non-Black/non-AA, Hispanic/LA, and non-Hispanic/non-LA patients. Non-Black/non-AA patients were all patients who did not identify as Black/AA; non-Hispanic/non-LA patients encompasses all patients who did not identify as Hispanic/LA.

Endpoints

- The effectiveness of DMF on MS relapses was evaluated by assessment of annualized relapse rate (ARR), proportion of patients without relapse, and distribution of the number of relapses.
- The incidence of treatment discontinuation was also assessed.

Analysis

- ARRs were obtained by fitting a repeated-measure negative binomial model (ARR = total number of relapses during the study for all patients / total number of patient-years followed in the study).
- Descriptive summary statistics were calculated. For absolute lymphocyte count (ALC), median values were deemed more appropriate due to the outliers in this real-world observational study.

Results

Study Population

- Overall, 220 (4.2%) Black/AA, 5031 non-Black/non-AA, 105 (2.0%) Hispanic/LA, and 5146 non-Hispanic/non-LA patients received ≥ 1 dose of DMF and were included in the analysis, with follow-up over 60 months.
- Baseline characteristics are shown in Table 1.

ARR and Relapses

- Unadjusted ARRs up to 5 years were: Black/AA, 0.054 (95% CI, 0.038–0.078); non-Black/non-AA, 0.077 (95% CI, 0.072–0.081); Hispanic/LA, 0.069 (95% CI, 0.043–0.112), and non-Hispanic/non-LA, 0.076 (95% CI, 0.072–0.081), representing reductions ranging from 91% to 92%, compared with ARR 12 months prior to study entry ($p < 0.0001$ for all subgroups; Figure 1).
- The estimated proportion of patients without relapse at 5 years ranged from ~80% to 85% (Table 2).

Safety and Discontinuations

- An AE leading to treatment discontinuation occurred in 39 (18%) Black/AA patients and 29 (28%) Hispanic/LA patients (Table 3).
- Gastrointestinal (GI) disorders were the most common reason for discontinuation in both subgroups.

Absolute Lymphocyte Counts

- Median lymphocyte counts decreased over time, and percentage decrease from baseline to Months 12–60 are shown in Figure 2A and B and Table 4.
- In the first year, median lymphocyte counts declined 24% in Black/AA, 36% in non-Black/non-AA, 34% in Hispanic/LA, and 35% in non-Hispanic/non-LA patients, and then remained stable.

Table 1. Patient Baseline Characteristics[a,b]

Characteristic	Black/AA n = 220	Non-Black/ Non-AA n = 5031	Hispanic/LA n = 105	Non-Hispanic/ Non-LA n = 5146
Age category, y, n (%)				
< 40	89 (40)	2555 (51)	56 (53)	2588 (50)
≥ 40	131 (60)	2475 (49)	49 (47)	2557 (50)
Female, n (%)	184 (84)	3703 (74)	83 (79)	3804 (74)
Region, n (%)				
United States, including Puerto Rico	214 (97)	1302 (26)	103 (98)	1413 (27)
Western Europe, Canada, New Zealand, and Australia	6 (3)	2842 (56)	1 (1)	2847 (55)
Eastern Europe	0 (0)	887 (18)	1 (1)	886 (17)
Duration between most recent relapse and enrollment, mo, median (range)	6 (0–182)	6 (0–410)	6 (0–410)	6 (0–325)
No. of relapses in prior year, median (range)	1 (0–4)	1 (0–10)	1 (0–4)	1 (0–10)
No. of relapses in prior 2 years, median (range)	1 (0–6)	1 (0–30)	1 (0–10)	1 (0–30)
No. of relapses in prior 3 years, median (range)	1 (0–20)	1 (0–45)	1 (0–13)	1 (0–45)
DMF treatment duration, mo, mean (SD)	26 (20)	31 (20)	25 (20)	31 (20)
Patients with minimum 1-year follow-up, n (%)	101 (53)	2742 (56)	50 (51)	2793 (56)
Patients with minimum 2-year follow-up, n (%)	63 (33)	2004 (41)	32 (33)	2035 (41)
Patients with minimum 3-year follow-up, n (%)	36 (19)	1155 (24)	15 (15)	1176 (24)
Patients with minimum 4-year follow-up, n (%)	14 (7)	410 (8)	5 (5)	419 (8)
Duration in ESTEEM, mo, median (range)	32 (0–72)	41 (0–85)	29 (1–77)	41 (0–85)
EDSS score, mean (SD)[c]	n = 36 2.9 (2.1)	n = 3364 2.1 (1.5)	n = 32 2.3 (2.3)	n = 3368 2.1 (1.5)
Prior MS treatments, n (%)				
Glatiramer acetate	87 (55)	1259 (39)	31 (42)	1315 (40)
IFN beta-1a intramuscular	58 (37)	971 (30)	29 (40)	1000 (30)
IFN beta-1a subcutaneous	40 (25)	937 (29)	15 (21)	962 (29)
IFN beta-1b	25 (16)	606 (19)	17 (23)	614 (19)
Fingolimod	15 (9)	287 (9)	9 (12)	293 (9)
Teriflunomide	7 (4)	217 (7)	2 (3)	222 (7)
Natalizumab	21 (13)	286 (9)	17 (23)	290 (9)

AA = African American; DMF = delayed-release dimethyl fumarate; EDSS = Expanded Disability Status Scale; IFN = interferon; LA = Latino American; MS = multiple sclerosis
[a]Patients could be included in > 1 subgroup.
[b]Percentages are calculated using non-missing data as the denominator.
[c]EDSS assessment at enrollment.

Table 2. Proportion of Patients Without MS Relapses in the 5 Years After DMF Initiation

	Black/AA n = 220	Non-Black/Non-AA n = 5031	Hispanic/LA n = 105	Non-Hispanic/Non-LA n = 5146
Proportion of patients without MS relapse, %[a]	85.2	79.9	84.7	80.0

AA = African American; DMF = delayed-release dimethyl fumarate; LA = Latino American; MS = multiple sclerosis
[a]Kaplan-Meier analyses.

Table 3. Most Common AEs Leading to DMF Discontinuation

Category, n (%)[a]	Black/AA n = 220	Non-Black/ Non-AA n = 5031	Hispanic/LA n = 105	Non-Hispanic/ Non-LA n = 5146
Any AE leading to treatment discontinuation	39 (18)	1126 (22)	29 (28)	1136 (22)
GI disorders	17 (8)	429 (9)	11 (10)	435 (8)
Abdominal pain	1 (0.5)	171 (3)	4 (4)	168 (3)
Diarrhea	7 (3)	120 (2)	3 (3)	124 (2)
Nausea	5 (2)	116 (2)	3 (3)	118 (2)
Vomiting	4 (2)	65 (1)	1 (1)	68 (1)
Vascular disorders	2 (1)	182 (4)	1 (1)	183 (4)
Flushing	2 (1)	165 (3)	1 (1)	166 (3)
Skin and subcutaneous tissue disorders	5 (2)	117 (2)	6 (6)	116 (2)
Pruritis	2 (1)	23 (0.5)	1 (1)	24 (0.5)
Rash	0 (0)	28 (1)	4 (4)	24 (0.5)
Infections and infestations	0 (0)	41 (1)	3 (3)	38 (1)
Investigations	6 (3)	118 (2)	3 (3)	121 (2)
Lymphocyte count decreased	4 (2)	69 (1)	2 (2)	71 (1)
White blood cell count decreased	2 (1)	8 (0.2)	0 (0)	10 (0.2)
Respiratory, thoracic, and mediastinal disorders	1 (0.5)	23 (0.5)	2 (2)	22 (0.4)
Dyspnea	0 (0)	10 (0.2)	2 (2)	8 (0.2)
Nervous system disorders	7 (3)	117 (2)	4 (4)	120 (2)
MS relapse	2 (1)	23 (0.5)	0 (0)	25 (0.5)
Headache	1 (0.5)	20 (0.4)	2 (2)	19 (0.4)
Blood and lymphatic system disorders	4 (2)	191 (4)	3 (3)	192 (4)
Lymphopenia	3 (1)	170 (3)	1 (1)	172 (3)
Injury, poisoning, and procedural complications	2 (1)	7 (0.1)	0 (0)	9 (0.2)
General disorders and administration site conditions	3 (1)	79 (2)	2 (2)	80 (2)
Asthenia	2 (1)	10 (0.2)	1 (1)	11 (0.2)
Fatigue	1 (0.5)	30 (1)	2 (2)	29 (1)

AA = African American; AE = adverse event; DMF = delayed-release dimethyl fumarate; GI = gastrointestinal; LA = Latino American; MS = mutiple sclerosis
[a]AEs coded using Medical Dictionary for Regulatory Activities (MedDRA), System Organ Class (SOC), and Preferred Term (PT).
Percentages calculated based on total number of patients. A patient was counted only once in each MedDRA, SOC, or PT. AEs reported in > 1 patient in either the Black/AA or Hispanic/LA subgroups are listed.

Table 4. Median Percentage Change in ALC Over Time

	Median Percentage Change			
Timepoint	Black/AA	Non-Black/Non-AA	Hispanic/LA	Non-Hispanic/ Non-LA
From Baseline to Month 12	–24 (n = 61)	–36 (n = 2037)	–34 (n = 27)	–35 (n = 2071)
From Baseline to Month 24	–27 (n = 43)	–35 (n = 1575)	–34 (n = 18)	–35 (n = 1600)
From Baseline to Month 36	–20 (n = 21)	–37 (n = 1064)	–39 (n = 12)	–36 (n = 1073)
From Baseline to Month 48	–17 (n = 14)	–35 (n = 631)	–36 (n = 7)	–34 (n = 638)
From Baseline to Month 60	–20 (n = 6)	–36 (n = 334)	--[a]	–36 (n = 338)

AA = African American; ALC = absolute lymphocyte count; LA = Latino American
n = number of patients with baseline ALC assessment and an ALC assessment at the specified follow-up visit. [a]Not listed because only 2 patients had data points at both baseline and Month 60.

Figure 1. ARR in the 12 Months Before and up to 5 Years After DMF Initiation

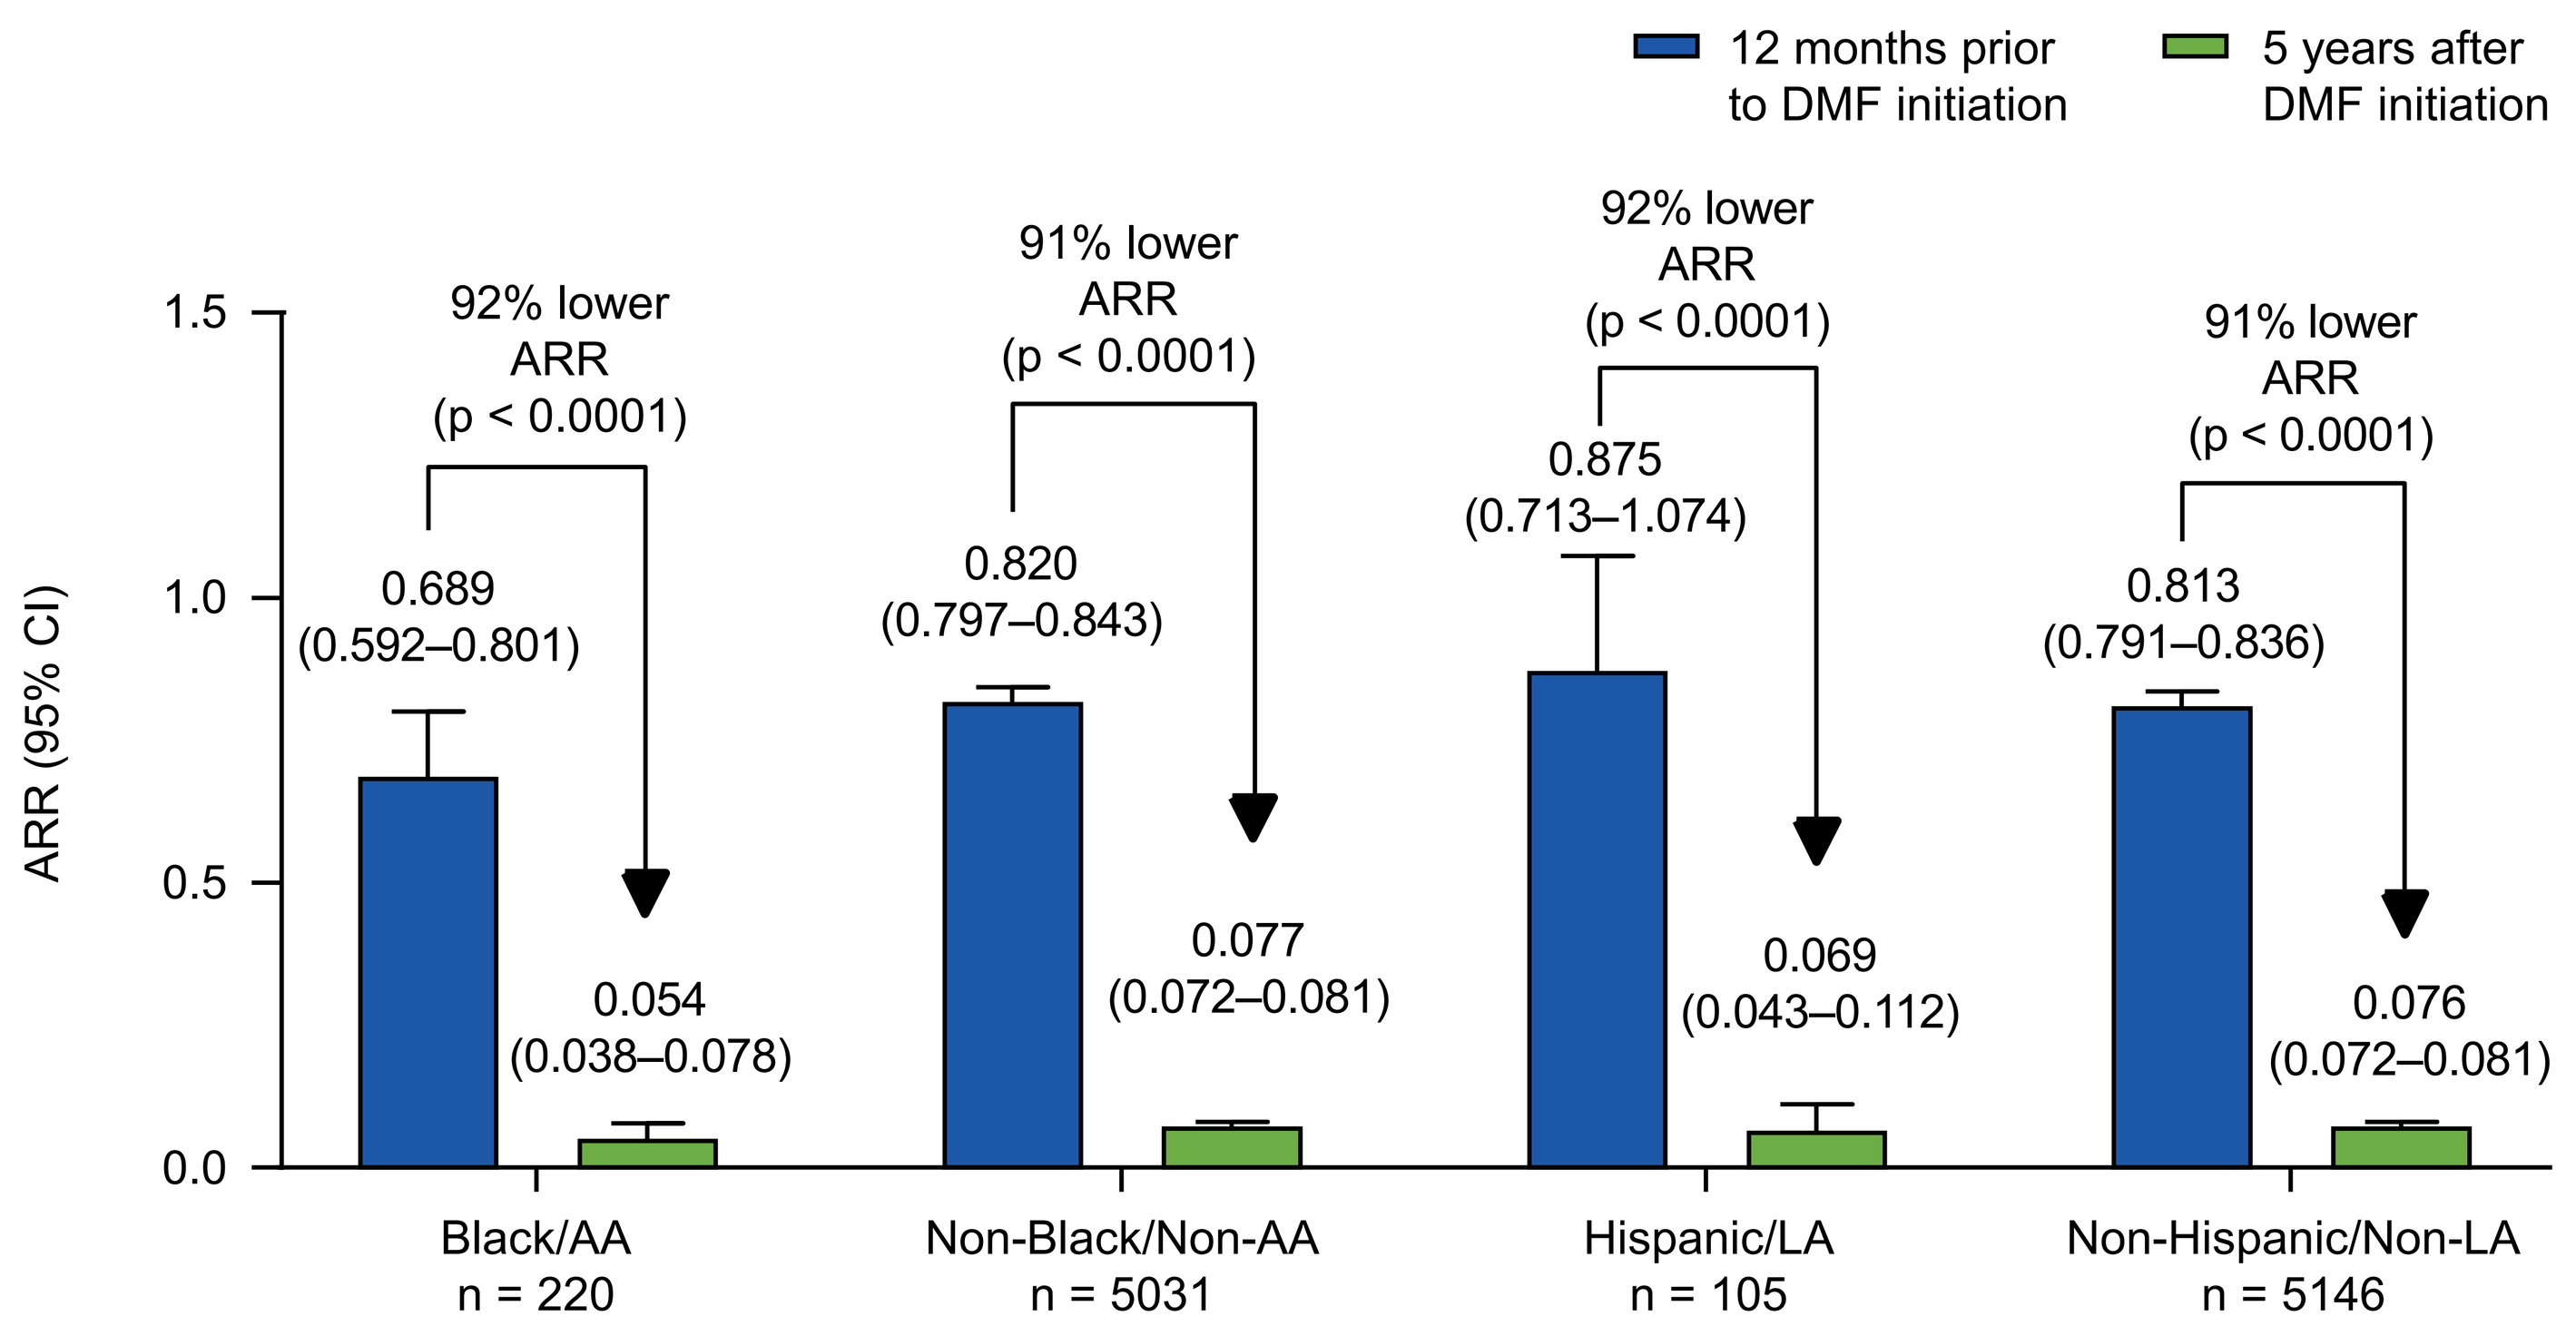

12 months prior to DMF initiation
5 years after DMF initiation
ARR (95% CI)
1.5
1.0
0.5
0.0
92% lower ARR (p < 0.0001)
0.689 (0.592–0.801)
0.054 (0.038–0.078)
91% lower ARR (p < 0.0001)
0.820 (0.797–0.843)
0.077 (0.072–0.081)
92% lower ARR (p < 0.0001)
0.875 (0.713–1.074)
0.069 (0.043–0.112)
91% lower ARR (p < 0.0001)
0.813 (0.791–0.836)
0.076 (0.072–0.081)
Black/AA n = 220
Non-Black/Non-AA n = 5031
Hispanic/LA n = 105
Non-Hispanic/Non-LA n = 5146

AA = African American; ARR = annualized relapse rate; DMF = delayed-release dimethyl fumarate; LA = Latino American

Figure 2. Median Lymphocytes Over Time

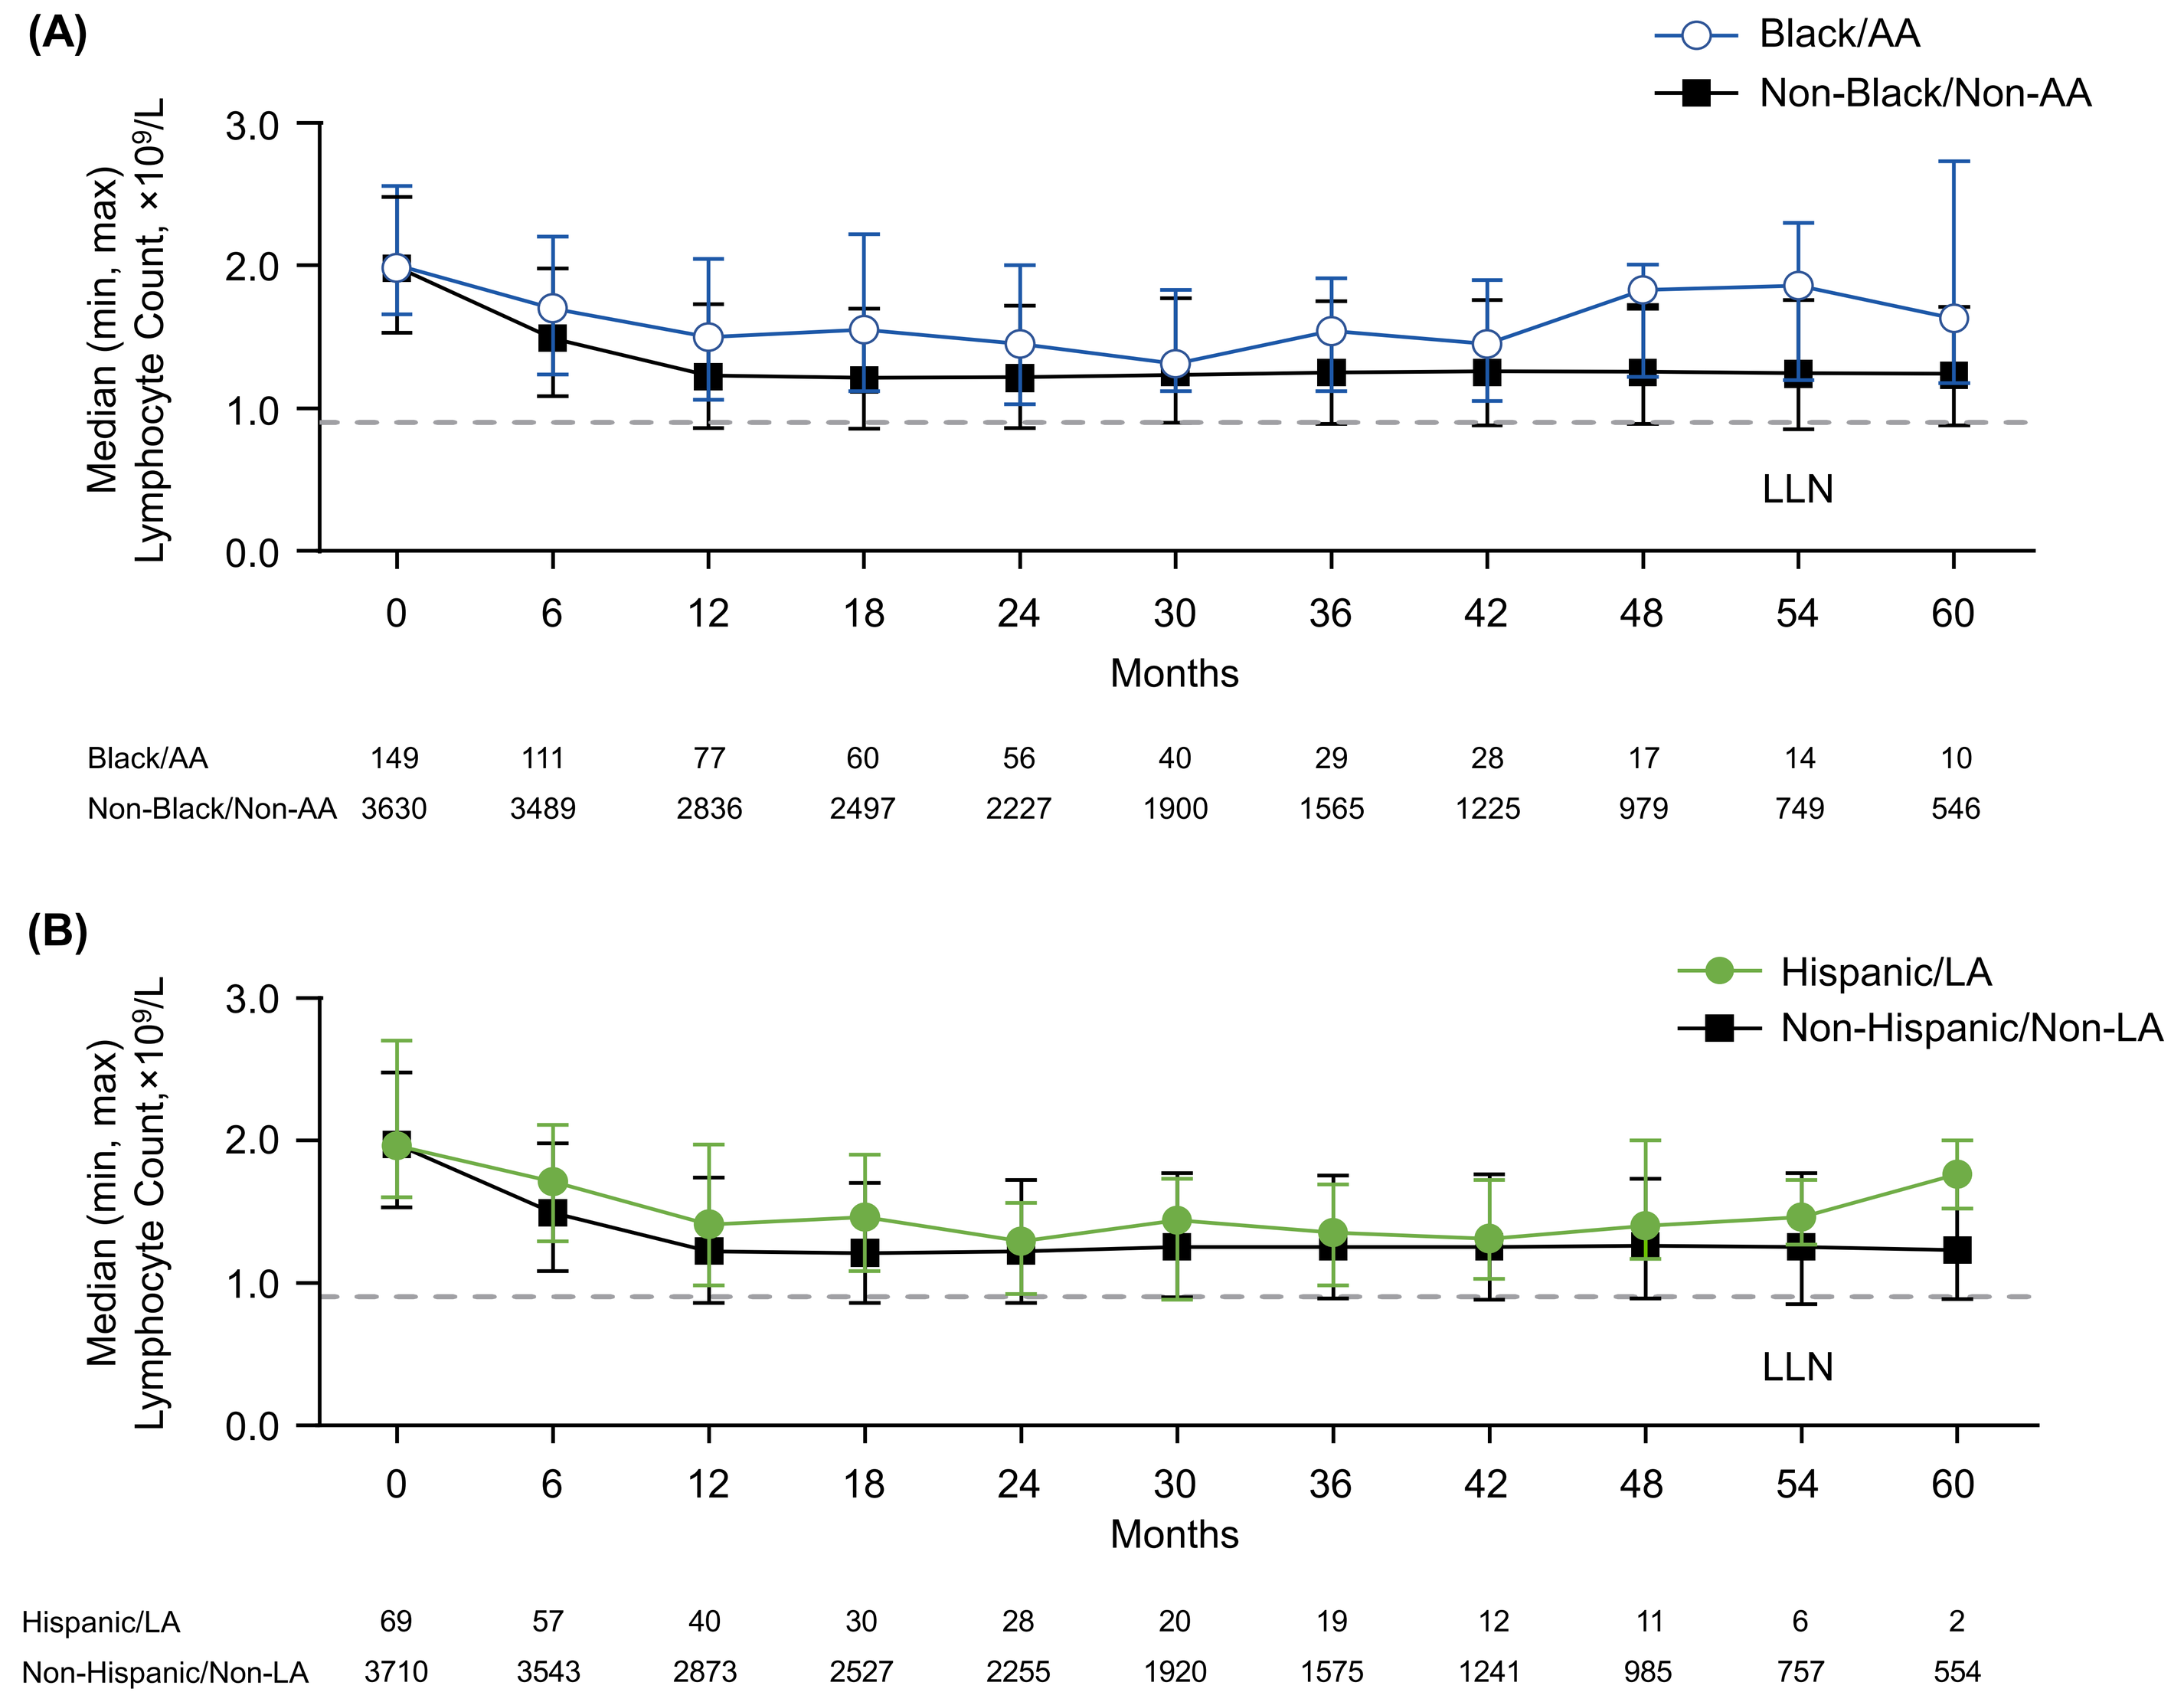

(A)
Black/AA
Non-Black/Non-AA
Median (min, max) Lymphocyte Count, ×10⁹/L
3.0
2.0
1.0
0.0
LLN
0 6 12 18 24 30 36 42 48 54 60
Months
Black/AA 149 111 77 60 56 40 29 28 17 14 10
Non-Black/Non-AA 3630 3489 2836 2497 2227 1900 1565 1225 979 749 546
(B)
Hispanic/LA
Non-Hispanic/Non-LA
Median (min, max) Lymphocyte Count,×10⁹/L
3.0
2.0
1.0
0.0
LLN
0 6 12 18 24 30 36 42 48 54 60
Months
Hispanic/LA 69 57 40 30 28 20 19 12 11 6 2
Non-Hispanic/Non-LA 3710 3543 2873 2527 2255 1920 1575 1241 985 757 554

AA = African American; LA = Latino American; LNN = lower limit of normal
n = number of patients with absolute lymphocyte count assessment at the specified timepoint.

References 1. Fox RJ, et al; CONFIRM Study Investigators. *N Engl J Med.* 2012;367(12):1087-1097. 2. Gold R, et al; DEFINE Study Investigators. *N Engl J Med.* 2012;367(12):1098-1107. 3. Kister I, et al. *Neurology.* 2010;75(3):217-223. 4. Langer-Gould A, et al. *Neurology.* 2013;80(19):1734-1739. 5. Clinical Research Pathways. https://clinicalresearchpathways.org/diversity/diversity-statistics-infographic. Accessed April 19, 2022. 6. Fox RJ, et al. *Neurol Ther.* 2017;6(2):175-187. 7. Zhovtis Ryerson L, et al. *Ther Adv Neurol Disord.* 2016;9(6):454-461. 8. Chinea A, et al. *Neurol Ther.* 2020;9(2):495-504. 9. Williams MJ, et al. *Neurol Ther.* 2020;9(2):483-493.

Disclosures MJW: advisory/consulting fees from Biogen, Celgene, EMD Serono, Genentech, Sanofi Genzyme, and Teva Neuroscience; speaker feesfrom Biogen, Genentech, Sanofi Genzyme, and Teva; LA: advisory/consulting fees from EMD Serono, Genzyme, and Novartis; research support from Biogen and MedDay; AC: advisory/consulting fees from Biogen, Genentech, Novartis, Sanofi Genzyme, and Teva Neuroscience; research support from Biogen, Novartis, and Sanofi Genzyme; speaker fees from Biogen, Genentech, Novartis, Sanofi Genzyme, and Teva; SC: advisory/consulting fees from Biogen, Celgene, Novartis, Pear Therapeutics, Roche Genentech, Sage Therapeutics, and Sanofi Genzyme; research support from AbbVie, Adamas, Alithios, Biogen, EMD Serono, MedDay, Novartis, Roche Genentech, and Sanofi Genzyme; speaker fees from Biogen, Roche Genentech, and Sanofi Genzyme; AO: advisory/consulting fees from Biogen, Celgene, EMD Serono, Genentech, Novartis, and Sanofi Genzyme; research support from Biogen, Novartis, Sanofi Genzyme, and TG Therapeutics; speaker fees from Biogen, Genentech, Novartis, Sanofi Genzyme, and Teva; DTO: personal compensation for consulting and advisory services from Biogen, Celgene/Bristol Myers Squibb, EMD Serono, Genentech, Genzyme, Janssen Pharmaceuticals, Novartis, Osmotica Pharmaceuticals, RVL Pharmaceuticals, TG Therapeutics, and Viela Bio; research support from Biogen and EMD Serono/Merck; royalties received for intellectual property licensed by The Board of Regents of The University of Texas System; WV: advisory/consulting fees from Alexion, Biogen, Genentech, and Octapharma; research support from Teva; NB, IB, XJ, JBL, JL, CS, SME, and NG: employees of and hold stock/stock options in Biogen. Acknowledgments This study was sponsored by Biogen (Cambridge, MA, USA). Writing and editorial support for the preparation of this poster was provided by Excel Scientific Solutions (Fairfield, CT, USA): funding was provided by Biogen.